OP3

THE UPGRADE OF EDUCATIONAL SYSTEM FOR IMPROVING PRACTICAL SKILLS OF ECONOMICS GRADUATE STUDENTS IN HTA IN UKRAINE

Irynchyna I.[1], Piniazhko O.[2], Zaliska O.[3], Haidukov Y.[4], Khmelovska M.[2], Solodkovskyi Y.[1]

[1]*Kyiv National Economic University named after Vadym Hetman, Kyiv, Ukraine*
[2]*State Expert Center of the Ministry of Health of Ukraine, Kyiv, Ukraine*
[3]*Danylo Halytsky Lviv National Medical University, Lviv, Ukraine*
[4]*AstraZeneca Ukraine LLC, Kyiv, Ukraine*

OBJECTIVES

Global challenges such as VUCA (volatility, uncertainty, complexity, ambiguity) require the preparation of a new generation of experts for the transformation of the national healthcare system. It demands a restrictive national educational system. The upgrade of the educational system for students in the Kyiv National Economic University named after Vadym Hetman (KNEU) requires innovative approaches.

METHODS

This research is based on the analytical and statistical methods of the effectiveness of the educational system in Ukraine for economics graduate students in the field of HTA and pharmacoeconomics. We reviewed HTA and pharmacoeconomic guidelines to borrow effective international practice experience.

RESULTS

An innovative academic discipline “Forming of Global Health Economics: New Opportunities and Challenges” is already launched in KNEU. After this course, students can analyze market access for pharmaceuticals and medical technologies and perform cost-effectiveness analyses. They obtain such analytical skills as determining effectiveness, cost-effectiveness, demand evaluation, budget impact analysis. The analysis of the qualifications of KNEU students in the HTA field shows that their academic skills require more practical experience. That is why the processes of academics and industry collaborations were launched. Essential achievements in this way are Memorandums of cooperation. As of today, KNEU and global pharmaceutical industry stakeholders signed the Memorandum to promote HTA cooperation in Ukraine. Also, in plans is a memorandum of KNEU with the responsible body for HTA in Ukraine to guarantee a high level of professional qualification of students as future experts in HTA and pharmacoeconomics. As a result, we predict the availability of an increased number of HTA experts who would assist decision-making processes to promote patient access to effective, safe, and cost-effective medicines.

Figure 1: Students of the academic discipline “Forming of Global Health Economics: New Opportunities and Challenges”

