EE68

A UK Cost-utility Analysis of Ferric Carboxymaltose versus Ferric Derisomaltose in Patients with Iron Deficiency Anemia and Inflammatory Bowel Disease: Incorporation of Fracture Data

Zoller H[1], Pammer LM[1], Nottmeier M[2], Pöhlmann J[3], Pollock RF[3]

[1] Medizinische Universität Innsbruck, Innsbruck, Austria [2] Pharmacosmos A/S, Holbæk, Denmark [3] Covalence Research Ltd, Harpenden, UK

Background

In the United Kingdom (UK), up to 725 per 100,000 people in primary care have inflammatory bowel disease (IBD), implying that there are hundreds of thousands of IBD cases in the UK [1]. The incidence of IBD ranks among the highest globally, ranging between 28.5–69.5 cases per 100,000 people. Substantial increases in incidence have been observed in recent years, most notably in adolescents in whom incidence increased by 94%, or approximately 3% per year, between 2000 and 2018 [1,2]. Iron deficiency (ID) and iron deficiency anemia (IDA) is one of the most common extraintestinal manifestations of IBD, which severely affects comorbidity and quality of life (QoL) and is often attributable to iron deficiency [3]. Regular monitoring for anemia is strongly recommended [3,4] but, given the frequency with which IDA occurs, needs to be complemented with effective treatment options.

Iron replacement is the cornerstone of ID/IDA treatment. Oral iron is typically the first-line treatment but, due to low treatment adherence, frequent side effects, and insufficient iron replenishment in more severe cases, intravenous (IV) iron is often recommended [3–5]. Several IV iron formulations are available, differing in their posology and safety profile. While hypersensitivity reactions are uncommon with all modern high-dose iron formulations, the formulations differ in their propensity to induce symptomatic hypophosphatemia and bone complications including osteomalacia and fractures [6–8]. Understanding and characterizing these differences is important for patients but also, given their effects on costs and QoL, for payers trying to establish the cost-effectiveness of the IV iron formulations. In the UK, a recent cost-utility analysis (CUA) comparing ferric derisomaltose (FDI) with ferric carboxymaltose (FCM) showed that fewer FDI infusions were needed, and hypophosphatemia occurred significantly less frequently with FDI versus FCM, leading to gains in quality-adjusted life expectancy (QALE) at lower costs and establishing FDI as the dominant treatment option in patients with IBD and IDA [9].

The present analysis extended the CUA by Iqbal *et al.* [9] by also incorporating recent data on the clinical consequences of hypophosphatemia — namely risk of fractures with FDI and FCM — to contribute to a more complete understanding of the relative merits of these IV iron formulations in treating IDA in people with IBD.

Methods

The analysis was based on a previously published, patient-level, discrete-time, decision analytic model implemented in MS Excel that captured first- and second-order uncertainty as well as patient heterogeneity [9,10]. The PHOSPHARE-IBD randomized controlled trial (RCT) informed the risk of hypophosphatemia associated with each treatment and equivalent hematological response [7]. Characteristics of the modelled patient cohort were also taken from the PHOSPHARE-IBD RCT.

Data on fracture incidence in 110 and 179 Austrian patients with ID treated with FDI and FCM, respectively, were used to derive parametric survival models of fracture-free survival after the first IV iron administration [11]. Eight distributions – exponential, gamma, generalized gamma, generalized F, Gompertz, Weibull, log-normal, and log-logistic – were evaluated using standard goodness-of-fit criteria (Akaike Information Criterion [AIC] and Bayesian Information Criterion [BIC]). The best-fitting distribution was chosen to model fracture rates used in the CUA and predict fractures for up to 20 years. All analyses were performed in R v4.3.2.

Costs of iron and phosphate infusions as well as costs associated with fractures were taken from the 2021/22 National Schedule of National Health Service (NHS) costs, weighted by activity (Table 1). Drug costs were sourced from the British National Formulary, including for IV iron and oral phosphate, while phosphate test costs were obtained from the Personal Social Services Research Unit (PSSRU). Data pertaining to QoL, including disutilities associated with fracture, were obtained from the literature and a prior National Institute for Health and Care Excellence technology appraisal (TA464).

The analysis was conducted from a societal perspective, over a 10-year time horizon and costs and benefits were discounted at 3.5% per annum.

