EE457

The Economic Burden Impact of Thyroid Eye Disease in The United Arab Emirates

Eatamadi H[1], Elkonaissi I[1], **Al Dallal S[2]**, Shaheen R[3], Alharbi M[3], Abdelaal M[4], Sehly K[4]

[1]Sheikh Shakhbout Medical City, Abu Dhabi, United Arab Emirates, [2]Dubai Health Authority, Dubai, United Arab Emirates, [3]HEPA Solutions, Riyadh, Saudi Arabia, [4]Amgen, Riyadh, Saudi Arabia

Copies of this eposter obtained through QR Code are for personal use only and may not be reproduced without written permission from the authors.

Background

- Thyroid eye disease (TED) is characterized by progressive inflammation and remodeling of the tissue surrounding the orbits, especially the extraocular muscle, connective and fatty tissue [1].
- TED has an active phase during which usually manifests as a variety of symptoms including pain, redness, swelling, bulging of the eyes (proptosis) and double vision (diplopia). This is often followed by an inactive phase where disease progression ceases[2-4].
- TED imposes significant clinical, psychological and economic burdens on patients and their caregivers due to its debilitating and vision-impairing nature [5].
- The estimated prevalence of TED is reported at 18.9 per 100,000 people[6].
- Studies reporting the burden of TED are limited, particularly, in the United Arab Emirates (UAE) where there's a scarcity of reported prevalence and practice behaviors.

Objective

The aim of this study was to assess the economic burden and the cost of illness of TED amongst UAE patients from societal and payers perspectives.

Methods

- A cost of illness study was conducted to evaluate the economic burden of TED in the UAE.

Model Structure:

- Population: TED patients in the UAE of all severity stages
- Approach: bottom-up and prevalence based
- Perspective: payers and societal perspectives
- Time Horizon: 1-5 years
- Parameters: direct and indirect costs
- Data collection: a structured questionnaire-guided interviews

