Patient-Centric Care in Higher-Risk Myelodysplastic Syndromes: A Global Systematic Literature Review and Gap Analysis

PCR123

Edward P. Armstrong,[1] Duska Franic,[2] Daniel C. Malone,[1] Sissi Pham,[2] Cosmina Hogea[3]

[1]Strategic Therapeutics, Tucson, AZ, USA; [2]AESARA, Chapel Hill, NC, USA; [3]Patient Engagement, Global Medical Strategic Operations, Gilead Sciences, Inc., Foster City, CA, USA

Scan for a copy of the poster and full reference list

Conclusions

More studies and programs need to be conducted to advance patient-centric care (PCC) in patients with HR-MDS. PCC can help patients make decisions, and implement behavioral changes that result in improved satisfaction, better outcomes, and a lower cost of care[1,2]

Patient-lived experiences can be captured with patient-reported outcome (PRO) and patient-reported experience measures (PREMs),[3] however HR-MDS targeted measures were lacking

As new therapies for HR-MDS emerge, it is critical to actively engage with patients in designing evidence-based solutions that can empower them to interact and engage within the healthcare ecosystem and elucidate outcomes/experiences that are meaningful to them

Call to Action

Critical to customize currently available MDS PREMs for HR-MDS to include salient factors such as personal treatment goals, satisfaction, and maintenance of autonomy.[4,5] *It is essential to quantify and communicate patient preferences for treatment in a real-world setting, promote shared decision-making (SDM), and thereby improve the HR-MDS patient care experience*[4,6,7,8]

Actively engage patients in the validation of the available HR-MDS PROs to ensure the inclusion of outcomes that are meaningful to patients with HR-MDS. *The inclusion of the patients' voice to the same level as objectively measured data*[3] *in a real-world setting requires the uptake of validated HR-MDS PROs to promote SDM and facilitate PCC*

Customization of fit-for-purpose HR-MDS educational materials to support and activate providers, patients, and caregivers. *Patients/caregivers are seeking unvalidated sources. However, an understanding of HR-MDS, how it is being treated, and what happens if treatment is delayed remains important to reduce anxiety and confusion, and improve emotional well-being and engagement of patients/caregivers; thereby, promoting shared decision-making (SDM) and improving PCC*[5,6]

Develop standardized best practices and increase evidence-based evaluation tools for HR-MDS.[5,9] *Patient experience and outcomes can be improved with more efficient HR-MDS diagnosis, referral, and treatment frameworks*[10]

References: **1.** Bosworth HB, et al. *J Participat Med*. 2017;9:e7. **2.** Schuster ALR, et al. *Patient*. 2023;16(2):89-93. **3.** Wu Z, et al. *Diabetes Metab*. 2023; 49(2):101417. **4.** Messina AJ, et al. *Blood*. 2021;138(suppl 1):4982. **5.** Wintrich S, et al. *Br J Haematol*. 2019;185:48. **6.** Booth A, et al. *J Med Internet Res*. 2019;21(11):e14285. **7.** Lynch KA, et al. *BMC Palliat Care*. 2022;21(1):29. **8.** Poynton M, et al. *HemaSphere*. 2022;6:3032-3033. **9.** Kasprzak A, et al. *Int J Environ Res Public Health*. 2021;18(14):7629. **10.** Becker MN, et al. *Blood*. 2021;138(suppl 1):1944. **11.** Greenberg P, et al. *Blood*. 1997;89(6):2079-2088. **12.** Vallow S, et al. *Blood*. 2021;138(suppl 1):4654. **13.** Rochau U, et al. *Br J Haematol*. 2020;191(3):405-417. **14.** Troy JD, et al. *J Natl Compr Canc Netw*. 2018;16(3):267-273. **15.** Tinsley-Vance S, Johnson N. *Leuk Res*. 2021;108(suppl):S1-S60. **16.** Efficace F, et al. *Cancer*. 2018;124(6):1251-1259. **17.** Amitai I, et al. *Br J Haematol*. 2021;194(2):319-324. **18.** Wan BA, et al. *Blood*. 2019;134:4245. **19.** King D, et al. *Leuk Res*. 2020;96:106425. **20.** Vijenthira A, et al. *Transfusion*. 2022;62(7):1355-1364. **21.** Horodniceanu EG, et al. *Health Qual Life Outcomes*. 2017;15(1):129. **22.** Zeidan AM, et al. *Clin Lymphoma Myeloma Leuk*. 2022;22(3):e185-e198. **23.** Abuhadra N, Nazha A. *Curr Hematol Malig Rep*. 2018;13(6):570-575. **24.** Lauritsen TB, et al. *Hematol Oncol*. 2022;40(5):1056-1066.