Table 1. Unit costs employed in the base case analysis

Item	Cost (£)	Source
Drug costs		
FDI, per gram	169.50	British National Formulary
FCM, per gram	167.60	British National Formulary, weighted using Secondary Care Medicines Data
Oral phosphate, per dose	0.16	British National Formulary
Procedure and test costs		
IV iron administration, per administration	322.09	Activity-weighted costs across HRG SA04G–H, SA04J–L
Phosphate infusion, per infusion	304.00	Activity-weighted costs across HRG KC05M–N
Serum phosphate test, per test	12.50	£4.00 per laboratory test + 10 minutes Band 6 nurse time at £51.00 per hour (PSSRU)
Event costs		
Average cost of fracture treatment	4,981.97	Weighted fracture HRGs across all fractures in input dataset, scaled based on 61% of fracture treatment costs occurring in the index year

FDI, ferric derisomaltose; FCM, ferric carboxymaltose; HRG, healthcare resource group; PSSRU, Personal Social Services Research Unit.

Results

Fracture rates were similar before and after treatment with FDI, while fracture rates were higher after treatment with FCM. When censored at the first fracture event, the observed fracture rates were lower with FDI (0.455 per 100 person-years) versus those treated with FCM (0.997 per 100 person-years).

Visual inspection and both goodness-of-fit statistics suggested that the log-normal distribution had the best fit to the observed trial data, followed by the log-logistic and Weibull distributions. The log-normal distribution was therefore selected.

Over 10 years, FDI resulted in 4.85 quality-adjusted life-years (QALYs), compared with 4.70 QALYs for FCM, equivalent to a gain of 0.157 QALYs (Table 3). The differences in fracture incidence accounted for 13.8% of the overall difference in QALE. Total per-patient costs were £4,460 with FDI and £6,280 with FCM (Figure 3). Based on a per-fracture cost of £4,982, fractures contributed £122 to total costs with FDI and £442 with FCM (Figure 2).

Combining costs and effects, FDI was found to dominate FCM in people with IBD in the UK as FDI was associated with improved QALE at lower cost. The net monetary benefit at a willingness-to-pay threshold of £20,000 per QALY was £4,969.

Table 2. Absolute and incremental life expectancy, quality-adjusted life expectancy, and cost outcomes in the base case

Item	Life expectancy (years)	QALE (QALYs)	Cost (£)
FDI	9.732	4.854	4,460
FCM	9.732	4.686	6,280
Incremental (FDI vs FCM)	0.00	+0.157	-1,821
Incremental cost-utility ratio (£ per QALY gained)	FDI dominant		
Net monetary benefit (£)	4,969		

FDI, ferric derisomaltose; FCM, ferric carboxymaltose; QALE, quality-adjusted life expectancy; QALY, quality-adjusted life year.

