Quantifying the relationship between time to diagnosis and potential system level burden in obstructive hypertrophic cardiomyopathy (HCM)

Poster code: EE172

Teresa Lemmer,[1] Kevin G. Pollock,[1] Michael Hurst,[1] Belinda Sandler,[1] Corinne Sadlowski,[1] Dimitra Lambrelli,[2] Harmony Omeife,[2] Robert Donaldson,[2] Ashwin Rai,[3] Faizel Osman[4]

[1]Bristol Myers Squibb, Uxbridge, UK; [2]Evidera Inc, London, UK; [3]Evidera Inc, Waltham, MA, US; [4]Institute for Cardiometabolic Medicine, University Hospitals Coventry and Warwickshire, Coventry, UK

Introduction

- Hypertrophic cardiomyopathy (HCM) is a chronic cardiovascular disease in which hypercontractility results in left ventricular hypertrophy and cardiac dysfunction[1,2]
- HCM is one of the most common cardiomyopathies, with an estimated prevalence rate of 1 in 500; however, expert consensus assumes a significant undiagnosed patient population, owing to non-specific symptom burden[3]
- Patients with a left ventricular outflow tract obstruction (LVOTO)[4] are categorized as living with obstructive HCM, with approximately two-thirds of patients living with the obstructive subtype[5]
- Prior studies have identified a strong relationship between increased symptomatic burden (denoted by increases in New York Heart Association [NYHA] class) and increases in system level burden and associated costs[6-8]
- The aim of this study is to explore the association between time to diagnosis and healthcare resource utilization (HRU) and the associated economic burden to understand potential savings that can be achieved through earlier diagnosis and improved clinical management

Methods

- A retrospective observational study was undertaken utilizing linked primary and secondary care data in the UK from the Clinical Practice Research Datalink (CPRD) and Hospital Episode Statistics (HES), respectively (ISAC protocol 21_000342 on March 4, 2022)
- Adult patients (aged >18 years) were included if they were eligible for HES linkage and had a diagnosis of obstructive HCM (defined as an International Classification of Diseases, Tenth Revision [ICD-10], Read, and/or Systematized Nomenclature of Medicine [SNOMED] code for obstructive HCM or a code for HCM with an additional code for LVOTO and/or septal reduction therapies [SRT]) between 2007 and 2019 with at least 1 year of continuous registration prior to index date (date of diagnosis)
- Patient records were extracted for all eligible patients, with a pre-index period starting from the date a patient started contributing data to CPRD (from April 1, 1997 onwards)
- When investigating HRU, the pre-index period was stratified into three distinct periods (**Figure 1**)
 - Period A, between date patient contributed to CPRD and 2 years prior to diagnosis
 - Period B, in the second year prior to diagnosis
 - Period C, in the year prior to diagnosis
- Event rates were summarized as mean per patient-year (PY)

Table 1. Baseline demographics and clinical characteristics

	Obstructive HCM population (N = 3,988)
Baseline demographics at index date	
Age, years	62.3 (16.0)
Gender: male	2,295 (57.5)
Ethnicity: White	3,071 (77.0)
Smoking status: current smoker	798 (20.0)
Region in England	
London	931 (23.3)
South-East	907 (22.7)
North-West	645 (16.2)
West Midlands	578 (14.5)
South-West	459 (11.5)
Other	468 (11.7)
Duration of pre-index registration, years	12.3 (6.1)
Clinical characteristics	
Clinical measurements (latest measurement in year prior to index)	
SBP, mm Hg	133.2 (18.4)
DBP, mm Hg	76.4 (11.0)
BMI, kg/m^2	29.3 (6.2)
Total cholesterol, mmol/L	4.7 (1.2)
HDL cholesterol, mmol/L	1.4 (0.4)
LDL cholesterol, mmol/L	2.7 (1.0)
Triglycerides, mmol/L	1.6 (1.1)
Family history (entire pre-index period)	
Obstructive HCM diagnosis	70 (1.8)
MI, HF, sudden death, or unexplained death	186 (4.7)
Diagnosis history (entire pre-index period)[a]	
Hypertension	2,600 (65.2)
Cardiac dysrhythmias	1,391 (34.9)
Coronary heart disease	1,359 (34.1)
Non-obstructive HCM diagnosis	1,259 (31.6)
Hypercholesterolemia	1,218 (30.5)

BMI, body mass index; DBP, diastolic blood pressure; HDL, high-density lipoprotein; HF, heart failure; LDL, low-density lipoprotein; MI, myocardial infarction; SBP, systolic blood pressure; SD, standard deviation.
[a]Shown when experienced by more than 30% of patients.
Note: Mean (SD) for continuous variables; n (%) for categorical variables.

