EE373

The Economic Impact of Adrenomyeloneuropathy (AMN) on Direct Medical Costs in Commercially Insured US Adults: A Retrospective Study

Josh Bonkowsky[1,2], Bridget Healey[3], Naomi C. Sacks[3], Ronaé K. McLin[3], Philip Cyr[3], Eileen Sawyer[4], Christopher D. Stephen[5,6], Florian Eichler[5,6]

[1]Primary Children's Hospital, Intermountain Healthcare, Salt Lake City, UT, [2]Division of Pediatric Neurology, Department of Pediatrics, University of Utah School of Medicine, Salt Lake City, UT,[3]Heath Economics and Outcomes Research, PRECISIONheor, Boston, MA, [4]Clinical Medical Affairs, SwanBio Therapeutics, Boston, MA, [5] Department of Neurology, Massachusetts General Hospital, Boston, MA,[6]Harvard Medical School, Boston, MA

SwanBio THERAPEUTICS

INTRODUCTION

- X-linked adrenoleukodystrophy (X-ALD) is a genetic, metabolic condition with an incidence of 1:16,800 caused by a *ABCD1* gene mutation.[1,2]
- Males with the mutation develop adrenomyeloneuropathy (AMN) in adulthood. Symptom onset is typically observed around the late 20s to 30s. Women may also develop symptoms, though onset is observed later in life.[3-5]
- AMN is neurodegenerative – characterized by progressive myeloneuropathy that causes spastic paraparesis, sensory ataxia, incontinence, and sexual dysfunction. Eventually walking is severely affected and mobility is impaired.[3,6-8]
- There are currently no effective treatments in stabilizing or reversing the progression of AMN. Moreover, suggested and practiced care varies based on the symptoms experienced by each person.[9]
- AMN's impact on healthcare resource utilization (HRU) and mortality is unknown.

Objective

- This goal of this study was to quantify for healthcare costs for commercially insured male and female adults with AMN in the US.

Methods

- Direct medical costs were assessed using IQVIA's PharMetrics Plus commercial claims database (1/01/2006-6/30/2021), with nationally representative demographic, enrollment and claims data.
- Study subjects were individuals ages 18-64y with ≥1 inpatient or ≥2 outpatient claims containing an AMN or X-ALD diagnosis (ICD-10-CM: E71.52x; ICD-9-CM 277.86), no evidence of childhood cerebral adrenoleukodystrophy or other peroxisomal disorders and no pregnancy or labor and delivery.
- AMN study patients were 1:4 propensity score matched to non-AMN controls on demographic characteristics and enrollment window.
- Patients and controls were followed for as long as they were observable in the claims data.
- Costs paid to providers by payers were estimated per patient per month (PPPM) and annualized to represent costs per patient per year (PPPY).

Results

- A total of 806 individuals with evidence of AMN (Males:303; Females: 503) were matched to 2,726 (Males: 1,037; Females: 1,689) similar in age, sex, geography, and length of enrollment.
- Most individuals with AMN were between ages 18-35y, mean age 35.1y ±13.81 (males) and 35.0 ±11.56 (females).
- Mean follow-up in males was 29.0 ±27.69 months and female 18.9±19.74 months.
- Mean Charlson Comorbidity Index (CCI) scores were 0.67 ±0.94 (males) and 0.34 ±1.33 (females).

