Impact of Definitions of Vaso-occlusion in Clinical Studies of Sickle Cell Disease (SCD) on Efficacy Outcome Assessment

CO197

Haydar Frangoul[1]; Suzan Imren[2]; Fengjuan Xuan[2]; Nanxin Li[2]; Jaime Rubin Cahill[2]; Bill Hobbs[2]; Franco Locatelli[3]

[1]Sarah Cannon Research Institute at The Children's Hospital at TriStar Centennial, Nashville, TN, USA; [2]Vertex Pharmaceuticals Incorporated, Boston, MA, USA; [3]IRCCS, Ospedale Pediatrico Bambino Gesù Rome, Catholic University of the Sacred Heart, Rome, Italy

INTRODUCTION

- Frequent episodes of vaso-occlusion are a clinical hallmark of sickle cell disease (SCD). Vaso-occlusion results from blood flow blockages that deprive tissues and organs of adequate oxygen and nutrients, resulting in severe pain and contributing to progressive organ dysfunction and failure.[1]
- A variety of therapeutic options are aimed at reducing or eliminating these episodes; however, the definition of vaso-occlusion is not consistent across different clinical trials.
- As the SCD treatment landscape evolves and includes the promise of potentially curative therapies, it is important to understand the impact of differing definitions for vaso-occlusion episodes on efficacy outcome data.
 - This information may be of particular interest to payers and health technology assessment (HTA) agencies as they evaluate potentially curative therapies to ensure a balanced assessment across available products.

OBJECTIVE

- To compare vaso-occlusion endpoint definitions from clinical trials for various therapies, including genetic therapies, and use clinical trial data to demonstrate the potential for outcomes differences based on differing definitions of vaso-occlusive episodes.

METHODS

- We reviewed completed and ongoing clinical trials and published articles using endpoints assessing the reduction or elimination of vaso-occlusive crisis/events, including trials of
 - exagamglogene autotemcel (exa-cel, CRISPR/Cas-9 based genome editing treatment)[2]
 - lovotibeglogene autotemcel (lovo-cel, gene addition therapy)[3]
 - hydroxyurea (HU)[4]
 - crizanlizumab[5]
 - l-glutamine[6]
 - voxelotor[7]
- Outcomes related to vaso-occlusion were compared across variables including care setting, care duration, treatments, and complications.
- Data from the CLIMB SCD-121 trial of exa-cel in patients aged 12 to 35 years with severe SCD (data cut as of 14 Jun 2023) were evaluated using different published definitions of vaso-occlusion.

RESULTS

Comparison of vaso-occlusion endpoint definitions

- All vaso-occlusion endpoints evaluated involved a health care facility visit. The most substantial differences in vaso-occlusion endpoint definitions were associated with frequency and duration of visits to health care facilities for acute pain and inclusion of specific SCD specific complications.
- Definitions of vaso-occlusions for acute pain related events differed across studies (**Table 1**)
 - The definitions of severe vaso-occlusive crises for exa-cel (severe VOC; exa-cel), sickle cell pain-related crises (SCPC; crizanlizumab and L-glutamine), and VOC (VOC; voxelotor) include events with medical facility visits of any duration (severe VOC and SCPC) or medical records of a patient being seen or contacting a physician within 1 business day of an event (VOC).
 - Definition of severe vaso-occlusive events (severe VOE; lovo-cel) requires a ≥ 24-hour hospital or emergency room (ER) observation unit visit or ≥2 visits to a day unit or ER over 72 hours.
 - Vaso-occlusion definition for HU (painful crisis) required a facility visit of ≥4 hours duration.
- All endpoints include acute chest syndrome (**Table 1**).
- All but one vaso-occlusion endpoint (VOC; voxelotor) include priapism, for these endpoints that include priapism, the frequency of visits to health care facility varies (**Table 1**).
 - The definition of severe VOE (lovo-cel) requires ≥4 visits to a medical facility for priapism to meet criterion.
 - The definition of several VOC (exa-cel), painful crisis (HU), and SCPC (crizanlizumab and L-glutamine) require only a single visit.
- The exa-cel phase 3 trial in SCD employs a broad and inclusive definition for severe VOC which counts each individual medical facility visit, regardless of frequency or duration of hospitalization (**Table 2**).

