CO3

Prediction of Long-Term Factor IX Durability Among Hemophilia B Patients Treated with Fidanacogene Elaparvovec Gene Therapy

Morris A[a], Ortega L[b], Thakkar S[c], Wilcox L[d], Cappelleri JC[e], Incerti D[f], Ines M[g], Deb P[h], Chhabra A[i], Vytlacil E[j], Batt K[k], Lakdawalla D[l]

[a] ENTITYRISK, INC., San Diego, CA, USA, [b] ENTITYRISK, INC., Washington, DC, USA, [c] PFIZER, INC., Cambridge, MA, USA, [d] PFIZER Canada, Kirkland, QC, Canada, [e] PFIZER, INC., Groton, CT, USA, [f] ENTITYRISK, INC., San Francisco, CA, USA, [g] PFIZER, INC., Porto Salvo, Portugal, [h] HUNTER COLLEGE New York, NY, USA, [i] PFIZER, INC., New York, NY, USA [j] YALE UNIVERSITY, New Haven, CT, USA, [k] ENTITYRISK, INC., Raleigh, NC, USA, [l] UNIVERSITY OF SOUTHERN CALIFORNIA, Los Angeles, CA, USA

INTRODUCTION

Disease studied: Hemophilia B

- Hemophilia B is a rare inherited bleeding disorder caused by a deficiency or dysfunction of a blood clotting factor known as Factor IX (FIX).
- Patients with Hemophilia B can experience spontaneous and prolonged bleeding, which, if left unmanaged, can result in joint damage.
- Prevalence of hemophilia B is reported as 3.7 cases per 100,000 males in the U.S.[1]

Novel therapy: fidanacogene elaparvovec gene therapy

Standard of care: FIX prophylaxis therapy

Health outcomes: FIX activity levels

Data source: Phase 1/2 (NCT02484092 and NCT03307980) and 3 (NCT03861273) clinical trials for fidanacogene elaparvovec.

OBJECTIVE

Predict the long-term distribution of Factor IX activity among moderately-severe and severe hemophilia B (FIX ≤ 2%) patients treated with fidanacogene elaparvovec out to 30 years after treatment and generalize predictions across two common FIX measurement techniques to maximize comparability.

METHODS

I. Data

- This study used data from patients treated with fidanacogene elaparvovec who participated in Phase 1/2 ("Ph1/2") and Phase 3 ("Ph3") clinical trials.
- As of Nov. 2023, the Ph1/2 trial included 15 participants who were followed for up to 6 years, whereas Ph3 enrolled 45 participants followed for up to 4 years.
- Inclusion criteria:
 - At least two FIX level measurements taken 6 months after gene therapy infusion.
 - At least one FIX level measurement taken 18 or more months after gene therapy infusion.
 - FIX level measurements occurring within 7 days of a FIX infusion were excluded.
- 14 patients from Ph1/2 and 42 patients from Ph3 met this inclusion criteria and were part of the analytic dataset used for the predictive modeling.
- The analysis assumed that FIX activity level reaches a steady state within the first few months post gene-therapy infusion, and used 6 months as the starting point for inclusion of FIX activity measurements in the predictive modeling.
 - This approach was used to ensure all patients have reached steady state when included in the modeling and to mitigate any influence of corticosteroids.
 - This approach was also consistent with a similar analysis by Shah et al. (2023).[2]

Table 1: Summary of the analytic dataset

	Phase 1/2	Phase 3	Pooled sample
Data type	**Longitudinal; varied measurement timing**		
Patients	14	42	56
Observations	262	433	695
Maximum follow-up as of Nov. 2023	Up to 73 months post-gene therapy infusion	Up to 48 months post-gene therapy infusion	Up to 73 months post-gene therapy infusion

Copyright @ 2024

II. Pooling clinical trials

- We conducted an exploratory data analysis to account for potential differences in patient selection between the two clinical trials.
- Patient age, body mass index (BMI), and baseline FIX activity measured 6 months after gene therapy infusion were identified as significant contributors to the observed variation across FIX measurements observed in the two trials over time.[3,4]

Figure 1: FIX activity levels for patients in Ph1/2 and Ph3 trials over time after controlling for key explanatory variables

