Poster #CO164

Yong Mei Yin
ymyin@njmu.edu.cn

Ribociclib versus CDK4/6 inhibitors and endocrine therapies (ET) for first line (1L), metastatic or advanced HR+ HER2- pre- and post-menopausal breast cancer in Asia: A systematic review and network meta-analysis

Yin Y[1], Liang Y[1], Sun C[1], Yang F[1], Li C[2], Yu J[2], Sun R[2], Mondal S[3], Tai T[4], Ting B[5]

[1]Department of Oncology, The First Affiliated Hospital of Nanjing Medical University, Nanjing, China; [2]Novartis Pharmaceuticals, Shanghai, China; [3]Novartis Healthcare Pvt. Ltd, Hyderabad, India; [4]Novartis Global Service Centre, Dublin, Ireland; [5]Novartis Corporate Center, Kuala Lumpur, Malaysia

Scan to obtain:
- Poster

https://www.medicalcongressposters.com//Default.aspx?doc=78d4a

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

BACKGROUND

- Among all subtypes of breast cancer, approximately 68% are HR+ (i.e. estrogen receptors [ERs] and progesterone receptors [PRs])/HER2-[1].
- Treatments used to treat HR+/HER2– advanced breast cancer include cyclin-dependent kinase 4/6 (CDK4/6) inhibitors, endocrine therapies (ET), chemotherapies, and targeted agents[2].
- Studies have showed that the addition of a CDK4/6 inhibitor to ET provided a greater benefit regarding progression-free survival (PFS) than ET alone in pre/post- menopausal patients with advanced HR+/HER2– breast cancer[3].
- The magnitude of PFS benefit is ethnicity-dependent and it is different between Asian and non-Asian population in first line treatment[4]
- In China four CDK 4/6 inhibitors abemaciclib (ABE), palbociclib (PAL), ribociclib (RIBO) and dalpiciclib (DAL) approved for treating breast cancer[5].

OBJECTIVE

- The objective was to compare RIBO versus CDK4/6 inhibitors and ET in patients with the first line pre/post-menopausal HR+/HER2-metastatic/advanced breast cancer patients in Asia.

METHODS

- Literature search was conducted using key words in English (Embase, MEDLINE, Cochrane, and MEDLINE in-process) and Chinese (Wanfang) language literature databases from 2007 onwards up to September 2022 (**Table 1**).
- Data reported on Asia population was extracted and considered for Bayesian network meta-analysis (NMA), comparing overall survival (OS) and PFS
- Both fixed and random effects models were fitted to account for different assumptions regarding heterogeneity of treatment effects for post-menopausal whereas for pre-menopausal there was only one trial per contrast thus fixed effect was feasible.

Table 1. Inclusion criteria for study selection

Criteria	Pre/peri menopausal	Post-menopausal
Patient population	• Disease: HR+/HER2- advanced/metastatic, pre/peri menopausal breast cancer • Age: Adults (≥18 years) • Line of therapy: First line • Gender: Any • Race: Any	• Disease: HR+/HER2 advanced/metastatic post-menopausal breast cancer • Age: Adults (≥18 years) • Line of therapy: First line • Gender: Any • Race: Any
Intervention/ Comparators	• CDK4/6 inhibitors: abemaciclib, ribociclib, dalpiciclib, palbociclib • Endocrine therapies:letrozole, anastrozole, exemestane, tamoxifen, fulvestrant	
Trial design	Randomized controlled trials (RCT)	
Language restrictions	English & Chinese	

RESULTS

Evidence identified from review

- Eight randomized controlled trials included in systematic literature review contributed to NMA for both post-menopausal (n=7) and pre-menopausal (n=2) inclusively (**Table 2**)
- In post-menopausal, PFS data was available for CDK4/6 inhibitors, but OS was reported only for ABE and RIBO in the Asia subgroup.
- For pre-menopausal only PFS data was available for RIBO and DAL among CDK 4/6 inhibitors whereas DAL have no OS data, so analysis was not conducted.

Table 2. An overview of survival outcome of available RCT studies in Asia region

Study	Treatment	Comparator	PFS (HR; 95% CI)	OS (HR; 95% CI)
Post-menopausal population				
MONALEESA-2[6,7]	RIBO+AI	AI	0.26 (0.13-0.52)	0.64 (0.35-1.16)
FALCON[8]	FUL500	AI	0.81 (0.43-1.50)	-
PALOMA-2[9]	PAL+AI	AI	0.49 (0.27-0.87)	-
PALOMA-4[10]	PAL+AI	AI	0.68 (0.53-0.87)	-
DAWNA-2[11]	DAL+AI	AI	0.52 (0.36-0.75)	-
MONARCH plus[12]	ABE+AI	AI	0.39 (0.26-0.59)	-
MONARCH 3[13,14]	ABE+AI	AI	0.32 (0.20-0.53)	0.63 (0.37-1.07)
Pre-menopausal population				
MONALEESA-7[15,16]	RIBO+ET	ET	0.42 (0.27-0.66)	0.64 (0.40–1.03)
DAWNA-2 [11]	DAL+ET	ET	0.53 (0.33-0.85)	-

ABE = abemaciclib, AI = aromatase inhibitors, DAL = dalpiciclib, ET = endocrine therapy, FUL500 = fulvestrant 500mg, HR= hazards ratio, PAL = palbociclib, PFS = progression free survival, RCT = randomized control trial, RIBO = ribociclib.

NMA results

Post-menopausal patients

- In post-menopausal patients, significant PFS benefit was observed with RIBO plus aromatase inhibitors (AI) compared to AI (HR: 0.26 [0.13, 0.52]), fulvestrant (HR: 0.33 [0.13, 0.81]), and PAL plus AI (HR: 0.41 [0.20, 0.83]) whereas results were numerically favouring RIBO plus AI against DAL plus AI (HR: 0.51 [0.24, 1.08]) and ABE plus AI (HR: 0.72 [0.34,1.52]) (**Figure 1** & **Figure 2**).

