Systematic Literature Review (SLR) of Randomized Controlled Trials (RCTs) of Immuno-Oncology (IO) for First-Line (1L) Treatment of Esophageal Squamous Cell Carcinoma (ESCC) in Adult Patients

Elizabeth Smyth,[1] Alexa Sibiga,[2] JeanPierre Coaquira Castro,[3]* Kirk Szafranski,[2] Samantha Craigie,[2] and Lin Zhan[3]*

[1]Oxford University Hospitals, NHS Foundation Trust, Oxford, UK; [2]EVERSANA, Burlington, ON, Canada; [3]BeiGene USA, Inc., Emeryville, CA, USA

*Affiliation at time of study

Poster No: CO17 Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Annual Conference; May 5–8, 2024; Atlanta, GA, USA

Conclusions

- **This systematic literature review (SLR) provided a comprehensive overview of trials investigating the addition of immuno-oncology (IO) therapies to standard chemotherapy (CT) in patients with previously untreated locally advanced, unresectable, or metastatic esophageal squamous cell carcinoma (ESCC)**
- **Across the 8 randomized controlled trials (RCTs) included, there were clear advantages to adding IO agents to CT in terms of overall survival (OS), progression-free survival (PFS), overall response rate (ORR), and duration of response (DoR)**
- **This treatment benefit was reflected in programmed death-ligand 1 (PD-L1)-positive, metastatic first-line (1L) patients with ESCC regardless of race or geographic locations**
- **Similar rates of all-grade treatment-related adverse events (TRAEs) were noted between the IO + CT and CT arms; however, favorable health-related quality of life (HRQoL) outcomes were noted among patients treated with IO + CT versus those treated with CT**

Background

- Esophageal squamous cell carcinoma (ESCC) accounts for approximately 90% of esophageal cancer (EC) cases globally[1]
- The prognosis for ESCC is poor, with a 5-year survival rate of <20%[2]
- Overexpression of PD-L1 is common in ESCC patients and is associated with poor prognosis[3]
- The addition of programmed cell death protein-1 (PD-1) inhibitors to CT improved outcomes in patients with previously untreated 1L unresectable, locally advanced, or metastatic ESCC[4,5]
- In this population, worldwide clinical practice guidelines recommend treatment with PD-1 inhibitors; however, limited PD-1-targeted therapeutic options are available, highlighting a need for global access to novel IO treatments[4-8]

Objective

- This SLR was conducted to summarize the efficacy, safety, and HRQoL data from RCTs assessing IO agents in patients with 1L unresectable, locally advanced, or metastatic ESCC

Methods

- The SLR followed Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines. Publications from inception to June 23, 2023 were searched in Embase, Ovid MEDLINE®, and Cochrane CENTRAL on June 23, 2023 to identify English-language RCTs of IO regimens for 1L ESCC
- Hand searches of health technology assessment agencies, conference proceedings, and trial registries were also conducted to supplement database searches
- Study selection was performed in duplicate and was assessed according to the following eligibility criteria:
 - Adult patients with previously unresectable, locally advanced or metastatic ESCC
 - Interventions: IO agents alone or in combination with CT, targeted therapy, or any other immunotherapy
 - Comparators: IO agents alone or in combination with CT, targeted therapy, or any other immunotherapy; CT alone; or placebo
 - Outcomes of interest: OS, PFS, ORR, DoR, TRAEs, and HRQoL
 - Only phase 2 and/or 3 RCTs were included
- Study details, patient characteristics, and outcomes of interest were extracted into a standardized form in Microsoft® Excel (Microsoft Corporation, Seattle, USA). In addition to the overall trial population, key subgroups of interest included PD-L1 subgroups, race/geographic region, and disease status at trial entry
- Study quality of included RCTs with available full-text publications was assessed using the NICE Single Technology Appraisal Evidence Submission Checklist for assessment of risk of bias in RCTs[8]