Figure 1. Research design for cost estimation model

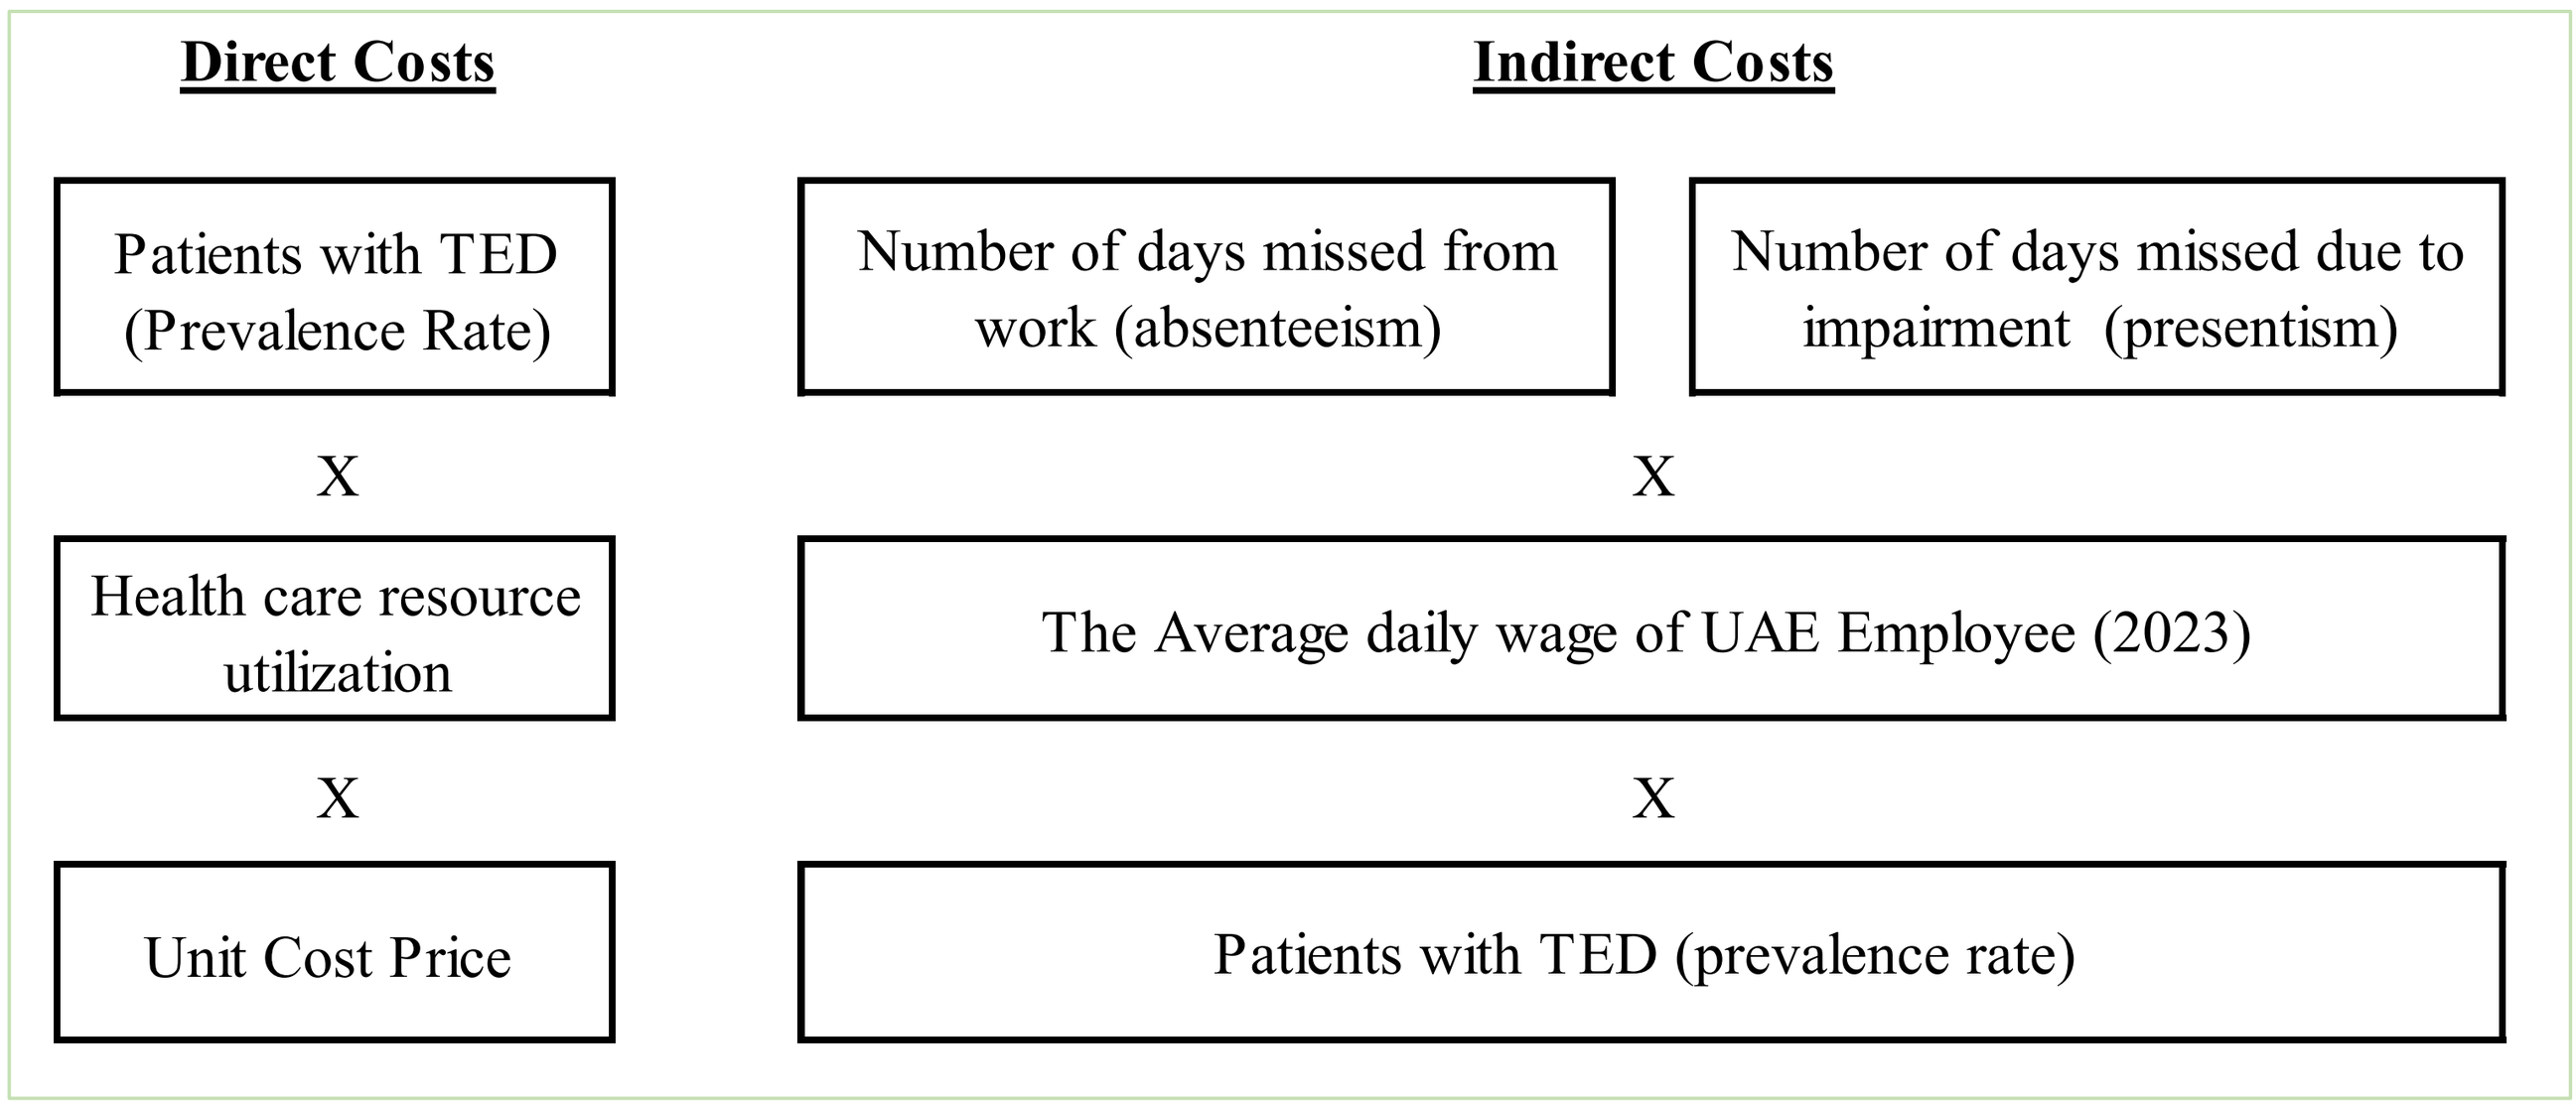
Direct Costs
Indirect Costs
Patients with TED (Prevalence Rate)
Number of days missed from work (absenteeism)
Number of days missed due to impairment (presentism)
X
X
Health care resource utilization
The Average daily wage of UAE Employee (2023)
X
X
Unit Cost Price
Patients with TED (prevalence rate)

