EPH169

Persons Diagnosed with COVID-19 in Linked Clinical Practice Research Datalink (CPRD) – Hospital Episode Statistics (HES) Data: A Cohort Description

Kathleen M. Andersen,[1] Maya Reimbaeva,[1] Leah J. McGrath,[1] Jennifer L. Nguyen,[1]
Kiran Rai,[3] Theo Tritton,[3] Carmen Tsang,[2] Jingyan Yang[1,4]

[1] Pfizer Inc., New York, NY; [2] Pfizer Ltd., Tadworth, UK; [3] Adelphi Real World, Bollington, UK; [4] Institute for Social and Economic Research and Policy, Columbia University, New York, NY.

INTRODUCTION

CPRD Aurum captures primary care electronic healthcare records for ~28% of the population in England and can be linked to HES secondary care data.

From August 2020 – March 2022, all SARS-CoV-2 PCR tests were reported back to the patient's GP, making the CPRD a closed system for COVID research questions.

OBJECTIVE

To describe the demographic and clinical characteristics, as well as social deprivation, of persons **hospitalized with COVID in England between August 1, 2020 - March 31, 2021.**

METHODS

We defined persons with COVID as those recorded in primary care from August 1, 2020-March 31, 2021.

We required continuous GP practice registration for ≥365 days prior to diagnosis, and eligibility for linkage to HES Admitted Patient Care data.

Hospitalizations for COVID were defined as persons admitted with a primary diagnosis of COVID (ICD-10 U07.1) within 12 weeks of the primary care diagnosis record.

RESULTS

Our cohort included 535,453 persons diagnosed in primary care (outpatient setting) with COVID, **with 2% later hospitalized for COVID.**

Consistent with other studies, hospitalized COVID patients were disproportionately:

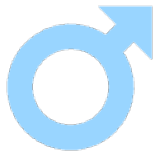

Male (57%)

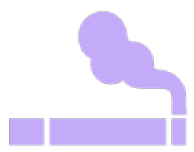

Current or former smokers (42%)

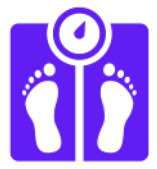

Overweight or obese (53%)

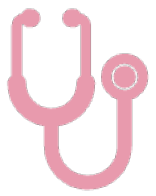

People with comorbid conditions
(46% Charlson Comorbidity Index ≥1)

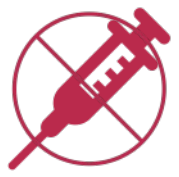

Unvaccinated
for COVID (98%)

Hospitalizations increased with age.

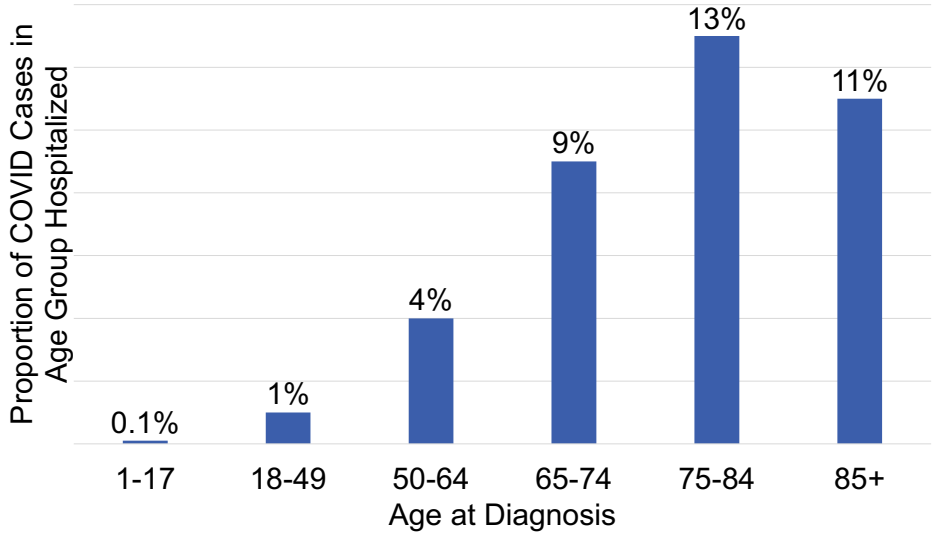
Proportion of COVID Cases in Age Group Hospitalized
0.1%
1%
4%
9%
13%
11%
1-17
18-49
50-64
65-74
75-84
85+
Age at Diagnosis

Persons living in socially disadvantaged areas were overrepresented.

