Poster Code: PCR49

Mapping and Linking between the EQ-5D-5L and the PROPr in the United States

Xiaodan Tang[1], Ron D. Hays[2], David Cella[1], Sarah Acaster[3], Benjamin David Schalet[4], Asia Sikora Kessler[5], Montserrat Vera Llonch[5], Janel Hanmer[6]

[1]Department of Medical Social Sciences, Northwestern University Feinberg School of Medicine; [2]Department of Medicine, Division of General Internal Medicine & Health Services Research, University of California, Los Angeles; [3]Acaster Lloyd Consulting Ltd; [4]Department of Epidemiology and Data Science, Amsterdam UMC, Vrije Universiteit; [5]Ionis Pharmaceuticals; [6]Department of Medicine, University of Pittsburgh

Background

Two **commonly-used** preference-based measures which summarize multiple domains of health-related quality of life into a single score anchored at 0 (dead) and 1 (full health):

- EQ-5D-5L:
 - 5 domains each assessed with 1 item and 5 response options (see Table for specific domains)
- Patient-Reported Outcomes Measurement Information System (PROMIS®) preference score (PROPr)
 - Derived from **PROMIS domains**
 - 7 domains scored using **Item Response Theory** (see Table for specific domains)
 - Ceiling effects are less common

Similar content allows for mapping or linking scores between the EQ-5D-5L and the PROPr. [1-2]

Research Objectives

- Evaluate **content domain coverage** between the two preference-based measures when PROPr is collected using the PROMIS 29+2
- Align scores of the EQ-5D-5L and PROPr using **US value sets**
- **Compare mapping and linking** approaches in prediction accuracy

Methods

- 983 adults recruited from the US general population completed an online survey (detailed demographic information in Hanmer et al.)
- **Regression-based mapping**
 - **Indirect mapping**: Logistic regression mapped EQ-5D-5L item responses onto PROMIS domain scores
 - **Weighted kappa**: agreement between predicted and observed EQ-5D-5L item responses
 - **Direct mapping**: Ordinary least-squares regression mapped EQ-5D-5L preference scores onto PROMIS domains and PROPr preference scores
 - Intraclass correlations (**ICC**s) and root-mean-squared deviations (**RMSD**s): agreement between predicted and observed EQ-5D-5L preference scores
- **IRT-based linking**: bidirectional crosswalks between EQ-5D-5L item responses and corresponding PROMIS domain scores.
 - **Fixed parameter calibration**
 - **ICCs and RMSDs:** agreement between predicted and observed PROMIS domain T scores

Table. Mapping and Linking Matrix

		EQ-5D-5L Domains					EQ-5D-5L Preference Score
		Mobility	Self-care	Usual activities	Pain/ Discomfort	Anxiety/ Depression	
PROMIS Domains	Anxiety					M	
	Cognitive Function						
	Depression					M, L	M
	Fatigue	M				M	
	Pain Interference	M	M	M	M, L		M
	Physical Function	M, L	M, L	M, L	M		M
	Sleep Disturbance				M		M
	Ability to Participate in Social Roles and Activities	M		M			M

*Note. **M** denotes a **mapping** relationship was **significant**; **L** denotes a **linking** relationship was modeled. **NO mapping or linking** was significant for **cognitive function**.*

