HSD113

Treatment Patterns, Comorbidities, Healthcare Resource Utilization and Related Costs of Dermatomyositis and Polymyositis in the United States: A Cohort Study in the Merative™ MarketScan® Database

Caroline Foch[1], Sabrina de Souza[1], Luisa Henkel[2], **Xiaoxue Chen**[3], Deborah Denis[3], Jeffrey M. Muir[4]

[1]the healthcare business of Merck KGaA, Darmstadt, Germany; [2]Merck Healthcare Germany GmbH, Weiterstadt, Germany, an affiliate of Merck KGaA, Darmstadt, Germany; [3]EMD Serono, Rockland, MA, USA; [4]Cytel Inc., Toronto, Canada

GET POSTER PDF

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

CONCLUSIONS

Glucocorticoids were the predominate treatment for DM and PM in the United States

Overall costs, driven largely by outpatient charges, were higher than previously reported

These data provide an important update on the treatment landscape and economic burden of DM and PM in the United States

INTRODUCTION

Idiopathic inflammatory myopathies (IIM)

- IIM are a heterogeneous group of disorders, classified into DM, PM and inclusion body myositis, which typically feature **chronic inflammation of skeletal muscles** that leads to muscle weakness, with other organs also frequently affected[1]

Management of IIM

- First-line treatment is **high-dose glucocorticoids**, often combined with at least one **immunosuppressive agent** at early stages to better control disease activity and enable steroid tapering[2]
- IVIg and biologics may be used in severe cases or as escalation therapy[2]

Costs of DM and PM in the United States

- Costs of DM have been estimated to be 5.4x higher than that for matched controls at **$39,508/patient** based on health insurance data (2014–2020)[3]
- A systematic literature review (2011–2021) found that mean hospital costs to payers for patients with DM/PM were **$55,000/patient** – which was $13,000 higher than inpatients without DM/PM – with total DM annual hospitalization costs of **$168 million**[4]

OBJECTIVE

Describe the treatment patterns and economic burden of DM and PM using data from a large, commercial insurance database in the United States

METHODS

Observational cohort study using data from the Merative™ MarketScan® Commericial Database and the Merative™ MarketScan® Medicare Database

Study design

Cohort entry (index date)	First DM or PM diagnosis between January 1, 2018 and June 30, 2021 (prevalent cases)
	Data collected from cohort entry to the end of follow-up
End of follow-up	Earliest of loss of insurance coverage, end of data availability, or end of study period (December 31, 2021)

Inclusion criteria

- Age ≥18 years at cohort entry
- Diagnosis of DM (ICD-10: M33.1, M33.9) or PM (ICD-10: M33.2), defined as ≥1 inpatient diagnosis or ≥2 outpatient encounters (minimum 30 days and maximum 365 days apart)

- The frequency and rate of dispensed prescriptions, comorbidities, annualized health service utilization, and annualized direct healthcare costs were described descriptively
- Patients with diagnoses of DM and PM were included in both cohorts

RESULTS

Patient demographics and characteristics

- The study included 3693 patients with DM and 2841 with PM; 388 patients were included in both cohorts
- The median (p25, p75) age was 53 (42, 60) years in the DM Cohort and 55 (47, 62) years in the PM Cohort, and 77% and 66% of patients were female, respectively
- The median duration of follow-up was 1.39 years in both cohorts

Comorbidities

	DM Cohort, n (%) n = 3693	PM Cohort, n (%) n = 2841
Hypertension	1633 (44)	1659 (58)
Hyperlipidemia	1448 (39)	1416 (50)
Respiratory infection	1433 (39)	1087 (38)
Vitamin D deficiency	1161 (31)	934 (33)
Obesity	1049 (28)	984 (35)
Gastro-esophageal reflux disease	1047 (28)	913 (32)

Comorbidities that occurred in over 30% of patients in either cohort shown

Treatments for DM and PM

- 78% and 79% of patients in the DM and PM cohorts, respectively, received ≥1 prescription for glucocorticoids (primarily systemic)
- Antimalarial use was higher in the DM Cohort (36%) than the PM Cohort (16%), and immunosuppressants were used in over half of patients in each cohort
- Use of IVIg and biologics (primarily rituximab) was similar between cohorts