Figure 1. Pre-index period

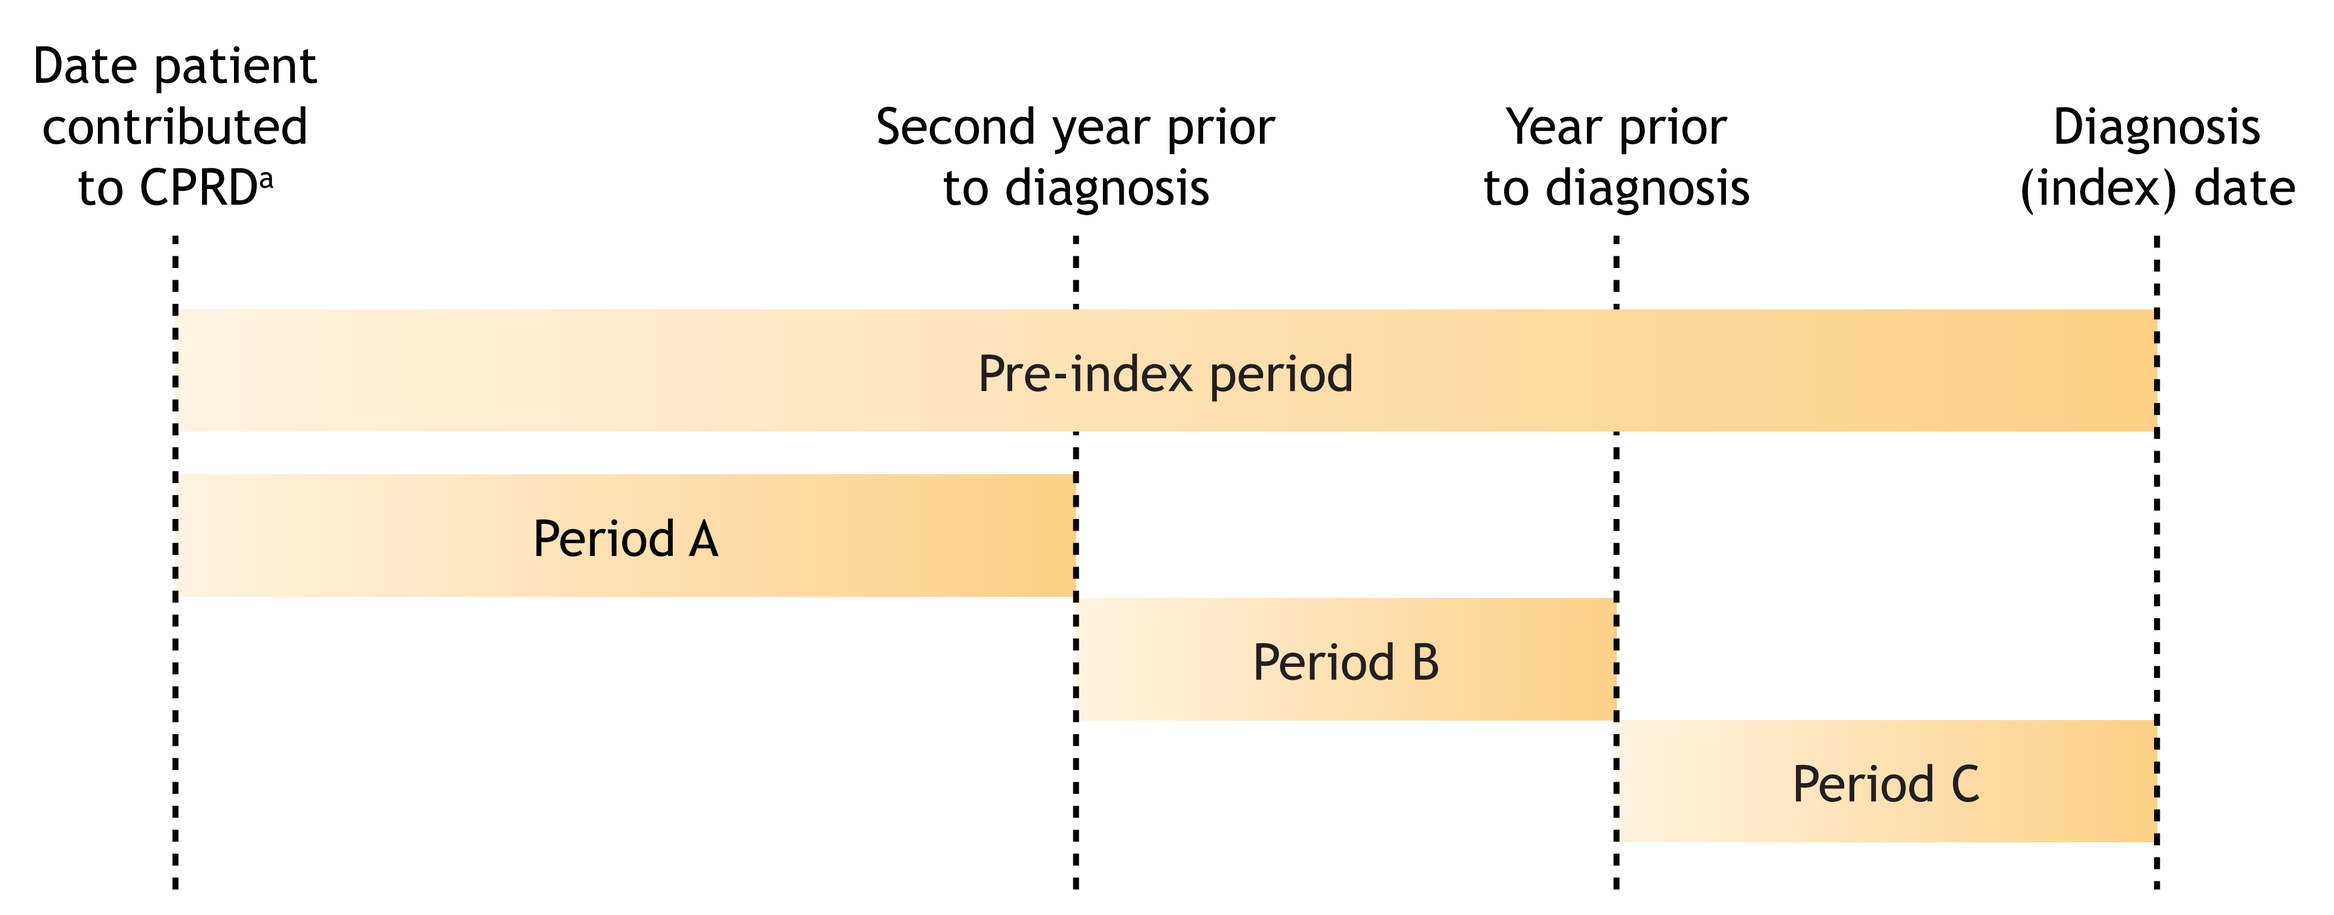
Date patient contributed to CPRD[a]
Second year prior to diagnosis
Year prior to diagnosis
Diagnosis (index) date
Pre-index period
Period A
Period B
Period C

[a]From April 1, 1997 onwards.

