Clinical Outcomes of Patients with Obesity in a US Employer-funded Medical Weight Management Program: A Pilot Study

Poster Code: HSD92

Kelly Olsson, MA[1]; Ronald DeVizia, PharmD, CDES[2]; Caroline Lewis, BPharm[2]
Presenting author: Kelly Olsson

Introduction and Aims

- An estimated 42% of adults in the United States (US) have obesity.[1] Obesity and obesity-related comorbidities represent a large burden on society and on the US healthcare system.
- Interventions to treat obesity are generally aimed at making lifestyle modifications to diet and physical activity levels, but weight loss is difficult to maintain over time, particularly with these methods alone.[2-3]
- In addition to lifestyle modifications, multiple evidence and consensus-based guidelines for the treatment of obesity recommend the use of anti-obesity medication (AOM) in combination with diet and exercise in patients with obesity.[4-8]
- Understanding the benefit of treating patients with obesity with AOMs, in addition to a weight management program, will help to better provide care for this patient population.
- We aimed to present and descriptively compare obesity-related clinical outcomes among three cohorts of patients participating in a 12-month pilot medical weight management program (MWMP) offered by Piedmont Pharmaceuticals Care Network, LLC (PPCN), to two self-insured employers. By evaluating outcomes for patient cohorts defined by AOM use, we aim to provide insights regarding the benefits of a weight management program combined with AOMs.

Methods

- This was an observational, retrospective cohort analysis of health plan members.
- Inclusion criteria:
 - Aged ≥18 years
 - Body mass index (BMI) of ≥40 kg/m^2
 - Continuous enrollment in the participating benefit program
- Exclusion criteria:
 - Patients with pre-existing type 2 diabetes
- Clinical data was collected at biometric screening events/quarterly visits from April 2021 to March 2022 and August 2021 to July 2022; data was combined for both employers. Comorbidities were obtained from medical claims data using ICD-10 diagnosis codes.
- Three patient cohorts were analyzed:
 - **Branded AOM (bAOM) Use**
 - Participated in the MWMP pilot
 - Were prescribed branded glucagon-like peptide 1 receptor agonists ([GLP-1 RA] liraglutide or semaglutide injection indicated for obesity, any dose) by a clinician
 - Had ≥3 fills of the prescribed medication
 - **AOM Mix Use**
 - Participated in the MWMP pilot
 - Use of all other AOMs (non bAOM) or received fewer than 3 fills of branded GLP-1 RA (liraglutide or semaglutide injection indicated for obesity, any dose)
 - **Non AOM/MWMP Use**
 - Not enrolled or participating in the MWMP pilot
 - Did not receive AOMs
- bAOM Use and AOM Mix Use cohorts voluntarily participated in the MWMP, consisting of virtual coaching, weekly check-ins, and quarterly clinical pharmacist visits.
- Patient demographic characteristics and clinical outcomes were characterized at the start and end of the pilot MWMP.

Results

- Eighty-four patients were included in the analysis: 31 bAOM Use, 8 AOM Mix Use, and 45 Non AOM/MWMP Use.
- Baseline demographic and clinical characteristics are presented in **Table 1**.
- Between the start and end of the program, mean weight loss and decrease in BMI were greatest for the bAOM Use cohort. Changes in key clinical characteristics are shown in **Figure**.
- Reductions were observed in the number of some obesity-related comorbidities present at the end of the MWMP for the bAOM Use group **(Table 2)**.

Table 1. Baseline demographics and clinical characteristics of health plan members included in the analysis

Characteristic	bAOM Use (n=31)	AOM Mix Use (n=8)	Non AOM/MWMP Use (n=45)
Age, mean (SD)	46.45 (9.95)	44.50 (10.52)	40.60 (11.34)
Sex (female), n (%)	23 (74%)	7 (88%)	27 (60%)
Race/ethnicity, n (%)*			
Black/African American	7 (23%)	2 (25%)	0 (0%)
White , non-Hispanic	24 (77%)	6 (75%)	45 (100%)
BMI (kg/m^2), mean (SD)	43.94 (4.05)	52.20 (8.92)	46.75 (6.10)
Body weight (pounds), mean (SD)	279.10 (46.19)	318.55 (58.55)	304.24 (48.40)

*Race/ethnicity were self-reported by the patient.