Figure 1. Parametric models of fracture-free survival time

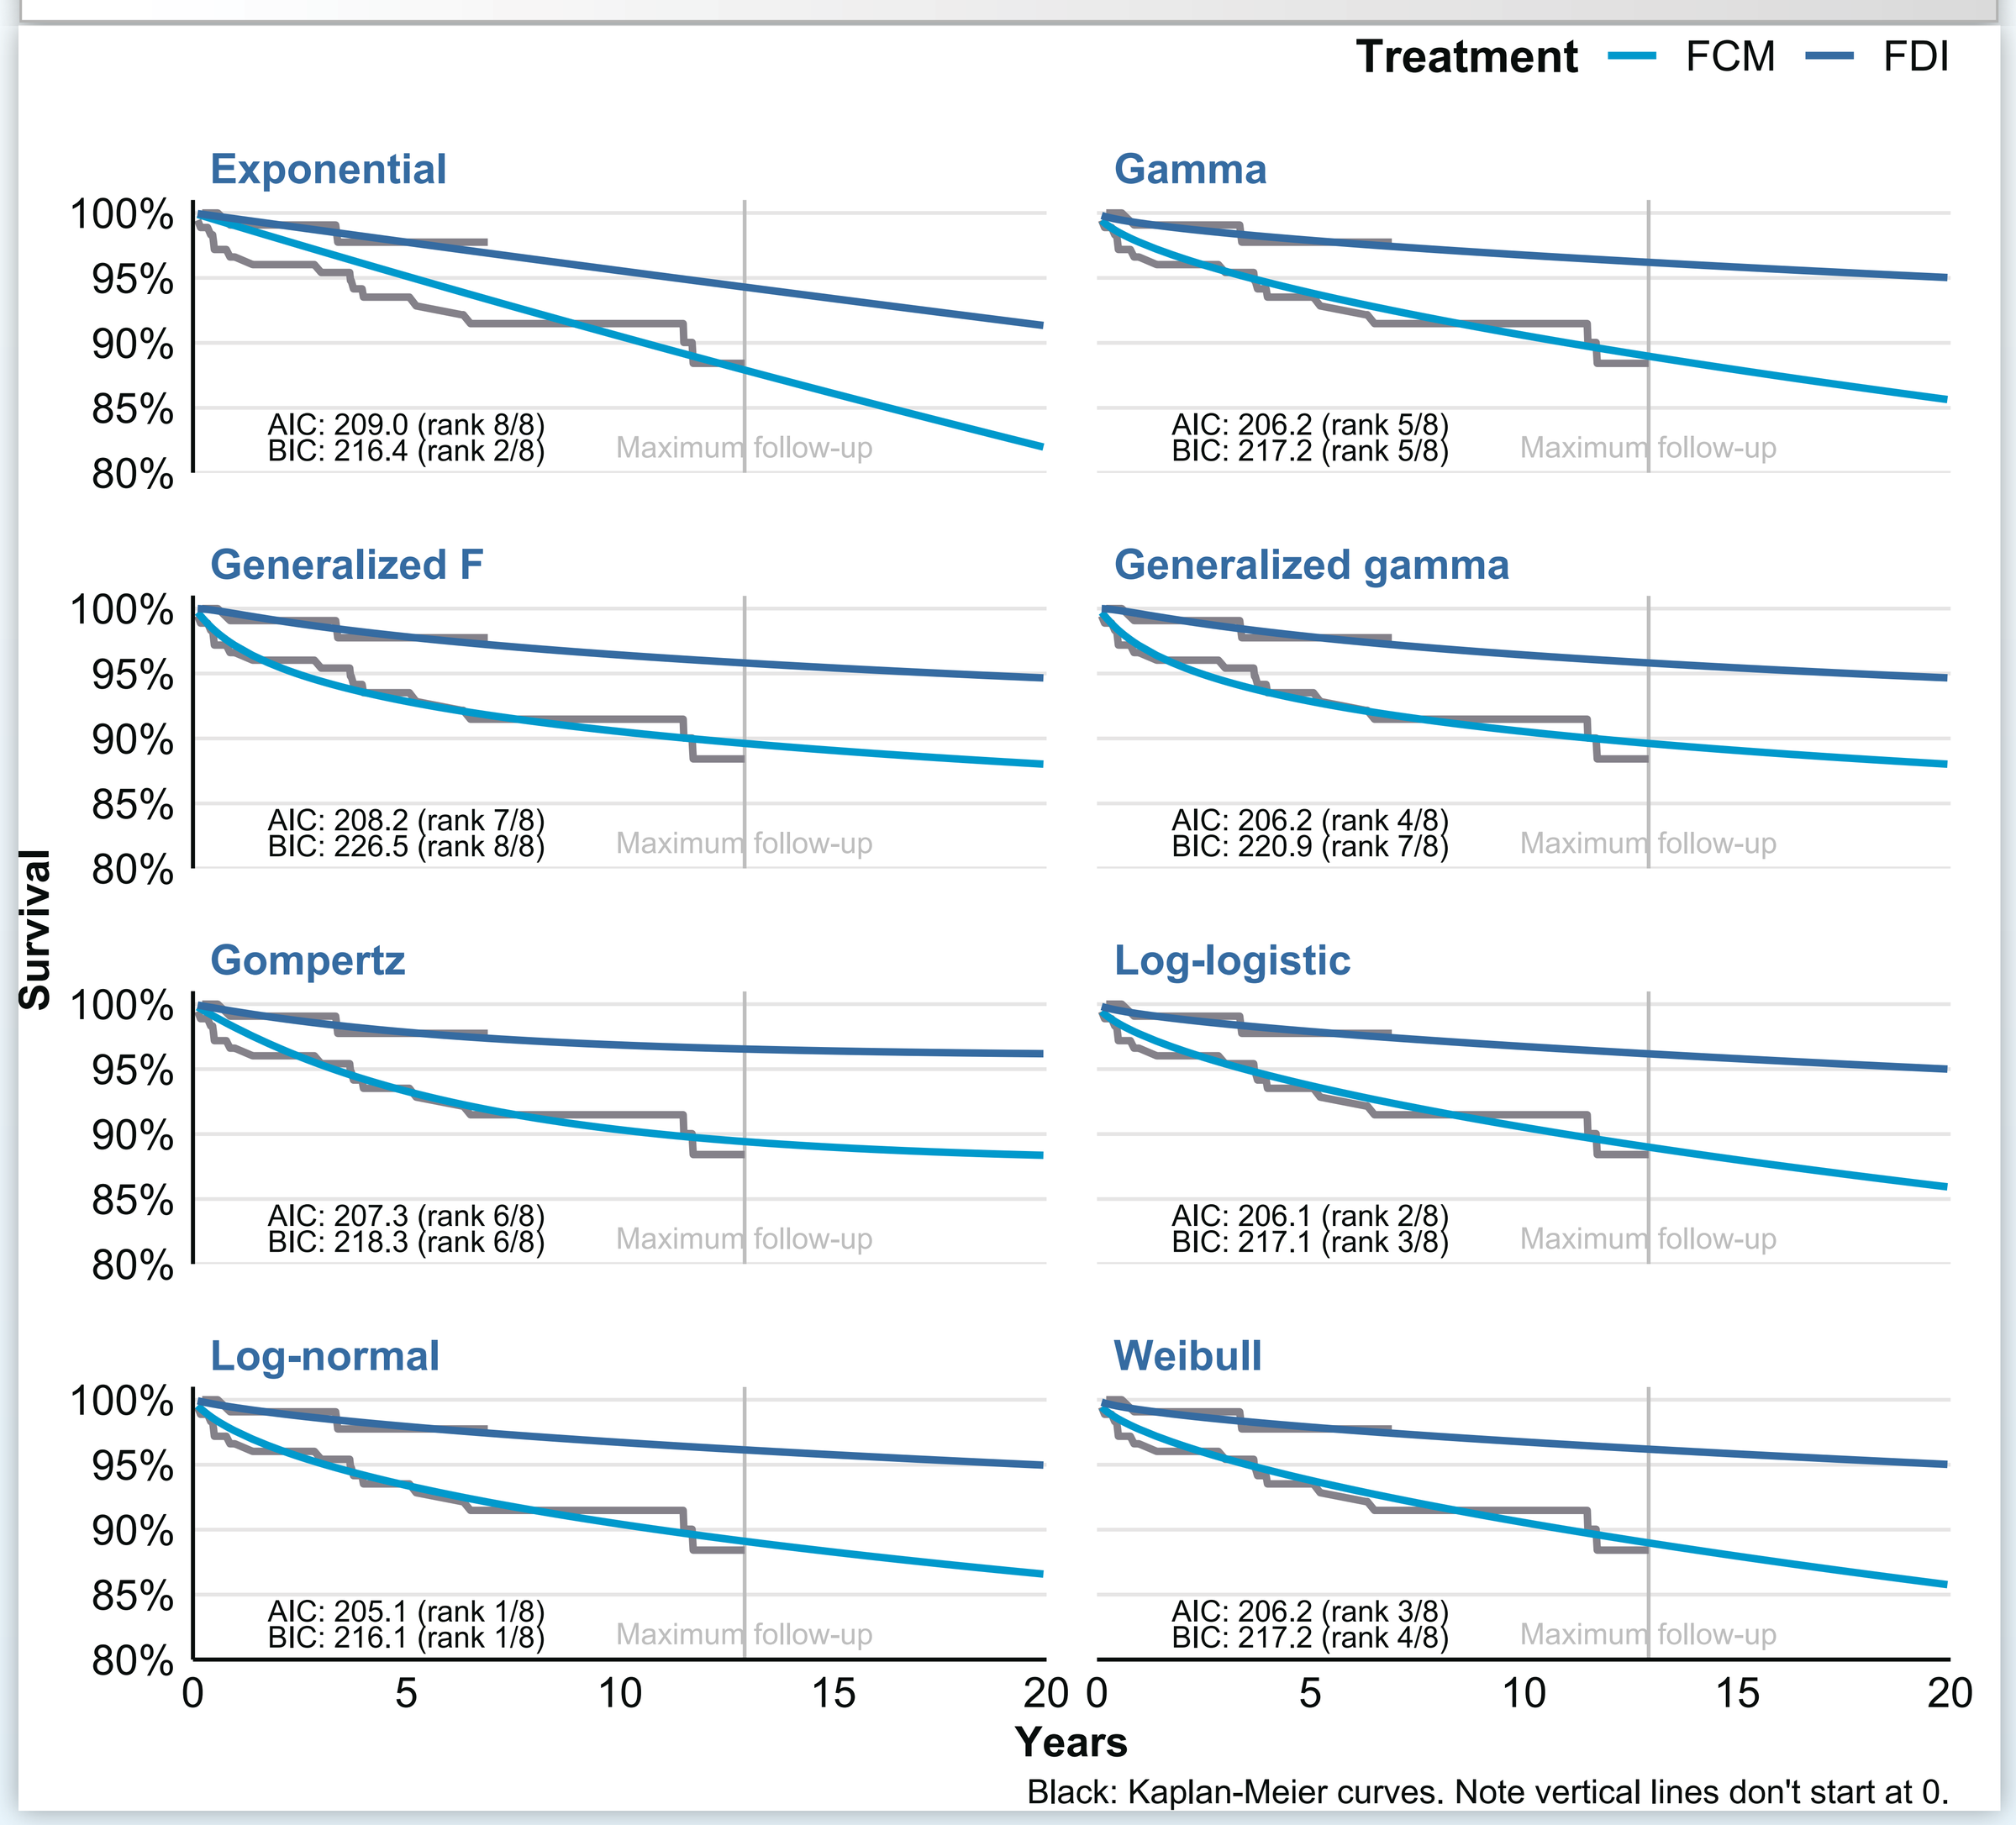
Treatment FCM FDI
Exponential
AIC: 209.0 (rank 8/8)
BIC: 216.4 (rank 2/8)
Gamma
AIC: 206.2 (rank 5/8)
BIC: 217.2 (rank 5/8)
Generalized F
AIC: 208.2 (rank 7/8)
BIC: 226.5 (rank 8/8)
Generalized gamma
AIC: 206.2 (rank 4/8)
BIC: 220.9 (rank 7/8)
Gompertz
AIC: 207.3 (rank 6/8)
BIC: 218.3 (rank 6/8)
Log-logistic
AIC: 206.1 (rank 2/8)
BIC: 217.1 (rank 3/8)
Log-normal
AIC: 205.1 (rank 1/8)
BIC: 216.1 (rank 1/8)
Weibull
AIC: 206.2 (rank 3/8)
BIC: 217.2 (rank 4/8)
Maximum follow-up
Survival
100%
95%
90%
85%
80%
0
5
10
15
20
Years
Black: Kaplan-Meier curves. Note vertical lines don't start at 0.