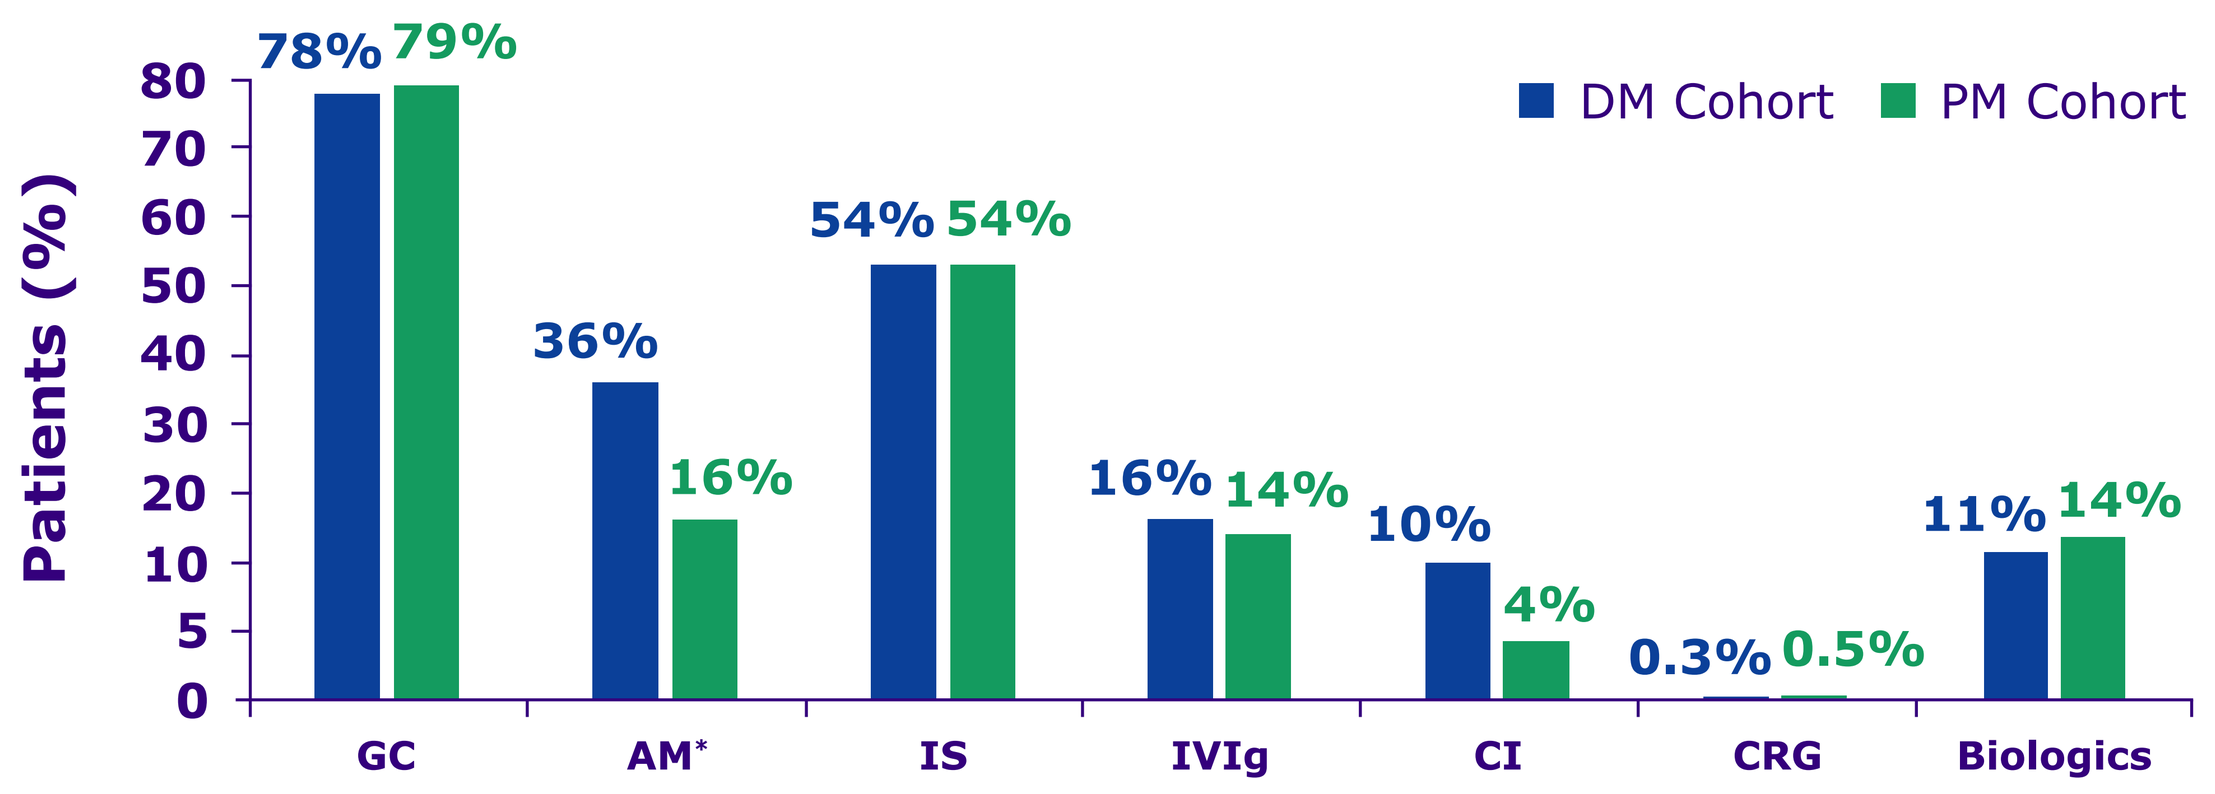

DM Cohort
PM Cohort
Patients (%)
78%
79%
36%
16%
54%
54%
16%
14%
10%
4%
0.3%
0.5%
11%
14%
GC
AM*
IS
IVIg
CI
CRG
Biologics

