Methodological Approaches to Compare Treatment Options for Chronic Hepatitis D: An Early Network Meta-Analysis

SA8

MYR301

Barinder Singh[1], Shubhram Pandey[2], Sumeet Attri[2], Marvin Rock[3], Chong Kim[3]

[1]Pharmacoevidence, London, UK; [2]Pharmacoevidence, Mohali, India; [3]Gilead Sciences, Foster City, CA, USA

Conclusions

- Early network meta-analysis (NMA) enables timely identification of data and methodological challenges, allowing proactive mitigation, and guiding prioritization
- The suggested risk difference NMA approach proves valuable when conventional NMA faces challenges due to convergence issues related to rare events and single-study connections

Plain Language Summary

- The goal of this study was to understand how effective chronic hepatitis delta (CHD) treatments are and what challenges exist in comparing them
- The studies included in the systematic literature review (SLR) were organized to develop a visually connected network to help compare treatments
- This process faced initial challenges due to difficulties in linking the studies together and dealing with different statistical hurdles, such as convergence issues due to single study connections and rare data points
- A comparator similarity assumption was adopted to connect the network; further, the convergence issue was resolved by using the risk difference model
- In addition to the advanced population-adjusted meta-analysis methods, the risk difference method can be useful to facilitate the treatment comparisons in the presence of convergence issues

References:

1. Lempp FA, et al. *Nat Rev Gastroenterol Hepatol*. 2016;13; 580–89. 2. Hepcludex Assessment Report. *European Medicines Agency*. 2020. 3. Wedemeyer H, et al. *N. Engl. J. Med*. 2023; 389(1):22-32. 4. Buti M, et al. *J Hepatol*. 2022;77(S1):S4. 5. Warn DE, *Stat Med*. 2002; 21:1601–1623.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Editing and production assistance were provided by Pharmacoevidence

Disclosures: BS, SP, and SA are employees of Pharmacoevidence, and CK and MR are employees of Gilead Sciences, Inc.

Correspondence: Barinder Singh, barinder.singh@pharmacoevidence.com

Introduction

- CHD is the most severe form of viral hepatitis and is associated with progression to cirrhosis, decompensation, hepatocellular carcinoma, and end-stage liver disease[1]
- In 2020, the European Medicines Agency granted conditional marketing authorization to bulevirtide (BLV) 2 mg, as the first treatment approved for CHD[2]
- The Phase 3 MYR301 trial showed that BLV 2 mg administered for 48 weeks as a treatment for CHD was well tolerated and associated with significant reductions in hepatitis Delta virus (HDV) RNA and biochemical disease activity, with improvements in quality of life[3,4]

Objective

- Despite the comparative clinical studies evaluating BLV, Pegylated interferon (PEG-IFN), and anti-viral medications for CHD, an NMA is needed to systematically synthesize and consolidate the existing evidence
- This early NMA aims to elucidate the methodology, challenges encountered, and the corresponding mitigation strategies, enabling an effective comparison of treatments for CHD

Methods

- Following the guidelines outlined by the ISPOR Task Force, NICE UK, and the Cochrane Handbook, the initial stage of the NMA framework involved constructing network geometry and evidence network diagrams based on a systematic literature review
- Subsequent steps encompassed assessing heterogeneity, inconsistency, statistical models, assumptions, and model fit, followed by validation. Assumptions and modeling approaches were explored in pursuit of convergence and reliable NMA estimates

Results

- The first challenge in the NMA was the missing interconnected network due to the lack of a common comparator
- This issue was resolved by assuming similar efficacy of Nucleoside/nucleotide analogs (NATs) and control groups, as NATs are part of the background therapy among most CHD patients **(Figure 1)**
- Owing to the connected network involving closed loops, the conventional Bayesian NMA was feasible but encountered difficulties in achieving convergence for binomial outcomes due to single-study connections and rare or zero events
- Specifically, convergence challenges persisted in Bayesian odds ratio models, even when employing informative priors
- Furthermore, the Frequentist NMA approach produced confidence intervals with substantial width, indicative of pronounced uncertainty in the estimates and thus limiting the evidence synthesis to qualitative comparisons only