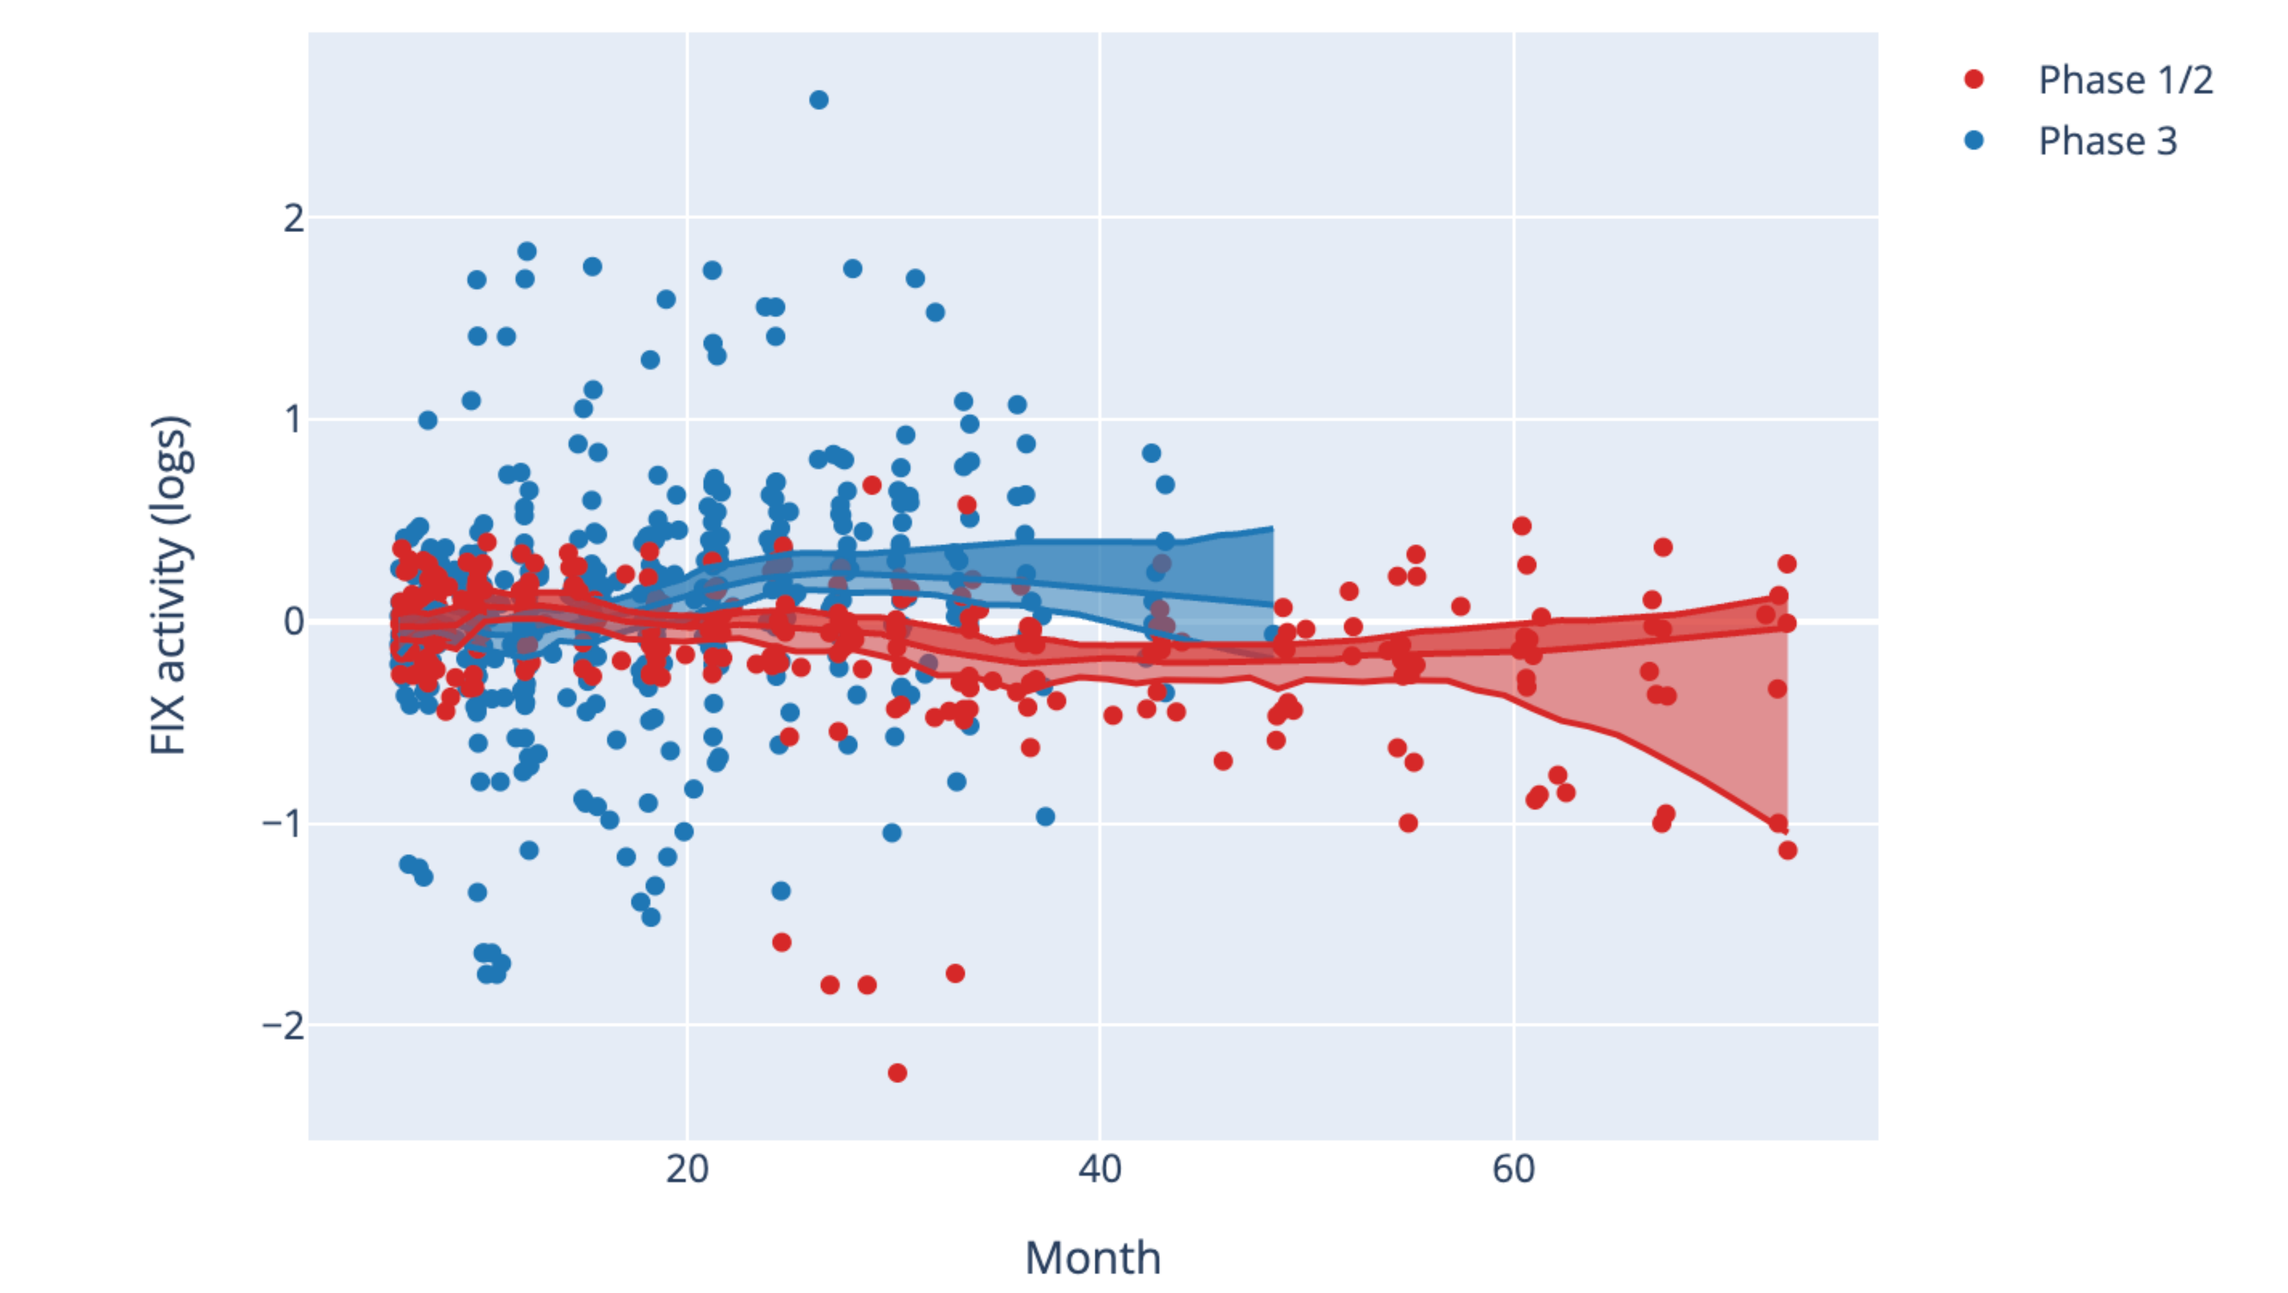
Phase 1/2
Phase 3
FIX activity (logs)
2
1
0
−1
−2
20
40
60
Month

