DIGITAL ENGAGEMENT TO CAPTURE INSIGHTS INTO PATIENT PERSPECTIVES ON CARE AND QUALITY OF LIFE: UTILIZING AN ONLINE SURVEY TO UNDERSTAND THE BURDEN ON PATIENTS POST-ACUTE MYOCARDIAL INFARCTION

PCR3

M. Cecilia Bahit[1,2], Serge Korjian[2,3], Gerald Chi[3], Yazan Daaboul[2,3], Ginger Jiang[2,3], Deepak L. Bhatt[4], Roxana Mehran[4], Paul Nara[5], **Alka Shaunik[5]**, C. Michael Gibson[2,3]

[1]INECO Neurociencias, Rosario, Argentina; [2]Baim Institute for Clinical Research, Boston, MA, USA; [3]Beth Israel Deaconess Medical Center, Harvard Medical School, Boston, MA, USA; [4]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [5]CSL Behring, King of Prussia, PA, USA.

Background and objectives

- Patients who have had an acute myocardial infarction (AMI) are at a very high risk of having recurrent cardiovascular (CV) events in the following year[1]
- Given the need for long-term lifestyle changes and post-AMI care, patients often experience reduced health-related quality of life[2,3], which rarely returns to pre-infarction levels
- The importance of tailored care to address patient preferences has been increasingly recognized in recent years[4]
- Here, we designed an online survey designed to gather patient perspectives post-AMI in order to assess how digital engagement can identify treatment preferences/goals

Methods

- This survey was designed by an advisory board of medical experts and patients, and hosted on an online patient platform, collecting GDPR-compliant data
- Adults in the USA who had an AMI in the previous 12 months were recruited via the patient platform and associated online partners
- Patients who were members of the online community were invited to participate in the survey via email and received weekly digital reminders to complete it
- The survey approved by an institutional review board (IRB), it was unincentivized and self-administered, without direct influence of a clinician or interviewer
- Survey questions were defined under five topics, as shown in **Figure 1**
- Target enrollment was 150 patients

Results

- Recruitment was completed within a two-month time period, between October and December 2022
- In total, 151 patients completed the survey
- Basic demographic characteristics are shown in **Figure 2**
 - Of the respondents, 58 (38%) patients had their latest AMI in the past 90 days (**Figure 3**), indicating that patient engagement is feasible in the early period post-AMI
- Although survey studies can be subject to different types of bias (e.g., recall bias) several strategies were employed to reduce the impact of bias on the results (**Figure 4**)

Figure 1. Survey question topics

1. Screening

- Age/gender
- Residence
- Experience of an AMI
- Number of AMIs experienced
- Date of first AMI
- Date of last AMI

2. Medical profile

- Comorbidities
- Treatments received for comorbidities

3. Patient pathway

- Surgery required for the AMI
- Hospitalization/length of stay for AMI
- Treatments received after discharge
- Duration of treatment
- Perceptions of/involvement in cardiac rehabilitation program
- Experience of complications as a result of AMI
- HCPs providing follow-up care for AMI, etc.

4. Burden of AMI and unmet needs

- Most prominent feelings immediately after AMI and at time of survey completion
- Impact of AMI on patients' life/diet/physical activities
- Current employment status
- Impact of AMI on the professional life immediately after AMI and at time of survey completion
- Impact of AMI on household finances and social/family-related spending
- Out-of-pocket costs related to AMI

5. Treatment burden/expectations of new treatments

- Perceived benefits and drawbacks of current treatments
- Perceived treatment burdens
- Perceived changes in adherence between hospitalization and the current situations
- Expectations regarding new treatment
- Perceived relevant features of a new treatment
- Preferred treatment regimen for a new treatment
- Perceptions of digital services, etc.

