US Cost-effectiveness of Lenacapavir plus Optimized Background Regimen versus Other Treatments for Multi-drug Resistant HIV

EE472

Vardanega V,[1] New E,[2] Mezzio D,[3] Eddowes LA[1]

[1]Costello Medical, London, United Kingdom, [2]Costello Medical, Singapore, [3]Gilead Sciences, California, United States

Key Findings

- **Lenacapavir (LEN) + optimized background regimen (OBR) was associated with additional quality-adjusted life-years (QALYs) and lower costs vs fostemsavir (FTR)+OBR and ibalizumab (IBA)+OBR and was therefore dominant (Table 1).**
- **LEN+OBR remained below a $50,000/QALY willingness-to-pay threshold for pairwise comparisons vs FTR+OBR and IBA+OBR in all deterministic sensitivity analyses (DSAs). Figure 2 shows the inputs with the largest impact on the incremental cost-effectiveness ratio (ICER) for LEN+OBR vs FTR+OBR and IBA+OBR.**
- **In probabilistic sensitivity analyses (PSAs), LEN+OBR was dominant or below the $50,000/QALY threshold for 91.5% of runs vs FTR+OBR and 90.3% of runs vs IBA+OBR (Figure 3), indicating LEN+OBR had a high probability of being cost-effective vs relevant comparators.**

Conclusions

LEN+OBR dominated FTR+OBR and IBA+OBR; LEN was more favorable in terms of both costs and health outcomes compared with other available therapies when considering options for optimizing treatment for heavily treatment-experienced (HTE) adults with multi-drug resistant (MDR) HIV-1.

Background

- LEN is a first-in-class multi-stage HIV capsid inhibitor administered subcutaneously as a long-acting injectable every 6 months, indicated in combination with an OBR for the treatment of HIV-1 in HTE adults with MDR HIV-1 failing their current antiretroviral regimen due to resistance, intolerance, or safety considerations.[1]
- With limited treatment options available, this population has historically received highly individualized, complex and often ineffective treatment regimens, leading to high risk of virologic failure and mortality.[2]
- In the Phase 2/3 CAPELLA study (NCT04150068), LEN demonstrated high rates of virologic suppression for HTE people with HIV (PWH) with MDR through to Week 52.[3,4]
- In a previous analysis, LEN+OBR was associated with improved long-term clinical benefits in terms of virologically-suppressed life-years (LYs), AIDS-free LYs and QALYs gained vs relevant comparators in this population.[5]

References: 1. Food and Drug Administration. SUNLENCA® (lenacapavir) prescribing information 2023; **2.** Priest J, et al. Open Forum Infect Dis 2021; **3.** Segal-Maurer S, et al. New Engl J Med 2022;386:1793–1803; **4.** Gilead Data on File 2022; **5.** Vardanega V, et al. ISPOR 2022; **6.** Chatzidaki I, et al. Value in Health 2022.

Acknowledgements: This study was funded by Gilead Sciences. The authors acknowledge Xanthe Wignall for medical writing and editorial assistance. All costs associated with development of this poster were funded by Gilead Sciences.

Disclosures: **DM** is an employee of Gilead Sciences. **VV**, **EN** and **LAE** are employees of Costello Medical, which was funded to conduct the study by Gilead Sciences.

Correspondence: Dylan Mezzio (dylan.mezzio@gilead.com).

Objective

- To determine the cost-effectiveness of LEN+OBR vs FTR+OBR and IBA+OBR for the treatment of HTE PWH from a US commercial healthcare payer perspective.

Methods

Model framework

- A Markov state-transition model compared LEN+OBR vs FTR+OBR and IBA+OBR in PWH with MDR, from a US commercial healthcare payer perspective.
- The base case model used a lifetime time horizon (70 years) and a 3.0% annual discount rate for costs and benefits.
- As shown in Figure 1, PWH in the model occupied health states stratified by two to three viral load categories and four CD4 cell count categories within two lines of therapy: intervention/comparator and salvage therapy.
- PWH in the model accrued costs associated with treatment, disease management and AIDS-related events. These costs were determined by CD4 cell count.

Model inputs

- Transition probabilities between viral load categories were based on extrapolated viral load data from the CAPELLA study for LEN+OBR and on relative efficacy parameters obtained from indirect treatment comparisons (ITCs) for comparators.[4,6]
- Transition probabilities for CD4 cell count were dependent on viral load (PWH with viral load >200 copies/mL are more likely to transition to lower CD4 cell counts than virologically suppressed PWH) and were assumed to remain constant over time.
- Costs considered were drug acquisition costs (wholesale acquisition cost [WAC]), drug administration costs, disease management costs and AIDS-related event costs. The cost year of the study was 2021, with costs from previous years inflated to their 2021 value. Drug acquisition costs were updated in 2022 when the LEN WAC became publicly available.
- Other key inputs included AIDS-defining events, (dis)utilities, and mortality rates. Inputs were informed by literature reviews, ITCs, clinical opinion and internal sources.

Model outputs

- Primary outputs were total and incremental costs, LYs, QALYs, and ICERs, generated for each pairwise comparison.
- DSAs and PSAs were performed to address elements of uncertainty in the model and to explore the robustness of the results.

Limitations and Challenges

- Due to the small population size and heterogeneity within the PWH with MDR population, there are some uncertainties related to the derived probabilities for transition between different health states, as clinical data in this population are limited.
- The treatment of PWH with MDR is highly complex, and it was not possible to capture all possible OBR compositions, treatment switches, costs associated with switching treatments and effects on treatment efficacy in the model. However, the inputs were carefully chosen to simulate conditions that closely reflect the clinical reality of treating PWH with MDR and these can be easily adapted to the changing HIV treatment landscape.

