Estimands in EQ-5D-5L:
How to Deal with the "Worse Than Death" and the Intercurrent Events of "Death"

MSR53

Kunika Kikumori[1,2], Satoru Shinoda[1], Shoichi Ohwada[2], and Kouji Yamamoto[1]

[1] Department of Biostatistics, School of Medicine, Yokohama City University, Japan; [2] Daiichi Sankyo Co., Ltd., Japan

Background

Importance of the EQ-5D assessment

- Recently, health-related quality of life (HR-QOL) assessment has increasingly been considered important in both health technology assessments (HTAs) and clinical trials.
- The EuroQol 5 dimensions (EQ-5D) is one of the questionnaires commonly used for HR-QOL assessment. Among the EQ-5D, the EQ-5D-5L questionnaire consists of five questions rated on a five-level (1 to 5) scale.
- In clinical trials, EQ-5D-5L scores are often analyzed after they have been converted to Index Values based on a country-specific tariff [1][2][3] (Fig 1).

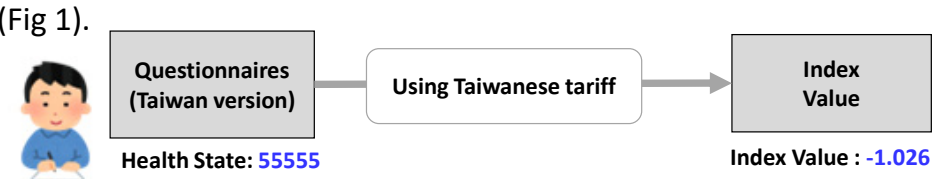
Questionnaires (Taiwan version)
Using Taiwanese tariff
Index Value
Health State: 55555
Index Value : -1.026

Although Index Values lies on a scale between 0 (state as bad as being dead) and 1 (full health state) but can be negative values. Negative Index Values are referred to as **worse than death (WTD)**.

Fig 1. Flow from EQ-5D-5L Questionnaire to Index Value.

Problems in the analysis of EQ-5D-5L Index Value

The problems in the analysis of EQ-5D-5L Index Values :

If both 'WTD' and 'Actual Death' occur in a trial, how should they be handled?

- In the field of cost-effectiveness analysis, an Index Value of an Actual Death case is often defined as zero (0) for quality-adjusted life years estimations [4].
- Also, in the field of clinical trials, an Index Value of an "Actual Death" is defined as zero (0) for some analyses [5].

The health state of the alive participants (WTD, Index Value < 0) appears to be worse than that of dead participants (Actual Death, Index Value = 0).

Example

Parallel-design placebo-controlled comparative study of a treatment A under development.

Clinical Question: EQ-5D-5L Index Values at Week T are compared between the patients who received treatment A and a Treatment B (placebo).

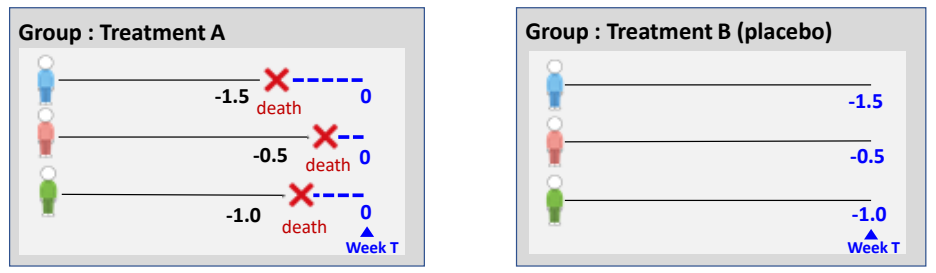
Group : Treatment A
-1.5
death
0
-0.5
death
0
-1.0
death
0
Week T
Group : Treatment B (placebo)
-1.5
-0.5
-1.0
Week T

Fig 2. Example.

- The QOL score in all patients died group is rated higher than that in all patients survived group.
- Consequently, the QOL in Treatment A is interpreted as being higher than that in Treatment B.
- This outcome might lead to misinterpretation of the statistics of Index Values in comparative clinical trials.

