EE495

A targeted literature review of lung function decline in idiopathic pulmonary fibrosis to improve survival predictions in cost-effectiveness analyses

Olivia M. Dong,[1] William L. Herring,[1] Pooja Chopra,[2] Brandon Elpers,[2] Kasper M. Johannesen[3]

[1] RTI Health Solutions, Research Triangle Park, NC, United States; [2] Bristol Myers Squibb, Lawrenceville, New Jersey, United States; [3] Bristol Myers Squibb, Stockholm, Sweden

Background

- Idiopathic pulmonary fibrosis (IPF) is a rare, debilitating, chronic, progressive fibrotic interstitial lung disease of unknown etiology.[1] In IPF, fibrosis causes irreversible lung function loss, which has been shown to impact patients' quality of life and overall survival.[2,3]
- Mean survival for patients with IPF has been estimated to be 3 to 5 years,[4] although disease progression among patients is highly variable and difficult to predict.[5]
- Cost-effectiveness (CE) models for IPF treatments that have been submitted to the National Institute for Health and Care Excellence in the United Kingdom for health technology assessments have not explicitly linked lung function decline to survival, with companies citing a lack of suitable data.[6]
- In its appraisals of antifibrotic treatments for IPF, the National Institute for Health and Care Excellence expressed concern about this limitation, suggesting a need to improve mortality modeling approaches in future CE analyses for IPF treatments in development.[6]

Objective

- Our objective was to review the IPF disease progression literature for compatibility with existing IPF mortality prediction models to inform approaches explicitly linking lung function to survival in future CE analyses.

Methods

- Clinical experts identified 3 IPF mortality prediction models that link lung function decline to survival for consideration based on their real-world clinical utility: the Gender-Age-Physiology (GAP) model,[7] the longitudinal GAP model,[8] and a model by du Bois et al.[9]
 - These 3 mortality prediction models relied on percent predicted forced vital capacity (ppFVC); at least 1 of history of respiratory hospitalization, percent predicted diffusing capacity of the lung for carbon monoxide (ppDLCO), or 6-minute walking distance (6MWD); and demographic variables (Figure 1).
- We completed a targeted literature review of the IPF disease progression literature as of 1 October 2023 for studies reporting demographic data (gender, age), baseline clinical variables (ppFVC, ppDLCO, 6MWD, history of respiratory hospitalization or acute exacerbation), and longitudinal clinical variables (rates of ppFVC, ppDLCO, and 6MWD change over 24 weeks, and acute exacerbation or respiratory hospitalization risks), as well as correlations among these model variables.
 - Searches were conducted on PubMed and Google Scholar using various combinations and variations of the terms "idiopathy pulmonary fibrosis," "sex," "gender," "age," "ppFVC," "ppDLCO," "6MWD," "history of," "acute exacerbation," "change in," "correlation," "rates," "risk," "respiratory hospitalization," "24-week change," and "lung function." Results were restricted to references published in English.
 - Although change in ppDLCO is not directly used in any of the 3 mortality prediction models, it was included in the targeted literature review because it may be needed to predict ppDLCO progression when reevaluating mortality risk over time using the GAP and longitudinal GAP models.
 - When assessing the literature, acute exacerbations were assumed to result in respiratory-related hospitalizations.
- Studies were included if they provided key parameter inputs or provided evidence to support assumptions needed to link lung function to mortality in a potential CE model. Preference was given to recent systematic reviews, meta-analyses, randomized controlled trials, seminal references, and studies that used robust study methods.

Results

Overview of Included Studies

- We identified 10 key studies reporting relevant demographic data (gender, age), baseline variables (ppFVC, ppDLCO, 6MWD, history of acute exacerbation), or longitudinal variables (rates of ppFVC, ppDLCO, and 6MWD change over 24 weeks, and acute exacerbation risks), including correlations among select model variables (Table 1).
 - These studies suggested a heterogeneous disease course, especially in patterns of ppFVC decline and acute exacerbations.[11-14]
 - Study findings indicated that baseline ppFVC level was fairly and poorly correlated with baseline ppDLCO and 6MWD levels, respectively.[10,15] The rate of ppFVC change was poorly correlated with rates of change for ppDLCO and 6MWD and associated with acute exacerbation risk.[10,14,15]
 - Correlation coefficients that were reported among mortality prediction variables ranged from 0.12 to 0.38 (Table 1).
 - Annual incidence rate per patient of acute exacerbation risk ranged from 0.26 to 0.74 depending on the rate of ppFVC change over 6 months (Table 1).
- Limited data were available to inform other potential relationships (notably, the correlation between baseline ppFVC level and change in ppFVC).