Figure 1. Top-line overview of the model structure, indicating how PWH move through the therapy lines

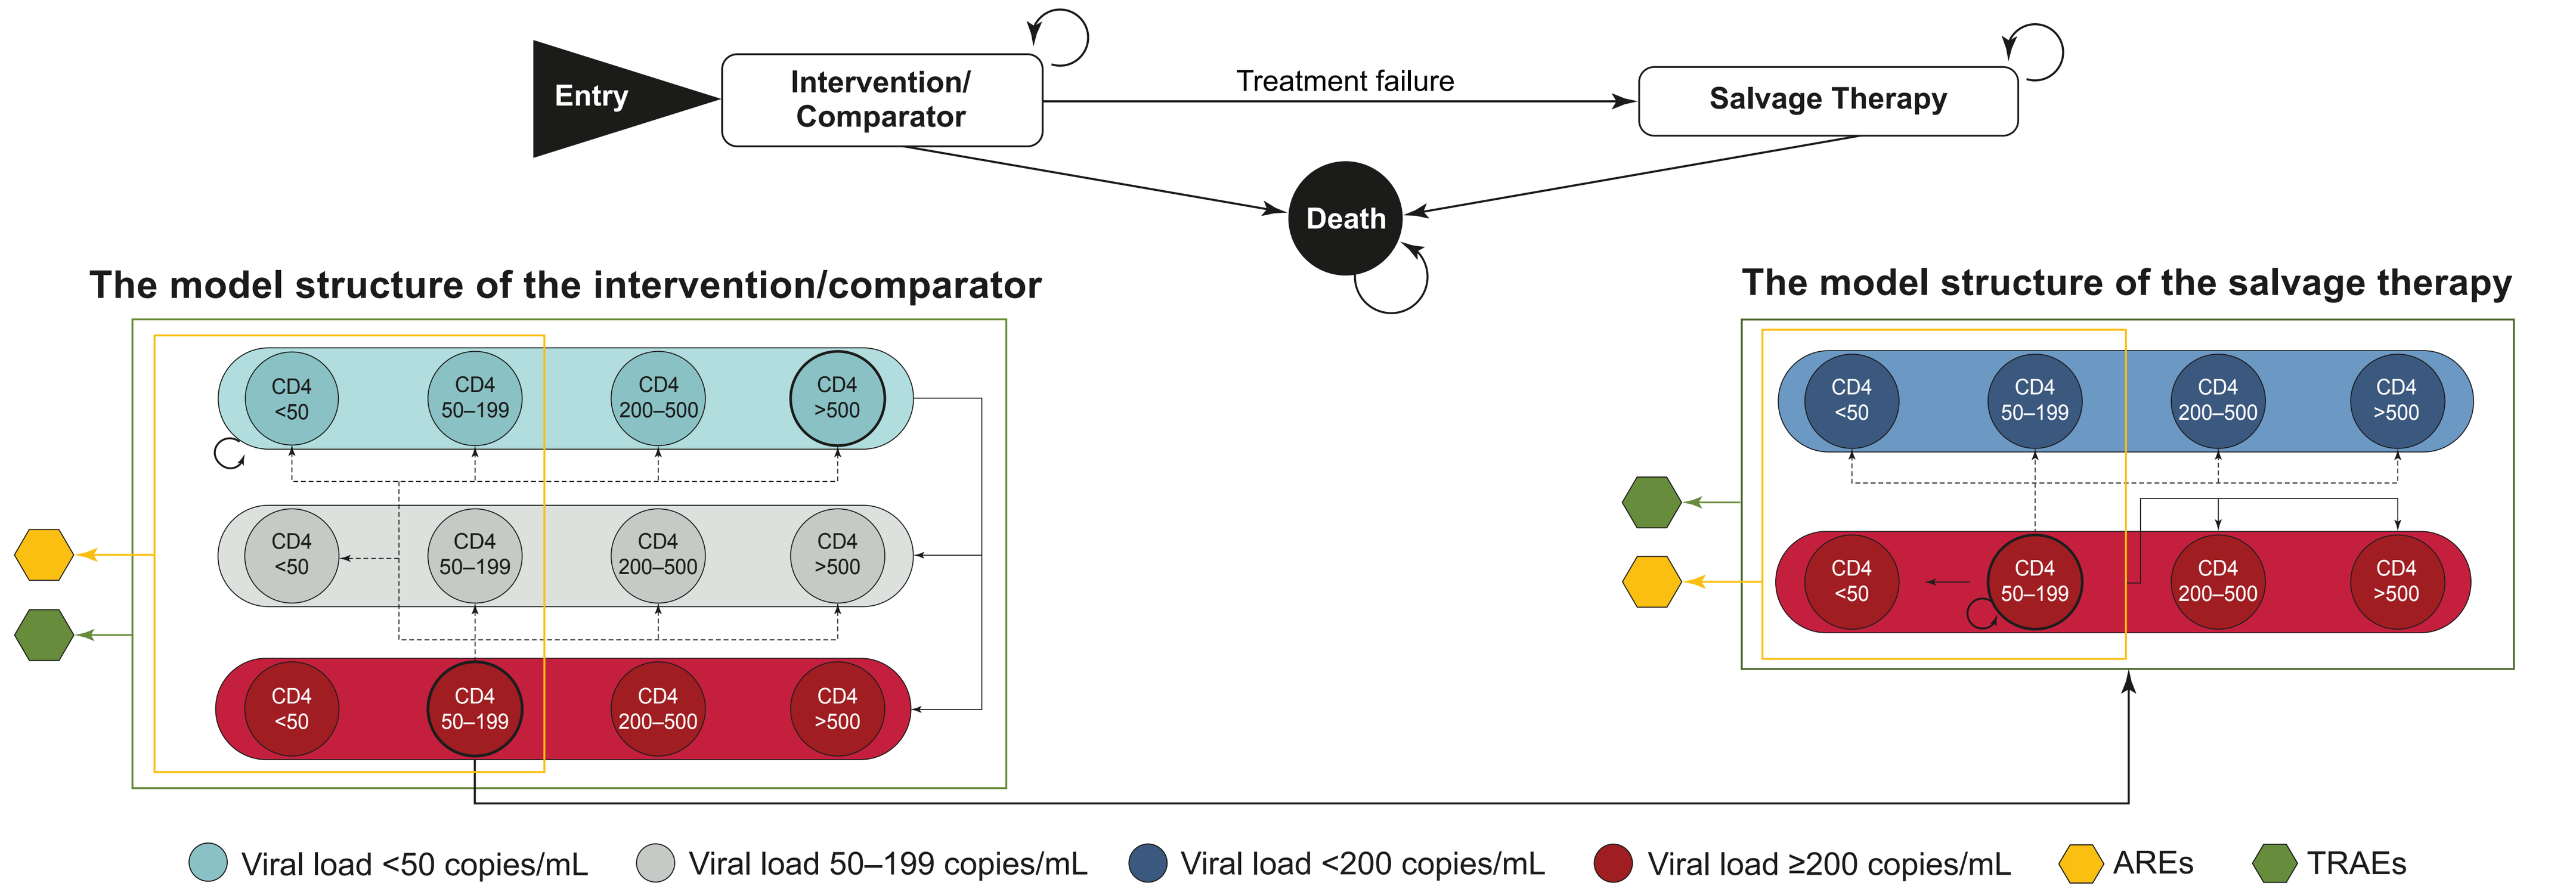
Entry
Intervention/ Comparator
Treatment failure
Salvage Therapy
Death
The model structure of the intervention/comparator
The model structure of the salvage therapy
CD4 <50
CD4 50–199
CD4 200–500
CD4 >500
Viral load <50 copies/mL
Viral load 50–199 copies/mL
Viral load <200 copies/mL
Viral load ≥200 copies/mL
AREs
TRAEs