Acknowledgments: This study was funded by Gilead Sciences, Inc., Foster City, CA, USA. Editorial and design support were provided by Michele Salernitano and Shannon Begley of Ashfield MedComms, an Inizio company, and funded by Gilead Sciences, Inc.

Disclosures: Drs Malone, Armstrong, Pham, and Franic are consultants to Gilead Sciences, Inc.

Correspondence: ed.armstrong@strategictherapeutics.com

Introduction

- Myelodysplastic syndromes (MDS) is a group of hematologic disorders characterized by abnormal proliferation of bone marrow cells
- MDS can be further categorized into lower-risk (LR)- or higher-risk (HR)-MDS. Median survival of LR-MDS and HR-MDS are 3.5 to 5.7 and 0.4 to 1.2 years, respectively, if only treated with supportive care[11]
- HR-MDS is more aggressive than LR-MDS and likely to progress to acute myeloid leukemia (AML), warranting urgent treatment and therefore diagnosis
- Patient-centric, evidence-based solutions in conjunction with emerging therapies can help advance overall care and improve outcomes
- This is the first global systematic literature review (SLR) utilizing real-world data about HR-MDS to provide calls to action to improve PCC

Objective

- The objective of this study was to evaluate PCC status in the treatment of HR-MDS, and identify opportunities to develop solutions to address care gaps for an optimal patient care experience

Methods

- A global SLR was conducted by cross-referencing MDS/HR-MDS terms with PCC terms (**Table 1**), using PubMed, Embase, and Cochrane Collaboration databases (2017-2022), in accordance with Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines

Table 1: Article Inclusion/Exclusion Criteria

Characteristic	Inclusion	Exclusion
Patient Population	HR-MDS or MDS	LR-MDS only, AML only, non-MDS
Outcomes	**PCC, Clinical Outcomes, COAs:** Terms included, but were not limited to, patient experience, patient engagement, patient journey (including barriers or facilitators to diagnosis and treatment), patient-centric evidence, patient education, patient-centered outcomes, shared decision-making, patient activation, patient preference, patient satisfaction with care, quality improvement, deficiencies and improvement in patient access to treatments, factors affecting access to treatment, disparities or access to treatment, PRO measures, HRQOL, unmet needs, patient meaningful outcomes, or caregiver experiences Articles that discuss unmet needs in provider education thereby improving the patient experience/journey	Pharmacoeconomic studies (eg, cost effectiveness analysis, cost of illness, productivity) Solely clinical efficacy studies Recruitment gaps in clinical trials
Article Type	Peer-reviewed publications and meeting abstracts: review papers, commentaries, meta-analysis, clinical trials, SLR, or targeted literature review Article reference lists	Gray literature
Country	All	NA
Language	English articles	Non-English articles

AML, acute myeloid leukemia; COA, clinical outcome assessment; HR-MDS, higher-risk MDS; HRQOL, health-related quality of life; LR-MDS, lower-risk MDS; MDS, myelodysplastic syndromes; NA, not applicable; PCC, patient-centric care; PRO, patient-reported outcome.

Results

Identification of Articles via PubMed, Embase, and Cochrane Collaboration (Figures 1-2)