Implications for Linking Lung Function to Survival in CE Analyses

- Figure 2 presents the lung function variables that are linked to mortality and the relationships among the variables identified in the included studies.
- Table 2 outlines key implications when implementing IPF mortality prediction models in CE analyses for IPF treatments.

Figure 1. Characteristics of IPF mortality prediction models

Model	Description	Mortality prediction timepoints
GAP[7]	Validated model that uses commonly measured clinical baseline variables	1-year, 2-year, and 3-year risks
Longitudinal GAP[8]	Model that includes GAP model baseline variables plus the addition of 2 longitudinal variables	1-year and 2-year risks
du Bois et al.[9]	Model that includes baseline and longitudinal variables	1-year risk

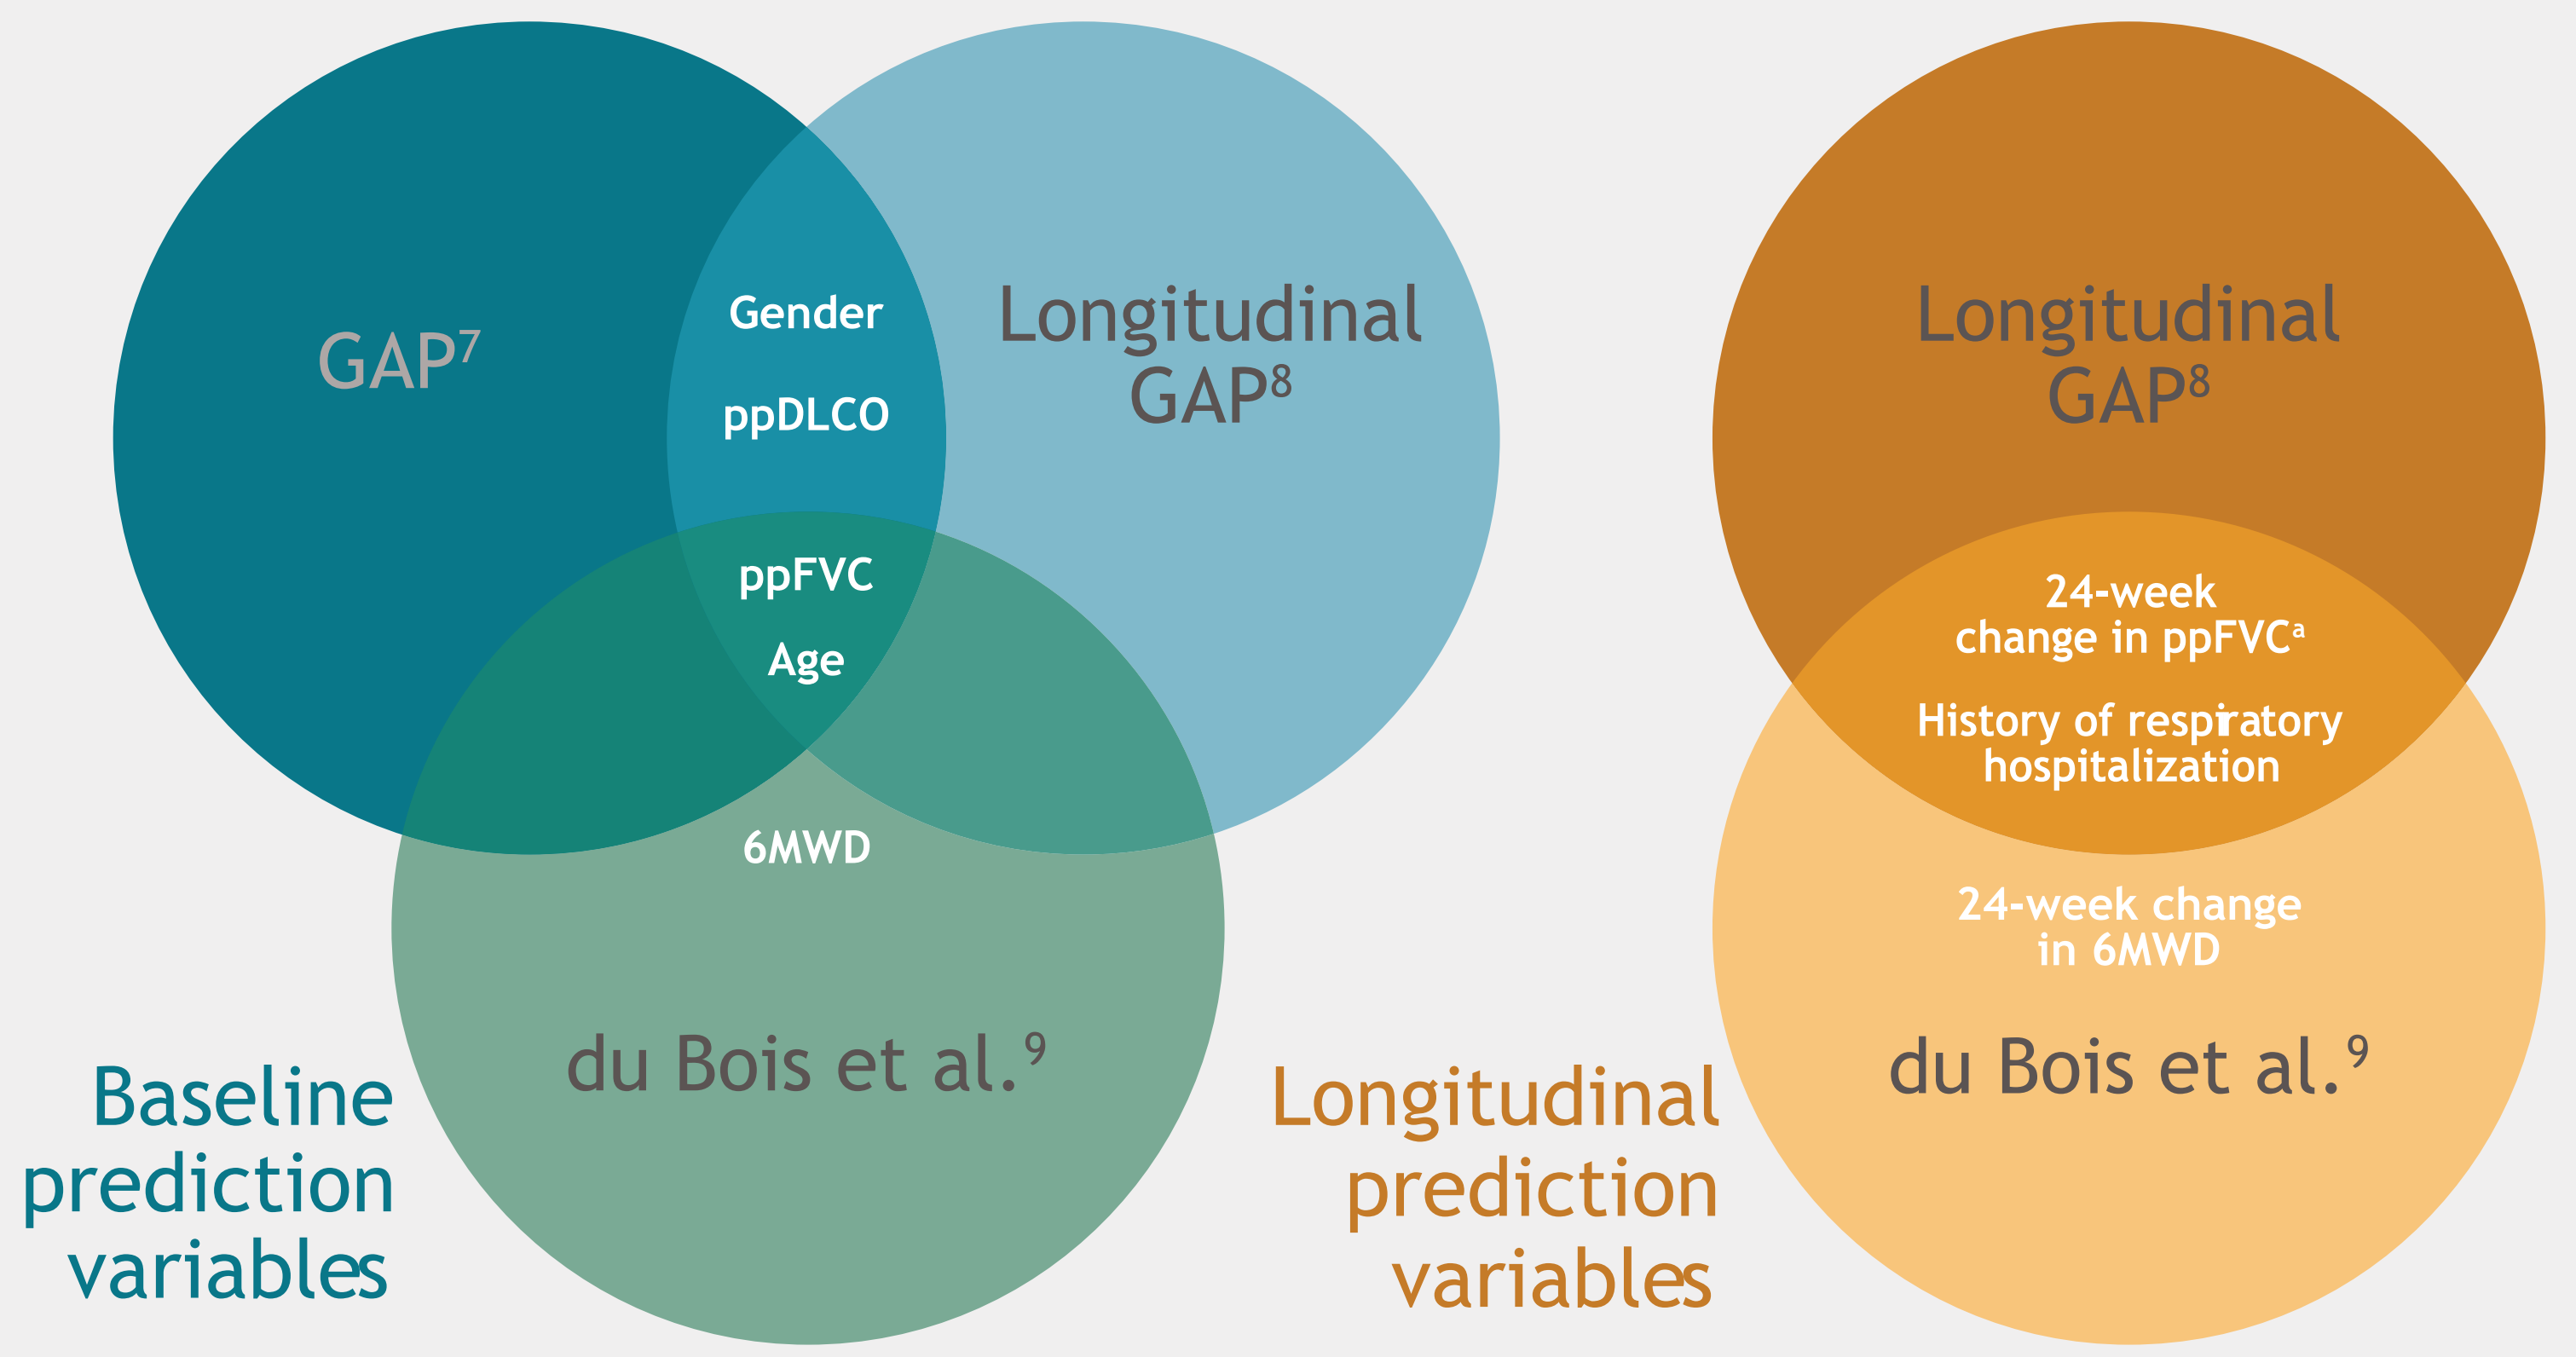
GAP[7]
Longitudinal GAP[8]
Gender
ppDLCO
ppFVC
Age
6MWD
du Bois et al.[9]
Baseline prediction variables
Longitudinal GAP[8]
24-week change in ppFVC[a]
History of respiratory hospitalization
24-week change in 6MWD
du Bois et al.[9]
Longitudinal prediction variables