Abbreviations: ARE, AIDS-related event; TRAE, Treatment-related adverse event. **Footnotes:** Dashed arrows show transitions that can occur only in the first model cycle after initiation of the treatment line; circular arrows indicate that PWH can remain in the state for more than one cycle. CD4 cell count-related health states are color coded to indicate the viral load category. Health states are represented with a circle shape, events with a hexagon shape. PWH can transition out the intervention/comparator line and move into salvage therapy. Transitions allowed from two health states (i.e. CD4 cell count 50–199 cells/μL with viral load ≥200 copies/mL, and CD4 cell count >500 cells/μL with viral load <50 copies/mL, both shown with a bold outline) are shown as an example of how PWH could flow through the model for the intervention/comparator line and salvage therapy lines; similar transitions from other health states were also possible. AREs and TRAEs are events that PWH can experience without leaving their current health state. Experiencing events, as opposed to transition to a different health state, are represented with a non-black line.

Figure 2. DSA ICER tornado diagrams for LEN+OBR pairwise comparisons vs FTR+OBR and IBA+OBR

LEN+OBR vs FTR+OBR

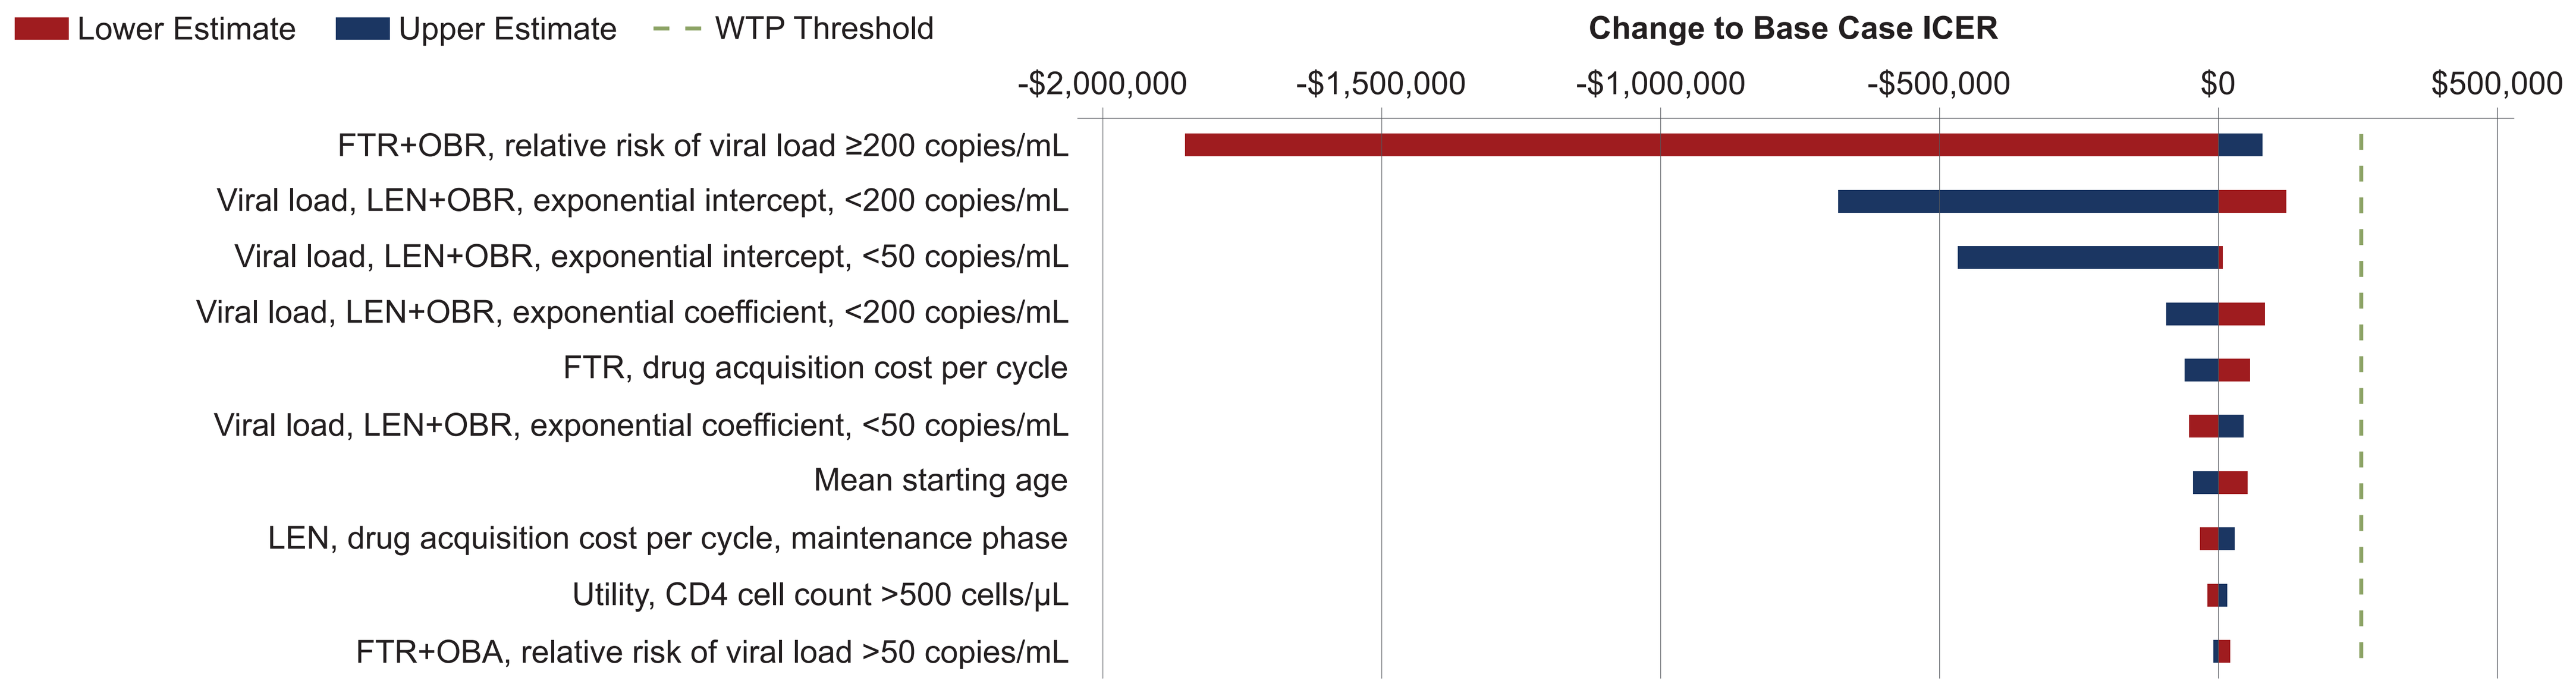
Lower Estimate
Upper Estimate
WTP Threshold
Change to Base Case ICER
-$2,000,000
-$1,500,000
-$1,000,000
-$500,000
$0
$500,000
FTR+OBR, relative risk of viral load ≥200 copies/mL
Viral load, LEN+OBR, exponential intercept, <200 copies/mL
Viral load, LEN+OBR, exponential intercept, <50 copies/mL
Viral load, LEN+OBR, exponential coefficient, <200 copies/mL
FTR, drug acquisition cost per cycle
Viral load, LEN+OBR, exponential coefficient, <50 copies/mL
Mean starting age
LEN, drug acquisition cost per cycle, maintenance phase
Utility, CD4 cell count >500 cells/μL
FTR+OBA, relative risk of viral load >50 copies/mL