Figure 1. Forest plot of Progression-Free Survival among post-menopausal population in Asia region

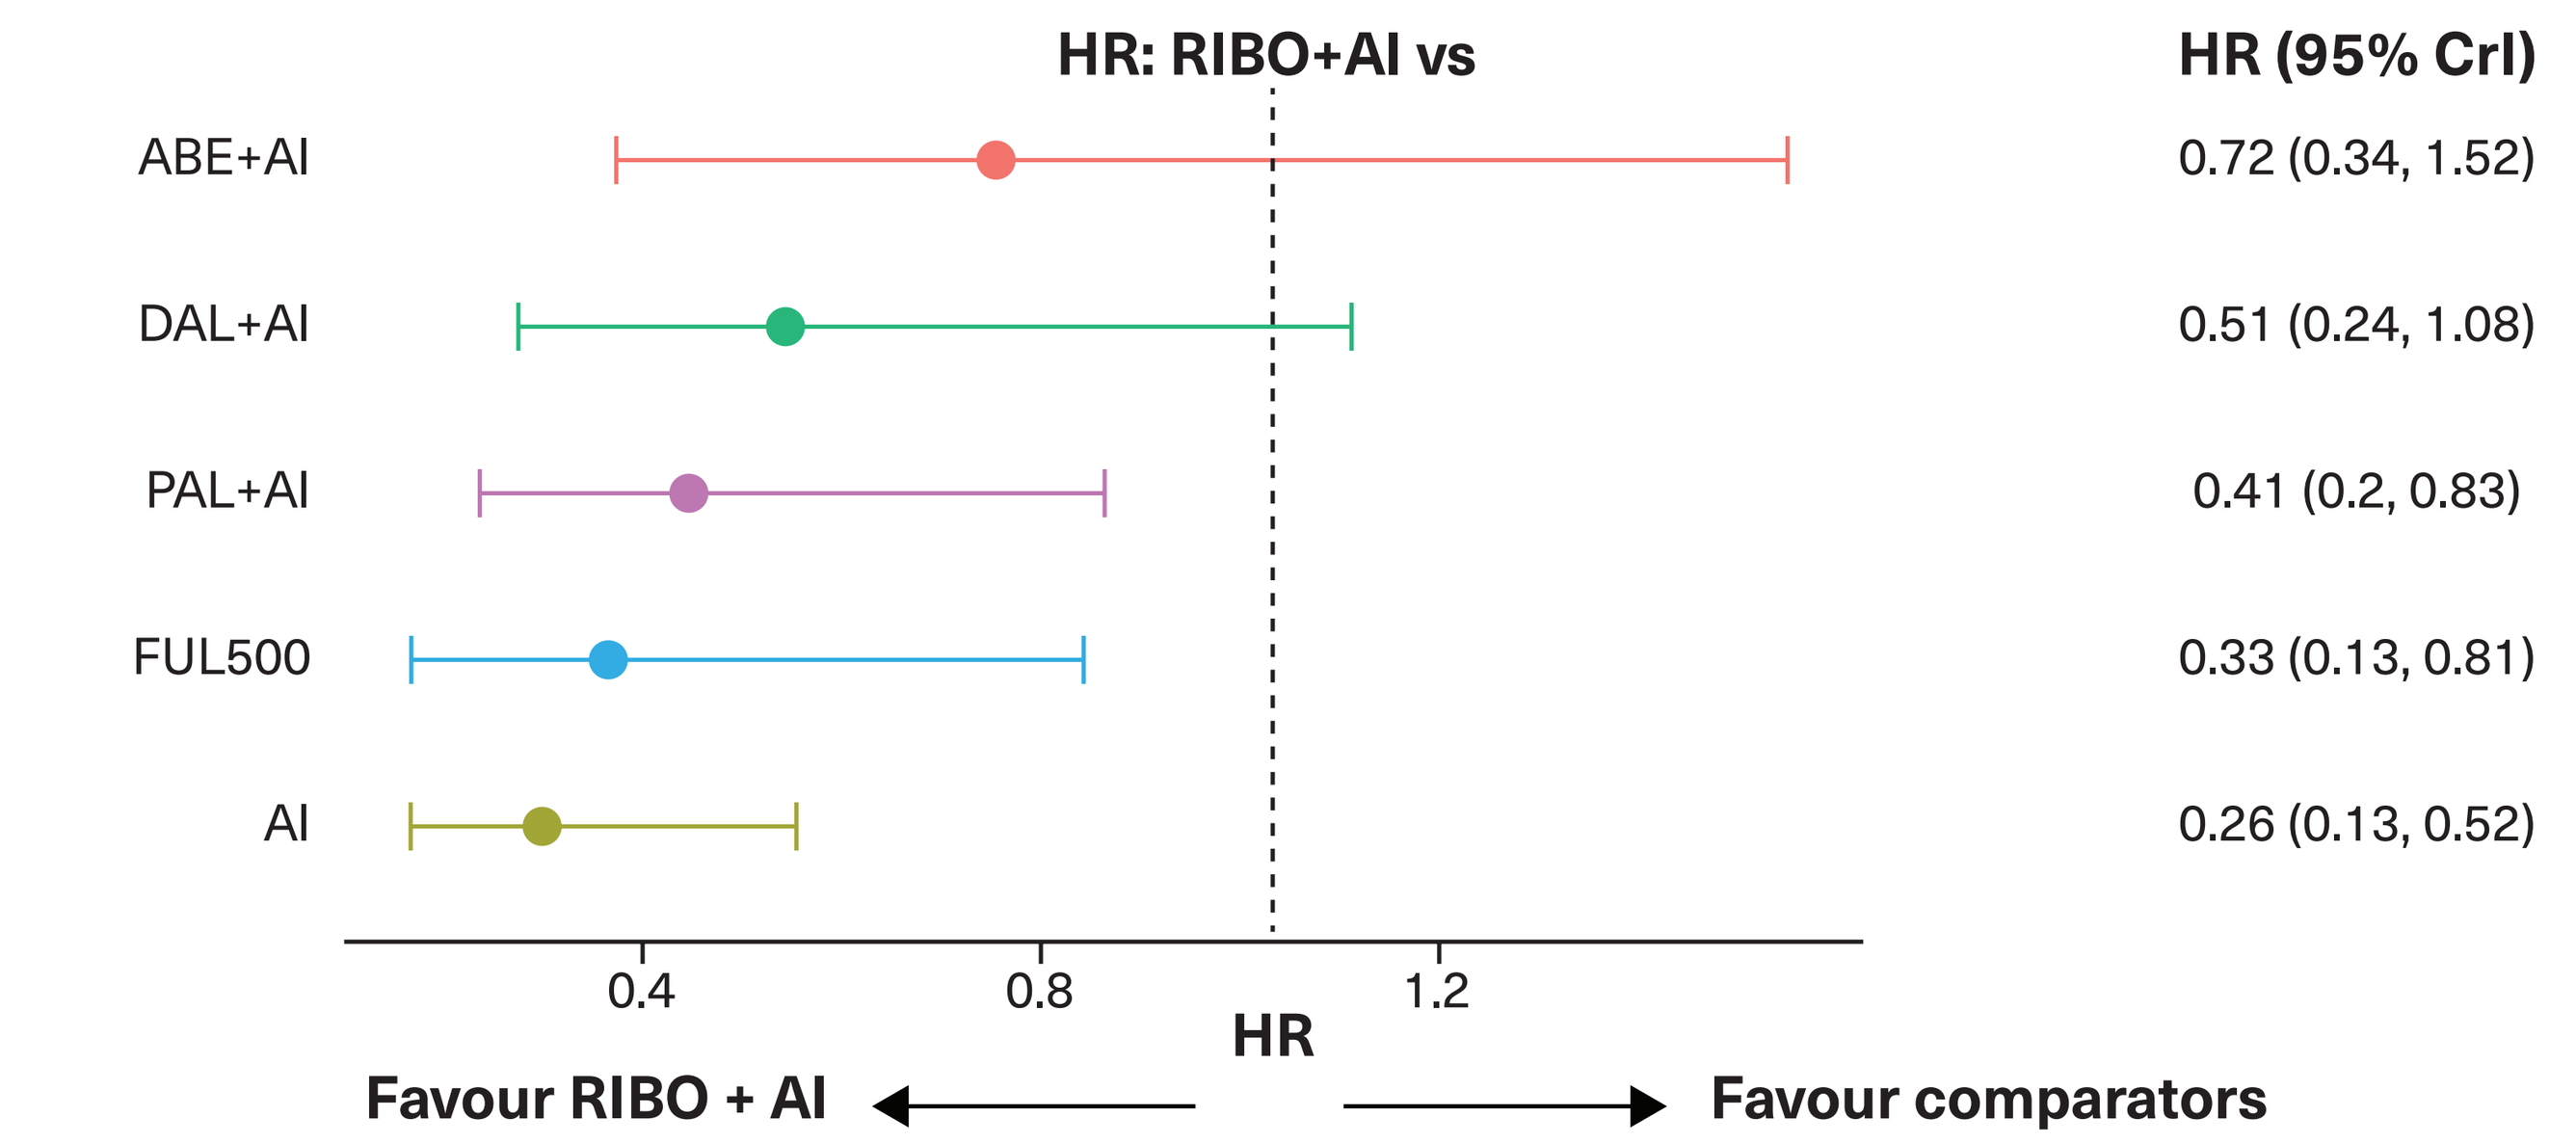
HR: RIBO+AI vs
HR (95% CrI)
ABE+AI
0.72 (0.34, 1.52)
DAL+AI
0.51 (0.24, 1.08)
PAL+AI
0.41 (0.2, 0.83)
FUL500
0.33 (0.13, 0.81)
AI
0.26 (0.13, 0.52)
0.4
0.8
1.2
HR
Favour RIBO + AI
Favour comparators

