Healthcare resource utilization and health-related quality of life in patients with resectable non-metastatic non-small cell lung cancer treated with neoadjuvant chemotherapy in China: a real-world survey

RWD89

[1]Bailey H, [2]Wang X, [1]Burlison H, [1]Forshaw C, [3]Wang P, [4]Lucherini S

[1]Adelphi Real World, Bollington, UK; [2]Bristol-Myers Squibb, Shanghai, China; [3]Fudan University, Shanghai, China; [4]Bristol-Myers Squibb, Middlesex, UK

Background

- With over 2.4 million new cases diagnosed every year, lung cancer accounts for around 12.4% of all new cancer diagnoses[1], with non-small cell lung cancer (NSCLC) accounting for 80-85% of these cases[2].
- In China, lung cancer accounts for 22% of all new cancer diagnoses[3] and is the leading cause of cancer-related deaths[3,4]. As one of the most populous countries in the world, distributing resources and managing the health of its population represents a significant challenge.
- Depending on many factors, including but not limited to the stage, genetics, and condition of the patient, standard of care for patients with resectable NSCLC is typically surgical resection with curative intent accompanied by either neoadjuvant or adjuvant chemotherapy[5].
- Specifically, neoadjuvant chemotherapy has demonstrated benefits such as a 10% reduction in distant metastases at five years and 5% absolute improvement in 5-year overall survival[6].
- The Phase 3 CheckMate 816 trial, approved in the US in March 2022, has shown statistically significant and clinically meaningful improvement in pathologic complete response and event-free survival with neoadjuvant immunotherapy (NIVO) + chemotherapy versus chemotherapy alone and has become the standard of care for patients with resectable NSCLC[7].
- It is estimated that nearly 0.65% of the total annual health expenditure in China is spent on lung cancer treatments annually, totalling around $5,360 per patient[8].
- With the rise of new therapeutic options for resectable NSCLC, it is important to establish of a real-world 'baseline' against which future changes in patient management can be compared, allowing accurate measurement of the effect of the introduction of new treatment approaches in China.

Scope

- This study aims to describe the real-world characteristics, healthcare resource utilization (HCRU) and health-related quality of life (HRQoL) in patients receiving, or those who have received neoadjuvant chemotherapy for resectable non-metastatic non-small cell lung cancer (nmNSCLC) in China.

Methods

- Data were drawn from the Adelphi nmNSCLC Disease Specific Programme™ (DSP), a cross-sectional survey of physicians in China from September – November 2022.
- Physicians provided information on their next four consulting patients with resectable nmNSCLC and an additional four patients who were specifically receiving/had received any neoadjuvant and/or adjuvant treatment
- Only patients who were receiving/had received neoadjuvant treatment were included in this analysis.
- Physicians completed patient record forms (PRFs) detailing data on patients' demographics, clinical characteristics, treatment history, and HCRU.
- The same patients were invited to complete a voluntary questionnaire capturing their perspective on their nmNSCLC, using the EQ-5D-5L, the NSCLC-Symptom Assessment Questionnaire (NSCLC-SAQ) and the Work Productivity and Activity impairment questionnaire General Health version (WPAI-GH).
- The EQ-5D-5L consists of a descriptive system and a Visual Analogue Scale (VAS). The descriptive system consists of five dimensions: mobility, self-care, usual activities, pain/discomfort and anxiety/depression and is scored on five levels from no problems to extreme problems.
 - Utility scores range from 1 (indicative of full health) to below 0 (indicative of a health state equivalent to death), with higher scores representing better HRQoL.
 - The VAS records the patients self-rated health on a visual scale ranging from 0 to 100; higher scores indicate better health status/quality of life.
- The NSCLC-SAQ assesses five NSCLC symptoms: cough, pain, dyspnea, fatigue, and poor appetite, in the past seven days.
 - Symptom intensity is measured on a scoring scale of 0 – 4: ranging from 0 "No symptom at all" to 4 "Very severe symptom".
 - Symptom frequency is measured on a scoring scale of 0 – 4: ranging from 0 "Never" to 4 "Always".
 - A total score out of 20, by combining their 5 scores from the symptoms assessment, is then calculated.
- The WPAI-GH assesses the impact of disease on work productivity and daily activities, including absenteeism, presenteeism and activity impairments, over the past 7 days.
 - Scores reflect impairment and productivity loss, where higher percentages indicate greater impairment and lower productivity.

Table 1. Patient/physician inclusion criteria

Physician Inclusion Criteria	Patient Inclusion Criteria
1. The physician must be a medical oncologist, pulmonologist or respiratory surgeon. 2. The physician must see in a typical month: • At least two patients with nmNSCLC • One nmNSCLC patient who were receiving/had received neoadjuvant treatment • One nmNSCLC patient who were receiving/had received adjuvant treatment 3. The physician must be personally responsible for prescribing decisions for patients with nmNSCLC.	1. The patient must be diagnosed with stage I/II/III NSCLC at time of data collection. 2. The patient must be ≥18 years old. 3. The patient must not be involved in a clinical trial. 4. Additionally for the neoadjuvant/adjuvant cohort: The patient must have received/be currently receiving neoadjuvant treatment or adjuvant treatment for their nmNSCLC.

Results

Neoadjuvant treatments

- A total of n=50 physicians reported on a total of 170 patients receiving neoadjuvant chemotherapy.
- Out of all the patients who received neoadjuvant treatment in China (n=170), 53% were receiving/had received chemotherapy only and 24% were receiving/had received chemotherapy in combination with a non-chemotherapy neoadjuvant agent (of which 73% received immunotherapy, n=30). The remaining 23% were receiving/had received any other non-chemotherapy neoadjuvant treatment (Table 2).

Table 2. Treatment groups

All neoadjuvant patients	n = 170
Patient who were receiving/had received neoadjuvant chemotherapy, n (%)	131 (77)
Patients who were receiving/had received neoadjuvant chemotherapy only (either alone or in combination), n (%)	90 (53)
Patients who were receiving/had receiving/had received neoadjuvant chemotherapy in combination with a non-chemotherapy neoadjuvant treatment, n (%)	41 (24)
Patient who were receiving/had received any other non-chemotherapy neoadjuvant treatment, n (%)	39 (23)

Demographics and Clinical Characteristics

- Over half of patients who were receiving/had received neoadjuvant chemotherapy (n=131) were male (63%). The mean (standard deviation; SD) age of patients was 56.3 (10.5) years. Most patients had a squamous cell histology (50%) and the highest proportion of patients had a TNM stage of IA-IB at the time of data collection (44%) and IIIA-IIIC at their initial nmNSCLC diagnosis (47%). Most patients (92%) had and an ECOG score of 0-1 at both their initial diagnosis and at the time of data collection, whereas in patients receiving non-chemotherapy neoadjuvant treatment (51%), the majority had an ECOG score of 2+ (Table 3).