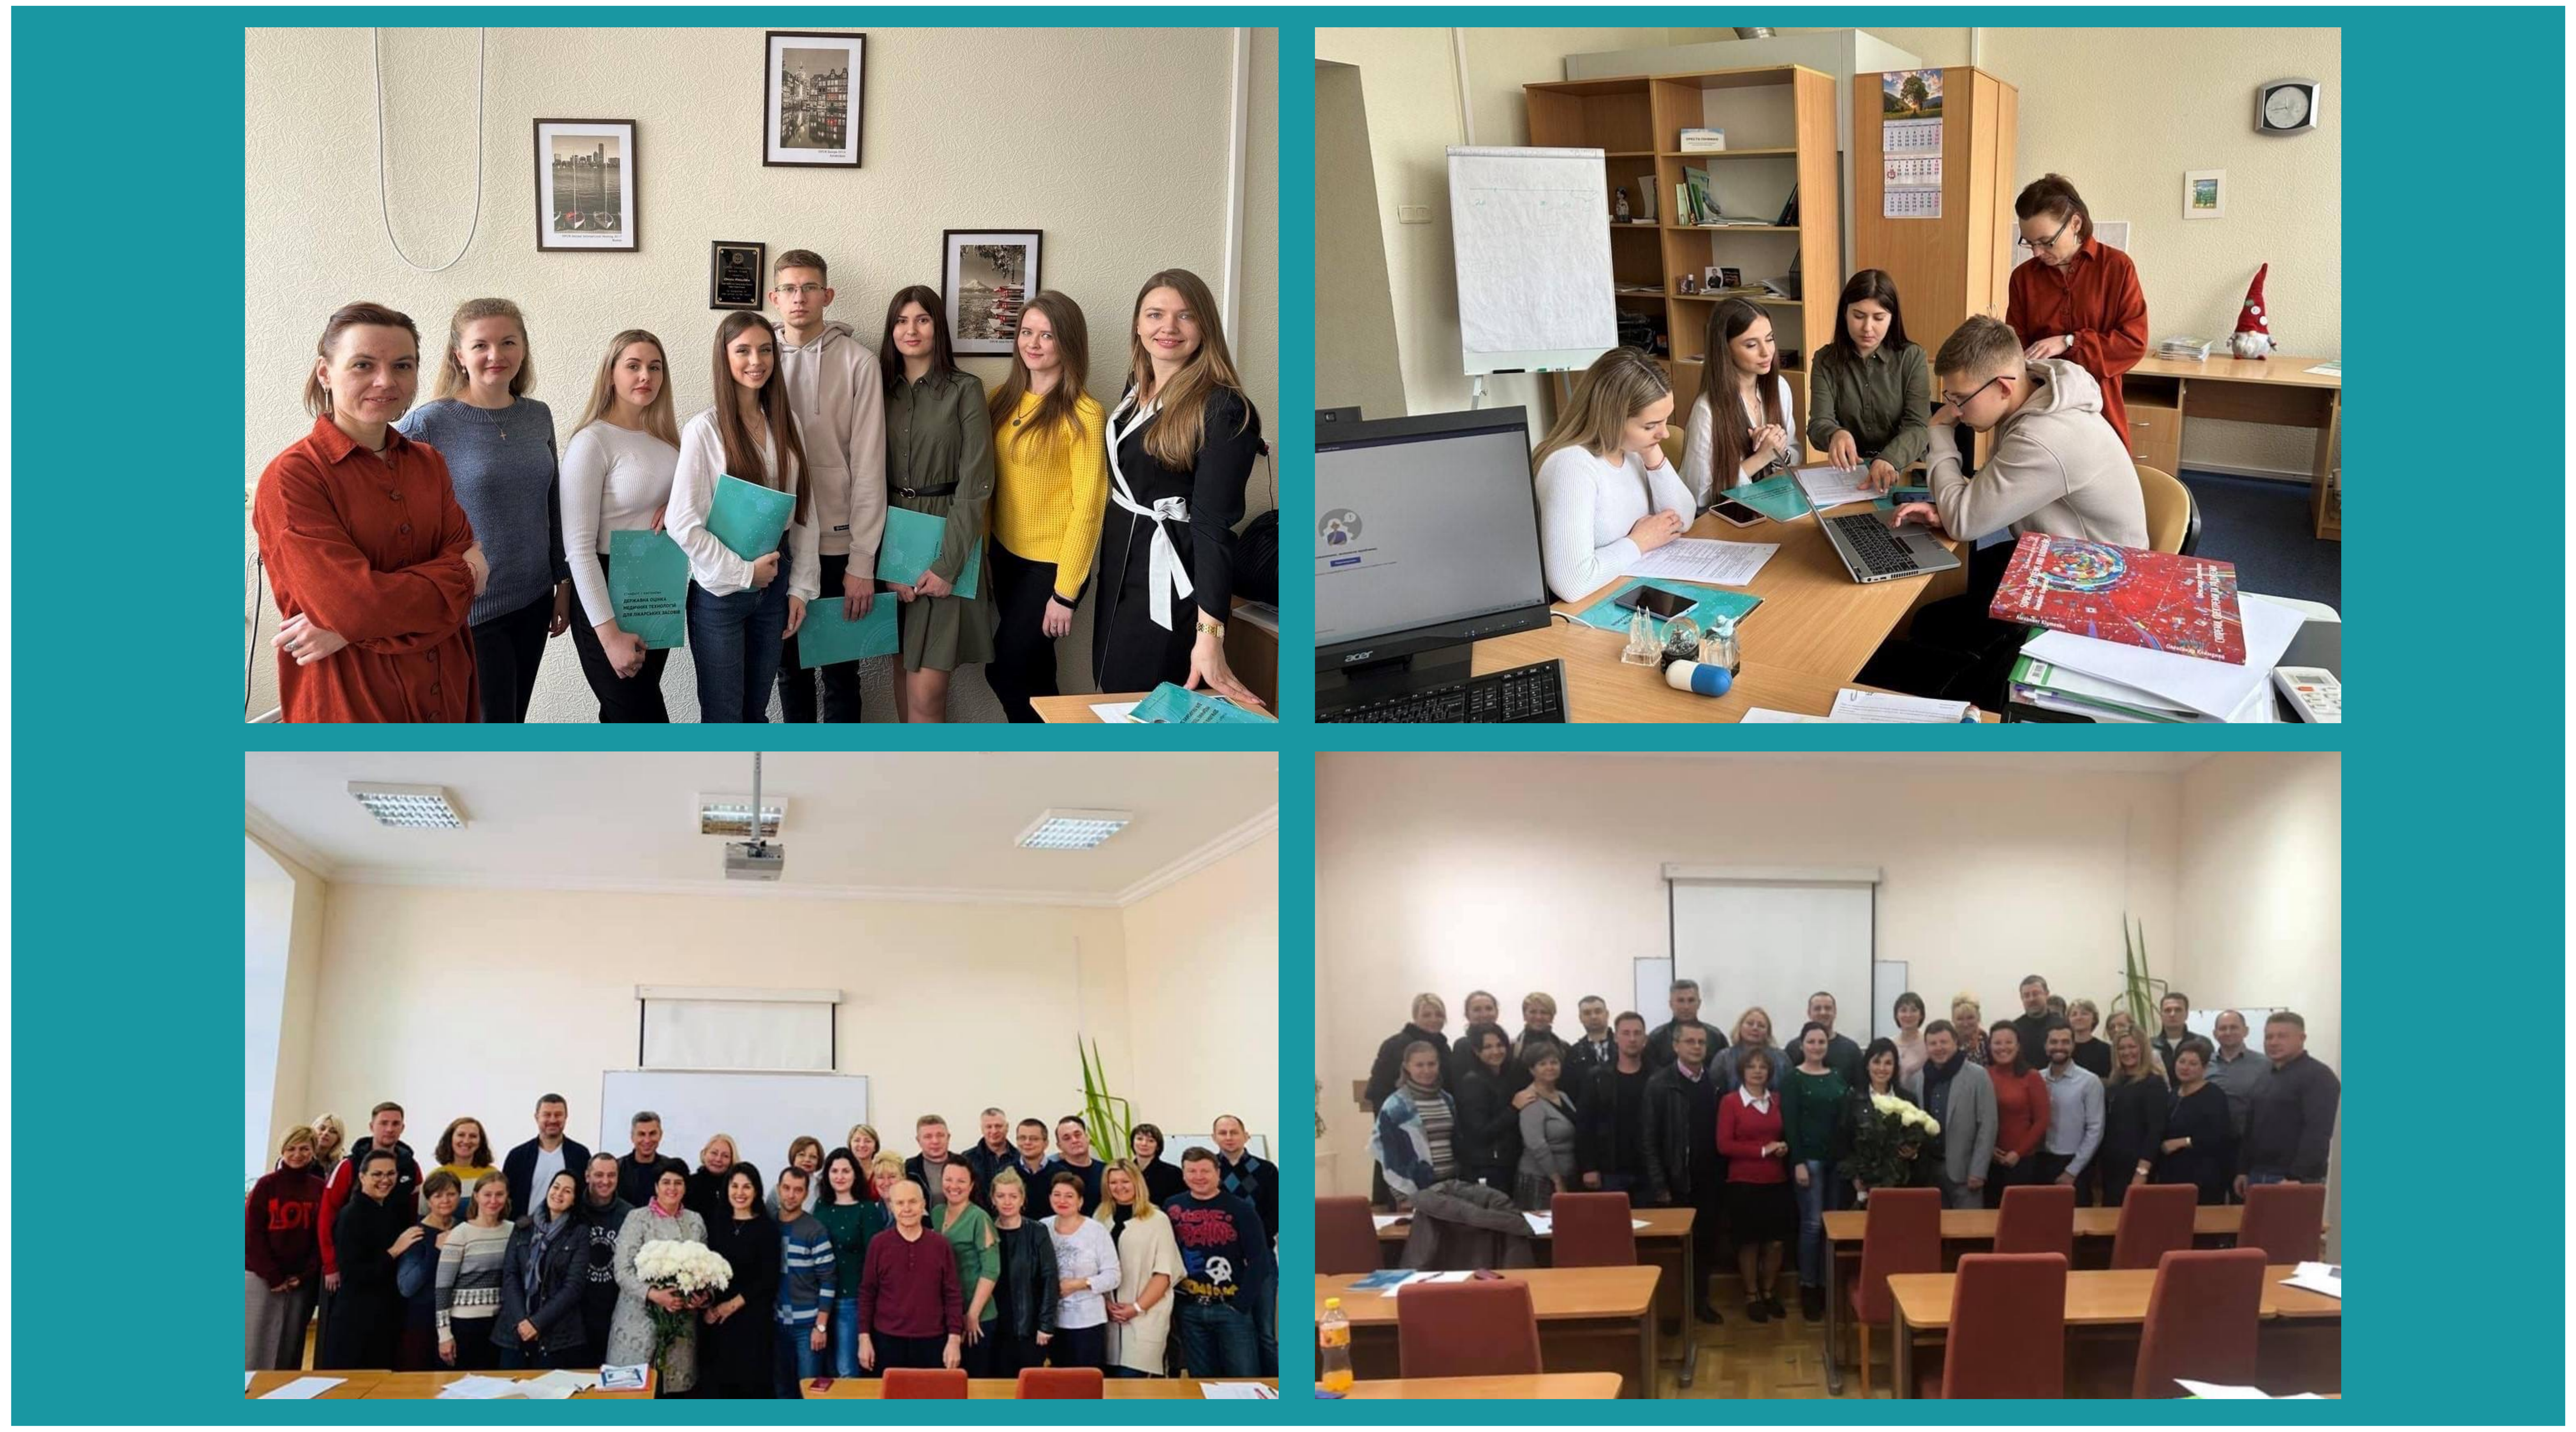

CONCLUSIONS

The process of upgrading the educational system in the field of HTA in KNEU through the stakeholders' involvement is an effective platform for students to gain practical experience.

REFERENCES

1. Kyiv National Economic University named after Vadym Hetman https://kneu.edu.ua/
2. State Expert Center of the Ministry of Health of Ukraine https://www.dec.gov.ua/
3. Danylo Halytsky Lviv National Medical University https://new.meduniv.lviv.ua/
4. AstraZeneca Ukraine LLC https://www.astrazeneca.ua/

International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 2024, Atlanta, GA, USA, May 5-8, 2024