Stakeholders Perspectives About Patient Participation in HTA in Latin America: Best Practices, Main Gaps, and Recommendations

HTA335

Aline Silveira Silva
Researcher, University of Brasilia/University of British Columbia, Vancouver, Canada
dr.alinesvsilva@gmail.com

ISPOR
Improving healthcare decisions

OBJECTIVES

Latin America (LATAM) has seen a transition to democracy, leading to experiments with participatory governance. **Argentina, Brazil, and Colombia** have implemented formal mechanisms to **involve patients and citizens in Health Technology Assessment (HTA)**. This study aimed to explore this involvement.

METHODS

Interviews with **different stakeholders** from these **03 countries** and an **online survey** from **10 LATAM countries** were conducted to identify and rank **good practices, gaps, and recommendations**.

RESULTS

Good practices and gaps identified by the **interviewees** from **Argentina, Brazil and Colombia** are depicted beside. The **top 4 practices**, ranked by the online survey, were training, continuous patient involvement, participation guides, and patient participation in committees and meetings. However, respondents felt that **patients and patient groups were unprepared** and had **little impact on health decision-making**. The **most pressing gap** was undervaluing of patient involvement by HTA institutions, followed by lack of patient knowledge, transparency and information, and unclear methodology. The **top recommendations** were clear participation methods, continuous patient involvement, better government support and dialogue, and guidance for participation.

Patient participation is crucial in healthcare, and efforts should be made to involve patients and bridge the identified gaps. Implementing the identified recommendations can lead to a genuine patient-centered healthcare system in LATAM.

Good Practices

Argentina:
- Patient committee within CONETEC, featuring four patient representatives continuously involved in the process

Brazil:
- Public consultations
- Patient testimonials
- Public hearings
- Broadcasting meetings

Colombia:
- Release of a handbook for participation by the IETS that incorporated patient feedback

Gaps

Top 4:
1. Undervaluing of patient involvement
2. Lack of patient knowledge about the process
3. Lack of transparency and information
4. Unclear methodology

Recommended Actions

Top 4:
1. Clear and transparent methods
2. Continuous patient involvement
3. Government support & dialogue
4. Training & Improved communication

Others:
- Better representation & diversity
- Transparent expectations and roles
- Sharing experiences among countries
- Evaluation

Top 4 Practices

1. Training for patients
2. Continuous patient involvement
3. Participation guides
4. Patient participation in committees and meetings

Take a picture to download the position paper

This study was sponsored by FIFARMA