Employer Perspectives on Alternative Funding Programs

HPR132

Laura Huff BA[1], James Motyka PharmD[2], Kimberly Westrich MA [2]

[1]Gallagher Research and Insights, St Louis, MO, USA, [2]National Pharmaceutical Council, Washington, DC, USA

BACKGROUND

- Payers and employers continue to explore various programs to mitigate specialty drug costs.
- One controversial strategy is alternative funding programs (AFP), which remove specialty medications from the list of covered drugs (i.e., formulary), leaving the patients "uninsured" for the drugs that are not covered.
- Employers with AFPs in place require employees to enroll in a patient assistance program run by an AFP vendor that attempts to obtain the drug from an alternative source, like a drug manufacturer program for uninsured patients.
- AFPs prompt more questions and pose greater challenges for employer plan sponsors and the effect of these programs on patient adherence and treatment experience is unknown.

OBJECTIVES

- To evaluate employer perspectives on AFPs, which often controversially tap into other sources (e.g., advocacy groups, foundations, or grants) to pay for patients' medication costs.

METHODS

- A national web-based survey of U.S. employer drug benefit decision-makers was fielded in February 2023.

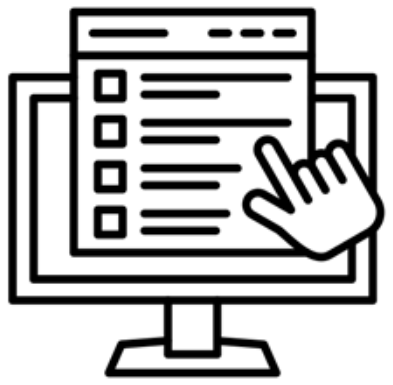

- 50 self-insured employers with 2,000 or more employees, participated in the survey.

METHODS, CONT.

Figure 1. Participants by title

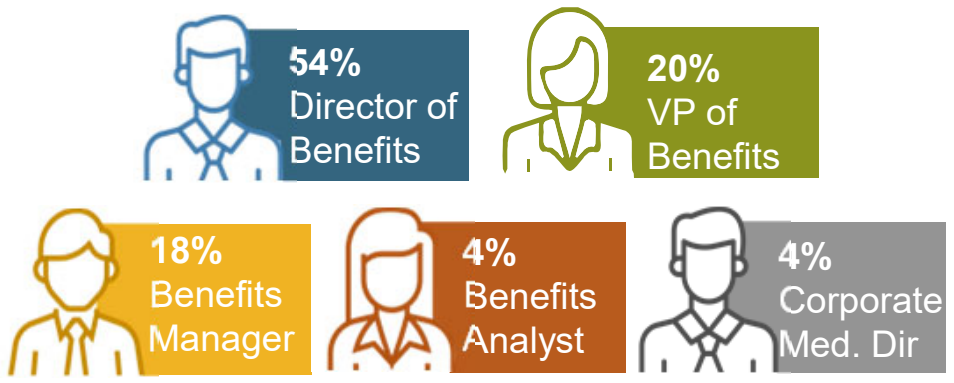

54% Director of Benefits
20% VP of Benefits
18% Benefits Manager
4% Benefits Analyst
4% Corporate Med. Dir

Figure 2. Participants by number of U.S. employees

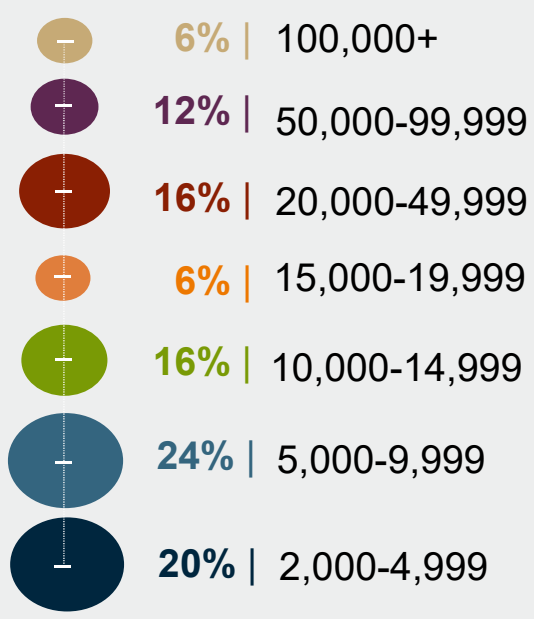

6% | 100,000+
12% | 50,000-99,999
16% | 20,000-49,999
6% | 15,000-19,999
16% | 10,000-14,999
24% | 5,000-9,999
20% | 2,000-4,999