Figure 1: PRISMA Flow Diagram Identifying MDS/HR-MDS Articles

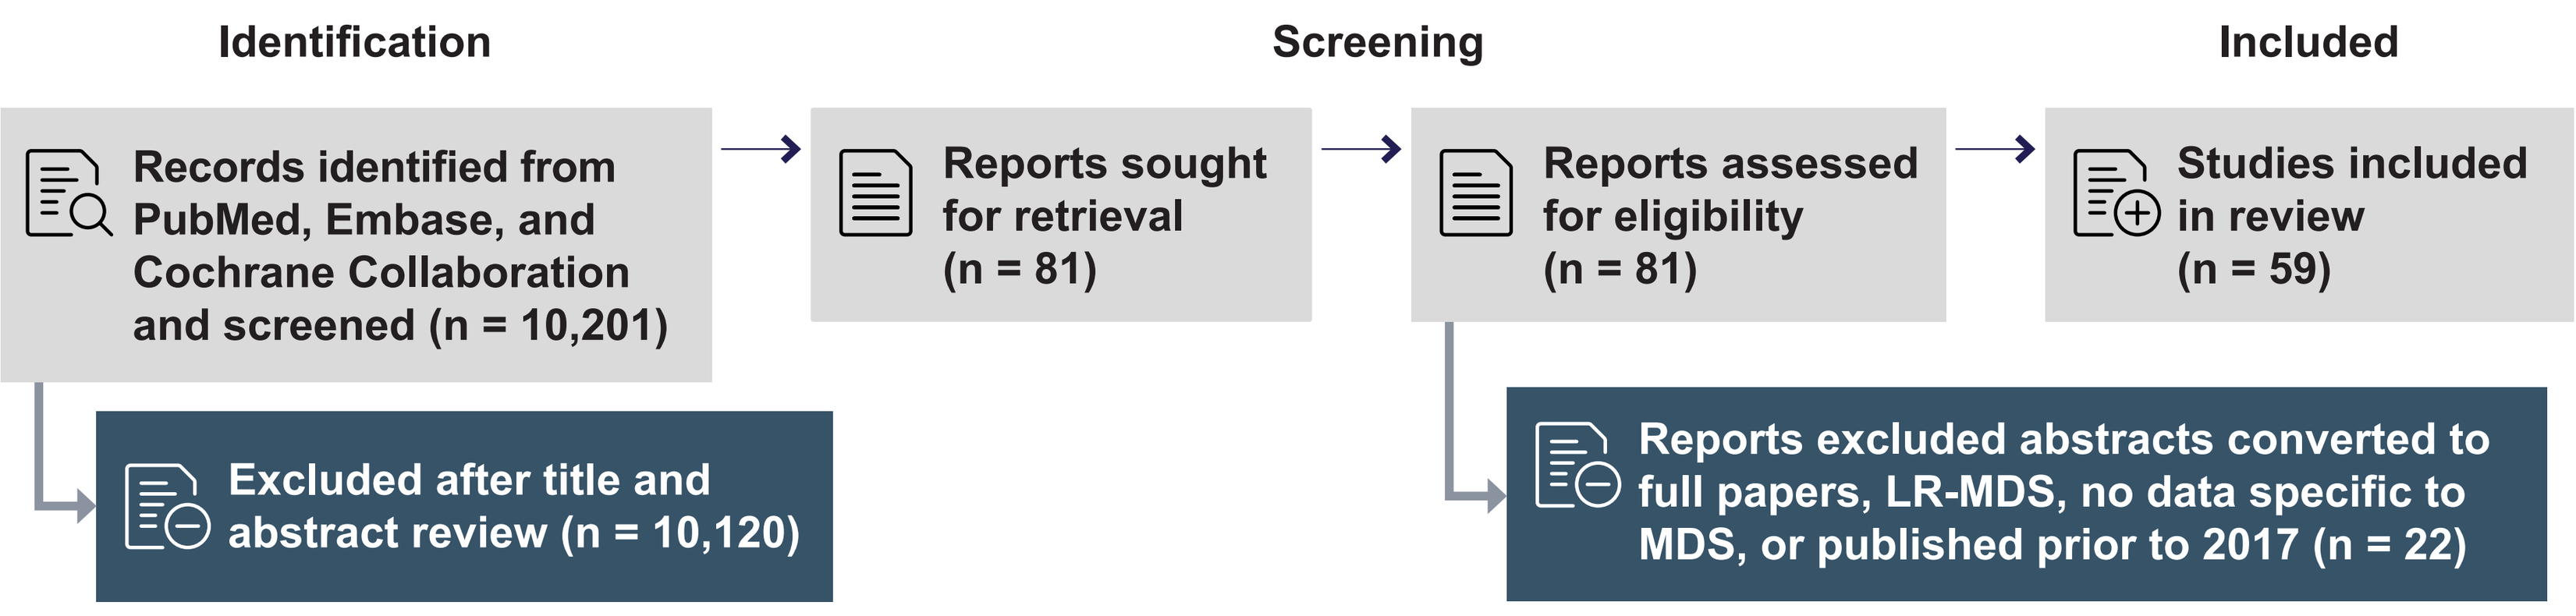

Identification
Screening
Included
Records identified from PubMed, Embase, and Cochrane Collaboration and screened (n = 10,201)
Reports sought for retrieval (n = 81)
Reports assessed for eligibility (n = 81)
Studies included in review (n = 59)
Excluded after title and abstract review (n = 10,120)
Reports excluded abstracts converted to full papers, LR-MDS, no data specific to MDS, or published prior to 2017 (n = 22)

HR-MDS, higher-risk MDS; LR-MDS, lower-risk MDS; MDS, myelodysplastic syndromes; PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses.

Results (continued)

Figure 2.

