Willingness to Pay Study for Migraine Medications in Argentina

Rey Ares L[1], Tabares MF[2], Vega C[1], Camacho Benavides A[3], Arrmesto I[4], Viozzi MF[1]

Affiliations:
1 Pfizer. Buenos Aires, Argentina.
2 Real World Insights (RWI), IQVIA. Buenos Aires, Argentina.
3 Real World Insights (RWI), IQVIA. Bogotá, Colombia.
4 Real World Analytics Solutions (RWAS), IQVIA. Buenos Aires, Argentina.

EE 478

Study Objective

The aim of this study was to assess the willingness to pay (WTP) to understand migraine treatment preferences of patients with migraine in an upper middle-income economy and the relative importance of therapies attributes.

Study Key Takeaway

The efficacy of the treatment was the most weighted attribute according to importance, followed by the speed of action.

The study reflected a patient with high price sensitivity consistent with the current economic context.
Patients were willing to pay between 3% and 20% of their monthly income for an effective migraine treatment.

These insights highlighted the diverse challenges that patients encountered in accessing and adhering to treatment regimens, emphasizing the need to address these barriers to improve overall patient care and outcomes.

INTRODUCTION

- Migraine is a neurological disorder, typified by recurrent, throbbing headaches, generally localized on one side of the head, accompanied with symptoms such as nausea, sensitivity to light (photophobia), and sound (phonophobia) that further exacerbate the condition's debilitating impact on individuals.
- The World Health Organization (WHO) underscores the global magnitude of migraine, estimating its yearly prevalence to range between 3% and 24.6% of the world population. This prevalence emphasizes the widespread impact of migraine on individuals across various demographics and geographical regions.
- Despite increased awareness and medical advancements, diagnosing and treating migraine remains far from optimal. The multifaceted nature of migraine, including variations in symptoms, triggers, and response to treatments, presents a complex clinical landscape, often leading to challenges in effective management and patient care.

METHODS

Figure 1: Study Design

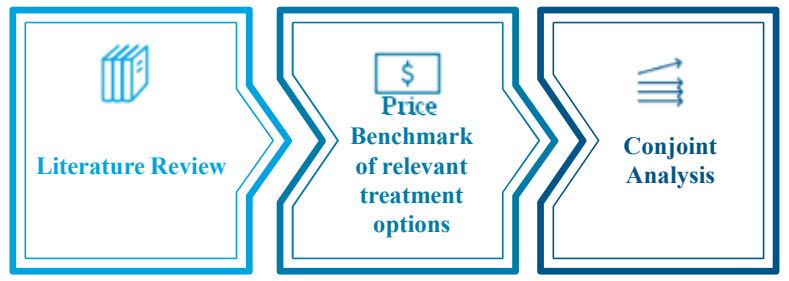
Literature Review
$
Price Benchmark of relevant treatment options
Conjoint Analysis

- The study was conducted in 3 stages: (i). a literature review focused on evaluating the efficacy and safety profiles of available antimigraine treatments (ii). a price benchmark of relevant treatment options (iii). a questionnaire to patients/customers to assess patients' preferences.
- We conducted a web questionnaire, participants recruited from a population-based database of headache sufferers' consumers and were shown assorted products, each of which differs in terms of combination of features. Each consumer performed the exercise 10 times, choosing from 3 hypothetical products profiles to the ideal one to shown preference. In the "conjoint" methodology, attributes are established with their respective options and presented to the participant randomly and in "blocks".
- Finally, the total selection of the blocks was analyzed according to which were the most selected combinations as well as the importance of each attribute in the decision of the participants.

RESULTS

Participant's profile

- Most of the patients surveyed were between 25 and 44 years of age.
- 83% of the patients surveyed were concentrated in 4 main cities.
- Most of the patients were employees in the private sector (micro-enterprises) and self-employed.
- Most respondents were between the middle and upper classes.
- 60% of patients had social coverage as their first medical coverage.
- 54% of patients who had a second coverage correspond to pre-paid.