Abbreviations. ABE = abemaciclib, AI = aromatase inhibitors, CrI = credible interval, DAL = dalpiciclib, ET = endocrine therapy, FUL500 = fulvestrant 500mg, HR= hazards ratio, PAL = palbociclib, RIBO = ribociclib

Figure 2. NMA for Progression-Free Survival among post-menopausal population in Asia region

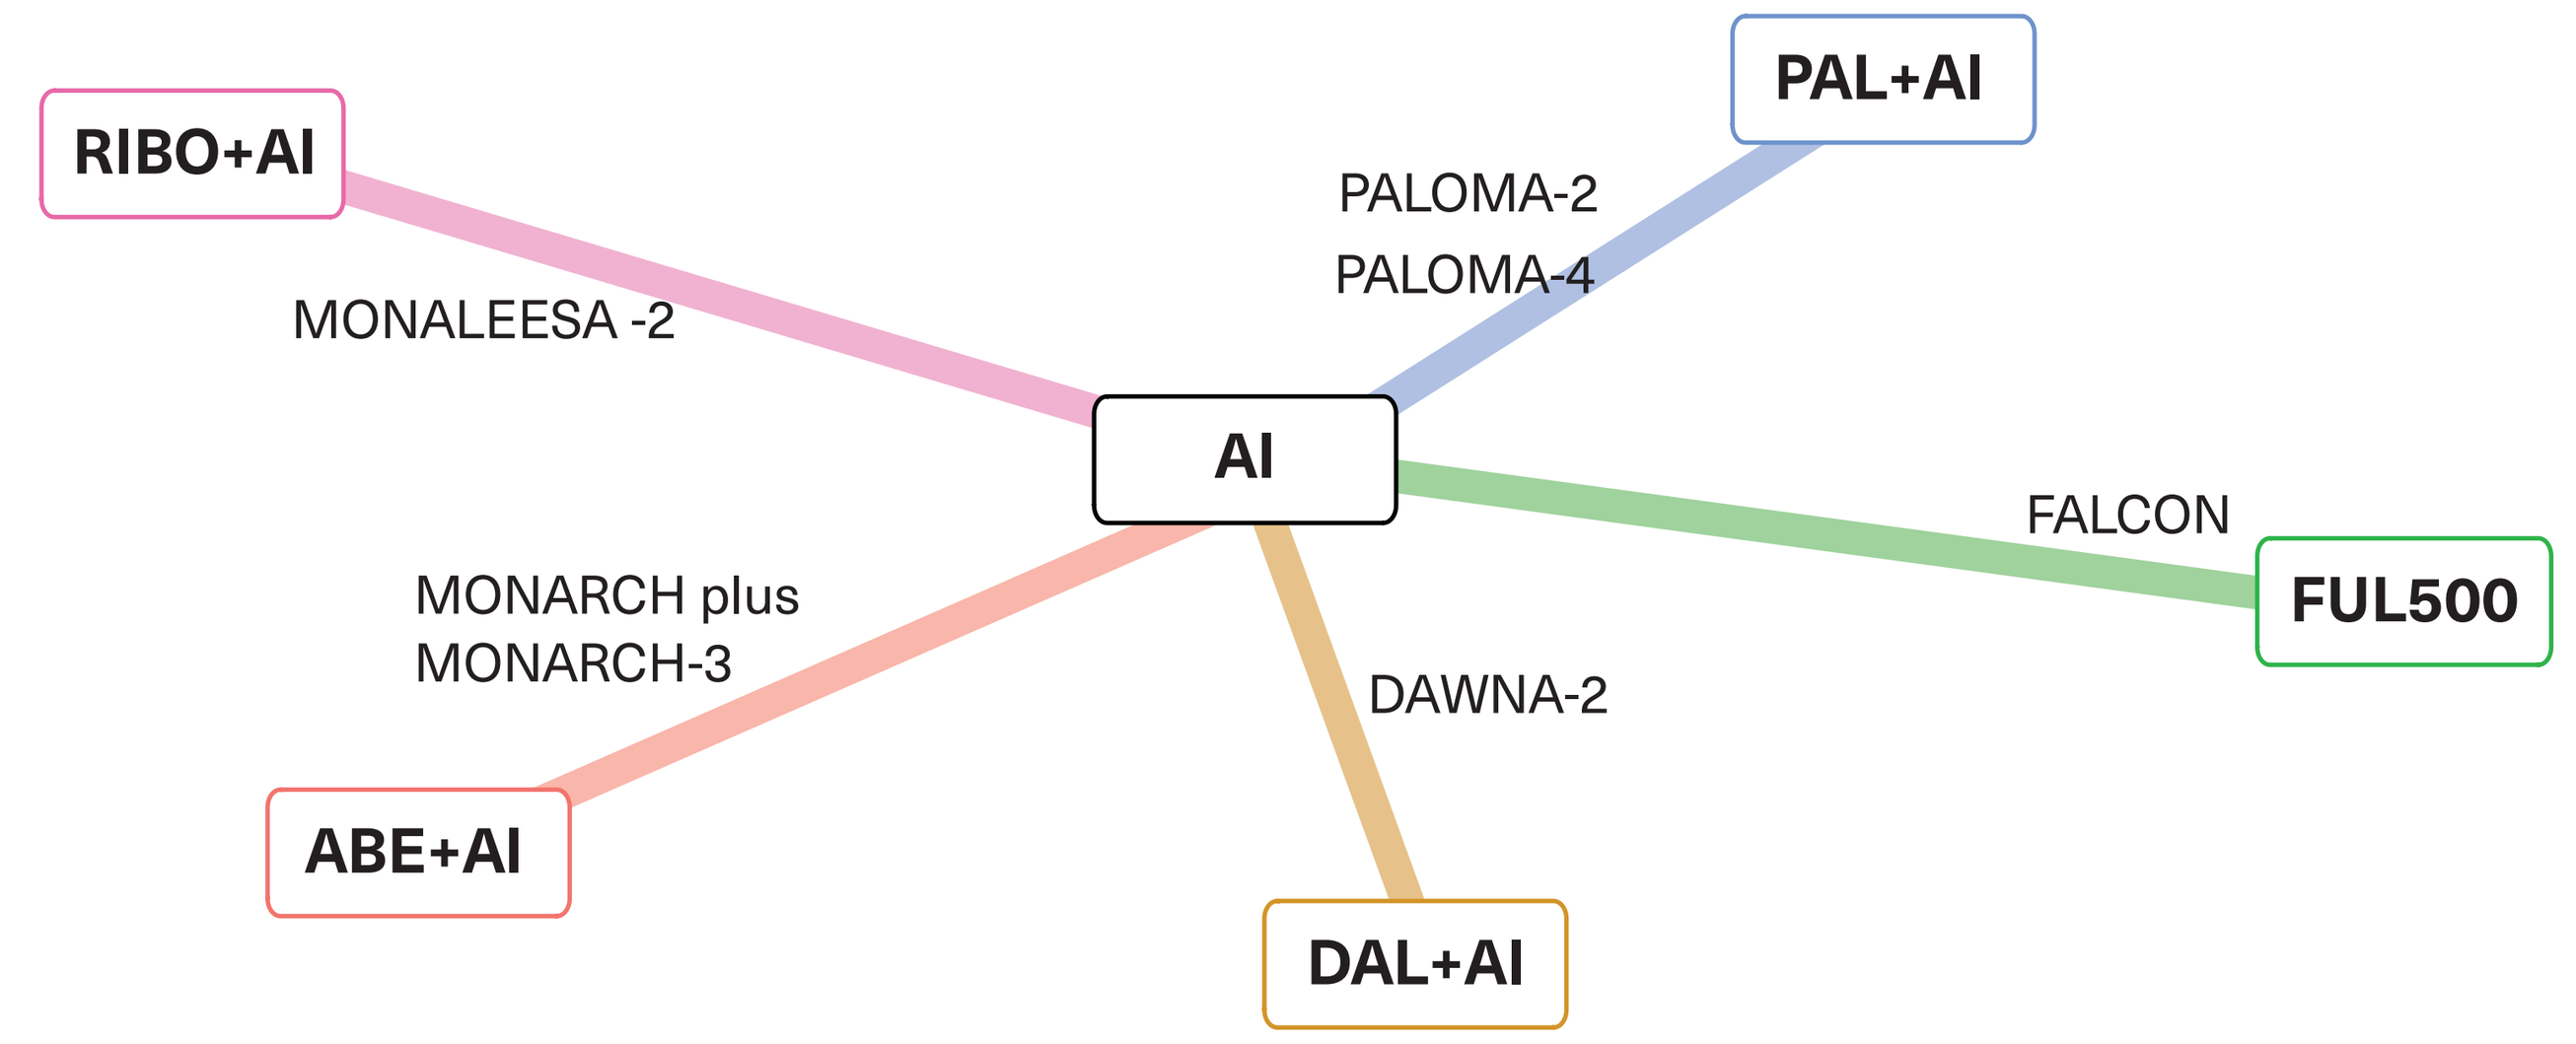
RIBO+AI
PAL+AI
PALOMA-2
PALOMA-4
MONALEESA -2
AI
FALCON
FUL500
MONARCH plus
MONARCH-3
DAWNA-2
ABE+AI
DAL+AI