Figure 1: Annual All-cause Healthcare Costs AMN vs. Controls, By Sex

Figure 1A: Males

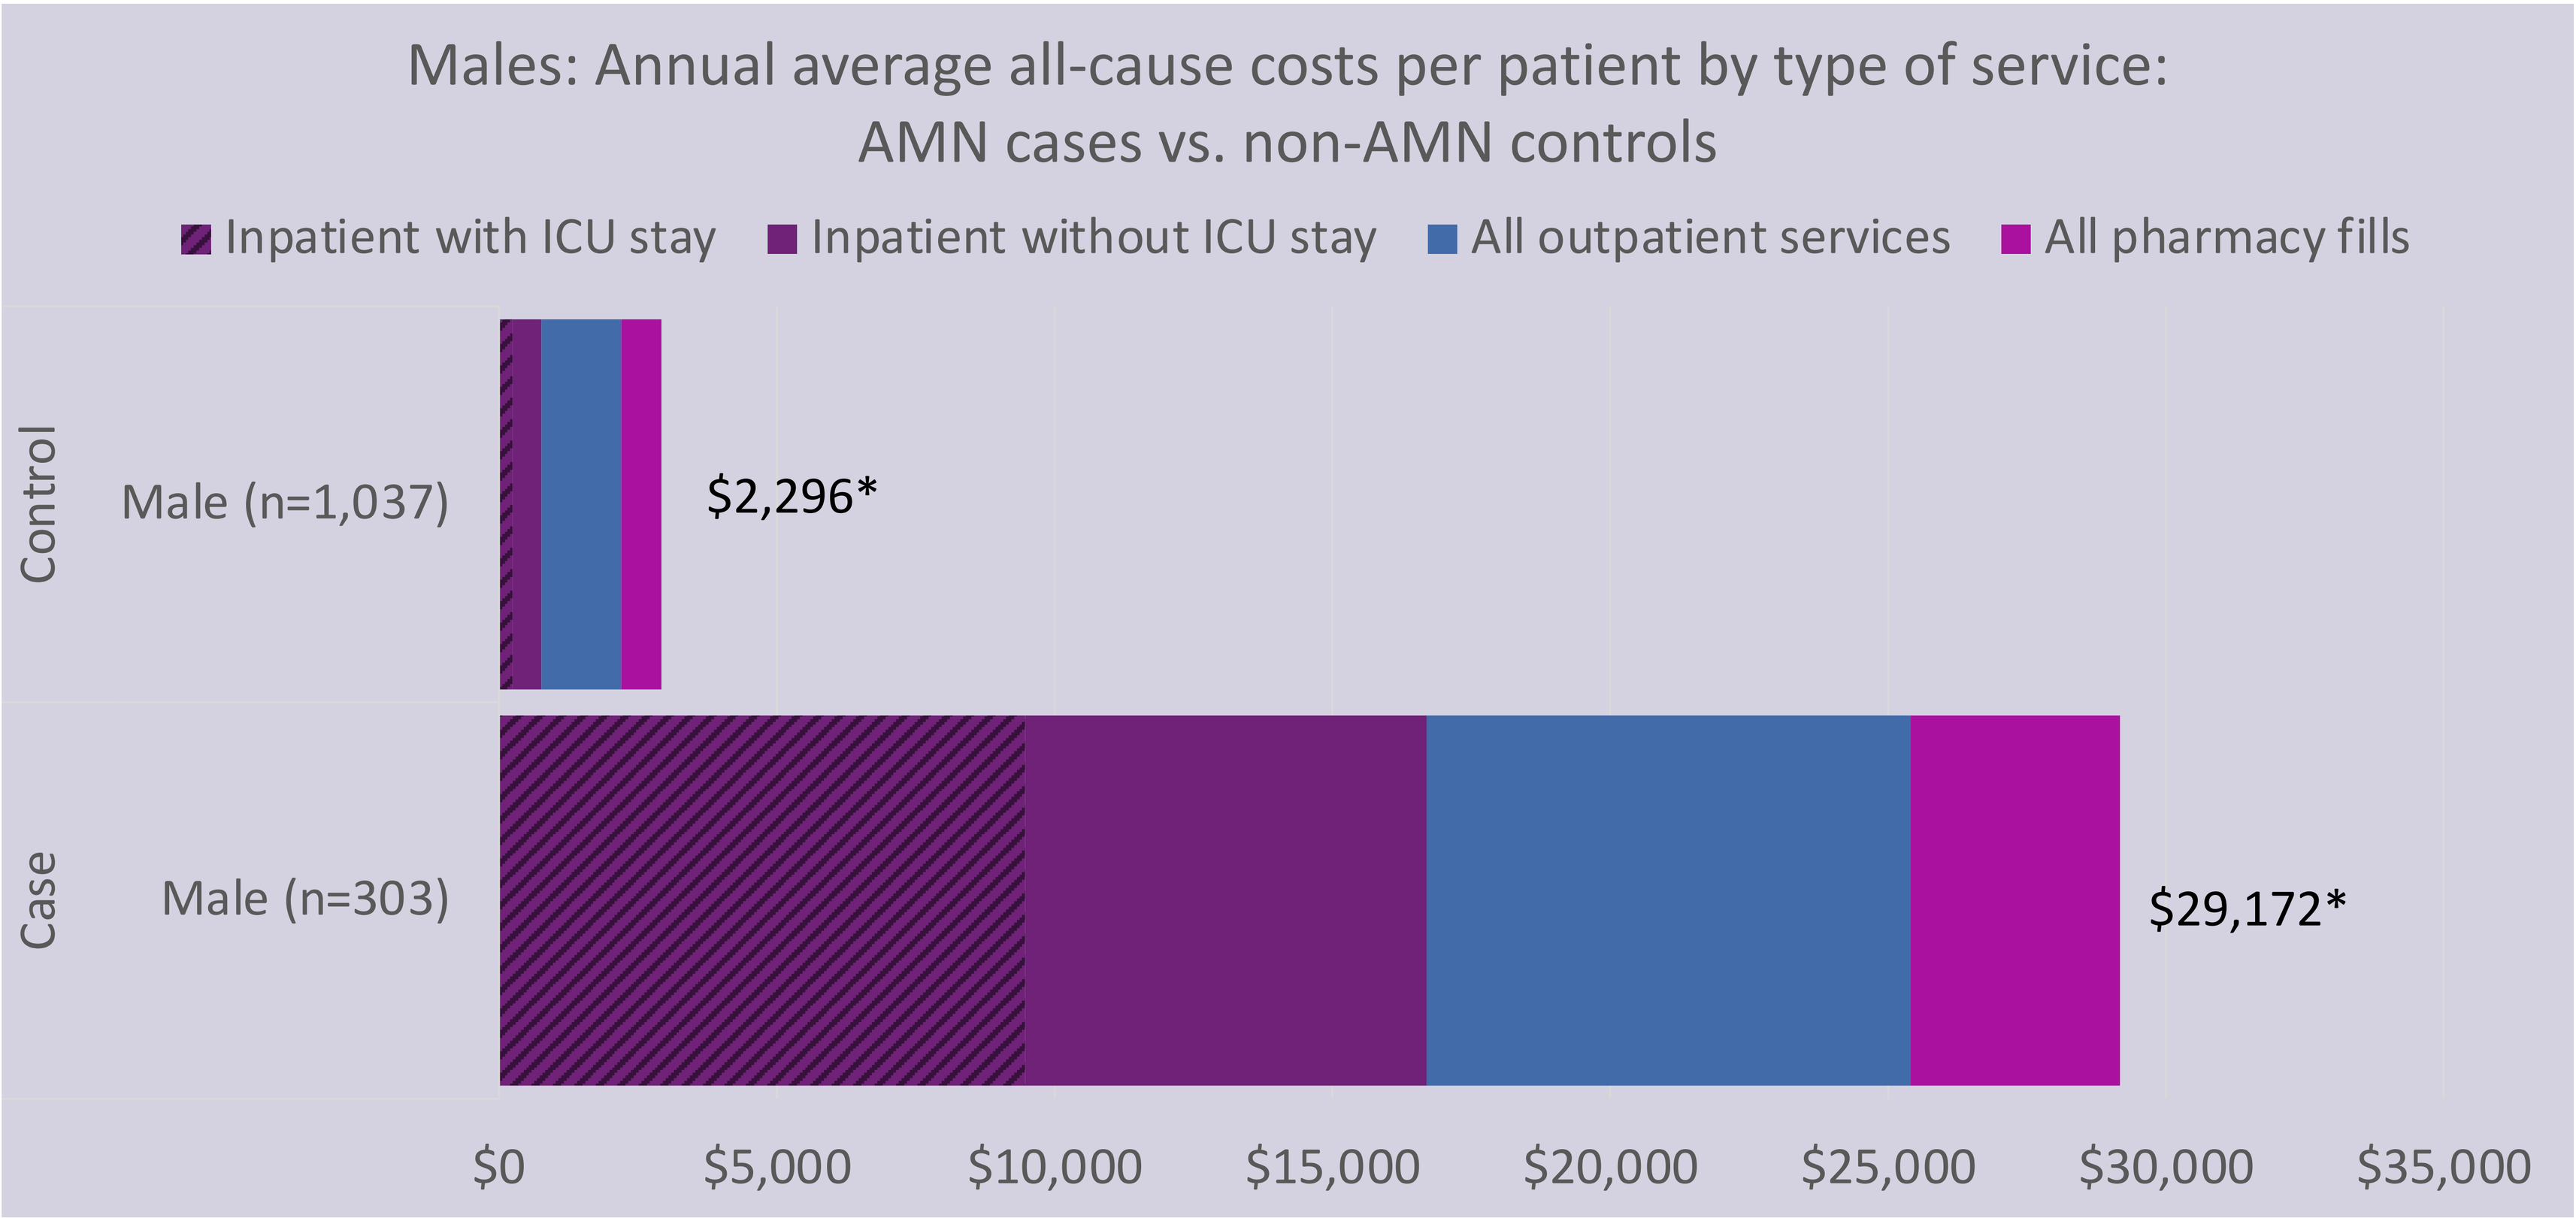
Males: Annual average all-cause costs per patient by type of service:
AMN cases vs. non-AMN controls
Inpatient with ICU stay
Inpatient without ICU stay
All outpatient services
All pharmacy fills
Control
Male (n=1,037)
$2,296*
Case
Male (n=303)
$29,172*
$0
$5,000
$10,000
$15,000
$20,000
$25,000
$30,000
$35,000

