PCR228

Healthcare Provider Preferences for Attributes of Influenza Vaccines in the United States: Results From a Discrete-Choice Experiment Study

Christine Poulos, PhD[1]; Alan McMichael, PhD[2]; Cannon Kent, MS[1]; Deborah Rudin, MD[3]; Philip Buck, PhD[3]; Parinaz Ghaswalla, PhD[3]; Darshan Mehta, PhD[3]

[1] RTI Health Solutions, Research Triangle Park, NC, United States; [2] RTI Health Solutions, Belfast, Northern Ireland, United Kingdom; [3] Moderna, Inc., Cambridge, MA, United States

BACKGROUND

- In the United States (US), the Centers for Disease Control and Prevention (CDC) updates influenza vaccination recommendations annually.
- The CDC's vaccination recommendations are informed by the Evidence to Recommendation Framework, which has increasingly taken stakeholder preferences into account.[1]
- There have been few studies of healthcare provider (HCP) preferences for influenza vaccination.[2]

OBJECTIVE

- The aim of this study was to quantify preferences for influenza vaccination attributes among HCPs in the US.

METHODS

- HCPs were recruited from an online panel if they met the following criteria: aged 25 years or older; recommended or administered influenza vaccines to adults; a physician, pharmacist, nurse practitioner, or physician assistant licensed to practice (and administer vaccines) in the US; able to read and understand English.
- Using an online discrete-choice experiment, US HCPs were presented with a series of experimentally-designed pairs of hypothetical influenza vaccines composed of the attributes and levels shown in **Table 1**.
- Each respondent was also randomly assigned to 1 of 2 patient profiles: (1) an adult aged 18-64 with at least 1 risk factor for serious flu complications and (2) an adult aged 65 or older with no additional risk factors for serious flu complications.
- In each choice question (see example in **Figure 1**), respondents were asked to choose 1 of the 2 influenza vaccines for the assigned patient profile, or the option to recommend no flu vaccine.

Table 1. Attributes and Levels Included in the Discrete-Choice Experiment

Attribute label	Attribute levels
Vaccine efficacy (absolute reduction in influenza risk, assuming a 10% baseline risk of influenza)	• Vaccine prevents flu in 15 of 100 people in the next year • Vaccine prevents flu in 25 of 100 people in the next year • Vaccine prevents flu in 50 of 100 people in the next year • Vaccine prevents flu in 60 of 100 people in the next year
Prevention of hospitalizations	• Same as standard-dose flu vaccines • More than standard-dose flu vaccines
Durability of influenza protection	• Fades within 6 months • Stays the same for at least 6 months
Risk of moderate to severe injection site reactions	• 0 injection site reactions in 1,000 people (0%) • 150 injection site reactions in 1,000 people (15%) • 300 injection site reactions in 1,000 people (30%) • 500 injection site reactions in 1,000 people (50%)
Risk of moderate to severe systemic side effects ("flu-like reactions")	• 0 systemic reactions in 1,000 people (0%) • 150 systemic reactions in 1,000 people (15%) • 300 systemic reactions in 1,000 people (30%) • 500 systemic reactions in 1,000 people (50%) [narrow scope range only][a] • 600 systemic reactions in 1,000 people (60%) [wide scope range only][a]

[a] The study included an assessment of respondents' sensitivity to absolute differences in risks, known as a scope test.[3] Respondents were randomly assigned to 1 of 2 ranges of the risk of systemic side effects: narrow or wide.

- Random-parameters logit analysis results were used to calculate conditional relative attribute importance (CRAI) out of 100% and maximum acceptable risks (MARs) of moderate to severe vaccine side effects in exchange for improvements in vaccine efficacy.