Results

Study Characteristics

- Of 1144 records identified in the database/registry searches and 2602 records across grey literature sources, 40 records pertaining to 8 unique phase 3, multicenter RCTs that investigated IO agents in combination with CT were identified (**Figure 1**)
- All trials were double-blind, except the open-label CheckMate 648 trial
- Across trials, the mean age of patients ranged from 62 to 64 years; the proportion of Asian patients ranged from 53% to 100%. ASTRUM-007, ESCORT-1st, GEMSTONE-304, JUPITER-06, were conducted exclusively in Asia[10,11,14,15]
- All trials required patients to have an Eastern Cooperative Oncology Group performance status of 0–1. 58% to 92% of patients had metastatic disease at baseline
- Proportion of PD-L1 positive patients (defined as tumor area positivity ≥10%, combined positivity score ≥10, or tumor positivity score ≥1%) ranged from 33% to 57%
- Seven out of 8 identified RCTs with available full-text publications were assessed for risk of bias (**Supplementary Figure 1**). Of the 7 trials, potential sources of bias included open-label design of CheckMate 648, and lack of clear description of intent-to-treat analyses or missing data in 6 trials. GEMSTONE-304 was not assessed, as trial data was only available in an abstract at the time of the SLR

Supplementary data

Figure 1: PRISMA Flow Diagram

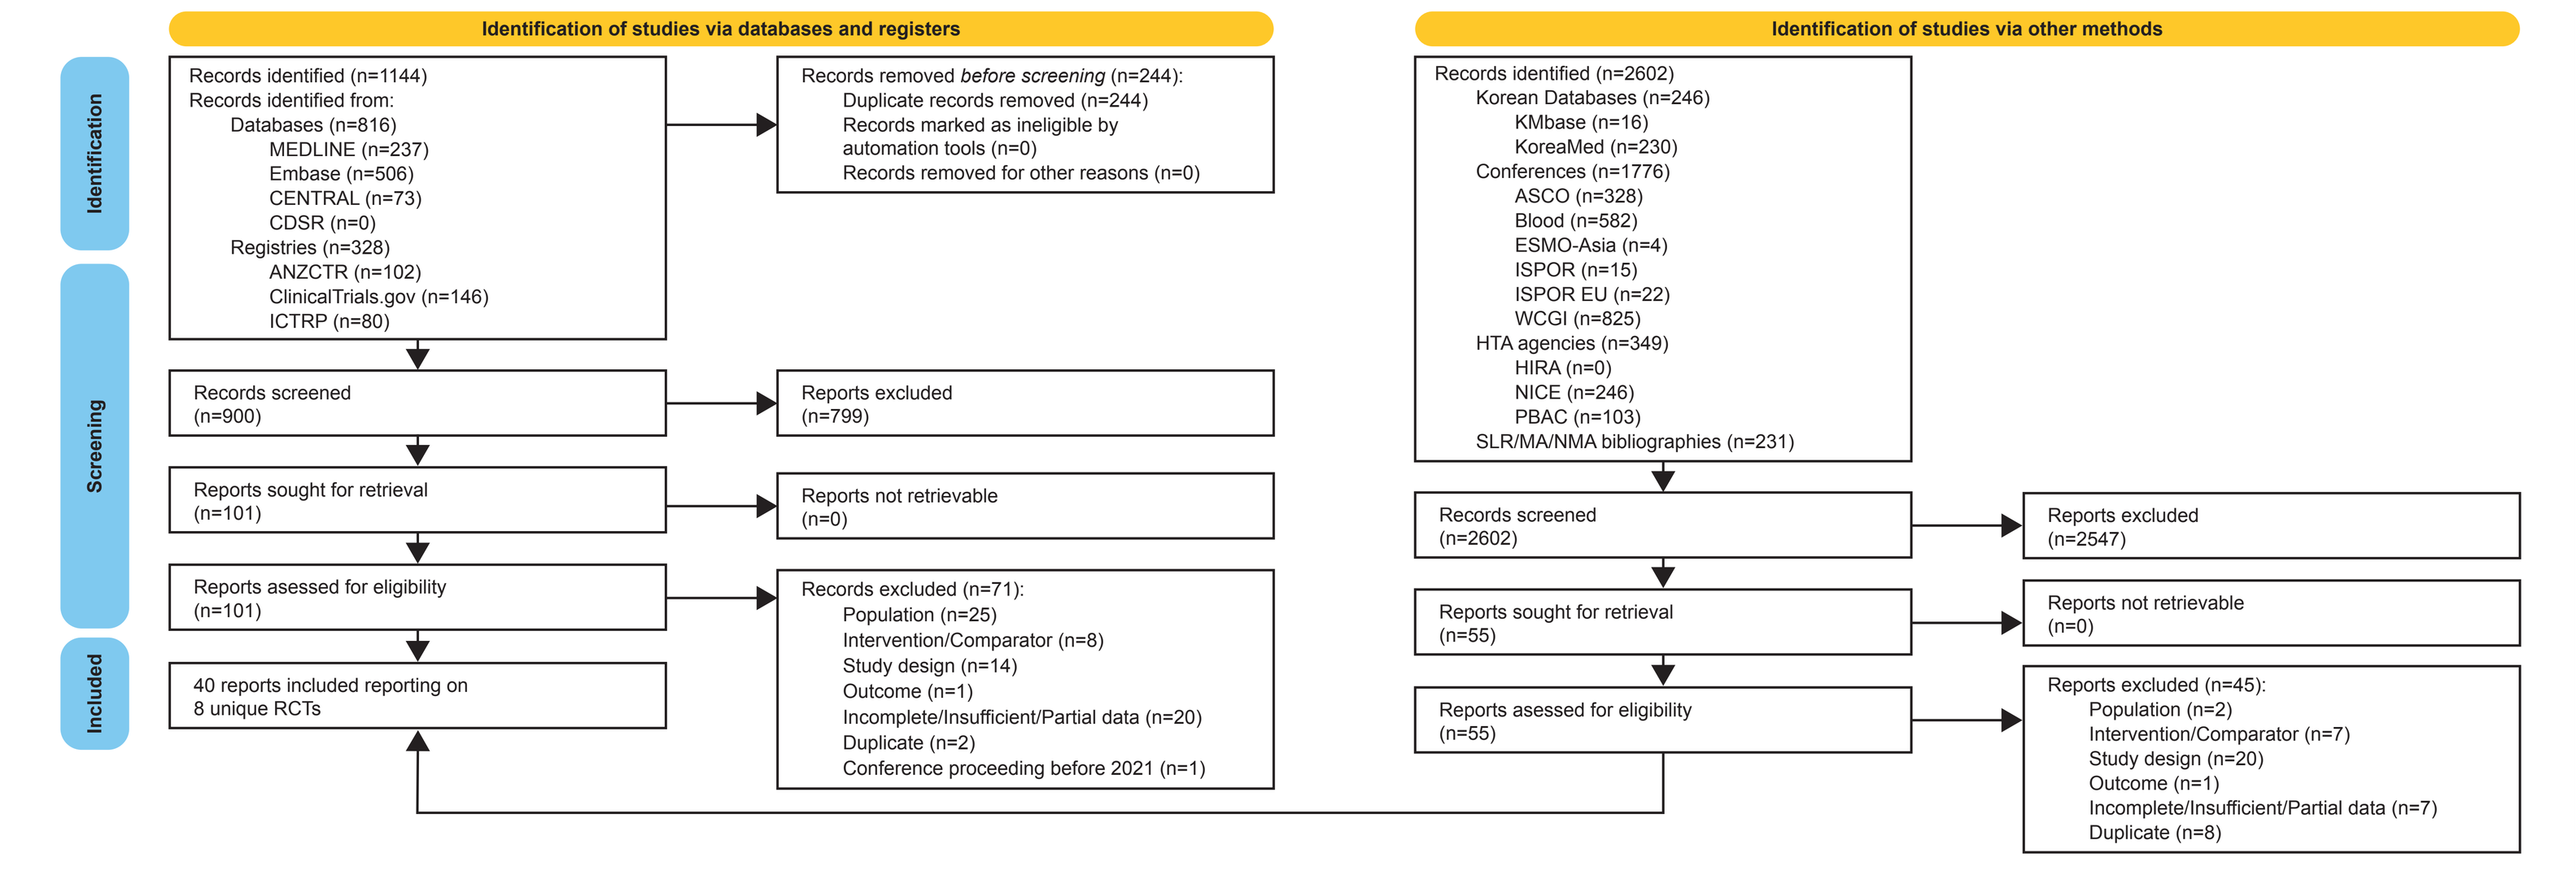
PRISMA 2020 flow diagram for new systematic reviews which included searches of databases, registries, and other sources
