Evaluation of Digital Mental Health Technologies in the United States: A Systematic Literature Review and Mixed-Methods Framework Synthesis

Authors: Malone DC,[1] Beaver S,[2] Catania J,[3] Kamath RS,[3] Worthington E,[4] Lu M,[5] Gandhi H,[5] Waters HC[5]

Affiliations: [1]University of Utah, Salt Lake City, UT, US; [2]Costello Medical, London, UK; [3]Costello Medical, Boston, MA, US; [4]Costello Medical, Cambridge, UK; [5]Otsuka Pharmaceutical Development & Commercialization Inc., Princeton, NJ, US

MT12

OBJECTIVES

To conduct a systematic literature review (SLR) and framework synthesis to understand how digital mental health technologies (DMHT) are valued in the United States (US).

BACKGROUND

- DMHT have the potential to enhance mental healthcare.[1]
- Some DMHT are subject to the Food and Drug Administration's (FDA) approval in the US, but most low-risk technologies, including mobile device apps, are not.[2] As such, there is little information on how DMHT are evaluated.

METHODS

- MEDLINE, Embase, the Cochrane Library, PsycINFO, and the Health Technology Assessment (HTA) Database were searched in June 2022. SLR reference lists, the FDA website, and the following congresses (2019 onwards) were hand-searched: HTA International, Drug Information Association Global Annual Meeting (AM), Society for Digital Mental Health (MH) AM, American Psychiatric Association (APA) AM, and American Society of Clinical Psychopharmacology AM.
- Records were assessed by two independent reviewers to identify studies from 2017 onwards reporting novel quantitative or qualitative data on theoretical valuation factors for DMHT targeting MH and central nervous system indications from US consumer, payer, and employer perspectives.
- Data were extracted by one individual and validated by a second. Quality assessments were conducted using the Critical Appraisal Skills Programme's Qualitative and Cohort Study Checklists.[3]
- For the framework synthesis, the APA MH App Evaluation Framework was adapted into three levels: themes, subthemes, and criteria.[4]
- Extracted data were coded and mapped by two individuals onto the adapted framework using Docear, a mind-map software, and novel themes, subthemes, and criteria were inductively added to the framework *post hoc*.

RESULTS

- Of 4,353 records screened, data from 26 studies were included (**Figure 1**).
- Eight studies were quantitative,[5–14] twelve qualitative,[15–26] and six mixed-methods.[27–32] All took a consumer perspective and assessed apps, except one that assessed an electronic pill monitor.[9–11]
- All APA-defined themes were reported (**Figure 2**). Under existing themes/subthemes, five novel criteria were identified (**Figure 3**).
- No relevant FDA appraisals were identified; however, FDA MH app guidance reiterated the value of most themes in the APA framework.
- Overall, risk of bias for included evidence was moderate.