Results

Patient population

- Data from 10,316 patients with an obstructive HCM diagnosis who were registered with a practice that was eligible for HES linkage were extracted
- After applying study exclusion criteria, data from 3,988 patients were used in these analyses, contributing 48,988 PYs, equating to a mean (SD) pre-index period of 12.3 (6.1) years (interquartile range: 7.3-17.4 years)
- The mean age at index date of the patient population was 62.3 years and the majority were men (57.5%) and White (77.0%). London and South-East England contributed 46.0% of the eligible patient population (**Table 1**)
- Nearly two-thirds (65.2%) of patients had a code for hypertension prior to index date, with cardiac dysrhythmias (34.9%), coronary heart disease (34.1%), non-obstructive HCM (31.6%), and hypercholesterolemia (30.5%) present in over 30% of patients
- Chest pain (48.9%) and dyspnea (37.4%) were the most common symptoms patients presented with prior to diagnosis

Table 2. Healthcare resource utilization, stratified by intervals within the pre-index period

	All pre-index period (PYs: 48,988)	Period A (PYs: 41,124)	Period B (PYs: 3,876)	Period C (PYs: 3,899)
Primary care (all-cause)				
Primary care physician visits	6.40 (6.38-6.42)	5.52 (5.49-5.54)	9.57 (9.48-9.67)	12.42 (12.31-12.53)
Nurse visits	1.59 (1.58-1.60)	1.37 (1.36-1.39)	2.57 (2.52-2.62)	2.88 (2.82-2.93)
Secondary care (all-cause)				
Outpatient visits	3.75 (3.73-3.76)	3.09 (3.07-3.11)	6.06 (5.98-6.14)	8.27 (8.18-8.36)
Day case visits	0.43 (0.43-0.44)	0.36 (0.35-0.36)	0.64 (0.61-0.66)	1.03 (1.00-1.06)
Inpatient visits	0.31 (0.30-0.31)	0.23 (0.23-0.24)	0.44 (0.42-0.46)	0.93 (0.90-0.96)
Emergency visits	0.81 (0.80-0.82)	0.66 (0.65-0.66)	1.30 (1.27-1.34)	1.95 (1.90-1.99)
Cardiovascular-related tests and visits				
ECG visits	0.16 (0.15-0.16)	0.08 (0.08-0.08)	0.29 (0.27-0.30)	0.82 (0.79-0.85)
Pacemaker-related visits	0.04 (0.04-0.04)	0.02 (0.02-0.02)	0.08 (0.07-0.09)	0.20 (0.18-0.21)
CMR visits	0.02 (0.02-0.03)	0.01 (0.01-0.01)	0.04 (0.03-0.05)	0.20 (0.19-0.22)
Urea/electrolytes tests	0.69 (0.68-0.70)	0.59 (0.58-0.60)	1.07 (1.04-1.11)	1.36 (1.33-1.40)
LFTs	0.61 (0.60-0.62)	0.52 (0.52-0.53)	0.96 (0.93-0.99)	1.18 (1.14-1.21)
Cardiac monitoring visits	0.02 (0.02-0.02)	0.01 (0.01-0.01)	0.03 (0.02-0.03)	0.06 (0.05-0.07)
CPET visits	0.02 (0.01-0.02)	0.01 (0.01-0.01)	0.02 (0.01-0.02)	0.06 (0.06-0.07)
Biomarker[a] testing	0.02 (0.01-0.02)	0.01 (0.01-0.01)	0.03 (0.03-0.04)	0.08 (0.07-0.09)
Cardiac rehabilitation visits	0.00 (0.00-0.00)	0.00 (0.00-0.00)	0.00 (0.00-0.00)	0.00 (0.00-0.01)

BNP, B-type natriuretic peptide; CI, confidence interval; CMR, cardiovascular magnetic resonance; CPET, cardiopulmonary exercise test; ECG, electrocardiogram; LFT, liver function tests.
[a]Includes BNP and troponin T tests.
Note: Results presented as mean burden (95% Poisson CI) per PY.

Table 3. Costs associated with healthcare resource utilization, stratified by intervals within the pre-index period

	Unit costs (NHS [2021/22] and PSSRU [2022])	All pre-index period (PYs: 48,988)	Period A (PYs: 41,124)	Period B (PYs: 3,876)	Period C (PYs: 3,899)
Primary care (all-cause)		**275 (274-276)**	**237 (236-238)**	**413 (409-417)**	**532 (527-537)**
Primary care physician visits	41.00[9]	262 (262-263)	226 (225-227)	392 (389-396)	509 (505-514)
Nurse visits	7.99[9]	13 (13-13)	11 (11-11)	21 (20-21)	23 (23-23)
Secondary care (all-cause)		**4,026 (3,945-4,045)**	**3,140 (3,118-3,216)**	**6,002 (5,789-6,208)**	**10,807 (10,510-11,098)**
Outpatient visits	235.00[9]	881 (877-884)	726 (721-731)	1,424 (1,405-1,443)	1,943 (1,922-1,965)
Day case visits	1,224.00[9]	526 (526-539)	441 (428-441)	783 (747-808)	1,261 (1,224-1,297)
Inpatient visits	7,076.00[9]	2,194 (2,123-2,194)	1,627 (1,627-1,698)	3,113 (2,972-3,255)	6,581 (6,368-6,793)
Emergency visits	523.99[9]	424 (419-430)	346 (341-346)	681 (665-702)	1,022 (996-1,043)
Cardiovascular-related tests and visits		**267 (257-277)**	**147 (138-155)**	**502 (453-555)**	**1,277 (1,198-1,360)**
ECG visits	351.36[10]	55 (54-56)	28 (27-29)	100 (95-106)	287 (278-298)
Pacemaker-related visits	3,989.97[10]	160 (153-168)	85 (79-91)	323 (288-361)	779 (726-836)
CMR visits	553.96[10]	14 (13-15)	3 (3-4)	22 (19-26)	112 (105-120)
Urea/electrolytes tests	20.00[11,b]	14 (14-14)	12 (12-12)	21 (21-22)	27 (27-28)
LFTs	20.00[11,b]	12 (12-12)	10 (10-11)	19 (19-20)	24 (23-24)
Cardiac monitoring visits	192.29[10]	3 (3-3)	2 (2-2)	5 (4-6)	12 (10-13)
CPET visits	487.05[10]	8 (7-8)	5 (5-6)	9 (7-11)	31 (28-35)
Biomarker[a] testing	20.00[11]	0 (0-0)	0 (0-0)	1 (1-1)	2 (1-2)
Cardiac rehabilitation visits	483.98[10]	1 (1-1)	0 (0-1)	1 (0-2)	2 (1-4)
Total costs		**4,567 (4,476-4,599)**	**3,524 (3,492-3,609)**	**6,917 (6,652-7,180)**	**12,616 (12,236-12,995)**