[a] 24-week change in ppFVC is relative for the longitudinal GAP model[8] and is absolute for the du Bois model.[10]

Table 1. Characteristics of included studies

Study	Year	Study design	Study population	Relevant data and assumptions for mortality prediction models	Details of inputs and assumptions
Demographic and baseline clinical variables					
Song et al.[11]	2011	Retrospective cohort study	461 patients with IPF in South Korea	Proportion with baseline history of acute exacerbation, ppFVC mean and SD with and without history of acute exacerbation	• Proportion with baseline history of acute exacerbation: 20.8% • Baseline mean ppFVC with history of acute exacerbation: 72.0 (SD, 15.7) • Baseline mean ppFVC with no history of acute exacerbation: 77.6 (SD, 17.0)
du Bois et al.[9]	2014	Post hoc analysis of RCT data	748 patients with IPF from multiple countries	Gender, age, ppDLCO mean and SD; 6MWD mean and SD	• Proportion of cohort female: 28.5% • Mean age: 66.0 years (SD, 7.6) • Baseline ppFVC: 72.5 (SD, 12.8) • Baseline mean ppDLCO: 47.5 (SD, 9.2) • Baseline mean 6MWD: 397 (SD, 107)
Ley et al.[7]	2012	Retrospective cohort study	228 patients in the US with IPF (derivation cohort), 330 patients in the US and Italy with IPF (validation cohort)	Assumptions to characterize patients that are no longer able to perform the DLCO	• Patients with very poor lung function may not be able to perform the DLCO test,[7] although the exact ppDLCO cutoff value has not been reported - ppDLCO values were collected for patients every 6 months with a median follow-up of 35.1 months.[16] When ppDLCO values were graphed, values ≤ 10 seem sparse, which could be due to not being able to perform the test - 12 of 66 patients with advanced IPF (ppFVC < 50% and/or ppDLCO < 30%) could not perform DLCO[17]
Durheim et al.[17]	2021	Retrospective cohort study	502 patients enrolled in IPF registries in 4 Nordic countries		
Neely et al.[16]	2023	Retrospective cohort study	941 patients with IPF enrolled in the US IPF-PRO registry		
Longitudinal clinical variables					
Collard et al.[18]	2013	Post hoc analysis of RCT data[18,19]	180 patients with IPF in the US[19]	Risk of acute exacerbation	• Incidence of definite acute exacerbation per patient-year with IPF: 0.04 (CI, 0.10-0.12) • Incidence of suspected acute exacerbation per patient-year with IPF[a]: 0.16 (CI, 0.09-0.26) • Using these inputs in a CE model requires assuming that acute exacerbations result in respiratory-related hospitalizations
Khor et al.[12]	2020	Systematic review and meta-analysis (154 cohort studies and 16 RCTs)	Various number of patients with IPF (n = 1000 for ppFVC input, n = 277 for ppDLCO input, n = 444 for 6MWD input)	ppFVC, ppDLCO, and 6MWD decline	For patients not on antifibrotic drugs: • At < 2 years, mean change in ppFVC was −6.76 (95% CI, −8.92 to −4.61) • At 1 year to < 2 years, mean change in ppDLCO was −3.33 (95% CI, −5.14 to −1.52) • At 1 year to < 2 years, mean change in 6MWD was −37 (95% CI, −88 to 15)
Lee et al.[13]	2021	Retrospective, observational study	295 patients with IPF in South Korea	ppFVC and ppDLCO decline	For patients not on antifibrotic drugs: • Annual mean ppFVC decline: −9.85 (SD, 11.43) • Annual mean ppDLCO decline: −11.695 (SD, 16.819)
Correlations among variables					
du Bois et al.[10]	2011	Post hoc analysis of RCT data	1156 patients with IPF from multiple countries[10,20,21]	Correlations: ppDLCO and ppFVC, ppFVC and 6MWD; change in ppDLCO and change in ppFVC, change in 6MWD, and change in ppFVC inputs	Correlation between ppFVC and: • ppDLCO: r = 0.38 • 6MWD: r = 0.12 Correlation between absolute 24-week change in ppFVC and: • 24-week absolute change in ppDLCO: r = 0.29 • 24-week change in 6MWD: r = 0.22
Reichmann et al.[14]	2015	Retrospective cohort study	490 patients with IPF in the US	Risk of acute exacerbation based on ppFVC decline	12-month incidence rate per patient of suspected acute exacerbation[b] stratified by 6-month relative change in ppFVC: • < 5% decline in ppFVC: 0.26 • ≥ 5% to < 10% decline in ppFVC: 0.47 • ≥ 10% decline in ppFVC: 0.74

IPF-PRO = idiopathic pulmonary fibrosis prospective outcomes; RCT = randomized controlled trial; SD = standard deviation; US = United States.
[a] Suspected acute exacerbation was defined in the study by an idiopathic acute respiratory worsening that could not be categorized as a definite acute exacerbation because of missing data or criteria.
[b] Suspected acute exacerbation was defined in the study by having pulmonologists evaluate outpatient visits, emergency room visits, or hospitalizations to determine whether they were related to an IPF acute exacerbation.

