EE73

Direct Healthcare Costs Associated With Primary Symptomatic Cytomegalovirus in the United States

Mihaela Georgieva,* Jane Wang, Tianyi Lu, John Diaz-Decaro, Colin Kunzweiler, Sue Hall,[†] Philip Buck

Moderna, Inc., Cambridge, MA, USA
*Presenting author.
[†]Affiliation at the time of study.

SUPPLEMENTARY MATERIAL

Figure 1. Sample Selection

≥ 1 primary or ≥ 2 secondary diagnoses for CMV disease, CMV mononucleosis, or cCMV between June 30, 2017 to February 29, 2020
N (Commercial) = 5,564 | N (Medicaid) = 3,500

≥ 6 months of continuous enrollment prior to the index date
N (Commercial) = 2,693 | N (Medicaid) = 974

Aged 9-50 years at index
N (Commercial) = 1,623 | N (Medicaid) = 615

No diagnoses of solid organ, stem cell or blood transplants at any point during the study period
N (Commercial) = 1,111 | N (Medicaid) = 420

No missing demographics* at index
N (Commercial) = 1,081 | N (Medicaid) = 379

≥ 6 months of continuous enrollment following the index date
N (Commercial) = 870 | N (Medicaid) = 319

Matched 1:1 to non-CMV controls on age, sex, region§, plan type, index year, race§, QCCI score, and IC status
N (Commercial) = 845 | N (Medicaid) = 295

CMV, cytomegalovirus; cCMV, congenital cytomegalovirus; QCCI, Quan-Charlson Comorbidity Index; IC, immunocompromised
*Demographics on the index date include sex, health plan indicator, race (Medicaid insured patients only), or geographic region (Commercially-insured patients only)
§ Region of residence is available for commercially-insured individuals only, while race is available for Medicaid-insured individuals only.

Supplementary Table 1. All-Cause HCRU and Costs – Commercially Insured Population