Table 1: Caractheristics of respondents

Number of patients surveyed: 185
Patient's gender
Female: 51% (94) Male: 49% (91)
Number of patients by age group
18-24 years: 6.5% (12) 25-34 years: 34.1% (63) 35-44 years: 31.9% (59) 45-54 years: 18.4% (34) 55-64 years: 8.6% (16) +65 years: 0.5% (1)
Place of residence
Buenos Aires: 57.3% (106) Cordoba: 10.3% (19) Mendoza: 8.1% (15) Santa Fe: 7.0% (13) Others: 17% (32)
Socioeconomic level
ABC1-Upper Class: 30.8% (57) C2-Upper Middle Class: 25.9% (48) C3-Middle Class: 29.2% (54) D1-Upper Lower Class: 11.4% (21) D2E-Lower Class: 2.7% (5)

Diagnosis and classification

- 34% of patients were diagnosed less than 1 year ago and 54% took less than 1 year to reach diagnosis.
- Most patients saw a clinician for the first time, but they were not necessarily the ones making the diagnosis.
- 80% of patients were diagnosed in less than a year, and the vast majority had migraine 2 to 7 days a month.
- 64% of patients had less than 8 monthly episodes.

Figure 2: Time since migraine diagnosis (n=185 patients)

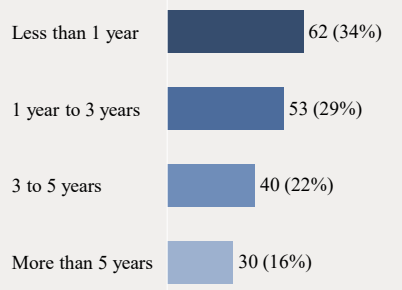
Less than 1 year
62 (34%)
1 year to 3 years
53 (29%)
3 to 5 years
40 (22%)
More than 5 years
30 (16%)

Figure 3: Time from onset of pain to migraine diagnosis (n=185 patients)

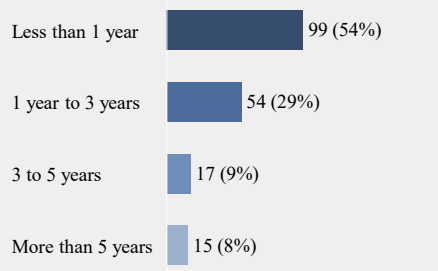
Less than 1 year
99 (54%)
1 year to 3 years
54 (29%)
3 to 5 years
17 (9%)
More than 5 years
15 (8%)

Figure 4: Distribution of patients by monthly migraine days (n=185 patients)

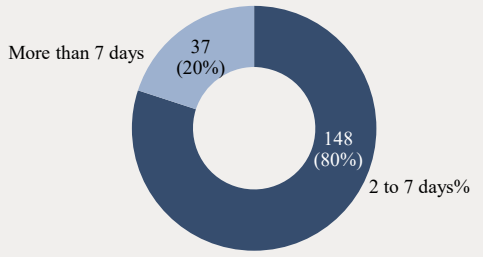
More than 7 days
37
(20%)
148
(80%)
2 to 7 days%

Figure 5: Patients by number of episodes in the last 3 months (n=185 patients)

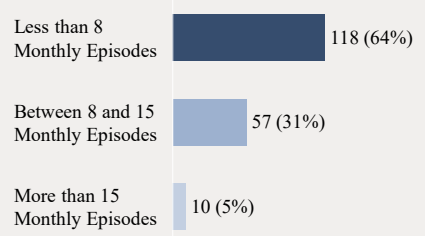
Less than 8 Monthly Episodes
118 (64%)
Between 8 and 15 Monthly Episodes
57 (31%)
More than 15 Monthly Episodes
10 (5%)

Treatment dynamics

- The efficacy of the treatment was the most weighted attribute according to importance, followed by the speed of action and duration of effect.
- 30% of patients were not under treatment. The main reason for this is that it has not been indicated.
- Paracetamol, Ergotamine+Dipyrone+Caffeine and Ibuprofen were the most used treatments. Sumatriptan stood out for its effectiveness.

