PCR209

Joint modeling of progression-free survival (PFS) and patient-reported symptoms among relapsed/refractory multiple myeloma (RRMM) patients

Knop S[1], Einsele H[2], Dhanda D[3], Marshall T[3], Eliason L[3], McLoone D[4], Caisip C[4], Chen J[4], Boehm D[5], Dhamane A[3], Ramasamy K[6], Cope S[4], Towle K[4]

[1]Nuremberg General Hospital, Dept. of Hematology and Oncology and Paracelsus Medical School, Nuremberg, Germany; [2]Department of Internal Medicine II of the University Hospital, Würzburg, Germany; [3]Bristol Myers Squibb, Lawrenceville, NJ, USA; [4]PRECISIONheor, Vancouver, BC, Canada; [5]Bristol Myers Squibb, Munich, Germany; [6]Oxford University & Oxford University Hospitals NHS Trust, Oxford, Oxfordshire, UK

Introduction

Background/motivation

- Increasing emphasis on the patient experience has led to an interest in considering patient-reported outcomes in combination with clinical outcomes (e.g., survival) when evaluating the effectiveness of new interventions
- For example, while progression-free survival (PFS) is a common primary endpoint in oncology studies given its established criteria based on laboratory diagnostics and imaging procedures, it has been critiqued for disregarding changes in patients' perceived health-related quality of life (HRQoL)[1-3]
- Patient-reported symptom burden is an important component for understanding the patient perspective and disease experience, which has led to interest in symptom-relevant disease progression (i.e., 'symptomatic progression')[1-3]
- The Institute for Quality and Efficiency in Health Care (IQWiG) recommends considering mortality, morbidity, HRQoL, and safety when evaluating the added benefit of an intervention[4]
 - 'Symptom-accompanied progression' was previously presented in a dossier to IQWiG for the benefit assessment of daratumumab + pomalidomide + dexamethasone for relapsed/refractory multiple myeloma (RRMM)[5]
 - While IQWiG reported that this concept was relevant to patients, it was noted that additional research was needed to assess the association between disease progression and symptoms in RRMM, including clear selection of relevant symptoms and transparent methods to assess the association

Case example

- Clinical trials usually evaluate outcomes as separate, independent analyses, typically using Cox proportional hazards models for time-to-event outcomes (i.e., PFS) and linear mixed-effect models for longitudinal data with repeated measures over time (i.e., HRQoL)
- Alternatively, joint models can simultaneously analyze time-to-event outcomes (i.e., PFS) and longitudinal outcomes (i.e., HRQoL), allowing for the impact of symptoms/HRQoL on PFS to be quantified
- Joint models of PFS and patient-reported symptoms were performed involving Bristol Myers Squibb (BMS) RRMM clinical trials (randomized clinical trials [RCTs] and single-arm trials) with sufficient follow-up (i.e., completed primary analysis) and with data availability for both PFS and patient-reported symptoms (as measured by European Organisation for Research and Treatment of Cancer [EORTC] Quality of Life questionnaires)
- Six clinical trials met this eligibility criteria and for which BMS provided individual patient data (IPD):
 - OPTIMISMM (RCT) evaluating PVd vs Vd[6]
 - ELOQUENT-2 (RCT) evaluating Ld vs ELd[7]
 - KarMMa-3 (RCT) evaluating ide-cel vs SoC[8]
 - MM-003 (RCT) evaluating HiDex vs Pom + LoDex[9]
 - KarMMa (single-arm) evaluating ide-cel[10]
 - CC-220-MM-001 (single-arm) evaluating iberdomide + Dex[11]
- These trials represented various treatment classes, including immunomodulatory agents, proteasome inhibitors, anti-CD38 antibodies, chimeric antigen receptor (CAR) T-cell therapy, and cereblon E3 ligase modulators (CELMoDs)

Abbreviations: PVd, pomalidomide + bortezomib + dexamethasone; Vd, bortezomib + dexamethasone; Ld, lenalidomide + dexamethasone; Eld, elotuzumab + lenalidomide + dexamethasone; ide-cel, idecabtagene vicleucel; SoC, standard of care; HiDex, high-dose dexamethasone; Pom, pomalidomide; LoDex, low-dose dexamethasone.