	Baseline				6 months follow-up				0-3 months follow-up				4-6 months follow-up			
	CMV cases N = 845	Matched controls N = 845	*P*-value		CMV cases N = 845	Matched controls N = 845	*P*-value		CMV cases N = 845	Matched controls N = 845	*P*-value		CMV cases N = 845	Matched controls N = 845	*P*-value	
All-cause HCRU ever during each time period																
Total visits, n (%)	830 (98.2)	658 (77.9)	<0.001	*	845 (100.0)	845 (100.0)	N/A		845 (100.0)	845 (100.0)	N/A		687 (81.3)	538 (63.7)	<0.001	*
Inpatient admissions, n (%)	57 (6.7)	27 (3.2)	<0.001	*	124 (14.7)	52 (6.2)	<0.001	*	94 (11.1)	33 (3.9)	<0.001	*	40 (4.7)	20 (2.4)	0.013	*
Outpatient visits, n (%)	829 (98.1)	651 (77.0)	<0.001	*	843 (99.8)	835 (98.8)	0.043	*	842 (99.6)	832 (98.5)	0.024	*	684 (80.9)	535 (63.3)	<0.001	*
Lab/imaging visits, n (%)	797 (94.3)	524 (62.0)	<0.001	*	799 (94.6)	638 (75.5)	<0.001	*	767 (90.8)	560 (66.3)	<0.001	*	541 (64.0)	369 (43.7)	<0.001	*
ED visits, n (%)	263 (31.1)	115 (13.6)	<0.001	*	227 (26.9)	122 (14.4)	<0.001	*	184 (21.8)	88 (10.4)	<0.001	*	90 (10.7)	44 (5.2)	<0.001	*
Pharmacy claims, n (%)	736 (87.1)	539 (63.8)	<0.001	*	732 (86.6)	618 (73.1)	<0.001	*	668 (79.1)	563 (66.6)	<0.001	*	614 (72.7)	435 (51.5)	<0.001	*
All-cause HCRU																
Total visits																
Mean ± SD	10.5 ± 9.9	7.4 ± 10.0			12.3 ± 11.7	8.2 ± 10.0			7.2 ± 6.5	5.3 ± 6.3			6.3 ± 6.6	4.7 ± 5.7		
Median [IQR]	8.0 [4.0-14.0]	4.0 [2.0-8.0]	<0.001	*	8.0 [5.0-16.0]	5.0 [2.0-10.0]	<0.001	*	5.0 [3.0-9.0]	3.0 [1.0-6.0]	<0.001	*	4.0 [2.0-8.0]	3.0 [1.0-5.8]	<0.001	*
Inpatient admissions																
Mean ± SD	1.2 ± 0.5	1.2 ± 0.5			1.3 ± 0.8	1.1 ± 0.4			1.2 ± 0.6	1.0 ± 0.2			1.1 ± 0.5	1.2 ± 0.4		
Median [IQR]	1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.925		1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.084		1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.053		1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.390	
Inpatient days																
Mean ± SD	6.6 ± 7.2	7.8 ± 8.9			13.0 ± 22.2	6.8 ± 13.4			13.5 ± 18.9	7.3 ± 15.7			8.3 ± 14.5	5.3 ± 5.3		
Median [IQR]	4.0 [3.0-7.0]	4.0 [3.0-8.0]	0.491		5.0 [3.0-11.0]	3.5 [3.0-5.3]	0.003	*	6.0 [3.0-13.5]	3.0 [3.0-4.0]	<0.001	*	4.0 [3.0-6.0]	4.0 [3.0-7.0]	0.844	
Outpatient visits																
Mean ± SD	9.8 ± 9.6	7.1 ± 9.7			11.7 ± 11.1	8.1 ± 9.9			6.7 ± 6.1	5.2 ± 6.2			6.1 ± 6.3	4.5 ± 5.6		
Median [IQR]	7.0 [4.0-13.0]	4.0 [2.0-8.0]	<0.001	*	8.0 [4.0-15.0]	5.0 [2.0-10.0]	<0.001	*	5.0 [3.0-9.0]	3.0 [1.0-6.0]	<0.001	*	4.0 [2.0-8.0]	3.0 [1.0-5.0]	<0.001	*
Lab/imaging visits																
Mean ± SD	4.9 ± 4.3	3.4 ± 4.4			5.6 ± 5.5	3.8 ± 5.2			3.6 ± 3.2	2.8 ± 3.6			3.2 ± 3.3	2.4 ± 2.9		
Median [IQR]	4.0 [2.0-6.0]	2.0 [1.0-4.0]	<0.001	*	4.0 [2.0-7.0]	2.0 [1.0-4.0]	<0.001	*	3.0 [1.0-5.0]	2.0 [1.0-3.0]	<0.001	*	2.0 [1.0-4.0]	1.0 [1.0-3.0]	<0.001	*
ED visits																
Mean ± SD	1.8 ± 1.3	1.7 ± 1.3			1.8 ± 1.3	1.5 ± 1.0			1.5 ± 1.1	1.4 ± 0.9			1.5 ± 1.0	1.3 ± 0.6		