* Hydroxychloroquine and chloroquine

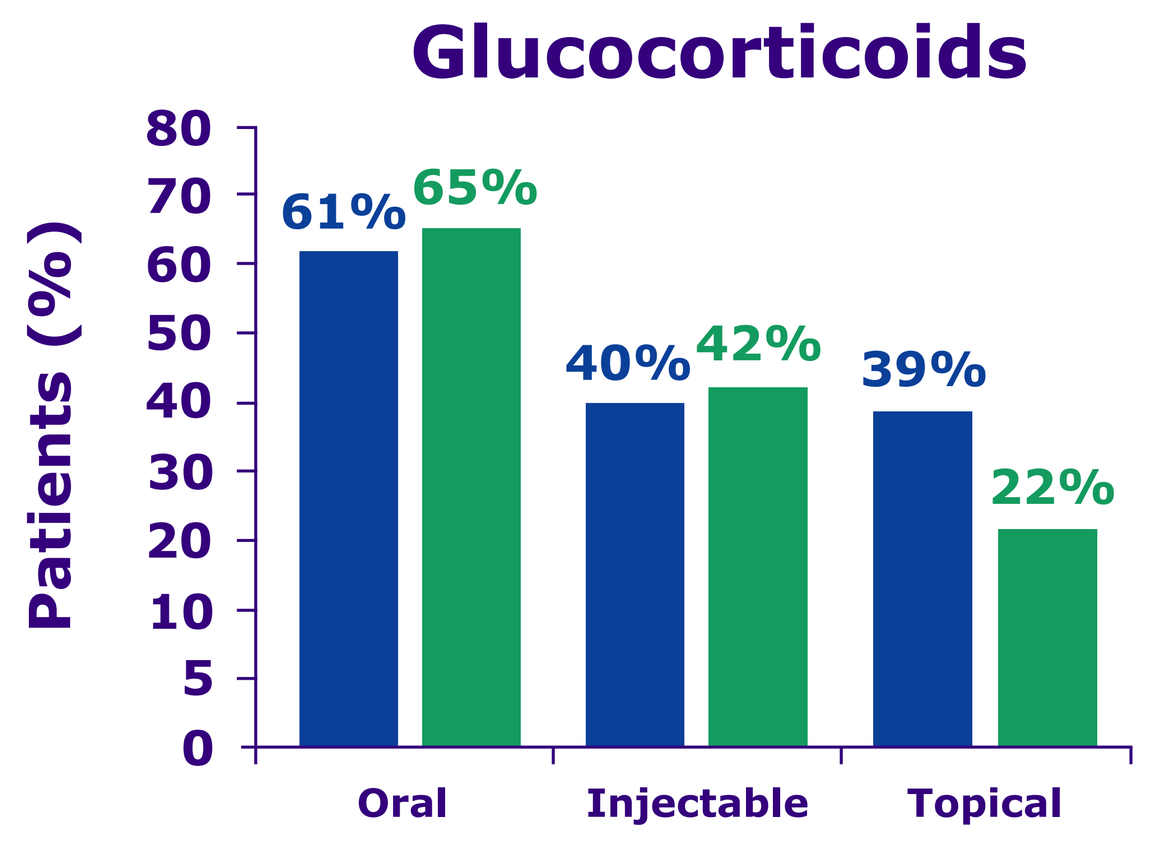

Glucocorticoids
Patients (%)
61%
65%
40%
42%
39%
22%
Oral
Injectable
Topical

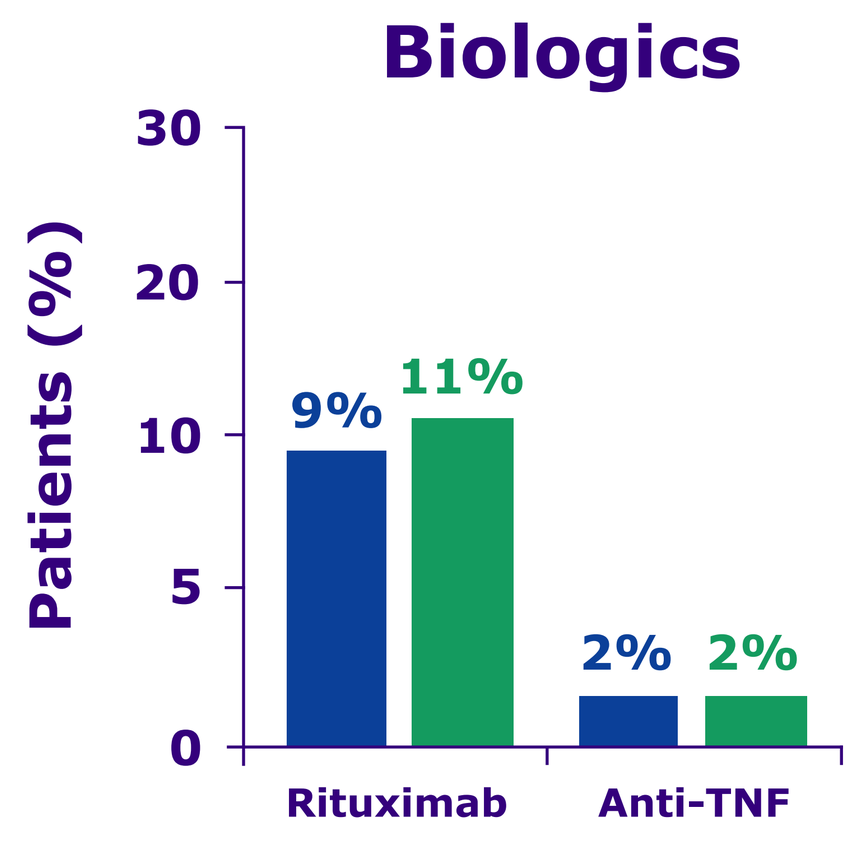

Biologics
Patients (%)
9%
11%
2%
2%
Rituximab
Anti-TNF

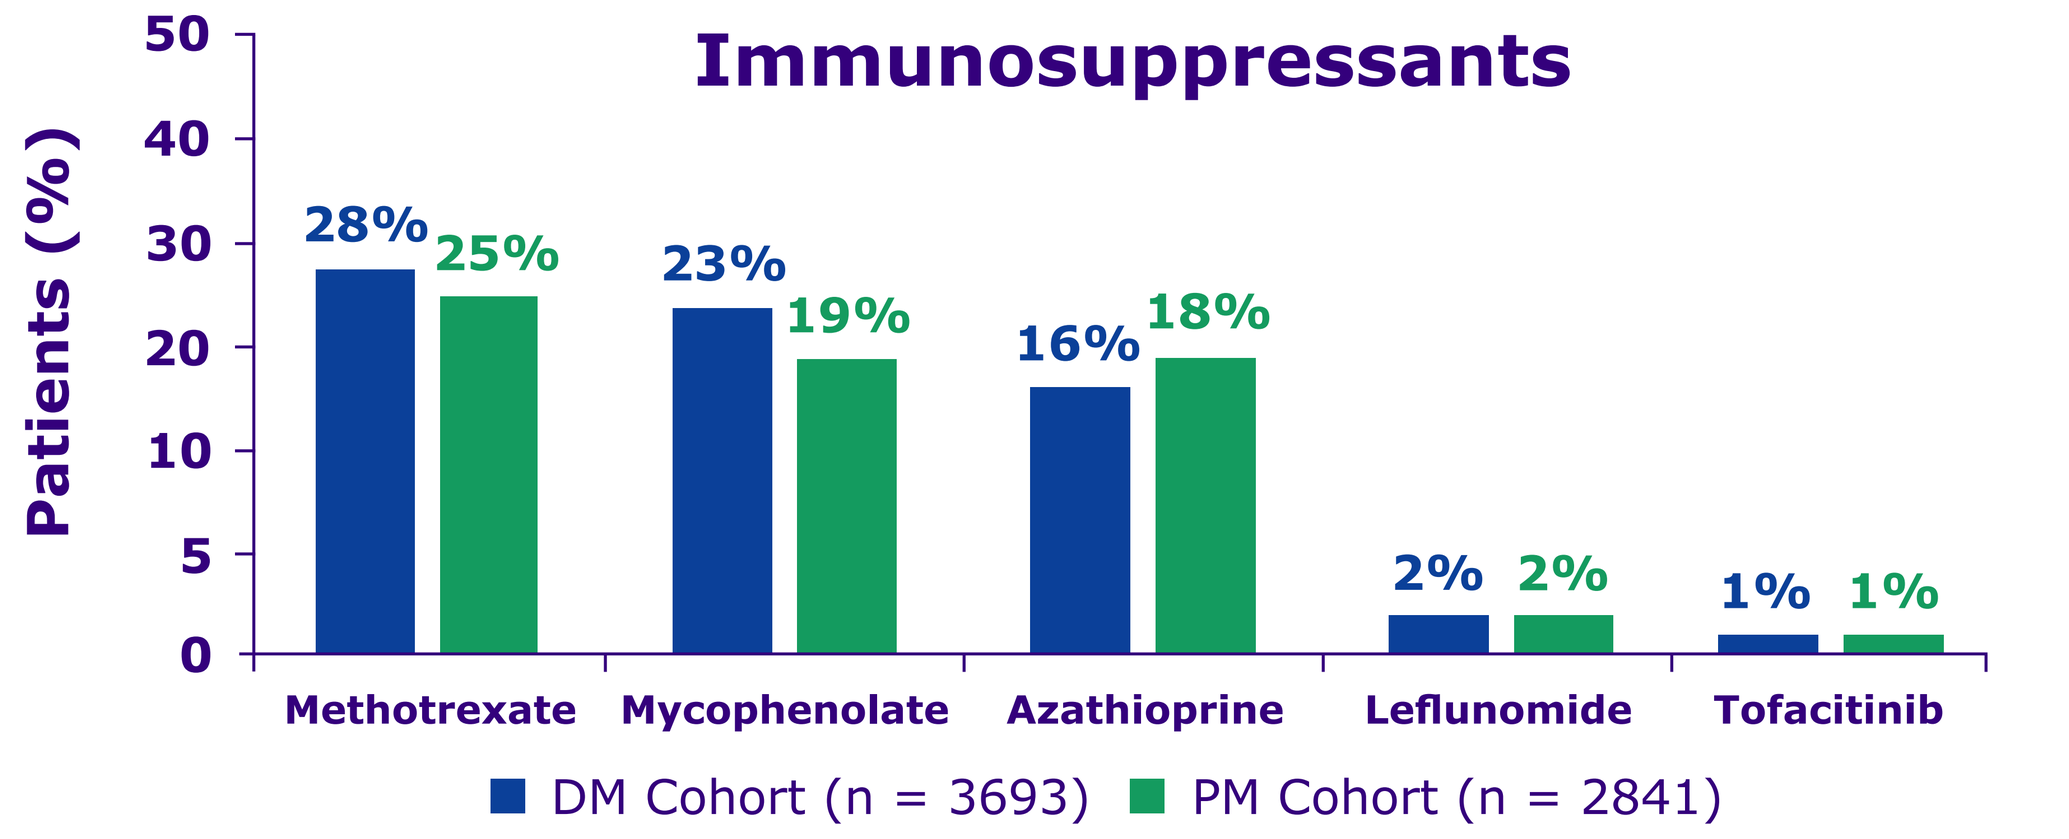

Immunosuppressants
Patients (%)
28%
25%
23%
19%
16%
18%
2%
2%
1%
1%
Methotrexate
Mycophenolate
Azathioprine
Leflunomide
Tofacitinib
DM Cohort (n = 3693)
PM Cohort (n = 2841)

Immunosuppressants apremilast, baricitinib, cyclophosphamide, cyclosporine, and everolimus, and biologics abatacept and tocilizumab were prescribed for <1% of patients in each cohort

Healthcare utilization and associated costs

- 20–30% of patients were admitted to hospital, for a median duration of approximately 5 days, and 30–40% visited the emergency department