Table 1. Detailed Comparison of Definition of Vaso Occlusive Endpoints Used in SCD Clinical Studies

	Exa-cel	Lovo-cel		Hydroxyurea	Crizanlizumab	L-glutamine	Voxelotor
Endpoint	Severe Vaso-occlusive Crisis "severe VOC"	Severe Vaso-occlusive Event "severe VOE"	Vaso-occlusive Event "VOE"	Painful Crisis	Sickle Cell Related Pain Crisis "SCPC"	Sickle Cell Related Pain Crisis "SCPC"	Vaso-occlusive Crisis "VOC"
Medical facility visit required?	Yes	Yes	Yes	Yes	Yes	Yes	Yes
Any duration of healthcare facility visit included?	Yes	No; if admission <24 hours, at least 2 visits to a day unit or ER over 72-hour period required	Yes	No; requires at least 4-hour duration	Yes	Yes	No; requires a patient being seen or contacting a physician within 1 business day of an event
Oral or IV narcotics or IV NSAIDs included?	Yes	No; requires IV treatment	Yes	No; requires IV treatment	Yes	Yes	Yes
Acute chest syndrome included?[a]	Yes	Yes	Yes	Yes	Yes	Yes	Yes
Single priapism event included?	Yes	No; requires 4 episodes to a medical facility	Yes	Yes	Yes	Yes	No
Splenic sequestration included?[b]	Yes	Yes	Yes	No	Yes	Yes	No
Hepatic sequestration included?[c]	No	Yes	Yes	Yes	Yes	Yes	No

[a]Acute chest syndrome definition is generally similar across the endpoints, however severe VOE (lovo-cel) further requires oxygen treatment and/or blood transfusion.
[b]Splenic sequestration definition is generally similar across the endpoints.
[c]Hepatic sequestration definition is generally similar across the endpoints.
Abbreviations: ER: emergency room; IV: intravenous; NSAIDs: non-steroidal anti-inflammatory drugs; SCPC: sickle cell pain-related crises; VOC: vaso-occlusive crises; VOE: vaso-occlusive events.

Table 2. Severe VOC Endpoint Definition Per Exa-cel CLIMB-121 Trial Protocol

	Severe VOC Requirements per Protocol Definition
The Definition of Severe VOC is Objectively Defined	• Acute pain event requiring a visit to a medical facility and administration of pain medications (oral or IV opioids or IV NSAIDs) or RBC transfusions • Acute chest syndrome, as indicated by the presence of a new pulmonary infiltrate on imaging associated with pneumonia-like symptoms, pain or fever • Priapism lasting >2 hours and requiring a visit to a medical facility • Splenic sequestration and an acute decrease in hemoglobin concentrations of ≥2 g/dL
The Definition of Severe VOC is Broad	Includes events that involve: • A short hospital stay (i.e., less than 24 hours) • Administration of oral narcotics • Single emergency room or outpatient clinic visits of any duration • Individual events of priapism (>2 hours)
The Definition of Severe VOC is Inclusive	• Events require either a medical facility visit or an evaluation by a healthcare professional • Events are not restricted to hospital admission

IV: intravenous; NSAIDs: non-steroidal anti-inflammatory drugs; RBC: red blood cell; VOC: vaso-occlusive crises.

Analysis of CLIMB-121 study based on different vaso-occlusion definitions

- Based on the protocol definition for severe VOC in the CLIMB-121, 29 out of 30 patients (96.7%) met the primary endpoint of freedom from severe VOCs for at least 12 consecutive months (VF12; 95% CI: 82.8%, 99.9%) (**Figure 1**).
 - When the primary endpoint was analyzed using the severe VOE definition (see lovo-cel severe VOE definition in Table 1), all patients (30/30; 100.0%) were free from severe VOEs for at least 12 consecutive months (95% CI: 88.4%, 100.0%).