Figure 1B: Females

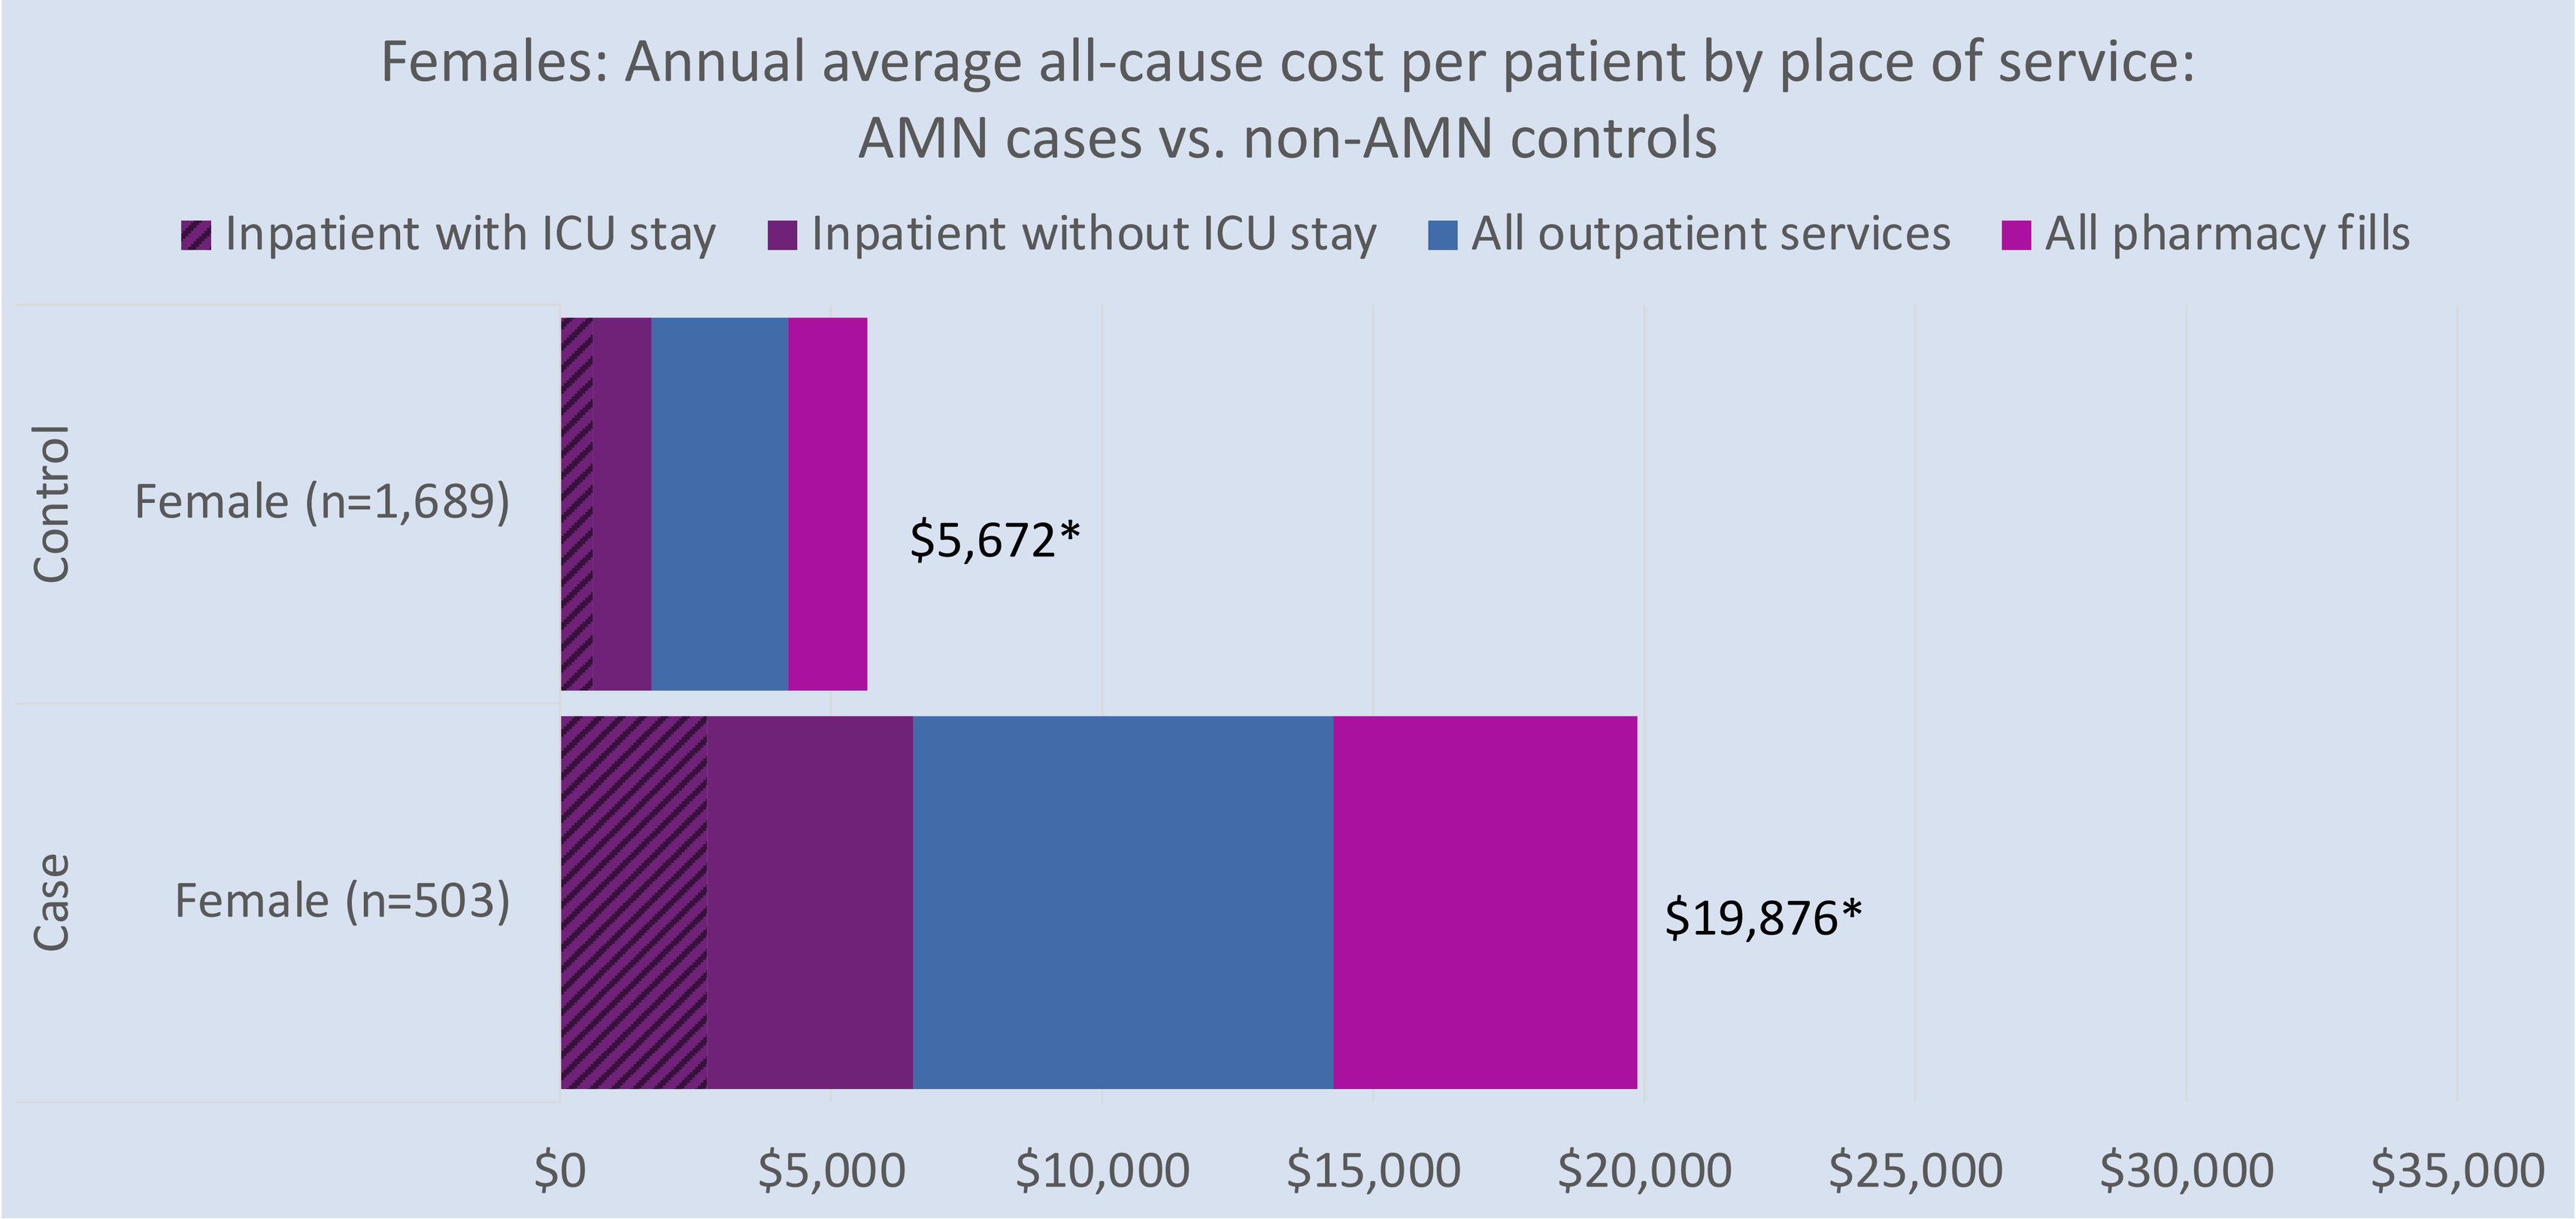
Females: Annual average all-cause cost per patient by place of service:
AMN cases vs. non-AMN controls
Inpatient with ICU stay
Inpatient without ICU stay
All outpatient services
All pharmacy fills
Control
Female (n=1,689)
$5,672*
Case
Female (n=503)
$19,876*
$0
$5,000
$10,000
$15,000
$20,000
$25,000
$30,000
$35,000