Figure 6: Number of patients under migraine treatment (n = 185 patients)

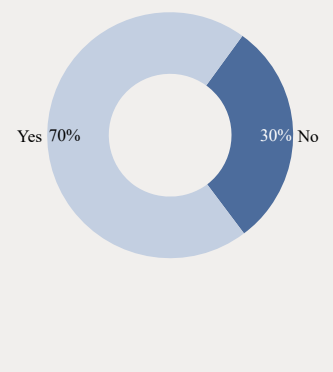
Yes 70%
30% No

Figure 7: Reason for not being under treatment (n = 55 patients)

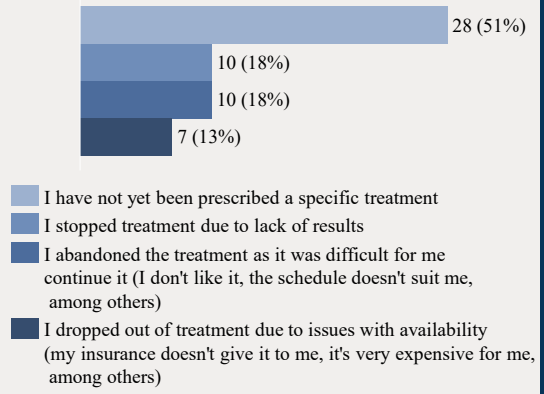
28 (51%)
10 (18%)
10 (18%)
7 (13%)
I have not yet been prescribed a specific treatment
I stopped treatment due to lack of results
I abandoned the treatment as it was difficult for me continue it (I don't like it, the schedule doesn't suit me, among others)
I dropped out of treatment due to issues with availability (my insurance doesn't give it to me, it's very expensive for me, among others)

Figure 8: Main treatments used for migraine (n = 185 patients, 280 mentions)

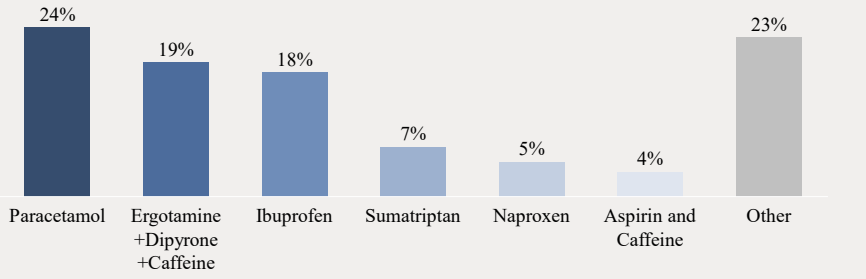
24%
Paracetamol
19%
Ergotamine +Dipyrone +Caffeine
18%
Ibuprofen
7%
Sumatriptan
5%
Naproxen
4%
Aspirin and Caffeine
23%
Other

Conjoint Analysis

Acute Treatment

- Price was the attribute that patients attached the most importance to when choosing a migraine product.
- When differentiating by time of diagnosis, both price and route of administration were attributes that represented greater importance for patients who had been diagnosed more than 3 years ago.
- There was no significant difference in the relative importance of attributes when analyzed by time of diagnosis.
- In terms of route of administration, oral presentation was the characteristic that they perceived to be most useful
- Intravenous or subcutaneous administrations were undesirable.
- Having a 15% chance of relief after 2 hours as an attribute generated more utility for patients who had had migraine for more than 3 years
- According to the price, the product that represented the greatest utility for the patient was the one that had a price of USD 34.15 per month and USD 145.90 per month.