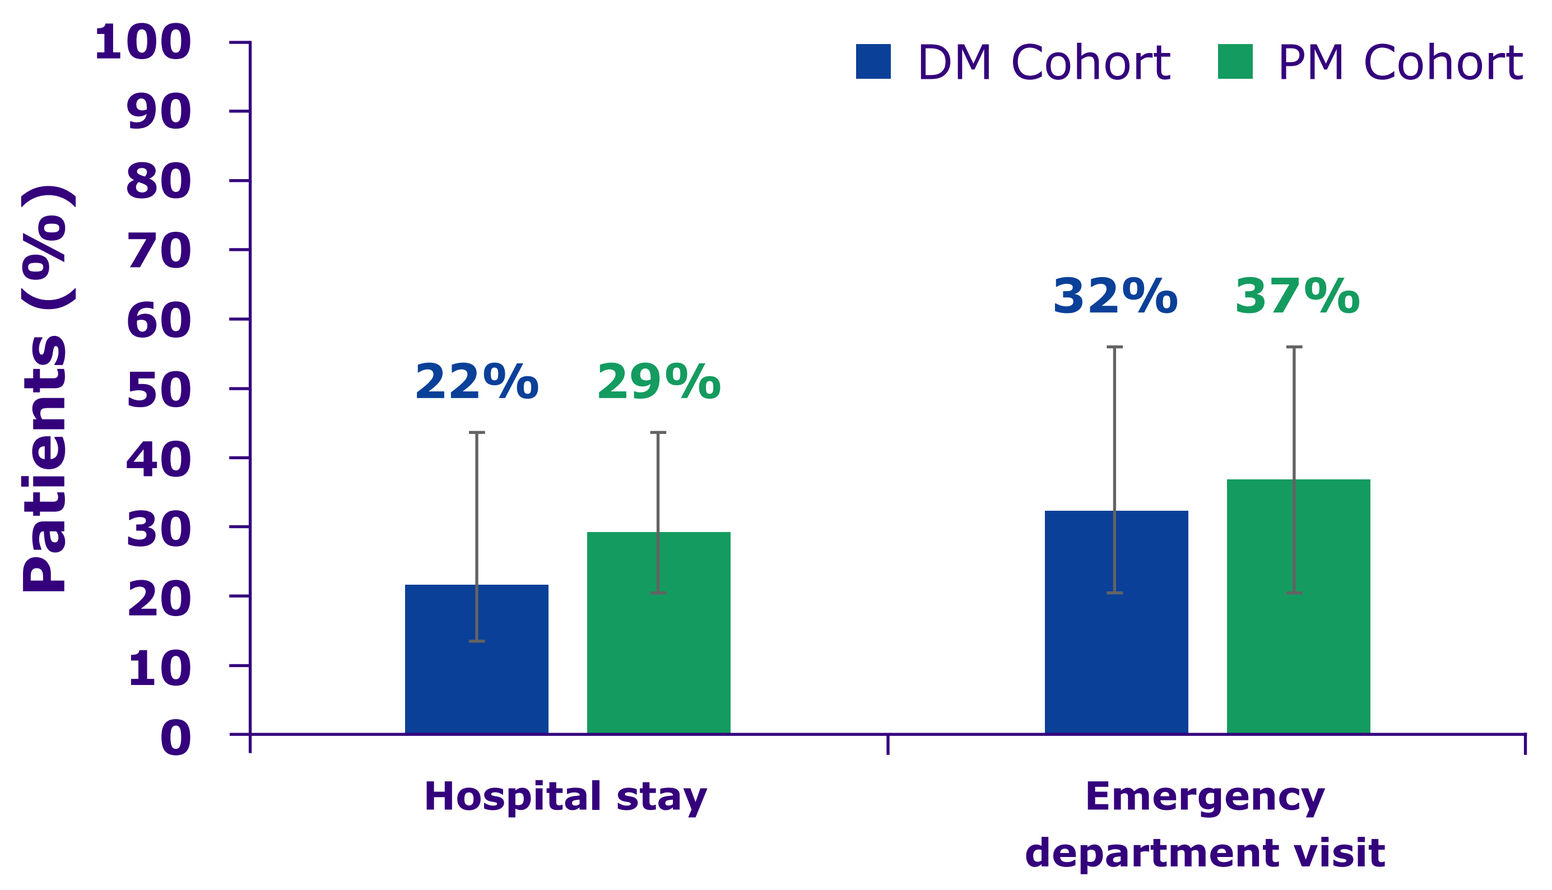

DM Cohort
PM Cohort
Patients (%)
22%
29%
32%
37%
Hospital stay
Emergency department visit