Table 3. Demographics and Clinical Characteristics

	All neoadjuvant patients	Patients who were receiving/had received neoadjuvant chemotherapy	Patients who were receiving/had received any other non-chemotherapy neoadjuvant treatment
Age, mean (SD), years	n=170	n=131	n=39
	56.7 (10.8)	56.3 (10.5)	57.8 (11.7)
Male, n (%)	n=170	n=131	n=39
	105 (62)	83 (63)	22 (56)
TNM staging at time of data collection (8th Edition), n (%)	n=170	n=131	n=39
IA-IB	65 (38)	57 (44)	8 (21)
IIA-IIB	49 (29)	34 (26)	15 (38)
IIIA-IIIC	45 (26)	34 (26)	11 (28)
Unknown	11 (6)	6 (5)	5 (13)
TNM staging at initial nmNSCLC diagnosis (8th Edition), n (%)	n=170	n=131	n=39
IA-IB	27 (16)	25 (19)	2 (5)
IIA-IIB	62 (36)	44 (34)	18 (46)
IIIA-IIIC	80 (47)	62 (47)	18 (46)
Unknown	1 (1)	0 (0)	1 (3)
Histology, n (%)	n=170	n=131	n=39
Squamous cell	78 (46)	65 (50)	13 (33)
Adenocarcinoma	85 (50)	63 (48)	22 (56)
Large cell carcinoma	5 (3)	1 (1)	4 (10)
Other	2 (1)	2 (2)	0 (0)
ECOG performance status at initial nmNSCLC diagnosis, n (%)	n=170	n=131	n=39
0 – 1	139 (82)	120 (92)	19 (49)
2+	30 (18)	10 (8)	20 (51)
Unknown / not assessed	1 (1)	1 (1)	0 (0)
Family history of lung cancer, n (%)	n=170	n=131	n=39
Yes	23 (14)	18 (14)	5 (13)
No	142 (84)	108 (82)	34 (87)
Don't know	5 (3)	5 (4)	0 (0)

Caregiver history

- Of patients who were receiving/had received neoadjuvant chemotherapy, 53% had a caregiver, which was a partner in 93% of cases. An average of 51.4 hours per week were spent by caregivers providing assistance for those patients who were receiving/had received neoadjuvant chemotherapy, compared to 24.6 hours by those patients who were receiving/had received non-chemotherapy neoadjuvant treatment (Table 4).

Table 4. Caregiver status

	All neoadjuvant patients	Patient who were receiving/had received neoadjuvant chemotherapy	Patient who were receiving/had received any other non-chemotherapy neoadjuvant treatment
Is anyone responsible for this patient's daily needs?, n (%)	n=170	n=131	n=39
Yes	114 (67)	91 (69)	23 (59)
No	45 (26)	34 (26)	11 (28)
Don't know	11 (6)	6 (5)	5 (13)
Relationship of the caregiver(s) to the patient, n (%)	n=88	n=70	n=18
Partner / spouse	71 (81)	65 (93)	6 (33)
Son / daughter over 18 years	26 (30)	20 (29)	6 (33)
Other relative(s)	20 (23)	20 (29)	0 (0)
Friend(s)	2 (2)	2 (3)	0 (0)
Don't know	10 (11)	3 (4)	7 (39)
Number of hours spent caregiving per week, mean (SD)	n=78	n=67	n=11
	47.6 (32.3)	51.4 (32.1)	24.6 (23.9)

Health-related Quality of Life – PRO tools

- Patients receiving neoadjuvant chemotherapy at the time of data collection (n=65) reported an EQ-5D-VAS of 60.6, compared to a population norm of 85.4 (Figure 1).
- Based on the EQ-5D-5L results, patients receiving neoadjuvant chemotherapy at the time of data collection reported moderate to extreme problems with mobility (18%), self-care (2%), doing usual activities (38%), pain/discomfort (14%), and anxiety/depression (32%).

Figure 1. Average EQ-5D-5L scores for patients who were receiving neoadjuvant chemotherapy at data collection

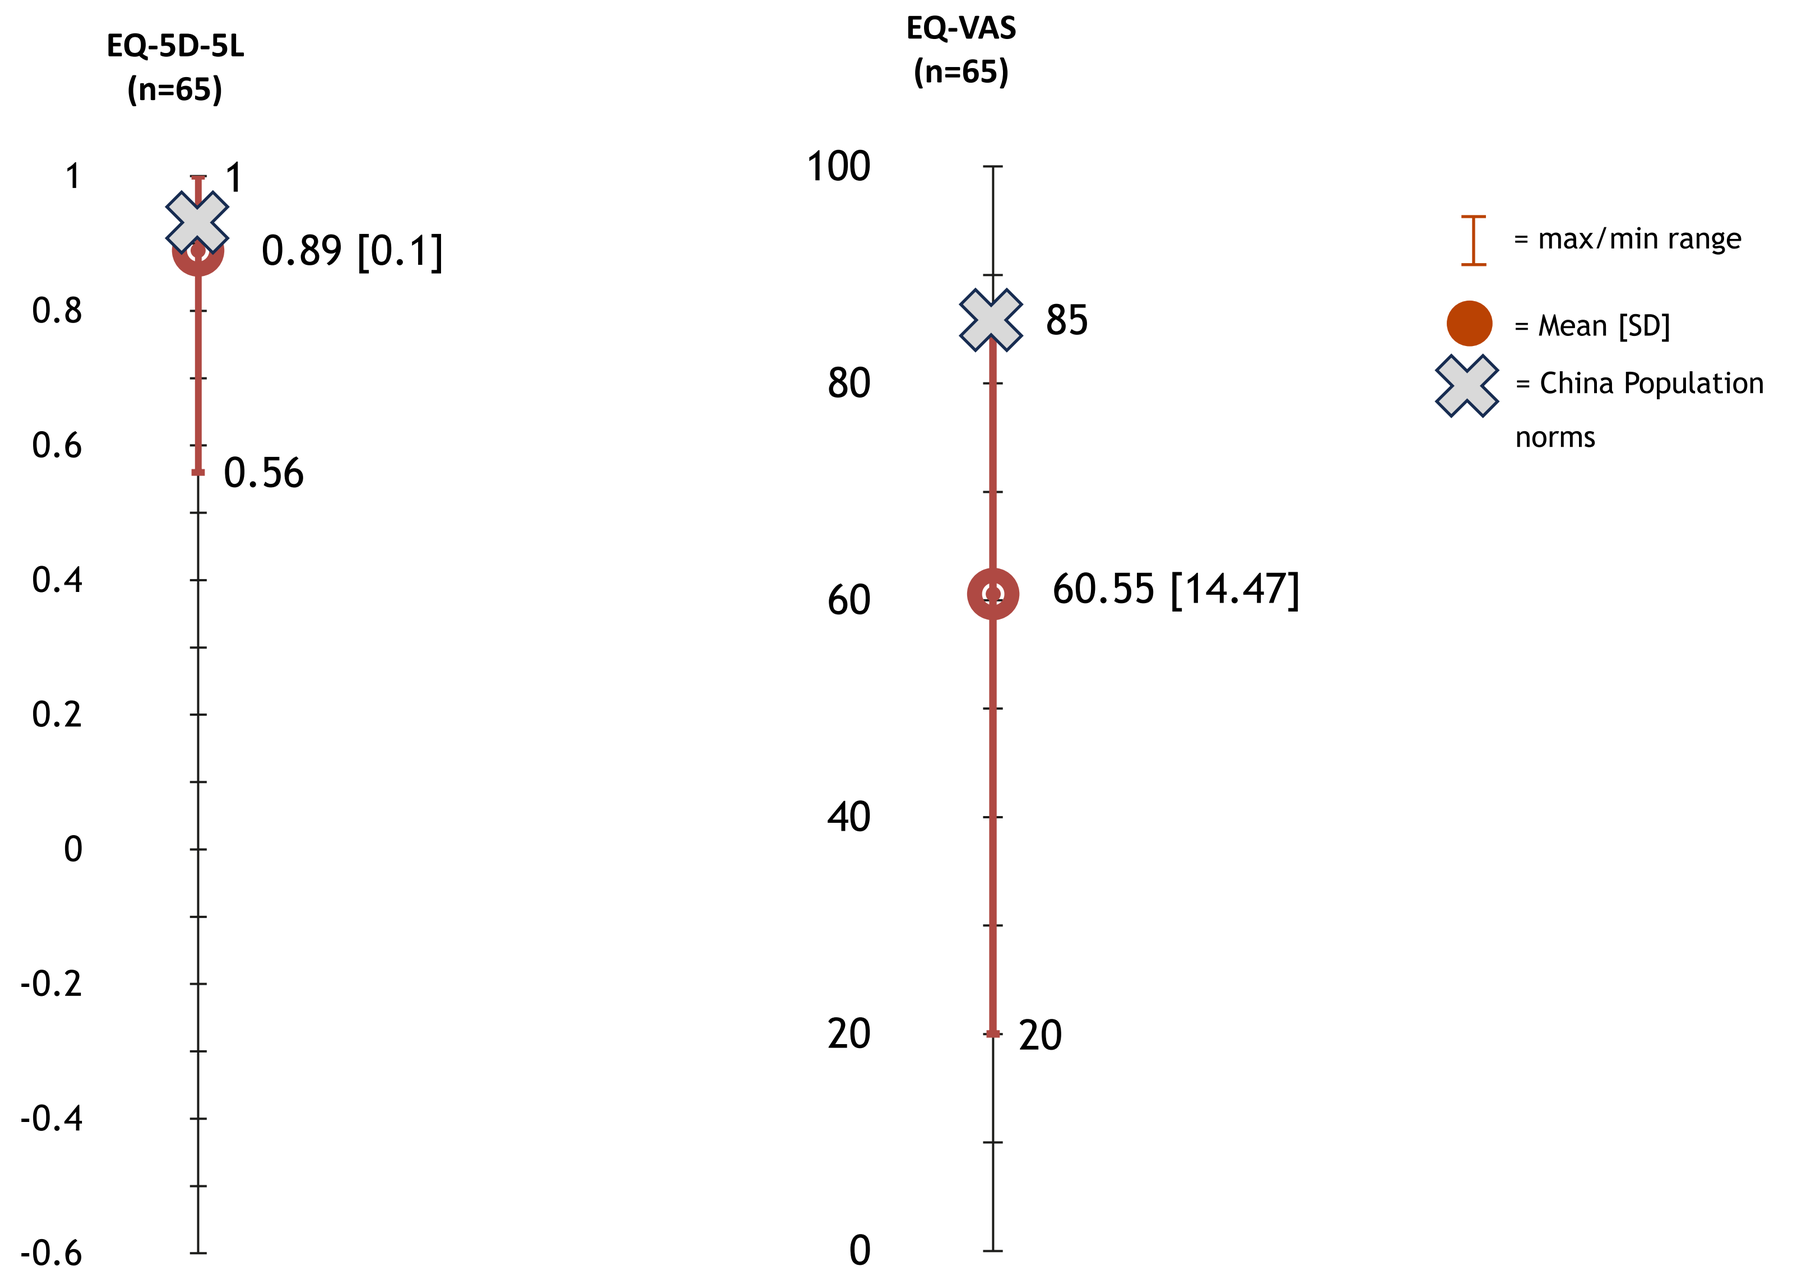
EQ-5D-5L (n=65)
1
0.89 [0.1]
0.56
EQ-VAS (n=65)
85
60.55 [14.47]
20
= max/min range
= Mean [SD]
= China Population norms