Identification of studies via databases and registers
Identification of studies via other methods
Identification
Screening
Included
Records identified (n=1144)
Records identified from:
Databases (n=816)
MEDLINE (n=237)
Embase (n=506)
CENTRAL (n=73)
CDSR (n=0)
Registries (n=328)
ANZCTR (n=102)
ClinicalTrials.gov (n=146)
ICTRP (n=80)
Records removed before screening (n=244):
Duplicate records removed (n=244)
Records marked as ineligible by automation tools (n=0)
Records removed for other reasons (n=0)
Records screened (n=900)
Reports excluded (n=799)
Reports sought for retrieval (n=101)
Reports not retrievable (n=0)
Reports assessed for eligibility (n=101)
Records excluded (n=71):
Population (n=25)
Intervention/Comparator (n=8)
Study design (n=14)
Outcome (n=1)
Incomplete/Insufficient/Partial data (n=20)
Duplicate (n=2)
Conference proceeding before 2021 (n=1)
40 reports included reporting on 8 unique RCTs
Records identified (n=2602)
Korean Databases (n=246)
KMbase (n=16)
KoreaMed (n=230)
Conferences (n=1776)
ASCO (n=328)
Blood (n=582)
ESMO-Asia (n=4)
ISPOR (n=15)
ISPOR EU (n=22)
WCGI (n=825)
HTA agencies (n=349)
HIRA (n=0)
NICE (n=246)
PBAC (n=103)
SLR/MA/NMA bibliographies (n=231)
Records screened (n=2602)
Reports excluded (n=2547)
Reports sought for retrieval (n=55)
Reports not retrievable (n=0)
Reports assessed for eligibility (n=55)
Reports excluded (n=45):
Population (n=2)
Intervention/Comparator (n=7)
Study design (n=20)
Outcome (n=1)
Incomplete/Insufficient/Partial data (n=7)
Duplicate (n=8)