ABE = abemaciclib, AI = aromatase inhibitors, DAL = dalpiciclib, FUL500 = fulvestrant 500mg, PAL = palbociclib, RIBO = ribociclib

- Favorable OS associated with ribociclib plus AI versus AI alone (HR: 0.64 [0.35,1.17]), while results were non-significant against abemaciclib plus AI (HR: 1.01 [0.45, 2.24]).

Pre-menopausal patients

- In pre-menopausal patients, PFS was significantly better with RIBO plus ET versus ET (HR: 0.42 [0.27, 0.66]) and numerically better compared to DAL plus ET (HR: 0.79 [0.41,1.52]) (**Figure 3** & **Figure 4**).

Figure 3. Forest plot of Progression-Free Survival among pre-menopausal population in Asia region

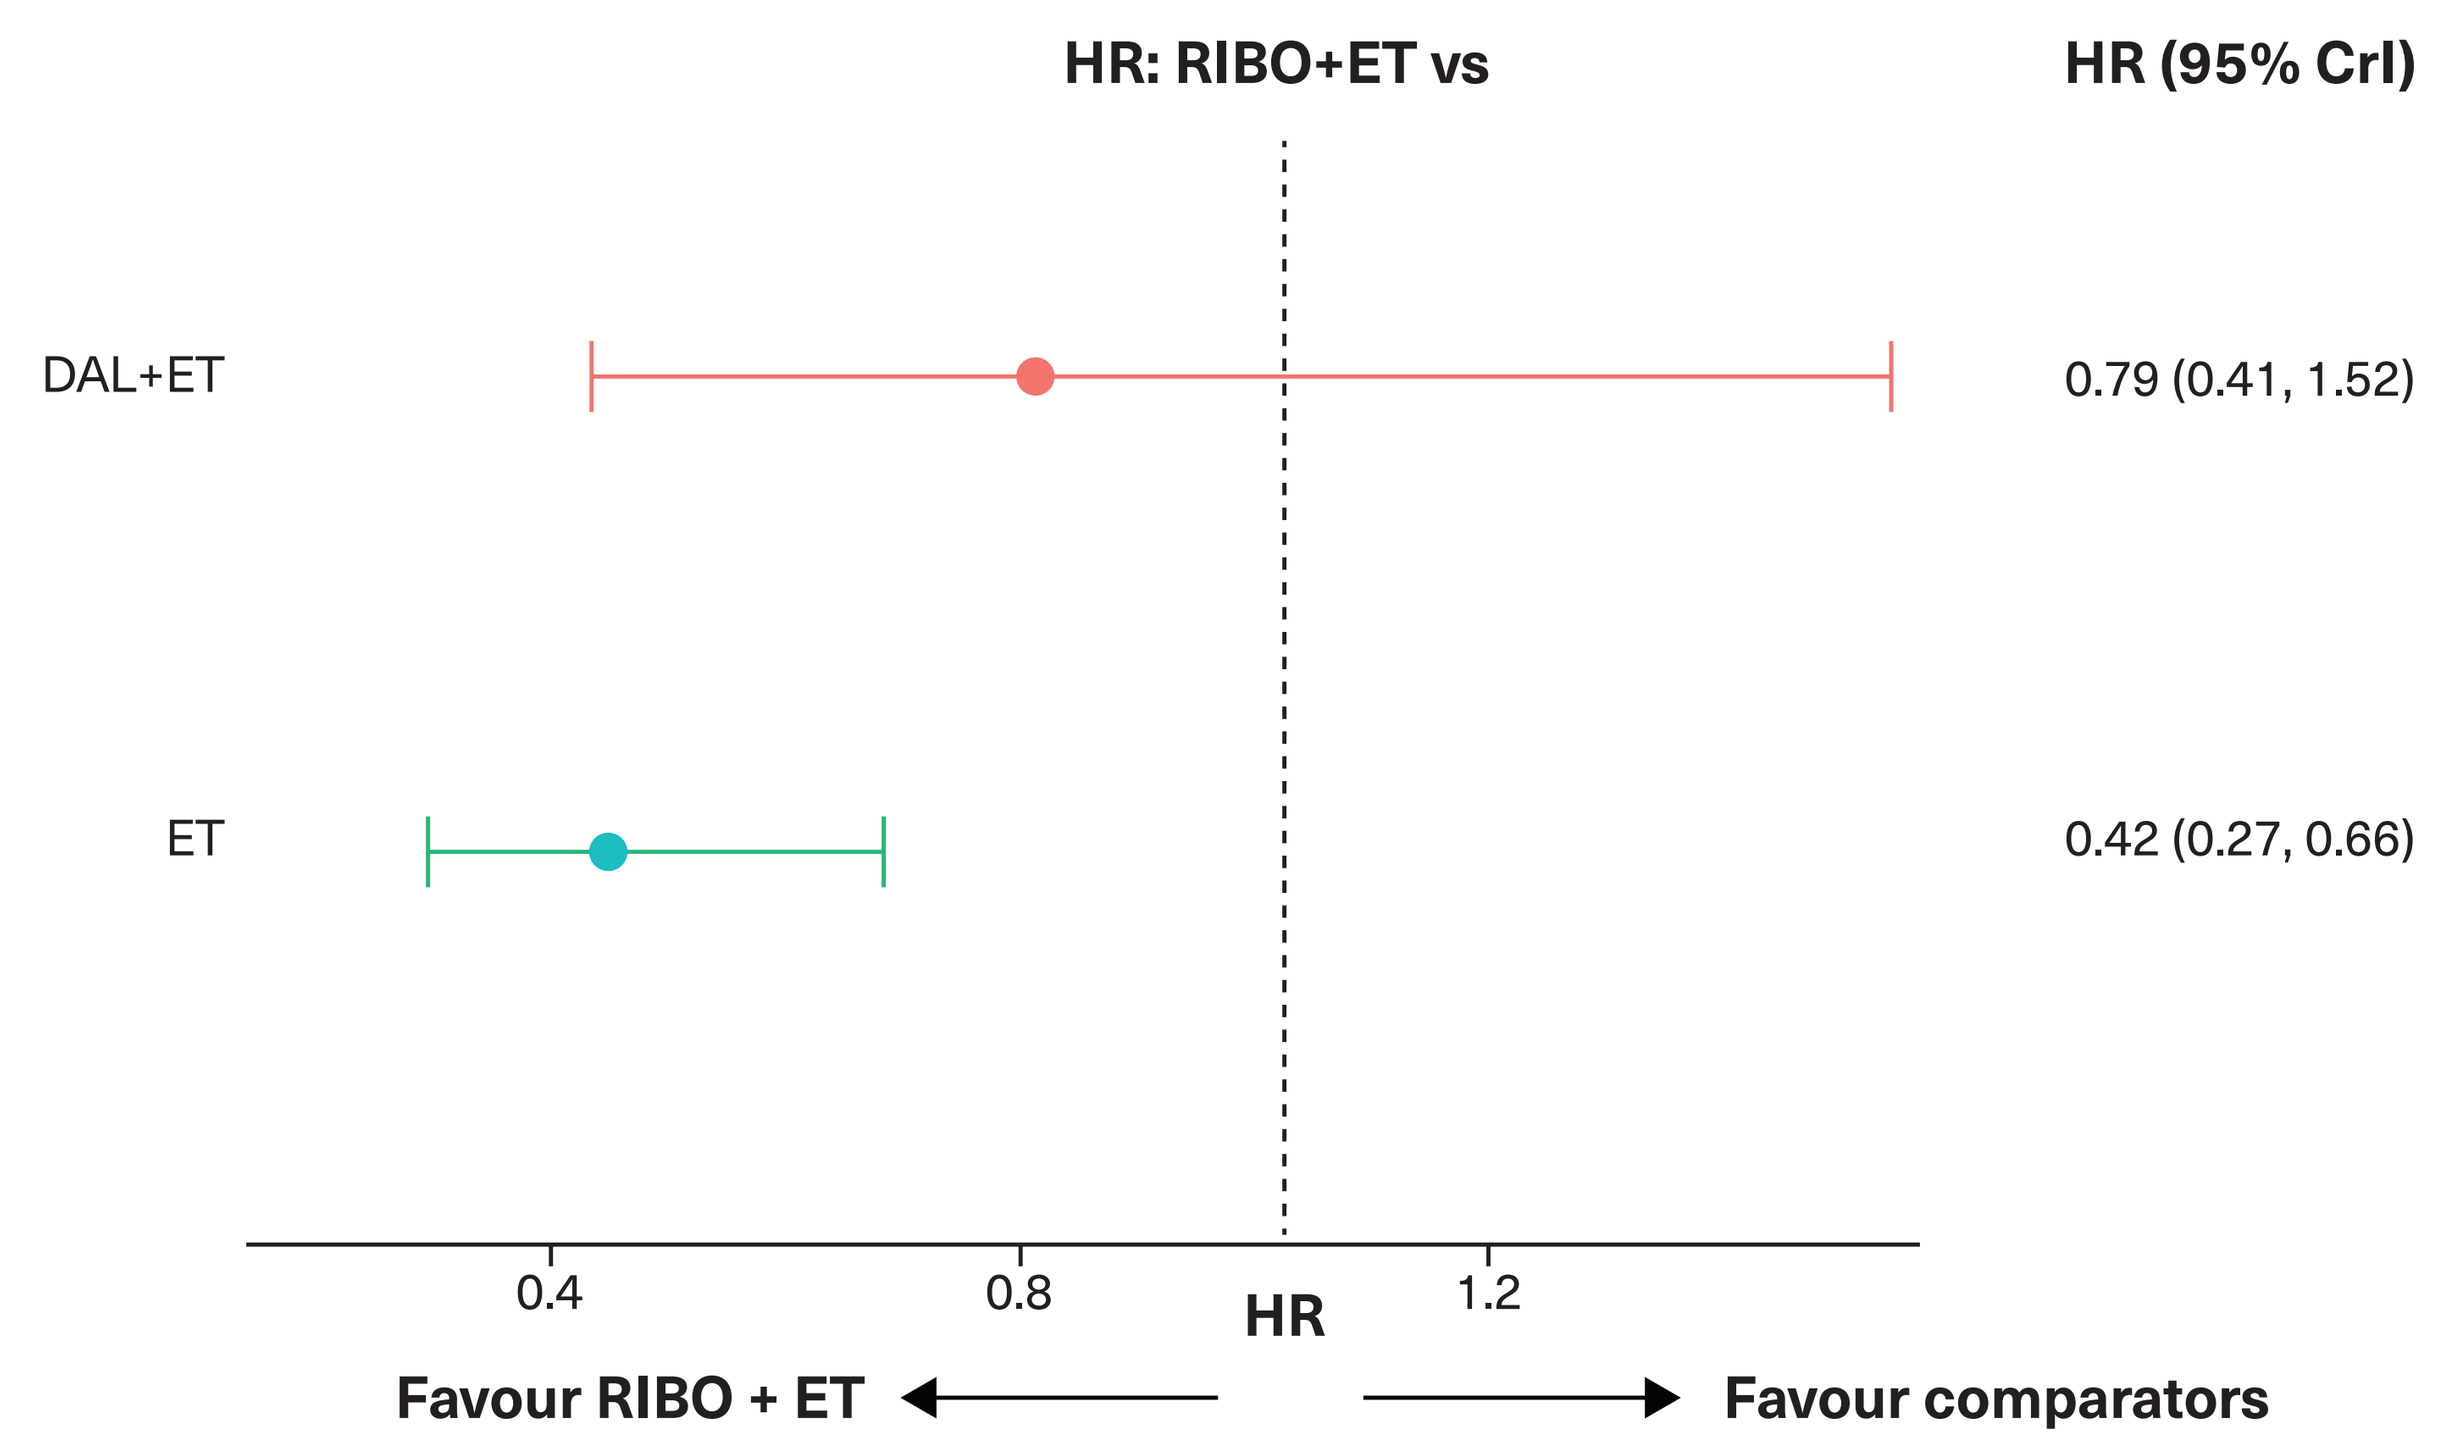
HR: RIBO+ET vs
HR (95% CrI)
DAL+ET
0.79 (0.41, 1.52)
ET
0.42 (0.27, 0.66)
0.4
0.8
1.2
HR
Favour RIBO + ET
Favour comparators