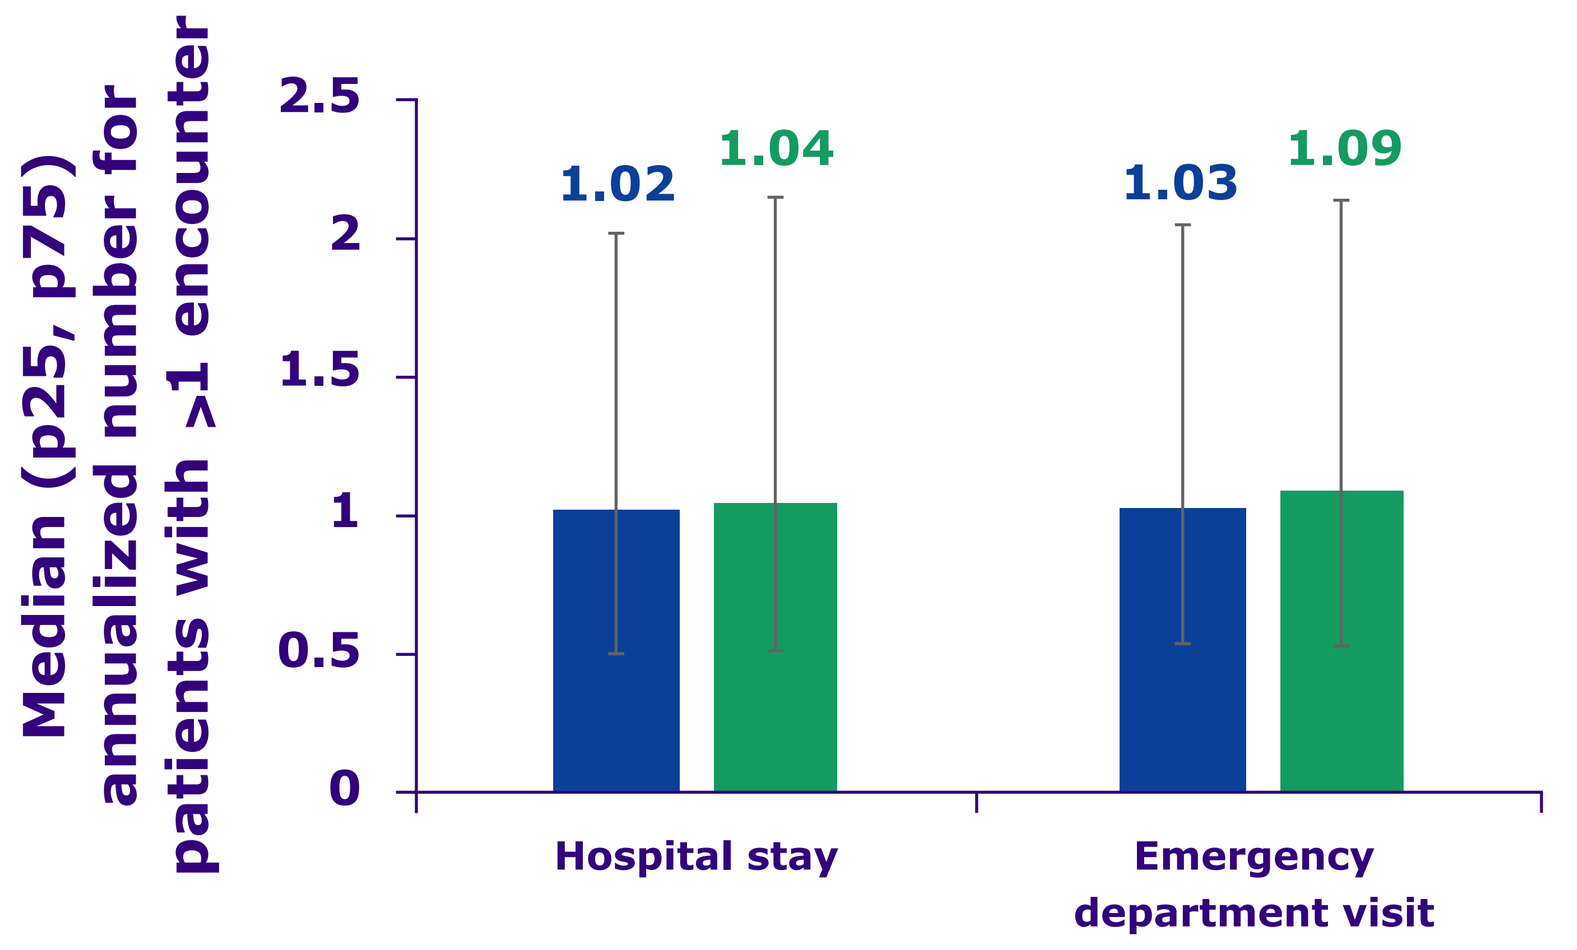

Median (p25, p75) annualized number for patients with >1 encounter
1.02
1.04
1.03
1.09
Hospital stay
Emergency department visit