Objective

1) To consider **Handling of the co-incidence of WTD and Actual Death** in a study.
2) To resolve Objective 1, **the handling of ICEs systematically established using the estimand framework**.

Method

Two objectives achieved using Estimand Flamework

- There is no comprehensive, established strategy to solve this problem.

 Knowledge of estimand and how to handle intercurrent events (ICEs), which is necessary to plan analyses of EQ-5D-5L Index Values according to estimand, is limited (see Appendix in handout).

- **Handling of the co-incidence of WTD and Actual death in a study** varies depending on the target of estimation ("What is to be estimated").

Handling **Actual Deaths as ICEs**

- Estimand (ICH-E9(R1) in 2019) is often defined as "what is to be estimated" and consists of five attributes. (see Table 2. in handout)
- ICEs are one of these attributes and are handled differently depending on the strategies and endpoints. (see Table 3. in handout)
- **Handling of ICEs needs to be systematically established using the estimand framework.**

Strategies and estimands (ICEs: Actual Deaths)

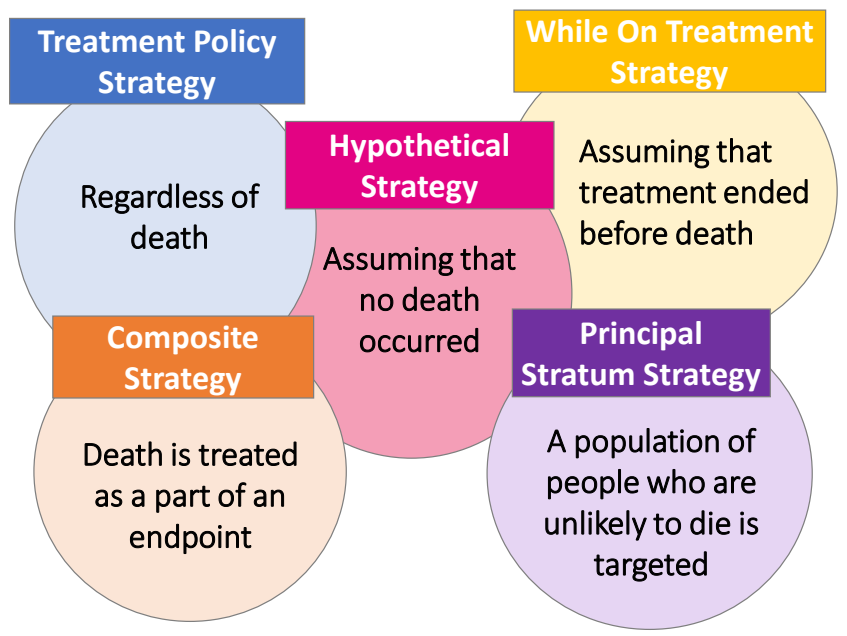
Treatment Policy Strategy
Regardless of death
Hypothetical Strategy
Assuming that no death occurred
While On Treatment Strategy
Assuming that treatment ended before death
Composite Strategy
Death is treated as a part of an endpoint
Principal Stratum Strategy
A population of people who are unlikely to die is targeted

Fig 3. Strategies and "What is to be estimated" (ICEs: Actual Death).

Evaluating EQ-5D-5L as Endpoint

Relationship between endpoints and estimands when EQ-5D-5L scores are evaluated as an endpoint.

Endpoints	What is to be estimated?
(1) Mean change from baseline at visit(t)	The subject of interest is the changes from baseline to Time Point t.
(2) Mean of value on treatment/study	The subject of interest is the means of values for **the whole treatment / study period**.
(3) Win-ratio	The subject of interest is the position of a certain factor X in the EQ-5D-5L scale when the factor X is considered.
(4) Responder Rate	The subject of interest is the percentage of study participants who achieved a response when Y is defined as the threshold.
(5) Time to deterioration (TTD)*	The subject of interest is the time until the score is worsened to threshold Z.