Figure 1. Exa-cel Trial Endpoint Based on Different Vaso-occlusion Definitions

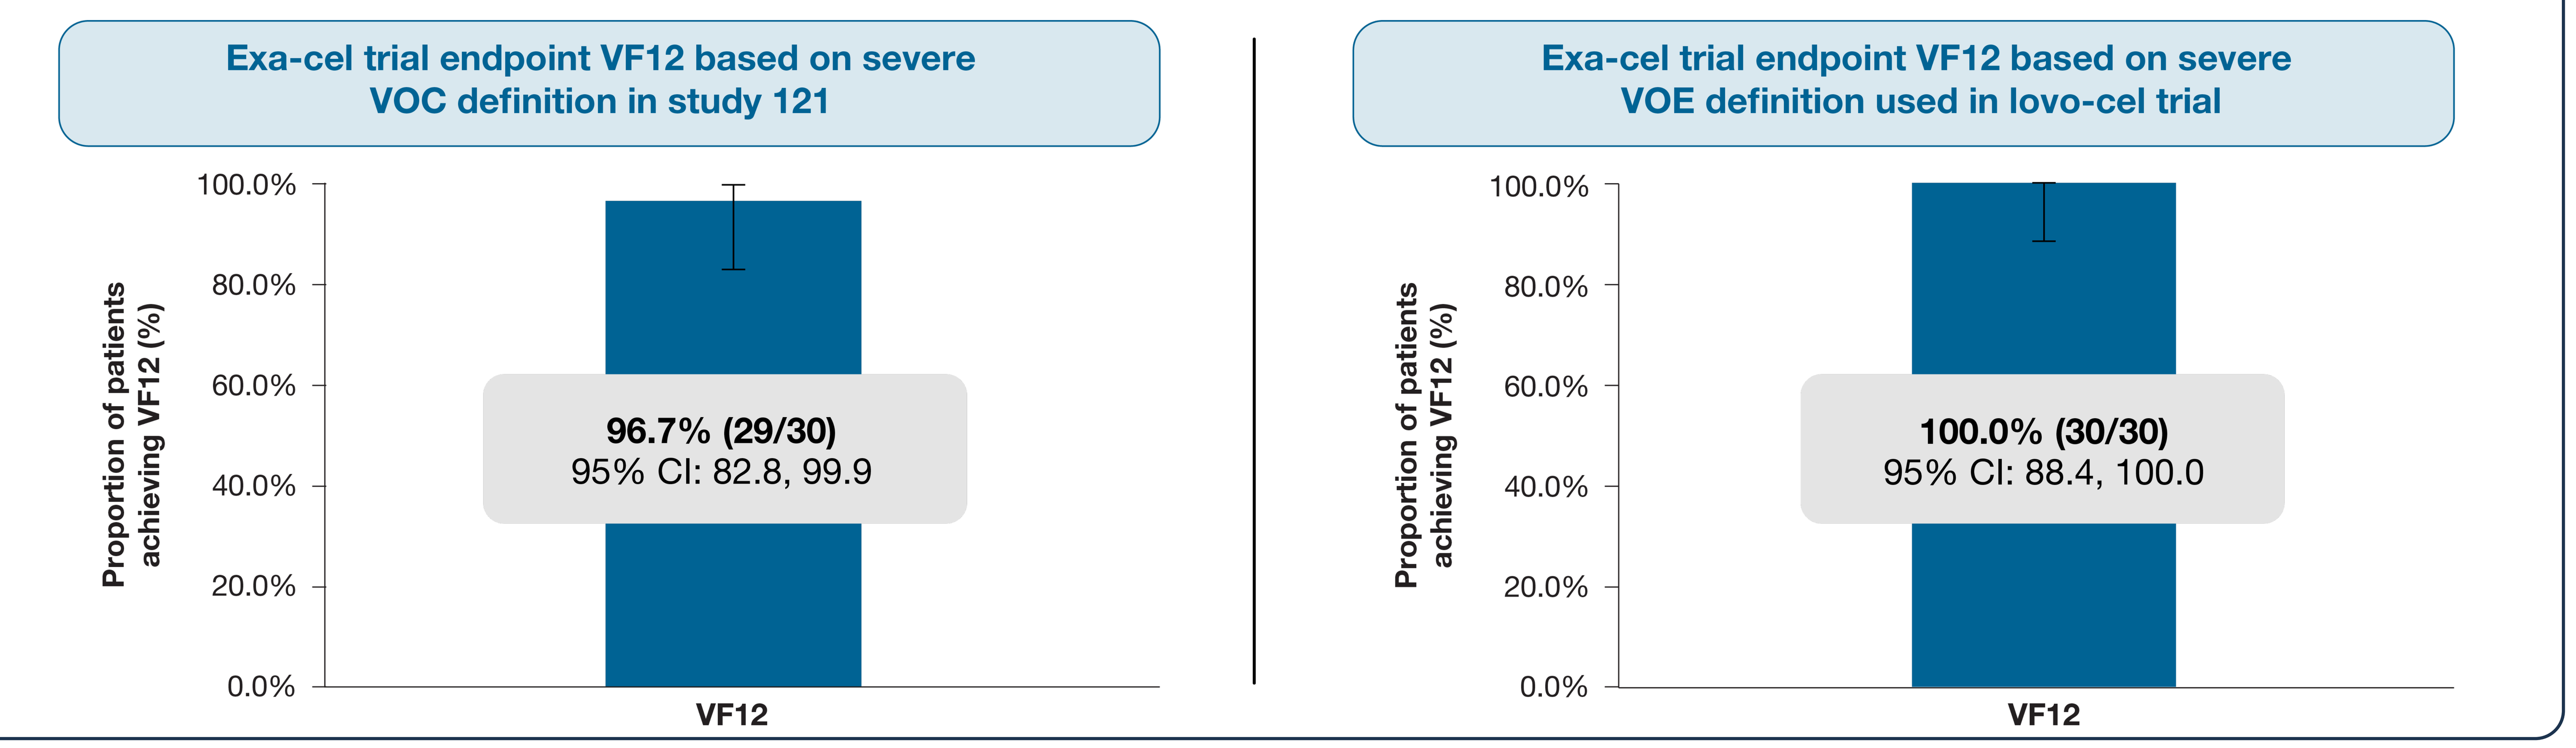
Exa-cel trial endpoint VF12 based on severe VOC definition in study 121
Proportion of patients achieving VF12 (%)
100.0%
80.0%
60.0%
40.0%
20.0%
0.0%
96.7% (29/30)
95% CI: 82.8, 99.9
VF12
Exa-cel trial endpoint VF12 based on severe VOE definition used in lovo-cel trial
Proportion of patients achieving VF12 (%)
100.0%
80.0%
60.0%
40.0%
20.0%
0.0%
100.0% (30/30)
95% CI: 88.4, 100.0
VF12