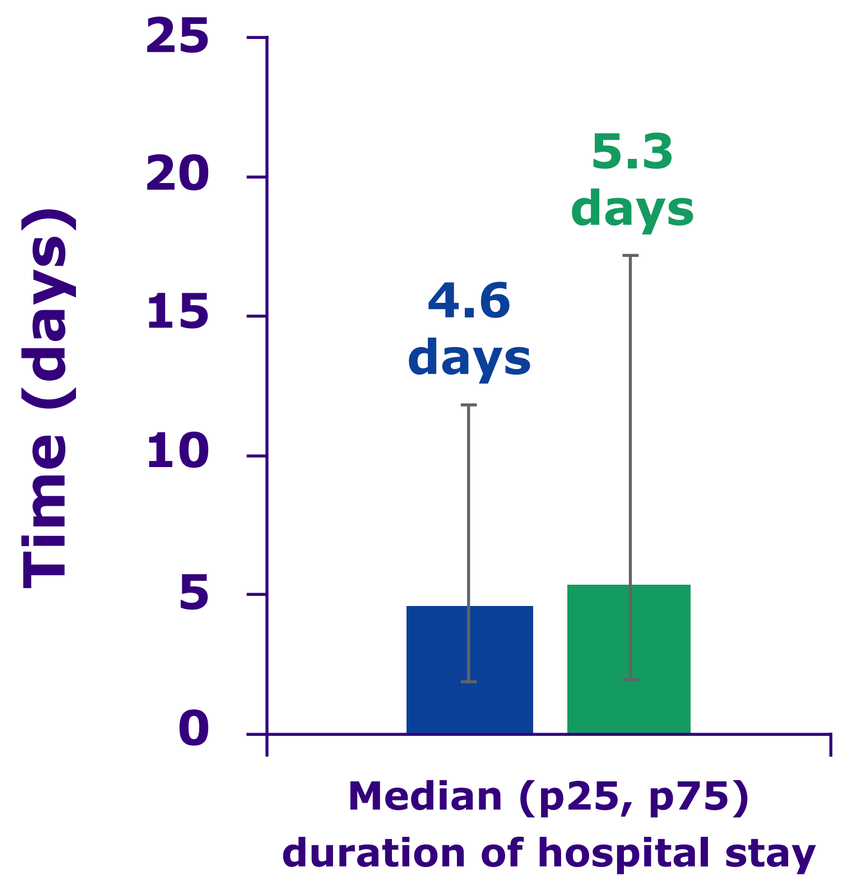

Time (days)
4.6 days
5.3 days
Median (p25, p75) duration of hospital stay

- Mean annualized direct costs were higher in the PM Cohort than the DM Cohort due to greater inpatient costs
 - Direct costs were driven largely by outpatient claims in both cohorts

DM Cohort

Mean (SD) total $73,673 ($187,688)

[median; p25, p75: $17,290; $6191, $67,536]

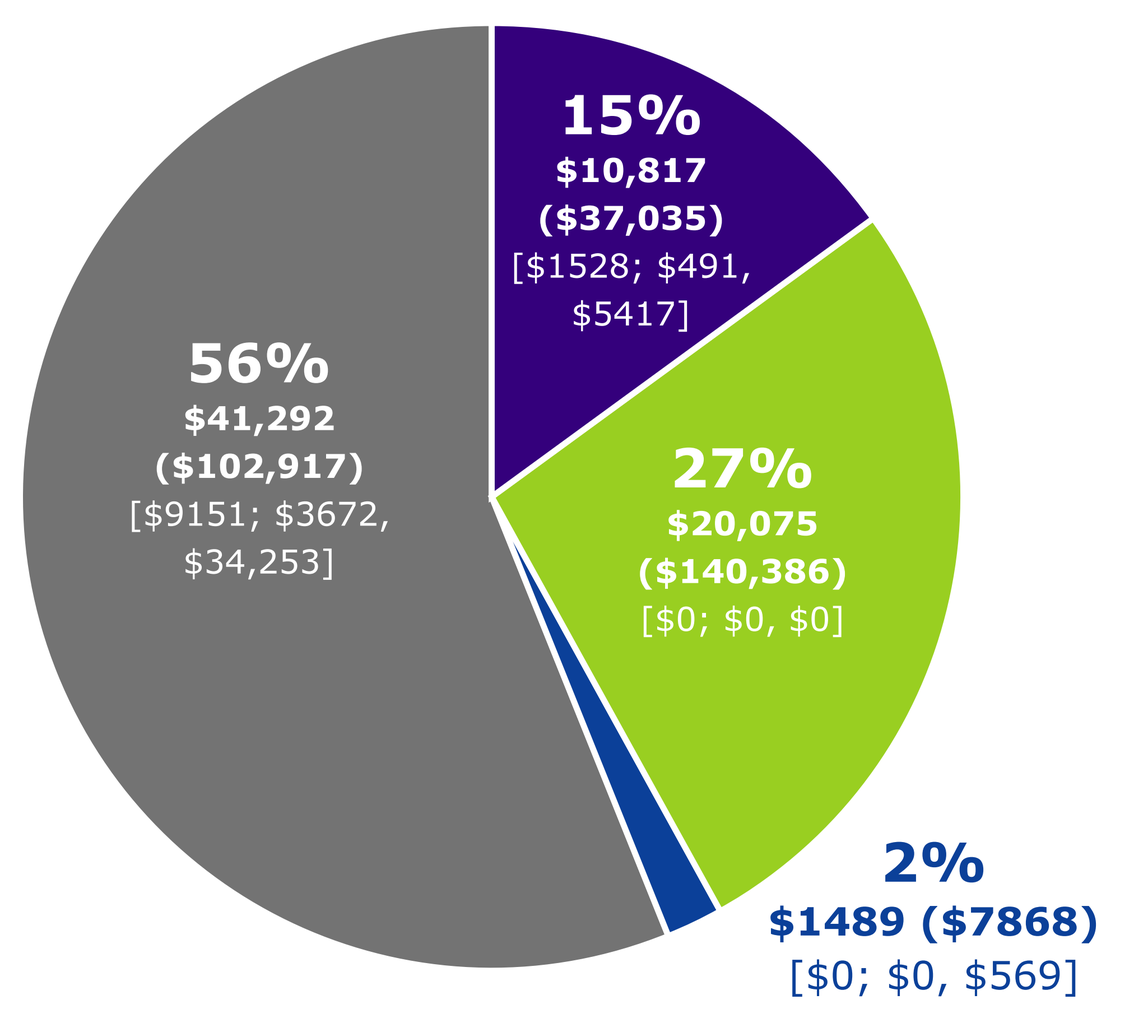

15%
$10,817
($37,035)
[$1528; $491, $5417]
27%
$20,075
($140,386)
[$0; $0, $0]
2%
$1489 ($7868)
[$0; $0, $569]
56%
$41,292
($102,917)
[$9151; $3672, $34,253]

PM Cohort

Mean (SD) total $92,380 ($304,170)

[median; p25, p75: $24,261; $7316, $78,895]