Figure. Changes in clinical measurements from start to end of 12 months of MWMP pilot participation

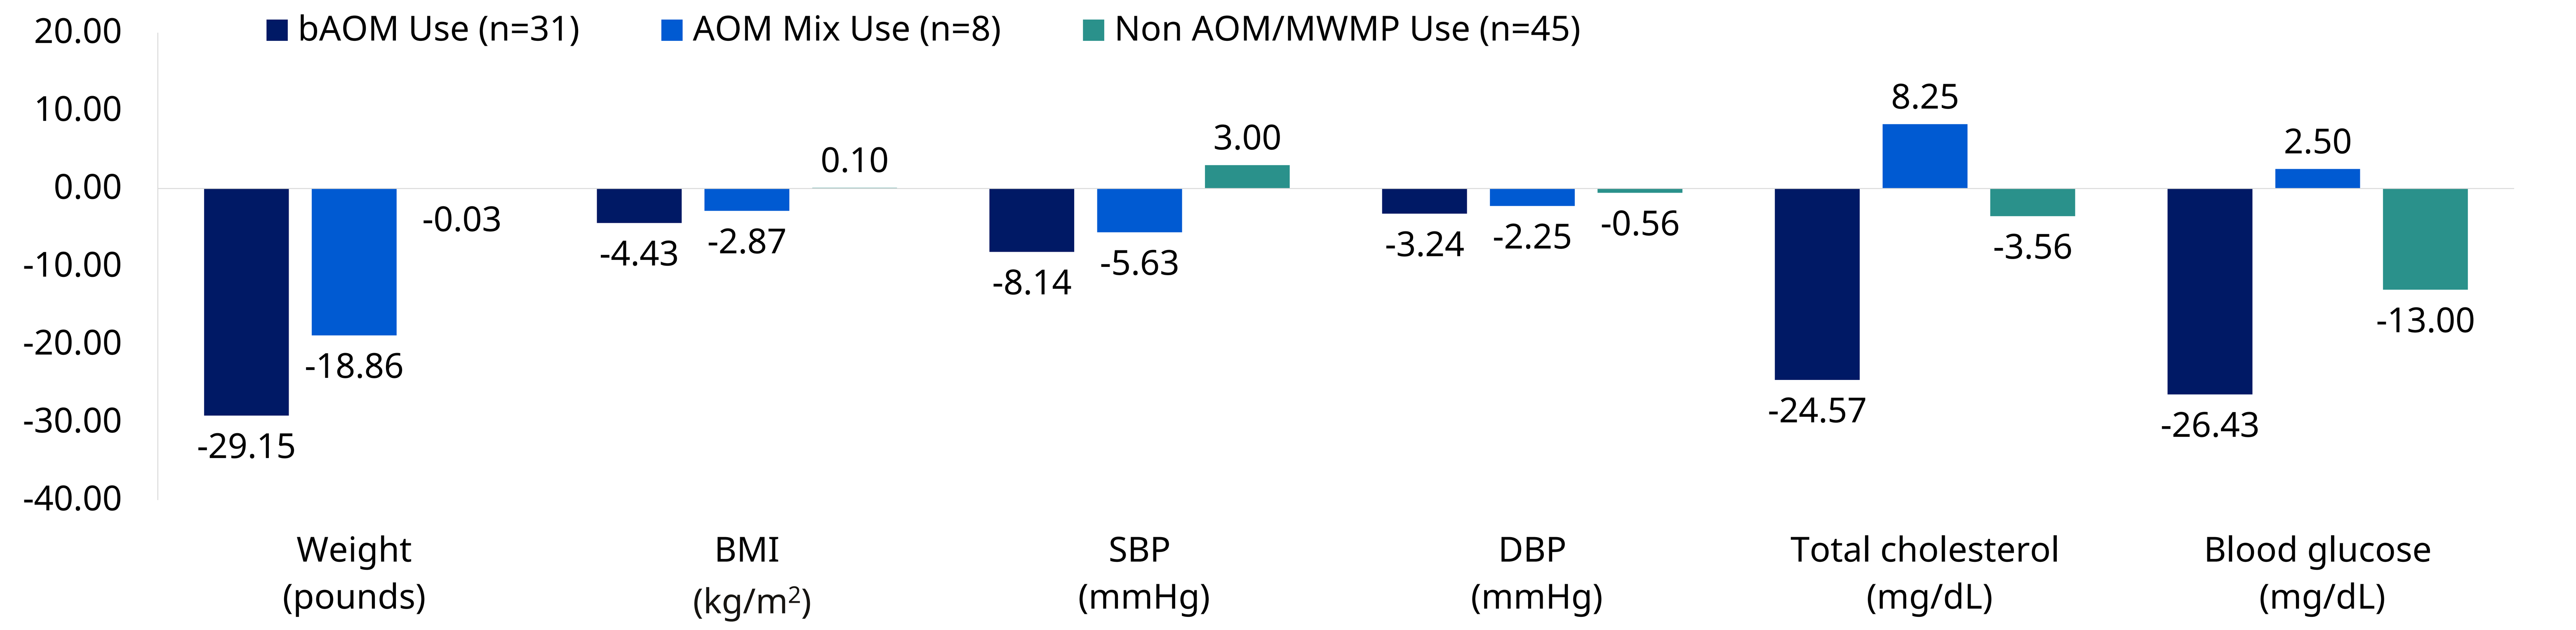
bAOM Use (n=31)
AOM Mix Use (n=8)
Non AOM/MWMP Use (n=45)
20.00
10.00
0.00
-10.00
-20.00
-30.00
-40.00
-29.15
-18.86
-0.03
-4.43
-2.87
0.10
-8.14
-5.63
3.00
-3.24
-2.25
-0.56
-24.57
8.25
-3.56
-26.43
2.50
-13.00
Weight (pounds)
BMI (kg/m2)
SBP (mmHg)
DBP (mmHg)
Total cholesterol (mg/dL)
Blood glucose (mg/dL)