Objective

Data from six RRMM clinical trials with different underlying patient populations (i.e., less severe to more severe patient populations) were analyzed using trial-specific joint models to assess the relationship between PFS and patient-reported symptoms (as measured by validated HRQoL questionnaires) in clinical trials evaluating RRMM treatments

Methods

Identification of patient-relevant symptoms

- A pre-defined literature review of patient preferences and experiences in RRMM was performed, including patient preference exploration or qualitative studies exploring patient symptoms and experiences in RRMM
- A total of 18 patient preference and qualitative studies were identified, with 12 studies reporting information on self-reported symptoms of concern
 - Pain (N=12 studies [100%]) and fatigue (N=9 studies [75%]) were the most commonly reported symptoms
- EORTC instruments were selected as they have been validated, accepted by health technology assessment agencies, and are specific to symptoms for cancer (core quality of life questionnaire [QLQ-C30]) and multiple myeloma patients (quality of life multiple myeloma questionnaire [QLQ-MY20])
 - Based on the literature review, the following domains were selected for this analysis: 1) QLQ-C30 pain, 2) QLQ-MY20 disease symptoms (assesses pain in different locations), and 3) QLQ-C30 fatigue
 - During a clinical consultation, experts in multiple myeloma agreed that pain and fatigue were the most relevant patient-reported symptoms related to disease progression rather than treatment-related effects

Summary of evidence base

- The clinical trial evidence base was summarized, including trial characteristics, intervention characteristics, patient characteristics, and PFS/HRQoL timing of assessments (**Table 1**; trials ordered according to increasing prior exposure criteria)

Table 1. Summary of trial characteristics

Characteristic	OPTIMISMM	ELOQUENT-2	KarMMa-3	MM-003	KarMMa	CC-220-MM-001
NCT	NCT01734928	NCT01239797	NCT03651128	NCT01311687	NCT03361748	NCT02773030
Trial design	Phase III RCT (open-label)	Phase III RCT (open-label)	Phase III RCT (open-label)	Phase III RCT (open-label)	Phase II single-arm (open-label)	Phase Ib/IIa multicohort single-arm (open-label)
Interventions (full trial N)	PVd (N=281) or Vd (N=278)	ELd (N=321) or Ld (N=325)	Ide-cel (N=254) or SoC (N=132)	POM + LoDex (N=302) or HiDex (N=153)	Ide-cel (N=128)	Iber + Dex (N=107)
Actual study start date	January 7, 2013	June 20, 2011	April 16, 2019	March 11, 2011	December 13, 2017	October 14, 2016
Age	≥18	≥18	≥18	≥18	≥18	≥18
Performance score	ECOG 0-2	ECOG 0-2	ECOG 0-1	ECOG 0-2	ECOG 0-1	ECOG 0-2
Exposure to prior regimens	Previously received 1-3 lines of therapy, including at least 2 consecutive cycles of a lenalidomide-containing regimen	Previously received 1-3 lines of therapy	2-4 lines of therapy, including an immunomodulatory agent, a PI, and an anti-CD38 monoclonal antibody	Previously received ≥2 lines of therapy, including at least 2 consecutive cycles of a lenalidomide and bortezomib-containing regimen	≥3 lines of therapy, including an immunomodulatory agent, a PI, and an anti-CD38 monoclonal antibody	≥3 lines of therapy, including an immunomodulatory agent, a PI, and an anti-CD38 monoclonal antibody
Refractoriness to prior regimens	Refractory to previous treatment Exclusion: PD during therapy or within 60 days of the last dose of a bortezomib-containing therapy	Refractory to previous treatment Exclusion: refractory to lenalidomide	Refractory to last line of therapy	Refractory to previous treatment	Refractory to last line of therapy	Refractory to an immunomodulatory agent, a PI, a glucocorticoid, and an anti-CD38 antibody, and last line of therapy
Crossover	Not permitted	Not permitted	Crossover permitted	Crossover permitted	Not applicable (single-arm)	Not applicable (single-arm)
PFS criteria	FDA censoring (IRAC, IMWG)	FDA censoring (IRC, EBMT)	FDA censoring (IRC, IMWG)	EMA censoring (IA, IMWG)	FDA censoring (IRC, IMWG)	FDA censoring (IRAC, IMWG)
Timing of response evaluations	Every 21 days (+/- 3 days) and time of discontinuation	Every 4 weeks (+/- 1 week)	Every 28 days (+/- 3 days) for months 1-25, every 3 months for month >25	Every 28 days (+/- 3 days)	Monthly evaluations (+/- 3 days) for the first 6 months and then every 3 months (+/- 14 days)	Every 28-days (+/- 2 days) and end of treatment
HRQoL Schedule of assessment	Day 1 of each 21-day treatment cycle (before treatment administration) and at the end-of-treatment visit	At baseline (prior to randomization), on Day 1 of each 4-week treatment cycle (28-day cycle), and at the end of treatment or study withdrawal	Screening, baseline, day 1, monthly between months 1 to 24, and every 3 months from 25 months	At baseline, on day 1 of each 28-day treatment cycle, and at treatment discontinuation	Screening, baseline, day 1, monthly between months 1 to 6, and every 3 months up to 24 months or study completion	Day 1 of each 28-day cycle 1 day 1 as baseline), as well as the end-of-treatment visit.