AIC, Akaike Information Criterion; BIC, Bayesian Information Criterion; FDI, ferric derisomaltose; FCM, ferric carboxymaltose.

Discussion

In addition to the clinical benefits to patients, incorporating the risk of fractures into a recently published patient-level simulation cost-utlity model from the UK showed that FDI improves QoL and reduces cost relative to FCM in patients with IBD and IDA [9].

Figure 2. Breakdown of modeled costs over 10 years

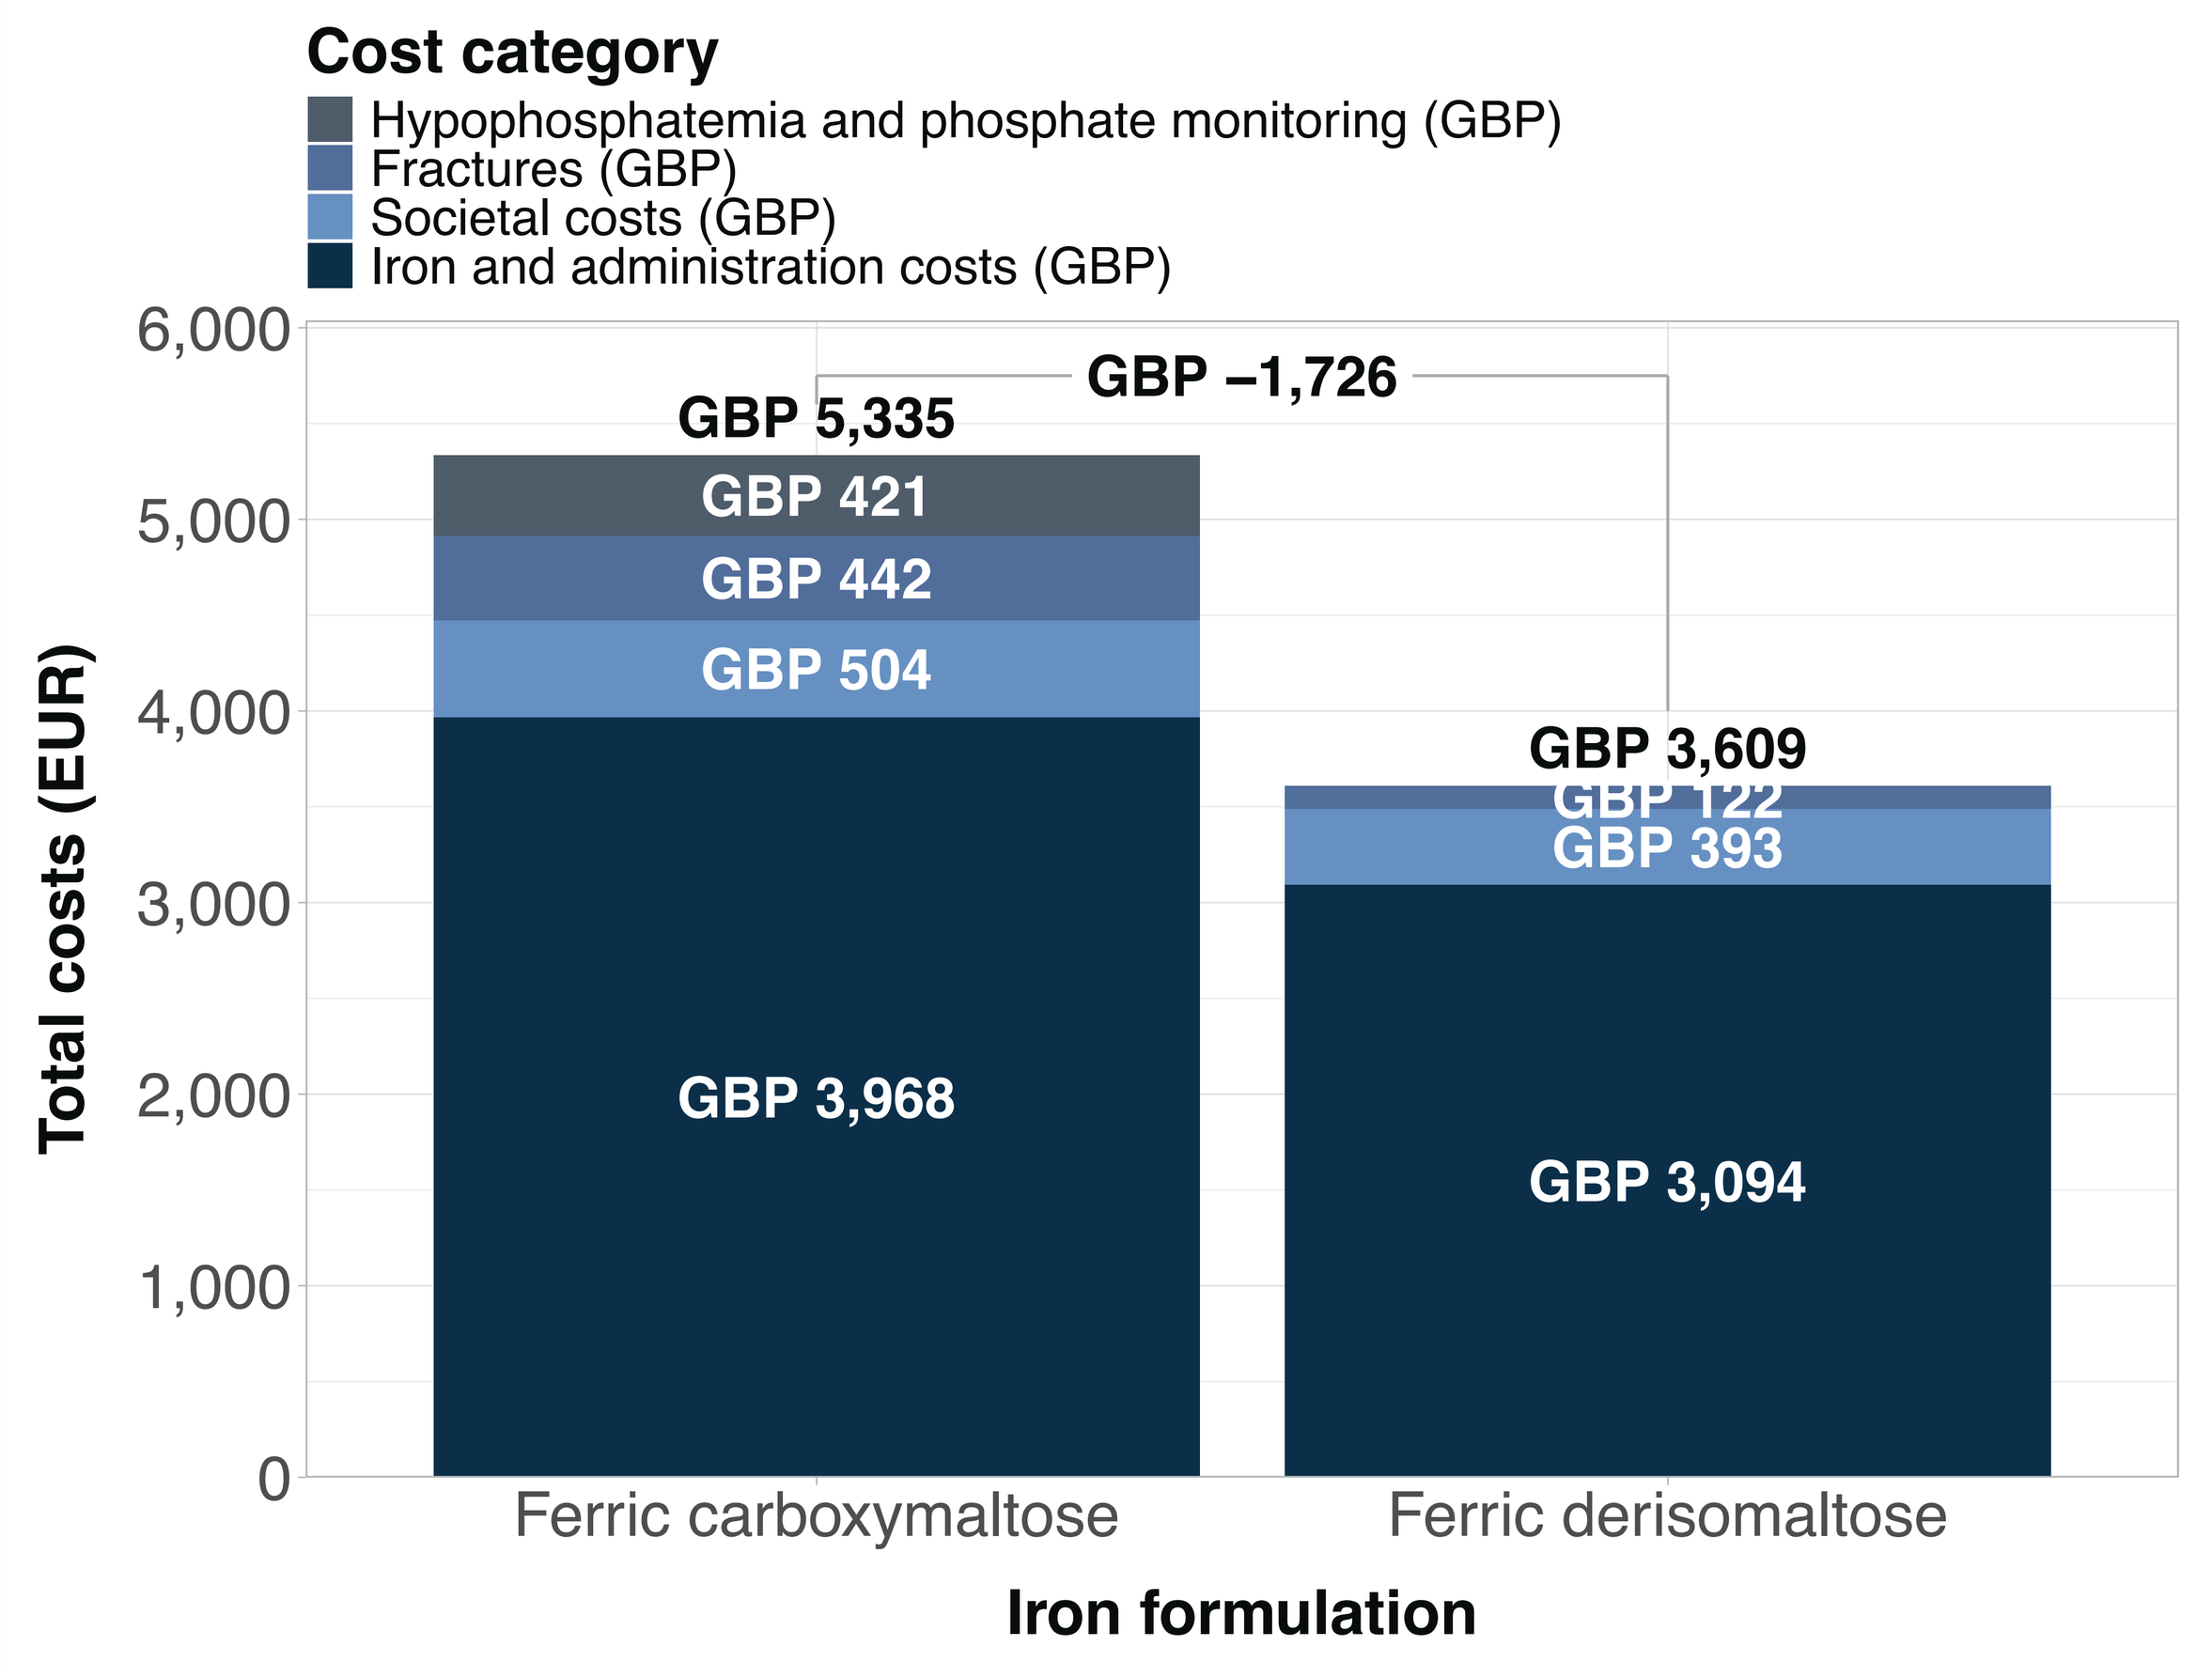
Cost category
Hypophosphatemia and phosphate monitoring (GBP)
Fractures (GBP)
Societal costs (GBP)
Iron and administration costs (GBP)
Total costs (EUR)
6,000
5,000
4,000
3,000
2,000
1,000
0
GBP 5,335
GBP –1,726
GBP 421
GBP 442
GBP 504
GBP 3,968
GBP 3,609
GBP 122
GBP 393
GBP 3,094
Ferric carboxymaltose
Ferric derisomaltose
Iron formulation