Health-related Quality of Life (Continued)

- Patients receiving neoadjuvant chemotherapy at the time of data collection had a NSCLC-SAQ cough domain score of 1.4 (0.9), a pain score of 1.0 (0.8), a dyspnea score of 1.8 (1.0), a fatigue score of 1.9 (1.0), and an appetite score of 2.0 (1.2). Patients appear to either rarely or sometimes experience coughing, pain in their chest and other areas, dyspnea, fatigue, and poor appetite. With a mean total score of 8.1 (3.2) out of the possible 20, suggesting slight overall symptomatology (Figure 2).

Figure 2. Average NSCLC-SAQ scores - Patients who were receiving neoadjuvant chemotherapy at the time of data collection

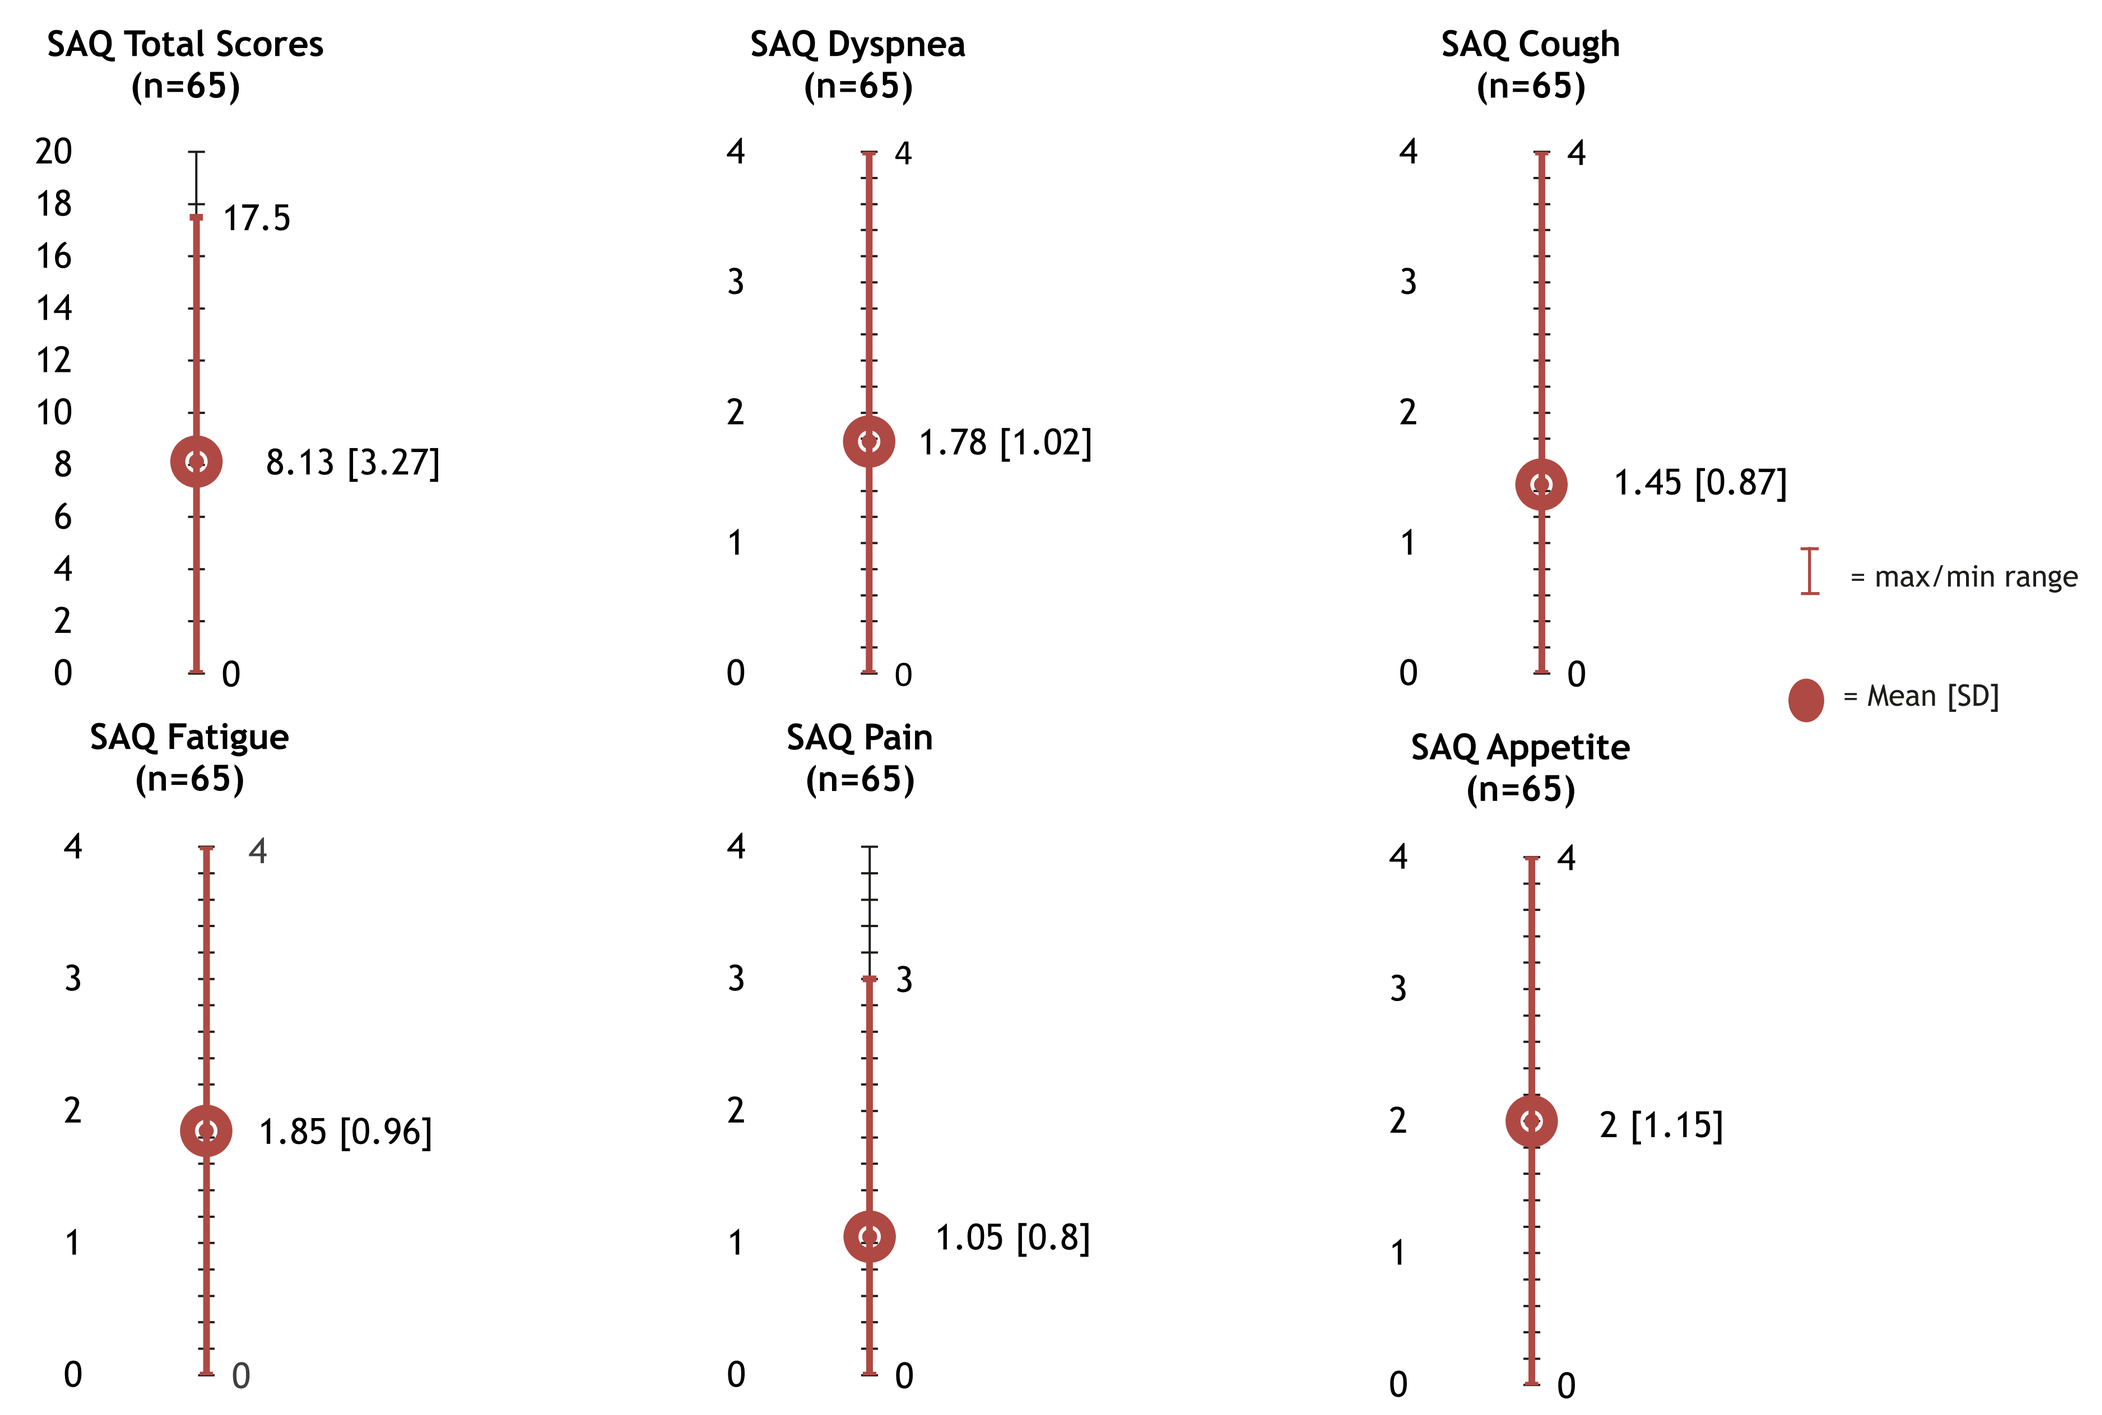
SAQ Total Scores (n=65)
17.5
8.13 [3.27]
SAQ Dyspnea (n=65)
1.78 [1.02]
SAQ Cough (n=65)
1.45 [0.87]
SAQ Fatigue (n=65)
1.85 [0.96]
SAQ Pain (n=65)
1.05 [0.8]
SAQ Appetite (n=65)
2 [1.15]
= max/min range
= Mean [SD]

- Out of the patients who were receiving/had received neoadjuvant chemotherapy and were employed at the time of data collection, patients (n=17) reported missing work in the past week due to their nmNSCLC, accounting for a mean (SD) of 60.47% (41.95) of their working time (absenteeism). While patients were working (n=9), 58.89% (17.64) of their work was impaired due to their nmNSCLC (presenteeism), with an overall work impairment of 68.36% (17.91) (n=9).
- Furthermore, 58.77% (19) of the patient's regular daily activities were impaired due to their nmNSCLC (activity impairment) (n=65) (Figure 3).

