Assessment of rare disease reimbursement levels and economic burden on patients in China

HPR65

Ye CHEN[1], Xinyang CHEN[1], Yi DENG[1], Jing YAN[2], Jiayu HUANG[2], Jinxi DING[1]

1. China Pharmaceutical University, Nanjing, Jiangsu, China; 2. Ipsen, Shanghai, China

Background

- China has established a comprehensive, multi-payer reimbursement system for rare diseases, in which the National Reimbursement Drug List (NRDL) serves as the cornerstone for the system and Medical Assistance Programs[1] (MAP) provide crucial support. Furthermore, efforts are underway to collaboratively develop the City Customized Commercial Medical Insurance[2] (CCCMI) to alleviate the economic burden on patients.

Objective

- The purpose of this study was to assess the reimbursement level and economic burden for Chinese patients with rare diseases within the current reimbursement system, and to explore the challenges faced by policymakers and stakeholders.

Methods

With Chinese reimbursement policies of 253 cities being reviewed, a descriptive statistical analysis was conducted to analyze the economic burden of urban and rural patients with 10 rare diseases with available medical treatments using the adjusted World Health Organization (WHO) / Health Action International (HAI)[3] standard survey method.

Selection criteria on diseases and drugs

- Rare diseases included in outpatient chronic and special disease lists of most cities were chosen, within which 1-2 drugs with the highest market share in China for each disease area were selected for the analysis.

Adjustments on WHO/HAI standard survey method

- The WHO/HAI standard survey method calculates the duration of **government non-technical personnel's minimum wage** required to cover the medication expenses associated with treating a specific disease at a standard dosage within the treatment course.
- Given the significant wealth disparity in China and the long-term nature of rare disease drug usage, adjustments were implemented following the framework proposed by Shin et al. to suit the Chinese context. In this adjustment, **the government non-technical personnel's minimum wage was substituted with the per capita annual disposable income of Chinese urban residents** (I_j) to calculate the number of working years necessary (N_{rj}) to cover the annual out-of-pocket (OOP) medication costs (E_r).
- N_{rj} <=1 means that the drug is deemed to be affordable, otherwise, it is unaffordable.

$$N_{rj} = \frac{E_r}{I_j} = \frac{n \times DDD \times P_r}{V_r \times I_j}$$

j refers to the city and r refers to the drug

N=working years required to cover the annual treatment cost

E=annual out-of-pocket treatment cost

I=Chinese urban resident's annual per capita disposable income

n=annual days spent using the drug

P=minimum retail price of the drug based on lowest national bidding price over last three years

V=packaged dosage of the drug

DDD=defined daily dose

Table 1 10 rare disease drugs – basic information

Rare disease	Parkinson Disease (Young-onset, Early-onset)		Systemic Sclerosis	Acromegaly	Multiple Sclerosis		Hemophilia		Niemann-Pick Disease	Fabry Disease
Medication	Drug 1	Drug 2	Drug 3	Drug 4	Drug 5	Drug 6	Drug 7	Drug 8	Drug 9	Drug 10
Annual cost (¥, CNY)	3,059	10,768	36,456	62,140	102,930	88,330	170,663	209,243	280,320	286,971
Enlisted in NRDL (Y/N)	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y
Average actual reimbursement ratio (%)	60.87%	40.90%	25.51%	64.89%	48.30%	55.06%	64.76%	64.12%	63.21%	73.51%
N (years)	0.03	0.15	0.64	0.41	1.11	0.96	1.46	1.17	2.19	1.78
Enlisted in NRDL & CCCMI (Y/N)	N	N	Y	N	Y	Y	Y	Y	Y	N
N (years)	-	-	0.58	-	0.98	0.86	1.05	0.89	1.13	-
Enlisted in NRDL & CCCMI & MAP (Y/N)	N	N	N	N	N	N	Y	N	Y	N
N (years)	-	-	-	-	-	-	0.8	-	0.82	-