Table 2. Changes in obesity-related comorbidities from start to end of 12 months of MWMP pilot participation

Comorbidity	bAOM Use (n=31)	AOM Mix Use (n=8)	Non AOM/MWMP Use (n=45)
ASCVD	-2	0	3
Asthma	1	2	0
Dyslipidemia	2	0	0
GERD	-4	1	-2
HFpEF	0	0	0
Hypertension	5	2	2
Knee Osteoarthritis	0	0	1
Musculoskeletal Pain	-2	2	4
Obstructive Sleep Apnea	-3	1	0
PCOS	0	1	0
Prediabetes	0	3	-3
Psoriasis	0	0	0
T2DM	1	0	5
Urinary Incontinence	0	0	1

Notes: The bAOM Use cohort was prescribed liraglutide or semaglutide injection (indicated for obesity), any dose (i.e., was not necessarily therapeutic dose). All patients had a BMI ≥40 kg/m^2. Comorbidities were obtained from medical claims data.

Limitations

- Limitations of our study include:
 - Small sample sizes (particularly for the AOM Mix Use group).
 - Only two employers were included in the analysis (and were both self-insured).
 - Short study duration period.
 - All patients had a BMI of ≥40 kg/m^2.
 - The bAOM Use cohort was prescribed liraglutide or semaglutide injection (indicated for obesity), any dose (i.e., was not necessarily therapeutic dose).
 - Comorbidities were determined based on presence of ICD-10 code for condition in claims data.

Summary and Conclusions

- Participants in a pilot MWMP who took bAOMs experienced substantial weight loss and change in BMI, similar to those observed in randomized controlled trials and other real-world evidence research.[9-11] We also saw improvements in key clinical measures.