LEN+OBR vs IBA+OBR

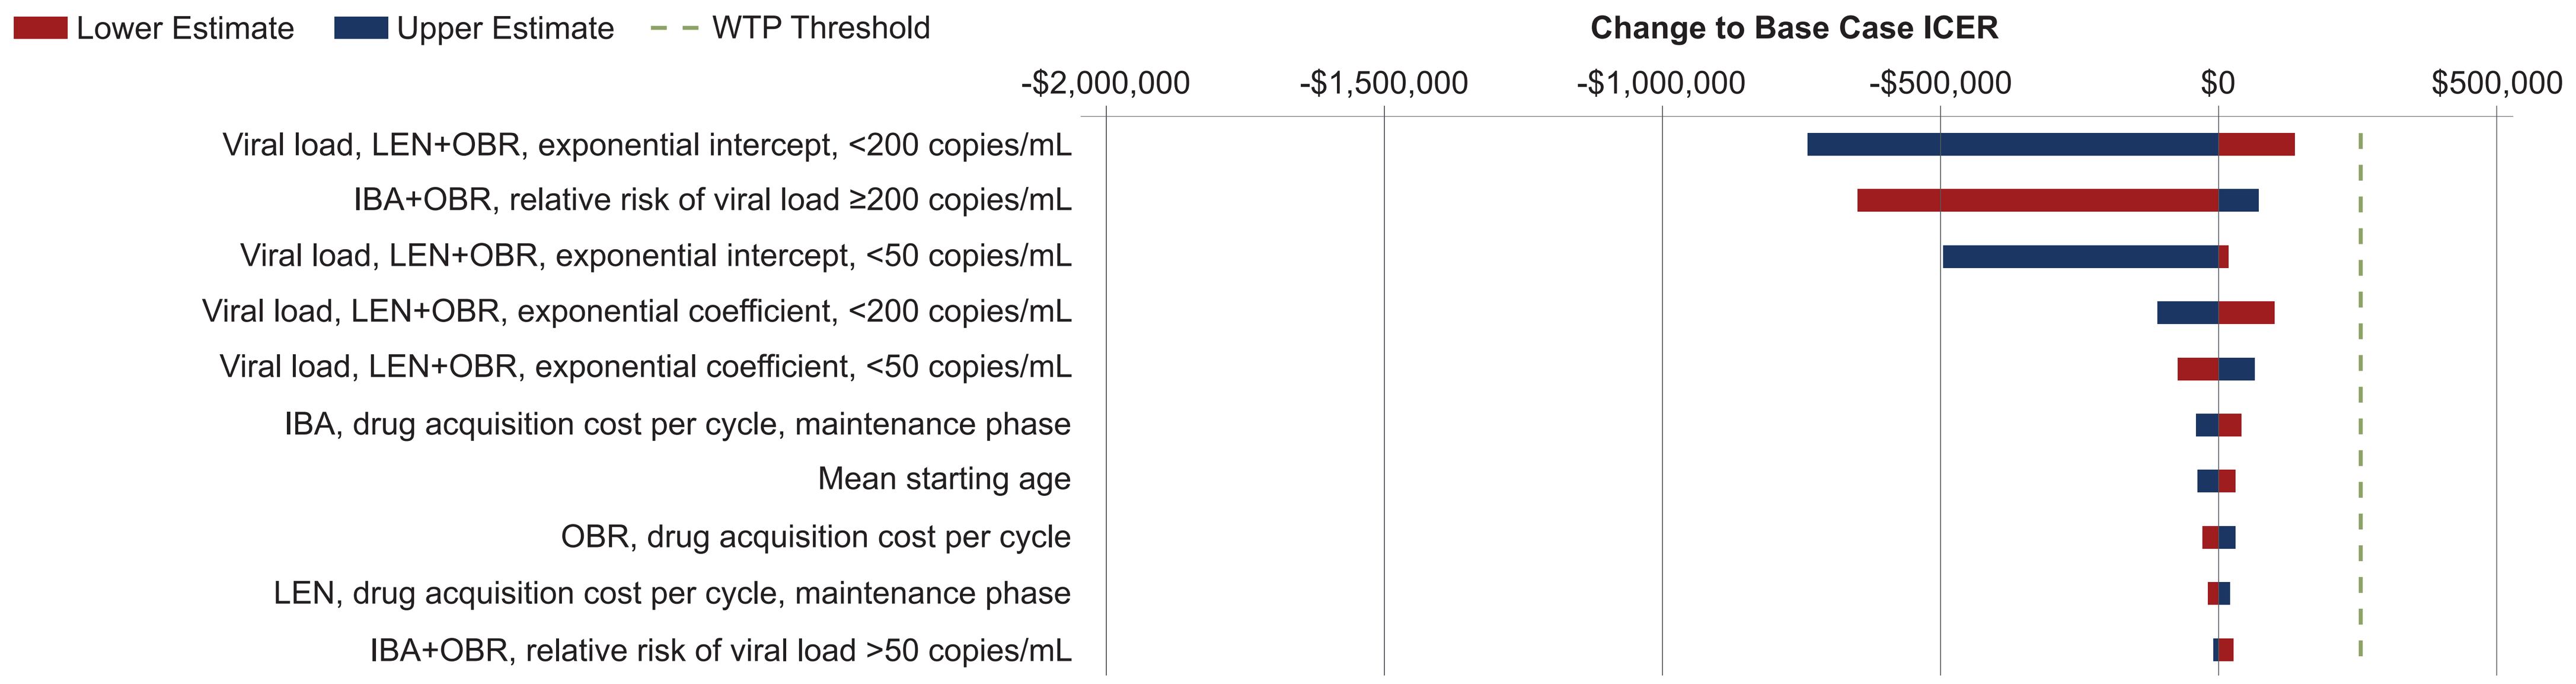
Lower Estimate
Upper Estimate
WTP Threshold
Change to Base Case ICER
-$2,000,000
-$1,500,000
-$1,000,000
-$500,000
$0
$500,000
Viral load, LEN+OBR, exponential intercept, <200 copies/mL
IBA+OBR, relative risk of viral load ≥200 copies/mL
Viral load, LEN+OBR, exponential intercept, <50 copies/mL
Viral load, LEN+OBR, exponential coefficient, <200 copies/mL
Viral load, LEN+OBR, exponential coefficient, <50 copies/mL
IBA, drug acquisition cost per cycle, maintenance phase
Mean starting age
OBR, drug acquisition cost per cycle
LEN, drug acquisition cost per cycle, maintenance phase
IBA+OBR, relative risk of viral load >50 copies/mL