Figure 2. Summary of associations among lung function variables

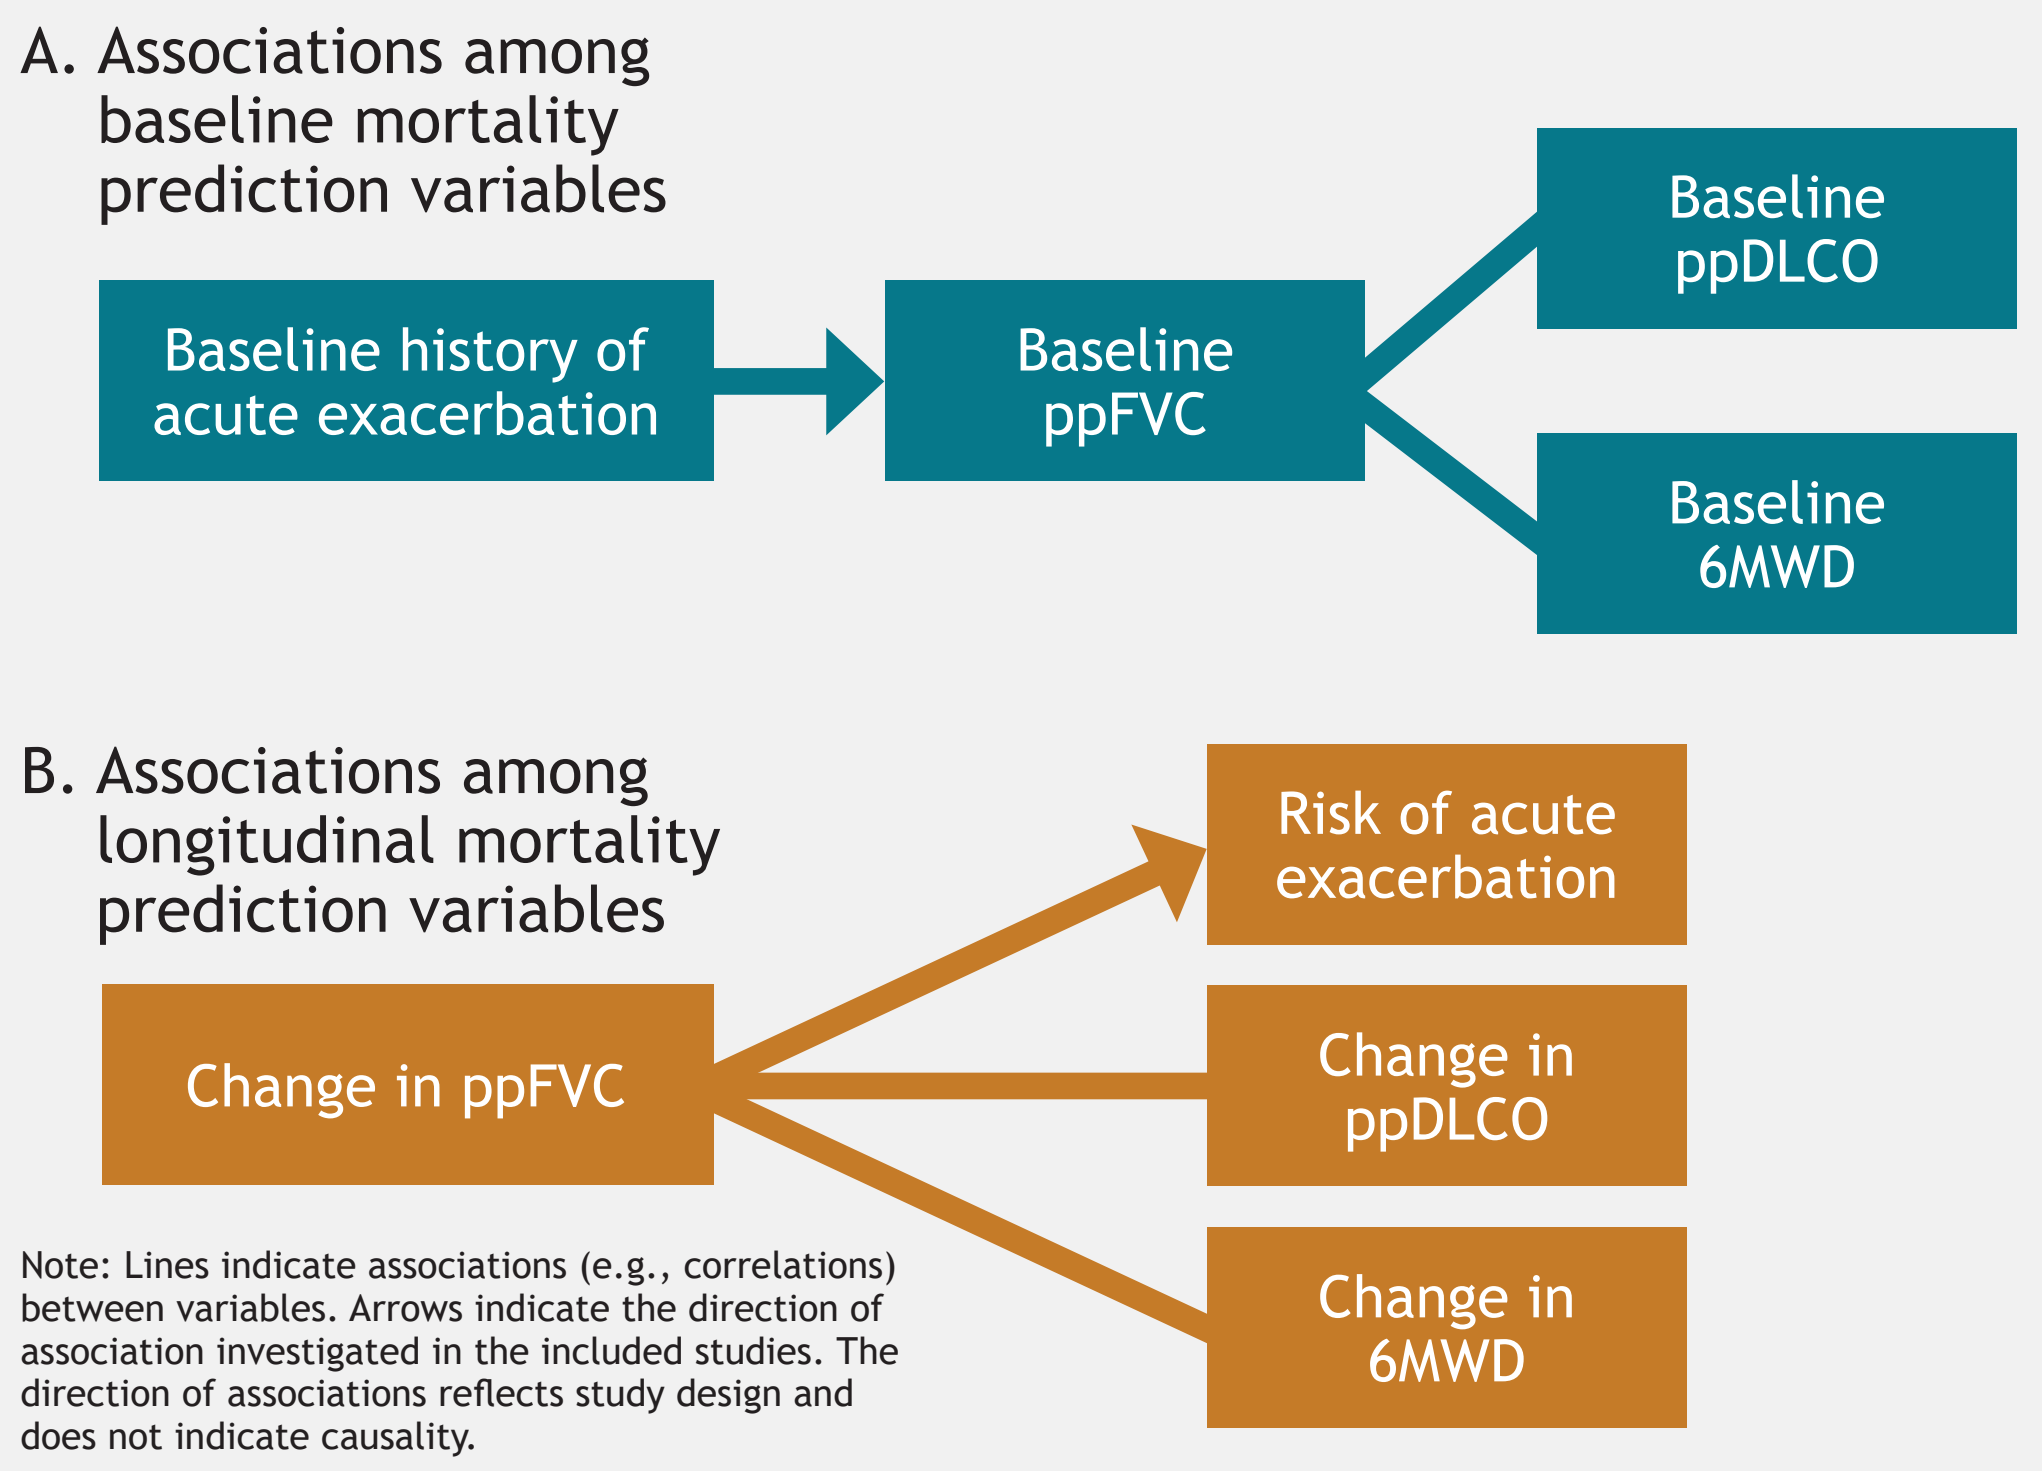
A. Associations among baseline mortality prediction variables
Baseline history of acute exacerbation
Baseline ppFVC
Baseline ppDLCO
Baseline 6MWD
B. Associations among longitudinal mortality prediction variables
Change in ppFVC
Risk of acute exacerbation
Change in ppDLCO
Change in 6MWD