HIRA, Hazard Identification and Risk Assessment; HTA, health technology assessment; MA, meta-analysis; NICE, National Institute for Health and Care Excellence; NMA, network meta-analysis; PBAC, Pharmaceutical Benefits Advisory Committee; PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses; RCT, randomized controlled trial; SLR, systematic literature review.

Survival Outcomes

- All 8 trials demonstrated statistically significant improvements in OS with IO + CT compared with CT alone
- Seven of 8 trials demonstrated statistically significant improvements in PFS with IO + CT compared with CT alone

Response Outcomes

- All 8 trials showed an ORR benefit with IO + CT compared with CT only; 2 trials (RATIONALE-306 and ASTRUM-007) demonstrated statistical significance of the ORR benefit with odds ratios
- DoR was longer in all IO + CT arms compared with CT alone

Subgroups

- Efficacy outcomes for subgroups based on PD-L1 positivity status are presented in **Table 1**. Efficacy outcomes for subgroups based on race or geographic location and disease status at trial entry (locally advanced, unresectable, metastatic) are presented in **Supplementary Table 1** (please see supplementary QR code from first column)
- Across all trials and subgroups presented, the benefits of IO + CT compared with CT alone were maintained. Outcomes with gain or loss of statistical significance in subgroup analyses compared with main analyses are highlighted in the corresponding tables
 - Survival and response benefits of IO agents were maintained in PD-L1-positive patients across trials
 - Clinical benefits of IO agents were consistent across trials in Asian and non-Asian patients
 - Survival benefits of IO + CT compared with CT alone were maintained in metastatic patients; benefits lost statistical significance in locally advanced/recurrent/unresectable patients in most trials. No trials reported response outcomes for patients based on disease status