Figure 3. Average WPAI scores for patients who were receiving neoadjuvant chemotherapy at the time of data collection

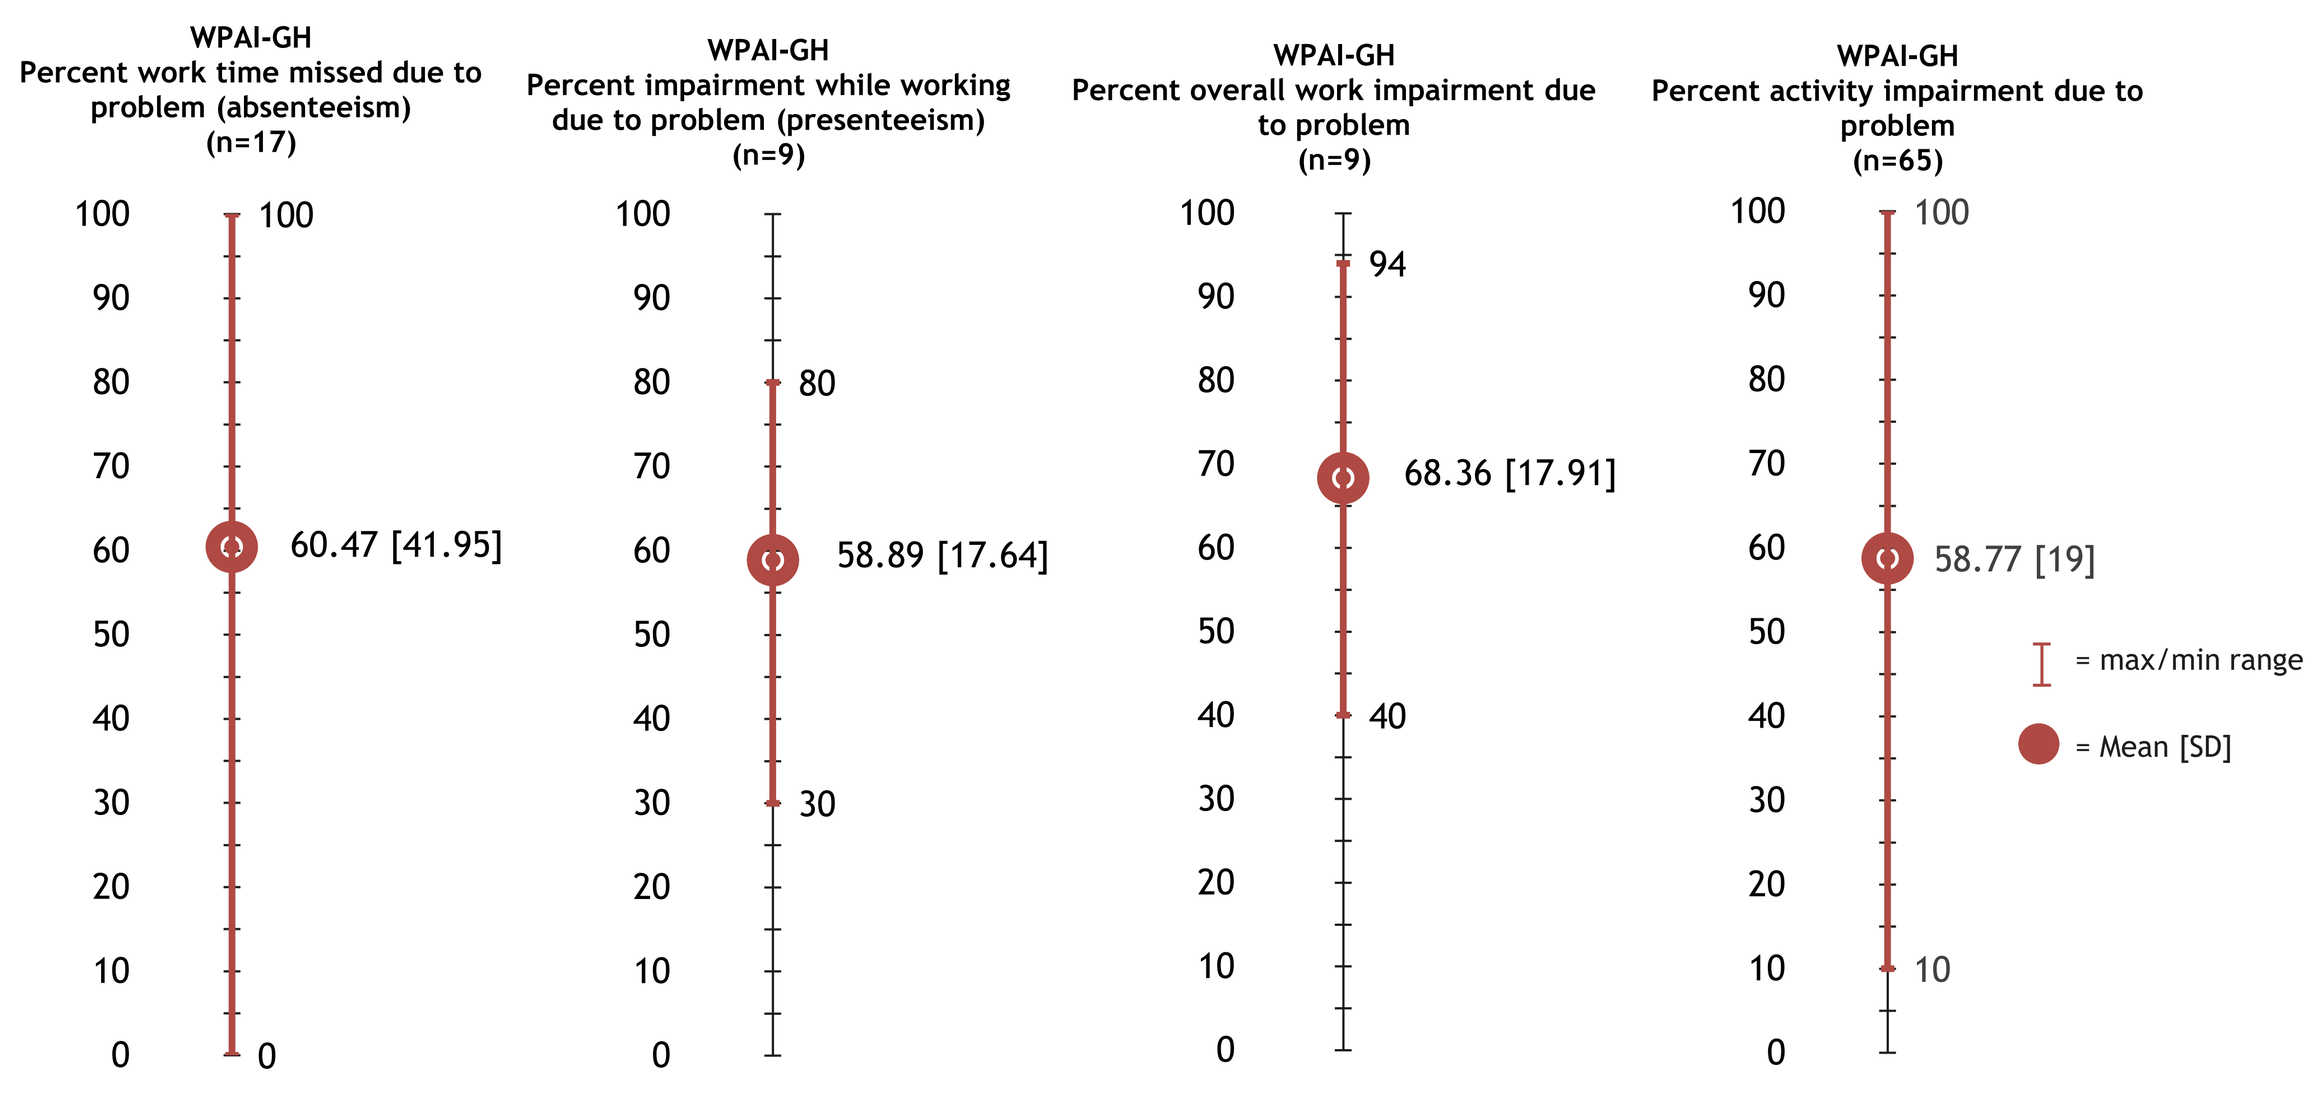
WPAI-GH Percent work time missed due to problem (absenteeism) (n=17)
60.47 [41.95]
WPAI-GH Percent impairment while working due to problem (presenteeism) (n=9)
58.89 [17.64]
WPAI-GH Percent overall work impairment due to problem (n=9)
68.36 [17.91]
WPAI-GH Percent activity impairment due to problem (n=65)
58.77 [19]
= max/min range
= Mean [SD]

Healthcare resource utilization

- Of n=106 patients who were receiving/had received neoadjuvant chemotherapy, 82% had at least one hospitalisation (including for surgery) in the 12 months prior to data collection, with a quarter (25%) having at least five hospitalisations (Figure 4a).
- Of patients who were receiving/had received neoadjuvant chemotherapy and were hospitalised (n=88), 6% patients were admitted to the ICU. Among patients who had to stay overnight as part of their hospitalisation (n=74), the average number of nights spent in total was 6.4 (3.2).
- The most frequently reported reasons for hospitalisation for patients who were receiving/had received neoadjuvant chemotherapy were for surgery ((20%), to treat a complication (11%), and for a biopsy (11%); Figure 4b).