Figure 1. Example Choice Question

Feature	Vaccine A	Vaccine B	No flu vaccine
Flu vaccine efficacy	Vaccine prevents flu in 15 of 100 people in the next year	Vaccine prevents flu in 60 of 100 people in the next year	No flu infections prevented, 100 of 1,000 people (10%) get the flu
Prevention of hospitalization due to severe flu-related complications	**Same** as standard dose flu vaccines	**More** than standard dose flu vaccines	Not applicable, no vaccine
Durability of flu protection in the 6 months after vaccination	**Fades** within 6 months	**Stays the same** for at least 6 months	Not applicable, no vaccine
Risk of moderate to severe injection site reaction	500 injection site reactions in 1,000 people (50%)	150 injection site reactions in 1,000 people (15%)	Not applicable, no vaccine
Risk of moderate to severe systemic (flu-like) reaction	150 systemic reactions in 1,000 people (15%)	300 systemic reactions in 1,000 people (30%)	Not applicable, no vaccine
Which option would you recommend?	○	○	○

Note: There were 48 vaccine pairs in the experimental design divided into 6 blocks of 8 vaccine choice questions. Each respondent was randomly assigned to 1 block of 8 vaccine choice questions.

RESULTS

Respondent Characteristics (N = 299)

Mean age 42.7 YEARS (SD, 10.9) Min, 25; Max, 72

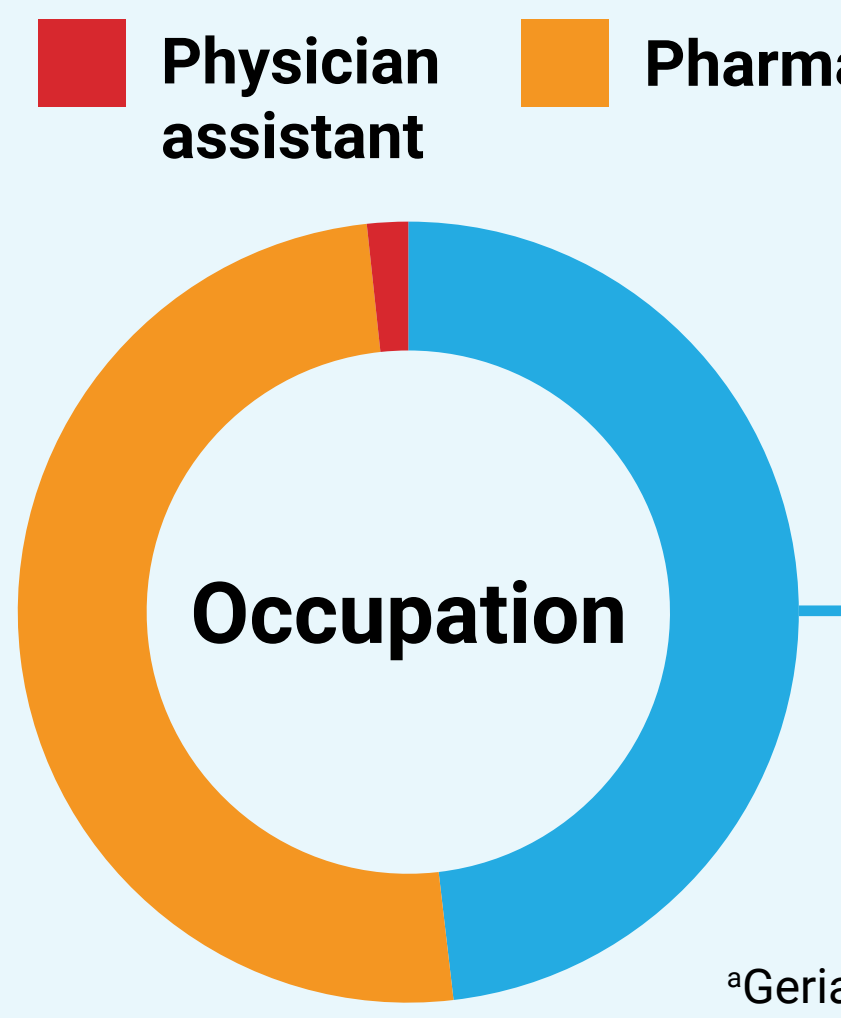
Physician assistant
Pharmacist
Physician
Occupation

SPECIALTY
63.8% Family practice
28.2% Primary care
4.0% Internal medicine
3.4% General practice
0.7% Geriatrics[a]

[a]Geriatric primary care physician [Eligible]

Years in practice (since training)