Table 1. Efficacy Results Across Trials Included by PD-L1-Positivity

Trial (NCT)	Subgroup (Patients, N)[a]	Study arm (Patients, N)	Median OS, months (95% CI)	OS HR (95% CI)	Median PFS, months (95% CI)	PFS HR (95% CI)	ORR, % (95% CI)	CR/PR (%)	Median DoR, months (95% CI)
ASTRUM-007 (NCT03189719) SER + CT vs. PBO + CT CT regimen: FLU + CIS	All patients[b,c] (551)[11]	SER + CT (368)	15.3 (14.0–18.6)	0.68 (0.53–0.87)	5.8 (5.7–6.9)	0.60 (0.49–0.74)	57.6 (52.4–62.7)	14/44	6.9 (5.6–8.3)
		PBO + CT (183)	11.8 (9.7–14.0)		5.3 (4.3–5.6)		42.1 (34.8–49.6)	7/36	4.6 (4.1–5.6)
	CPS ≥10 (241)[11]	SER + CT (162)	18.6 (15.3–20.9)	0.59 (0.40–0.88)	7.1 (5.8–9.1)	0.48 (0.34–0.68)	66.0 (58.2–73.3)	NR	7.9 (5.7–12.4)
		PBO + CT (79)	13.9 (8.3–18.2)		5.3 (4.1–6.0)		41.8 (30.8–53.4)	NR	5.6 (4.2–6.9)
CheckMate 648 (NCT03783442) NIV + CT vs. CT NIV + IPI vs. CT CT regimen: FLU + CIS	All patients (970)[12]	NIV + CT (321)	12.8 (11.1–15.7)	0.78 (0.65–0.93)	5.8 (5.5–7.0)	0.83 (0.68–1.00)	47 (NR–NR)	13/34	8.2 (6.9–9.7)
		NIV + IPI (325)	12.7 (11.3–15.5)	0.77 (0.65–0.92)	2.9 (2.7–4.2)	1.26 (1.04–1.51)	27 (NR–NR)	11/17	11.1 (7.1–14.3)
		CT (324)	10.7 (9.4–12.1)	Ref	5.6 (4.3–5.9)	Ref	27 (NR–NR)	6/21	7.1 (5.7–8.2)
	TPS ≥1% (473)[13]	NIV + CT (158)	15.0 (11.9–18.6)	0.59 (0.46–0.76)	6.8 (5.7–8.3)	**0.67 (0.51–0.89)**	53 (NR–NR)	16/37	8.4 (6.9–12.4)
		NIV + IPI (158)	13.1 (11.2–17.4)	0.62 (0.48–0.80)	4.0 (2.3–4.4)	1.04 (0.79–1.36)	35 (NR–NR)	18/18	11.8 (6.8–18.0)
		CT (157)	9.1 (7.7–10.0)	Ref	4.4 (2.9–5.8)	Ref	20 (NR–NR)	5/15	5.7 (4.4–8.7)
ESCORT-1st (NCT03691090) CAM + CT vs. PBO + CT CT regimen: PAC + CIS	All patients[b] (596)[10]	CAM + CT (298)	15.3 (12.8–17.3)	0.70 (0.56–0.88)	6.9 (5.8–7.4)	0.56 (0.46–0.68)	72.1 (66.7–77.2)	6.7/65.4	7.0 (6.1–8.9)
		PBO + CT (298)	12.0 (11.0–13.3)		5.6 (5.5–5.7)		62.1 (56.3–67.6)	3.7/58.4	4.6 (4.3–5.5)
	TPS ≥1% (329)[10]	CAM + CT (166)	15.3 (12.4–NR)	0.59 (0.43–0.80)	6.9 (5.7–7.8)	0.51 (0.39–0.67)	74.1 (66.7–80.6)	NR	6.9 (5.7–8.9)
		PBO + CT (163)	11.45 (10.3–13.3)		5.6 (5.4–5.7)		65.6 (57.8–72.9)	NR	4.3 (4.1–5.4)
GEMSTONE-304 (NCT04187352) SUG + CT vs. PBO + CT CT regimen: FLU + CIS	All patients[b] (540)[14]	SUG + CT (358)	15.3 (NR–NR)	0.70 (0.55–0.90)	6.2 (NR–NR)	0.67 (0.54–0.82)	60.1 (NR–NR)	NR	6.0 (NR–NR)