AMI, acute myocardial infarction; HCP, healthcare professional

Conclusions

- **Online surveys addressing post-AMI patient perspectives are feasible and can be a viable tool to better understand patient needs**
- **Future bigger and broader surveys in this area are needed to ensure patient preferences are addressed in their care**

Figure 2. Key demographics

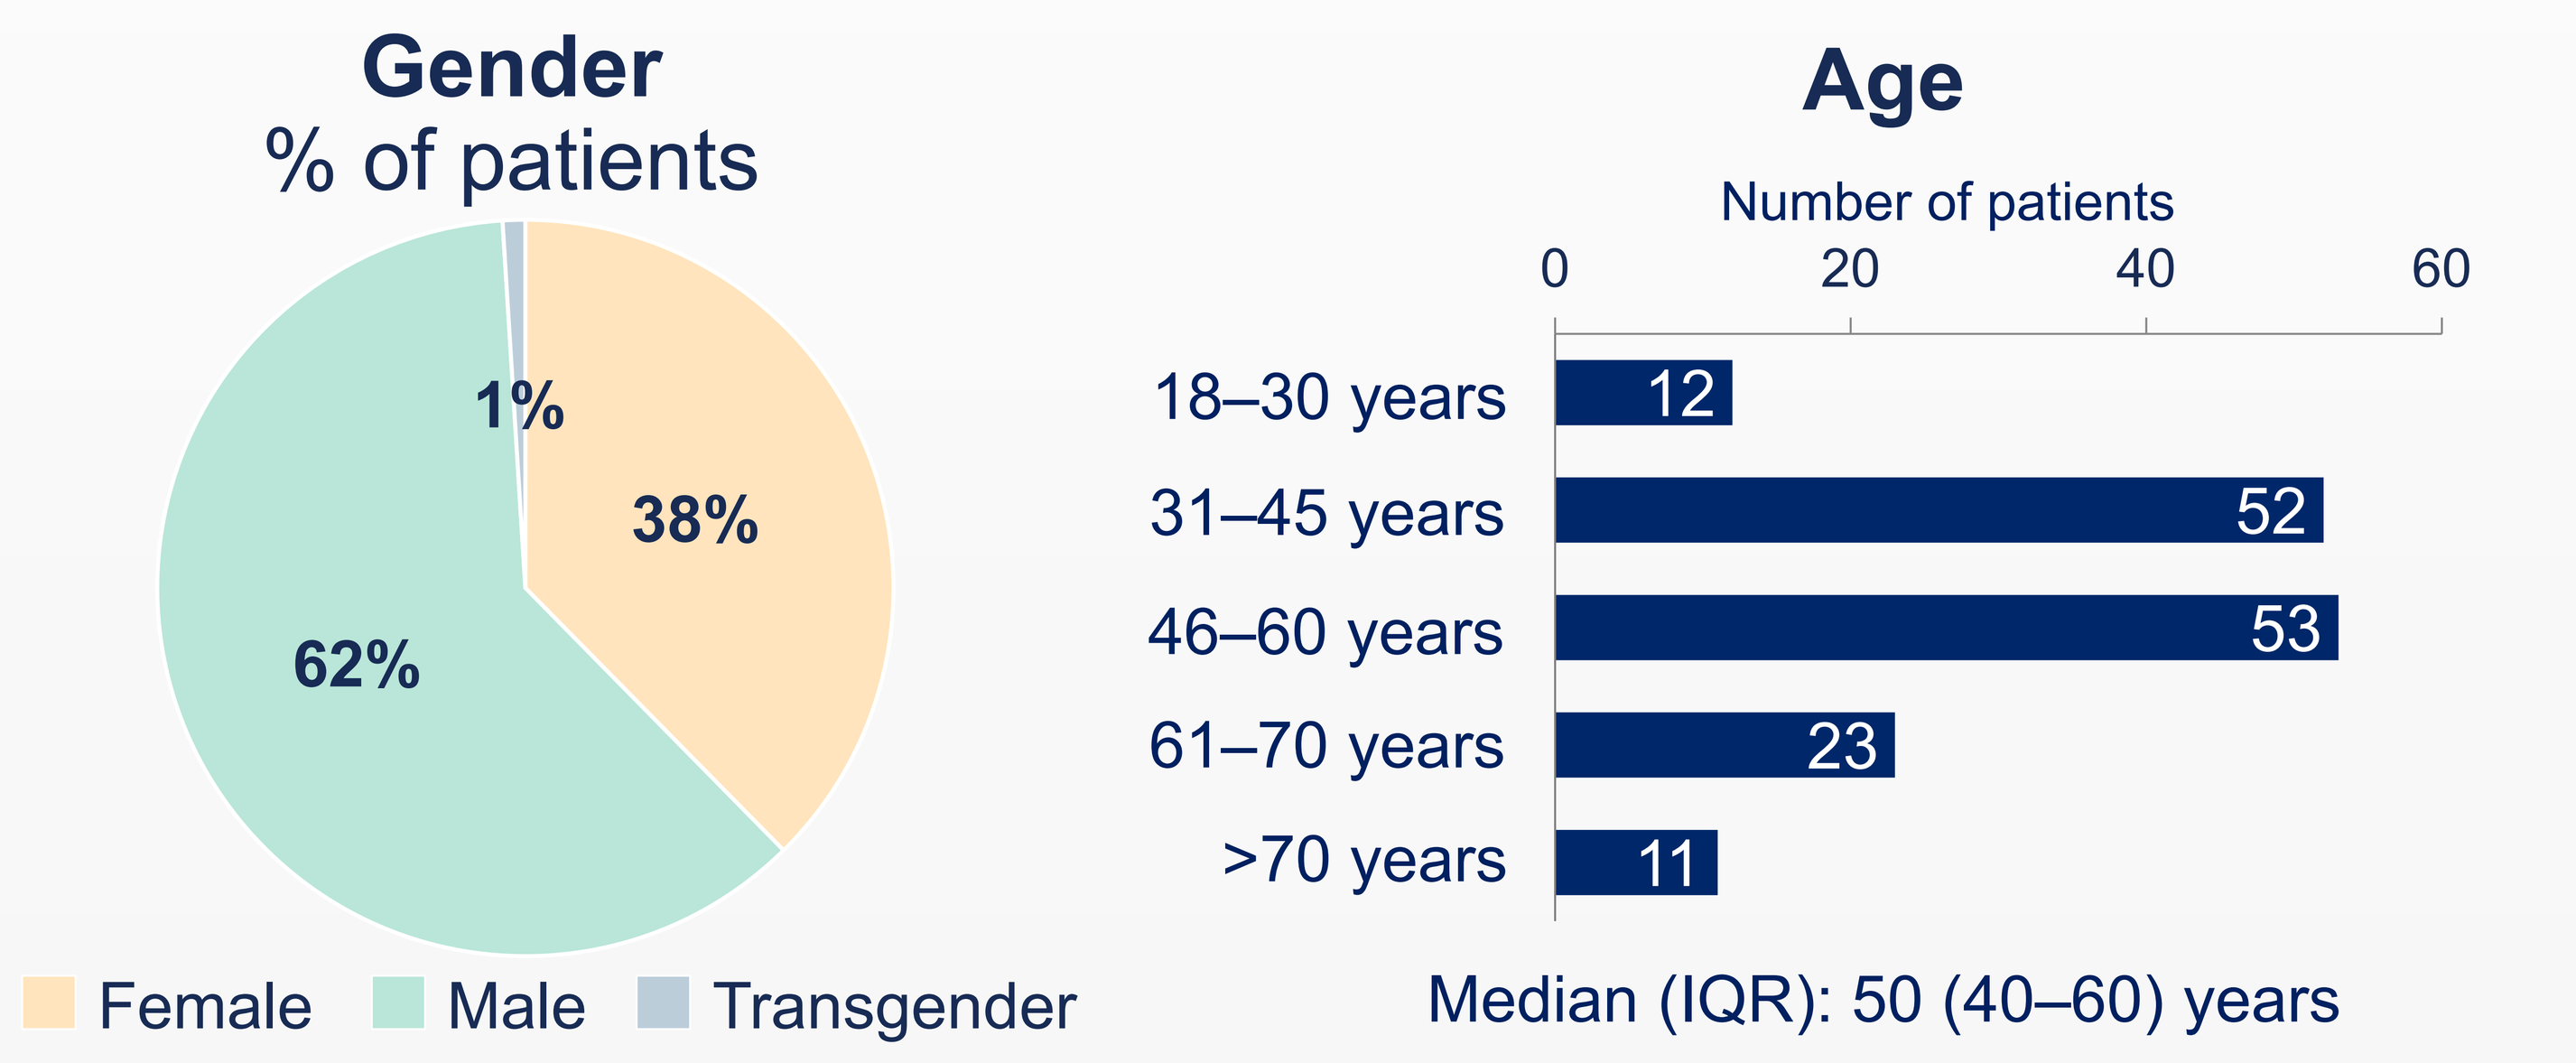
Gender
% of patients
1%
38%
62%
Female
Male
Transgender
Age
Number of patients
0
20
40
60
18–30 years
12
31–45 years
52
46–60 years
53
61–70 years
23
>70 years
11
Median (IQR): 50 (40–60) years