*Significantly different at $p<0.05$

- Across all AMN patients, mean all-cause healthcare monthly per patient costs paid by payers were $2,028±21,491, considerably higher than controls ($369±3,931), *p* <0.05
 - Inpatient admissions accounted for nearly half of all monthly per patient costs ($950±21,017, 47%), with outpatient services ($684±2,876; 34%) and prescription medications ($394±$1,850; 19%) constituting the remainder.
- Annualized costs for both males (n=303) and females (n=503) with AMN were substantially greater than costs for male and female controls.
 - All-cause annual average total costs were $26,247 higher per patient among males with AMN vs. male controls, *p* <0.05 (**Figure 1A**).
 - All-cause annual average total costs were $14,205 higher per patient among the females with AMN vs. female controls, *p* < 0.05 (**Figure 1B**).
 - Costs for inpatient admissions, outpatient services, and pharmacy fills were significantly higher for male and female AMN patients vs. controls, *p* < 0.05 (**Figures 1A and 1B**).
 - Annual hospitalization rates were much higher for individuals with AMN vs. controls, with the largest differences in male patients compared with controls (**Figure 2**).

Figure 2: Annual average hospitalizations PPPY: AMN vs. Controls

Figure 2A: Males

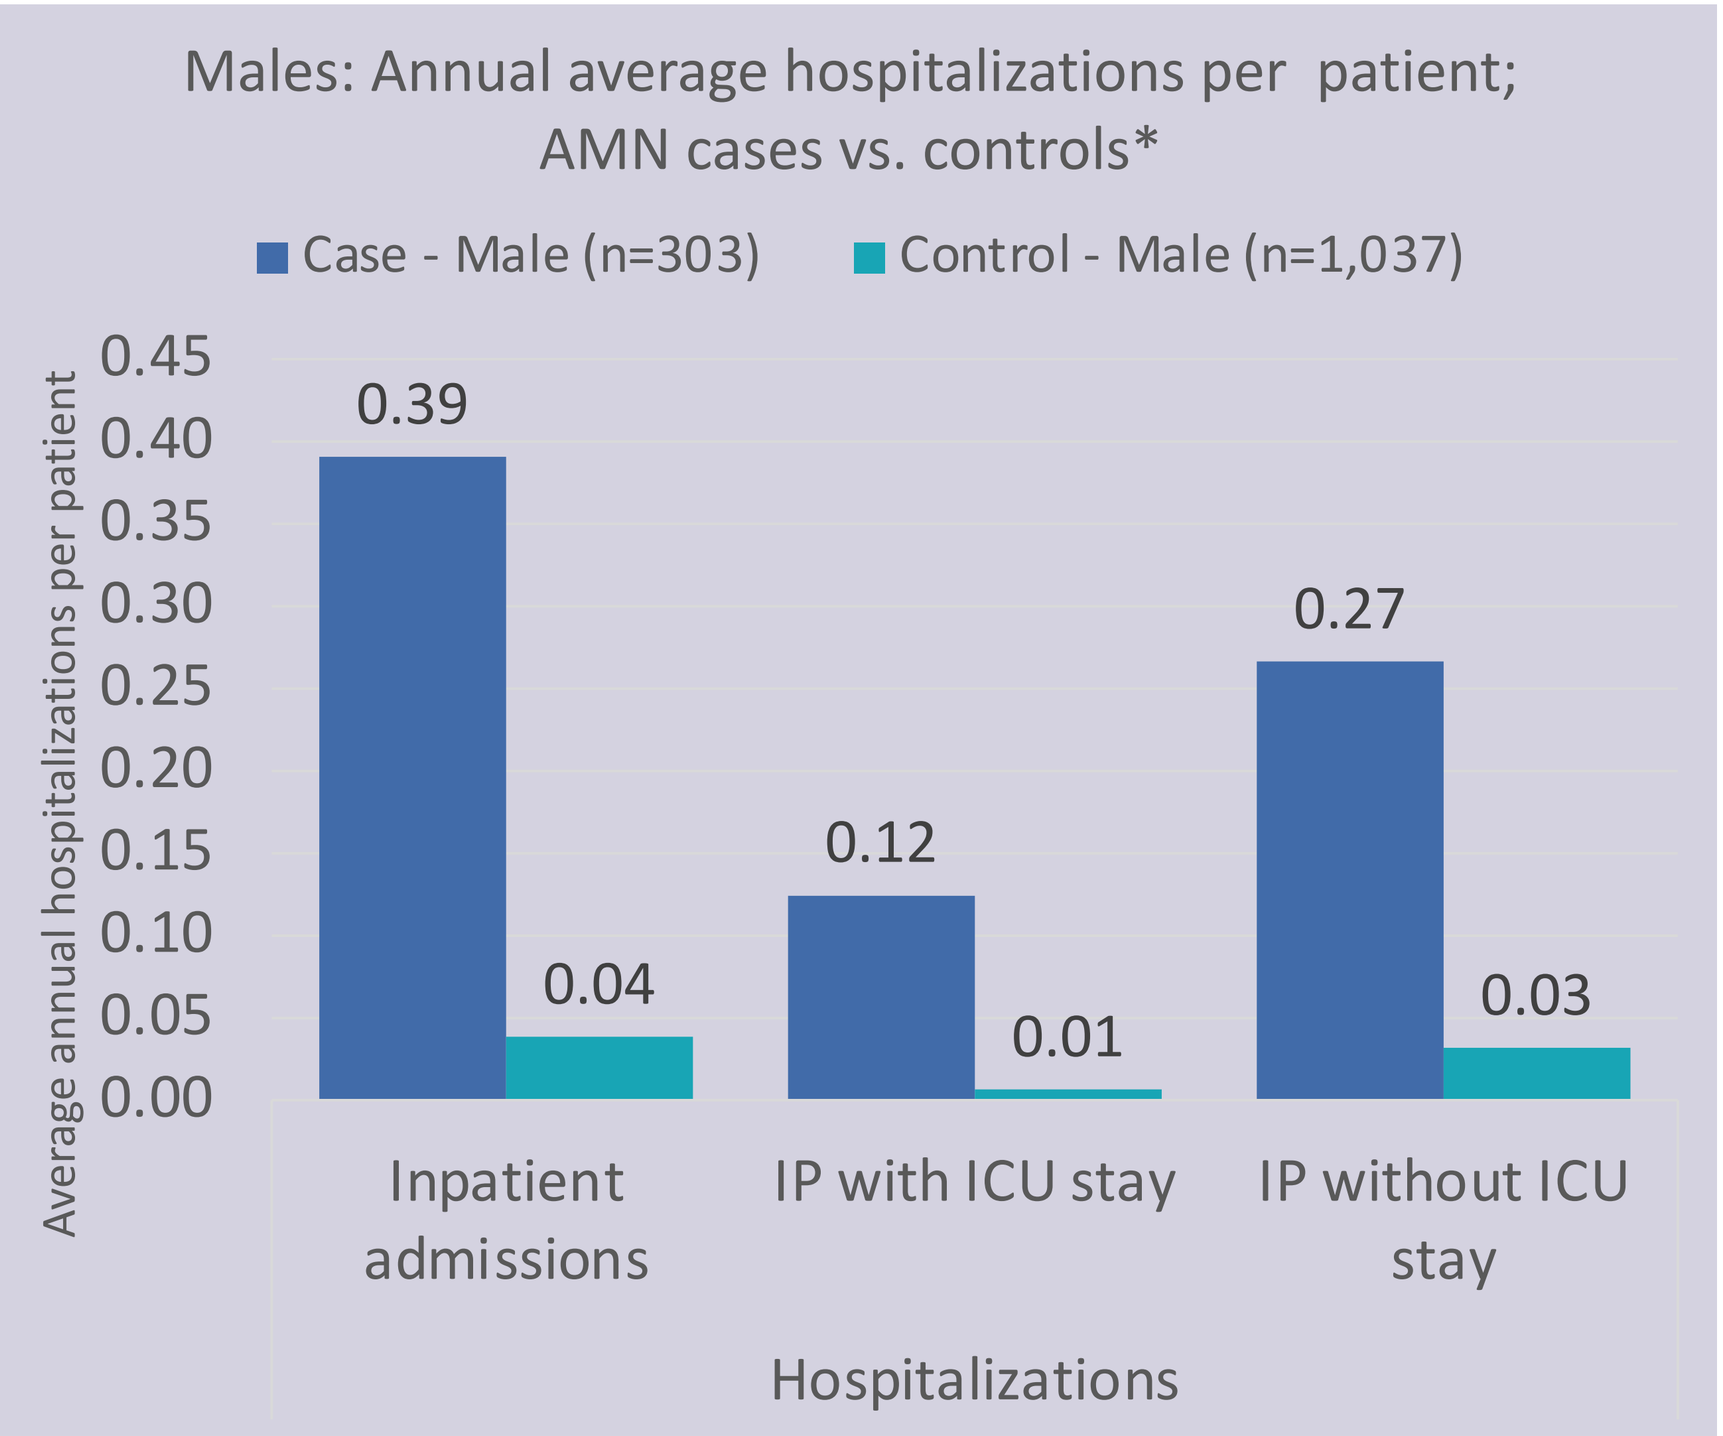
Males: Annual average hospitalizations per patient;
AMN cases vs. controls*
Case - Male (n=303)
Control - Male (n=1,037)
Average annual hospitalizations per patient
0.45
0.40
0.35
0.30
0.25
0.20
0.15
0.10
0.05
0.00
0.39
0.04
0.12
0.01
0.27
0.03
Inpatient admissions
IP with ICU stay
IP without ICU stay
Hospitalizations

