Clinical Outcomes of Second-Line (2L) Treatments in Locally Advanced or Metastatic Esophageal Squamous Cell Cancer (ESCC): A Systematic Literature Review (SLR)

Maximiliano Augusto Novis de Figueiredo,[1] Elizabeth Smyth,[2] JeanPierre Coaquira Castro,[3]* Tanushree Chaudhary,[4] Elaine Finn,[5] Weiwei Xu,[6] Ilia Ferrusi,[7] Rachid Rafia,[8] Kavita Rodha,[9] and Lin Zhan[3]

[1]Department of Internal Medicine, Faculty of Medical Sciences, Universidade Estadual de Campinas (UNICAMP), Campinas, Brazil; [2]Oxford University Hospitals, NHS Foundation Trust, Oxford, UK; [3]BeiGene USA, Inc., Emeryville, CA, USA; [4]IQVIA, Gurgaon, India; [5]IQVIA, London, UK; [6]IQVIA, Amsterdam, Netherlands; [7]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [8]Novartis Pharmaceuticals UK Limited, London, UK; [9]Novartis Ireland Limited, Dublin, Ireland

*Affiliation at time of study

Poster No: CO62 Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Annual Conference; May 5–8, 2024; Atlanta, GA, USA

Conclusions

- **In esophageal squamous cell carcinoma (ESCC), immuno-oncology (IO) monotherapy as a second-line (2L) treatment demonstrated a clinical benefit in overall survival (OS) with a more favorable safety profile compared with chemotherapy (CT)**
- **Immuno-oncology therapies demonstrated a better safety profile compared with CT, reflected by the percentage of grade ≥3 adverse events (AEs) and treatment-related AEs (TRAEs)**
- **Immuno-oncology therapy arms were associated with better overall response rates (ORRs) compared with CT, except nivolumab, which resulted in a numerically lower ORR**
- **Immuno-oncology therapy agents tislelizumab, nivolumab, and camrelizumab generally improved or maintained patients' quality of life versus CT**
- **Continued introduction of novel treatments for 2L ESCC may improve patient outcomes**

Background

- Esophageal squamous cell carcinoma (ESCC) comprises ~90% of the annual 456,000 esophageal cancers globally[1]
- Asia had the highest disease burden of esophageal cancer in 2020 followed by Europe and Africa[2]
- ESCC is most frequently diagnosed in people aged 64 to 74 years, with a median age of 68 years[3]
- Treatments for patients with locally advanced or metastatic ESCC include IO, chemoradiotherapy, CT, chemoimmunotherapy, or radiation therapy, depending on patients' Eastern Cooperative Oncology Group Performance Status (ECOG PS) and programmed death-ligand 1 (PD-L1) expression level[4]
 - The National Comprehensive Cancer Network guidelines recommend nivolumab and pembrolizumab (combined positive score ≥10) monotherapy as 2L treatments[5]
- Tislelizumab demonstrated statistically significant and clinically meaningful improvement in OS versus CT, with a tolerable safety profile, for 2L treatment in a global population with locally advanced or metastatic ESCC[6]
 - It is approved in the United States as monotherapy for the treatment of adult patients with unresectable or metastatic ESCC after prior systemic CT that did not include a PD-(L)1 inhibitor[6]

Objective

- This systematic literature review (SLR) was conducted to identify and summarize published data on the clinical efficacy and safety of existing and more recent 2L treatment regimens for patients with locally advanced or metastatic ESCC

Methods

- The SLR followed Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines
- Database searches: Embase®, MEDLINE®, Cochrane library, and EBM + non-indexed conferences and specific trial registries (2021–2023)
- Original SLR: Inception to May 2021. First SLR update: search limited to English language, May 2021 to November 24, 2022. Second SLR update: searches from November 24, 2022 to October 13, 2023
- Eligible publications: patients ≥18 years of age with locally advanced or metastatic 2L ESCC (Stage III or IV), and clinical trials (phase 2–4)/randomized clinical trials (RCTs)/single arm with English language
- Two independent reviewers screened titles, abstracts, and full texts of relevant records against pre-defined inclusion/exclusion criteria
- Population demographics were extracted alongside reported measures of median OS (mOS), median progression-free survival (mPFS), response rates, safety, and health-related quality of life (HRQoL) outcomes

Results

- Thirteen studies reported in 25 publications were included in the SLR (**Figure 1**)