Figure 4a. Number of hospitalisations in relation to nmNSCLC in the 12 months prior to data collection (including surgery)

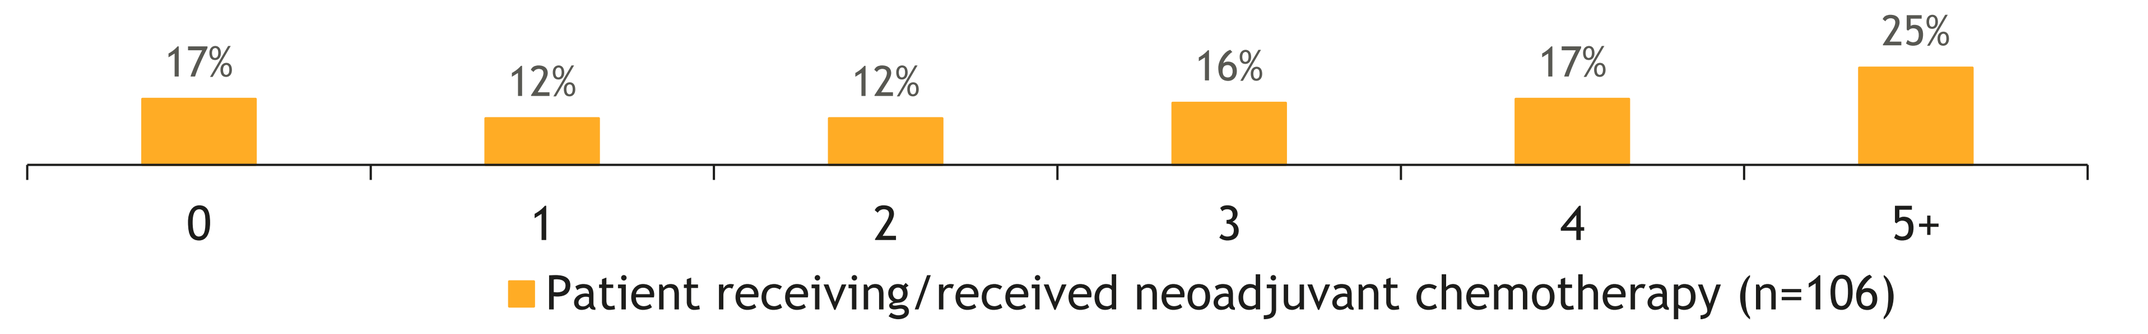
17%
12%
12%
16%
17%
25%
0
1
2
3
4
5+
Patient receiving/received neoadjuvant chemotherapy (n=106)

Figure 4b. Reason for most recent hospitalisation

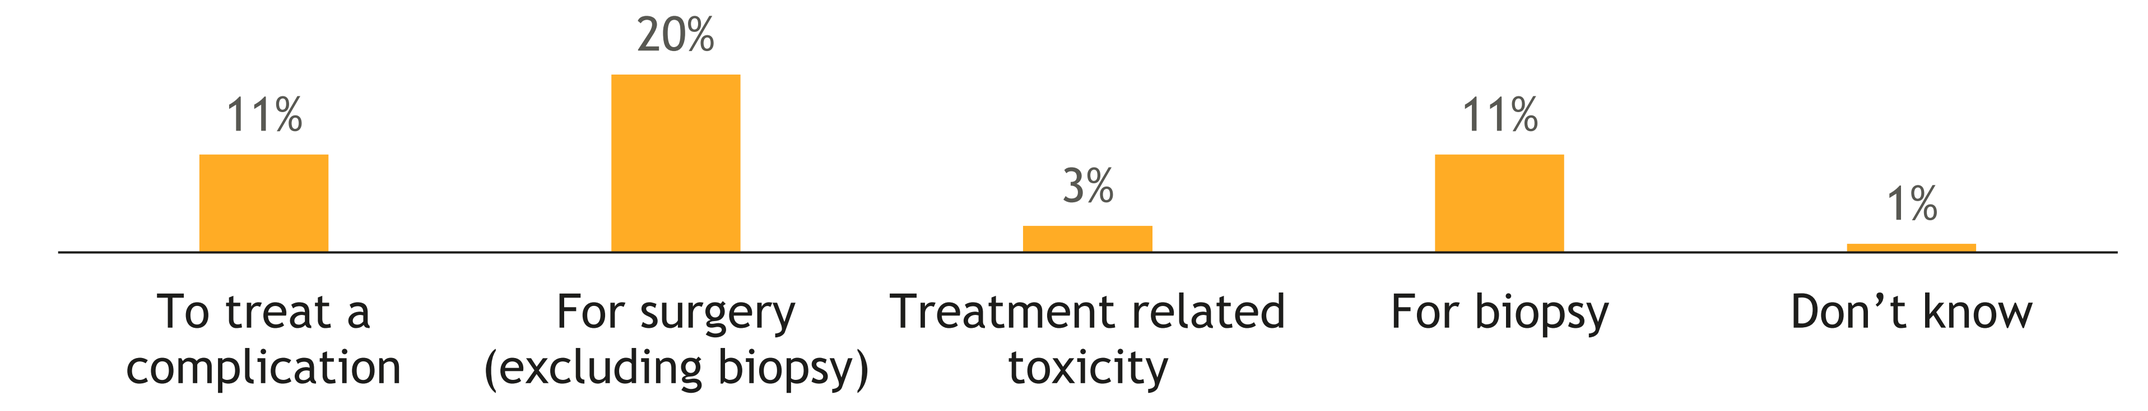
11%
20%
3%
11%
1%
To treat a complication
For surgery (excluding biopsy)
Treatment related toxicity
For biopsy
Don't know

n=46 (52%) patients reported having another, non-specified, reason for hospitalisation

Patient receiving/received neoadjuvant chemotherapy (n=88)

Healthcare resource utilization (Continued)

- Nearly half of patients who were receiving/had received neoadjuvant chemotherapy had either a pulmonologist (40%) or an oncologist (40%) involved in the ongoing management of their nmNSCLC.
- The most common tests used to aid diagnosis of patients who were receiving/had received neoadjuvant chemotherapy were a blood test (95%), CT scan (84%) or a pulmonary function test (82%). These were also the most common tests to monitor these patients' nmNSCLC (blood test: 89%, CT scan: 64%, pulmonary function test: 42%; Figure 5).
- In 12 months prior to data collection, patients who were receiving/had received neoadjuvant chemotherapy underwent a mean (SD) of 5.3 (4.8) blood tests, 2.6 (1.7) CT scans, and 2.1 (1.0) pulmonary tests.
- On average, patients receiving neoadjuvant chemotherapy at the time of data collection (n=65) were paying ¥3694.9 (approximately $510.72) per month for their lung cancer medicines and an extra ¥214.7 (approximately $29.68) per month for prescription medicines for any other conditions they may have.