		PBO + CT (182)	11.5 (NR–NR)		5.4 (NR–NR)		45.2 (NR–NR)	NR	4.5 (NR–NR)
JUPITER-06 (NCT03829969) TOR + CT vs. PBO + CT CT regimen: PAC + CIS	All patients[b] (514)[15]	TOR + CT (257)	17.0 (14.0–NE)	0.58 (0.425–0.783)	5.7 (5.6–7.0)	0.58 (0.461–0.738)	69.3 (63.2–74.8)	11.7/57.6	5.6 (4.4–8.7)
		PBO + CT (257)	11.0 (10.4–12.6)		5.5 (5.2–5.6)		52.1 (45.8–58.4)	7.0/45.1	4.2 (4.2–4.4)
	CPS ≥10 (212)[15]	TOR + CT (115)	17.0 (11.9–NE)	**0.64 (0.40–1.03)**	5.7 (5.6–7.0)	0.65 (0.45–0.92)	NR	NR	NR
		PBO + CT (97)	10.9 (9.0–13.0)		5.6 (4.5–5.7)		NR	NR	NR
KEYNOTE-590 (NCT03189719) PEM + CT vs. PBO + CT CT regimen: FLU + CIS	All patients (548)[16]	PEM + CT (274)	12.6 (10.2–14.3)	0.72 (0.60–0.88)	6.3 (6.2–6.9)	0.65 (0.54–0.78)	43.8 (37.8–49.9)	NR	9.1 (6.6–12.3)
		PBO + CT (274)	9.8 (8.6–11.1)		5.8 (5.0–6.1)		31.0 (25.6–36.9)	NR	6.1 (4.4–6.4)
	CPS ≥10 (286)[16]	PEM + CT (143)	13.9 (11.1–17.7)	0.57 (0.43–0.75)	7.3 (6.2–8.2)	0.53 (0.40–0.69)	51.0 (42.6–59.5)	NR	10.4 (8.0–16.2)
		PBO + CT (143)	8.8 (7.8–10.5)		5.4 (4.2–6.0)		28.0 (20.8–36.1)	NR	4.4 (4.1–6.2)
ORIENT-15 (NCT03748134) SIN + CT vs PBO + CT CT regimen: FLU + CIS or PAC + CIS	All patients (659)[17]	SIN + CT (327)	17.4 (16.0–19.8)	0.661 (0.554–0.788)[18]	7.2 (7.0–9.6)	0.56 (0.46–0.68)	66 (61–71)	2/64	9.7 (7.1–13.7)
		PBO + CT (332)	12.8 (11.3–14.5)		5.7 (5.5–6.8)		45 (40–51)	2/44	6.9 (5.6–7.2)
	CPS ≥10 (381)[17]	SIN + CT (188)	18.4 (16.2–24.6)	0.64 (0.50–0.80)	8.3 (6.9–12.4)	0.58 (0.45–0.75)	68 (61–74)	3/64	12.4 (7.2–15.4)
		PBO + CT (193)	14.5 (11.7–16.4)		6.4 (5.5–6.9)		49 (42–56)	2/47	5.7 (5.1–7.6)
RATIONALE-306 (NCT03783442) TIS + CT vs. PBO + CT CT regimen: FLU or CAP or PAC + CIS or OXA	All patients (649)[8]	TIS + CT (326)	17.2 (15.8–20.1)	0.66 (0.54–0.80)	8.4 (7.0–9.7)	0.60 (0.49–0.74)	68 (62–73)	15/52	7.1 (6.1–8.1)
		PBO + CT (323)	10.6 (9.3–12.1)		5.7 (5.5–6.8)		49 (43–55)	7/41	5.7 (4.4–7.1)
	TAP ≥10% (236)[8]	TIS + CT (123)	16.6 (15.3–24.4)	0.62 (0.44–0.86)	8.3 (7.0–10.2)	0.50 (0.37–0.69)	73 (64–81)	NR	NR
		PBO + CT (113)	10.0 (8.6–13.0)		5.6 (4.3–6.7)		40 (31–50)	NR	NR