* TTD: Time to deterioration. Defined as the time from the study enrollment until a score is worsened to a given threshold.

Fig 4. General endpoints and estimand in EQ-5D-5L.

Results

Estimand strategies and endpoints

Handling of actual deaths as ICEs under different combinations of strategies (refer to Fig. 3) and endpoints (refer to Fig. 4)

Table1. Methods of ICEs treatment by study endpoints and estimand strategies.

Endpoint categories		Strategy: Treatment Policy	Hypothetical*3	Composite*1	While On Treatment	Principal Stratum
A. Value	(1) Change from Baseline at visit(t)	NA	Complemented by data obtained immediately before death.	Death is defined as zero (0).	NA	Populations of patients who are unlikely to die.
	(2) Mean of value on treatment on study	Data obtained immediately before death are included in the analysis.	- Death T(S)/D: Data of death are complemented. - Pre-death T(S)/D: Data are analyzed as they are.			
B. Ratio	(3) win-ratio at visit(t)	NA	Scores in the period after death are predicted.	Death is defined as the worst priority.*2		
	(4) Responder Rate at visit(t) by visit(t)	Patients who die after response are handled as responders.		Patients who die after response are handled as non-responders.		
C. Time To Event	(5) Time to deterioration	Data are analyzed as they are.	Death is ignored.	Death is deemed as an event.		

NA :not applicable; T/D : treatment discontinuation; S/D :study discontinuation

Estimand strategies and endpoints (cont'd)

***1,2) In the composite strategy,** the phenomenon of WTD < Actual Death is likely to occur because Actual Deaths are included in the analysis as a part of an endpoint.

Therefore, **a special consideration may be necessary** for handling ICEs.

Especially, the use of the **win-ratio as an endpoint** enables **deaths as the worst outcome** to be included in the analysis and enables analysis results to be provided under the assumption that Actual Deaths (ICEs) are worse than WTD.

3) In the hypothetical strategy, Actual Deaths are not related to EQ-5D-5L values because the analysis is conducted under the assumption that no Actual Death occurred.

Therefore, it is **not necessary to consider the influence of WTD**.

- Under the assumption that Actual Deaths are ICEs, We systematically tabulated the estimands under different combinations of strategy and endpoint.

 This table might be useful for selecting endpoints and determining how to handle endpoints according to what is to be estimated using the EQ-5D-5L.

- Using this comprehensive table, we could identify the strategy and endpoint that require us to consider the relationship between Actual Death and WTD.
- Our proposal might solve the WTD-related problem in analyses because the use of the win-ratio in the composite strategy allows for dealing with the health status of dead participants being a worse status than that of being alive participants with WTD.

Conclusion

The systematical summary of the endpoints and the handling of "Actual Death" using the estimand framework is useful to assess the health state based on EQ-5D-5L data in comparative clinical trials that may have patients with actual death and patients with WTD state.

Reference

[1] Ashe.et al.(2024).Perceived Health Status and Capability after Hip Fracture: Secondary Outcomes from a Randomized Controlled Trial

[2] Corral.et al.(2023).Minimal Clinically Important Differences in EQ-5D-5L Index and VAS after a Respiratory Muscle Training Program in Individuals Experiencing Long-Term Post-COVID-19 Symptoms

[3] Ushida.et al.(2023).Mirogabalin for Central Neuropathic Pain After Spinal Cord Injury : A Randomized, Double-Blind, Placebo-Controlled, Phase 3 Study in Asia

[4] Singh.et al.(2022).An Analysis of 5-Level Version of EQ-5D Adjusting for Treatment Switching: The Case of Patients With Epidermal Growth Factor Receptor T790M-Positive Nonsmall Cell Lung Cancer Treated With Osimertinib

[5] Sampson.(2020).Drop Dead: Is Anchoring at 'Dead' a Theoretical Requirement in Health State Valuation?

Contact

Kunika Kikumori

<Kunika.Kikumori@daiichisankyo.com>