Note: N: The number of working years required to cover the annual treatment cost

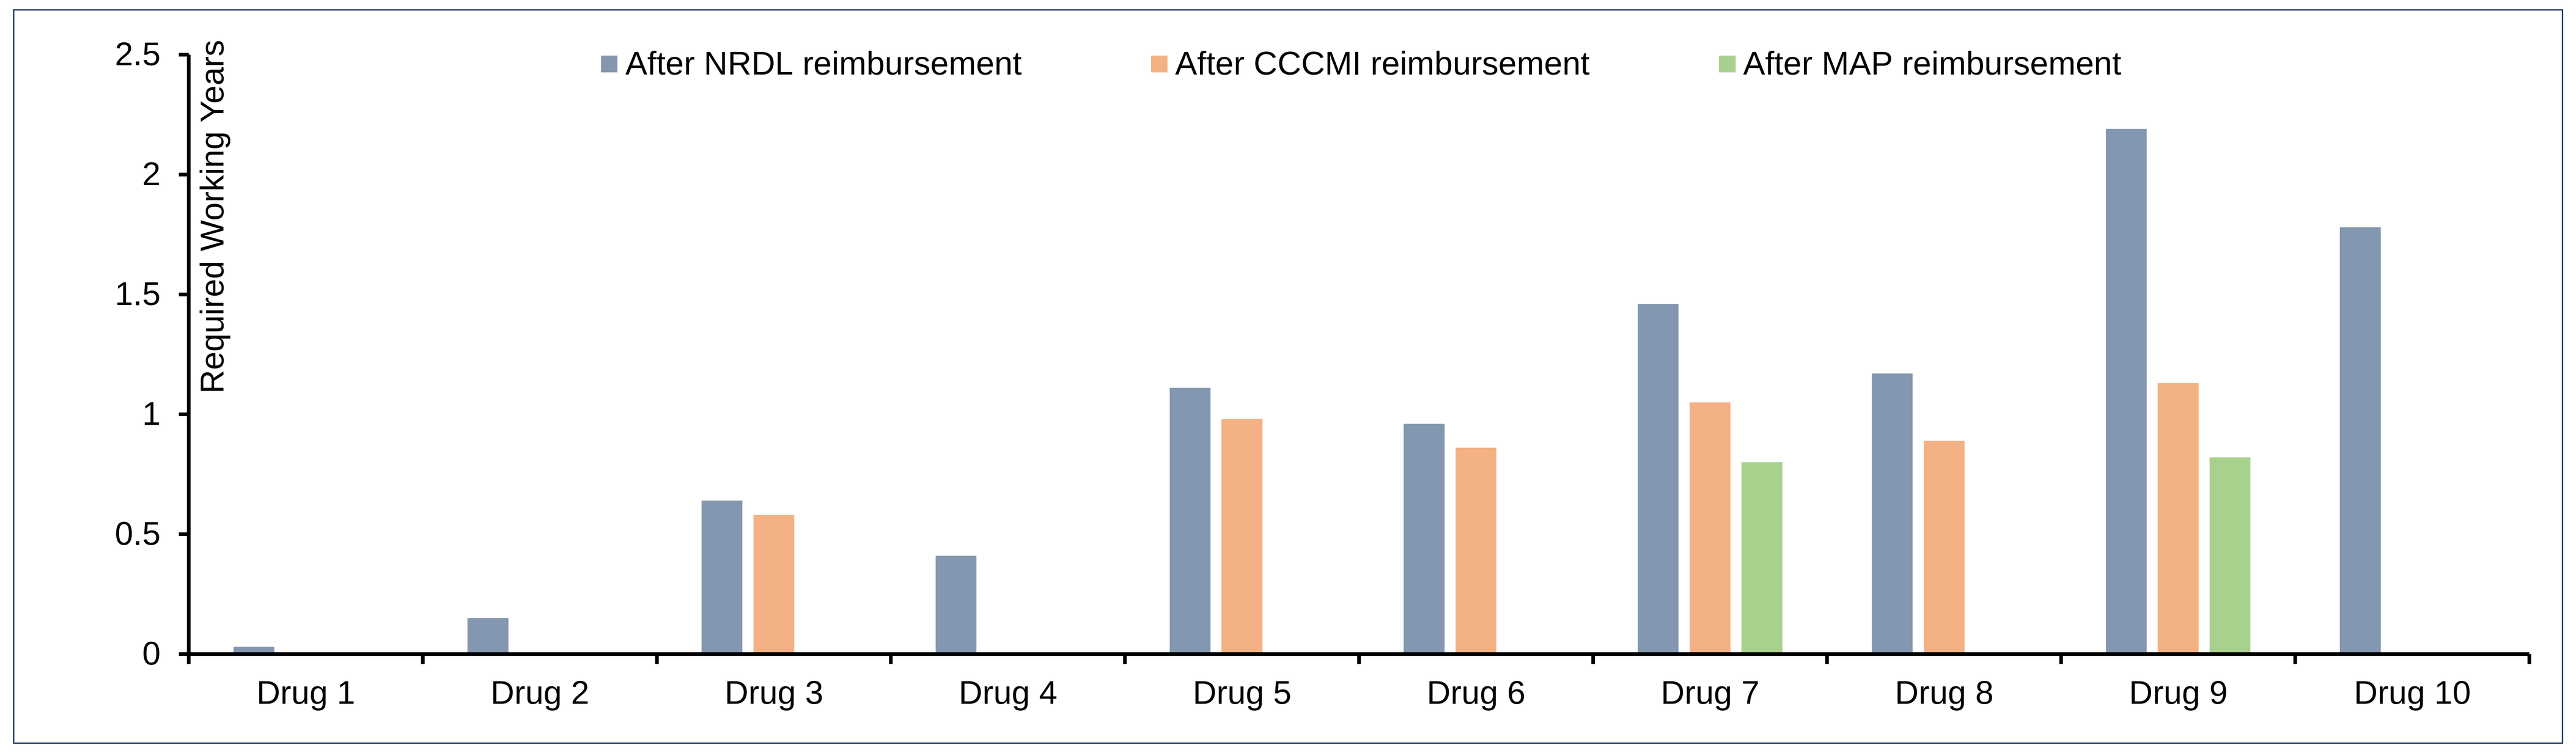
After NRDL reimbursement
After CCCMI reimbursement
After MAP reimbursement
Required Working Years
2.5
2
1.5
1
0.5
0
Drug 1
Drug 2
Drug 3
Drug 4
Drug 5
Drug 6
Drug 7
Drug 8
Drug 9
Drug 10