*Significantly different at $p<0.05$

Figure 2B: Females

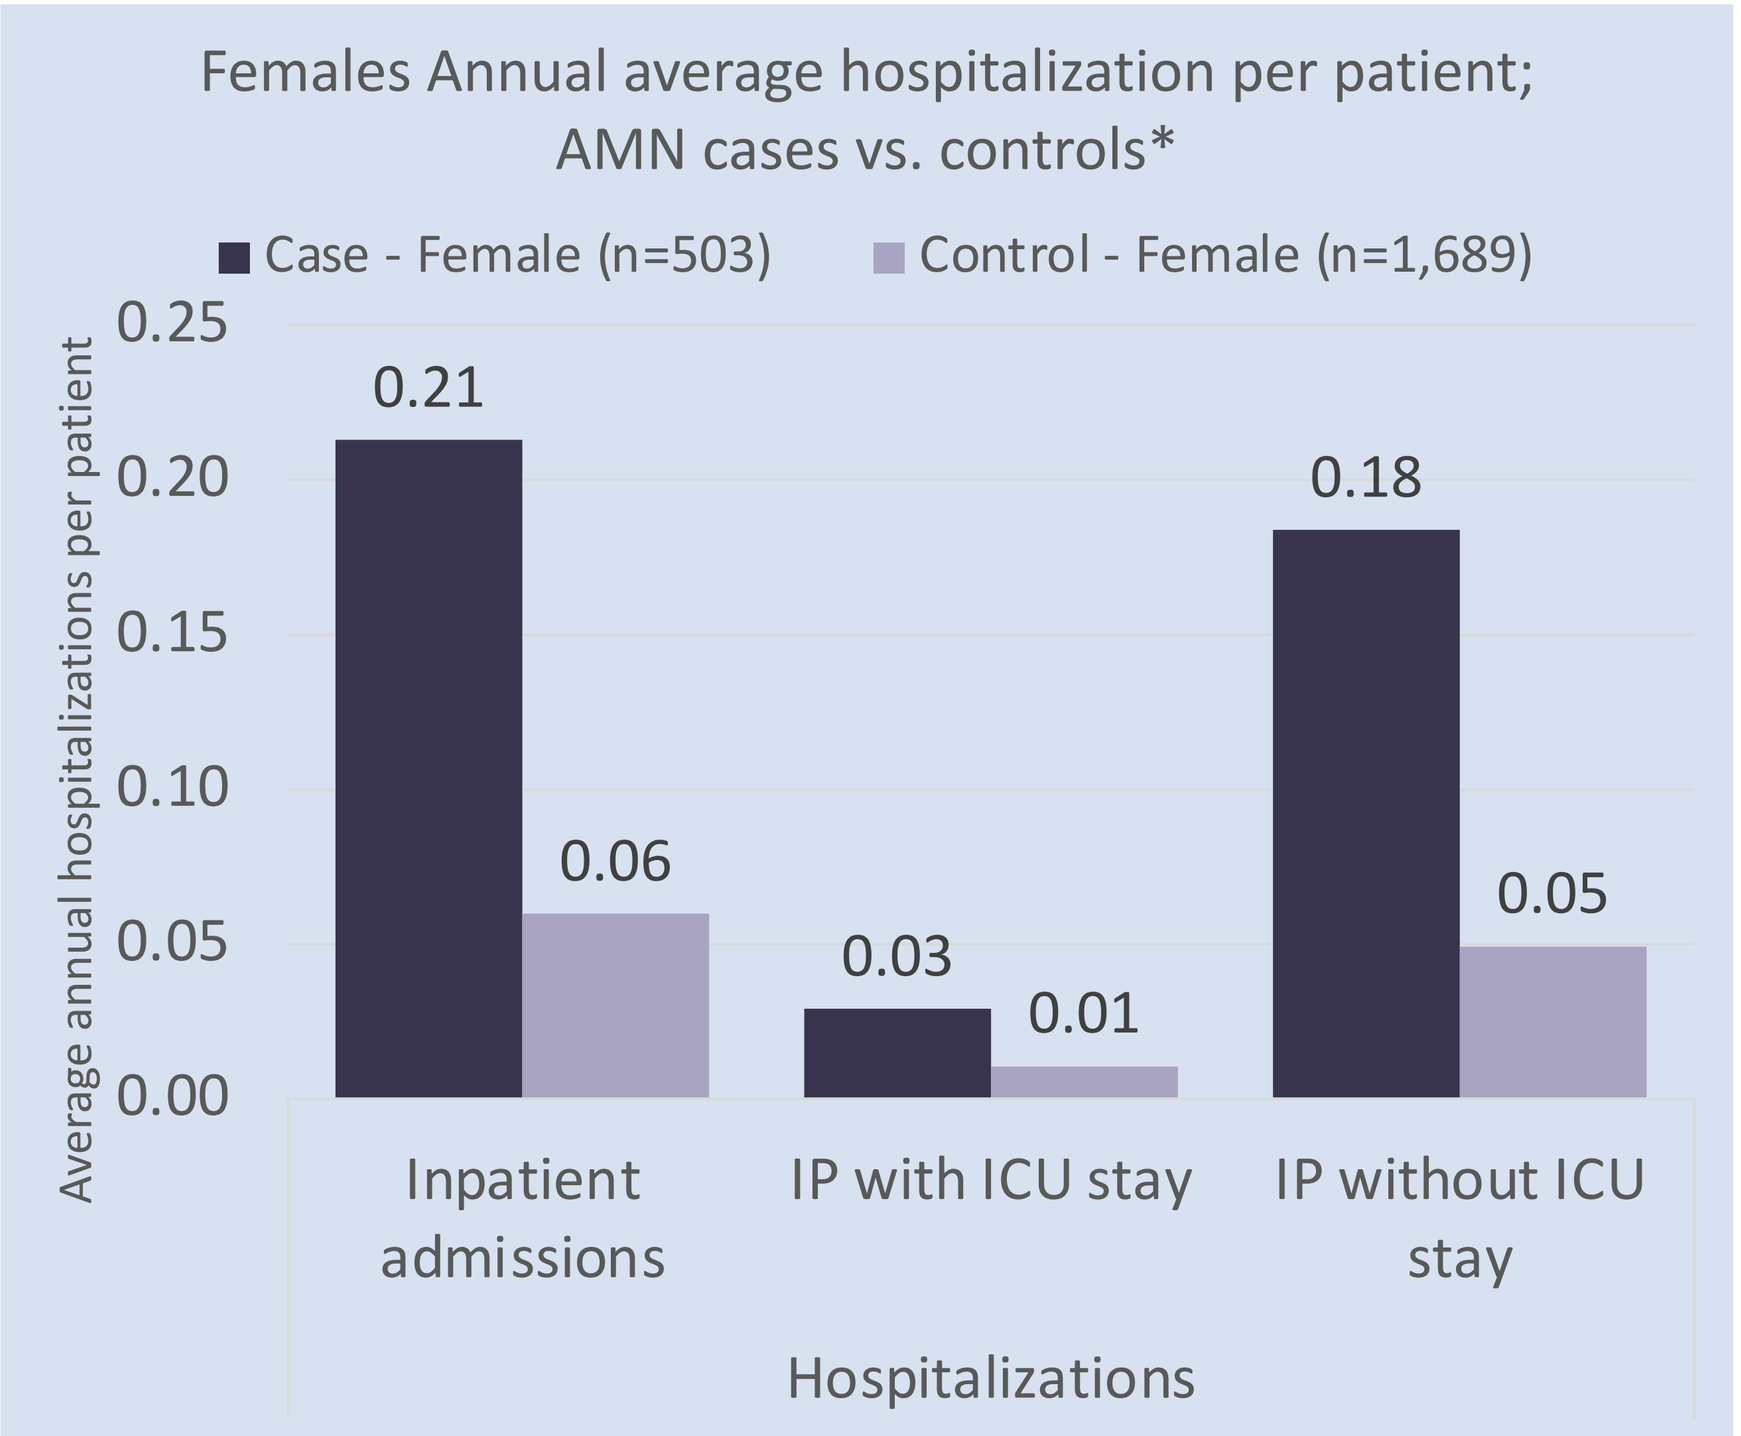
Females Annual average hospitalization per patient;
AMN cases vs. controls*
Case - Female (n=503)
Control - Female (n=1,689)
Average annual hospitalizations per patient
0.25
0.20
0.15
0.10
0.05
0.00
0.21
0.06
0.03
0.01
0.18
0.05
Inpatient admissions
IP with ICU stay
IP without ICU stay
Hospitalizations