Note: Lines indicate associations (e.g., correlations) between variables. Arrows indicate the direction of association investigated in the included studies. The direction of associations reflects study design and does not indicate causality.

Table 2. Key implications for implementing mortality prediction models in CE analyses

Item	Implication
The disease progression for IPF is heterogeneous	Capturing variations in changes in ppFVC, ppDLCO, and 6MWD and in risk of acute exacerbation over time is important when modeling the trajectories of these variables. It is also important for future CE analyses to consider the interconnectedness of lung function variables over time and the impact they can have on mortality risk
Mortality prediction models are designed for use at the patient level	Mortality prediction models can handle the heterogeneity of IPF disease progression that is observed at the patient level and can be considered for use in future CE analyses
Short-term mortality risks are estimated from the mortality prediction models (i.e., 1- to 3-year timeframe)	Validation may be needed to evaluate whether prediction models are designed for repeat usage across cycles in a CE model
Data were limited to inform potential relationships between model variables (e.g., baseline variables and longitudinal variables)	Future studies may provide evidence of additional relationships between model variables for inclusion in future CE analyses

Conclusions

- This targeted literature review identified evidence on IPF disease progression that supports clinically relevant mortality prediction approaches for future CE analyses.
- The heterogeneous course of IPF progression, observed correlations among measures of decline, and patient-level mortality risk prediction models suggest that a patient-level simulation modeling approach may be appropriate for assessing CE for future IPF treatments.

References

1. Mooney J, et al. J Manag Care Spec Pharm. 2021;27(12):1724-33.
2. Maher TM, et al. Respir Res. 2021;22(1):197.
3. Wu X, et al. Front Pharmacol. 2022;13:878764.
4. Caminati A, et al. Intern Emerg Med. 2021;16(7):1793-802.
5. Kim HJ, et al. Respir Med. 2015;109(6):661-70.
6. Ramos MCP, et al. Value in Health. 2023;26(12):S382.
7. Ley B, et al. Ann Intern Med. 2012;156(10):684-91.
8. Ley B, et al. Eur Respir J. 2015;45(5):1374-81.
9. du Bois RM, et al. Eur Respir J. 2014;43(5):1421-9.
10. du Bois RM, et al. Am J Respir Crit Care Med. 2011;184(12):1382-9.
11. Song JW, et al. Eur Respir J. 2011;37(2):356-63.
12. Khor YH, et al. Eur Respir Rev. 2020;29(157).
13. Lee EG, et al. PLoS One. 2021;16(12):e0261684.
14. Reichmann WM, et al. BMC Pulm Med. 2015;15:167.
15. Akoglu H. Turk J Emerg Med. 2018;18(3):91-3.
16. Neely ML, et al. Respir Res. 2023;24(1):209.
17. Durheim MT, et al. Respirology. 2021;26(10):982-8.
18. Collard HR, et al. Respir Res. 2013;14(1):73.
19. Idiopathic Pulmonary Fibrosis Clinical Research Network Investigators, et al. N Engl J Med. 2010;363(7):620-8.
20. Raghu G, et al. N Engl J Med. 2004;350(2):125-33.
21. King TE, Jr., et al. Lancet. 2009;374(9685):222-8.

Presented at ISPOR 2024; 5-8 May 2024; Atlanta, Georgia, United States

Contact information: Olivia Dong (odong@rti.org)

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.