Note: This figure shows FIX activity levels for both trial phases after netting (or "partialling") out age, BMI, and baseline treatment FIX levels according to the Frisch-Waugh-Lovell theorem. The plot also displays locally weighted scatterplot smoothing (LOWESS) trend lines and their corresponding 95% confidence intervals for each trial. All FIX activity levels displayed in logs and based on measurements taken using a one-stage assay with Actin FSL reagent.

III. FIX activity measurement assays and reagents

- Recent evidence suggests that using different assays and/or reagents when measuring FIX activity level may yield different results.[5,6,7]
- To account for potential variability between measurement techniques, our analysis aimed to predict long-term FIX activity measurements in terms of one-stage assays leveraging both Synthasil and Actin FSL reagents.
 - *Key challenge*: FIX activity levels in Ph1/2 were solely measured with a one-stage assay using the Actin FSL reagent, while Ph3 included simultaneous FIX measurements using both Actin FSL and Synthasil reagents.
 - *Solution*: We fit a linear regression model on the natural log scale (base e) using simultaneous measurements based on both the Actin FSL and Synthasil reagents from the Phase 3 trial. The model was then used to impute predictions in terms of the Synthasil reagent based on Actin FSL predictions generated with the model described in Section IV. The model was specified as follows:

$$ln(FIX_{Synthasil}) = \beta\, ln(FIX_{Actin}) + u$$

One-stage Actin FSL and Synthasil measurements of FIX activity are highly correlated with one another (see Figure 2, Panel A). The model made predictions that closely resembled observed values from the Phase 3 trial (see Figure 2, Panel B), though prediction error was larger for outlier FIX values, particularly at the right tail of the distribution.