Figure 1. PRISMA diagram

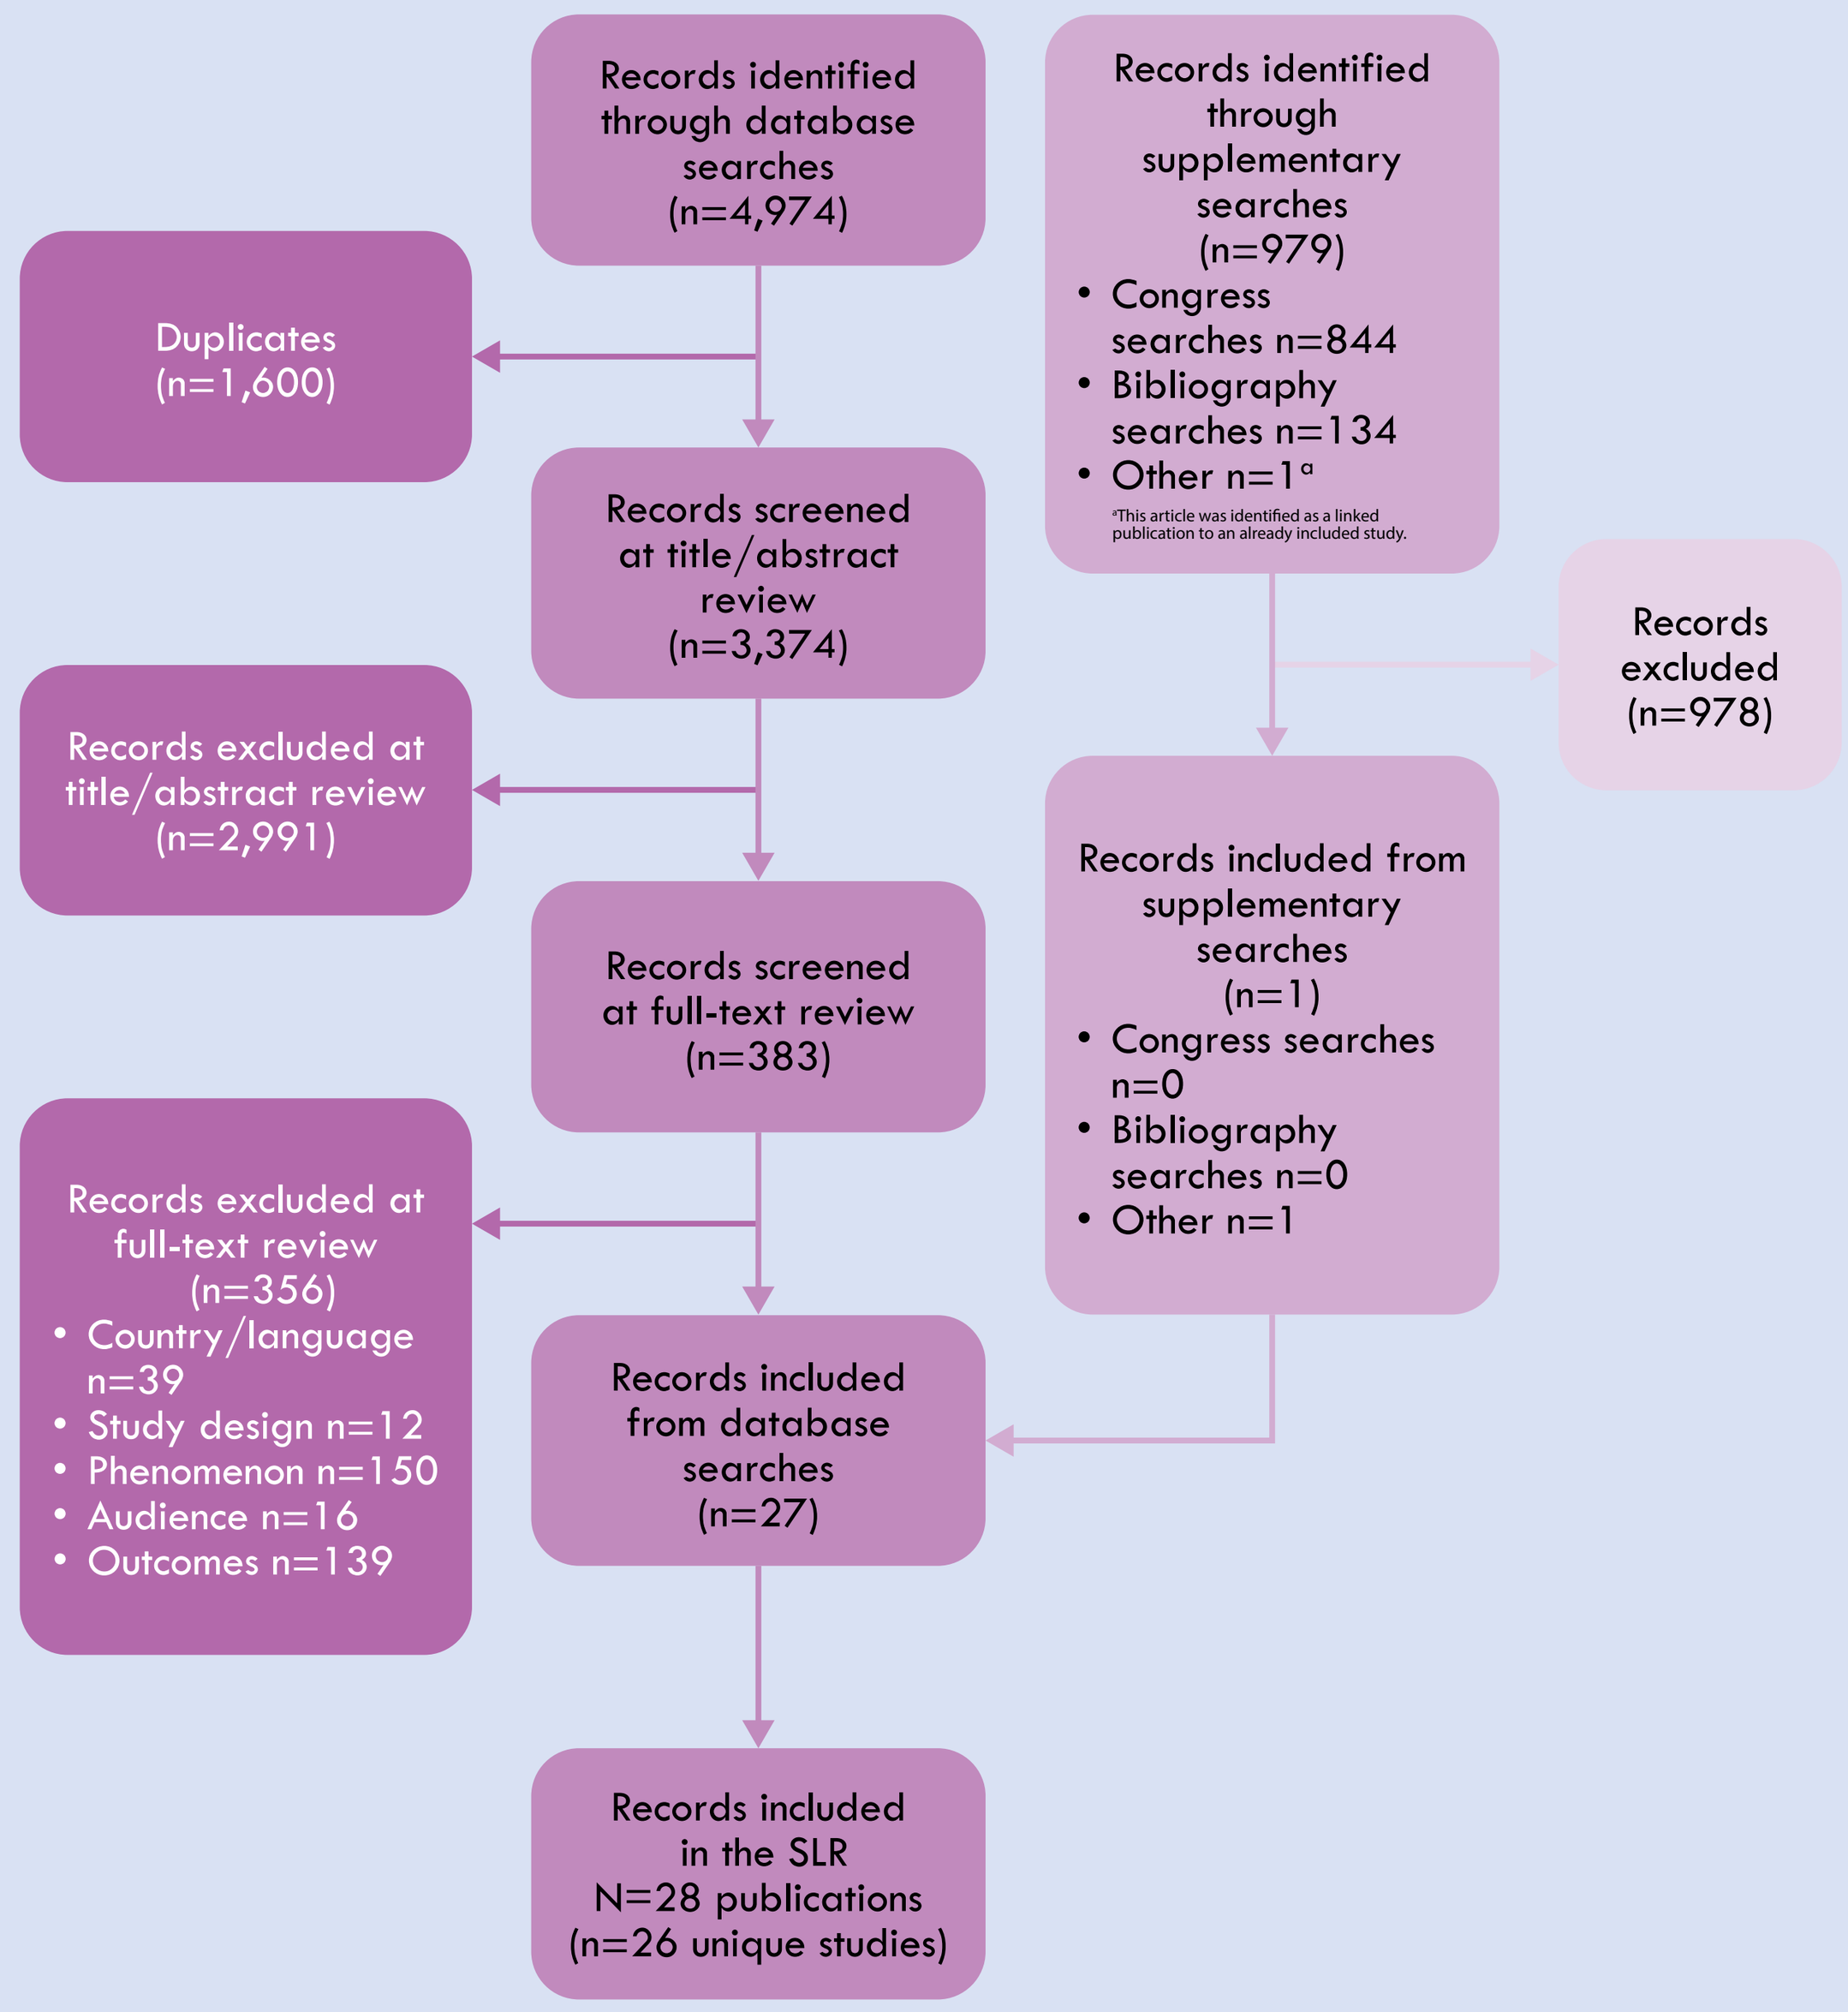
Records identified through database searches (n=4,974)
Records identified through supplementary searches (n=979)
• Congress searches n=844
• Bibliography searches n=134
• Other n=1a
aThis article was identified as a linked publication to an already included study.
Duplicates (n=1,600)
Records screened at title/abstract review (n=3,374)
Records excluded at title/abstract review (n=2,991)
Records excluded (n=978)
Records screened at full-text review (n=383)
Records included from supplementary searches (n=1)
• Congress searches n=0
• Bibliography searches n=0
• Other n=1
Records excluded at full-text review (n=356)
• Country/language n=39
• Study design n=12
• Phenomenon n=150
• Audience n=16
• Outcomes n=139
Records included from database searches (n=27)
Records included in the SLR N=28 publications (n=26 unique studies)

Figure 2. Adapted APA MH App Evaluation Framework themes and subthemes, including the number of studies reporting on each