VF12 was calculated based on study 121 primary efficacy set based on data cut as of 14 Jun 2023.
Abbreviations: VF12: freedom from severe VOCs for at least 12 consecutive months; VOC: vaso-occlusive crises; VOE: vaso-occlusive events.

CONCLUSIONS

- Requirements for health care facility visits in the definitions of severe VOC (exa-cel), VOC (voxelotor), and SCPC (crizanlizumab and L-glutamine) were more broadly inclusive and include events that would not be counted in the definitions of severe VOE (lovo-cel) or painful crisis (HU).
- Clinically, support for this observation comes from our analysis of exa-cel SCD pivotal clinical trial data using these different definitions, which showed the number of patients free from vaso-occlusive episodes changed depending on the definition of vaso-occlusion employed.
- The definition of severe VOC in the exa-cel program is one of the most inclusive compared to other approved gene therapies and disease modifying therapies.
- These results show differences in vaso-occlusion definitions have the potential to impact assessments of treatment efficacy, which should be considered by broad stakeholders including payers and HTA agencies in their evaluation of different SCD therapies.

REFERENCES

1. Kato GJ, et al. *Nat Rev Dis Primers*. 2018;4:18010.
2. Frangoul H et al. *N Engl J Med*. 2021;384(3):252-60.
3. Kantar J, et al. *N Engl J Med*. 2022;386(7):617-28.
4. Charache S, et al. *N Eng J Med*. 1995;332(20):1317-22.
5. Ataga KI, et al. *N Engl J Med*. 2017;376(5):429-39.
6. Niihara Y, et al. *N Engl J Med*. 2018;379(3);226-35.
7. Vichinsky E, et al. *N Engl J Med*. 2019;381(6);509-19.

AUTHOR DISCLOSURES

HF is a consultant to Editas Medicine, Rocket Pharmaceuticals, and Vertex Pharmaceuticals (steering committee); FL is consultant to Amgen, bellicum Pharmaceuticals, Novartis, Novimmune, Sobi, Inc; speaker bureau for Bluebird Bio and Gilead Sciences; steering committee Vertex Pharmaceuticals. SI, FX, NL, JRC, and BH are employees of Vertex Pharmaceuticals.

ACKNOWLEDGMENTS

Thank you to study participants and their families, as well as sites, investigators, nurses, and the entire exa-cel team. Medical writing, graphic support, and editorial coordination were provided by Nathan Blow, PhD, and Concetta G. Marfella, PhD, who are employees of Vertex Pharmaceuticals Incorporated and may hold stock and/or stock options at the company. CLIMB SCD-121 is sponsored by Vertex Pharmaceuticals Incorporated and CRISPR Therapeutics.

Presented at The Professional Society for Health Economics and Outcomes Research (ISPOR) Annual Scientific Meeting, May 5–8, 2023, Atlanta, GA, USA

Sponsored by Vertex Pharmaceuticals Incorporated