Abbreviations: DSA, Deterministic sensitivity analysis; FTR, Fostemsavir; IBA, Ibalizumab; ICER, Incremental cost-effectiveness ratio; LEN, Lenacapavir; OBR, Optimized background regimen; WTP, Willingness-to-pay.

Table 1. Base case cost-effectiveness results

	Treatment		
	LEN+OBR	**FTR+OBR**	**IBA+OBR**
Total Costs	$1,481,737	$1,660,940	$1,734,783
Total LYs	12.90	11.91	11.32
Total QALYs	10.06	9.27	8.80
Inc. Costs	–	-$179,203	-$253,046
Inc. LYs	–	0.99	1.58
Inc. QALYs	–	0.79	1.26
ICER	–	LEN+OBR is Dominant	LEN+OBR is Dominant

Abbreviations: FTR, Fostemsavir; IBA, Ibalizumab; ICER, Incremental cost-effectiveness ratio; Inc, Incremental; OBR, Optimized background regimen; LEN, Lenacapavir; LY, Life year; QALY, Quality-adjusted life year.

Figure 3. PSA scatter plots for LEN+OBR pairwise comparisons vs FTR+OBR and IBA+OBR

LEN+OBR vs FTR+OBR

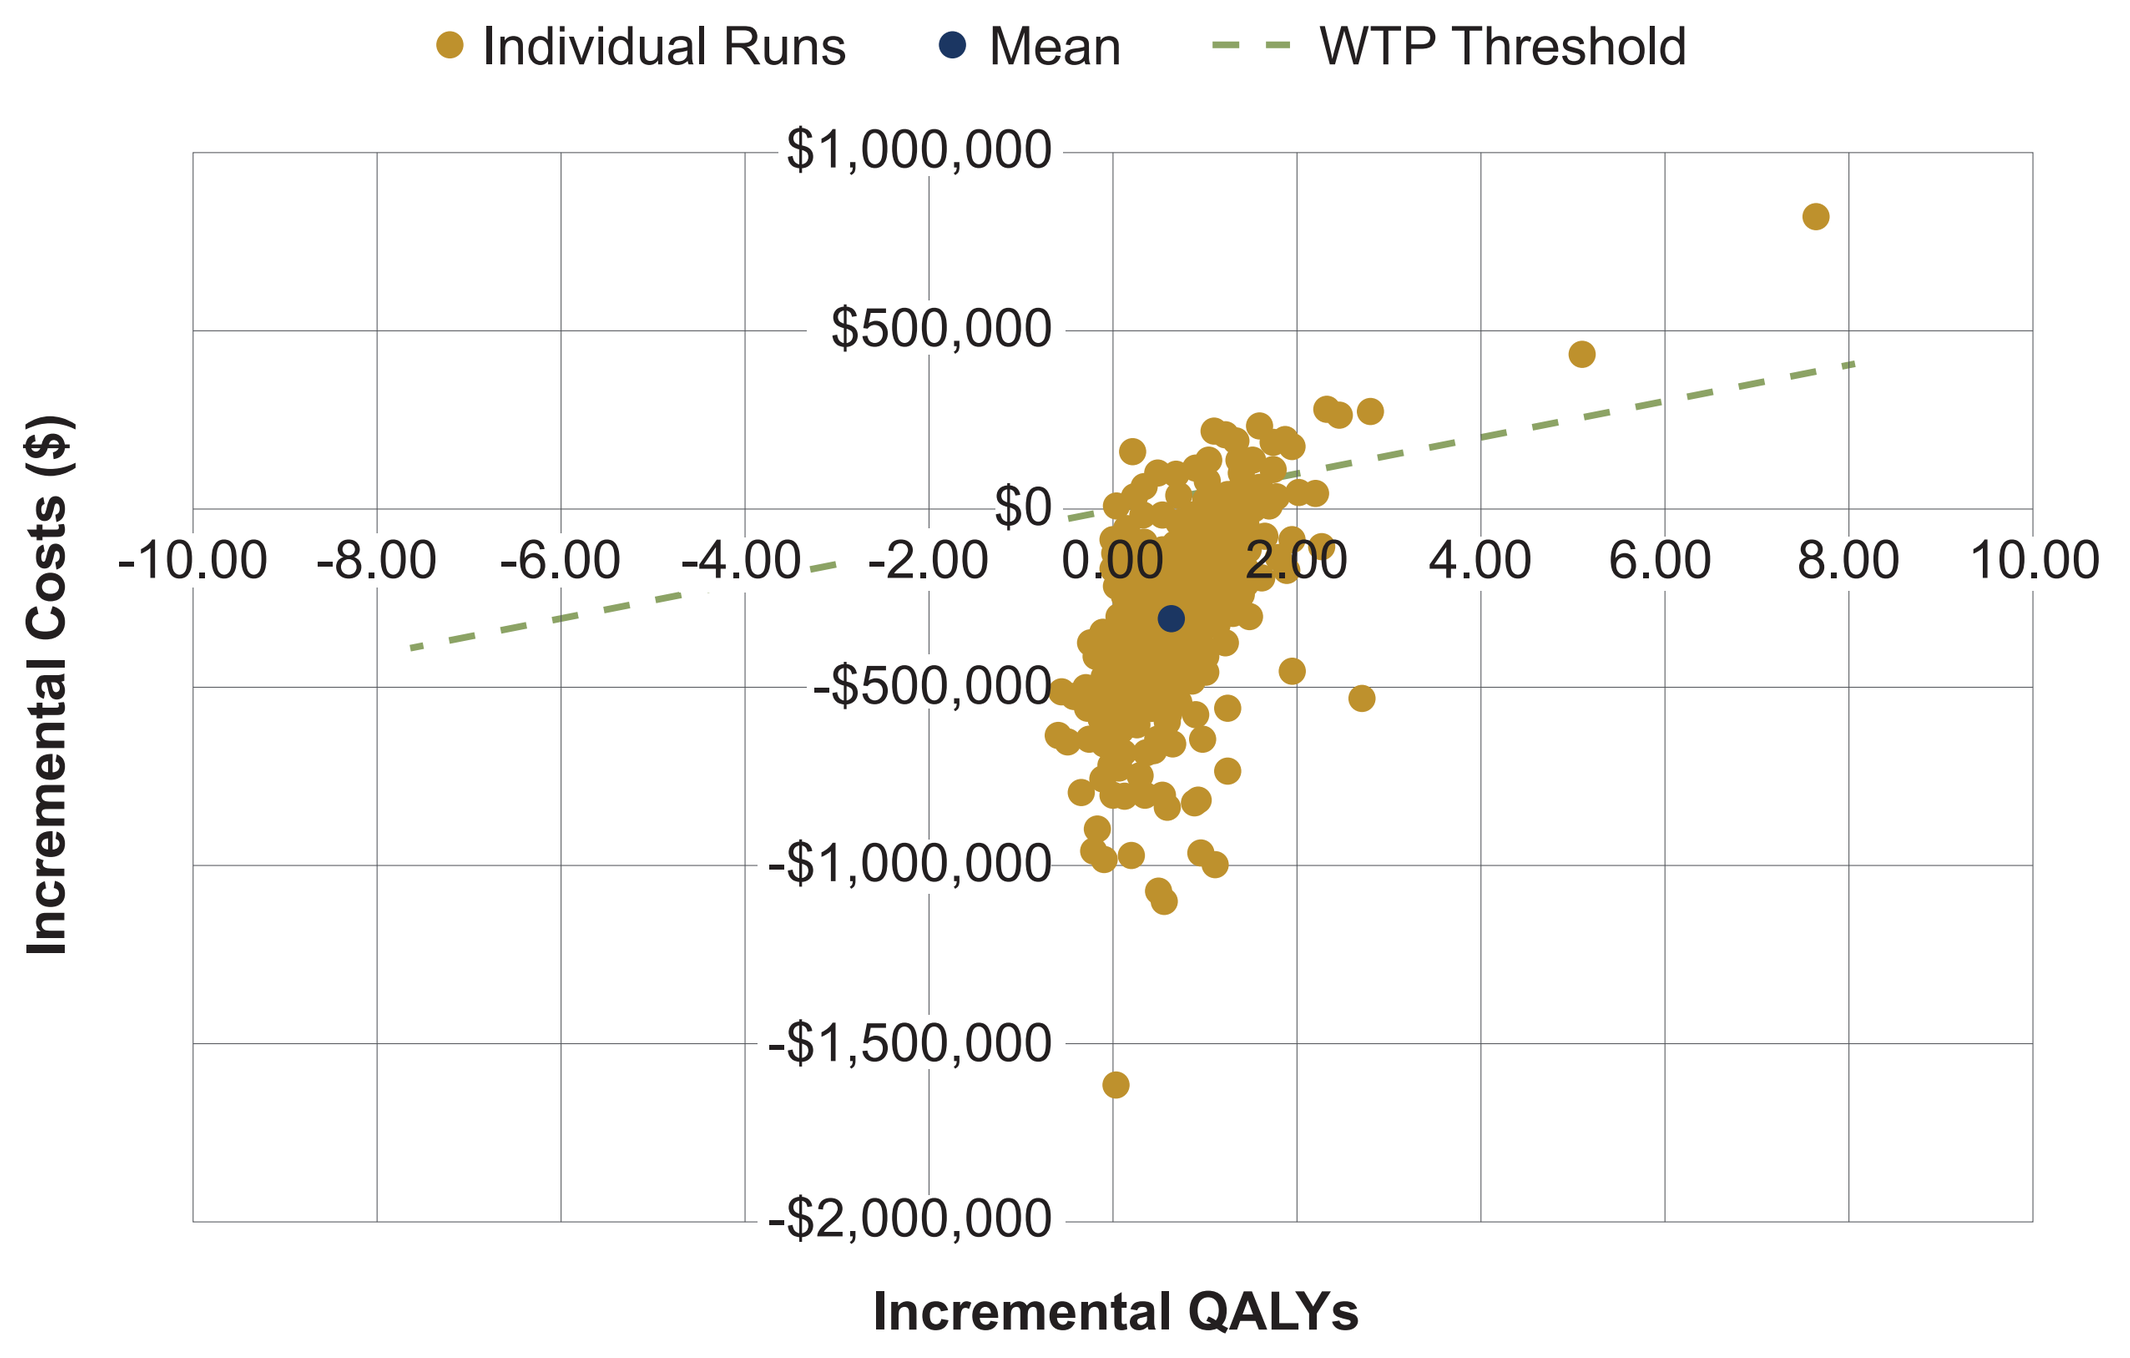
Individual Runs
Mean
WTP Threshold
$1,000,000
$500,000
$0
-$500,000
-$1,000,000
-$1,500,000
-$2,000,000
-10.00
-8.00
-6.00
-4.00
-2.00
0.00
2.00
4.00
6.00
8.00
10.00
Incremental Costs ($)
Incremental QALYs