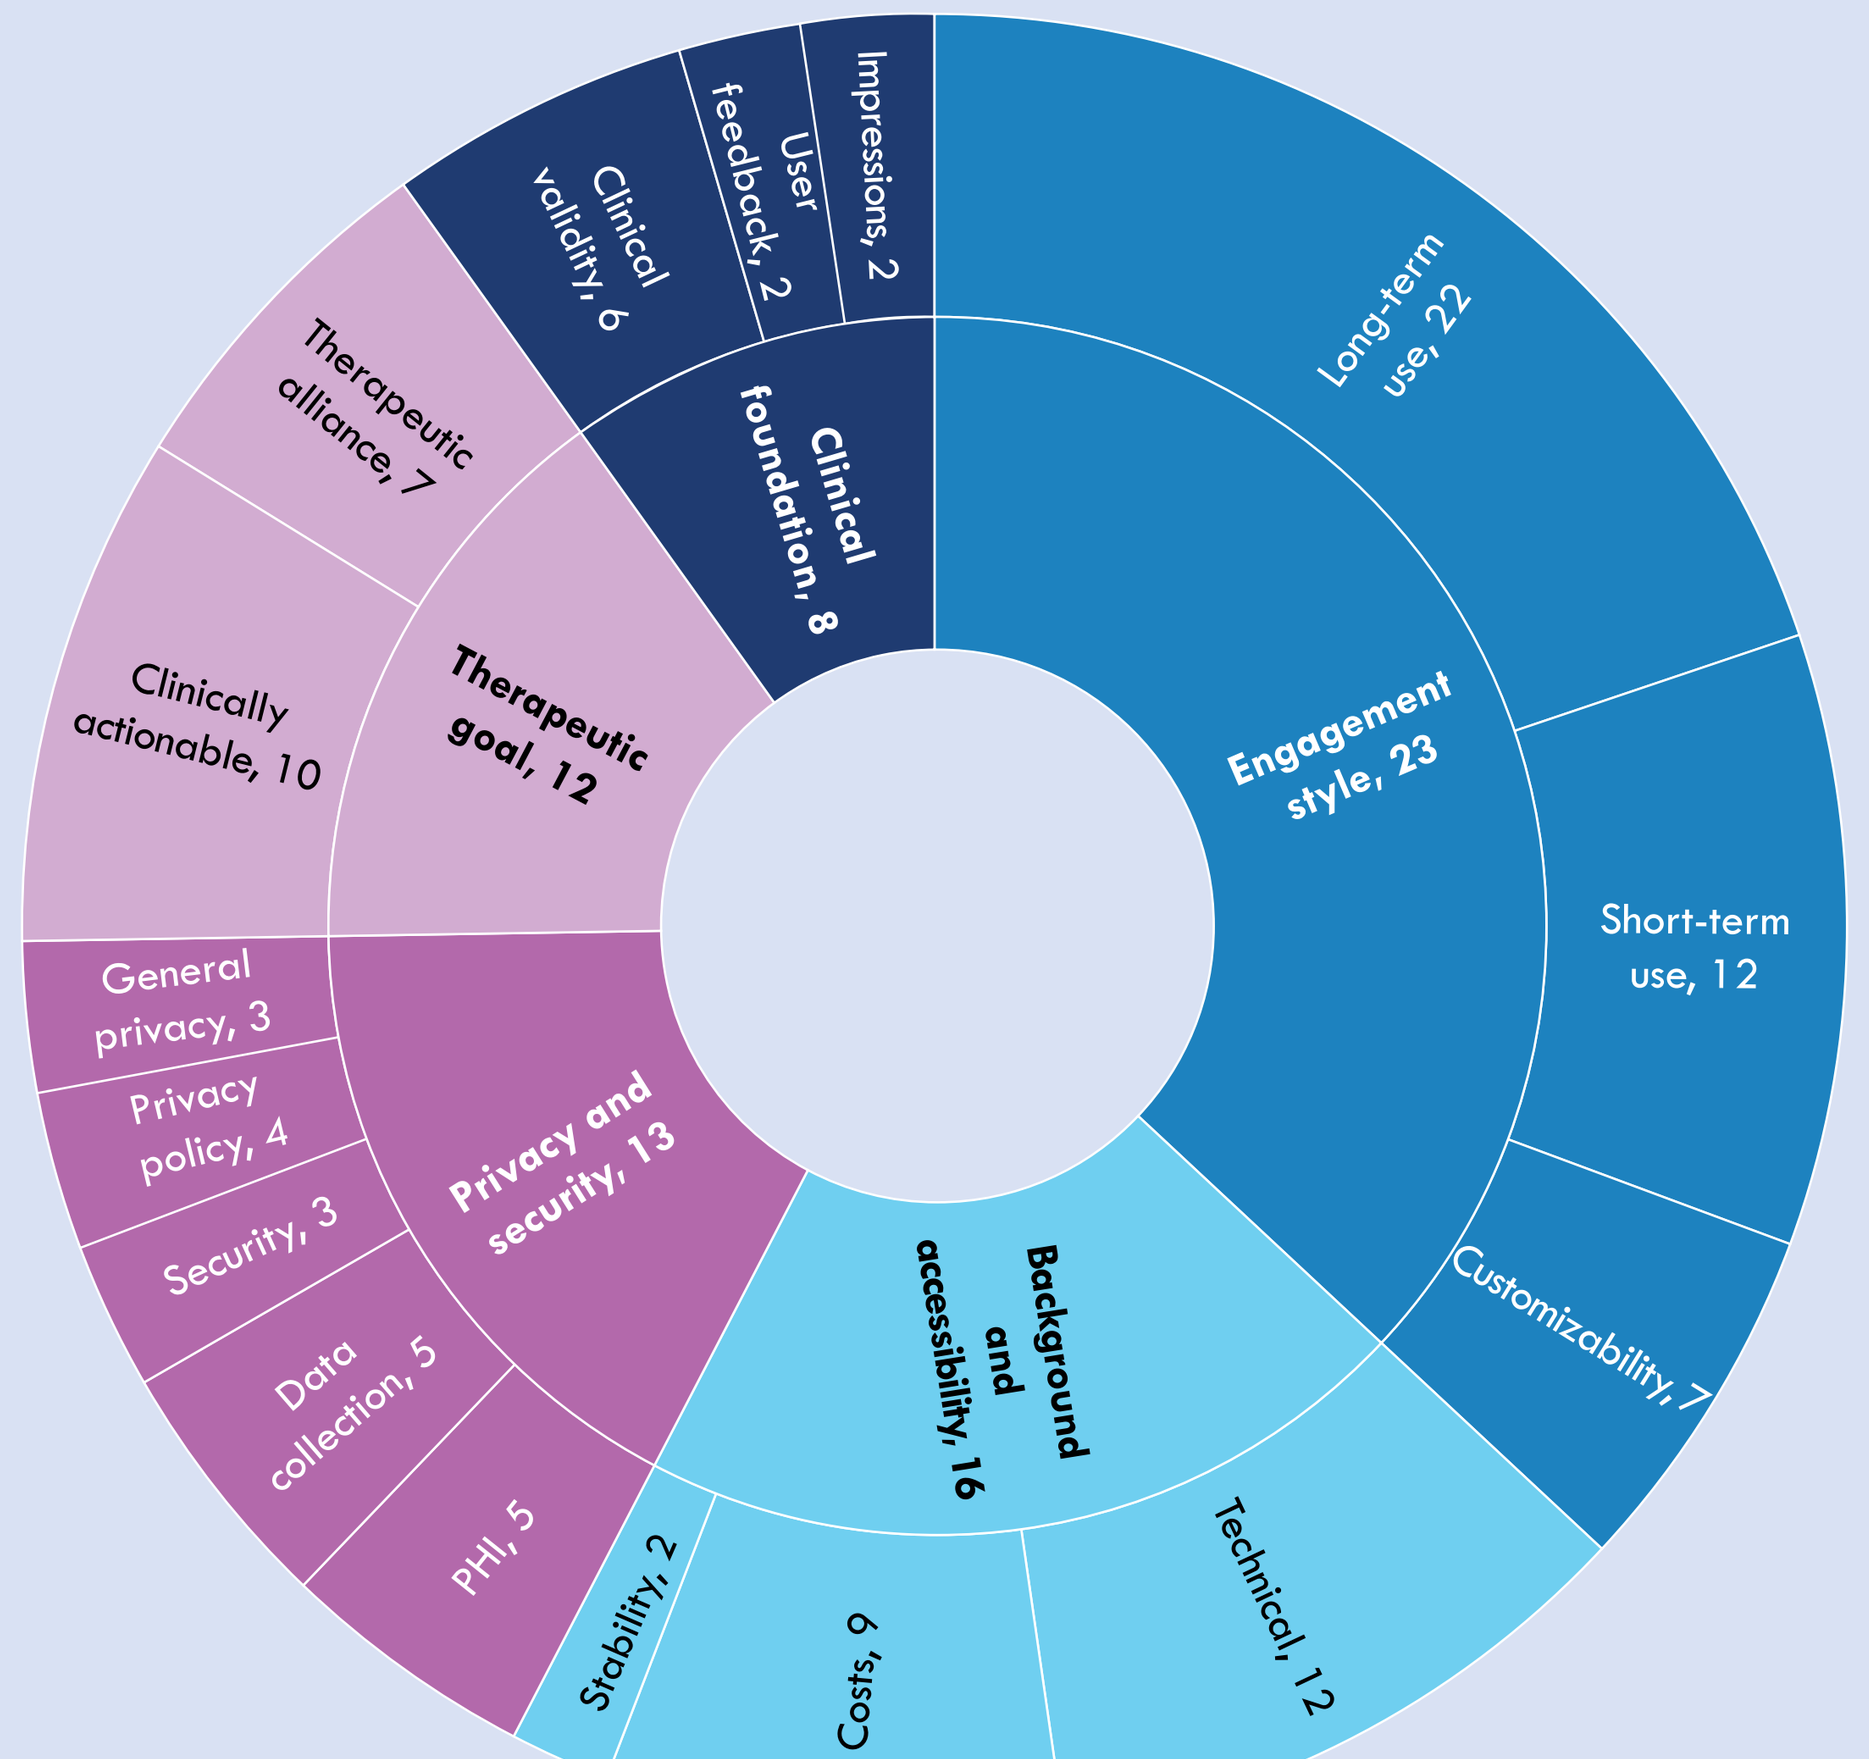
Engagement style, 23
Long-term use, 22
Short-term use, 12
Customizability, 7
Background and accessibility, 16
Technical, 12
Costs, 9
Stability, 2
Privacy and security, 13
PHI, 5
Data collection, 5
Security, 3
Privacy policy, 4
General privacy, 3
Therapeutic goal, 12
Clinically actionable, 10
Therapeutic alliance, 7
Clinical foundation, 8
Clinical validity, 6
User feedback, 2
Impressions, 2