RESULTS

- 62% of survey respondents rated the cost of specialty medications as very or extremely concerning for their organizations.
- Almost all respondents (92%) agreed that medication costs were prompting employers to consider services they would normally avoid.
- Maintaining patient access to medications was a priority when making specialty pharmacy benefits decisions, with 92% of respondents saying patient access to medications was a factor of high importance.
- 64% of respondents did not have a clear understanding of how AFPs differed from specialty medication management programs like copay accumulators or maximizers (Figure 3).

RESULTS, CONT.

Figure 3. Respondents' understanding of specialty medication management programs

How well do you understand the differences between copay accumulator adjustment programs, copay maximizers, and alternative funding programs?

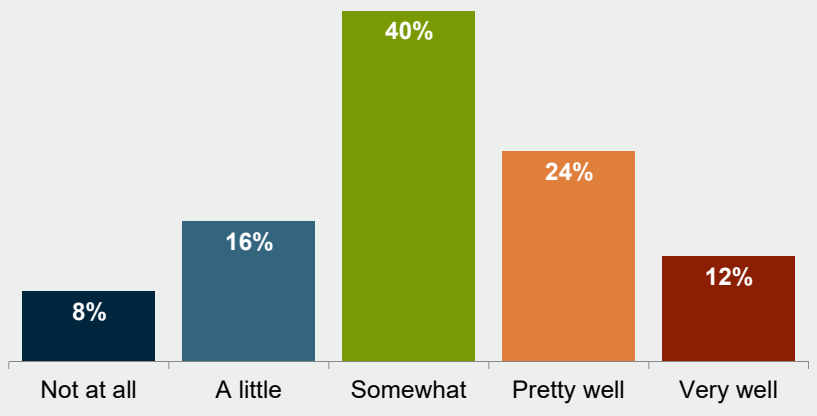

8%
16%
40%
24%
12%
Not at all
A little
Somewhat
Pretty well
Very well

- Two-thirds or more of respondents did not consider themselves knowledgeable about how AFPs affect charitable resources (74%), pharmaceutical rebates (68%), or the timing of patients receiving medications (66%) (Figure 4).

Figure 4. Respondents' understanding of alternative funding programs

How knowledgeable is your company about the following factors?