Figure 1. PRISMA Flow Diagram

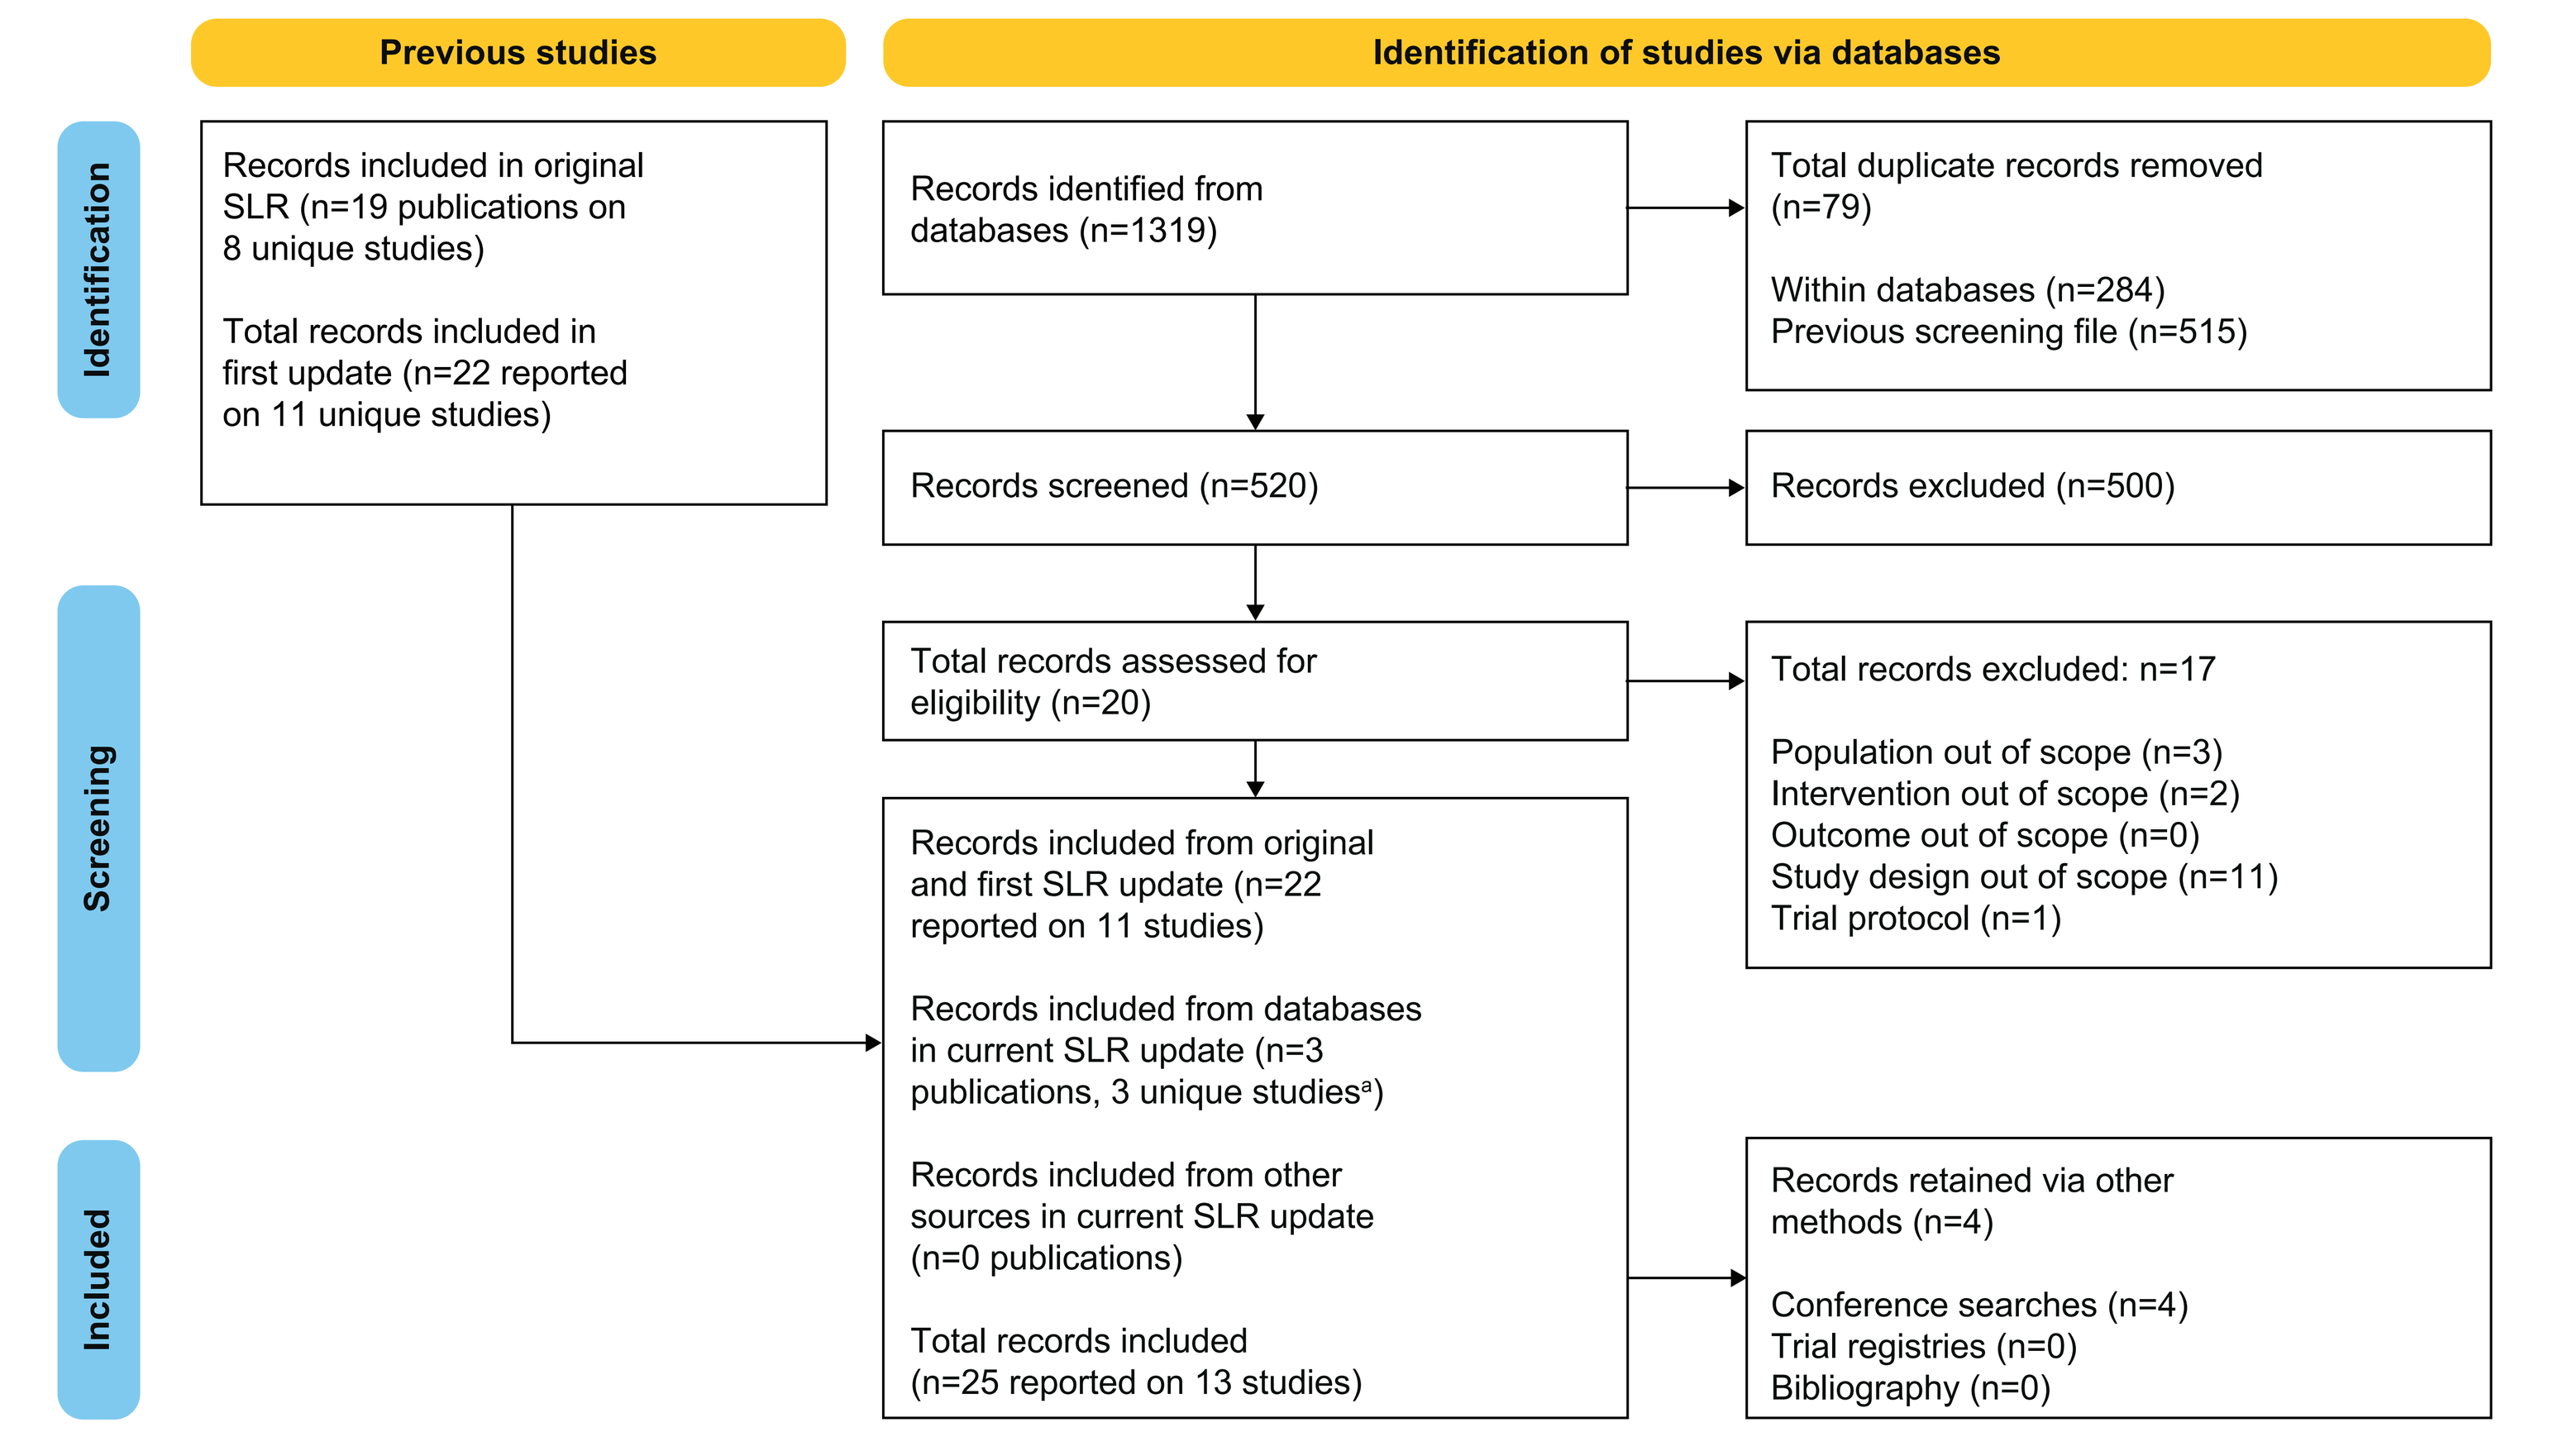
Previous studies
Identification of studies via databases
Identification
Screening
Included
Records included in original SLR (n=19 publications on 8 unique studies)
Total records included in first update (n=22 reported on 11 unique studies)
Records identified from databases (n=1319)
Total duplicate records removed (n=79)
Within databases (n=284)
Previous screening file (n=515)
Records screened (n=520)
Records excluded (n=500)
Total records assessed for eligibility (n=20)
Total records excluded: n=17
Population out of scope (n=3)
Intervention out of scope (n=2)
Outcome out of scope (n=0)
Study design out of scope (n=11)
Trial protocol (n=1)
Records included from original and first SLR update (n=22 reported on 11 studies)
Records included from databases in current SLR update (n=3 publications, 3 unique studies[a])
Records included from other sources in current SLR update (n=0 publications)
Total records included (n=25 reported on 13 studies)
Records retained via other methods (n=4)
Conference searches (n=4)
Trial registries (n=0)
Bibliography (n=0)