Engagement Style

- Psychoeducation, peer and professional support, automated positive affirmations, ease of use, and feature customizability promoted short- and long-term engagement (**Figure 4**).[5, 7, 8, 12–15, 17, 19, 21, 23–31]
- Users valued features that aligned with therapeutic goals, including anxiety management strategies and reminders for medication refills and appointments.[5–14, 17, 19, 21–23, 25, 26, 28, 29]
- Therapeutic games/activities interested many users; however, some caregivers and healthcare providers (HCPs) expressed concerns about distraction for child users.[5, 15, 22, 29]

Privacy and Security

- Concerns for the privacy and security of DMHT varied, likely due to heterogeneity in study populations (**Figure 5**).[12, 13, 18, 20, 22, 27, 28, 32]
- In a novel finding, some users were concerned about confidentiality and DMHT discreetness due to MH stigma (**Figure 3**).[12, 18, 28, 32]
- Despite some heterogeneity in opinion, data protection measures and accessible, transparent privacy policies were valued across studies.[18, 22, 25, 26, 28, 30–32]

Background and Accessibility

- Studies reported accessibility concerns for users with physical disabilities, low technical literacy, limited understanding of English, and limited device storage and data plans.[16, 18, 20–22, 25, 27, 28]
- Three novel cost-related criteria emerged from this SLR: willingness to pay, insurance restrictions, and cost savings (**Figure 3; Figure 6**).[9–12, 15, 18, 30, 31]
- Ancillary costs, such as device data packages, were qualitatively identified as a major cost barrier, suggesting that hidden costs influence value.[19, 25]

Therapeutic Goal

- Patients, caregivers, and HCPs valued clinically actionable functions including management strategies, symptom trackers, therapeutic exercises like mindfulness, and easy data sharing.[6, 18–24, 28, 30]
- Many HCPs reported active use or interest in use of DMHT in clinical practice to facilitate asynchronous communication and therapeutic interventions outside of formal appointments; however, some preferred traditional care strategies that afford more personalization and flexibility.[19, 22, 28]

Clinical Foundation

- Individuals valued positive user feedback and recommendations or evidence of benefit from HCPs and academic, research, and medical institutions.[13, 18–20, 24, 25, 30, 31]
- Redundant, irrelevant, or stigmatizing content discouraged DMHT use.[18, 19]

Figure 3. Novel criteria identified by this SLR

Theme	Subtheme	Novel Criterion
Engagement style	Long-term usability	Forgetting or being unmotivated to use DMHT (n=4)
Privacy and security	Personal health information	Desire to keep conditions private due to personal image and stigma surrounding MH (n=4)
Background and accessibility	Costs	Willingness to pay (n=4)
		Devices purchased under insurance may limit other device uses (n=1)
		Cost saving vs professional care (n=1)

Figure 4. Select studies reporting features that promote short- and long-term engagement