Methods Cont'd

- Direct medical costs were retrieved from national insurance for outpatient visits, medications, laboratory tests, imaging, and surgery costs.
- Indirect costs included absenteeism and productivity loss based on the experts' inputs.

Statistical Approach

- Descriptive statistics were used in data analysis to determine the prevalence of TED, the healthcare resource utilization and the associated costs.
- Through extrapolation of cost and prevalence data, the economic burden was calculated for each disease state.
- One-way sensitivity analyses were conducted in which input parameter values were varied by ±10%.

Results

- The estimated TED prevalence is 1500 patients in the UAE.
- The patients are distributed among active mild to moderate disease, moderate to severe disease, and very severe cases (Sight threatening) as presented in table 1.

Table 1. Distribution of patients amongst TED disease states

TED Disease States	%	N
Active Mild to Moderate Disease	60%	900
Active Moderate to Severe Disease	30%	450
Active Severe Disease	7%	105
Sight Threatening	3%	45

- Disease severity had varying levels of impact on total cost directly and indirectly.
- The annual cost per patient was AED 103,836 ($28,293), AED 239,768 ($65,332), and AED 256,167 ($69,800) for active mild to moderate, moderate to severe disease, and very severe cases, respectively.
- Over 5 years with diseases transition, the average cost per patient was AED 926,976 ($252,582) with moderate to severe disease.
- Indirect costs significantly contributed to cost accounting for 55.7-59.5% of the overall costs from the total patients with active moderate to severe and severe to very severe cases.
- By extrapolating costs based on prevalence rate of moderate to severe cases, the overall projected costs were between AED 424.5 million ($115.6 million) and AED 554.2 million ($151 million) over 5 years.