[a]Disease status subgroups report n by subgroup rather than by arm.
[b]All patients in this trial were recruited from China.
[c]All randomized patients were those with PD-L1 CPS ≥1.
Hazard ratio is not statistically significant; this is different from main trial analysis.
Hazard ratio is statistically significant; this is different from main trial analysis.
CAM, camrelizumab; CAP, capecitabine; CI, confidence interval; CIS, cisplatin; CPS, combined positivity score; CR, complete response; CT, chemotherapy; DoR, duration of response; FLU, fluorouracil; HR, hazard ratio; IPI, ipilimumab; NE, not estimable; NR, not reported; ORR, objective response rate; OS, overall survival; NIV, nivolumab; OXA, oxaliplatin; PAC, paclitaxel; PBO, placebo; PEM, pembrolizumab; PFS, progression-free survival; PR, partial response; SER, serplulimab; SIN, sintilimab; SUG, sugemalimab; TAP, tumor area positivity; TIS, tislelizumab ; TOR, toripalimab; TPS, tumor positivity score.

Treatment-Related Adverse Events

- 96% to 98% of patients in IO + CT and CT arms experienced ≥1 TRAE of any grade (**Table 2**)
- All but 1 trial (ESCORT-1st)[10] reported higher incidences of Grade ≥3 TRAEs among those treated with IO + CT versus those treated with CT only

Table 2. Adverse Events Among Trials Included

Trial (NCT)	Safety reporting period from first dose	Study arm (Patients, N)	All grade TRAE, n (%)	Grade ≥3 TRAE, n (%)
ASTRUM-007 (NCT03189719)[11]	90 days after last dose or before starting a new therapy	SER + CT (382)	376 (98)	201 (53)
		PBO + CT (168)	165 (98)	81 (48)
CheckMate 648 (NCT03783442)[13]	30 days after last dose	NIV + CT (310)	297 (96)	147 (47)
		NIV + IPI (322)	256 (80)	102 (32)
		CT (304)	275 (90)	108 (38)
ESCORT-1st (NCT03691090)[10]	90 days after last dose	CAM + CT (298)	296 (99.3)	189 (63.4)
		PBO + CT (297)	288 (97.0)	201 (67.7)
GEMSTONE-304 (NCT04187352)	NR	SUG + CT (NR)	NR	NR (51.3)
		PBO + CT (NR)	NR	NR (48.4)
JUPITER-06 (NCT03829969)[15]	60 days after last dose or before starting a new therapy	TOR + CT (257)	250 (97.3)	166 (64.6)
		TOR + CT (257)	250 (97.3)	144 (56.0)
KEYNOTE-590 (NCT03189719)[16]	30 to 90 days after treatment discontinuation[a]	PEM + CT (370)	364 (98)	266 (72)
		PBO + CT (370)	360 (97)	250 (68)
ORIENT-15 (NCT03748134)[17]	90 days after last dose	SIN + CT (327)	321 (98.1)	196 (59.9)
		PBO + CT (332)	326 (98.1)	181 (54.5)
RATIONALE-306 (NCT03783442)[8]	30 days after last dose or before starting a new therapy	TIS + CT (324)	313 (97.0)	216 (66.7)
		PBO + CT (321)	309 (96.0)	207 (64.5)

[a]AEs were evaluated at 30 days; serious AEs and events of interest to pembrolizumab were assessed at 90 days.
AE, adverse event; CAM, camrelizumab; CPS, combined positivity score; CT, chemotherapy; IPI, ipilimumab; NIV, nivolumab; NR, not reported; PBO, placebo; PD-L1, programmed death-ligand 1; PEM, pembrolizumab; SER, serplulimab; SIN, sintilimab; SUG, sugemalimab; TIS, tislelizumab; TOR, toripalimab; TRAE, treatment-related AE.

Health-Related Quality of Life

- Four trials (CheckMate 648, ESCORT-1st, KEYNOTE-590, ORIENT-15) reported HRQoL outcomes (**Table 3**)