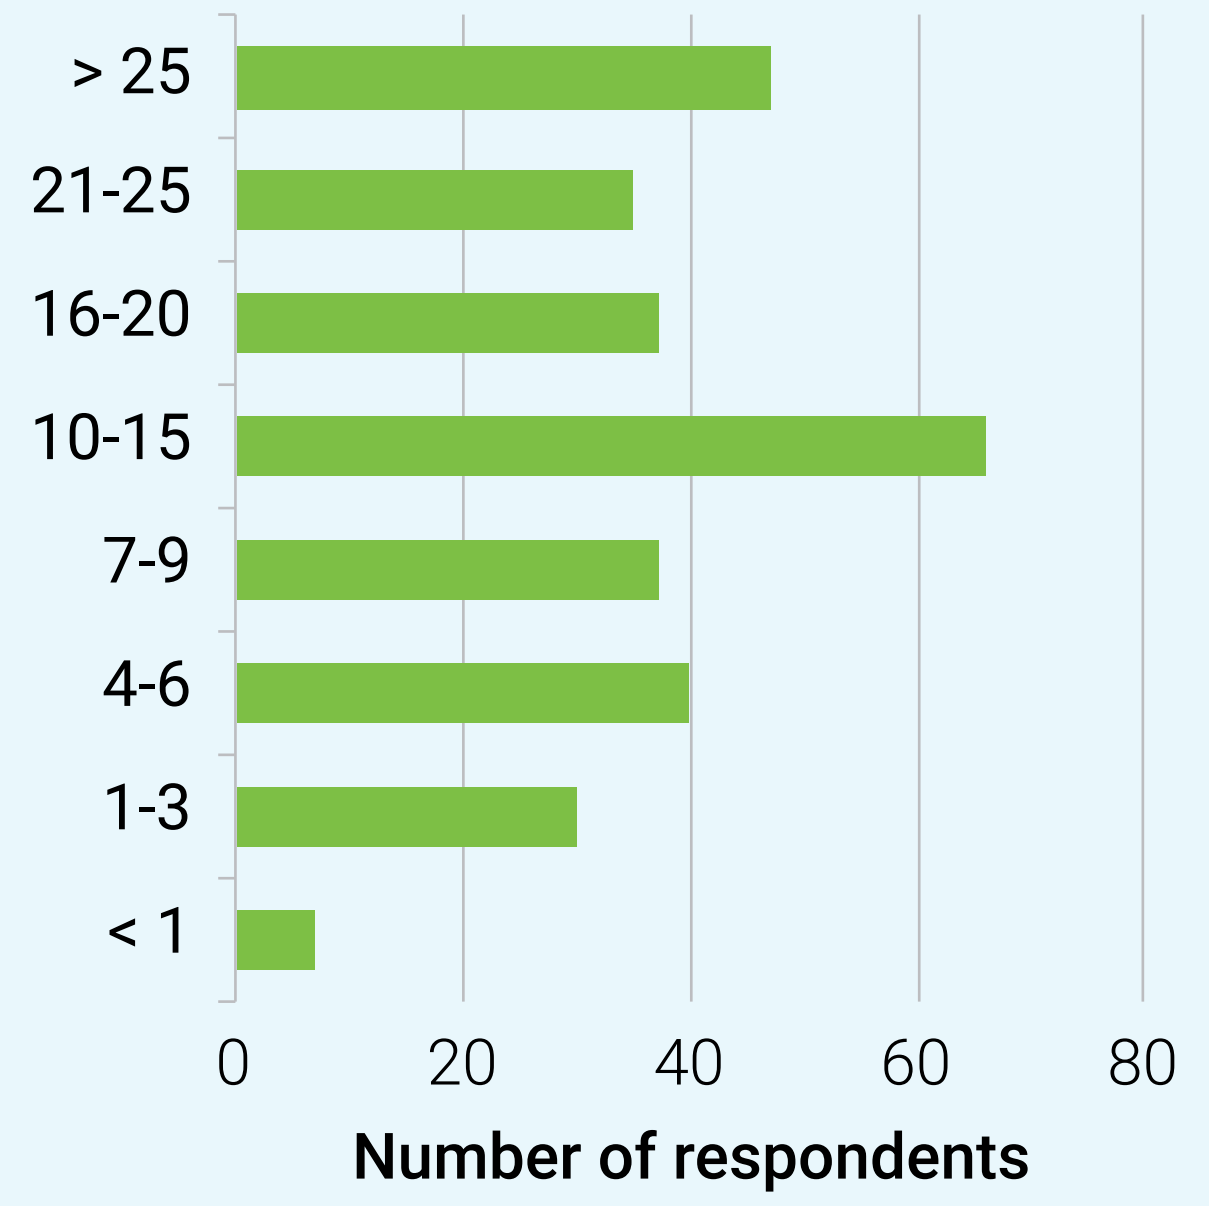
> 25
21-25
16-20
10-15
7-9
4-6
1-3
< 1
0
20
40
60
80
Number of respondents