Results

Figure 2. Total cost breakdown per patient for active mild disease

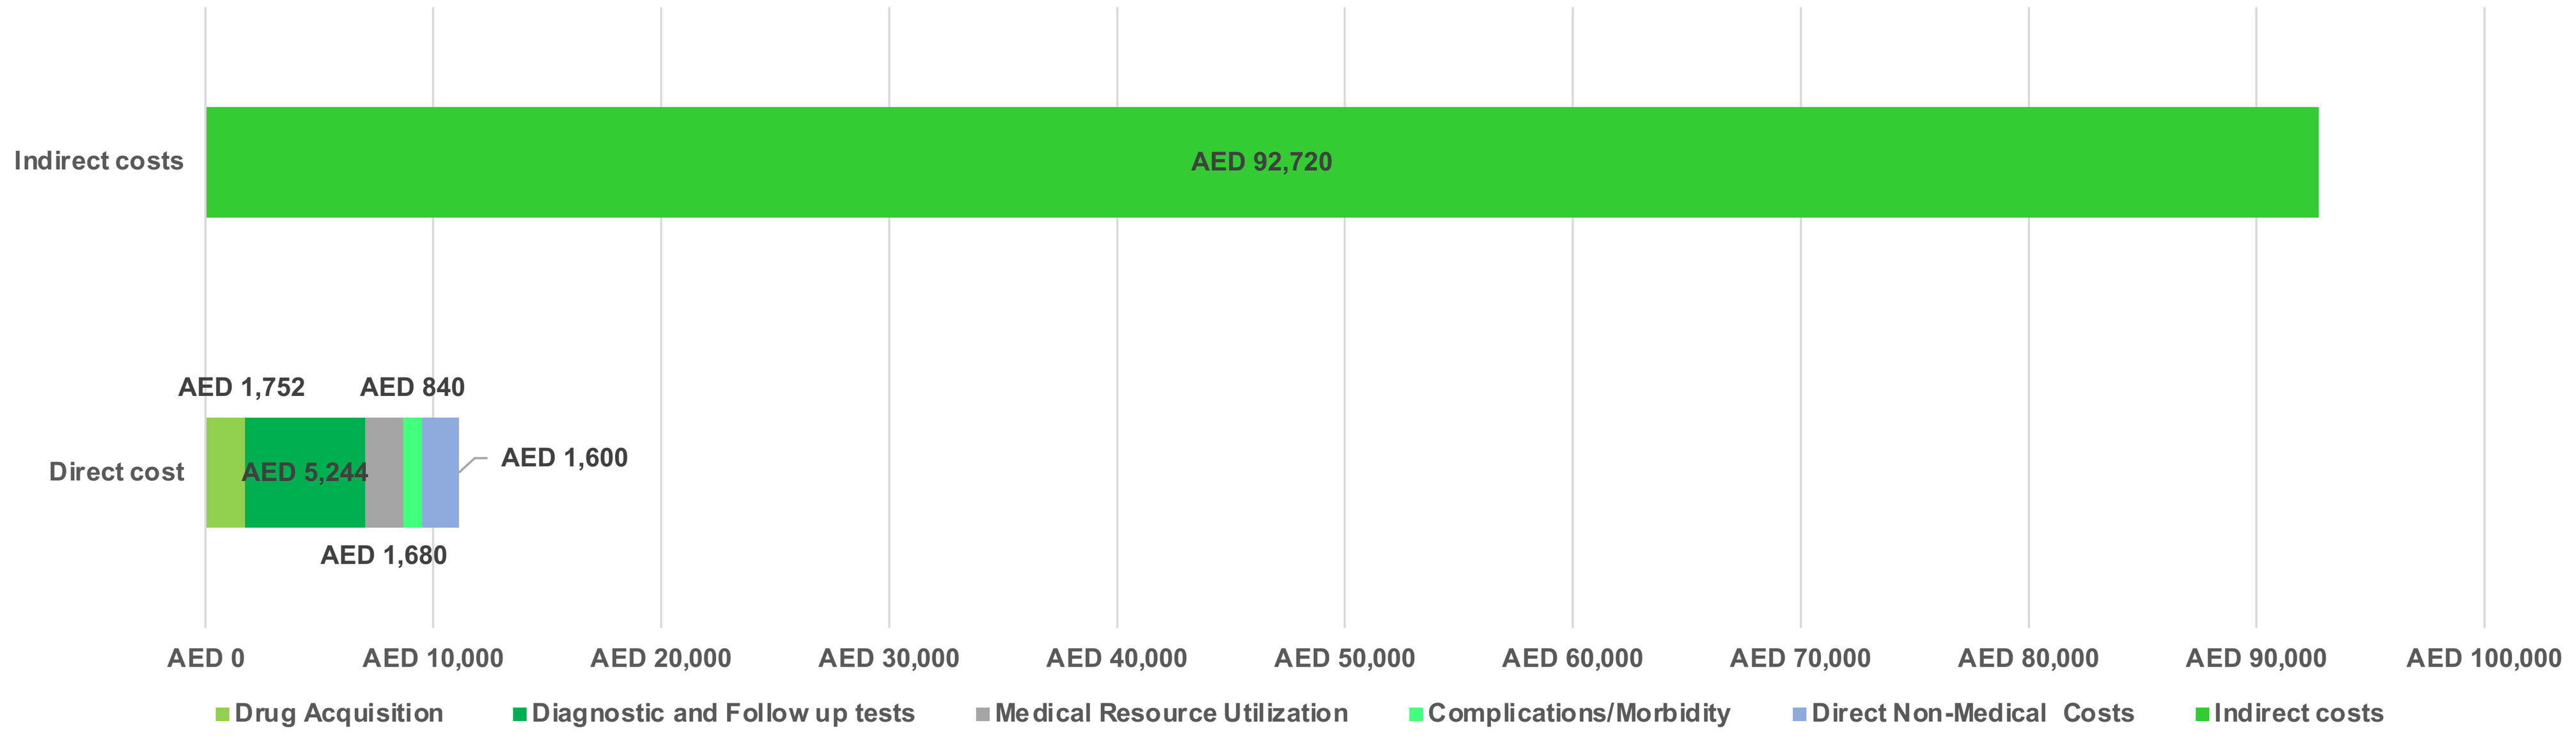
Indirect costs
AED 92,720
Direct cost
AED 1,752
AED 840
AED 5,244
AED 1,600
AED 1,680
AED 0
AED 10,000
AED 20,000
AED 30,000
AED 40,000
AED 50,000
AED 60,000
AED 70,000
AED 80,000
AED 90,000
AED 100,000
Drug Acquisition
Diagnostic and Follow up tests
Medical Resource Utilization
Complications/Morbidity
Direct Non-Medical Costs
Indirect costs