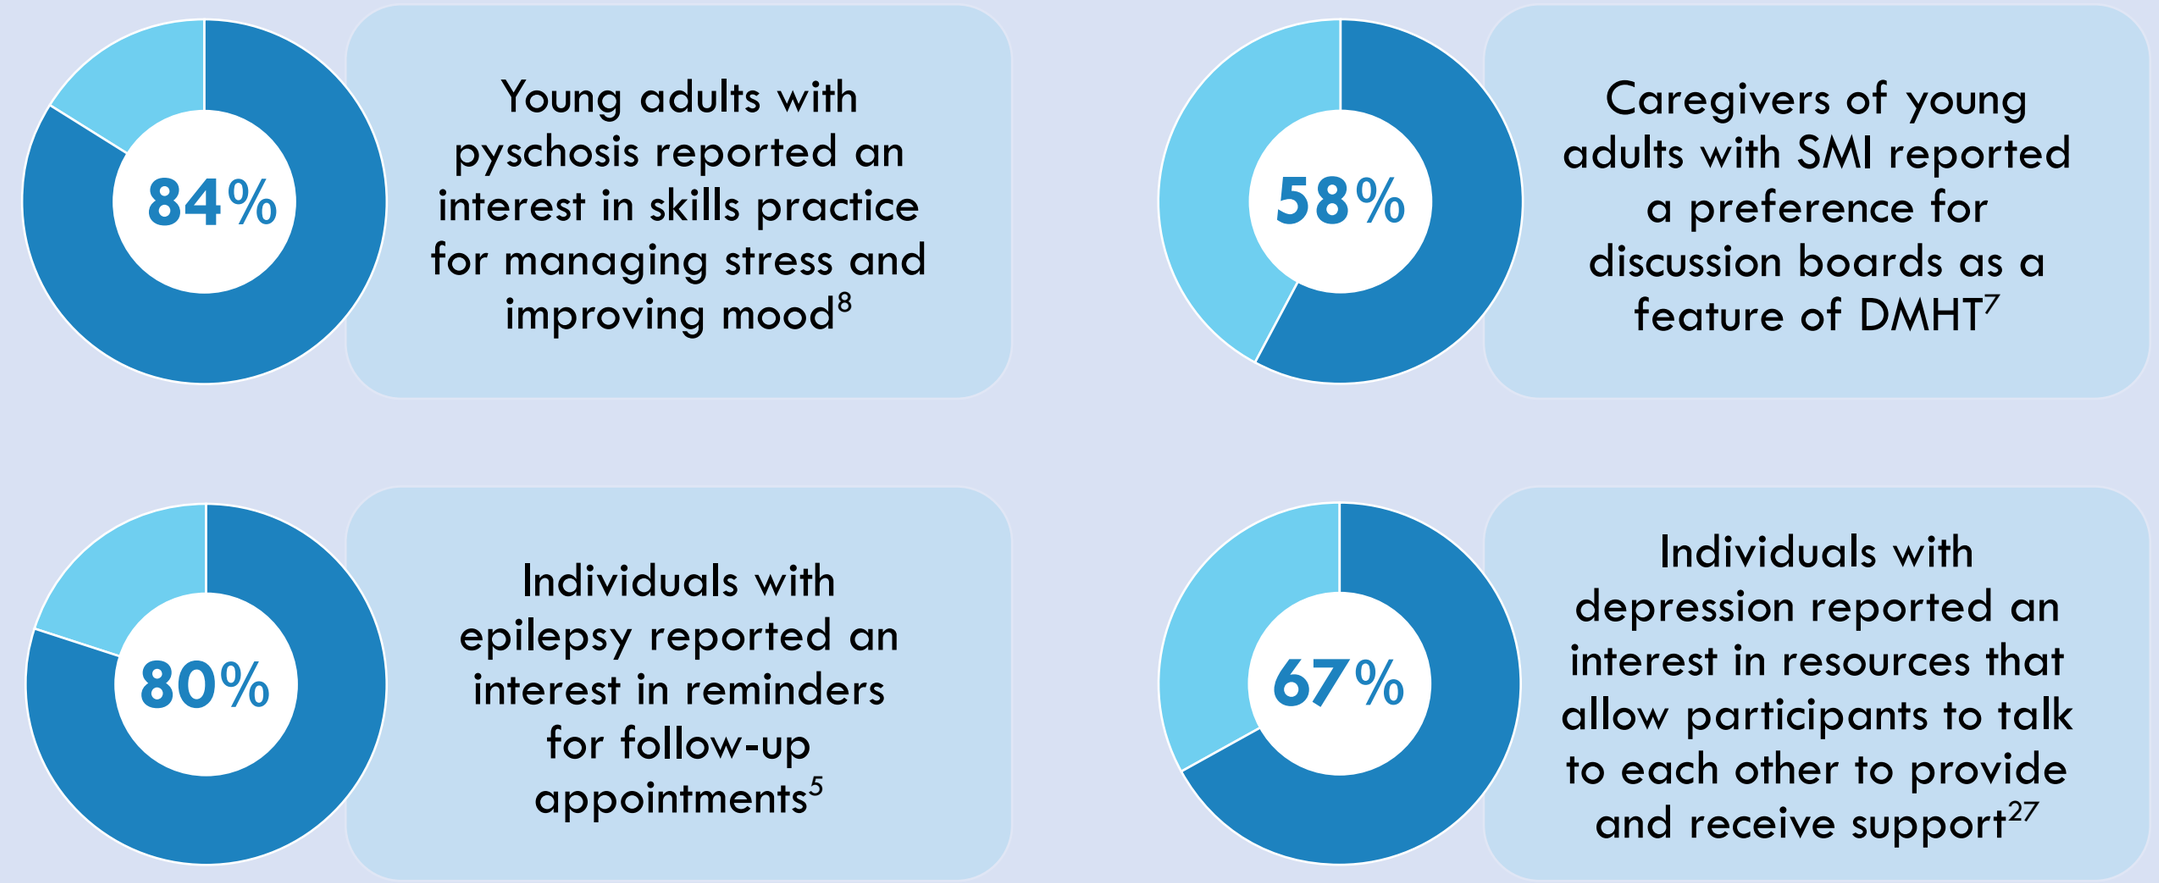
84%
Young adults with pyschosis reported an interest in skills practice for managing stress and improving mood8
58%
Caregivers of young adults with SMI reported a preference for discussion boards as a feature of DMHT7
80%
Individuals with epilepsy reported an interest in reminders for follow-up appointments5
67%
Individuals with depression reported an interest in resources that allow participants to talk to each other to provide and receive support27

"I no longer feel alone […] 'cause there's other people that are in the same boat with me. And so I think those stories are really important."
- Cancer patient/survivor, Huberty 2022[21]

Figure 5. Select studies reporting privacy and security concerns expressed by various consumer populations