Panel A | Of 10,201 Screened

MDS/HR-MDS articles	59 articles met study inclusion criteria	
	45 empirical and 14 review	
HR-MDS articles	Discussed in	**14**
	Target/focus in	**6**
AML	Discussed in	**32**
	Target/focus in	**8**

AML, acute myeloid syndrome; HR-MDS, higher-risk MDS; MDS, myelodysplastic syndromes.

Panel B | Global Study Representation: North America, South America, Europe, Australia, and Asia

Austria, Australia, Belgium, Brazil, Canada, China, Croatia, Czech Republic, Denmark, France, Germany, Greece, India, Israel, Italy, The Netherlands, Pakistan, Poland, Portugal, Romania, Serbia, South Korea, Spain, Sweden, Switzerland, United Kingdom, United States

Panel C | Regions Mostly Presented

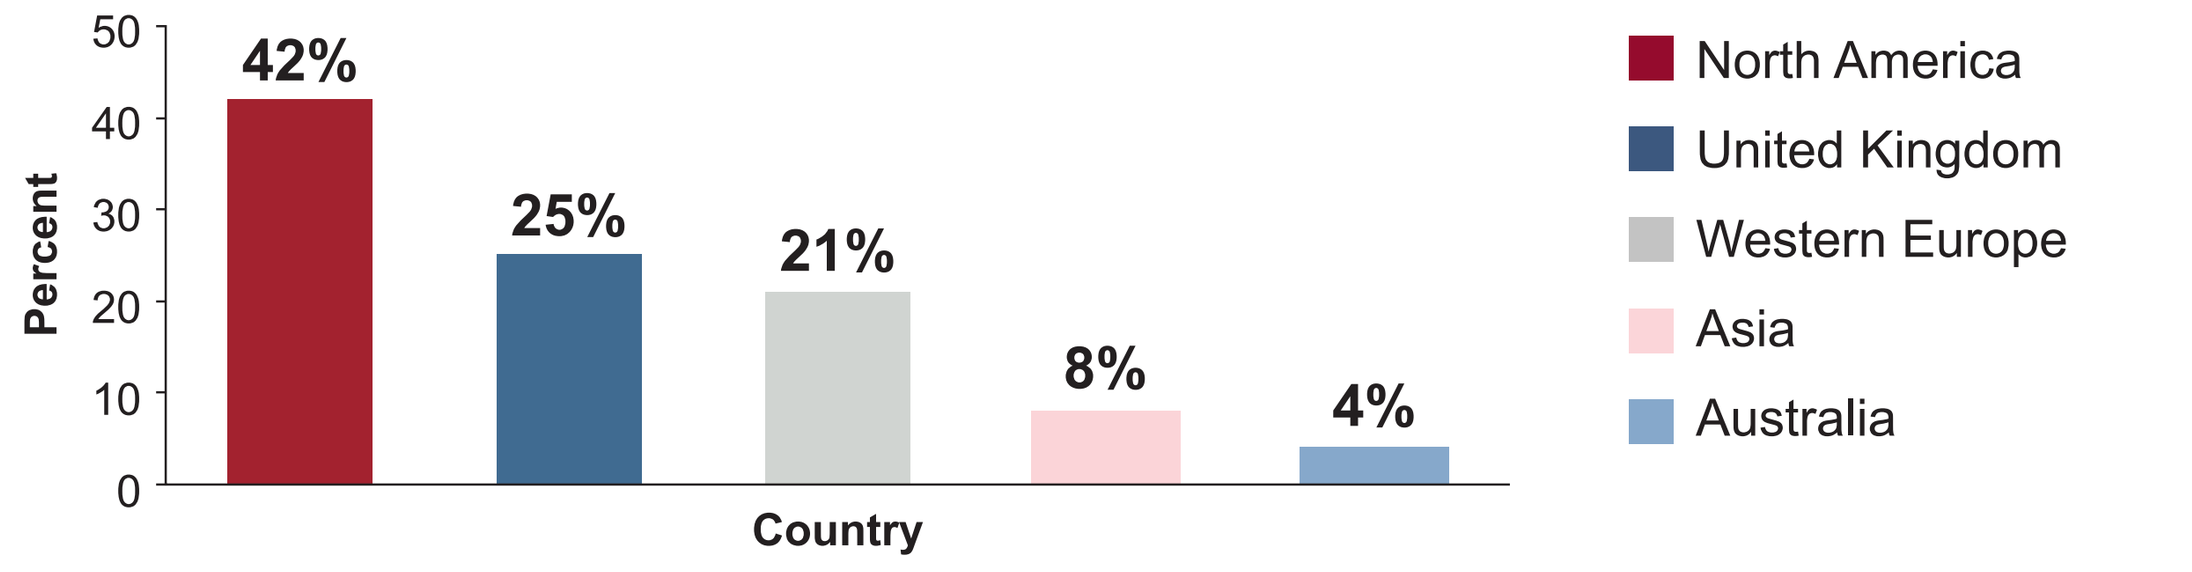

50
40
30
20
10
0
Percent
42%
25%
21%
8%
4%
Country
North America
United Kingdom
Western Europe
Asia
Australia