Figure 2: One-stage Actin FSL/Synthasil Reagent conversion model

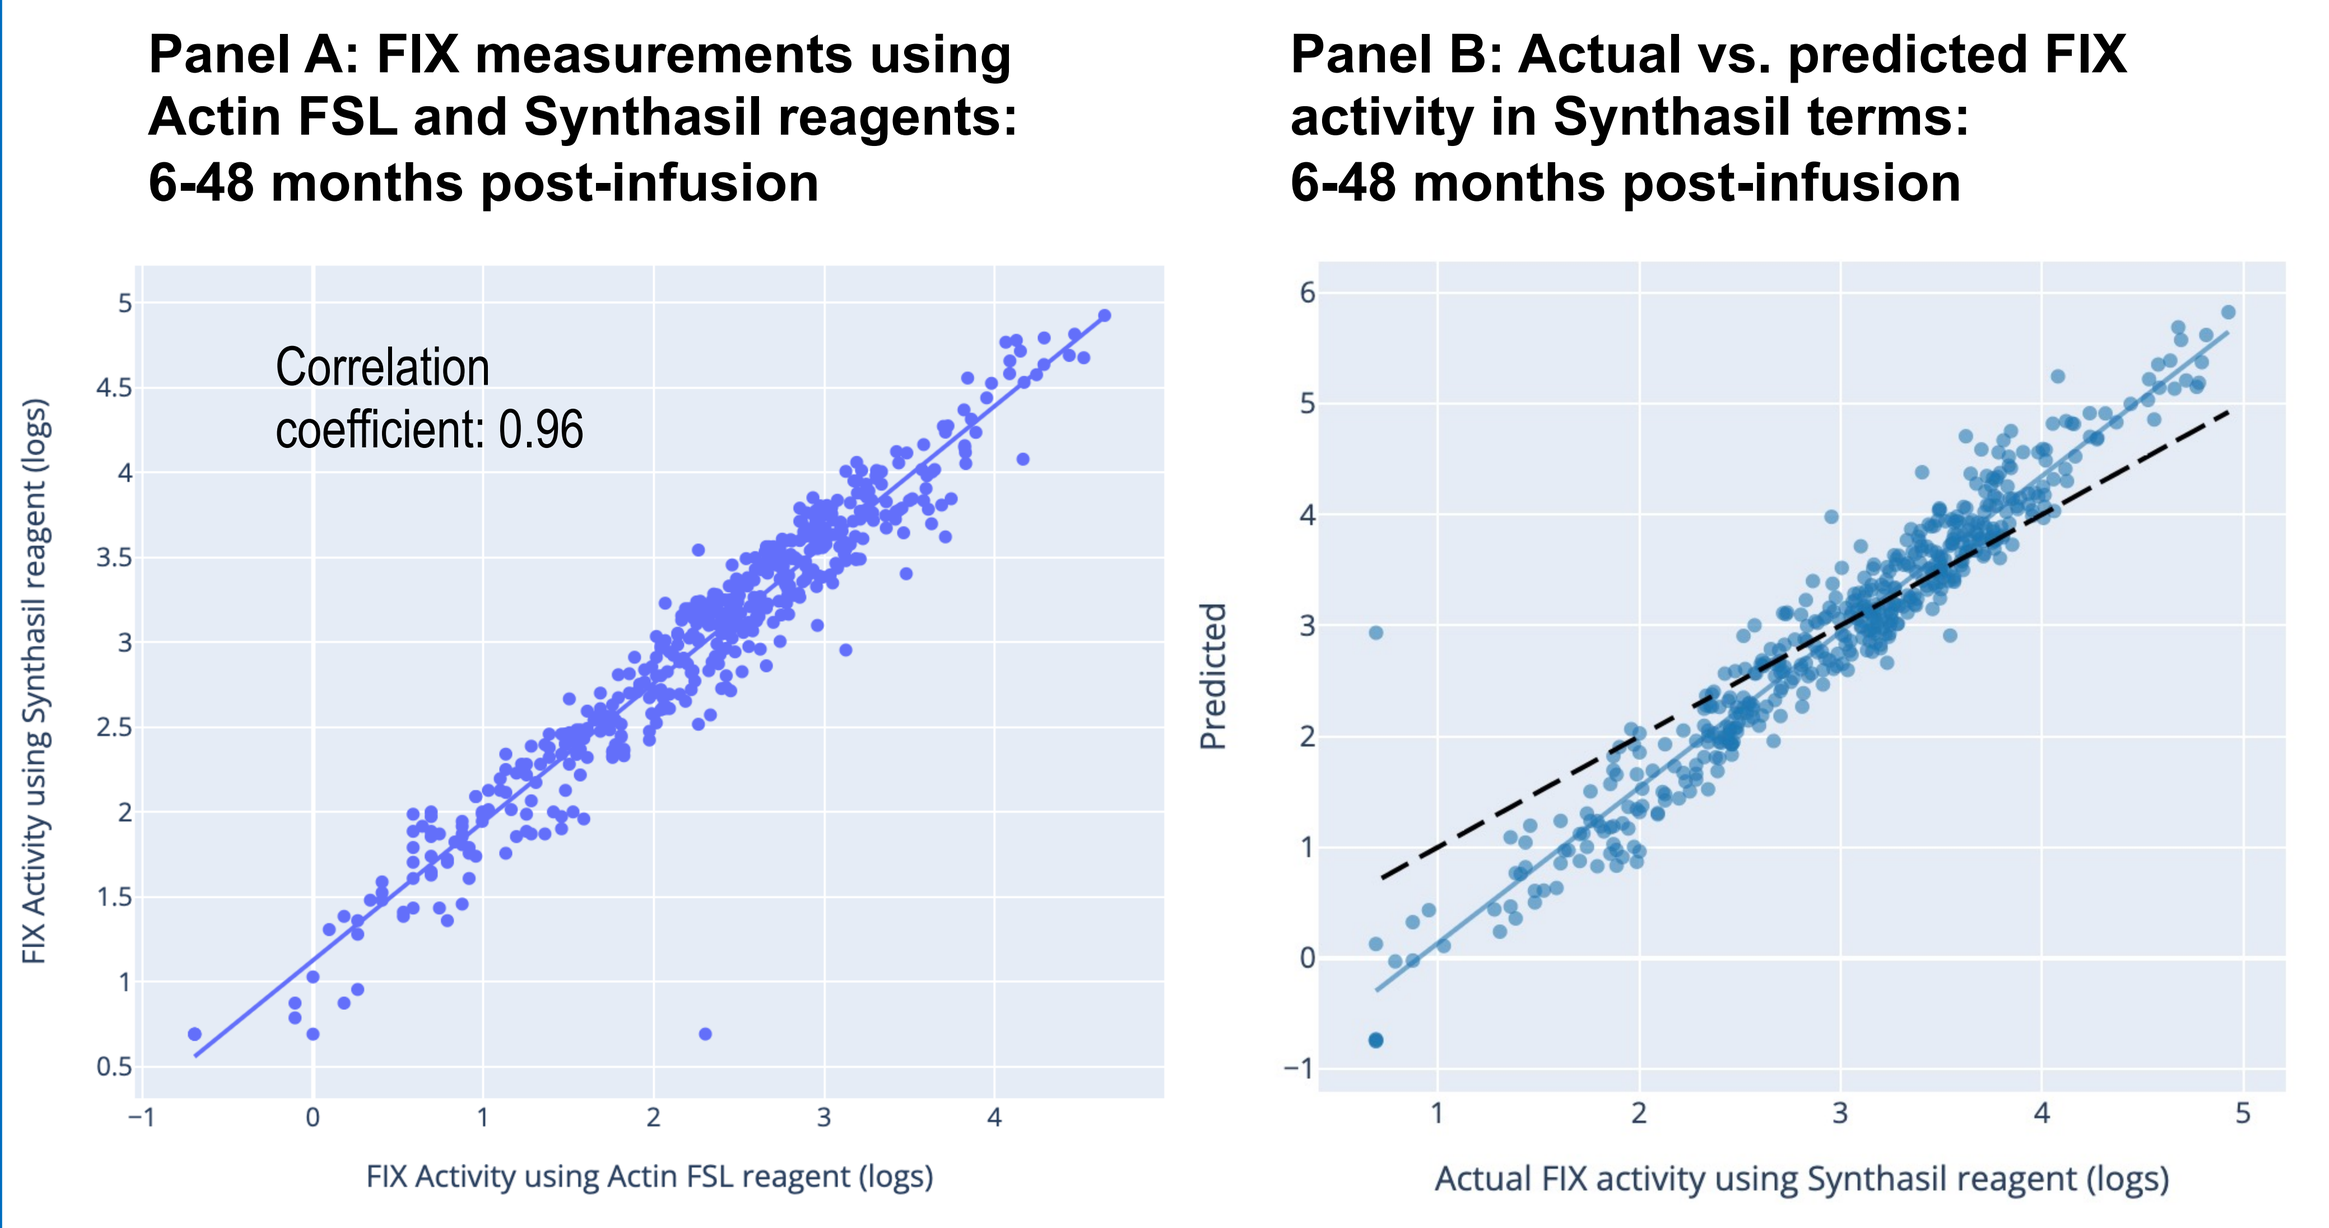
Panel A: FIX measurements using Actin FSL and Synthasil reagents: 6-48 months post-infusion
Correlation coefficient: 0.96
FIX Activity using Synthasil reagent (logs)
FIX Activity using Actin FSL reagent (logs)
Panel B: Actual vs. predicted FIX activity in Synthasil terms: 6-48 months post-infusion
Predicted
Actual FIX activity using Synthasil reagent (logs)

IV. Statistical methods: Long-term prediction of FIX activity levels in terms of Actin FSL

Step 1: Model specification

- We employed a Bayesian lognormal model with weakly informative priors because:
 - The lognormal model closely replicates the distribution of observed FIX activity levels from the clinical trial data.
 - A Bayesian framework allowed us to quantify uncertainty in the model parameters and predictions in an easily interpretable way.
- The lognormal model of FIX activity levels was parametrized as follows:

$$FIX \sim Lognormal(X\beta + \alpha, \sigma)$$

With X as the input dataset (detailed below in Table 2), β the vector of regression coefficients used to predict μ, the mean of the normal distribution on the log scale, α the intercept, and σ the standard deviation of the lognormal distribution.