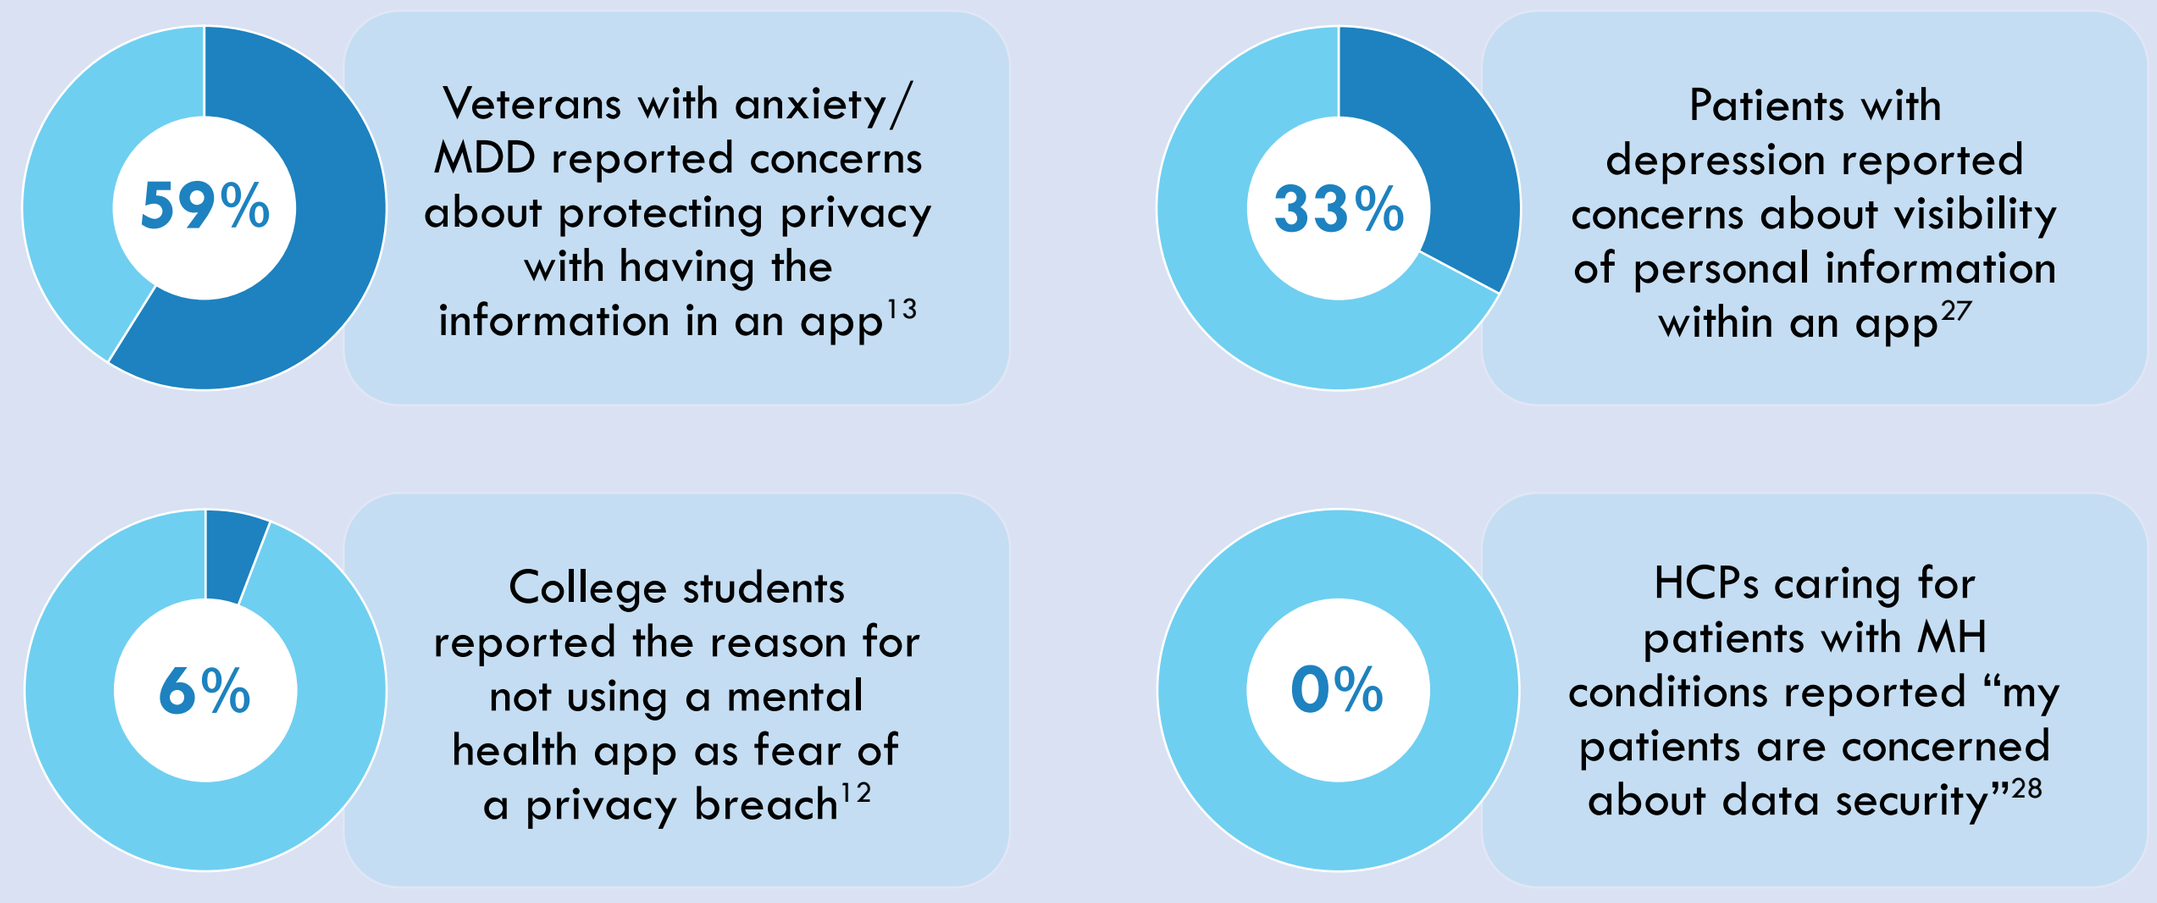
59%
Veterans with anxiety/MDD reported concerns about protecting privacy with having the information in an app13
33%
Patients with depression reported concerns about visibility of personal information within an app27
6%
College students reported the reason for not using a mental health app as fear of a privacy breach12
0%
HCPs caring for patients with MH conditions reported "my patients are concerned about data security"28

"I worry about my virtual image. I'd feel more comfortable using an app […] that is protected in the same way my electonic medical record is protected."
- Behavioral health patient, Hoffman 2019[28]