NHS, National Health Service; PSSRU, Personal Social Services Research Unit.
[a]Includes BNP and troponin T tests. [b]Unit cost assumed equivalent to BNP testing.
Note: Results presented as £ (95% Poisson CI) per PY.

Figure 2. Healthcare resource utilization, stratified by intervals within the pre-index period and setting

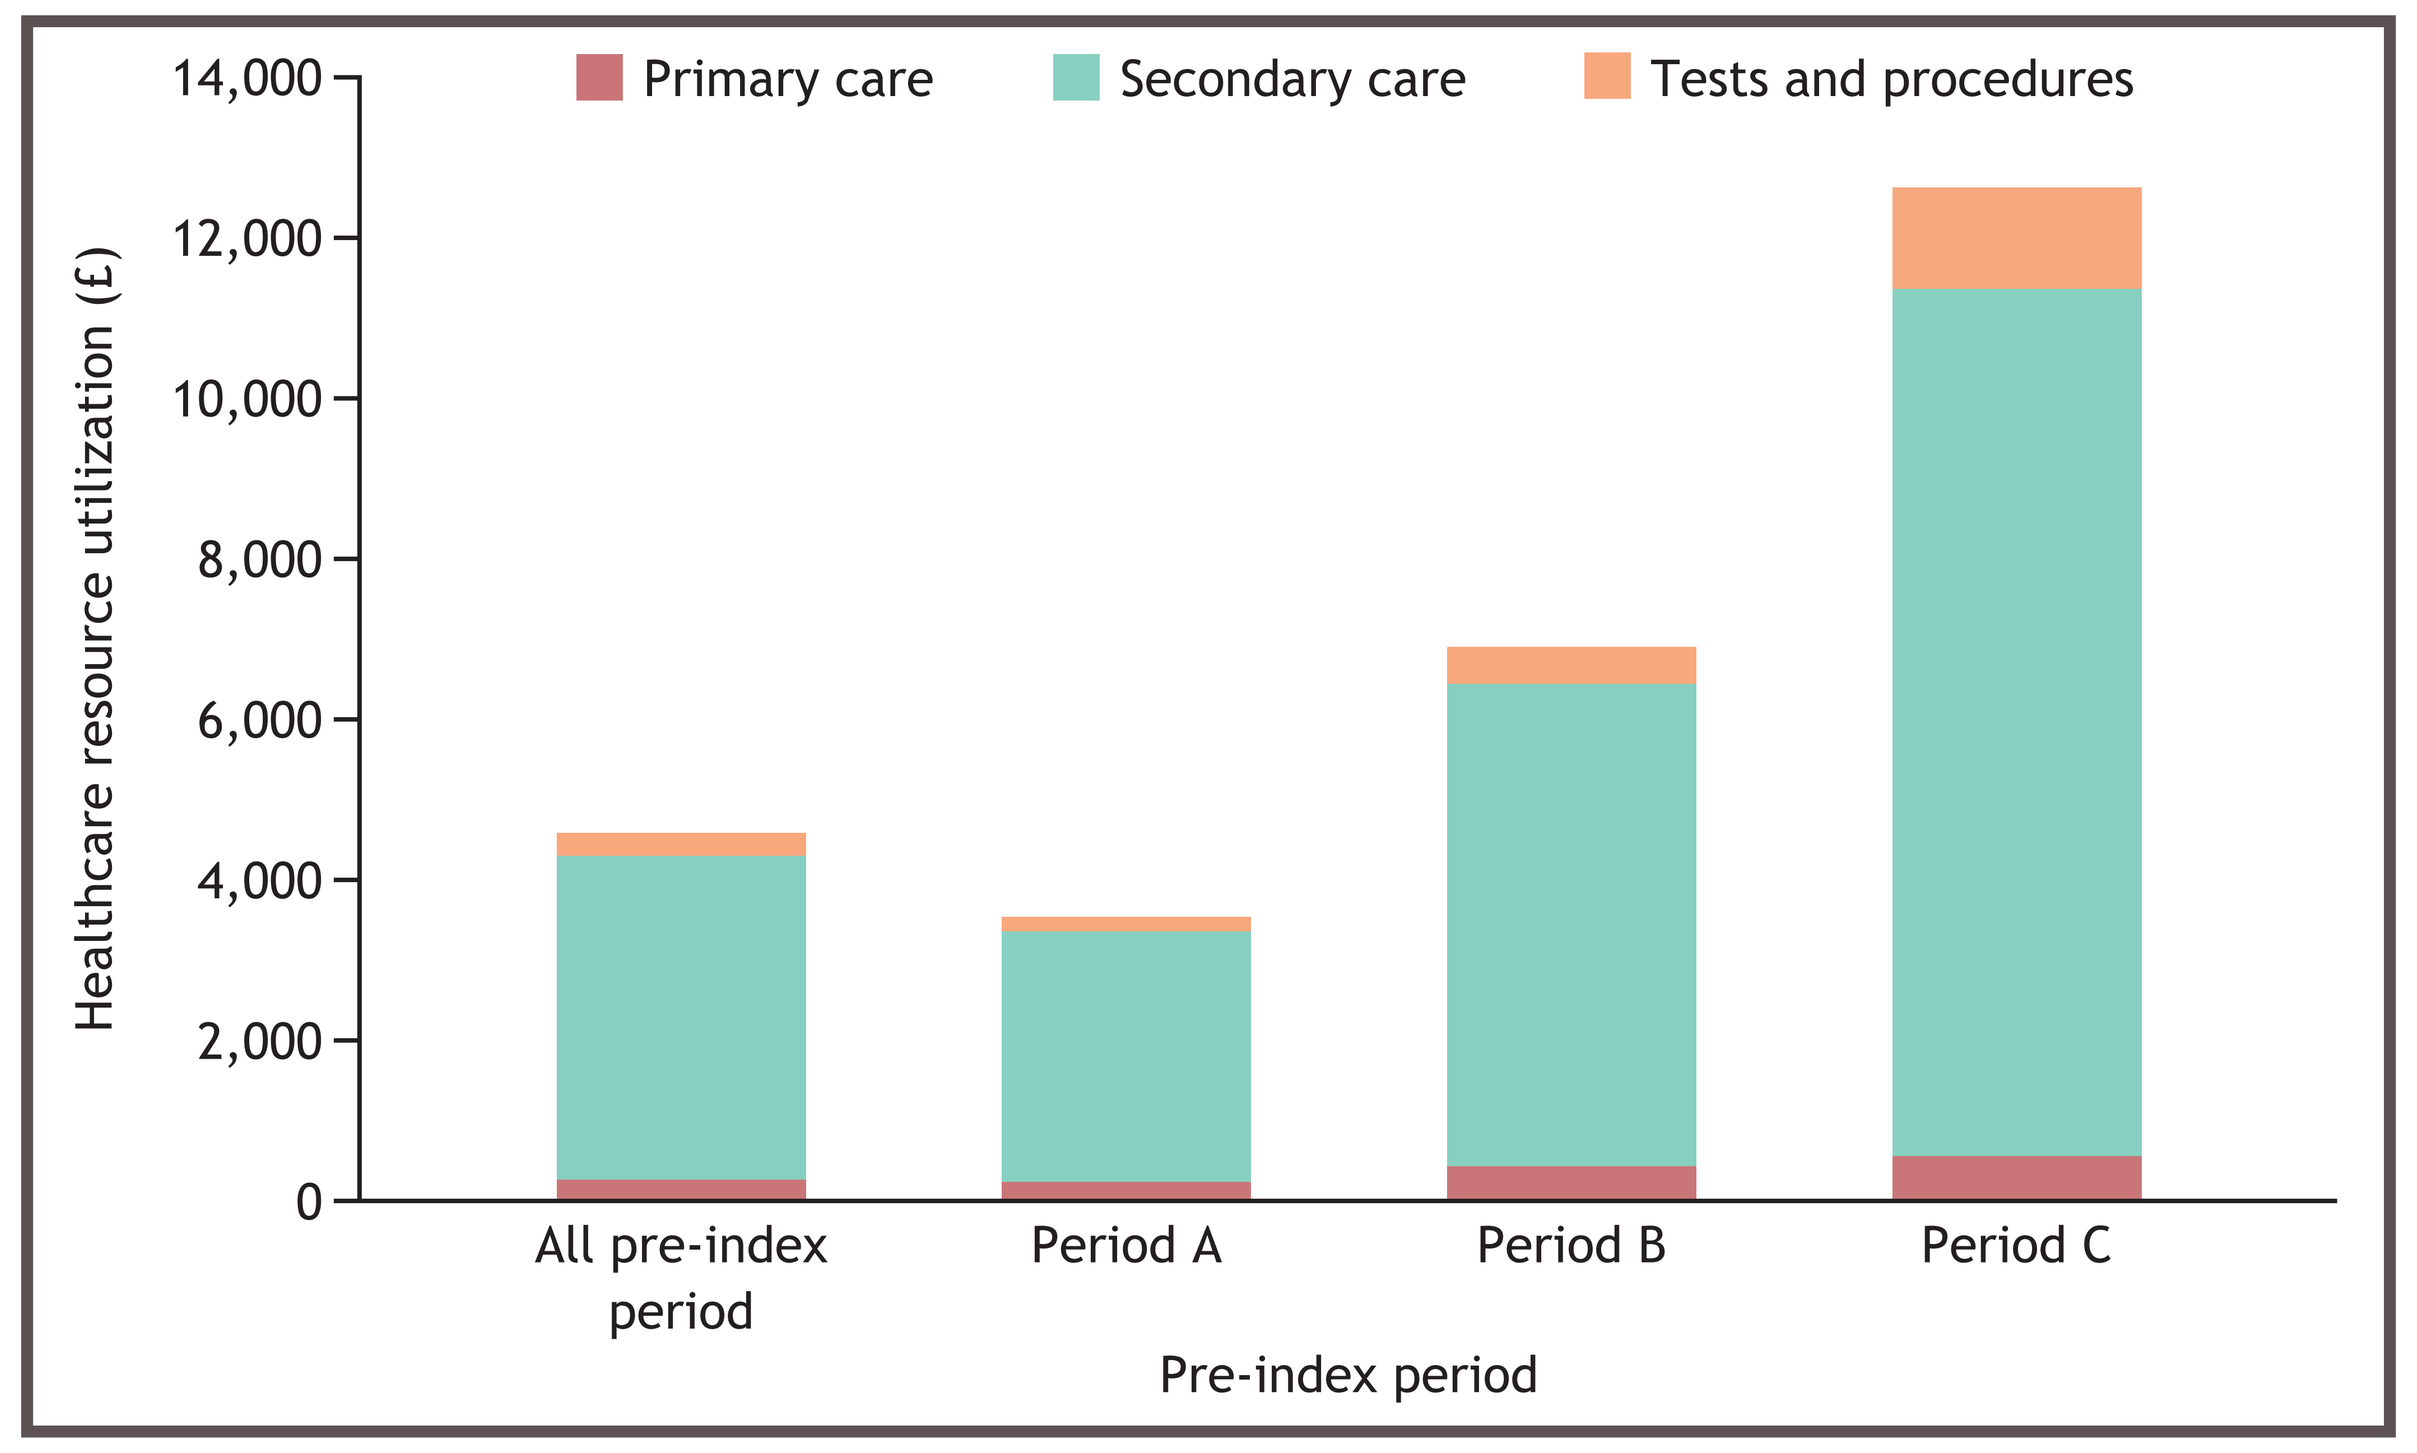
Primary care
Secondary care
Tests and procedures
Healthcare resource utilization (£)
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
All pre-index period
Period A
Period B
Period C
Pre-index period