Figure. Mapping vs Linking

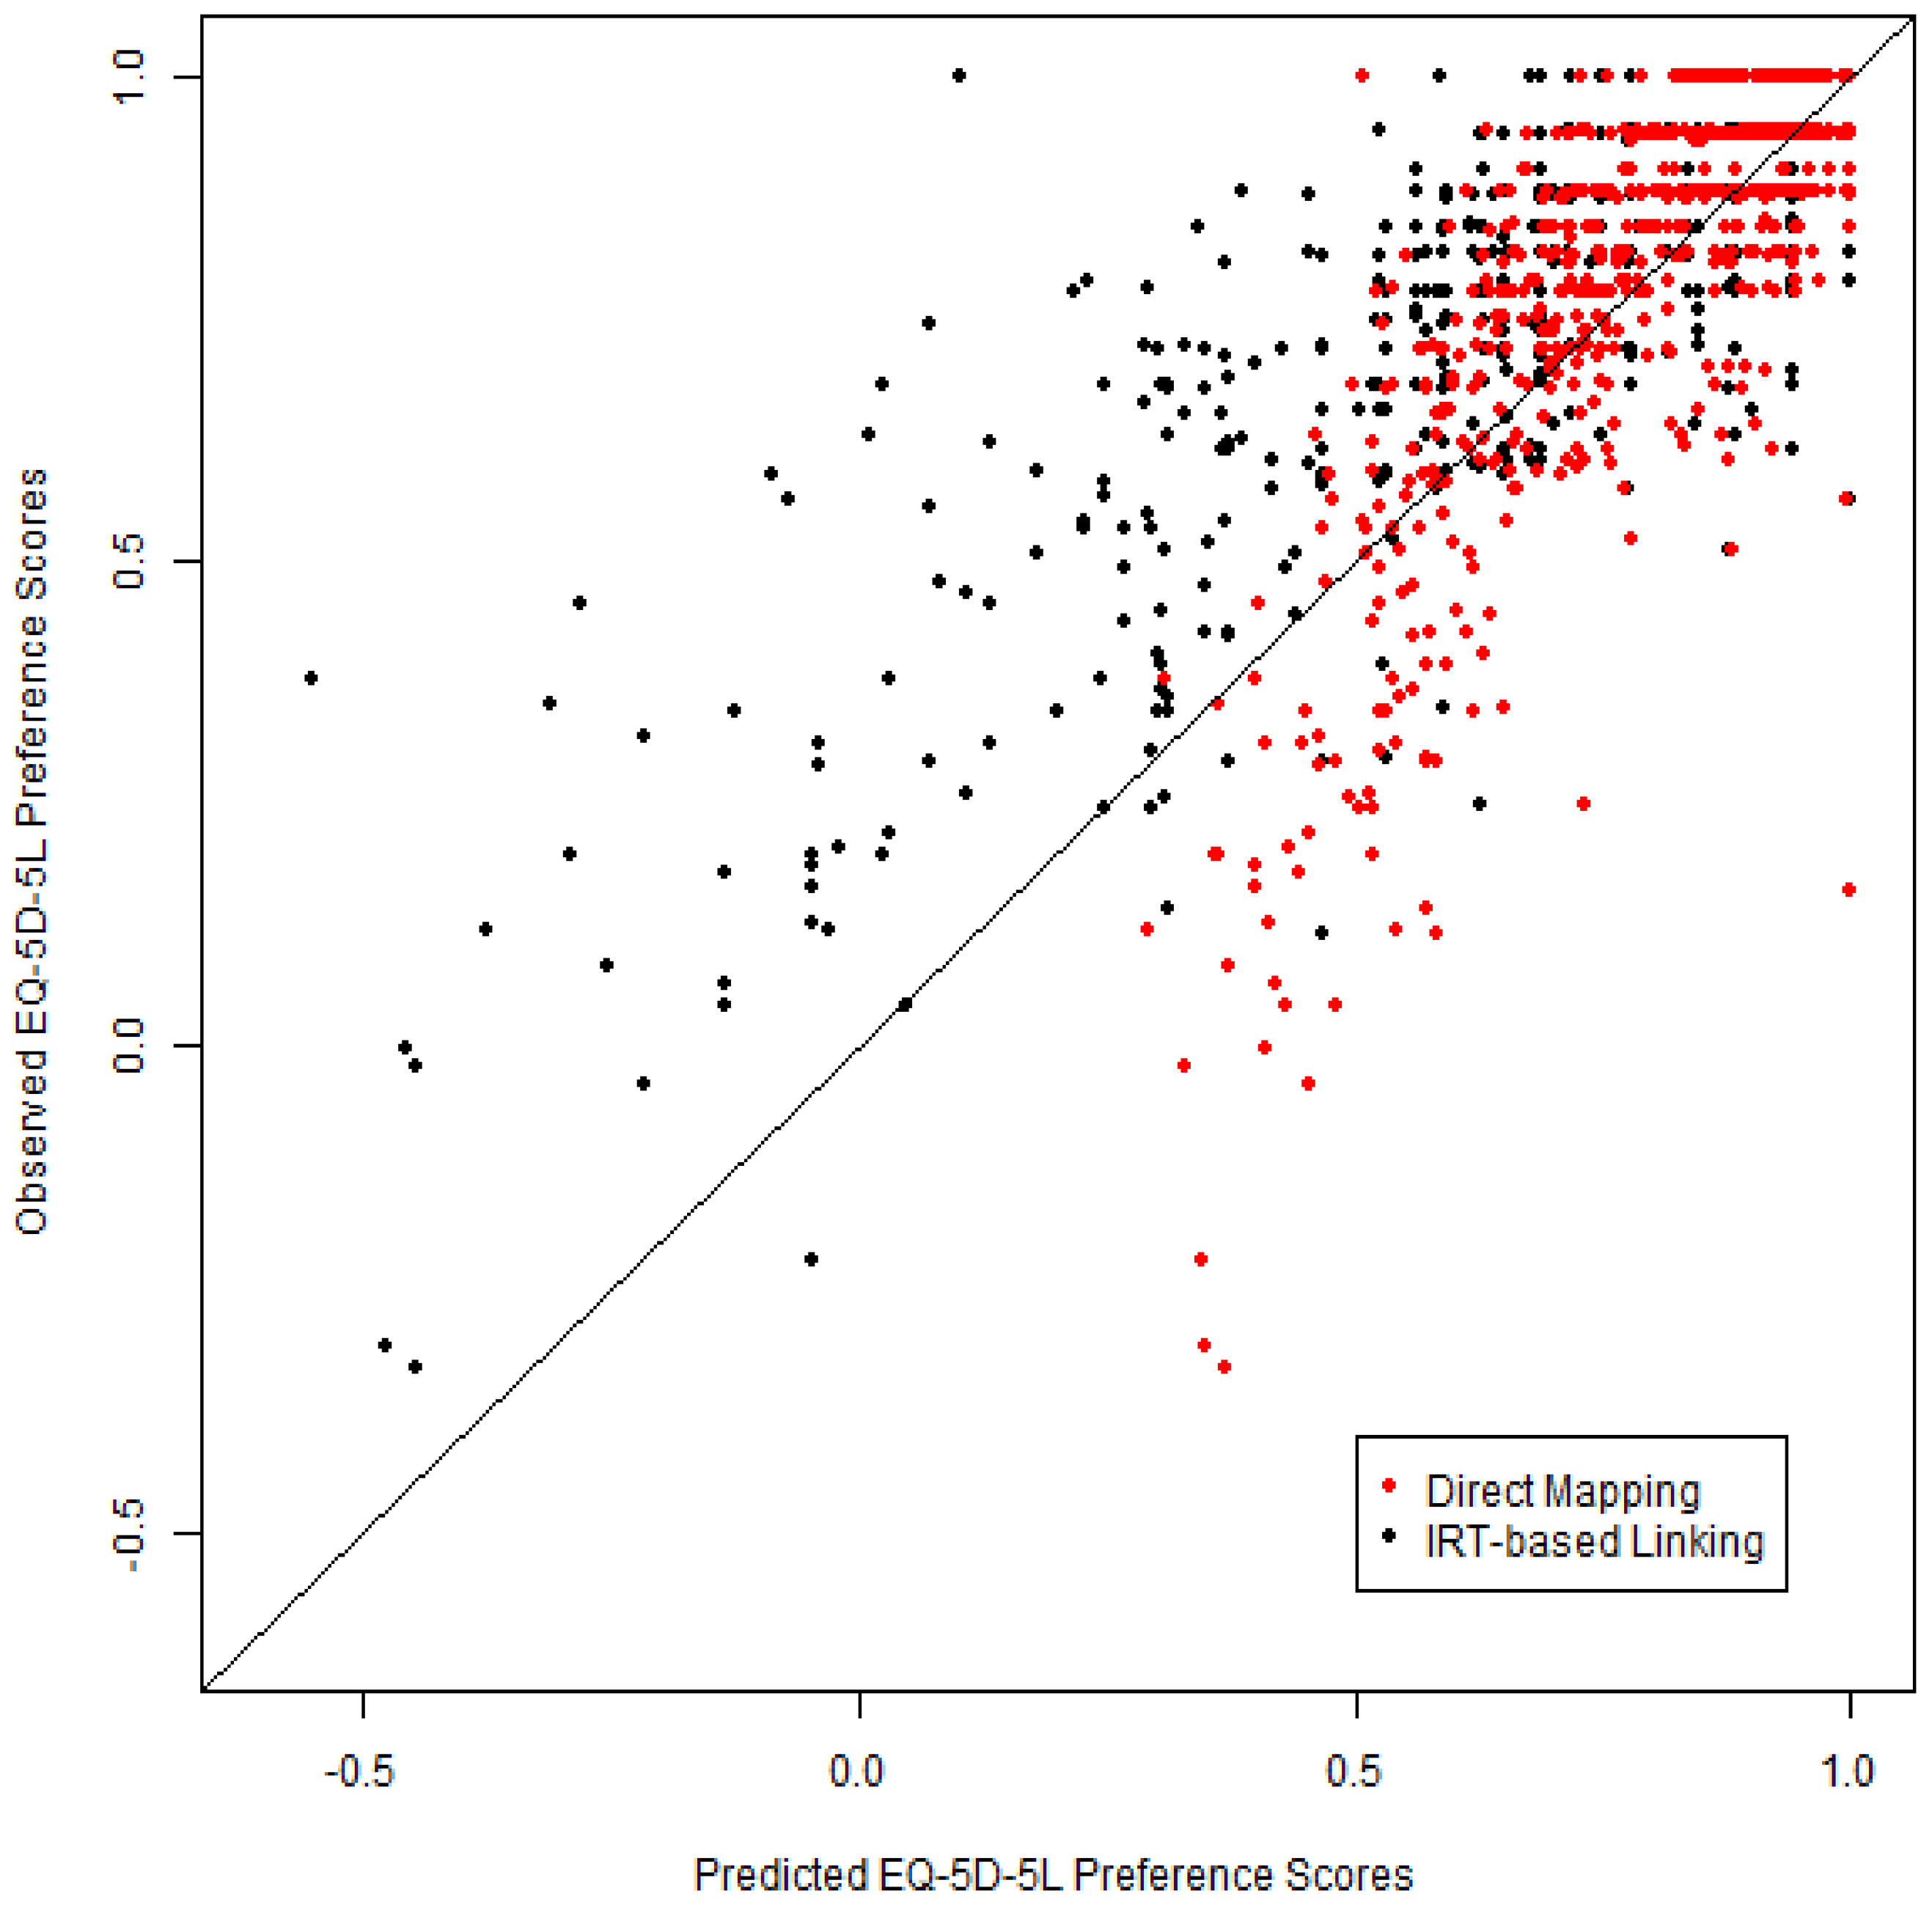
Observed EQ-5D-5L Preference Scores
Predicted EQ-5D-5L Preference Scores
-0.5
0.0
0.5
1.0
Direct Mapping
IRT-based Linking