LEN+OBR vs IBA+OBR

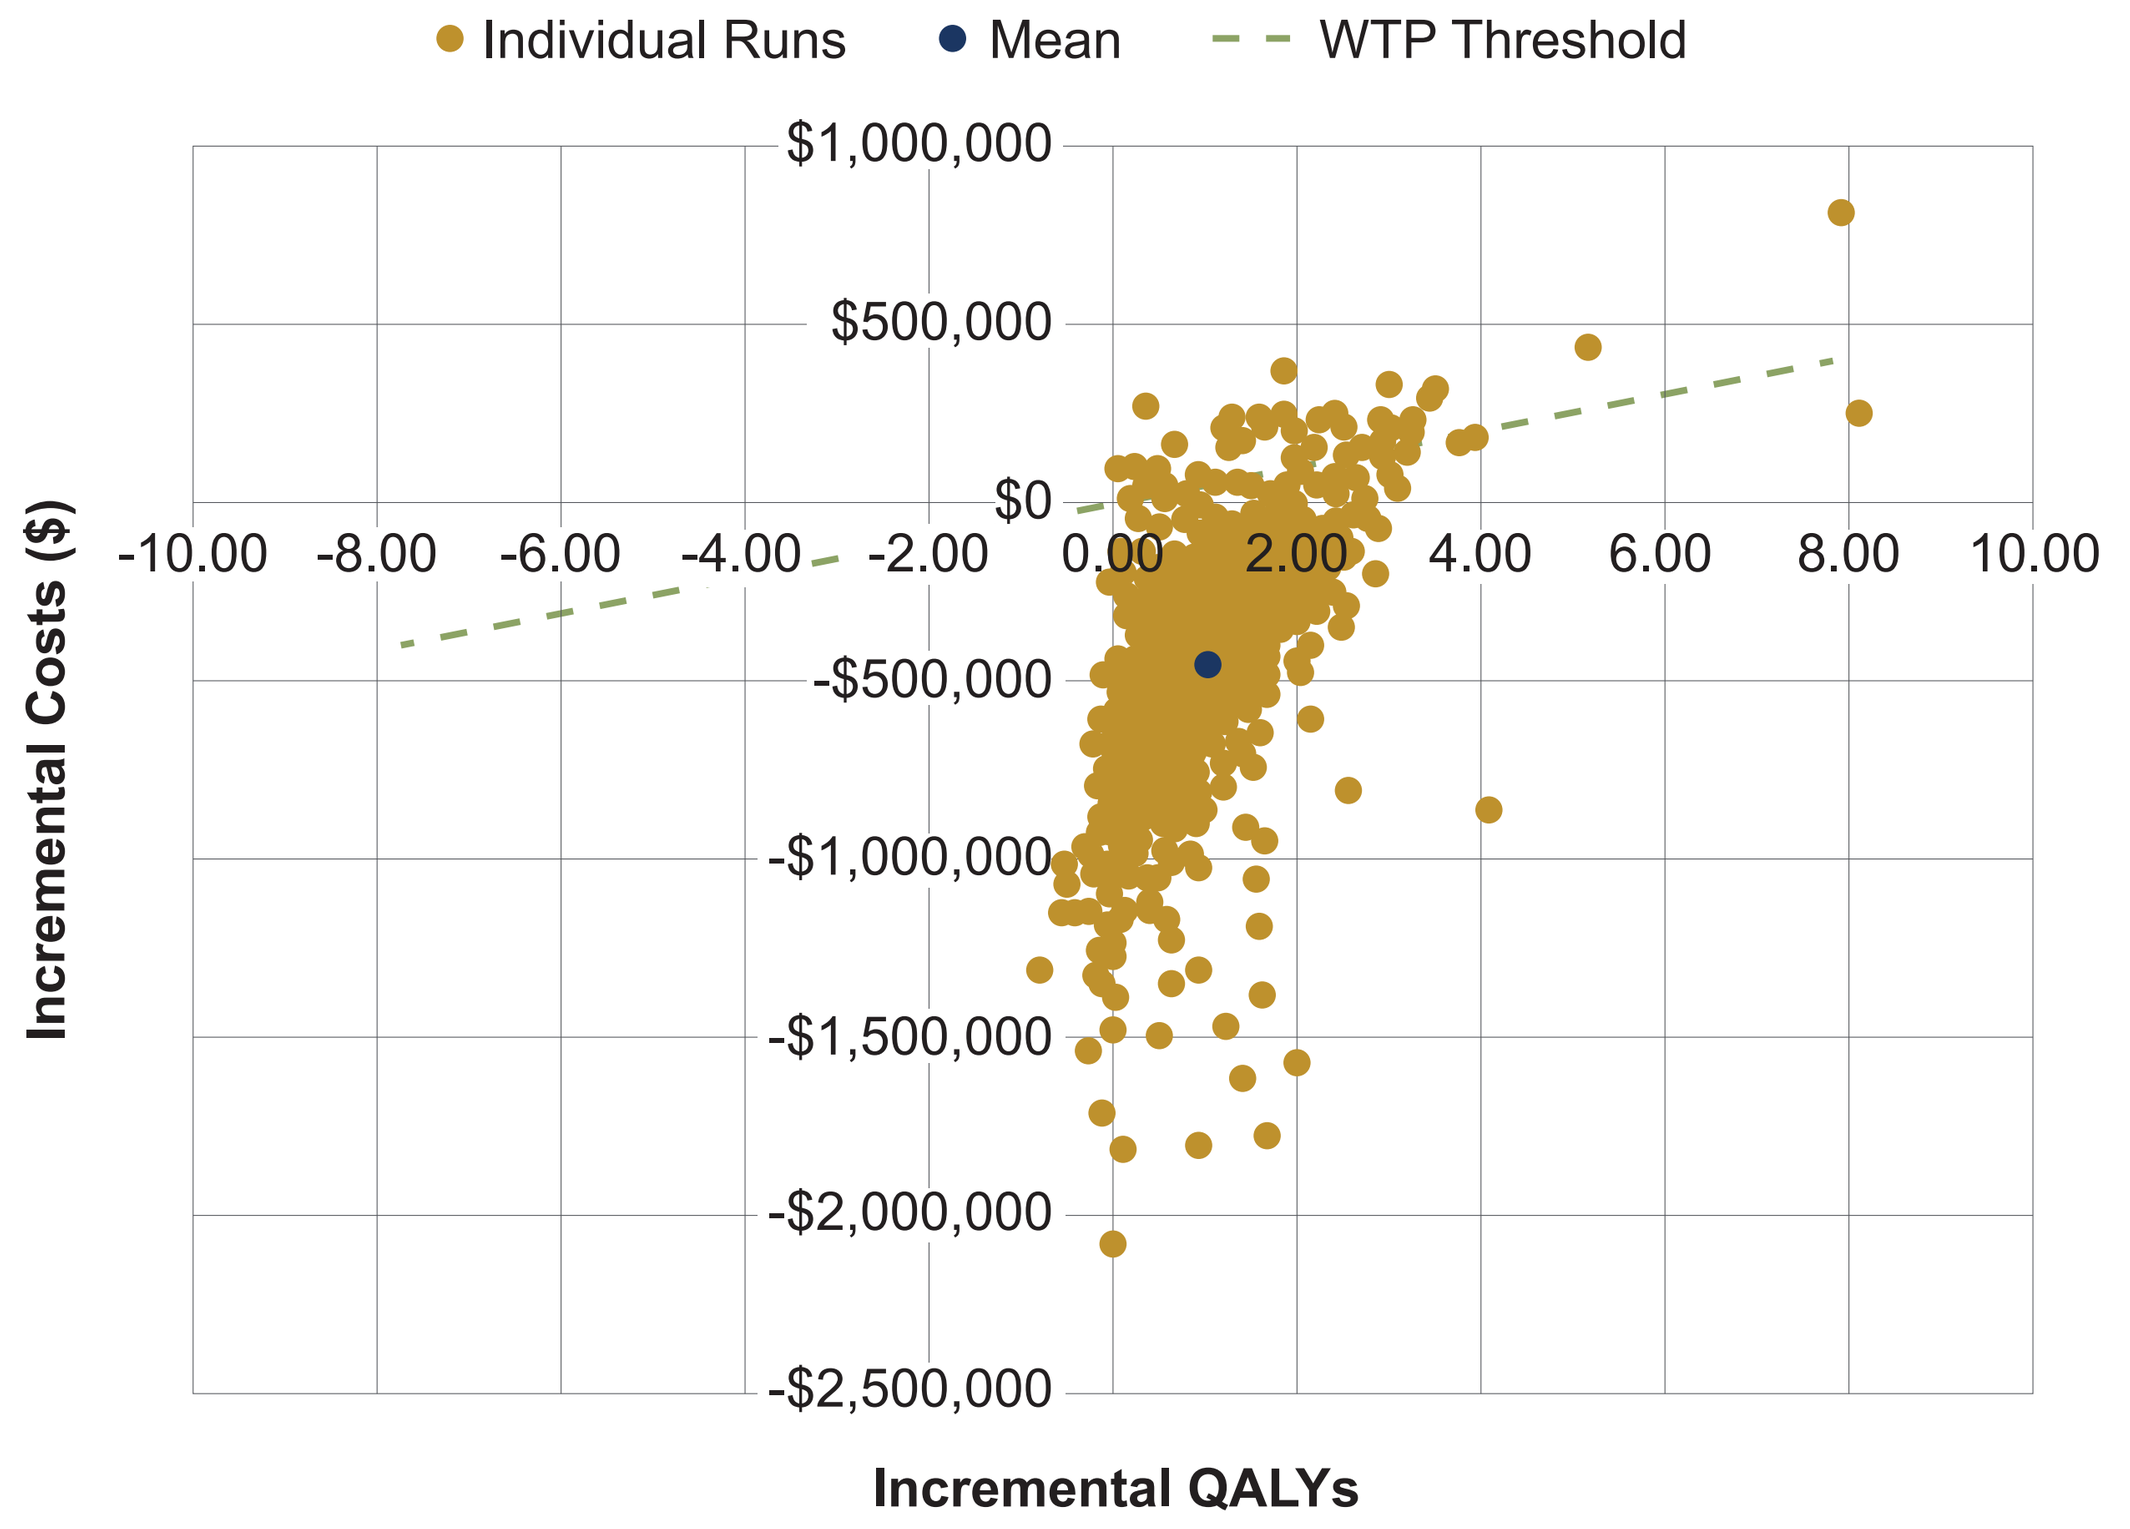
Individual Runs
Mean
WTP Threshold
$1,000,000
$500,000
$0
-$500,000
-$1,000,000
-$1,500,000
-$2,000,000
-$2,500,000
-10.00
-8.00
-6.00
-4.00
-2.00
0.00
2.00
4.00
6.00
8.00
10.00
Incremental Costs ($)
Incremental QALYs

Abbreviations: FTR, Fostemsavir; IBA, Ibalizumab; LEN, Lenacapavir; OBR, Optimized background regimen; PSA, Probabilistic sensitivity analysis; QALY, Quality-adjusted life year; WTP, Willingness-to-pay.

Presented at ISPOR: May 7–10, 2023; Boston, MA, USA