Note: [a]Two new studies were identified in the SLR update 2023, and 1 was a linked publication of study identified in the previous SLR update. PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses; SLR, systematic literature review.

- Six studies compared IO as monotherapy or in combination; 5 studies included CT as the intervention and comparator
- This poster identified 6 studies comparing IO with CT

Study Characteristics

- Five studies evaluated IO monotherapy (tislelizumab, pembrolizumab, nivolumab, camrelizumab, sintilimab),[7-11] and 1 study evaluated nivolumab monotherapy as well as nivolumab + ipilimumab[12]
- Four were phase 3 trials, 1 was a phase 2 trial, and 1 was a non-RCT
- The median age ranged from 60−73 years
- Across studies, most patients were Asian (78.5%−96.0%)
- ECOG PS was reported in 5 studies where most patients had PS=0 or 1. The percentage of patients with ECOG 0 was 20.0%−51.0%; ECOG 1 49.0%−80.0%. Two studies reported ECOG ≥2 for 0.3% patients
- Two studies reported smoking status at baseline; both classified patients either as having never smoked or being a current or former smoker
- Four studies reported the proportion of patients with metastases at baseline (73.0%−98.0%)

Median OS

- In all 6 studies (**Figure 2**), mOS ranged from 7.2−10.9 months for immunotherapy, and 6.2−8.5 months for CT. A statistically significant improvement in mOS versus chemotherapy was demonstrated for tislelizumab (RATIONALE-302),[7] nivolumab (ATTRACTION-3),[9] camrelizumab (ESCORT),[10] sintilimab (ORIENT-2),[11] nivolumab monotherapy, and nivolumab-ipilimumab combination (RAMONA)[12]
 - In KEYNOTE-181,[8] pembrolizumab did not demonstrate statistically significant benefit in terms of mOS compared with CT for the trial population (including both squamous cell carcinoma and adenocarcinoma)