Healthcare resource utilization

- Considering the entirety of the pre-index period, patients visited their primary care physician 6.40 (95% CI: 6.38–6.42) times, had 3.75 (3.73–3.76) outpatient visits, and had 0.69 (0.68–0.70) tests for urea and electrolytes (a measure of kidney function) per PY (**Table 2**)
- When compared with the Period A, patient contact with the system increased in Period B and increased further in Period C
- Substantial increases in HRU (defined as non-overlapping CIs) over time were observed in all primary care and secondary care interactions studied, with inpatient contact increasing by 91% and 304% for Period B and Period C, respectively, when compared with Period A (0.44 and 0.93 vs 0.23 contacts PYs)

Figure 3. Healthcare resource utilization, stratified by intervals within the pre-index period and resources within secondary care

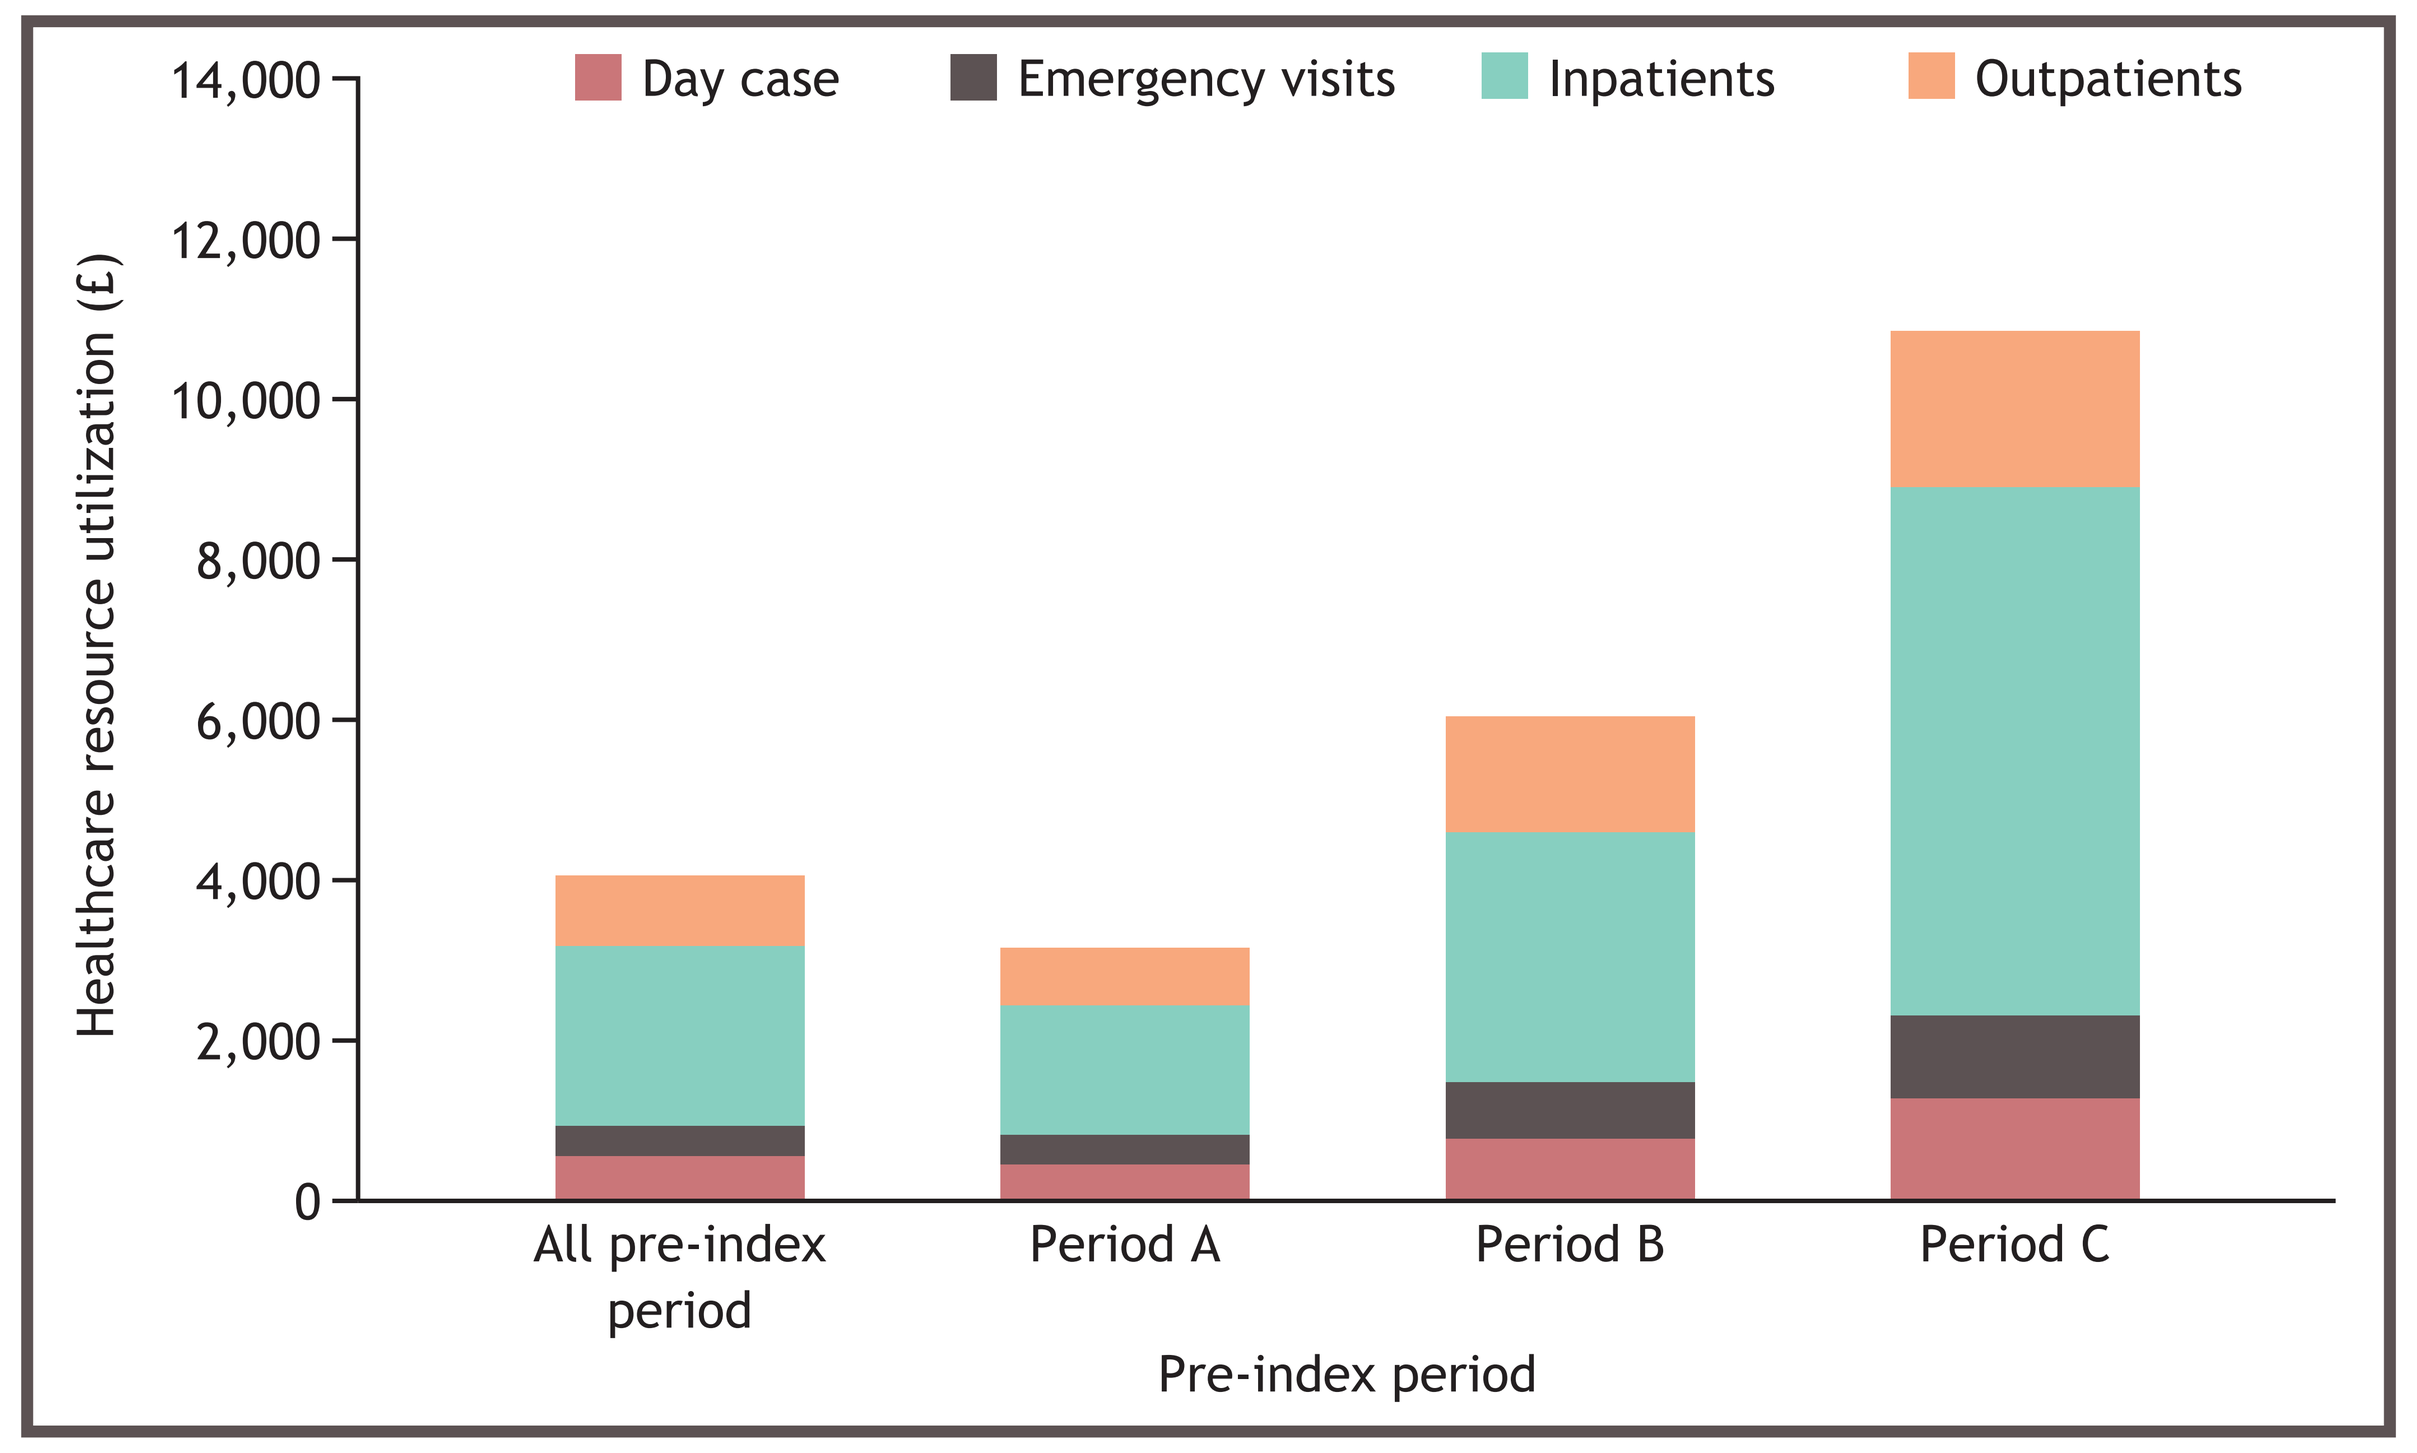
Day case
Emergency visits
Inpatients
Outpatients
Healthcare resource utilization (£)
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
All pre-index period
Period A
Period B
Period C
Pre-index period