Figure 6. Factors affecting willingness to pay for a mobile app

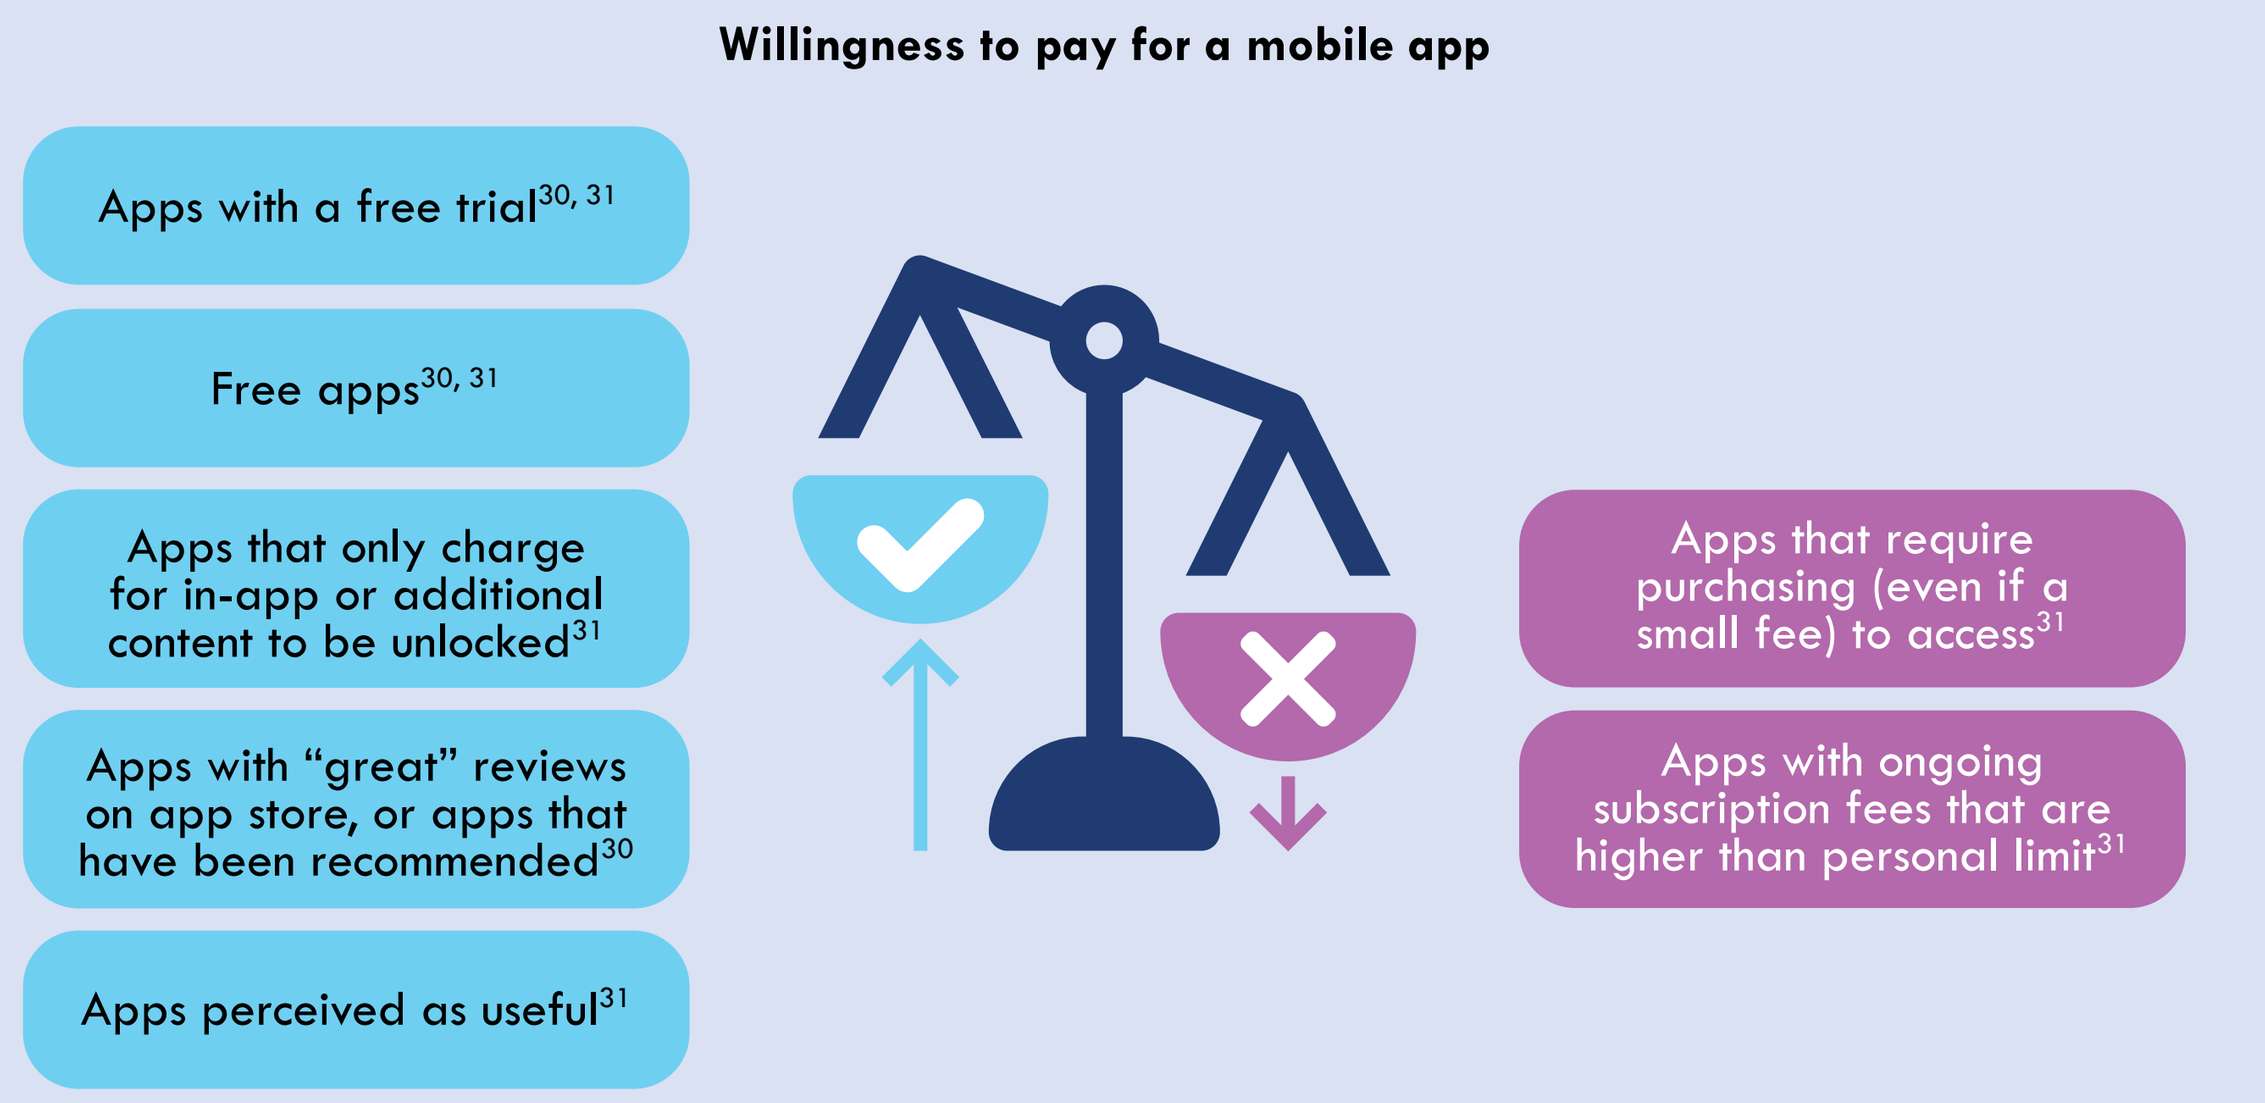
Willingness to pay for a mobile app
Apps with a free trial30, 31
Free apps30, 31
Apps that only charge for in-app or additional content to be unlocked31
Apps with "great" reviews on app store, or apps that have been recommended30
Apps perceived as useful31
Apps that require purchasing (even if a small fee) to access31
Apps with ongoing subscription fees that are higher than personal limit31