Figure 2. Summary of mOS Across IO Versus CT Trials

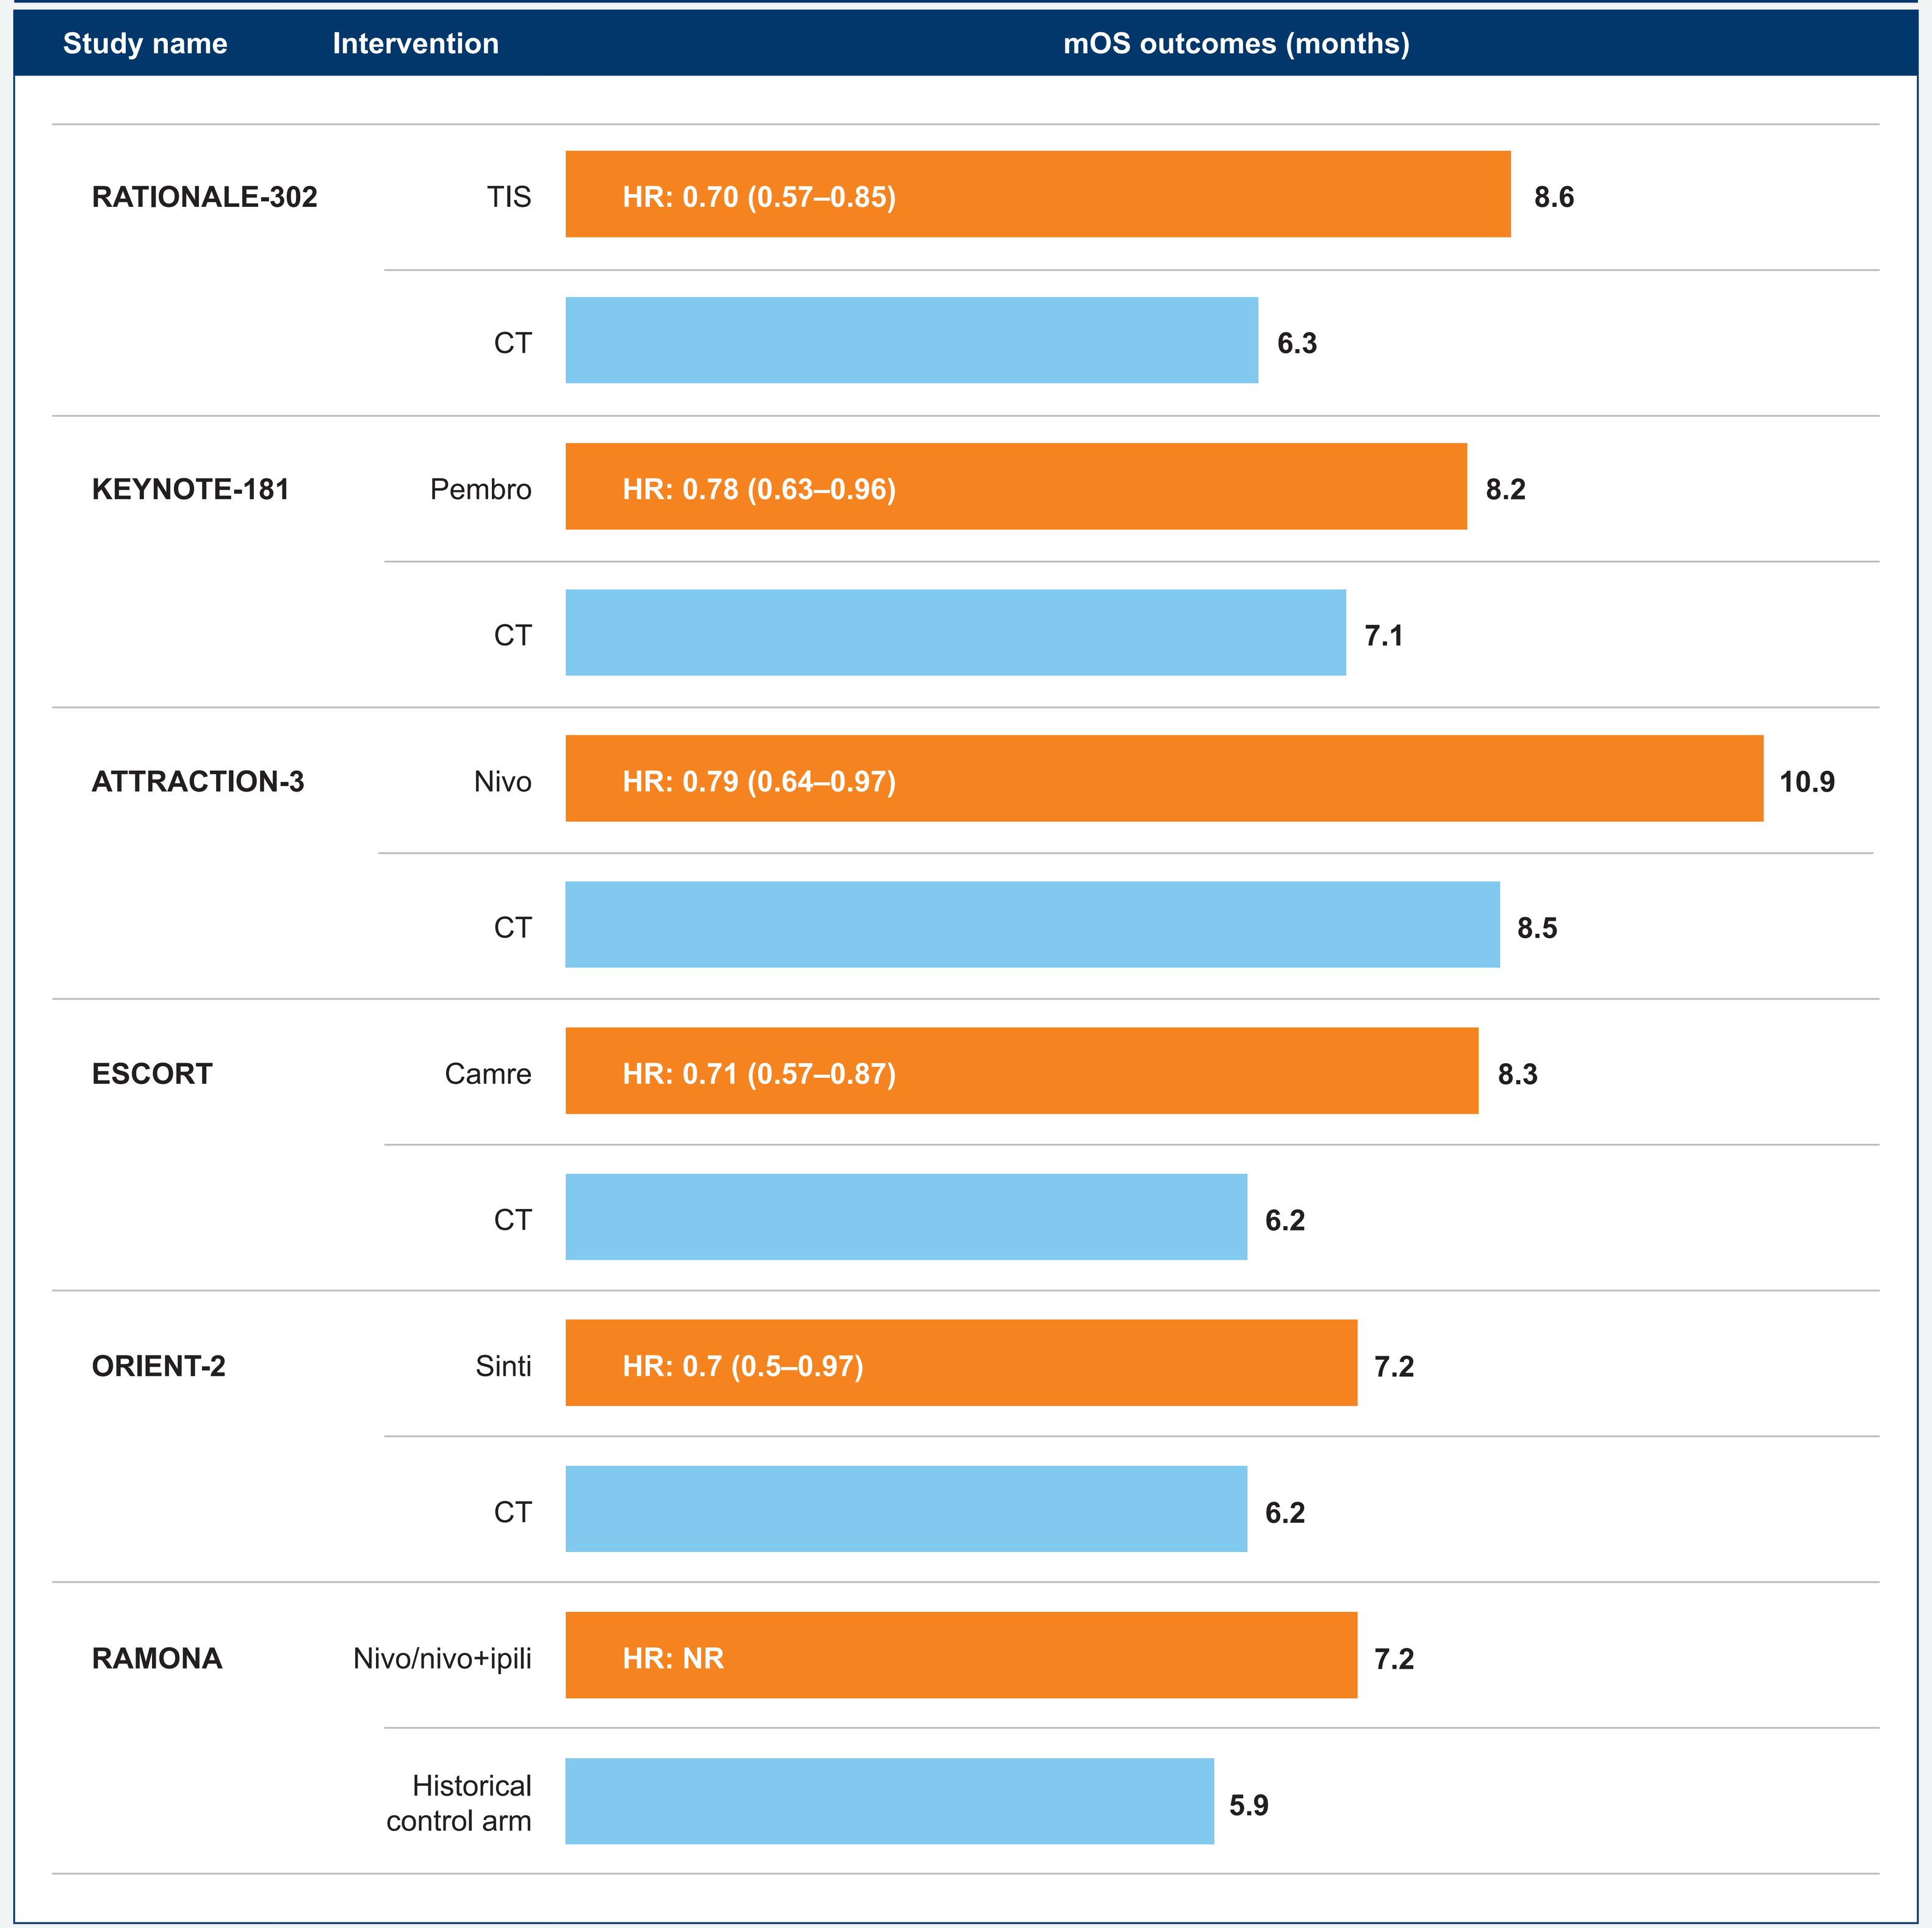
Study name
Intervention
mOS outcomes (months)
RATIONALE-302
TIS
HR: 0.70 (0.57–0.85)
8.6
CT
6.3
KEYNOTE-181
Pembro
HR: 0.78 (0.63–0.96)
8.2
CT
7.1
ATTRACTION-3
Nivo
HR: 0.79 (0.64–0.97)
10.9
CT
8.5
ESCORT
Camre
HR: 0.71 (0.57–0.87)
8.3
CT
6.2
ORIENT-2
Sinti
HR: 0.7 (0.5–0.97)
7.2
CT
6.2
RAMONA
Nivo/nivo+ipili
HR: NR
7.2
Historical control arm
5.9