Graph 1 Number of working years required to cover the annual treatment cost for rare disease drugs enlisted in NRDL

Table 2 6 rare disease drugs only included in CCCMI – basic information

Rare disease	Mucopolysaccharidosis			Glycogen Storage Disease	Gaucher's disease	
Medication	Drug 11	Drug 12	Drug 13	Drug 14	Drug 15	Drug 16
Annual cost (¥, CNY)	2,340,000	1,450,000	1,430,000	2,070,000	1,210,000	1,560,000
N (years)	37.1	19	17.2	31.3	15.1	21.4

Note: N: The number of working years required to cover the annual treatment cost

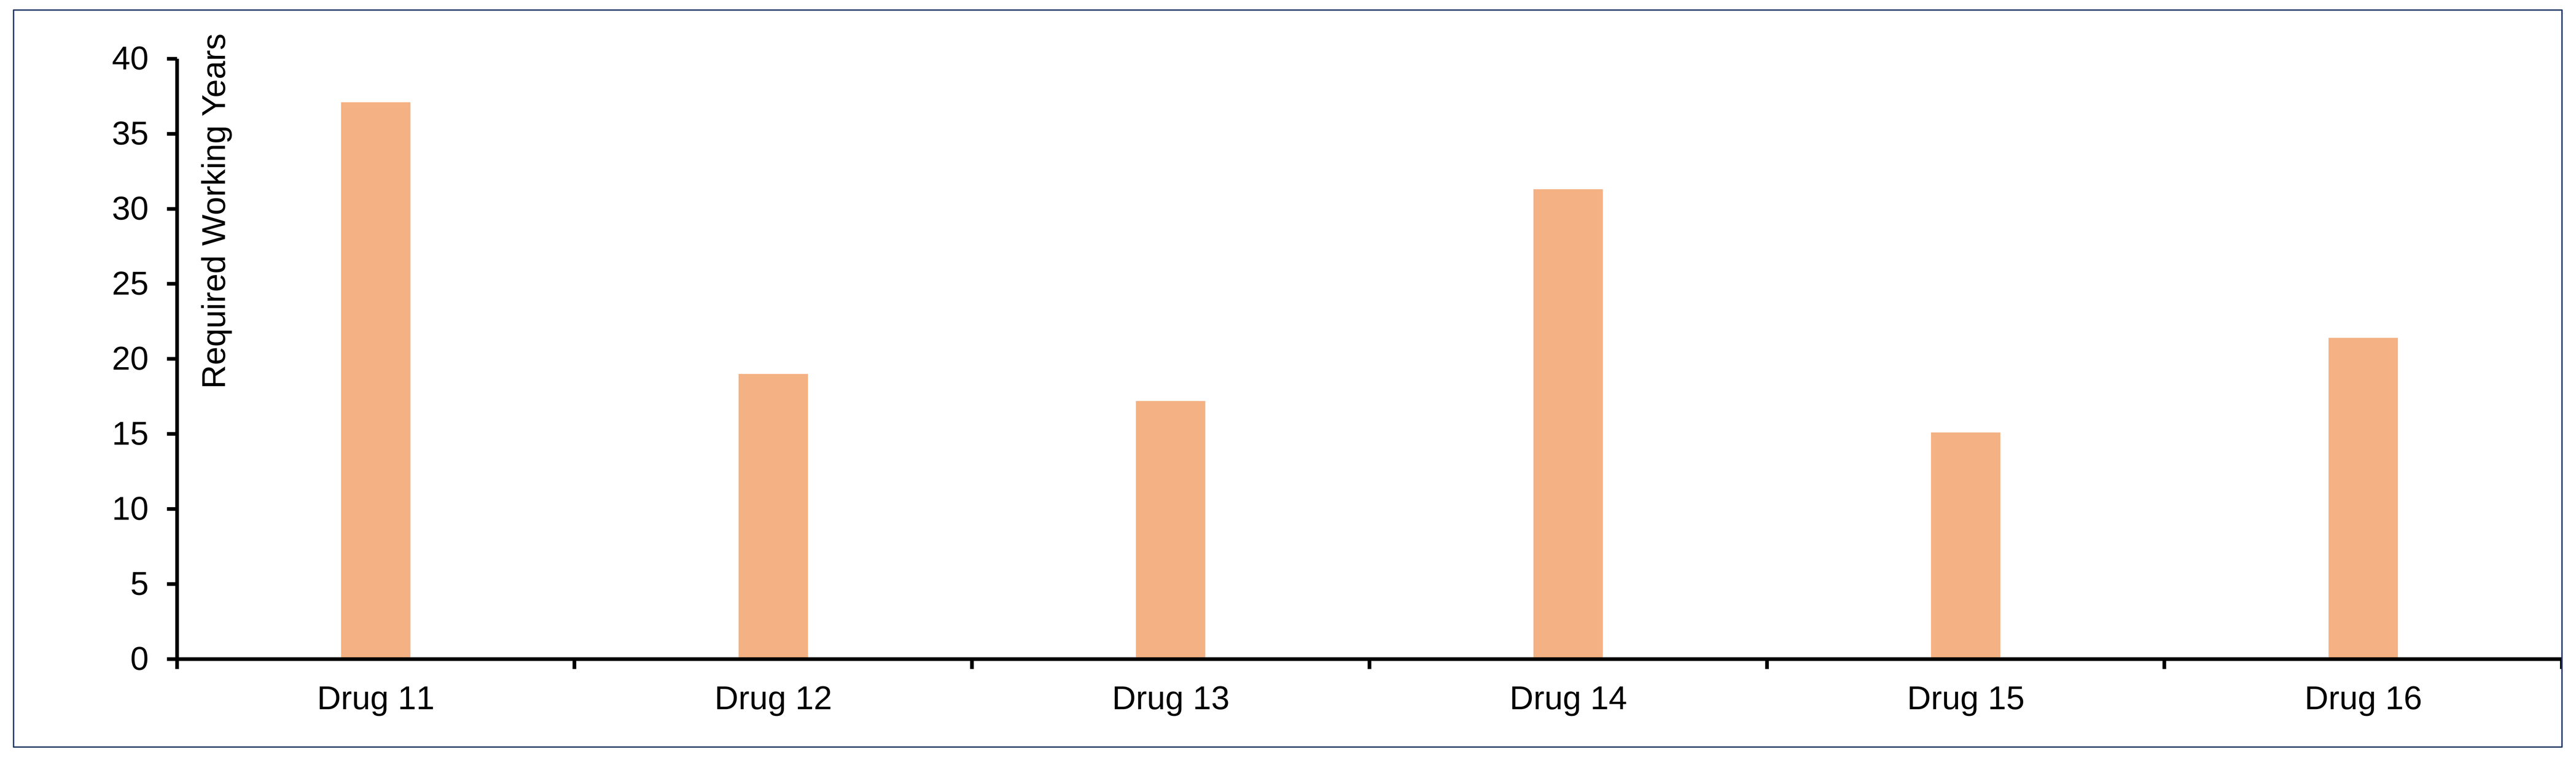
Required Working Years
40
35
30
25
20
15
10
5
0
Drug 11
Drug 12
Drug 13
Drug 14
Drug 15
Drug 16