*Significantly different at $p<0.05$

CONCLUSIONS

- All-cause direct medical costs for commercially insured adult men and women aged 18-64 years old with AMN are much higher than adults without AMN, particularly for male patients.
- Higher costs for AMN patients suggest greater disease-related healthcare issues for patients than has been previously recognized and constitute a substantial overall financial and healthcare burden.
- These costs do not include indirect, societal, or patient paid costs and thus do not represent a full cost assessment for patients with AMN.
- The increased costs for both men and women with AMN suggest a need for improved care options.

References: 1. Bezman L, Moser AB, Raymond GV, et al. Adrenoleukodystrophy: incidence, new mutation rate, and results of extended family screening. Ann Neurol. Apr 2001;49(4):512-7 2. Resende LL, de Paiva ARB, Kok F, da Costa Leite C, Lucato LT. Adult Leukodystrophies: A Step-by-Step Diagnostic Approach. Radiographics. Jan-Feb 2019;39(1):153-168. doi:10.1148/rg.2019180081 3. Engelen M, Kemp S, de Visser M, et al. X-linked adrenoleukodystrophy (X-ALD): clinical presentation and guidelines for diagnosis, follow-up and management. Orphanet J Rare Dis. Aug 13 2012;7:51. doi:10.1186/1750-1172-7-51 4. Kanakis G, Kaltsas G. Adrenal Insufficiency Due to X-Linked Adrenoleukodystrophy. [Updated 2018 Oct 12]. In: Feingold KR, Anawalt B, Blackman MR, et al., editors. Endotext [Internet]. 5. Zhu, J., Eichler, F., Biffi, A., Duncan, C. N., Williams, D. A., & Majzoub, J. A. (2020). The Changing Face of Adrenoleukodystrophy. Endocr Rev, 41(4), 577-593. https://doi.org/10.1210/endrev/bnaa013 6. Zhang YZ, G.: Chen, W.: Pu, Z.: Song, L.: Tang, X.: Liu, Z. A novel ABCD1 G1202A mutation in a Chinese patient with pure adrenomyeloneuropathy and literature review. Genes Dis. Sep 2021;8(5):709-714. doi:10.1016/j.gendis.2020.01.009 7. Dato C, Capaldo G, Terracciano C, et al. Late adult-onset adrenomyeloneuropathy evolving with atypical severe frontal lobe syndrome: Importance of neuroimaging. Radiol Case Rep. Mar 2019;14(3):309-314. doi:10.1016/j.radcr.2018.11.007 8. Kemp S, Berger J, Aubourg P. X-linked adrenoleukodystrophy: clinical, metabolic, genetic and pathophysiological aspects. Biochim Biophys Acta. Sep 2012;1822(9):1465-74. doi:10.1016/j.bbadis.2012.03.012 9. Eichler F, Duncan C, Musolino PL, et al. Hematopoietic Stem-Cell Gene Therapy for Cerebral Adrenoleukodystrophy. N Engl J Med. 2017;377(17):1630-1638.

Author contributions: FE, JB, BH, NS, RM, PC, ES, and CS contributed to the design and implementation of the study, and to the interpretation of results.

Disclosures: JB has received consultant fees from Bluebird bio, Neurogene, Passage Bio, Takeda, and Autobahn; owns stock in Orchard; received research support from NIH. RM, BH, NS, and PC received employee salary from PrecisionHEOR. ES received employee salary from and owns stock in uniQure Inc; received research support from Sanofi and NIH. CS has received consultant fees from SwanBio; received research support from Sanofi. FE has received consultant fees from SwanBio, Alnylam, Origin Biosciences, Orchard, Autobahn, Bluebird; received fees from UpToDate; received research support from Bluebird, Minoryx, and Sio; owns stock in SwanBio.

Poster presented at ISPOR 2023; Boston, MA, May 9, 2023

© 2023 SwanBio Therapeutics, Inc. All rights reserved

This study was sponsored by SwanBio Therapeutics Ltd