Median [IQR]	1.0 [1.0-2.0]	1.0 [1.0-2.0]	0.141		1.0 [1.0-2.0]	1.0 [1.0-2.0]	0.006	*	1.0 [1.0-2.0]	1.0 [1.0-1.0]	0.341		1.0 [1.0-2.0]	1.0 [1.0-2.0]	0.387	
All-cause costs																
Total costs (medical + pharmacy)																
Mean ± SD	$13,913 ± $42,142	$8431 ± $24,589			$27,745 ± $87,455	$9115 ± $24,001			$21,074 ± $76,512	$5745 ± $15,882			$7568 ± $20,091	$4604 ± $13,083		
Median [IQR]	$3944 [$1205-$12,688]	$1081 [$322-$3864]	<0.001	*	$4689 [$1378-$20,067]	$1506 [$532-$6188]	<0.001	*	$2354 [$722-$12,238]	$858 [$307-$3248]	<0.001	*	$1444 [$409-$5678]	$608 [$204-$2648]	<0.001	*
Medical costs																
Mean ± SD	$11,740 ± $40,929	$8027 ± $24,903			$23,995 ± $82,516	$7572 ± $22,429			$19,077 ± $74,474	$4979 ± $15,350			$6081 ± $17,396	$4099 ± $13,015		
Median [IQR]	$2937 [$966-$9263]	$926 [$304-$3043]	<0.001	*	$3223 [$1030-$15,466]	$1098 [$392-$4509]	<0.001	*	$1714 [$533-$8930]	$618 [$230-$2337]	<0.001	*	$1029 [$371-$3925]	$558 [$195-$1896]	<0.001	*
Inpatient costs																
Mean ± SD	$49,509 ± $122,440	$34,566 ± $29,281			$70,973 ± $148,694	$26,100 ± $31,768			$82,061 ± $161,538	$29,160 ± $38,735			$27,173 ± $38,240	$19,747 ± $12,545		
Median [IQR]	$19,575 [$12,663-$36,026]	$21,589 [$15,493-$51,761]	0.464		$20,778 [$14,026-$54,027]	$15,636 [$12,758-$29,204]	0.028	*	$26,149 [$15,533-$68,428]	$16,503 [$12,720-$29,696]	0.016	*	$16,990 [$9773-$24,942]	$14,582 [$12,369-$23,725]	0.913	
Outpatient costs																
Mean ± SD	$5428 ± $13,053	$4929 ± $16,390			$6087 ± $13,025	$5345 ± $18,999			$3362 ± $6740	$3474 ± $11,706			$3371 ± $8521	$2963 ± $11,112		
Median [IQR]	$1818 [$781-$4669]	$733 [$274-$2188]	<0.001	*	$2023 [$782-$5710]	$879 [$340-$2580]	<0.001	*	$1197 [$406-$3202]	$521 [$219-$1684]	<0.001	*	$933 [$345-$2744]	$477 [$177-$1270]	<0.001	*
Lab/imaging visits																
Mean ± SD	$2197 ± $3790	$1726 ± $4964			$2149 ± $4257	$1885 ± $7903			$1410 ± $2806	$1399 ± $6387			$1189 ± $2841	$1138 ± $5291		
Median [IQR]	$869 [$252-$2606]	$291 [$79-$1239]	<0.001	*	$821 [$238-$2146]	$368 [$104-$1234]	<0.001	*	$451 [$119-$1434]	$281 [$78-$916]	<0.001	*	$426 [$109-$1194]	$202 [$57-$713]	<0.001	*
ED costs																
Mean ± SD	$9233 ± $16,462	$9974 ± $22,512			$27,709 ± $69,244	$4685 ± $6812			$29,963 ± $74,631	$3953 ± $6165			$8632 ± $18,539	$5176 ± $7112		
Median [IQR]	$4474 [$1793-$9818]	$2476 [$1342-$5966]	0.004	*	$6934 [$2054-$25,237]	$2300 [$1199-$4471]	<0.001	*	$8770 [$2189-$25,709]	$1997 [$1096-$3745]	<0.001	*	$2514 [$1525-$7195]	$2922 [$1627-$4439]	0.970	
Pharmacy costs																
Mean ± SD	$2557 ± $8211	$1414 ± $4975			$4,390 ± $22,979	$2135 ± $7661			$2608 ± $13,640	$1175 ± $3972			$2396 ± $11,100	$1514 ± $5000		
Median [IQR]	$189 [$51-$1127]	$74 [$21-$291]	<0.001	*	$266 [$57-$1,752]	$107 [$31-$480]	<0.001	*	$154 [$33-$1010]	$67 [$18-$254]	<0.001	*	$132 [$30-$1021]	$71 [$22-$389]	<0.001	*