IQR, interquartile range

Figure 3. Patient AMI characteristics

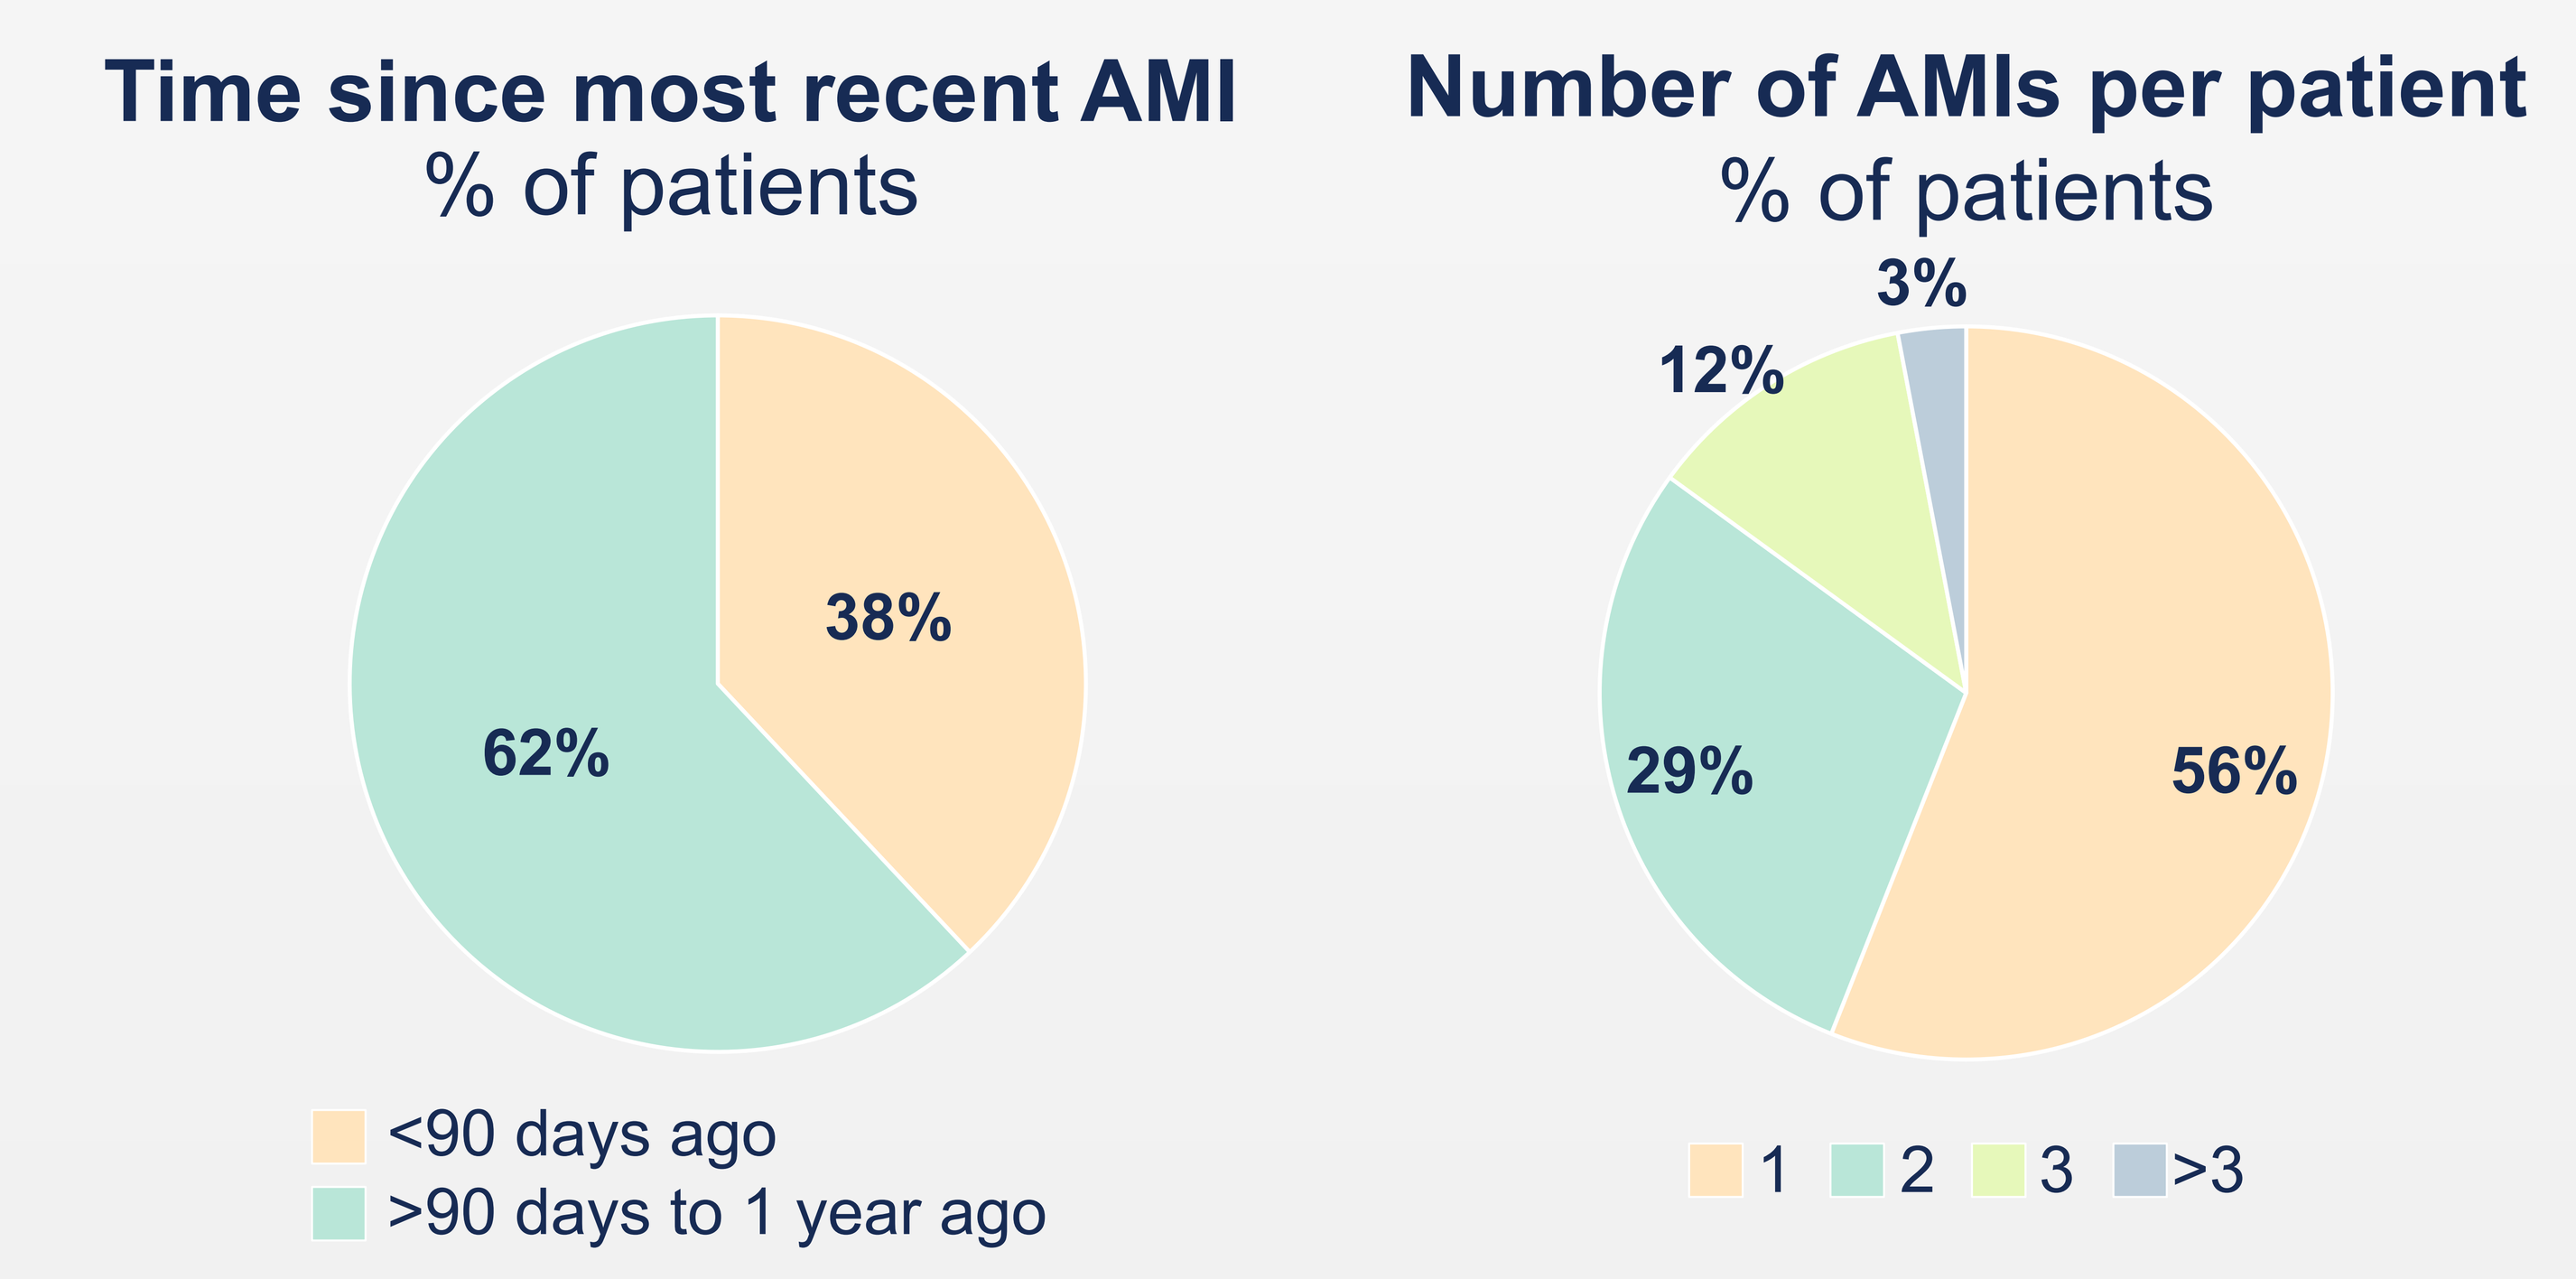
Time since most recent AMI
% of patients
38%
62%
<90 days ago
>90 days to 1 year ago
Number of AMIs per patient
% of patients
3%
12%
29%
56%
1
2
3
>3

AMI, acute myocardial infarction

Figure 4. Strategies used to minimize the impact of bias commonly associated with survey studies

Social desirability bias
- Patients were not questioned by HCPs or an interviewer, but answered the e-survey themselves

Recall bias
- Patients were aimed to be recruited within 90 days and always to within 1 year of the AMI episode

Selection bias
- Different channels of recruitment were used to avoid IT bias associated with online users

AMI, acute myocardial infarction; HCP, healthcare professional

References:

1. Chi G, et al. Clin Cardiol. 2022;45(3):299–307; 2. Pocock S, et al. Open Heart. 2021;8(1):e001499; 3. Doll JA. Heart. 2020;106(1):8–9; 4. Tringale M, et al. BMJ Open. 2022;12(11):e067268.

Presenter disclosures:

The study was sponsored by CSL Behring in collaboration with the Baim Institute.
Dr. Shaunik is an employee of CSL Behring.

Poster presented at The Professional Society for Health Economics and Outcomes Research (ISPOR), Atlanta, GA, May 5–8, 2024.