- Priors were specified as follows:
 - $\alpha \sim Normal(0,1)$
 - $\beta \sim Normal(0,1)$
 - $\sigma \sim Half\text{-}Normal(0,1)$

Table 2: Lognormal model specification

Variables	Description
Endpoints	
FIX activity levels	One-stage assay using Actin FSL reagent
Explanatory variables	
Age group indicators	• 18 ≤ Age ≤ 25 • 25 < Age ≤ 30 • 30 < Age ≤ 40 • 40 < Age ≤ 50 • Age > 50
BMI group indicators	• BMI < 18.5 • 18.5 ≤ BMI < 25 • 25 ≤ BMI < 30 • BMI ≥ 30
Baseline treatment FIX activity	FIX activity levels at month 6
Months since infusion	Monthly linear time trend

Step 2: Model fitting

- Raw data were pre-processed according to model specifications.
- Bayesian models were fit to the analytic dataset using Stan.[8]
- The distribution of FIX activity levels was predicted as follows:
 - A "bootstrapped" sample of 1,000 patients was drawn at random from the combined trial datasets with replacement.
 - For each of the 1,000 patients, 1,000 posterior predictive draws of FIX activity levels were drawn every quarter from month 6 up to 30.5 years post-infusion.

Step 3: Model checking

- We used posterior predictive checks to compare simulations from the model against the distribution of the underlying raw data and evaluate model performance.

Figure 3: Actual vs. predicted distribution of FIX activity levels in terms of Actin FSL: 6-73 months post-infusion

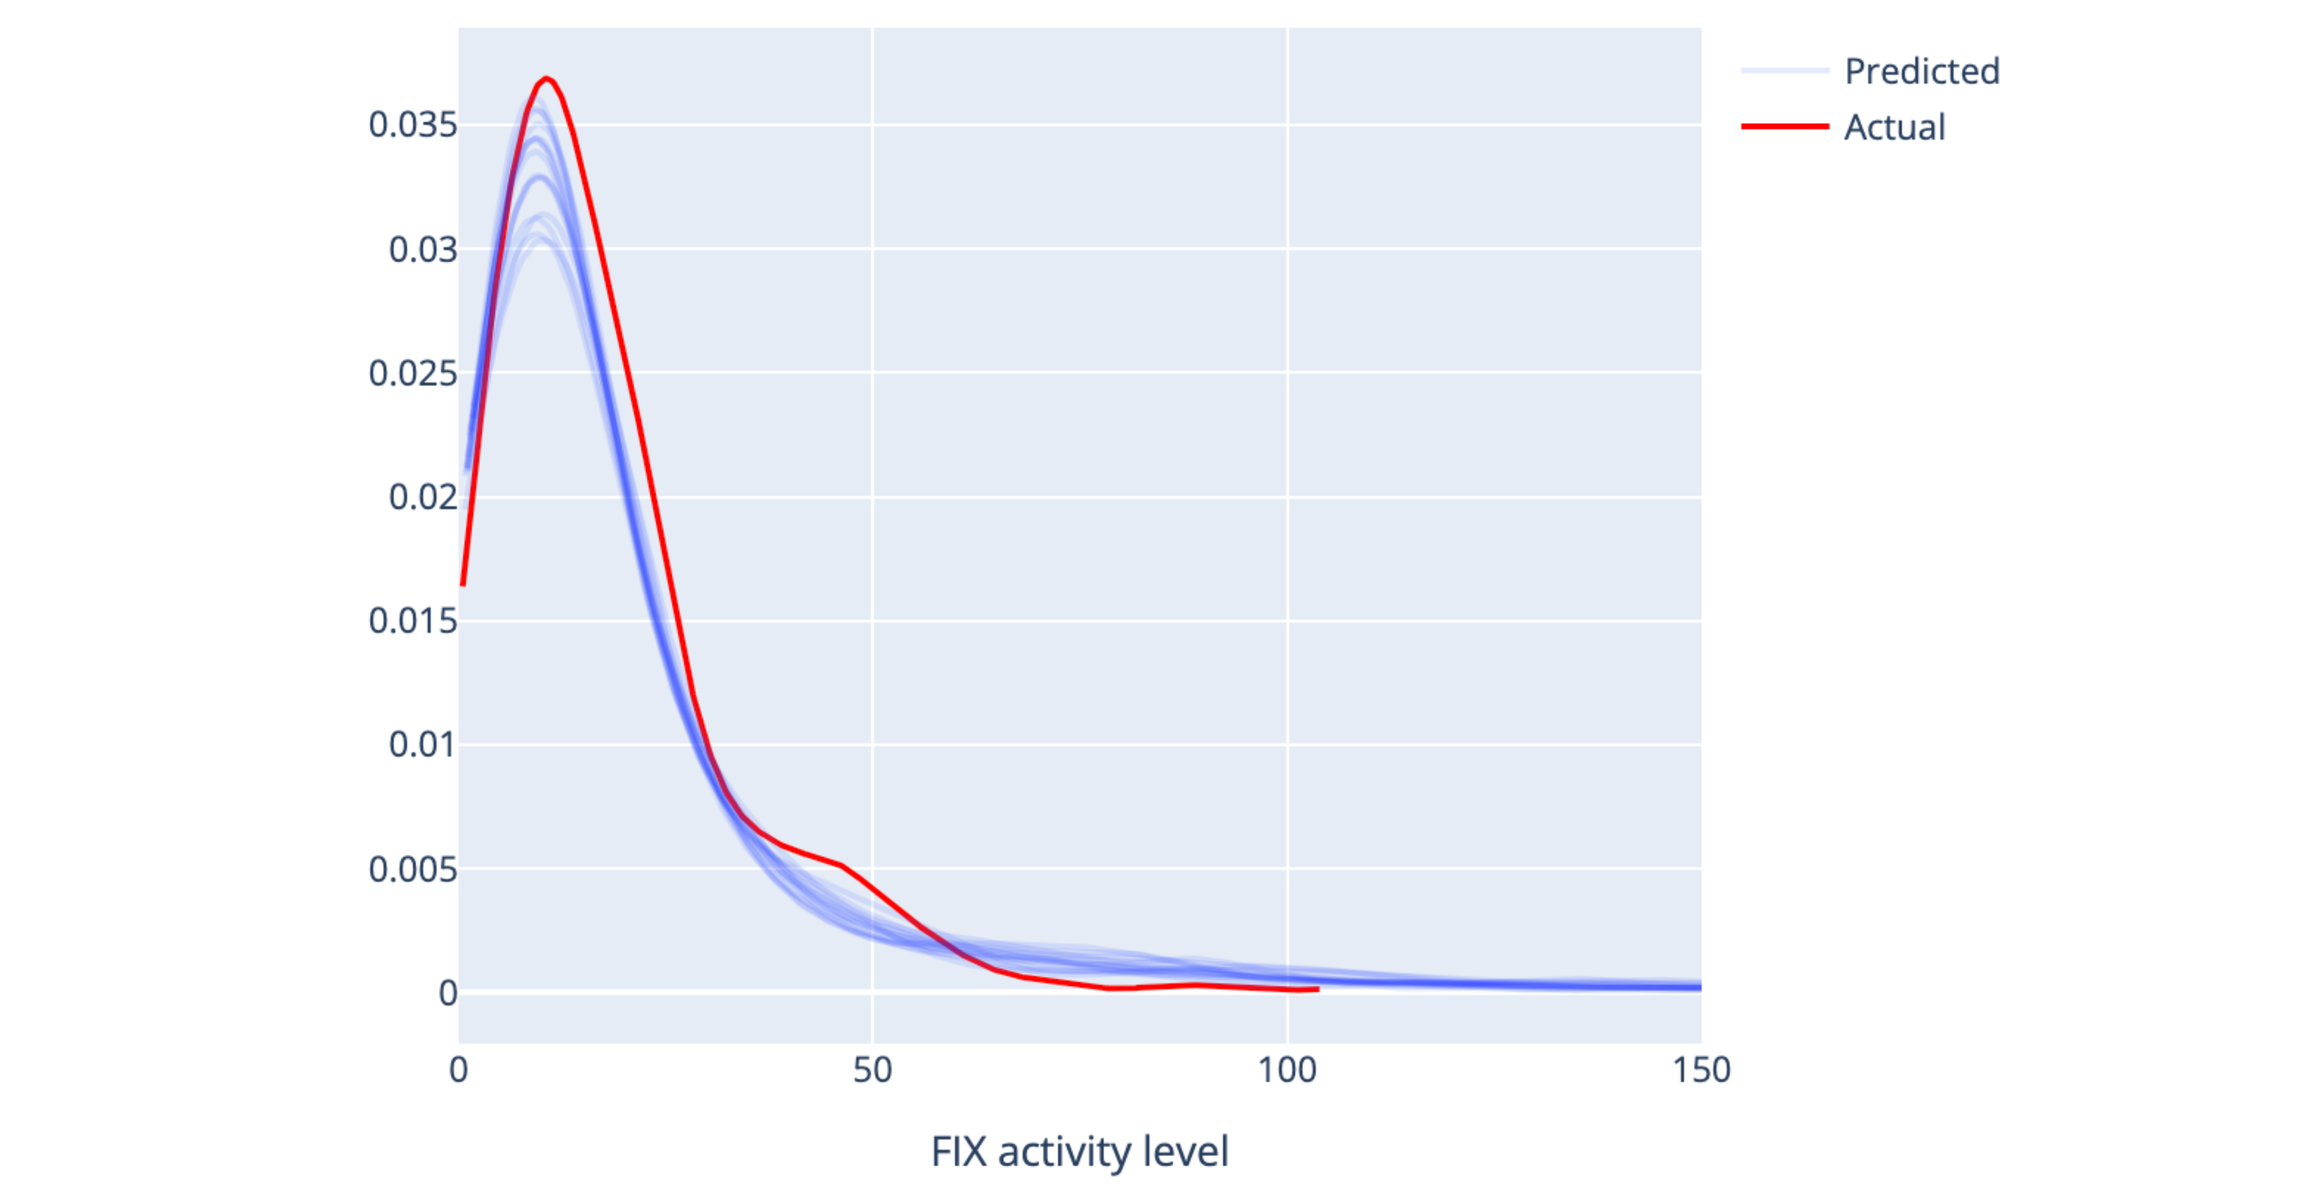
Predicted
Actual
0.035
0.03
0.025
0.02
0.015
0.01
0.005
0
0
50
100
150
FIX activity level