Figure 5. Tests/assessments in relation to nmNSCLC - Patients who were receiving/had received neoadjuvant chemotherapy

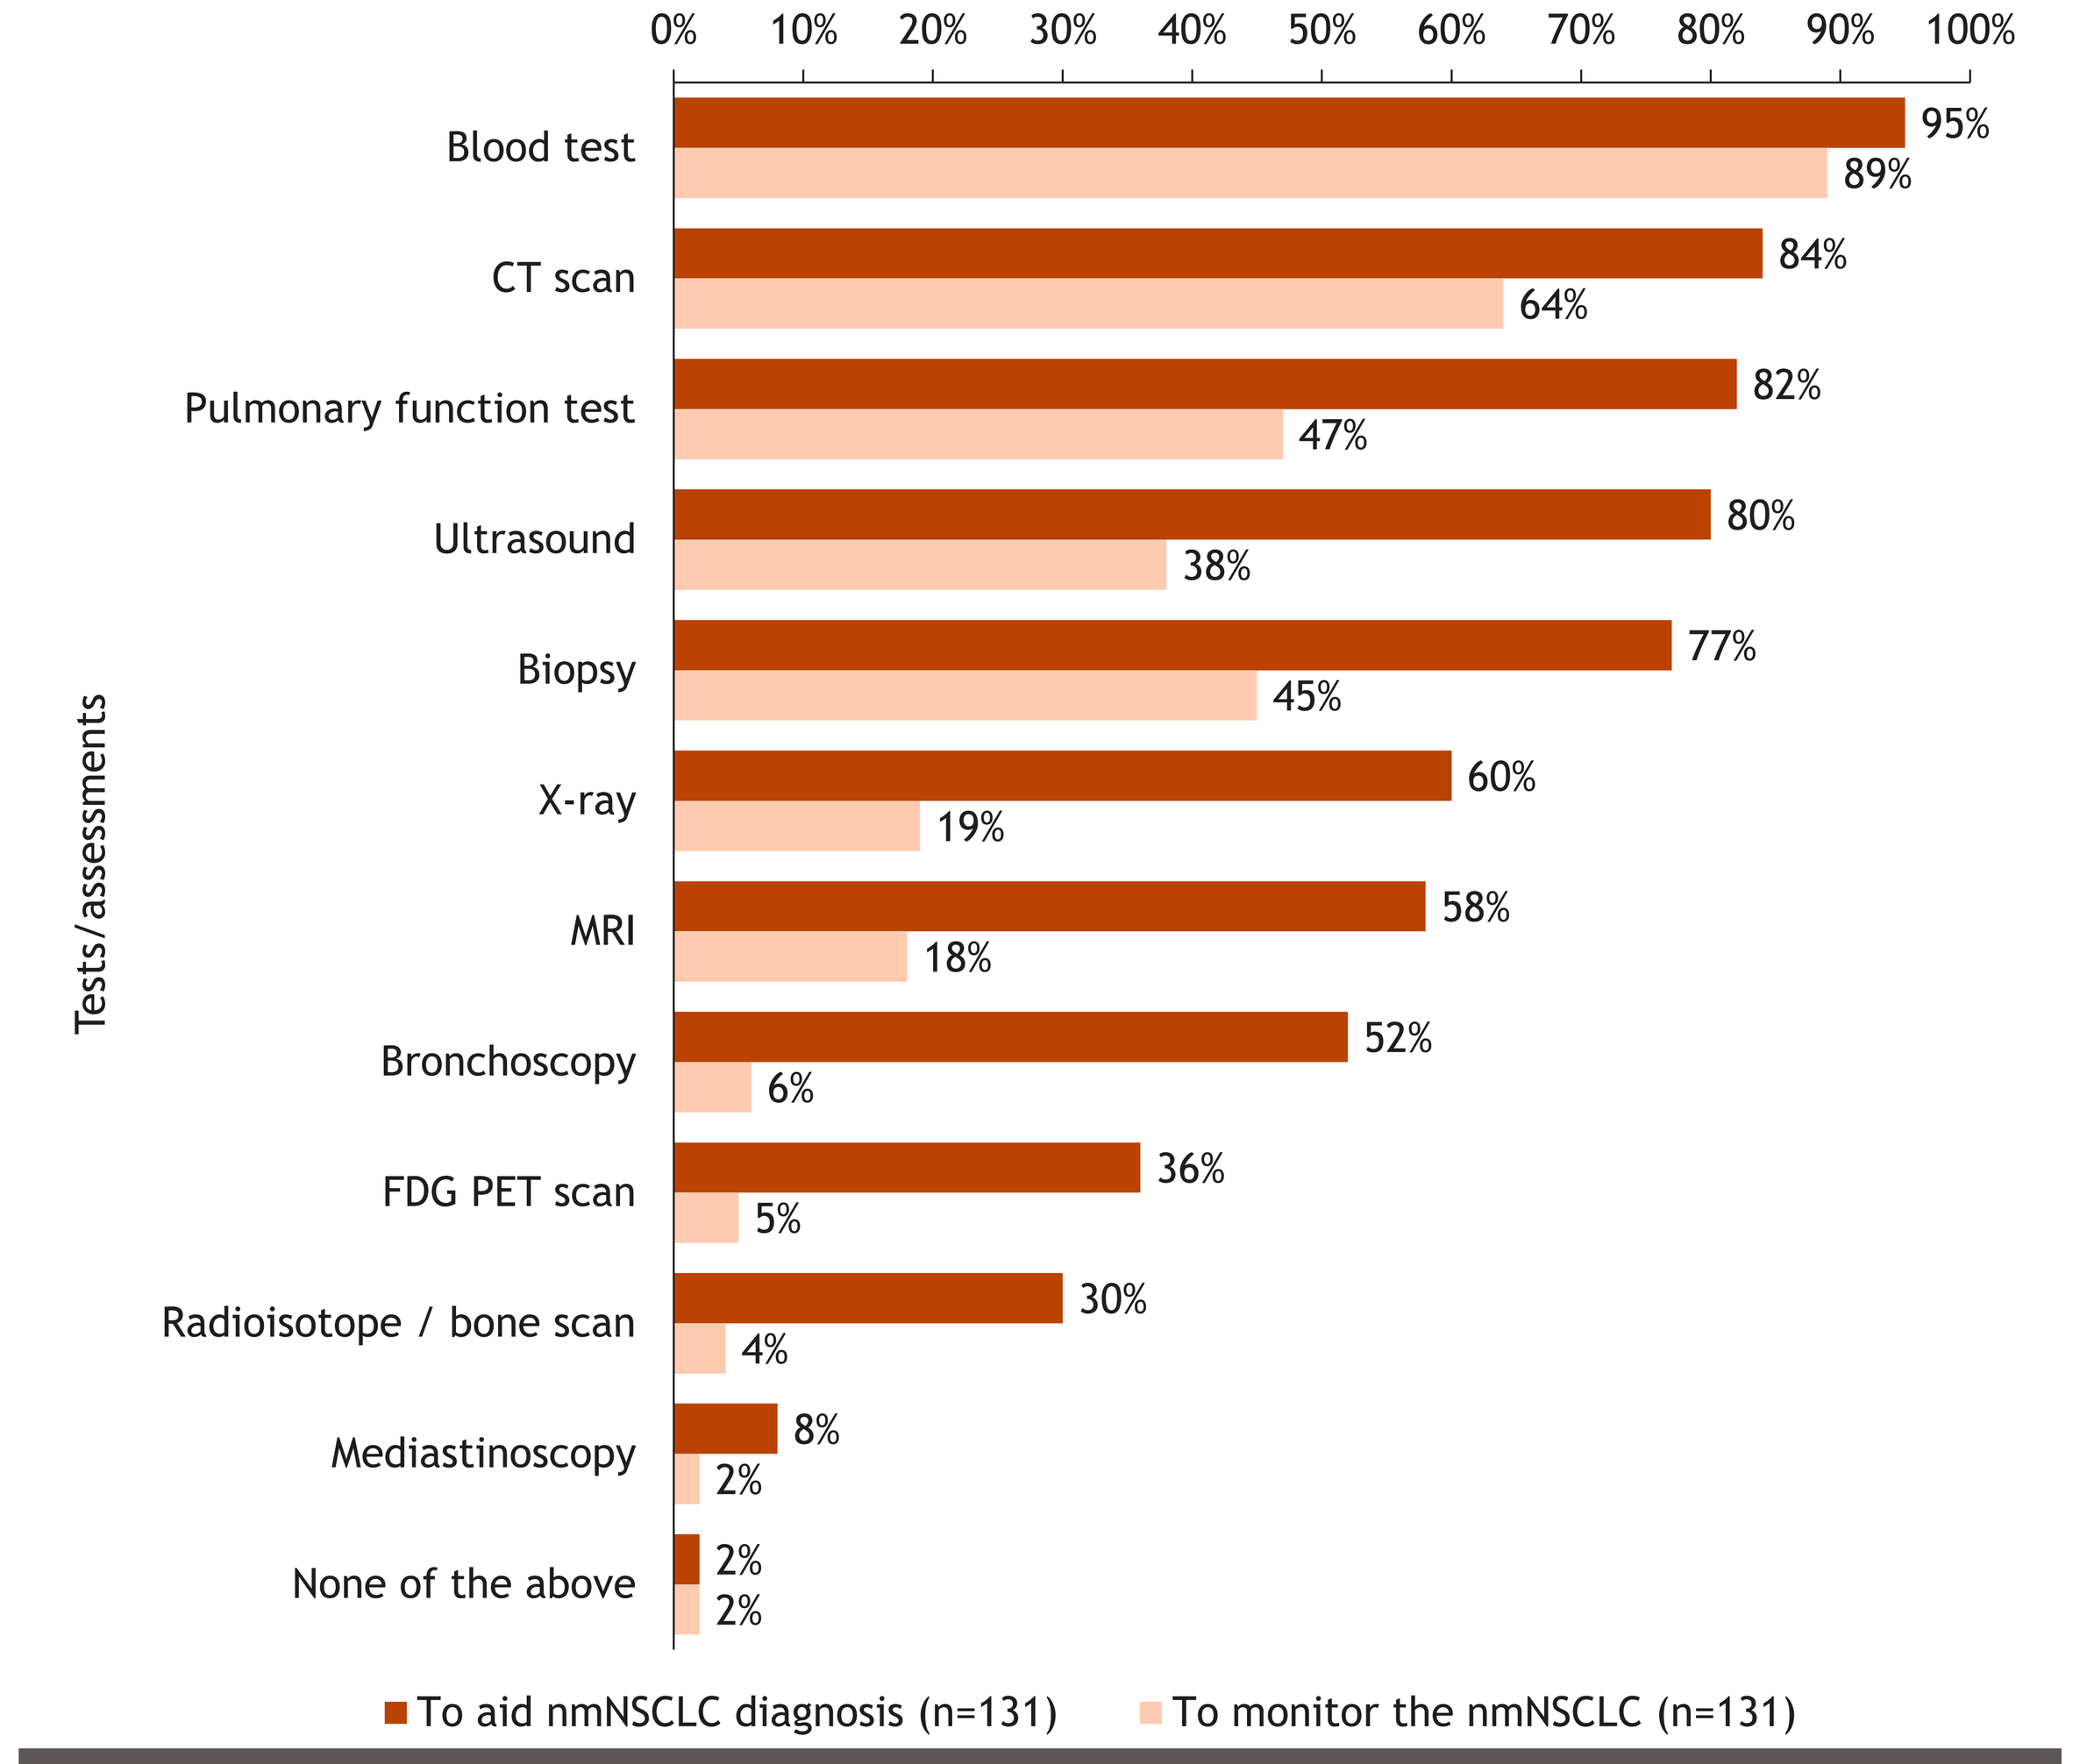
Tests/assessments
Blood test 95% 89%
CT scan 84% 64%
Pulmonary function test 82% 47%
Ultrasound 80% 38%
Biopsy 77% 45%
X-ray 60% 19%
MRI 58% 18%
Bronchoscopy 52% 6%
FDG PET scan 36% 5%
Radioisotope / bone scan 30% 4%
Mediastinoscopy 8% 2%
None of the above 2% 2%
To aid nmNSCLC diagnosis (n=131)
To monitor the nmNSCLC (n=131)