*Notes: pink font denotes differences across the evidence base. **Abbreviations**: Dex, dexamethasone; EBMT, European Society for Blood and Marrow Transplantation; ECOG, Eastern Cooperative Oncology Group; Eld, elotuzumab + lenalidomide + dexamethasone; EMA, European Medicines Agency; FDA, Food and Drug Administration; HiDex, high-dose dexamethasone; HRQoL, health-related quality of life; IA, investigator assessed; Iber, iberdomide; ide-cel, idecabtagene vicleucel; IMWG, International Myeloma Working Group; IRAC, Independent Response Adjudication Committee; IRC, Independent Response Committee; ITT, intention-to-treat; Ld, lenalidomide + dexamethasone; LoDex, low-dose dexamethasone; N, sample size; PI, proteosome inhibitor; PFS, progression-free survival; Pom, pomalidomide; PVd, pomalidomide + bortezomib + dexamethasone; RCT, randomized controlled trial; RRMM, relapsed/refractory multiple myeloma; SoC, standard of care; Vd, bortezomib + dexamethasone.*

Analyses

- Joint models were fit separately for each trial and each symptom (**Figure 1**)
- Each joint model involved two submodels: 1) a time-to-event model for PFS and 2) a longitudinal model for change from baseline in domain-specific HRQoL scores
- The two submodels were then linked using an association structure to provide insights on the relationship between PFS and patient-reported symptoms
- Model covariates were selected based on trial design/stratification factors, clinical input, and literature reviews of RRMM HRQoL prognostic factors

Figure 1. Summary of analyses

1 PFS submodel

Model: Cox proportional hazards model

Outputs: Coefficients (log HR) and HRs

Interpretation: positive coefficient (log PFS HR) values indicate an increase in hazard of a PFS event (i.e., HR>1)

2 Symptom submodel

Model: Linear mixed effect (change from baseline)

Outputs: Coefficients

Interpretation: positive coefficient values indicate that symptom worsened with an increase in the covariate

3 Joint model

Model: Current slope association structure

Outputs: Association parameter (log HR) and association HR

Interpretation: An association HR>1 indicates that an increase in the slope of change from baseline HRQoL score (worsening of symptom) corresponds to an increase in the hazard of a PFS event

Results

Joint models

- Deterioration in pain/fatigue was consistently associated with an increased hazard of PFS events (disease progression or death) based on the direction of the joint model association hazard ratios (HRs) (i.e. HR>1) across the trial-specific joint models (**Figure 2**)
- Joint model association HRs were statistically significant (p<0.05) for pain (OPTIMISMM, MM-003, CC-220-MM-001, KarMMa, KarMMa-3), disease symptoms (OPTIMISMM, MM-003, CC-220-MM-001, KarMMa-3), and fatigue (OPTIMISMM, ELOQUENT-2, MM-003, CC-220-MM-001, KarMMa-3)
- Joint model association HRs ranged from 1.00 (OPTIMISMM) to 1.32 (KarMMa-3) across all of the evaluated trials and symptoms
 - KarMMa-3 had the highest joint model association HR for QLQ-C30 pain (HR 1.30 [95% confidence interval (CI): 1.17, 1.44]), QLQ-MY20 disease symptoms (HR 1.32 [95% CI: 1.15, 1.52]), and QLQ-C30 fatigue (HR 1.26 [95% CI: 1.10, 1.45])
- Treatment effect PFS HRs from joint models (i.e., PFS modelled with symptoms) resulted in slight shifts away from the null effect with narrower 95% CIs in comparison to treatment effect PFS HRs from the PFS submodel (i.e., PFS modelled alone)