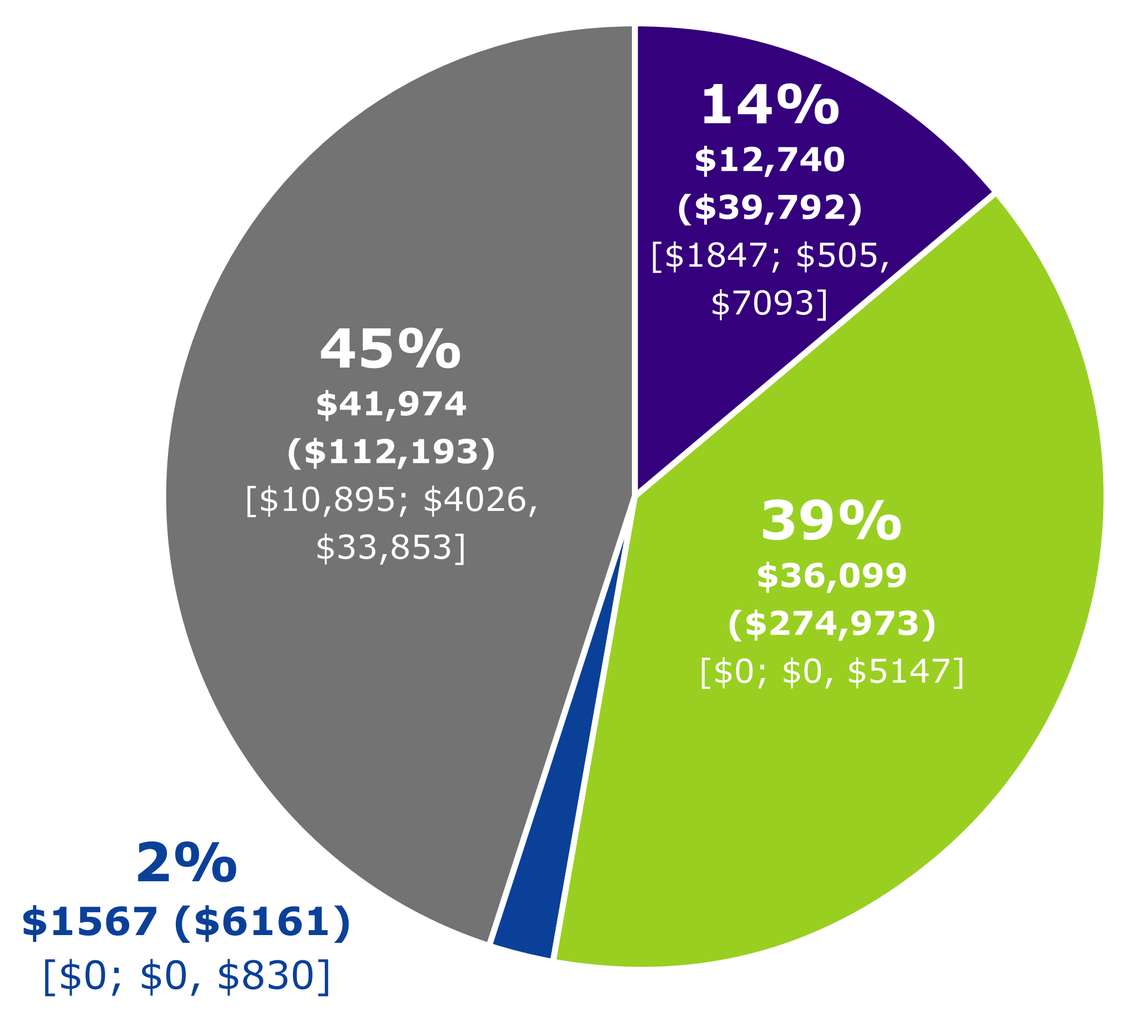

14%
$12,740
($39,792)
[$1847; $505, $7093]
39%
$36,099
($274,973)
[$0; $0, $5147]
2%
$1567 ($6161)
[$0; $0, $830]
45%
$41,974
($112,193)
[$10,895; $4026, $33,853]

■ Pharmacy ■ Inpatient ■ Emergency department ■ Outpatient

ABBREVIATIONS

AM, antimalarials; **CI**, calcineurin inhibitors; **CRG**, corticotropin repository gel; **DM**, dermatomyositis; **GC**, glucocorticoids; **ICD**, International Classification of Diseases; **IS**, immunosuppressants; **IIM**, idiopathic inflammatory myopathies; **IVIg**, intravenous immunoglobulin; **p25/p75**, percentile 25/75; **PM**, polymyositis; **SD**, standard deviation; **TNF**, tumor necrosis factor.

REFERENCES

1. Lundberg IE, et al. *Nat Rev Dis Primers*. 2021;7(1):86; 2. Barsotti S, et al. *Curr Treatm Opt Rheumatol*. 2018;4(4):299–315; 3. Bensimon A, et al. *Ann Rheum Dis.* 2022;81:716–717(POS0848); 4. Aggarwal R, et al. *Arthritis Rheumatol*. 2021;73(suppl 9):A1039.

CF and **SdS** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany. **LH** is an employee of Merck Healthcare Germany GmbH, Weiterstadt, Germany, an affiliate of Merck KGaA, Darmstadt, Germany. **XC** and **DD** are employees of EMD Serono. **JMM** is an employee of Cytel Inc., Toronto, Canada, and has worked as a consultant to EMD Serono.
Medical writing support was provided by Bioscript Group, Macclesfield, UK, and supported by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945).

Presented at the ISPOR 2024 meeting | 5–8 May 2024 | Atlanta, GA, USA

Copyright © 2024 remains with the authors

This study was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945)