Graph 2 Number of working years required to cover the annual treatment cost for rare disease drugs outside NRDL

Results

For drugs enlisted in NRDL

There were 16 drugs available for the 10 selected rare diseases. NRDL covered 10 of these drugs, with an average reimbursement rate of 56%.

- Both **Table 1** and **Graph 1** show that 5 low-cost drugs used to treat Parkinson's disease (young-onset, early-onset), systemic sclerosis, acromegaly, and partial multiple sclerosis were found to be affordable, with the required working years ranging from 0.03 to 0.96. However, the remaining 5 high-cost drugs, which were utilized for hemophilia, Niemann-Pick disease, Fabry disease, and partial multiple sclerosis treatment, were deemed unaffordable.
- Patients covered by CCCMI had secondary reimbursement after NRDL to further reduce OOP costs, but for drug 7 and drug 9, it took ≥ 1 working year to pay the OOP costs, as showed in **Table 1** and **Graph 1**.
- If patients qualified for MAP reimbursement, OOP costs for these 2 drugs could be further reduced (**Table 1**).

For drugs outside NRDL

- The remaining 6 non-NRDL drugs were covered by CCCMI, both **Table 2** and **Graph 2** show that these drugs require more than 15 working years for patients to cover the OOP payments.

Conclusion

- It is essential to optimize the existing Chinese healthcare reimbursement mechanism and enhance the coordination and supplement across various reimbursement levels to further reduce economic burden for patients with rare diseases in China.

References

1. Patients whose annual medical expenses comprise 40-60% of their household's income are eligible to apply for medical assistance through the local healthcare administration. The specific percentage of assistance provided varies from city to city.
2. CCCMI is a supplementary health insurance overseen by the respective local government authorities. This program is commercially administered by insurance companies and offers voluntary enrollment to residents.
3. WHO, HAI. Medicine prices: MEASURING MEDICINE PRICES, AVAILABILITY, AFFORDABILITY AND PRICE COMPONENTS (2ND ED.)[EB/OL].（2016） [2023-09-20].https://haiweb.org/publication/measuring-medicine-prices-availability-affordability-and-price-components-2nd-ed/

Author Contribution

Substantial contributions to study conception/design, acquisition/analysis/interpretation of data, drafting of the publication, revising it critically for important intellectual content and final approval of the publication: Y C, X C, Y D, J Y, J H, J D.

Presented at ISPOR 2024 | Atlanta, Georgia, USA | 5-8 May 2024.

Disclosures

Y C, X C, Y D, J D received: research grants from Ipsen; **J Y**, **J H** are employees of Ipsen.

Acknowledgements

The authors acknowledge the work of Jiahao LI, who was an author on the original abstract but was unable to take part in the poster development process.

This study was sponsored by Ipsen