Limitations

- Participating patients may not reflect the general nmNSCLC population as patients who consult their physicians more frequently had a greater chance of inclusion.
- The DSP is not based on a true random sample of physicians or patients. While minimal inclusion criteria governed the selection of the participating physicians, participation is influenced by willingness to complete the survey.
- The cross-sectional design of this study prevents any conclusions about causal relationships, which highlights the needs for further study.

Conclusions

- Patients in China in 2022 treated with neoadjuvant chemotherapy for their resectable nmNSCLC displayed high rates of HCRU and reported an unmet need for pain, mobility and anxiety/depression. Given the advent of immunotherapy regimens in this setting; this could lead to a decrease in HCRU and patient burden, which warrants further investigation.
- With just under a third of patients who were receiving/had received neoadjuvant chemotherapy not working due to their nmNSCLC and over half of these patients receiving support from non-professional caregiver, further research is needed to investigate the effects of the economy of both the patients and their caregivers not working due to their nmNSCLC.

References

1. Sung, H., et al., 2021. CA: a cancer journal for clinicians, 71(3), pp.209-249.
2. Chen, P., et al., 2022. Cancer Communications, 42(10), pp.937-970.
3. Globocan 2022 (2024) 160-china-fact-sheet.pdf. Available at: https://gco.iarc.fr/today/data/factsheets/populations/900-world-fact-sheets.pdf (Accessed: 11 March 2024).
4. Gao, S., et al., 2020. Journal of Thoracic Oncology, 15(10), pp.1567-1576.
5. Alexander, M et al., 2020. Lung, 198, pp.897-907.
6. NSCLC Meta-analysis Collaborative Group, 2014. The Lancet, 383(9928), pp.1561-1571.
7. Forde, P.M., et al., 2022 New England Journal of Medicine, 386(21), pp.1973-1985.
8. Zhu, D., et al., 2021. Cancer Medicine, 10(8), pp.2914-2923.

Acknowledgments

- All authors contributed to analysis and interpretation of data and reviewed and approved this poster.
- A substantial contribution to the analysis and review of the poster was made by Digby Barrowcliff, Adelphi Real World.
- Hollie Bailey has no conflicts of interest to declare.

Declarations of interests

- Stefano Lucherini – Employee of Bristol Myers Squibb
- Xingzhi Wang – Employee of Bristol Myers Squibb
- Pei Wang – Associate Professor of Fudan University
- Hollie Bailey- Employee of Adelphi Real World
- Helen Burlison- Employee of Adelphi Real World
- Cameron Forshaw – Employee of Adelphi Real World

Presented at ISPOR 2024; May 5–8, 2024; Atlanta, GA

Email: Hollie Bailey; hollie.bailey@adelphigroup.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.