Summary of Respondent's Vaccine Choices (N = 299)

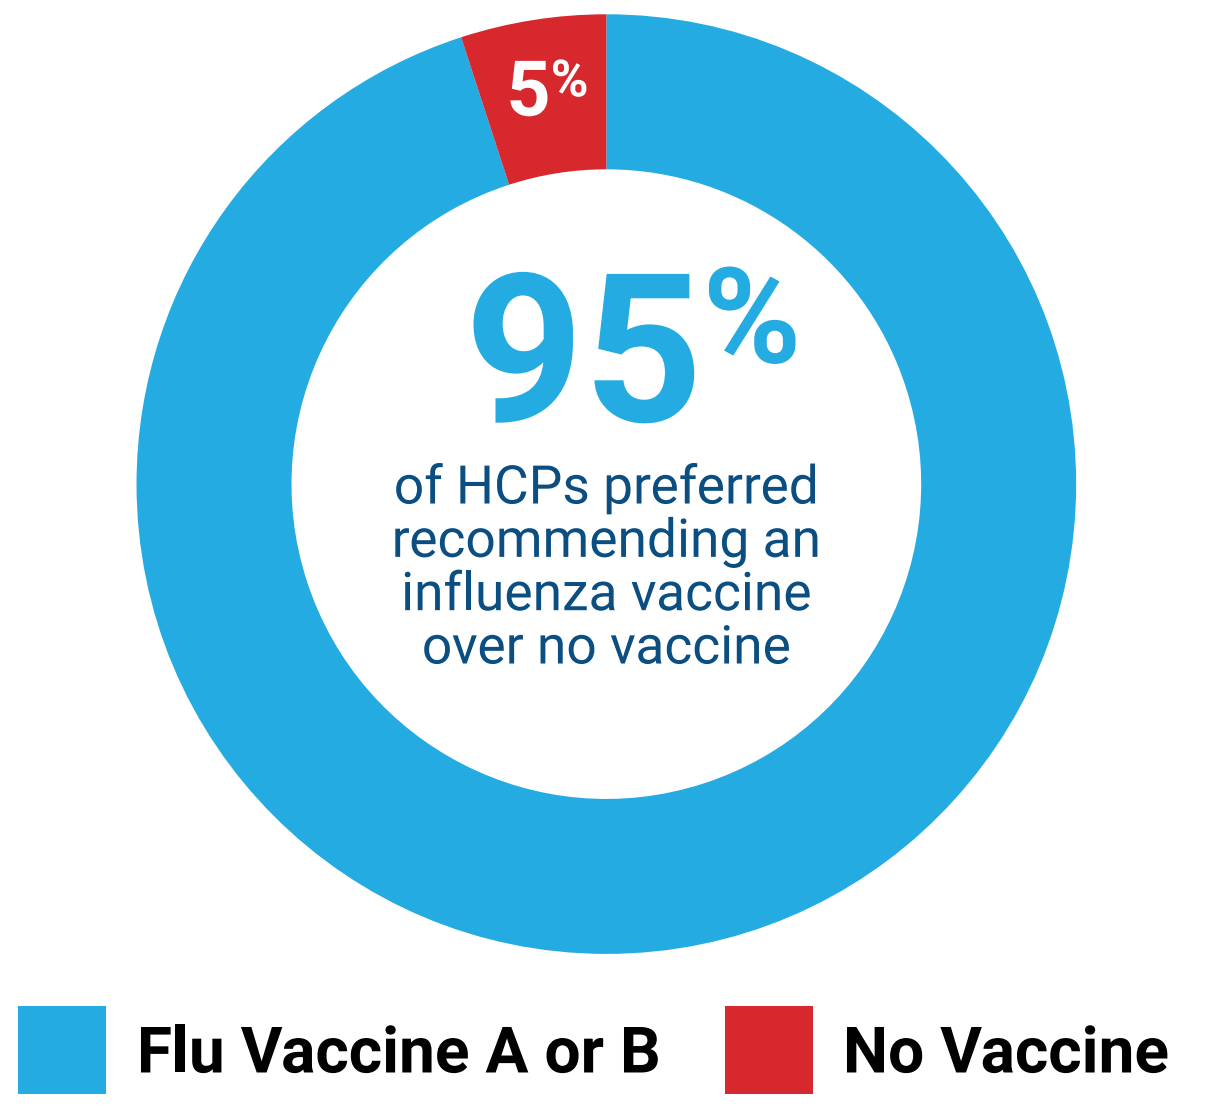
5%
95%
of HCPs preferred recommending an influenza vaccine over no vaccine
Flu Vaccine A or B
No Vaccine