Camre, camrelizumab; carbo, carboplatin; CT, chemotherapy; DCR, disease control rate; DoR, duration of response; HR, hazard ratio; IO, immuno-oncology therapy; ipili, ipilimumab; Nivo, nivolumab; Pembro: pembrolizumab; Sinti, sintilimab; TIS, tislelizumab.

Median PFS

- In all 6 studies (**Figure 3**) mPFS ranged from 1.6−2.7 months for IO, and 1.9−3.4 months for CT. No IO showed significant benefit compared with CT arms, except for camrelizumab, where mPFS was the same in both arms: 1.9 months; HR (95% CI), 0.69 (0.56−0.86); *P*=0.00063

Figure 3. Summary of mPFS Across IO Versus CT Trials

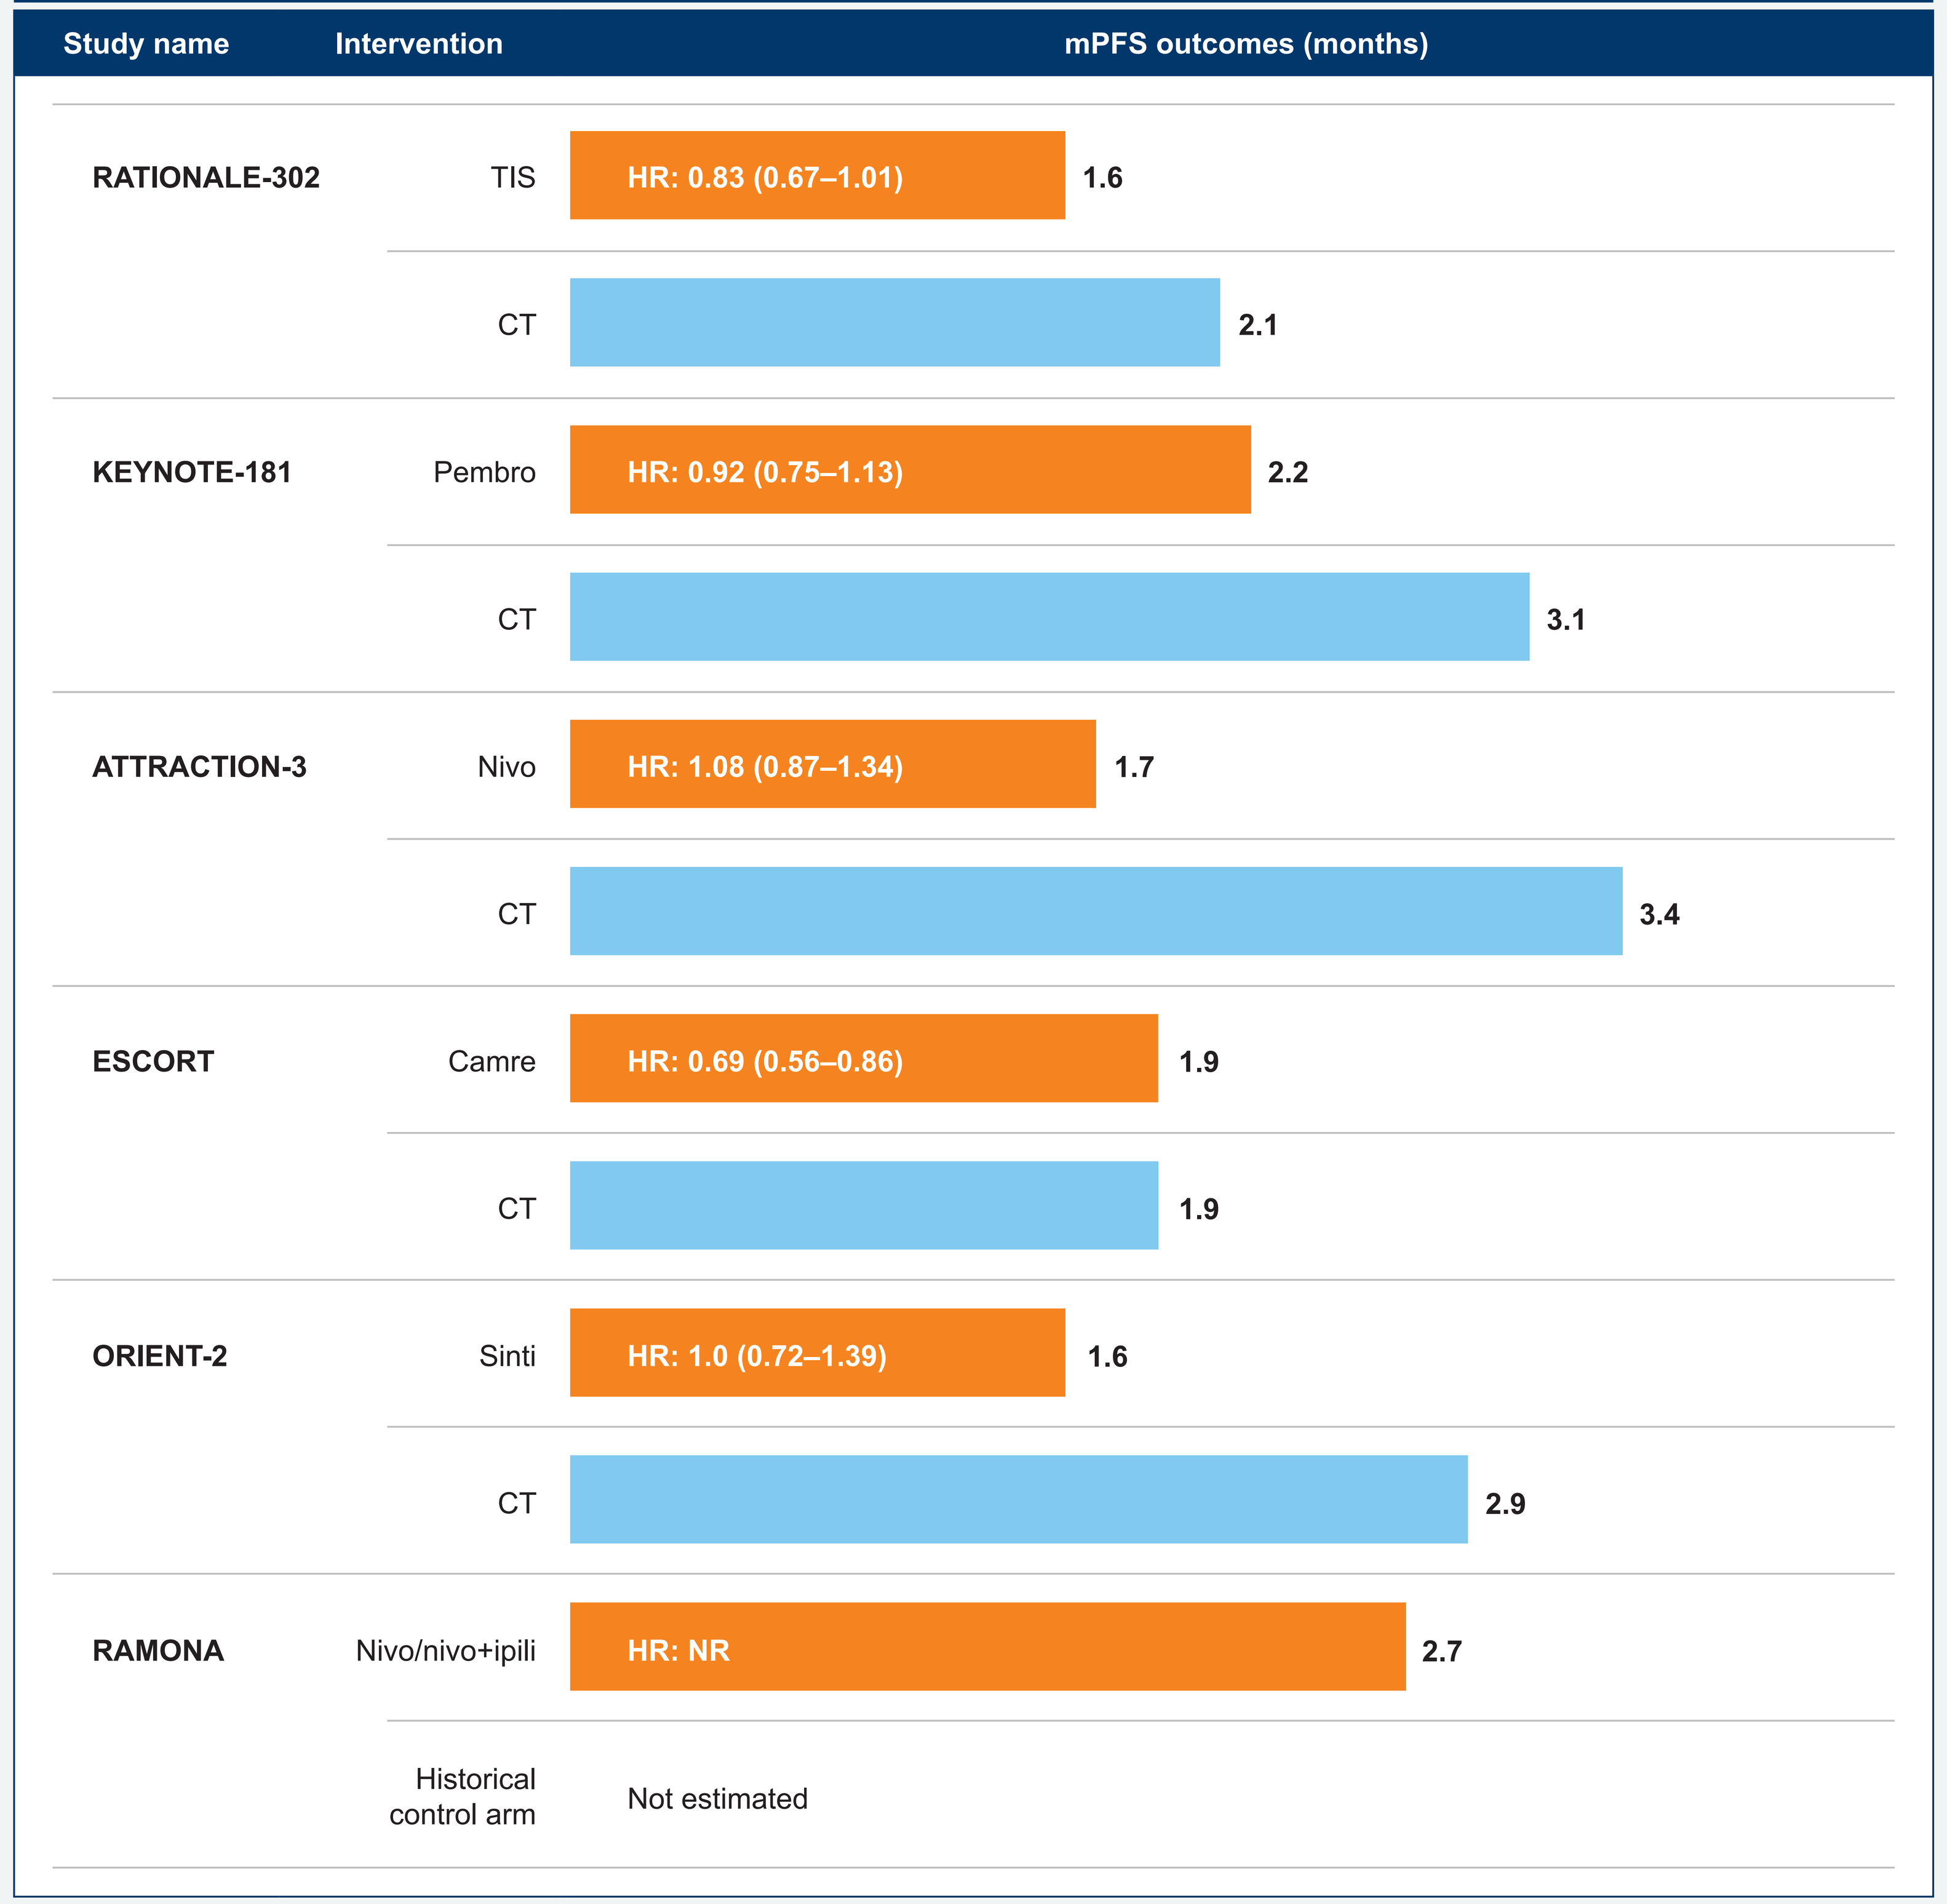
Study name
Intervention
mPFS outcomes (months)
RATIONALE-302
TIS
HR: 0.83 (0.67–1.01)
1.6
CT
2.1
KEYNOTE-181
Pembro
HR: 0.92 (0.75–1.13)
2.2
CT
3.1
ATTRACTION-3
Nivo
HR: 1.08 (0.87–1.34)
1.7
CT
3.4
ESCORT
Camre
HR: 0.69 (0.56–0.86)
1.9
CT
1.9
ORIENT-2
Sinti
HR: 1.0 (0.72–1.39)
1.6
CT
2.9
RAMONA
Nivo/nivo+ipili
HR: NR
2.7
Historical control arm
Not estimated