Figure 1: Network diagram of CHD treatments

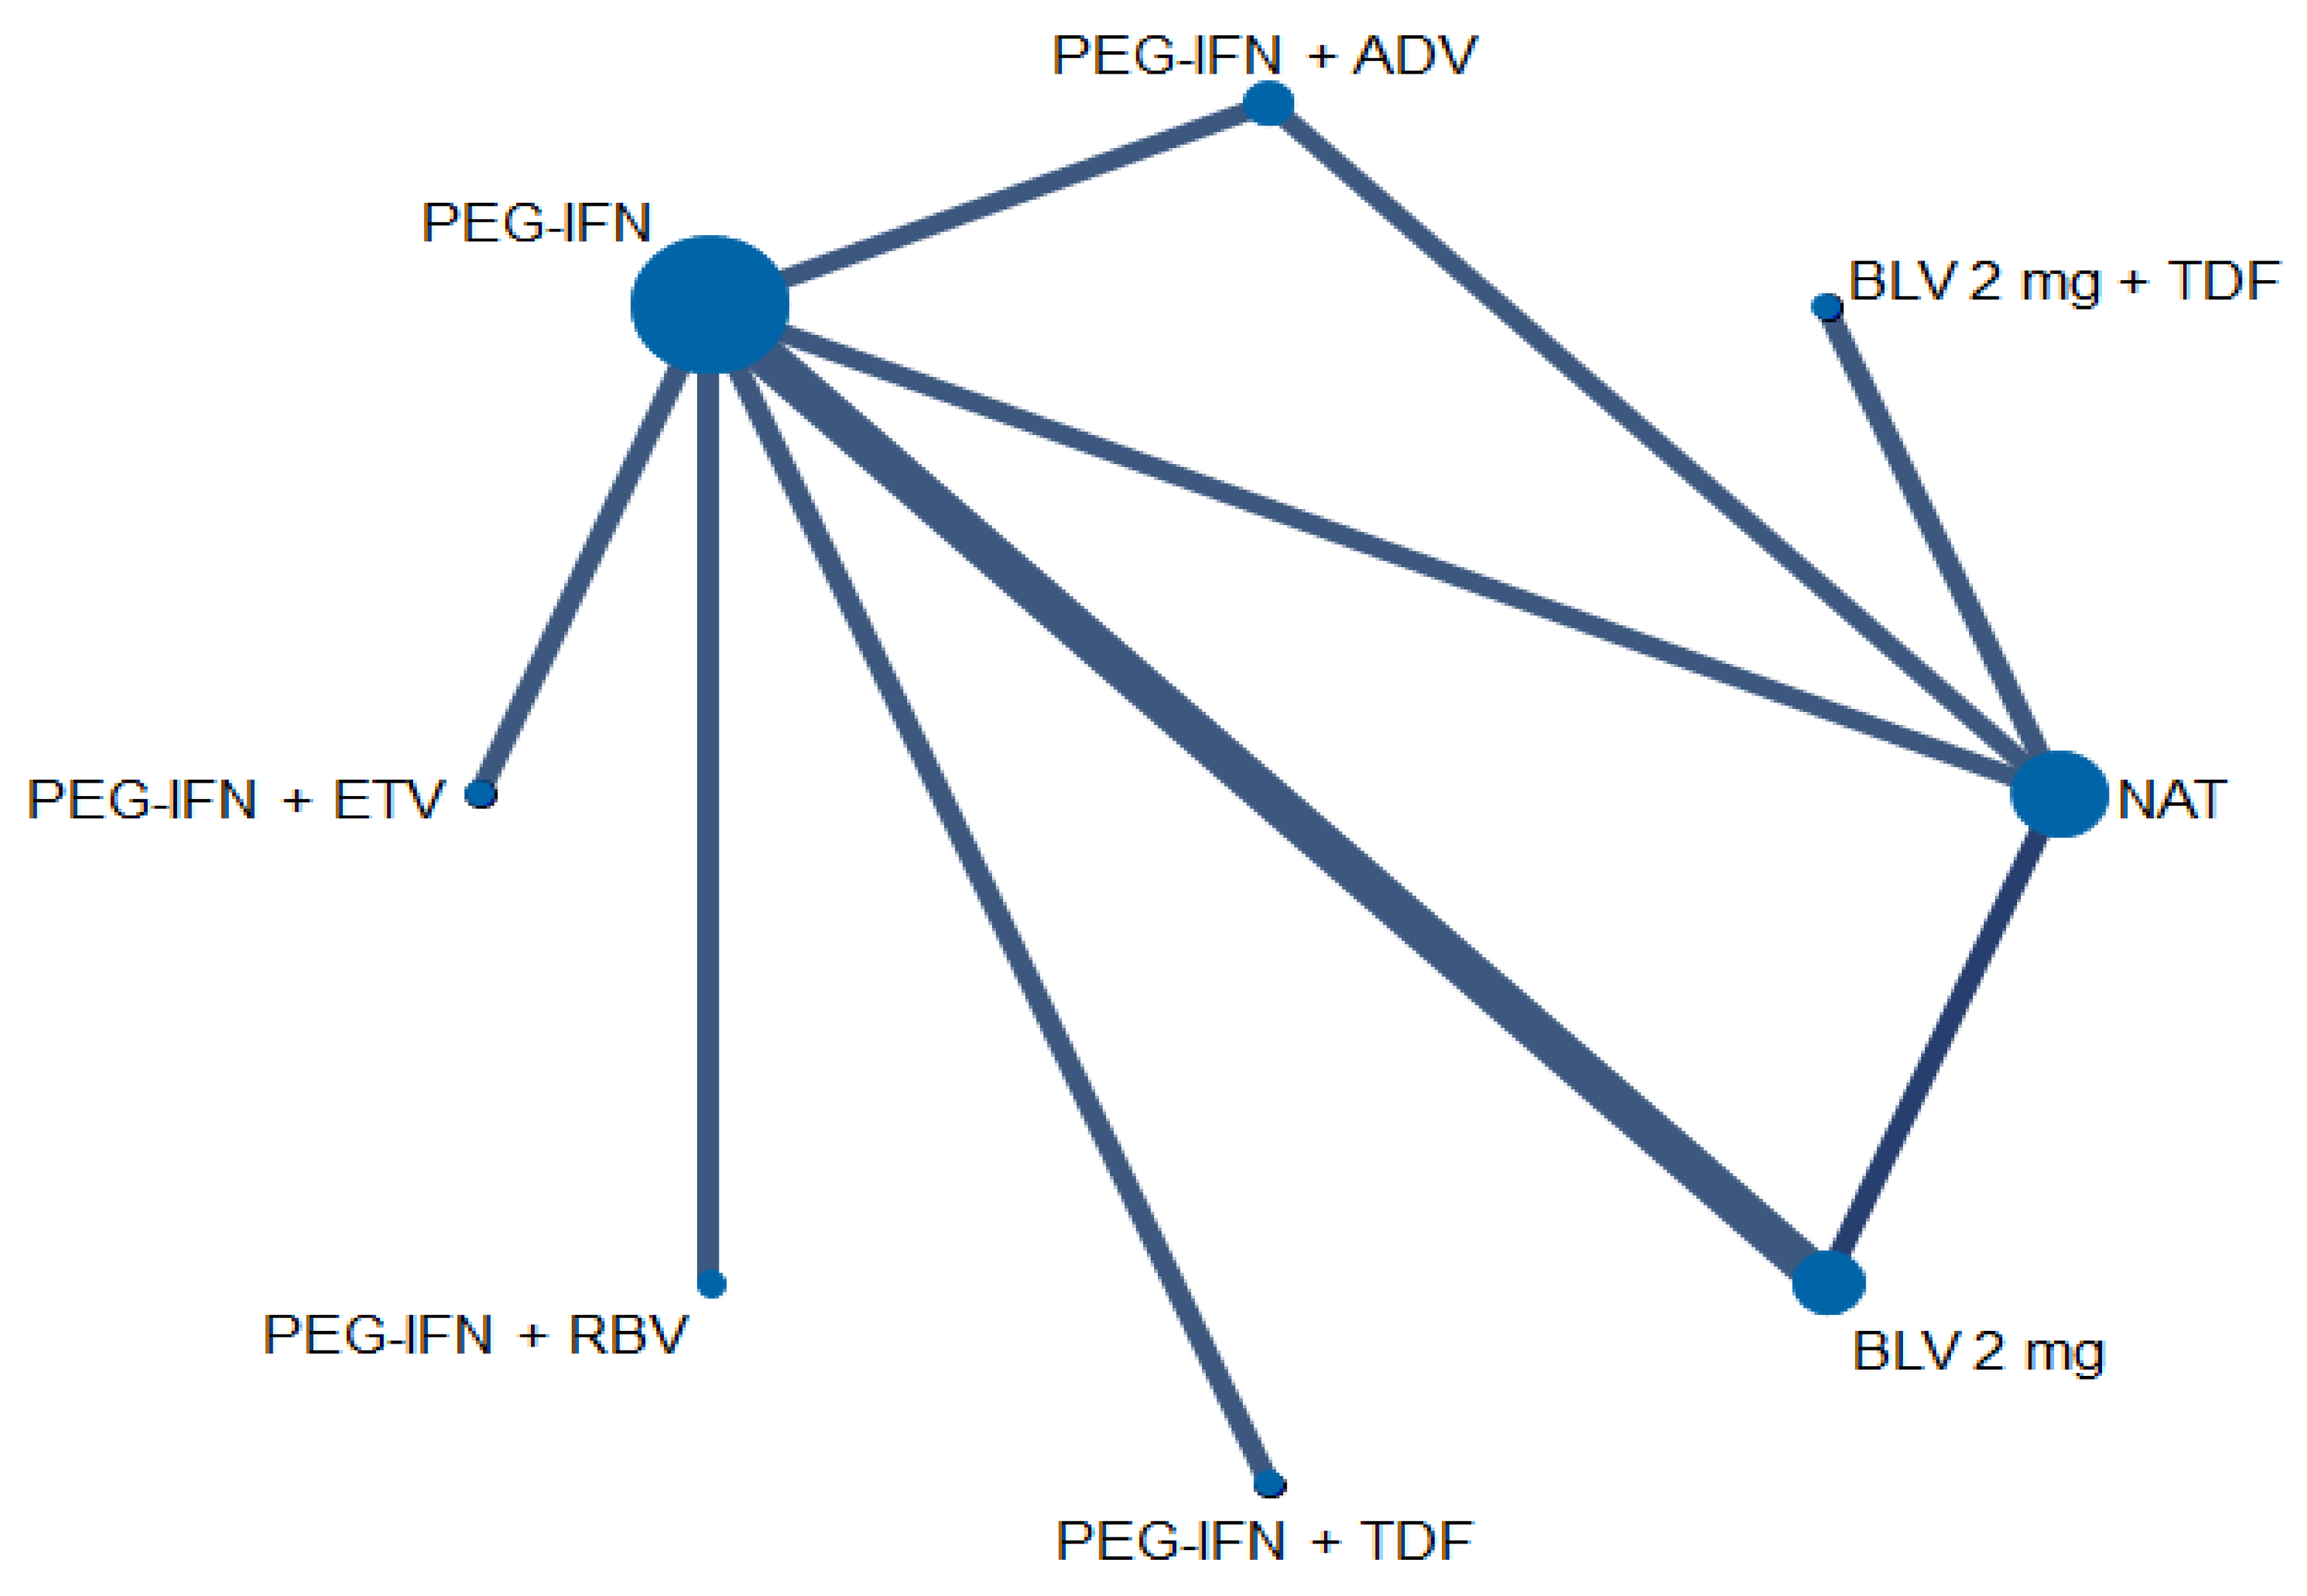
PEG-IFN + ADV
PEG-IFN
BLV 2 mg + TDF
PEG-IFN + ETV
NAT
PEG-IFN + RBV
BLV 2 mg
PEG-IFN + TDF