Note: The percentages in this graph are based on all of the discrete-choice experiment questions (i.e., 2391 questions) answered by respondents in the sample.

- HCPs had a strong preference for recommending an influenza vaccine over no vaccine. HCPs placed greatest importance on avoiding risk of moderate to severe systemic side effects (CRAI, 39.8%) and increasing vaccine efficacy (CRAI, 37.4%), followed by avoiding moderate to severe injection site reaction risk (CRAI, 14.1%) (**Figure 2**). Durability of protection (CRAI, 5.1%) and hospitalization prevention (CRAI, 3.5%) were least important.
- **Table 2** presents the MARs that HCPs were willing to accept in exchange for selected improvements in vaccine efficacy.

Figure 2. Conditional Relative Attribute Importance (N = 299)

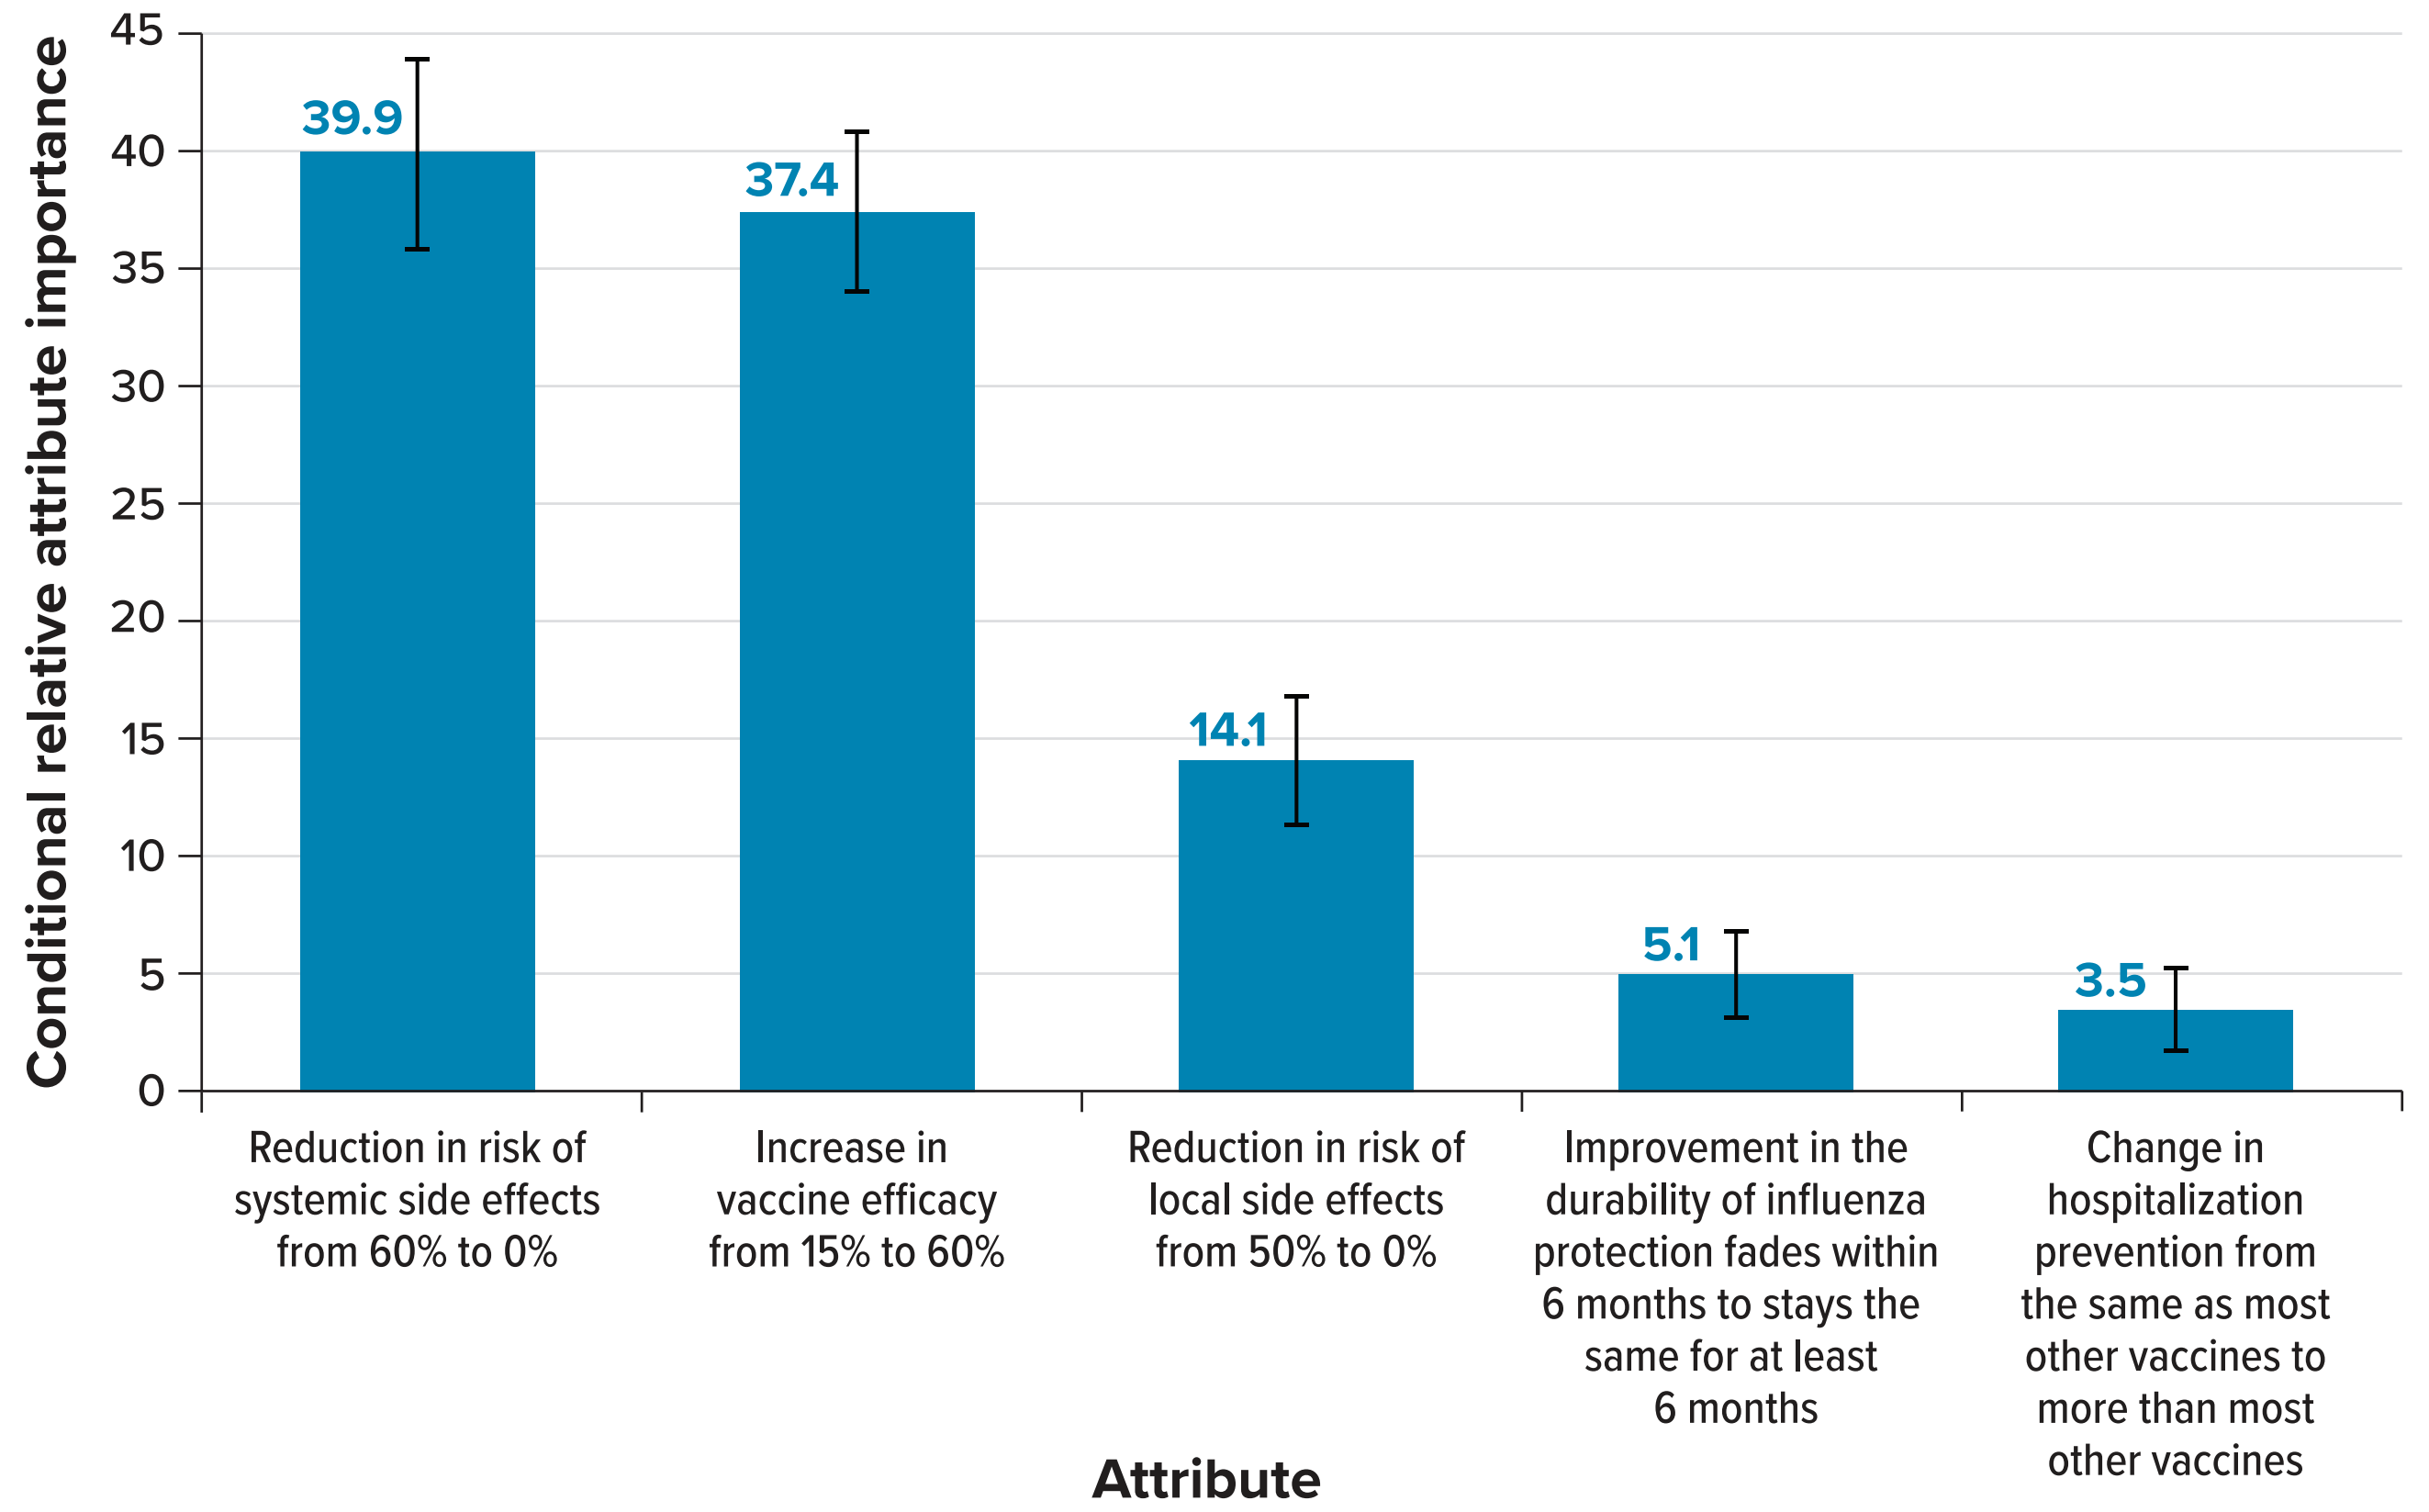
Conditional relative attribute importance
45
40
35
30
25
20
15
10
5
0
39.9
37.4
14.1
5.1
3.5
Reduction in risk of systemic side effects from 60% to 0%
Increase in vaccine efficacy from 15% to 60%
Reduction in risk of local side effects from 50% to 0%
Improvement in the durability of influenza protection fades within 6 months to stays the same for at least 6 months
Change in hospitalization prevention from the same as most other vaccines to more than most other vaccines
Attribute