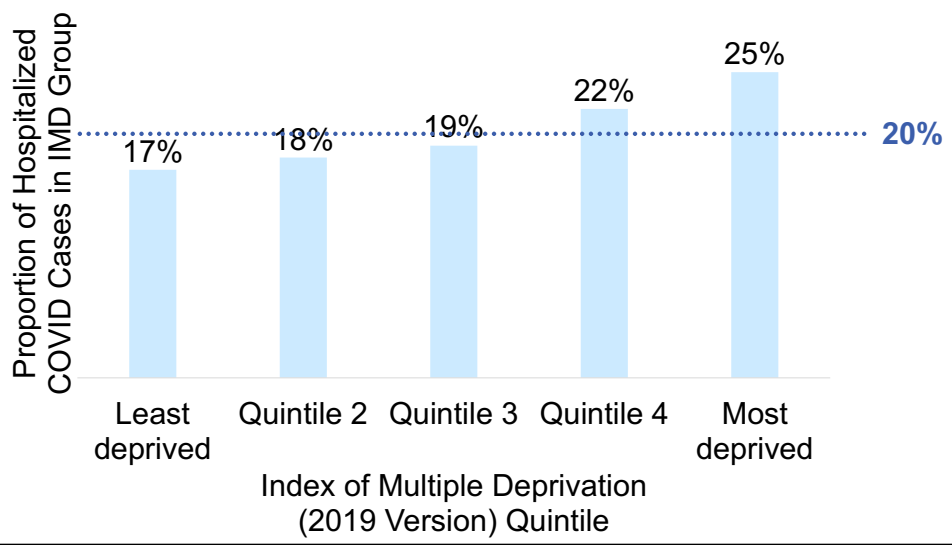
Proportion of Hospitalized COVID Cases in IMD Group
17%
18%
19%
22%
25%
20%
Least deprived
Quintile 2
Quintile 3
Quintile 4
Most deprived
Index of Multiple Deprivation (2019 Version) Quintile

Presented at ISPOR; May 9, 2023; Boston, Massachusetts

CONCLUSION

This study sample showed comparable characteristics to other published evidence of hospitalized COVID patients and thus indicates this CPRD-HES linked dataset is a valid source of data for studies on COVID.

This work can contextualize future efforts to describe healthcare utilization and costs in persons diagnosed with COVID in England.

LIMITATION OF HES DATA

Hospitalizations have changed since Spring 2021, with periods of increased (delta variant) & decreased (omicron) risk of hospital admission, as well the availability of therapeutics.

Acknowledgments

Bethany Backhouse, Poppy Payne, Elke Rottier and Robert Wood from Adelphi Real World, Tamuno Alfred, Darren Kailung Jeng, Deepa Malhotra, Tendai Mugwagwa, Qiao Mu and Chern Chuan Soo from Pfizer Inc., Agnieszka Gajewska, Tomasz Mikołajczyk and Ewa Śleszyńska-Dopiera from Quanticate, and Andy Surinach from Genesis Research.

Disclosures

This study was funded by Pfizer Inc. Dr Andersen, Ms Reimbaeva, Dr McGrath, Dr Mendes, Dr Mugwagwa, Dr Nguyen, Dr Tsang and Dr Yang are employees of Pfizer Inc or Pfizer Ltd and may hold Pfizer stock or stock options. Dr Rai and Mr Tritton are employees of Adelphi Real World which has received consulting fees from Pfizer Inc.

Contact and link to preprint

Kathleen M. Andersen, PhD MSc
Vaccines Real World Evidence
kathleen.andersen@pfizer.com