Figure 9: Relative importance of attributes for acute treatment (n=185 patients)

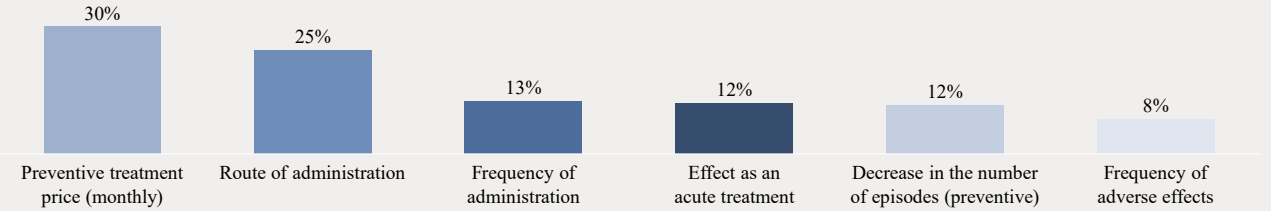
30%
Preventive treatment price (monthly)
25%
Route of administration
13%
Frequency of administration
12%
Effect as an acute treatment
12%
Decrease in the number of episodes (preventive)
8%
Frequency of adverse effects

Episodic Treatment

- Price was the attribute ,that they attached the most importance to when choosing a product for episodic migraine. When analyzed by time with diagnosis, price represented greater importance for patients over 3 years of age.
- In terms of price, between USD 24.83 and USD 41.91 was the range that generated some utility in the patients surveyed.
- That the medication did not have a preventive effect was rated as undesirable regardless of the time since the diagnosis.
- Higher prices generated negative utility, however, the longer the diagnosis with migraine, the lower it.
- The most desirable route of administration was oral; however, the nasal presentation was still an option that generated positive utility.
- Patients ruled out subcutaneous or intravenous treatment as an option.
- According to the price, the product that represented the greatest utility for the patient was the one that has a price of USD 24.83 per month.

Note: Average Exchange rate BCRA August 2023 -1 USD = 322,13 ARS

Figure 10: Relative importance of attributes for episodic treatment (n=185 patients)

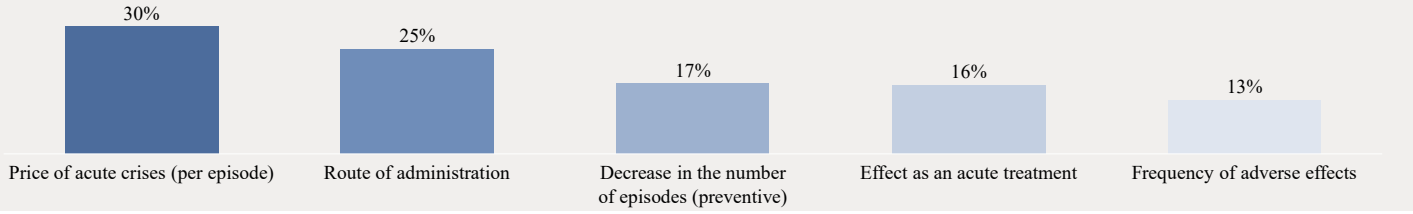
30%
Price of acute crises (per episode)
25%
Route of administration
17%
Decrease in the number of episodes (preventive)
16%
Effect as an acute treatment
13%
Frequency of adverse effects

CONCLUSIONS

According to our findings, respondents had a significant willingness to pay for medicines with higher efficacy and less-severe AEs. Considering a migraine treatment, improvement of these attributes would be valuable to migraine patients. Patients may be willing to trade off price for better attributes.

Topic subcategory

- Cost-comparison
- Effectiveness
- Utility
- Benefit Analysis
- Preference & Patient Satisfaction
- Stated
- Analysis Patient Behavior and Incentives

Disclosures:
Lucila Rey Ares, Celina Guadalupe Vega and Maria Florencia Viozzi are employed by Pfizer. Marina Fiorella Tabares, Alejandra Camacho Benavides and Ignacio Arrmesto are employed by IQVIA.

For more information about this work, please scan this QR code.

SCAN ME

Presented by Celina Vega at ISPOR; May 5th, 2024; Atlanta, Georgia, USA