Table 3. Contributors to quality-adjusted life expectancy

Item	FCM	FDI	Difference	% of total
Baseline disease-related quality of life (PHOSPHARE-IBD)	4.863	4.985	+0.122	77.6%
Infusion-related disutility	-0.137	-0.123	+0.014	8.7%
Fracture-related disutility	-0.030	-0.008	+0.022	13.8%
Total	**4.696**	**4.854**	**+0.157**	**100%**

FDI, ferric derisomaltose; FCM, ferric carboxymaltose.

Both the QALE gain and the cost savings when incorporating fracture data were larger than the gain and savings, respectively, observed in a previous analysis comparing these two IV iron formulations in the UK [9].

These findings suggest not only that FDI provides good value for money relative to FCM but also that the consideration of different IV iron safety profiles in health economic analysis can materially affect the magnitude of the benefit and cost estimates; in the present case, the finding that FDI was the dominant treatment option for IDA in IBD in the UK was unchanged from the previous analysis [9].

Strengths of the present analysis included the use of a published and peer-reviewed health economic model [9,10] as well as a contemporary treatment cohort. The clinical modelling used unambiguous events to define follow-up periods and outcomes and well-established, robust methods of time-to-event modeling. Limitations include the modeling of first fracture events only, and the potentially limited generalizability of Austrian real-world data to the UK, e.g., due to differences in fracture care between jurisdictions [12]. Including only first fractures is likely to be conservative from the FDI perspective, and it is plausible that the findings would generalize to different settings with broadly comparable levels of healthcare provision. Finally, not all patients in the Austrian data had IBD and IDA, which, especially given the chronic nature of IBD, may also limit the generalizability of the findings.

Conclusion

A cost-utility model was developed to incorporate iron dosing differences, phosphate monitoring, incidence of hypophosphatemia based on the PHOSPHARE-IBD RCT, and incidence of fractures based on a recent real-world dataset. The analysis showed FDI to reduce costs and improve QALE versus FCM, corroborating the high-level findings from previous analyses, but with greater cost savings and QALY gains associated with FDI.

References

1. Pasvol TJ, et al. BMJ Open 2020;10(7):e036584.
2. Freeman K, et al. BMC Gastroenterol 2021;21(1):139.
3. Gordon H, et al. J Crohns Colitis 2024;18(1):1–37.
4. Snook J, et al. Gut 2021;70(11):2030–51.
5. Van Doren L, Auerbach M. Hematology Am Soc Hematol Educ Program 2023;2023(1):622–9.
6. Kennedy NA, et al. Int J Clin Pharm 2023;45(3):604–12.
7. Zoller H, et al. Gut 2023;72(4):644–53.
8. Fukumoto S, et al. Adv Ther 2023;40(11):4877–88.
9. Iqbal TH, et al. J Med Econ 2024;27(1):392–403.
10. Hu S, et al. J Med Econ 2022;25(1):561–70.
11. Zoller H, et al. Blood 2023;(S1):3838.
12. Werner M, et al. Eur J Trauma Emerg Surg 2022;48(3):1625–38.

Poster presented at ISPOR 2024, Georgia World Congress Center, Atlanta, GA, USA. May 5–8, 2024.