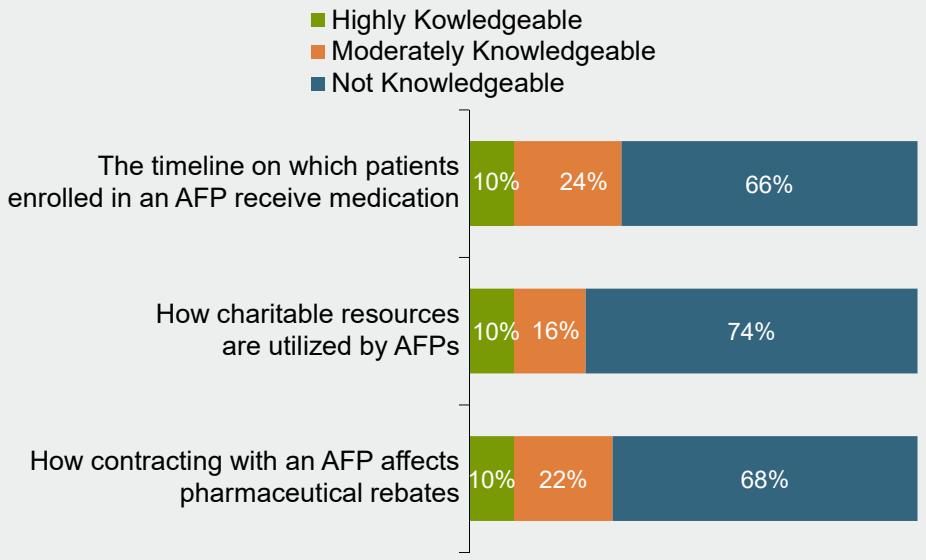

Highly Kowledgeable
Moderately Knowledgeable
Not Knowledgeable
The timeline on which patients enrolled in an AFP receive medication
10%
24%
66%
How charitable resources are utilized by AFPs
10%
16%
74%
How contracting with an AFP affects pharmaceutical rebates
10%
22%
68%

- 40% agreed that AFPs provide value to employers looking to control specialty pharmacy costs compared to 60% who had a neutral response.
- 68% considered it highly to moderately problematic if AFPs diverted resources from underinsured or uninsured patients (Figure 5).

Figure 5. Respondents' perceptions of AFPs

Below are some considerations employers might weigh when thinking about an AFP. Use the grid to indicate if your company would deem any of these considerations problematic.

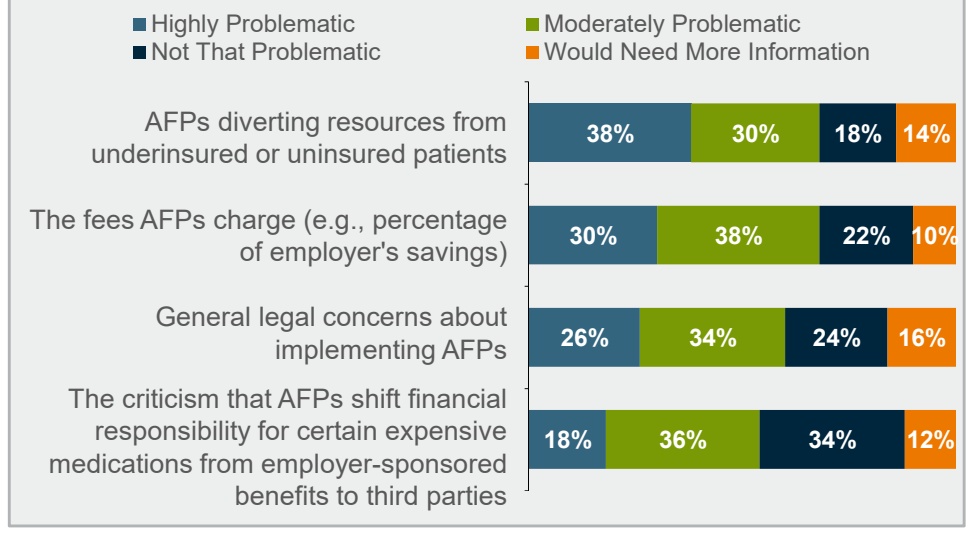

Highly Problematic
Not That Problematic
Moderately Problematic
Would Need More Information
AFPs diverting resources from underinsured or uninsured patients
38%
30%
18%
14%
The fees AFPs charge (e.g., percentage of employer's savings)
30%
38%
22%
10%
General legal concerns about implementing AFPs
26%
34%
24%
16%
The criticism that AFPs shift financial responsibility for certain expensive medications from employer-sponsored benefits to third parties
18%
36%
34%
12%

While 76% of survey respondents had not implemented an AFP, 26% were considering it.

CONCLUSIONS

- The high cost of specialty medications and a desire to sustain patient treatment access are prompting respondents to search for solutions they normally would not consider.
- This tension between trying to control pharmacy costs while still offering patients access to medications may fuel employer interest in AFPs, although most have not yet implemented the programs.
- Employer understanding of how the programs operate and differ from other specialty medication management programs is limited.
- Most survey respondents took a neutral stance on AFPs but recognized that aspects like diverting resources from under-insured patients can be problematic.

PhDPosters.com Your Research Poster, Printed For Less