Note. Scatterplots of predicted and observed EQ-5D-5L preference scores based on the direct mapping approach using PROMIS domains and IRT-based linking approaches. IRT = Item Response Theory.

Results

- When mapping is used, **5** PROMIS domains significantly predicted EQ-5D-5L **preference scores, 7** PROMIS domains significantly predicted EQ-5D-5L **item responses** (see Table)
- When linking is used, **3** PROMIS domains are needed to generate scores from EQ-5D-5L item responses, or **vice versa** (see Table)
- **Cognitive Function** was a **NOT** significant predictor for any EQ-5D-5L domain (see Table)
- **Small differences** (<10% EQ-5D-5L score range) **and moderate to high agreement** (>0.6 ICC for direct mapping, >0.6 kappa for both indirect mapping and linking except for Self-care) were found between observed and predicted scores for both mapping and linking methods
- **Mapping** exhibited **better precision** in estimating scores (see Fig)
- **Linking** demonstrated **a broader range** of score prediction (see Fig)

Conclusions

- **EQ-5D-5L** item scores and preference scores can be **estimated accurately** from **PROMIS health domains and PROPr**
- **Mapping equation** is **recommended** for using PROMIS domains scores to estimate EQ-5D-5L scores
- **Linking crosswalks** cover **a broader range of low preference scores** between EQ-5D-5L and PROPr/PROMIS health domains
- The mapping and linking results promote **the use of PROMIS** measures in **clinical studies**

References

[1]Aghdaee M, Gu Y, Sinha K, Parkinson B, Sharma R, Cutler H. Mapping the Patient-Reported Outcomes Measurement Information System (PROMIS-29) to EQ-5D-5L. *Pharmacoeconomics*. Feb 2023;41(2):187-198. doi:10.1007/s40273-022-01157-3

[2]Klapproth CP, van Bebber J, Sidey-Gibbons CJ, et al. Predicting EQ-5D-5L crosswalk from the PROMIS-29 profile for the United Kingdom, France, and Germany. *Health and Quality of Life Outcomes*. 2020/12/17 2020;18(1):389. doi:10.1186/s12955-020-01629-0

[3]Hanmer J, Dewitt B, Yu L, et al. Cross-sectional validation of the PROMIS-Preference scoring system. *PloS one*. 2018;13(7):e0201093-e0201093. doi:10.1371/journal.pone.0201093

Disclosures

ASK and MLV are employed by Ionis and own company stock. The rest of the authors have no relevant financial or non-financial interests to disclose.

Acknowledgement

This study was funded by Ionis Pharmaceuticals.

Northwestern Medicine® Feinberg School of Medicine

Contact: xiaodan.tang@northwestern.edu
Presented at: ISPOR 2024 Conference, May 5-8 2024, Atlanta, GA

IONIS®