Camre, camrelizumab; carbo, carboplatin; CT, chemotherapy; DCR, disease control rate; DoR, duration of response; HR, hazard ratio; IO, immuno-oncology therapy; ipili, ipilimumab; Nivo, nivolumab; Pembro: pembrolizumab; Sinti, sintilimab; TIS, tislelizumab.

Response Outcomes

- In all 6 studies, IO demonstrated a higher overall response rate (ORR) compared with CT. ORR ranged from 12.6%−20.3% for IO and 6.3%−9.8% for CT
- Nivolumab was the exception, where ORRs in ATTRACTION-3 were comparable between arms (19.3% nivolumab vs. 21.5% CT)
- IO demonstrated better complete and partial response rates compared with CT in most studies
- Longer duration of response (DoR) was reported for tislelizumab, nivolumab, camrelizumab, and sintilimab compared with CT. Median DoR was 7.1−8.5 months for IO arms, compared with 3.4−6.2 months in CT arms. The KEYNOTE-181 trial reported a median DoR of 8.5 months for pembrolizumab and 10.7 months for CT

Safety Outcomes

- All 6 studies investigated safety and tolerability (as AEs, serious AEs, and discontinuation rates) of IO versus CT, except the RAMONA trial which did not report AEs for the historical control arm
- Fewer grade ≥3 TRAEs were also reported for tislelizumab, camrelizumab, nivolumab, and pembrolizumab compared with CT
- The ORIENT-2 trial reported comparable safety profiles for sintilimab and CT

HRQoL Outcome

- All studies assessed HRQoL using the EORTC-QLQ (European Organisation for Research and Treatment of Cancer quality of life questionnaire), the EQ-5D (EuroQol 5-Dimension questionnaire), or both. Compared with CT, tislelizumab, nivolumab, and camrelizumab demonstrated better HRQoL

References

1. Abnet CC, et al. *Gastroenterology*. 2018;154:360-73.
2. Liu CQ, et al. *Thorac Cancer*. 2023;14:3-11.
3. National Cancer Institute—SEER Program Squamous Cell Carcinoma of the Esophagus—Cancer StatFacts. https://seer.cancer.gov/statfacts/html/esoph.html (accessed April 9, 2024).
4. Puhr HC, et al. *ESMO Open*. 2023;8:100789.
5. National Comprehensive Cancer Network Guidelines for Patients. *Esophageal Cancer*. 2022:68.
6. BeiGene. BeiGene Receives FDA Approval for TEVIMBRA® for the Treatment of Advanced or Metastatic Esophageal Squamous Cell Carcinoma After Prior Chemotherapy. https://ir.beigene.com/news/ beigene-receives-fda-approval-for-tevimbra-for-the-treatment-of-advanced-or-metastatic-esophageal-squamous/20eb032c-15ce-456a-a852-39c88a28d811/ (accessed March 27, 2024).
7. Shen L, et al. *J Clin Oncol*. 2022;40(26):3065-76.
8. Kojima T, et al. *J Clin Oncol*. 2020;38(35):4138-48.
9. Kato K, et al. *Lancet Oncol*. 2019;20(11):1506-17.
10. Huang J, et al. *Lancet Oncol*. 2020;21:832-42.
11. Xu J, et al. Nature Commun. 2022;13(1):857.
12. Ebert MP, et al. *Lancet Healthy Longev*. 2022;3:e417-e27.

Acknowledgments

This study was sponsored by BeiGene, Ltd. Medical writing support, under the direction of the authors, was provided by Rania Kairouz-Wahbe, PhD, and Renee E. Granger, PhD, of Envision Pharma Inc., and was funded by BeiGene.

Presenter disclosures

Lin Zhan is an employee of BeiGene and may own company stock/stock options.

Contact: lin.zhan@beigene.com (Lin Zhan)

Please scan the QR code to the right to download a digital copy of this poster. Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without written permission of the authors.