Figure 2. Forest plot of trial and symptom-specific joint model hazard ratios and 95% confidence intervals (summarizing association between progression-free survival and patient-reported symptoms)

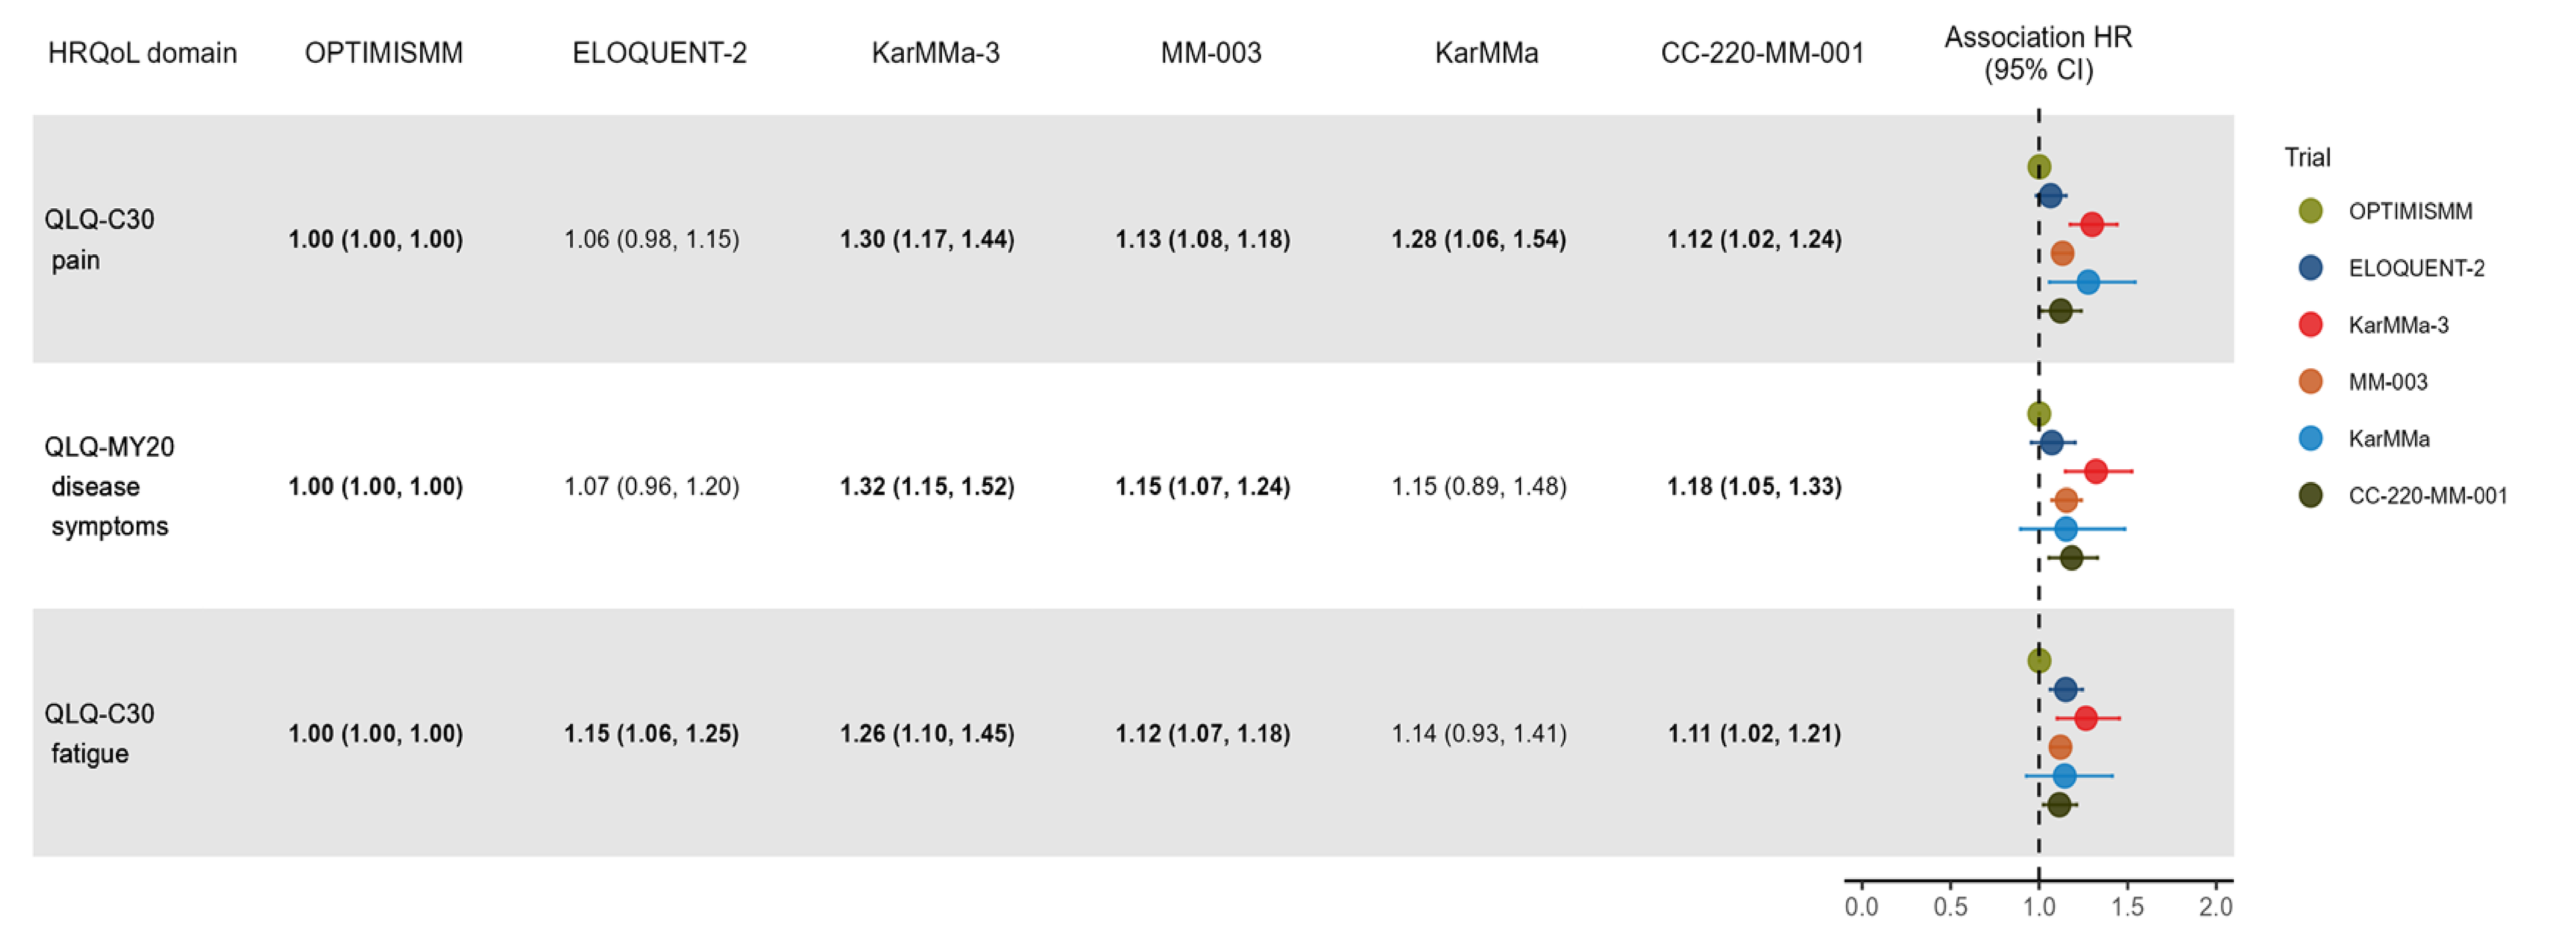
HRQoL domain
OPTIMISMM
ELOQUENT-2
KarMMa-3
MM-003
KarMMa
CC-220-MM-001
Association HR (95% CI)
QLQ-C30 pain
1.00 (1.00, 1.00)
1.06 (0.98, 1.15)
1.30 (1.17, 1.44)
1.13 (1.08, 1.18)
1.28 (1.06, 1.54)
1.12 (1.02, 1.24)
QLQ-MY20 disease symptoms
1.00 (1.00, 1.00)
1.07 (0.96, 1.20)
1.32 (1.15, 1.52)
1.15 (1.07, 1.24)
1.15 (0.89, 1.48)
1.18 (1.05, 1.33)
QLQ-C30 fatigue
1.00 (1.00, 1.00)
1.15 (1.06, 1.25)
1.26 (1.10, 1.45)
1.12 (1.07, 1.18)
1.14 (0.93, 1.41)
1.11 (1.02, 1.21)
0.0
0.5
1.0
1.5
2.0
Trial
OPTIMISMM
ELOQUENT-2
KarMMa-3
MM-003
KarMMa
CC-220-MM-001