Figure 3. Total cost breakdown per patient for active moderate to severe disease

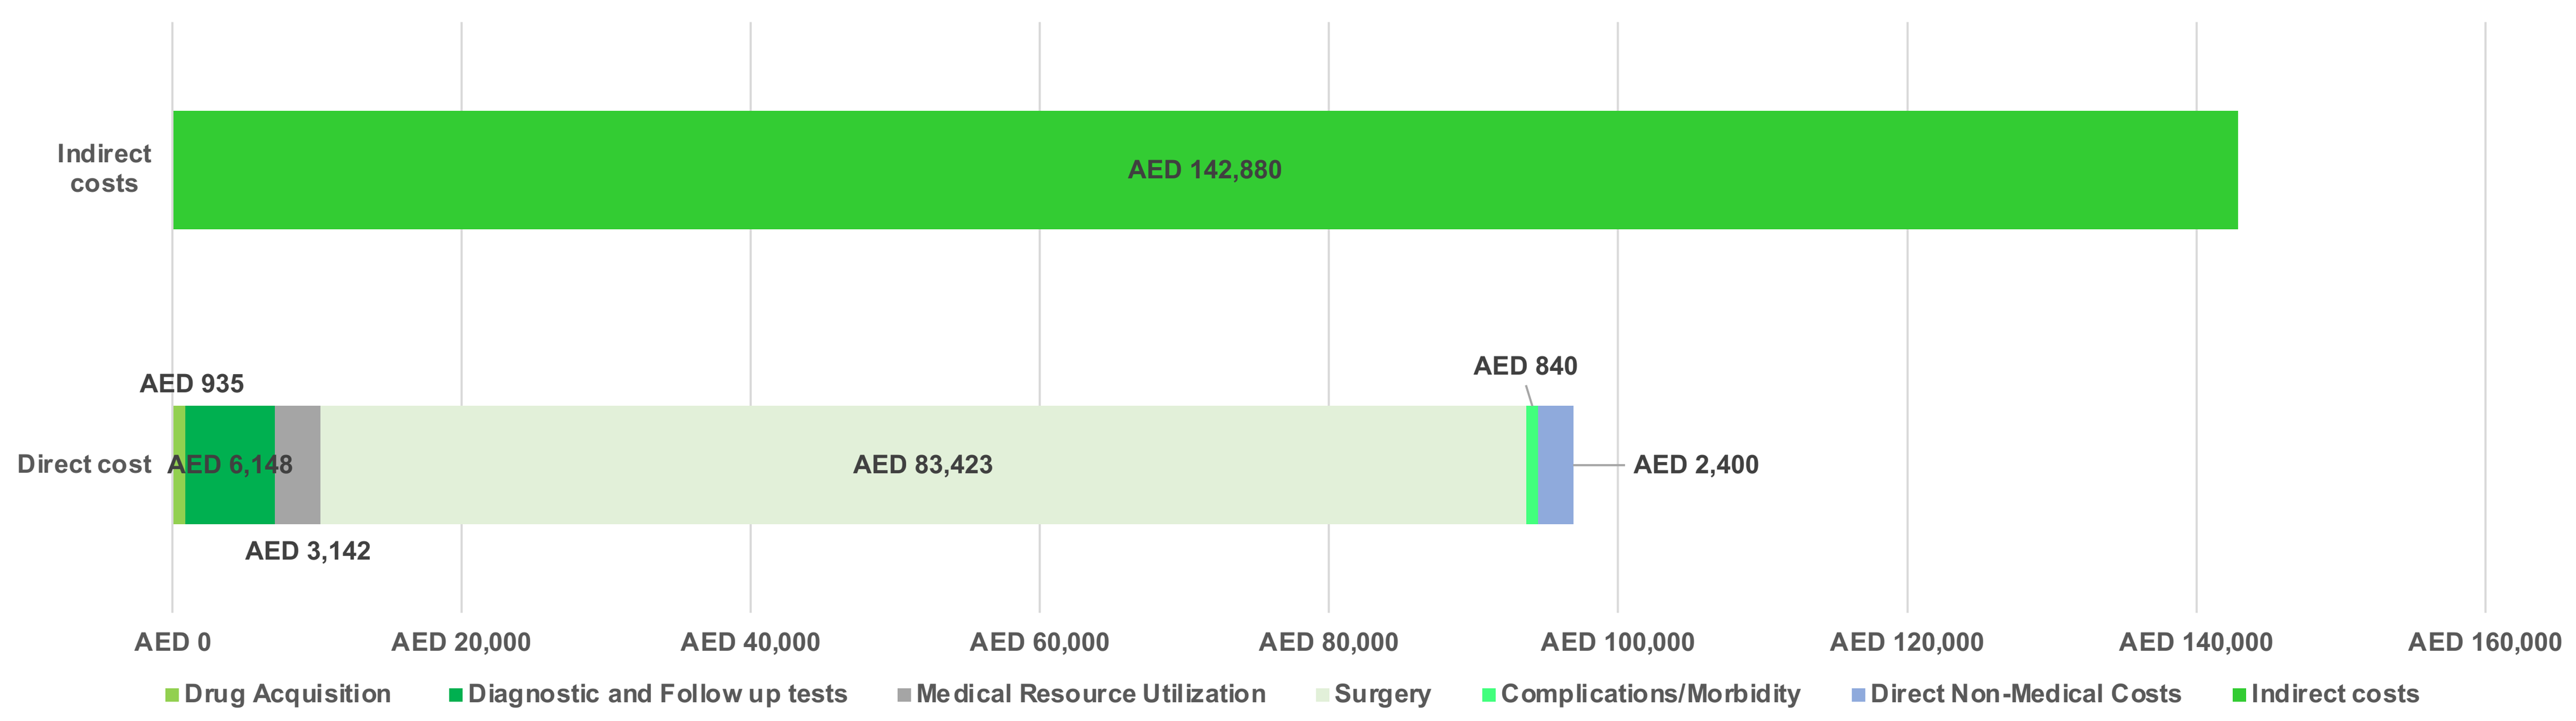
Indirect costs
AED 142,880
Direct cost
AED 935
AED 6,148
AED 3,142
AED 83,423
AED 840
AED 2,400
AED 0
AED 20,000
AED 40,000
AED 60,000
AED 80,000
AED 100,000
AED 120,000
AED 140,000
AED 160,000
Drug Acquisition
Diagnostic and Follow up tests
Medical Resource Utilization
Surgery
Complications/Morbidity
Direct Non-Medical Costs
Indirect costs