Note: The plot above displays actual and predicted probability density functions ("PDFs"). Predicted PDFs reflect simulations of the full distribution of FIX activity for all measurements from the analytic dataset, based on the Bayesian lognormal model detailed in Table 2.

- The posterior predictive checks suggested that simulation draws from the posterior predictive distribution of the Bayesian lognormal model produce probability density functions (PDFs) that fit the observed data well (see Figure 3).

RESULTS

- Predicted median FIX activity in the treated population at 30 years post infusion was **4.6%** (95% CrI: 1.8%, 11.5%) and **7.1%** (95% CrI: 2.2%, 22.4%) based on predictions in terms of a one-stage assay using Actin FSL and Synthasil reagents, respectively (see Figure 4, Panel A).
- At 30 years, **84.8%** (95% CrI: 53.1%, 99.3%) of the population was predicted to have FIX activity above 2% based on a one-stage assay using the Synthasil reagent (See Figure 4, Panel B).
- Results were similar when using alternative model specifications including (1) a gamma generalized linear model (GLM) and (2) a non-parametric ordinary least squares (OLS) model.

Figure 4: Predicted FIX activity among patients treated with fidanacogene elaparvovec: 0.5-30.5 years post-infusion

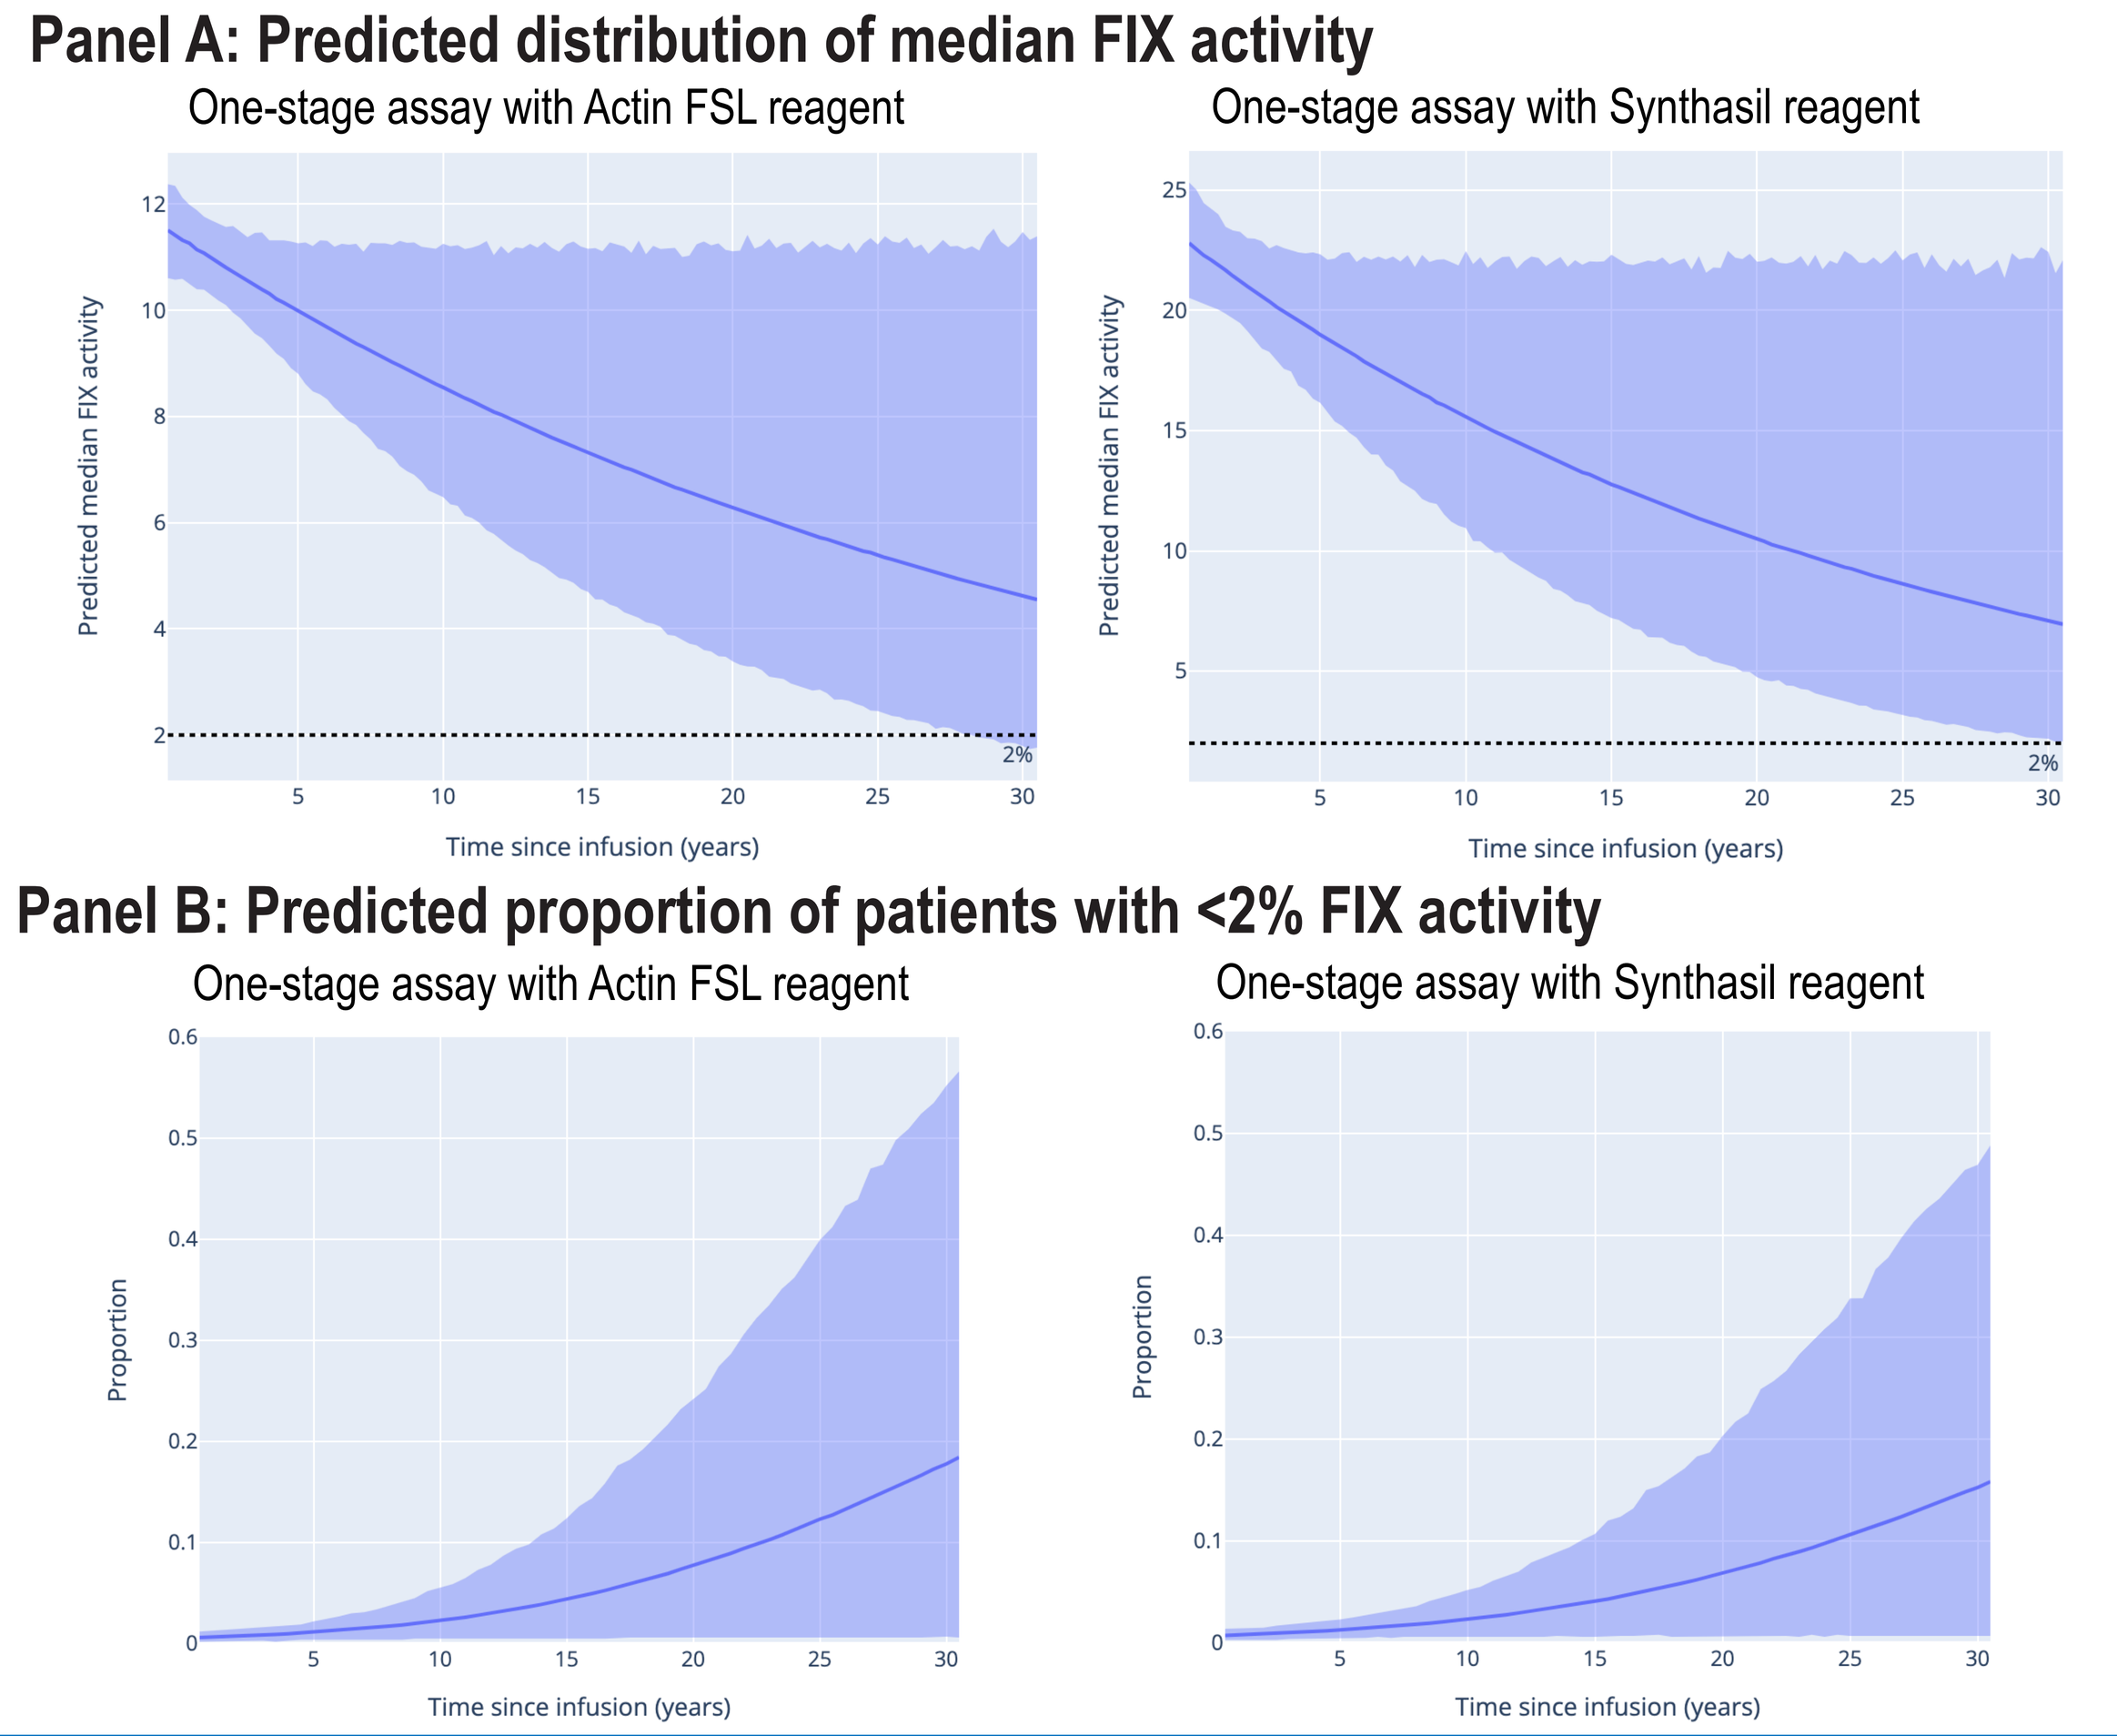
Panel A: Predicted distribution of median FIX activity
One-stage assay with Actin FSL reagent
One-stage assay with Synthasil reagent
Predicted median FIX activity
2%
Time since infusion (years)
Panel B: Predicted proportion of patients with <2% FIX activity
One-stage assay with Actin FSL reagent
One-stage assay with Synthasil reagent
Proportion
Time since infusion (years)