*Notes: bolded values are statistically significant at the 0.05 significance level. Analyses specific to trial-specific HRQoL-evaluable populations and model covariates. **Abbreviations**: CI, confidence interval; HR, hazard ratio; HRQoL, health-related quality of life; PFS, progression-free survival.*

Conclusions

- This study demonstrated that deteriorations in pain and fatigue were consistently associated with an increased hazard of disease progression or death events across numerous RRMM clinical trials
 - These associations were statistically significant in trial-specific models evaluating heterogenous RRMM patient populations with varying disease severity
- This analysis adds to the literature by demonstrating an approach to 1) identify and select patient-relevant symptoms based on a pre-defined literature review, 2) consult disease-specific clinical experts to assist with model selection, and 3) utilize joint models to assess PFS and symptoms simultaneously.
- Joint models were performed separately for six RRMM clinical trials. While trends in the magnitude of the association effect HRs from trial-specific joint models may provide clinical insights, it is important to note that the joint model association HRs are conditional on the inclusion of different covariates in the trial-specific models
- The timing and amount of patient-reported symptom follow-up is an important consideration, as longer time periods between evaluations and limited post-progression patient-reported symptom data may limit the ability to estimate the association between symptoms and disease progression
- Overall, findings from this study suggest that PFS is associated with disease-relevant symptom worsening and is therefore meaningful for patients with RRMM

References

1. He J, Duenas A, Collacott H, Lam A, Gries KS, Carson R, Potthoff D, Trevor N, Tervonen T. The Patient-Patient-Centered Outcomes Research. 2021 Sep;14:613-23.Janssens R, Lang T, Vallejo A, et al. Frontiers in Medicine. 2021:930
2. Janssens R, Lang T, Vallejo A, Galinsky J, Plate A, Morgan K, Cabezudo E, Silvennoinen R, Coriu D, Badelita S, Irimia R. Frontiers in medicine. 2021 Jul 6;8:686165.
3. Quinn B, Ludwig H, Bailey A, Khela K, Marongiu A, Carlson KB, Rider A, Seesaghur A.. Pain management. 2021 May;12(1):59-74.
4. IQWiG. General Methods. Version 6.0 of 5 November 2020. 2020.
5. G-BA, Benefit assessment according to Section 35a SGB V. 2022.
6. Richardson PG, Oriol A, Beksac M, Liberati AM, Galli M, Schjesvold F, Lindsay J, Weisel K, White D, Facon T, San Miguel J. The Lancet Oncology. 2019 Jun 1;20(6):781-94.
7. Lonial S, Dimopoulos M, Palumbo A, White D, Grosicki S, Spicka I, Walter-Croneck A, Moreau P, Mateos MV, Magen H, Belch A.. New England Journal of Medicine. 2015 Aug 13;373(7):621-31.
8. Rodriguez-Otero P, Ailawadhi S, Arnulf B, Patel K, Cavo M, Nooka AK, Manier S, Callander N, Costa LJ, Vij R, Bahlis NJ. New England Journal of Medicine. 2023 Mar 16;388(11):1002-14.
9. San Miguel J, Weisel K, Moreau P, Lacy M, Song K, Delforge M, Karlin L, Goldschmidt H, Banos A, Oriol A, Alegre A. The lancet oncology. 2013 Oct 1;14(11):1055-66.
10. Munshi NC, Anderson Jr LD, Shah N, Madduri D, Berdeja J, Lonial S, Raje N, Lin Y, Siegel D, Oriol A, Moreau P. New England Journal of Medicine. 2021 Feb 25;384(8):705-16.
11. Lonial S, Popat R, Hulin C, Jagannath S, Oriol A, Richardson PG, Facon T, Weisel K, Larsen JT, Minnema MC, Abdallah AO.. The Lancet Haematology. 2022 Nov 1;9(11):e822-32.

Presented at ISPOR 2024; May 5–8, 2024; Atlanta, GA

Email: shannon.cope@precisionvh.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.