ADV, Adefovir; BLV, Bulevirtide; ETV, Entecavir; mg, milligram; NAT, Nucleoside/nucleotide analogue therapy; PEG-IFN, Pegylated Interferon; RBV, Ribavirin; TDF, Tenofovir

Results (Contd.)

- The convergence issue was resolved by employing a risk difference two-stage model[5], allowing for reliable NMA estimates for relative efficacy and economic model inputs **(Figure 2)**
- It should be noted that the absolute treatment effects and treatment rankings were broadly similar between the different models tested during the early NMA

Figure 2: Process flow of selection of final model

Problem

- The convergence issue was identified in the Bayesian odds ratio NMA model by using:
 a. Trace and density plots and
 b. Gelman–Rubin–Brooks methods

Solution-I

- To resolve the issue, Bayesian NMA with informative priors and Frequentist NMA models were tested
- However, the convergence issues continued to persist

Solution-II (Final Model)

- The convergence issue was resolved by using the risk difference two-stage model based on the approach suggested by Warn and colleagues[5]
- The next steps include completing the NMA according to feasibility guidance, as well as exploring the application of advanced meta-analytical techniques to the evidence base

Limitations

- The efficacy assumption between NAT and control groups may introduce heterogeneity into the network
- The Bayesian risk difference model involves complex two-stage calculations and is subject to the same limitations as the Bayesian approach, specifically sensitivity to priors

Copies of this poster obtained through QR (Quick Response) and /or text key codes are for personal use only and may not be reproduced without written permission of the authors

ISPOR US 2024; May 5-8, 2024; Atlanta, GA, USA