Table 3. Health-Related Quality of Life Outcomes Among Trials Included

Trial (NCT)	HRQoL scales reported	Study arm (Patients assessed for HRQoL, n)	Summary of results
CheckMate 648 (NCT03783442)[19]	FACT-E	NIV + CT (310)	Changes in score from baseline showed better HRQoL for the NIV + IPI and NIV + CT arms versus CT alone, although results were not statistically significant.
		NIV + IPI (322)	
		CT (304)	
ESCORT-1st (NCT03691090)[10]	EORTC QLQ-C30 EORTC QLQ-OES18	CAM + CT (298)	Assessment up to 36 weeks showed statistically significant results in favor of CAM + CT for improvement from baseline in some subscales of the EORTC QLQ-C30 and EORTC QLQ-OES18
		PBO + CT (298)	
KEYNOTE-590 (NCT03189719)[20]	EORTC QLQ-30	PEM + CT (356)	• No significant differences in change from baseline between treatments groups were observed for EORTC QLQ-30 • At week 18, LSM change in EORTC QLQ-OES18 pain subscale score from baseline was higher for PEM+CT compared with PBO + CT
		PBO + CT (355)	
ORIENT-15 (NCT03748134)[17]	EQ-5D-5L VAS EORTC QLQ-30	SIN + CT (NR)	Compared with PBO + CT, • SIN + CT was associated with larger improvements in EQ-5D-5L VAS at 48 weeks from baseline • SIN + CT had a decreased risk of deterioration in global health status/quality of life domain of the EORTC QLQ-C30
		PBO + CT (NR)	

CAM, camrelizumab; CT, chemotherapy; EQ-5D-5L, EuroQoL 5 dimensions 5 levels; EORTC-QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire Core-30; EORTC QLQ OES18, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire-Oesophageal Cancer; FACT-E, Functional Assessment of Cancer- Esophageal; HRQoL, health-related quality of life; IPI, ipilimumab; NIV, nivolumab; NR, not reported; PBO, placebo; PEM, pembrolizumab; SIN, sintilimab; VAS, visual analog scale.

References

1. Abnet CC, et al. *Gastroenterology*. 2018;154(2):360-373.
2. Wu J, et al. *Transl Cancer Res*. 2021;10(5):2144-2152.
3. Zhang W, et al. *Ann Transl Med*. 2020;8(18):1193.
4. Ajani JA, et al. *J Natl Compr Canc Netw*. 2023;21(4):393-422.
5. Obermannova R, et al. *Ann Oncol*. 2022;33(10):992-1004.
6. Patel MA, et al. *J Clin Oncol*. 2022;40(24):2751-2762.
7. Thuss-Patience P, et al. *Curr Oncol*. 2022;29(4):2461-2471.
8. Xu J, et al. *Lancet Oncol*. 2023;24(5):483-495.
9. NICE. Quality assessment of the relevant clinical effectiveness evidence. 2022 February 10, 2024 [cited 2024; Available from: https://www.nice.org.uk/process/pmg24/chapter/clinical-effectiveness#quality-assessment-of-the-relevant-clinical-effectiveness-evidence
10. Luo H, et al. *JAMA*. 2021;326(10):916-925.
11. Zhang B, et al. *Nature Med*. 2023;29(2):473-482.
12. Kato K, et al. *J Clin Oncol*. 2023;41(4 Supplement):290.
13. Doki Y, et al. *N Engl J Med*. 2022;386(5):449-462.
14. Li J, et al. *Ann Oncol*. 2023;34(S1).
15. Wang ZX, et al. *Cancer Cell*. 2022;40(3):277-288.e273.
16. Sun JM, et al. *Lancet*. 2021;398(10302):759-771.
17. Lu Z, et al. *BMJ*. 2022.
18. Lu Z, et al. *Cancer Res*. 2023;83(8 Supplement).
19. Bridgewater JA, et al. *J Clin Oncol*. 2022;40(4 Supplement).
20. Metges JP, et al. *J Clin Oncol*. 2022;40(4 Supplement).

Acknowledgments

This study was sponsored by BeiGene, Ltd. Editorial support, under the direction of the authors, was provided by Thai Cao, MS, and Smitha Reddy, PhD, of Envision Pharma Inc., and was funded by BeiGene. The authors would like to thank Teresa Kangappaden and Dennis Gotthardt for their support with the SLR and Joanna M. Bielecki, MISt for the design of search strategies and other assistance.

Presenter disclosures

Lin Zhan is an employee of BeiGene and may own company stock/stock options.

Contact: lin.zhan@beigene.com (Lin Zhan)

Please scan the QR code to the right to download a digital copy of this poster. Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without written permission of the authors.