- Increased activity linked to cardiovascular-related tests and visits, specifically ECG-related visits, was also observed in the Period B and Period C, with an average (95% CI) of 0.29 (0.27–0.30) and 0.82 (0.79–0.85) visits per PY, a greater than 2-fold and 9-fold increase, respectively, when compared with Period A

System-level economic burden

- The mean economic burden per PY for the entirety of the pre-index period was £4,567 (95% CI: £4,476–£4,599) (**Table 3**, **Figure 2**)
- A significant increase in burden per PY was observed closer to diagnosis, with a cost (95% CI) of £6,917 (£6,652–£7,180) and £12,616 (£12,236–£12,995) in Period B and Period C, respectively, compared with £3,524 (£3,492–£3,609) in Period A
- Economic burden was driven by secondary care, independent of time to diagnosis, with secondary HRU accounting for 85–90% of the total burden throughout (**Figure 3**)
- In Period C, inpatient costs were £6,581 (95% CI: £6,368–£6,793) per PY, accounting for over half of the total economic burden of patients

Limitations

- Linked data are only available for patients registered with a primary care physician in England who are linkage-eligible. Despite this, the data are generally considered generalizable to the UK population; however, they may not be generalizable to other geographies
- Owing to the nature of retrospective data, this study has inherent limitations, including missing data and inconsistencies in coding and data entry
- This analysis for primary and secondary care captured all-cause HRU and associated costs, and as such included some non-HCM-related HRU
- As a descriptive retrospective study, this study does not imply that expedited diagnosis reduces HRU and associated economic burden, with additional research required to assess this impact

Conclusions

- This study is the first to demonstrate that a patient's pathway to diagnosis can be potentially associated with significant system-level HRU and associated economic burden
- Further research is needed to understand whether similar outcomes are observed in indirect costs and whether early proactive identification, diagnosis, and appropriate clinical management could realize the reduction of societal burdens

References

1. Ommen SR, et al. J Am Coll Cardiol. 2020;76(25):e159-e240.
2. Elliot PM, et al. Eur Heart J. 2014;35(39):2,733-79.
3. Maron BJ, et al. Circulation. 1995;92(4):785-9.
4. Schultze M, et al. Eur Heart J. 2022;43(Supplement_2):1,747.
5. Maron MS, et al. N Engl J Med. 2003;348(4):295-303.
6. Hurst M, et al. Value Health. 2022;25(12):S151.
7. Charron P, et al. Value Health. 2023;26(12):S80.
8. Osman F, et al. Value Health. 2023;26(12):S50.
9. Personal Social Services Research Unit. Unit Costs of Health and Social Care programme (2022-2027). Available at: https://www.pssru.ac.uk/unitcostsreport/. Accessed January 31, 2024.
10. NHS England. National Cost Collection for the NHS. Available at: https://www.england.nhs.uk/costing-in-the-nhs/national-cost-collection/#ncc1821. Accessed January 31, 2024.
11. NHS Improvement. Evidence for the introduction of Serum NP testing to the diagnostic pathway for Heart failure. Available at: https://www.nice.org.uk/media/default/sharedlearning/408_408evidencefortheintroofserumnptesting.doc. Accessed January 31, 2024.

Acknowledgments

- This work was supported by Bristol Myers Squibb.
- All authors contributed to and approved the poster; editorial assistance was provided by Nicolas Bertheleme of Oxford PharmaGenesis, funded by Bristol Myers Squibb.

Disclosures

- Teresa Lemmer, Kevin G. Pollock, Michael Hurst, Belinda Sandler, and Corinne Sadlowski are employees of Bristol Myers Squibb and own stock in Bristol Myers Squibb. Dimitra Lambrelli, Harmony Omeife, Robert Donaldson, and Ashwin Rai are employees of Evidera and have received funding from Bristol Myers Squibb for the study conduct. Faizel Osman has no disclosures.

Presented at: ISPOR 2024; May 5–8, 2024; Atlanta, GA, USA

www.globalbmsmedinfo.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.