CMV, cytomegalovirus; ED, emergency department; HCRU, healthcare resource utilization; IQR, interquartile range; SD, standard deviation.
*denotes a *P*-value of <0.05.

Supplementary Table 2. All-Cause HCRU and Costs -- Medicaid-Insured Population

	Baseline				6 months follow-up				0-3 months follow-up				4-6 months follow-up			
	CMV cases N = 295	Matched controls N = 295	*P*-value		CMV cases N = 295	Matched controls N = 295	*P*-value		CMV cases N = 295	Matched controls N = 295	*P*-value		CMV Cases N = 295	Matched controls N = 295	*P*-value	
All-cause HCRU, ever during each time period																
Total visits, n (%)	285 (96.6)	245 (83.1)	<0.001	*	295 (100.0)	295 (100.0)	N/A		295 (100.0)	295 (100.0)	N/A		260 (88.1)	213 (72.2)	<0.001	*
Inpatient admissions, n (%)	31 (10.5)	13 (4.4)	0.006	*	54 (18.3)	35 (11.9)	0.026	*	43 (14.6)	21 (7.1)	0.006	*	19 (6.4)	15 (5.1)	0.584	
Outpatient visits, n (%)	283 (95.9)	236 (80.0)	<0.001	*	292 (99.0)	286 (96.9)	0.114		292 (99.0)	281 (95.3)	0.010	*	252 (85.4)	206 (69.8)	<0.001	*
Lab/imaging visits, n (%)	246 (83.4)	179 (60.7)	<0.001	*	268 (90.8)	228 (77.3)	<0.001	*	252 (85.4)	211 (71.5)	<0.001	*	180 (61.0)	134 (45.4)	<0.001	*
ED visits, n (%)	151 (51.2)	108 (36.6)	<0.001	*	149 (50.5)	117 (39.7)	0.006	*	118 (40.0)	92 (31.2)	0.021	*	78 (26.4)	54 (18.3)	0.019	*
Pharmacy claims, n (%)	259 (87.8)	192 (65.1)	<0.001	*	265 (89.8)	218 (73.9)	<0.001	*	244 (82.7)	197 (66.8)	<0.001	*	228 (77.3)	163 (55.3)	<0.001	*
All-cause HCRU																
Total visits																
Mean ± SD	26.7 ± 41.4	8.6 ± 9.1			28.7 ± 42.0	12.1 ± 14.9			15.2 ± 20.9	7.3 ± 8.4			15.2 ± 22.5	6.6 ± 9.1		
Median [IQR]	12.0 [6.0-21.0]	6.0 [2.0-12.0]	<0.001	*	13.0 [7.0-26.0]	7.0 [3.0-15.0]	<0.001	*	7.0 [4.0-14.0]	5.0 [2.0-9.0]	<0.001	*	6.0 [3.0-14.3]	4.0 [2.0-8.0]	<0.001	*
Inpatient admissions																
Mean ± SD	1.4 ± 0.9	1.2 ± 0.6			1.4 ± 1.0	1.3 ± 0.9			1.2 ± 0.6	1.3 ± 0.7			1.4 ± 0.6	1.2 ± 0.8		
Median [IQR]	1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.571		1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.273		1.0 [1.0-1.0]	1.0 [1.0-1.0]	0.760		1.0 [1.0-2.0]	1.0 [1.0-1.0]	0.110	
Inpatient days																
Mean ± SD	8.4 ± 12.0	9.9 ± 10.4			12.4 ± 15.0	6.1 ± 10.4			10.7 ± 13.5	5.0 ± 3.7			10.8 ± 10.0	7.3 ± 14.1		
Median [IQR]	5.0 [3.0-7.0]	5.0 [3.0-10.0]	0.585		6.0 [3.0-15.0]	4.0 [3.0-5.0]	0.029	*	6.0 [3.0-10.0]	4.0 [3.0-5.0]	0.137		8.0 [3.5-13.0]	3.0 [3.0-4.5]	0.052	
Outpatient visits																
Mean ± SD	25.1 ± 41.4	7.9 ± 8.8			27.2 ± 41.9	11.4 ± 14.8			14.4 ± 20.9	7.0 ± 8.4			14.9 ± 22.6	6.3 ± 9.0		
Median [IQR]	10.0 [5.0-18.0]	5.0 [2.0-10.0]	<0.001	*	11.0 [6.0-24.0]	6.0 [3.0-13.8]	<0.001	*	7.0 [4.0-13.0]	4.0 [2.0-8.0]	<0.001	*	6.0 [3.0-13.3]	4.0 [1.0-7.8]	<0.001	*
Lab/imaging visits																
Mean ± SD	5.3 ± 4.6	4.3 ± 4.0			5.7 ± 5.6	4.6 ± 4.6			3.7 ± 3.3	3.3 ± 3.2			3.3 ± 3.2	2.7 ± 2.6		
Median [IQR]	4.0 [2.0-7.0]	3.0 [1.0-6.0]	0.001	*	4.0 [2.0-7.0]	3.0 [1.0-6.0]	0.015	*	2.0 [1.0-5.0]	2.0 [1.0-4.0]	0.132		2.0 [1.0-4.0]	2.0 [1.0-3.0]	0.019	*
ED visits																