CONCLUSIONS

Our model predicts that a majority of hemophilia B patients receiving fidanacogene elaparvovec would experience sustained long-term effects, maintaining FIX activity levels above 2% for up to 30 years after infusion.

REFERENCES

1. Soucie, J. M., Miller, C. H., Dupervil, B., Le, B., & Buckner, T. W. (2020). Occurrence rates of haemophilia among males in the United States based on surveillance conducted in specialized haemophilia treatment centres. *Haemophilia: The Official Journal of the World Federation of Hemophilia*, 26(3), 487–493. https://doi.org/10.1111/hae.13998
2. Shah, J., Kim, H., Sivamurthy, K., Monahan, P. E., & Fries, M. (2023). Comprehensive analysis and prediction of long-term durability of factor IX activity following etranacogene dezaparvovec gene therapy in the treatment of hemophilia B. *Current Medical Research and Opinion*, 39(2), 227–237. https://doi.org/10.1080/03007995.2022.2133492
3. Frisch, Ragnar; Waugh, Frederick V. (1933). "Partial Time Regressions as Compared with Individual Trends". *Econometrica*. 1(4): 387–401. doi:10.2307/1907330
4. Cleveland, W.S. (1979) "Robust Locally Weighted Regression and Smoothing Scatterplots". *Journal of the American Statistical Association* 74(368): 829-836.
5. Ovanesov, M. V., Jackson, J. W., Golding, B., & Lee, T. K. (2021). Considerations on activity assay discrepancies in factor VIII and factor IX products. *Journal of Thrombosis and Haemostasis*, 19(9), 2102–2111. https://doi.org/10.1111/jth.15425
6. Ovanesov, M. V., Williams, S. C., Nübling, C. M., Dodt, J., Hilger, A., Maryuningsih, Y., & Gray, E. (2020). Summary of the WHO hearing on the development of product-specific reference materials for coagulation factor VIII and factor IX products. *Biologicals*, 67, 88–93. https://doi.org/10.1016/j.biologicals.2020.07.009
7. Wilmot, H. V., & Gray, E. (2018). Potency estimates for recombinant factor IX in the one-stage clotting assay are influenced by more than just the choice of activated partial thromboplastin time reagent. *Haemophilia*, 24(5), e363–e368. https://doi.org/10.1111/hae.13556
8. Stan Development Team. 2024. Stan Modeling Language Users Guide and Reference Manual, v2.34. https://mc-stan.org

DISCLOSURES

Study sponsored by Pfizer, Inc.

Presented at ISPOR US, May 6 2024, Atlanta, GA, USA.