Figure 4. Total cost breakdown per patient for active severe to very severe disease

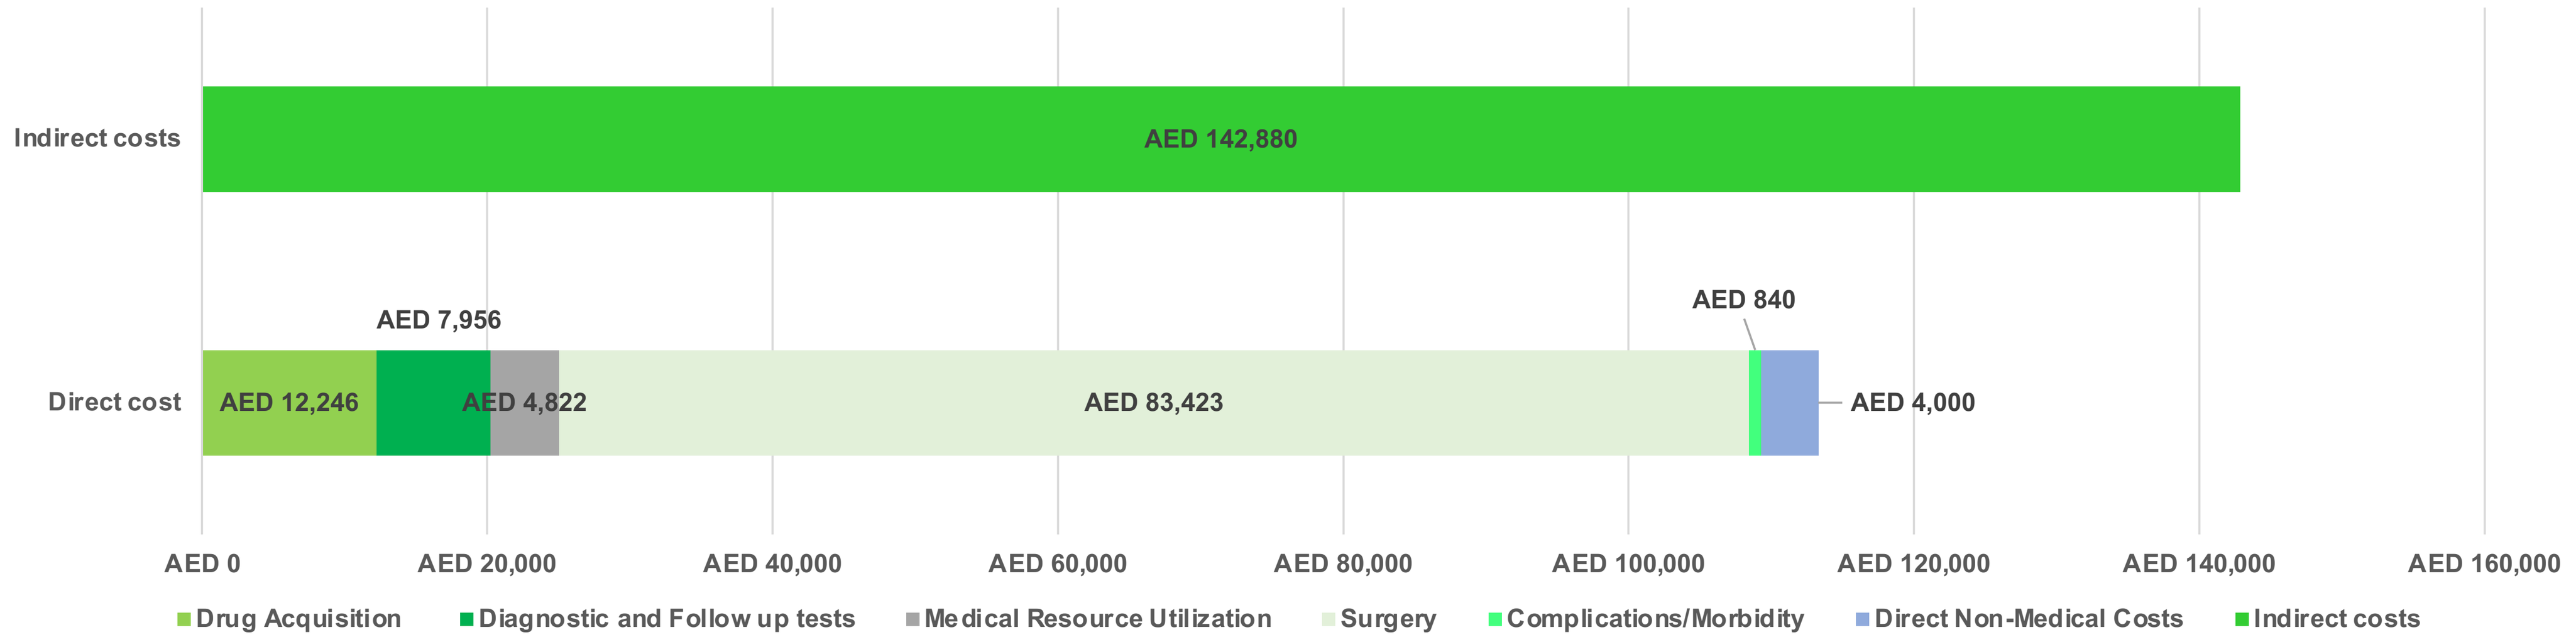
Indirect costs
AED 142,880
Direct cost
AED 12,246
AED 7,956
AED 4,822
AED 83,423
AED 840
AED 4,000
AED 0
AED 20,000
AED 40,000
AED 60,000
AED 80,000
AED 100,000
AED 120,000
AED 140,000
AED 160,000
Drug Acquisition
Diagnostic and Follow up tests
Medical Resource Utilization
Surgery
Complications/Morbidity
Direct Non-Medical Costs
Indirect costs