Table 2. Maximum Acceptable Risk of Injection Site and Flu-Like reaction (N = 299)

Improvement in vaccine efficacy: From this level	Improvement in vaccine efficacy: To this level	Mean MAR of injection site reaction	Mean MAR of systemic side effects
15%	25%	44.7%	21.0%
10% improvement			
50%	60%	38.9%	17.7%
10% improvement			
25%	50%	> 50%	26.8%
25% improvement			
15%	50%	> 50%	49.1%
35% improvement			
25%	60%	> 50%	44.0%
35% improvement			
15%	60%	> 50%	57.9%
45% improvement			

Note: Estimates of MAR were not extrapolated outside the risk range presented in the survey. Means that are reported as > 50% represent MAR estimates greater than 50%, which was the maximum level of risk of injection site reaction presented in the study.

CONCLUSIONS

- In this study, US HCPs strongly preferred to recommend an influenza vaccine for their patients to no vaccine, and their hypothetical vaccine choices were driven by higher vaccine efficacy and lower risks of systemic side effects. The risks of injection site reactions, vaccine durability, and hospitalization prevention had less influence on vaccine choices.
- US HCPs were more tolerant of injection site reactions than systemic side effects in exchange for improved vaccine efficacy.

References

1. Poulos C et al. Value Health. 2023;26(6):S347.
2. McMichael A et al. Value Health. 2023;26(6):S329-S330
3. Poulos C et al. Value Health. 2020; 23(1): S320.

Acknowledgments

Christine Poulos, Alan McMichael, and Cannon Kent are paid employees of RTI HS. Deborah Rudin, Philip Buck, Parinaz Ghaswalla, and Darshan Mehta are paid employees and own stocks/shares of Moderna, Inc.

Disclosures

This study was funded by Moderna, Inc.

Copies of this presentation obtained through QR codes are for personal use only and may not be reproduced without permission of the authors.

Presented at: ISPOR 2024; 5-8 May 2024; Atlanta, GA, United States