Abbreviations: AOM, anti-obesity medication; ASCVD, atherosclerotic cardiovascular disease; bAOM, branded AOM; BMI, body mass index; DBP, diastolic blood pressure; GERD, gastroesophageal reflux disease; GLP-1 RA, glucagon-like peptide 1 receptor agonists; HFpEF, heart failure with preserved ejection fraction; ICD-10, International Classification of Diseases, Tenth Revision; MWMP, medical weight management program; PCOS, polycystic ovary syndrome; SBP, systolic blood pressure; SD, standard deviation; T2DM, type 2 diabetes mellitus.

Disclosures:
This research was funded by Novo Nordisk Inc. (Plainsboro, NJ, USA).
Medical writing and editorial support was provided by Rebecca Hahn, MPH, of KJT Group, Inc. (Rochester, NY, USA), which was funded by Novo Nordisk Inc.
RD and CL are employees of Piedmont Pharmaceuticals Care Network, which received funding from Novo Nordisk Inc. to conduct the study.
KO is an employee of, and shareholder in, Novo Nordisk Inc.
Affiliations: [1]Novo Nordisk Inc., Plainsboro, NJ, USA; [2]Piedmont Pharmaceuticals Care Network, LLC, Greensboro, NC, USA
Presented at ISPOR 2024 (May 5-8, 2024), Atlanta, GA, USA.

References:
1. Hales C, Carroll M, Fryar C, Ogden C. Prevalence of obesity and severe obesity among adults: United States, 2017–2018. NCHS Data Brief, no 360. National Center for Health Statistics: Hyattsville, MD, USA. 2020.
2. Douketis JD, Macie C, Thabane L, Williamson DF. Systematic review of long-term weight loss studies in obese adults: clinical significance and applicability to clinical practice. *Int J Obes (Lond)*. 2005;29(10):1153-1167. doi:10.1038/sj.ijo.0802982
3. Ghanemi A, Yoshioka M, St-Amand J. Broken energy homeostasis and obesity pathogenesis: the surrounding concepts. *J Clin Med*. 2018;7(11):453. doi:10.3390/jcm7110453
4. Bray GA, Ryan DH. Evidence-based weight loss interventions: individualized treatment options to maximize patient outcomes. *Diabetes Obes Metab*. 2021;23(suppl 1):50-62. doi:10.1111/dom.14200
5. Friedrichsen M, Breitschaft A, Tadayon S, Wizert A, Skovgaard D. The effect of semaglutide 2.4 mg once weekly on energy intake, appetite, control of eating, and gastric emptying in adults with obesity. *Diabetes Obes Metab*. 2021;23(3):754-762. doi:10.1111/dom.14280
6. Khera R, Murad MH, Chandar AK, et al. Association of pharmacological treatments for obesity with weight loss and adverse events: a systematic review and meta-analysis. *JAMA*. 2016;315(22):2424-2434. doi:10.1001/jama.2016.7602
7. Garvey WT, Mechanick JI, Brett EM, et al. American Association of Clinical Endocrinologists and American College of Endocrinology comprehensive clinical practice guidelines for medical care of patients with obesity. *Endocr Pract*. 2016;22(suppl 3):1-203. doi:10.4158/EP161365.GL
8. Wharton S, Lau DCW, Vallis M, et al. Obesity in adults: a clinical practice guideline. *CMAJ*. 2020;192(31):E875-E891. doi:10.1503/cmaj.191707
9. Wilding JPH, Batterham RL, Calanna S, et al. Once-weekly semaglutide in adults with overweight or obesity. *N Engl J Med*. 2021;384(11):989-1002. doi:10.1056/NEJMoa2032183
10. Davies M, Færch L, Jeppesen OK, et al. Semaglutide 2·4 mg once a week in adults with overweight or obesity, and type 2 diabetes (STEP 2): a randomised, double-blind, double-dummy, placebo-controlled, phase 3 trial. *Lancet*. 2021;397(10278):971-984. doi:10.1016/S0140-6736(21)00213-0
11. Ruseva A, Michalak W, Zhao Z, Fabricatore A, Hartaigh BÓ, Umashanker D. Semaglutide 2.4 mg clinical outcomes in patients with obesity or overweight in a real-world setting: a 6-month retrospective study in the United States (SCOPE). *Obes Sci Pract*. 2024 Feb 8;10(1):e737. doi:10.1002/osp4.737