HR-MDS and AML articles were scarce and varied across study designs and themes (Table 2)

Table 2: Descriptive Characteristics of Identified Articles

Category	Theme	HR-MDS/MDS #Articles[a]	AML #Articles[a]	Country
Fatigue/Frailty/ HRQOL/SDM	Core Outcomes	0/3	0	
	HRQOL	3/5	2	
	Fatigue/Frailty	1/5	0	
	Patient Preferences	1/6	1	
	SDM	0/2	0	
	Social Media/Online Educational Material	0/4	1	
	Patient-Oriented Mobile Apps	0/4	0	
	Telehealth Intervention	0/1	1	
	Prescriber Gaps/CE Needs	0/1	1	Not stated
	Treatment Patterns	0/1	0	
Health and Economic Disparities	Disparities	1/8	1	and 1 not stated
Reviews	Commentaries or Reviews	0/14	2	

[a]Scan the poster QR code for the full reference list.
AML, acute myeloid leukemia; CE, continuing education; HRQOL, health-related quality of life; HR-MDS, higher-risk MDS; MDS, myelodysplastic syndromes; SDM, shared decision-making.

Two Major Theme Categories:

1. Impact of disease and treatment on health-related quality of life (HRQOL), fatigue, frailty, and SDM

HRQOL/PROs
- Two HR-MDS PROs under development were identified[4,12]
- Lack of a valid and reliable condition-specific HRQOL instrument to assess outcomes that are meaningful to HR-MDS patients

Core Outcomes
- Defined as a minimum set of outcomes (including HRQOL) aimed to enable comparisons across MDS studies and treatment efficacy as defined by patients and physicians[13]

Fatigue

Fatigue was the most commonly reported physical symptom that caused distress in patients with HR-MDS. The five most frequent symptoms contributing to distress were fatigue, pain, worry, sleep, and tingling in the hands or feet.[14,15] Clinical outcome assessments developed to measure fatigue included the following:
- Distress Thermometer (DT)[14]
- Fatigue International Prognostic Scoring System for higher-risk patients (FA-IPSS[h]) was found to be a significant predictor of overall mortality based on the IPSS and IPSS-revised (IPSS-R)[16,17]
- MDS-Frailty Index (MDS-FI) was developed using baseline demographic, disease-specific laboratory values, activities of daily living, the EQ-5D, and 3 physical fitness tests[18]

Patient Preferences/Treatment Decisions/SDM
- Significant variations in the experiences and preferences of patients with MDS/HR-MDS for blood transfusions were reported,[19] which varied by country[20] (**Figure 2C**)
- Patients were concerned about iron overload in chelation therapy[21]
- Patients reported a preference for oral treatments over injectable hypomethylating agents[22]
- Patients prioritized improving HRQOL, communication, access, and desired individualized care – beyond hemoglobin levels or a "numbers-led" approach. Patients valued maintaining independence and PCC.[7,8] Patients highlighted HRQOL and active participation in treatment decisions
- There was misalignment between patient and physician treatment goals

Education, Prescriber Gaps, Social Media
- Patients with either AML or MDS and their caregivers lack information on treatment options and humanistic impacts[6]
- Many patients and even some physicians continue to think of MDS as a benign blood "disorder" or a "syndrome" rather than a malignancy[10]
- A prescriber gap, a disparity between prescriber and the expert opinion of care, exists for older adult patients with MDS/HR-MDS or AML
- Online resources/social media appears to be reaching technology-oriented patients and caregivers and has removed many of the barriers to accessing timely information about their disease[23]

2. Disparities in health and care

- Disparities in regard to sex, race, economic status, and insurance coverage have been found to exist with respect to care for patients with MDS
- Only 1 study targeted HR-MDS treatment disparities[24] (in Denmark) and 8 studies targeted MDS
 - Individuals with less education (<12 years) were more likely to have transfusion-dependent (TD) disease at the time of diagnosis and more likely to be diagnosed with HR-MDS treatment disparities
 - Individuals with lower income were associated with a higher risk of mortality as compared with those with higher income
 - Individuals who live alone were more likely to be TD at the time of diagnosis

Presented at ISPOR 2023; May 7–10, 2023; Boston, MA