Conclusion

- The annualized per patient cost was positively associated with advancing disease severity with indirect parameters being the main cost driver across all disease states.
- Over 5 years, TED is estimated to impose a significant economic burden on the UAE patients and healthcare system.
- The findings postulate the considerable TED burden and provide valuable insights into key cost drivers for effective policymaking.

References

1. Dolman PJ. Evaluating Graves' orbitopathy. Best Pract Res Clin Endocrinol Metab 2012; 26(3): 229-48.
2. Patel A, Yang H, Douglas RS. A New Era in the Treatment of Thyroid Eye Disease. Am J Ophthalmol 2019; (208): 281-8.
3. Smith TJ. New advances in understanding thyroid-associated ophthalmopathy and the potential role for insulin-like growth factor-I receptor. F1000Res 2018; 7: 134.
4. Bahn RS, Heufelder AE. Pathogenesis of Graves' ophthalmopathy. N Engl J Med 1993; 329(20): 1468-75
5. Dickinson AJ, C.H., Clinical Manifestations. Graves' Orbitopathy: A Multidisciplinary Approach - Questions and Answers. Basel, Switzerland: Karger AG;. 2017: p. 1-23.
6. Lazarus, J.H. (2012) 'Epidemiology of graves' orbitopathy (go) and relationship with thyroid disease', Best Practice & Research Clinical Endocrinology & Metabolism, 26(3), pp. 273–279. doi:10.1016/j.beem.2011.10.005.

Presented at the [ISPOR 2024, Atlanta, GA, USA] | [05-2024]