Mean ± SD	3.0 ± 3.3	2.1 ± 1.5			2.8 ± 3.1	2.1 ± 1.6			2.1 ± 2.0	1.8 ± 1.1			2.3 ± 2.0	1.6 ± 1.0		
Median [IQR]	2.0 [1.0-3.0]	2.0 [1.0-2.3]	0.070		2.0 [1.0-3.0]	2.0 [1.0-2.0]	0.262		1.0 [1.0-2.0]	1.0 [1.0-2.0]	0.995		2.0 [1.0-2.0]	1.0 [1.0-2.0]	0.027	*
All-cause costs																
Total costs (medical + pharmacy)																
Mean ± SD	$19,117 ± $46,080	$4056 ± $9818			$23,079 ± $58,084	$5878 ± $25,355			$13,542 ± $38,556	$3350 ± $12,516			$10,997 ± $25,159	$3924 ± $16,379		
Median [IQR]	$2642 [$741-$15,483]	$680 [$196-$2236]	<0.001	*	$3500 [$770-$21,060]	$1060 [$281-$3766]	<0.001	*	$1998 [$456-$10,960]	$736 [$200-$2590]	<0.001	*	$1769 [$286-$10,379]	$632 [$186-$2,011]	<0.001	*
Medical costs																
Mean ± SD	$15,469 ± $42,672	$3441 ± $8727			$18,950 ± $55,415	$5149 ± $25,057			$11,592 ± $37,873	$2984 ± $12,513			$9279 ± $23,867	$3661 ± $16,791		
Median [IQR]	$2028 [$667-$8524]	$645 [$204-$2065]	<0.001	*	$2264 [$621-$14,127]	$851 [$299-$3023]	<0.001	*	$1397 [$385-$8150]	$590 [$174-$1796]	<0.001	*	$1022 [$310-$6108]	$500 [$214-$1699]	0.001	*
Inpatient costs																
Mean ± SD	$52,016 ± $119,632	$11,402 ± $14,727			$51,665 ± $106,524	$29,303 ± $76,842			$43,039 ± $95,813	$25,592 ± $45,200			$43,083 ± $68,311	$29,380 ± $60,933		
Median [IQR]	$4093 [$994-$4706]	$4219 [$2843-$15,187]	0.721		$9990 [$3779-$36,517]	$4050 [$1320-$8672]	0.150		$8339 [$3779-$27,276]	$6456 [$3398-$18,857]	0.713		$9990 [$6301-$42,048]	$3173 [$1430-$3899]	0.143	
Outpatient costs																
Mean ± SD	$9484 ± $21,956	$1640 ± $3579			$9925 ± $22,028	$2074 ± $5306			$5309 ± $11,380	$1239 ± $2358			$5767 ± $11,943	$1411 ± $4321		
Median [IQR]	$1519 [$569-$4454]	$457 [$161-$1114]	<0.001	*	$1461 [$536-$6107]	$572 [$216-$1821]	<0.001	*	$845 [$295-$3077]	$431 [$158-$1113]	<0.001	*	$843 [$234-$4198]	$394 [$175-$1184]	<0.001	*
Lab/imaging visits																
Mean ± SD	$779 ± $1835	$643 ± $1117			$861 ± $2272	$780 ± $2089			$534 ± $1348	$464 ± $985			$609 ± $1293	$675 ± $2356		
Median [IQR]	$303 [$132-$691]	$208 [$67-$499]	0.058		$322 [$125-$967]	$210 [$75-$674]	0.065		$214 [$78-$529]	$141 [$60-$397]	0.167		$268 [$99-$594]	$123 [$60-$374]	0.043	*
ED costs																
Mean ± SD	$5824 ± $16,404	$3358 ± $8875			$8871 ± $19,816	$2432 ± $5768			$7489 ± $18,917	$1296 ± $2076			$5485 ± $14,307	$2908 ± $6956		
Median [IQR]	$780 [$301-$1966]	$717 [$352-$1422]	0.734		$2200 [$579-$8358]	$590 [$341-$1630]	0.002	*	$1989 [$358-$6824]	$627 [$253-$1014]	0.004	*	$761 [$403-$2703]	$453 [$255-$2184]	0.305	
Pharmacy costs																
Mean ± SD	$5126 ± $16,969	$1438 ± $6090			$5838 ± $11,409	$1437 ± $4974			$3018 ± $5976	$763 ± $2629			$3469 ± $7181	$1011 ± $3029		
Median [IQR]	$576 [$60-$3694]	$83 [$14-$530]	<0.001	*	$806 [$100-$4900]	$150 [$17-$544]	<0.001	*	$617 [$95-$2878]	$91 [$14-$330]	<0.001	*	$653 [$41-$2620]	$74 [$22-$525]	0.001	*

CMV, cytomegalovirus; ED, emergency department; HCRU, healthcare resource utilization; IQR, interquartile range.
*denotes a *P*-value of <0.05.

Presented at The Professional Society for Health Economics and Outcomes Research (ISPOR) Annual Congress; May 5-8, 2024; Atlanta, Georgia, USA