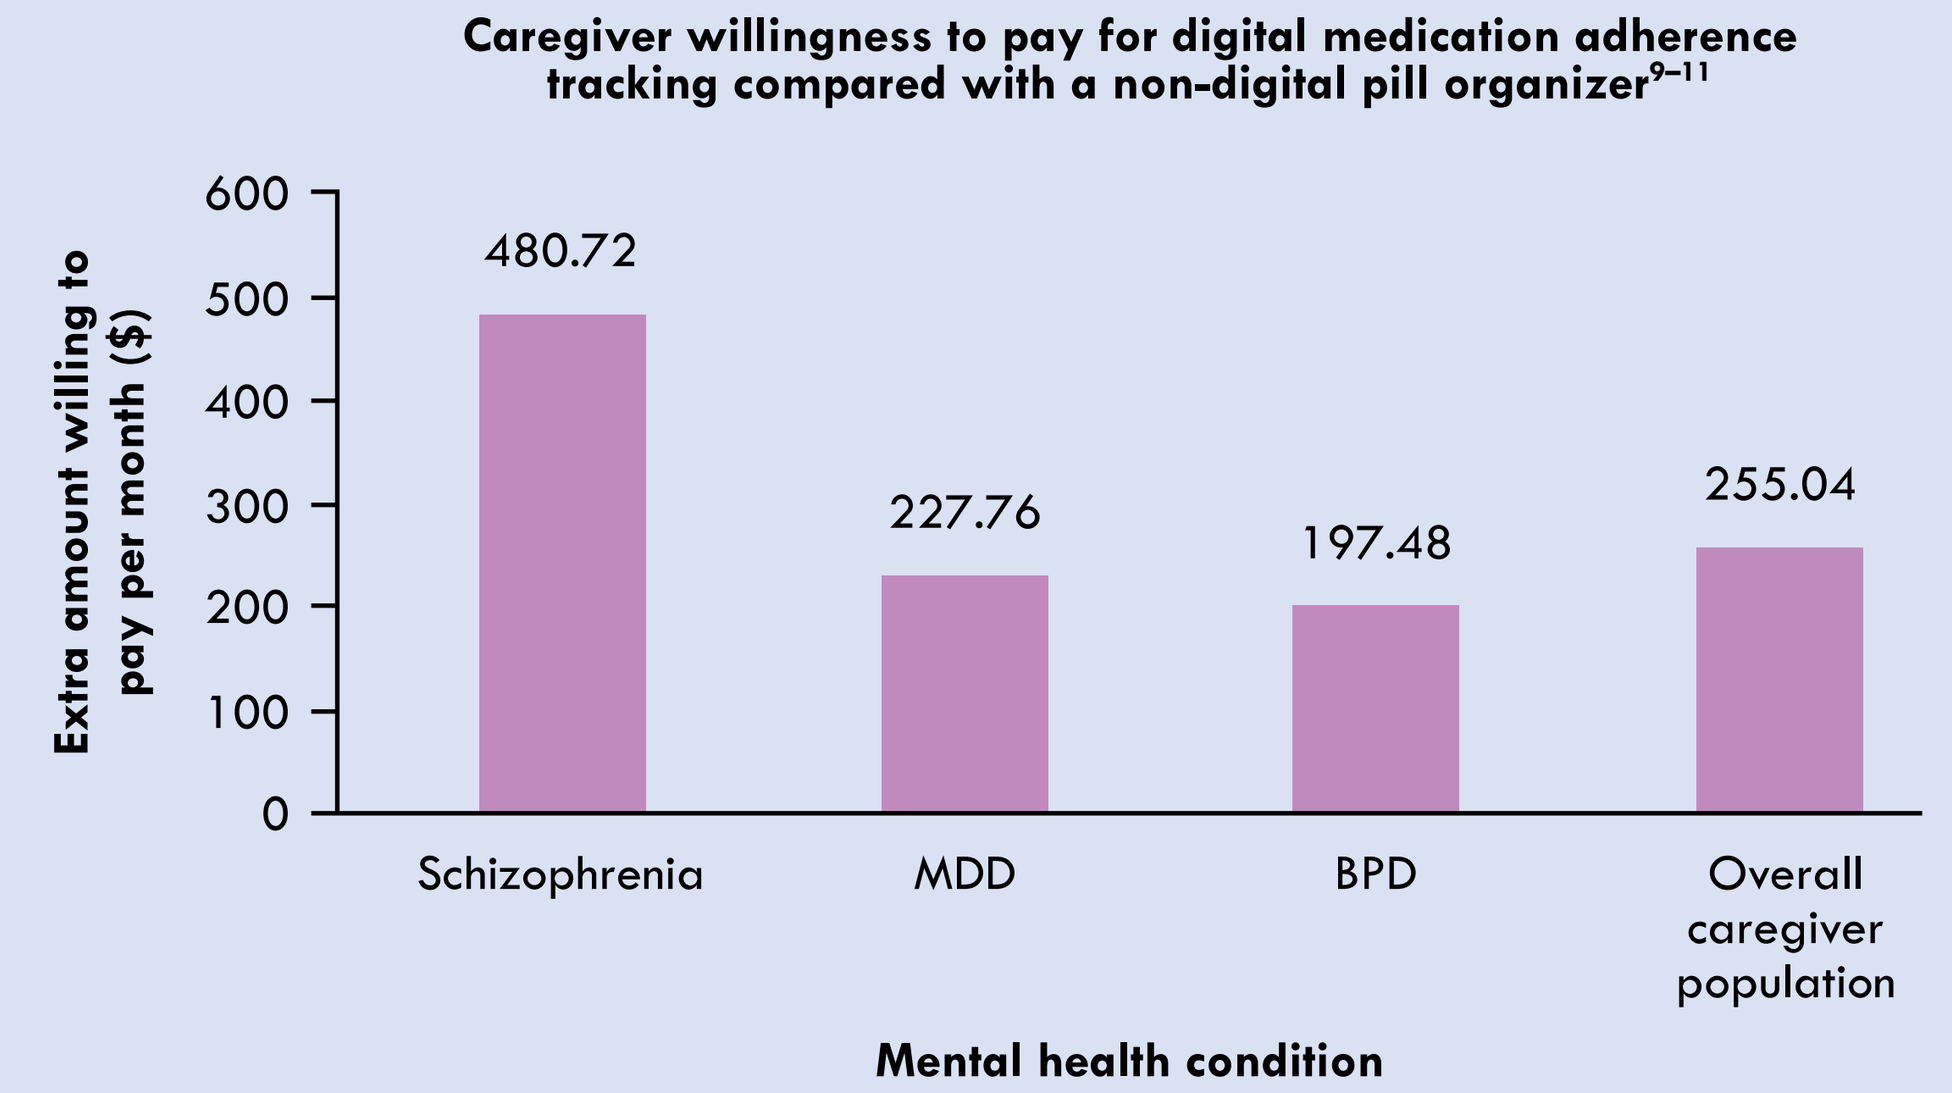
Caregiver willingness to pay for digital medication adherence tracking compared with a non-digital pill organizer9–11
Extra amount willing to pay per month ($)
600
500
400
300
200
100
0
480.72
227.76
197.48
255.04
Schizophrenia
MDD
BPD
Overall caregiver population
Mental health condition

CONCLUSIONS

- Short- and long-term usability, technical considerations, cost, and clinical actionability were the most reported subthemes, indicating their importance as valuation factors.
- DMHT valuation may differ among demographics and consumer types; differences among HCP, caregiver, and patient perspectives were identified in quantitative and qualitative data.
- Employer and payer perspectives and non-app DMHT were poorly represented in the literature and therefore warrant further research.

Abbreviations: AM, Annual Meeting; APA, American Psychiatric Association; BPD, bipolar disorder; DMHT, digital mental health technologies; FDA, Food and Drug Administration; HCP, healthcare provider; HTA, Health Technology Assessment; MDD, major depressive disorder; MH, mental health; PHI, personal health information; PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses; SLR, systematic literature review; SMI, serious mental illness; US, United States.

Acknowledgments: The authors acknowledge Max Lee, Costello Medical, US, for medical writing and editorial assistance, and Shaida Saharkhizian, Costello Medical, UK, for design assistance, based on the authors' input and direction. This research was funded by Otsuka Pharmaceutical Development & Commercialization (OPDC) Inc.

Disclosures: DCM is a consultant for OPDC for this project, and has received consulting funds from Pear Therapeutics, Sanofi, Avidity, Sarepta, Novartis, and Biomarin. LM, HG, and HCW are employees of OPDC.

References: [1]U.S. Food & Drug Administration. What is Digital Health? 2020; [2]Guo C et al. NPJ Digit Med 2020;3:110; [3]CASP. CASP Critical Appraisal Checklists, 2022; [4]Lagan S et al. Psychiatr Serv 2021;72:1095–1098; [5]Afra P et al. Front Hum Neurosci 2018;12; [6]Beard C et al. JMIR MHealth Uhealth 2019;7:e13364; [7]Buck B et al. Psychiatr Serv 2021;72(8):955–959; [8]Buck B et al. J Technol Behav Sci 2021; 6:667–676; [9]Forma F et al. Digit Health 2022;8:20552076221084472; [10]Forma F et al. Journal of Managed Care and Specialty Pharmacy 2021;27(4-A SUPPL):S58–S59; [11]Forma F et al. Value in Health 2021; 24(SUPPL1):S133–S13; [12]Kern A et al. Mhealth 2018 2018;4:20; [13]Lipschitz J et al. JMIR Ment Health 2019;6:e11334; [14]Torous J et al. JMIR Ment Health 2018;5:e11715; [15]Boster JB et al. Disabil Rehabil Assist Technol 2018;13(4):353–365; [16]Carpenter-Song E et al. JMIR Ment Health 2018;5:e10652; [17]Casarez RL et al. J Affect Disord 2019;250:391–396; [18]Connolly SL et al. JMIR MHealth UHealth 2018;6:e10748; [19]Cummings JR et al. J Technol Behav Sci 2019; 4:6–16; [20]Dinkel D et al. Transl Behav Med 2021; 11(9):1708–1716; [21]Huberty J et al. JMIR Form Res 2022;6:e32458; [22]Knapp AA et al. JMIR Ment Health 2021;8(8); [23]Kornfield R et al. Proc SIGCHI Conf Hum Factor Comput Syst 2022; [24]Schueller SM et al. JMIR Ment Health 2021;8(8); [25]Stiles-Shields C et al. Digit Health 2017;3:2055207617713827; [26]Storm M et al. Proc IEEE Glob Humanit Technol Conf 2021;2021:195–199; [27]Borghouts J et al. JMIR Hum Factors 2022;9:e35641; [28]Hoffman L et al. Front Psychiatry 2019;10(FEB); [29]Mata-Greve F et al. JMIR Ment Health 2021;8(8); [30]Melcher J et al. J Am Coll Health 2020:1–7; [31]Schueller SM et al. J Med Internet Res 2018;20(6):e10141; [32]Zhou L et al. Int J Telerehabil 2020;12:13–26.

Presented at ISPOR Annual Meeting, May 7–10, 2023, Boston, Massachusetts