CrI = credible interval, DAL = dalpiciclib, ET = endocrine therapy, HR= hazards ratio, RIBO = ribociclibribociclib

Figure 4. NMA of Progression-Free Survival among pre-menopausal population in Asia region

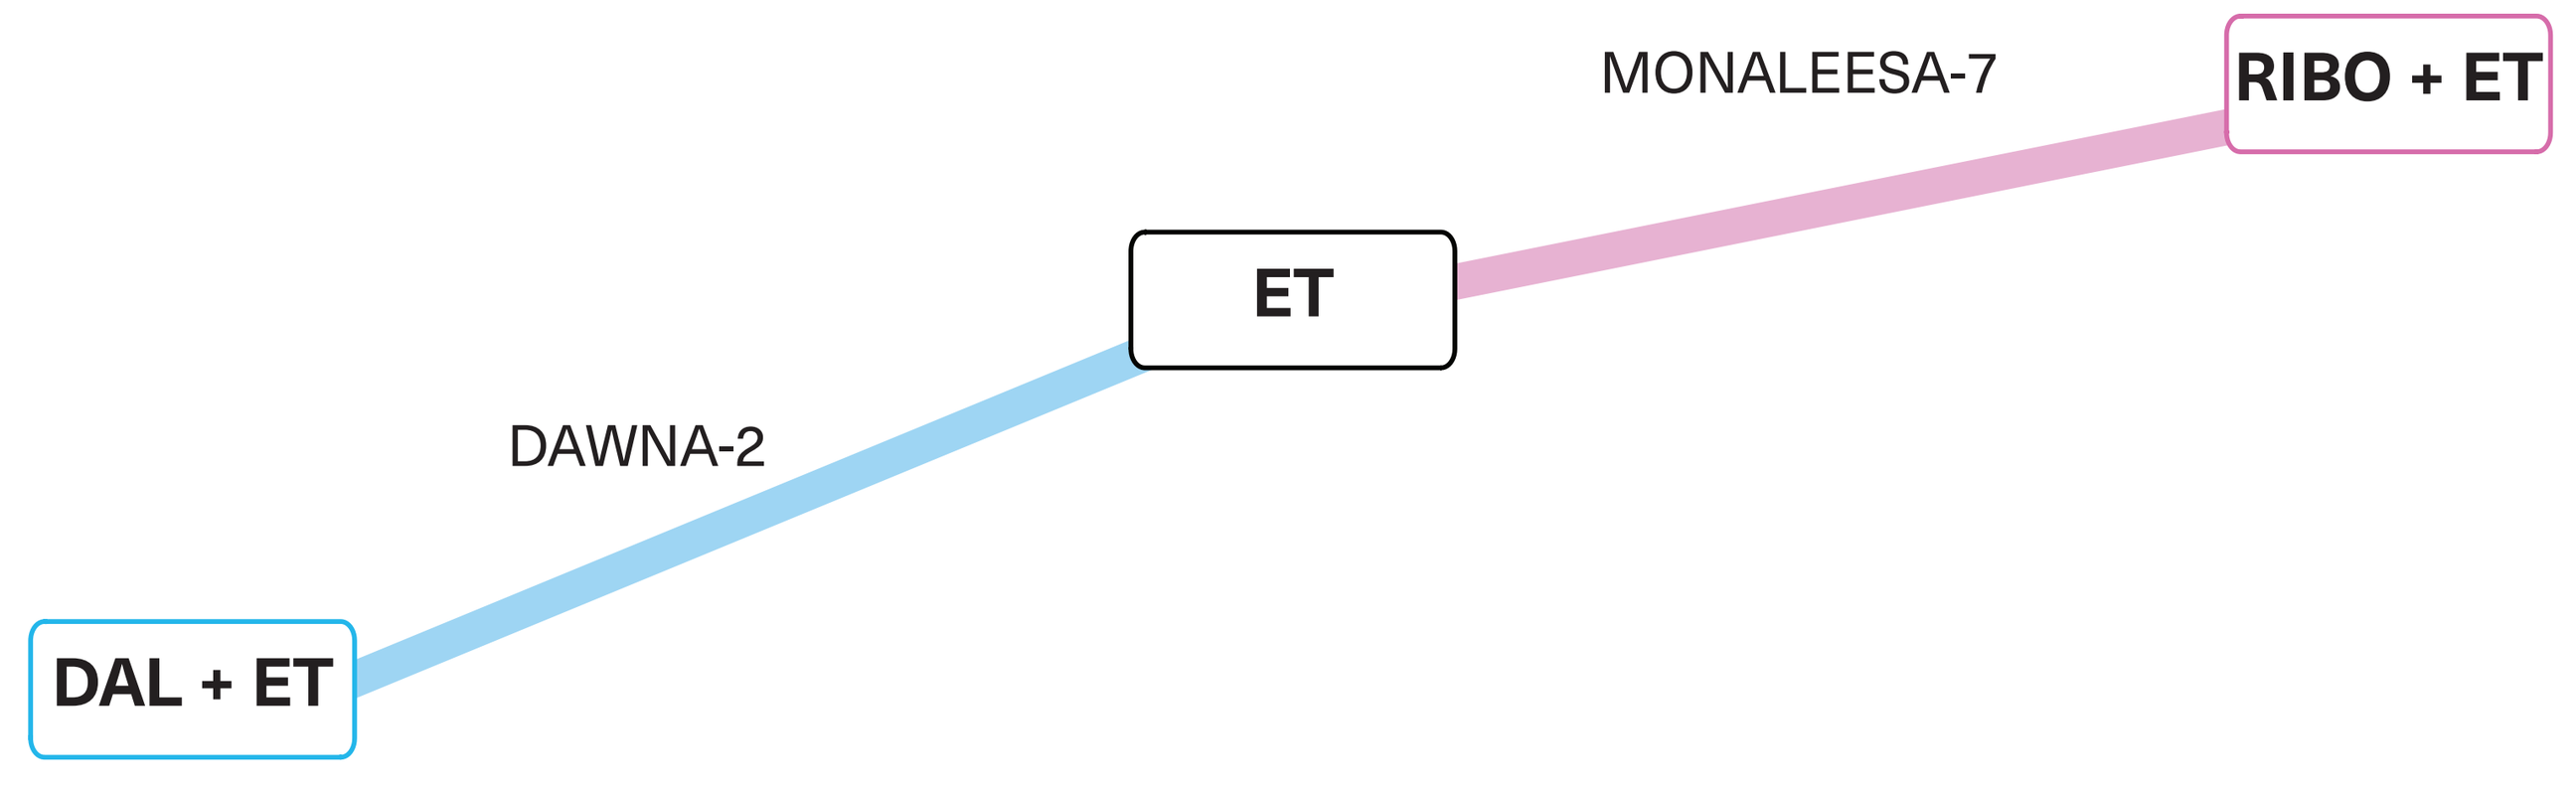
MONALEESA-7
RIBO + ET
ET
DAWNA-2
DAL + ET

DAL = dalpiciclib, ET = endocrine therapy, RIBO = ribociclib

CONCLUSION

- NMA results were estimated based on data of Asia region population and suggested that RIBO was associated with significant PFS benefit versus PAL in post-menopausal patients and was comparable or numerically better versus other CDK4/6 inhibitors (ABE and DAL) among pre/post-menopausal patients.

LIMITATIONS

- Limited data were reported for Asia subgroup in pre-menopausal patients; no trial was identified for PAL and ABE, while DAL trial reported only PFS.
- OS results should be interpreted with caution. RIBO has proven OS benefits in 1L phase 3 trial in both pre/peri menopausal and post-menopausal patients. However, for Asia region in pre-menopausal population only one study of RIBO whereas for post-menopausal RIBO and ABE reports OS data and thus we are exploring more options with chemotherapy studies to conduct analysis to draw conclusion on OS results.
- These results are based on trials' subgroup analysis from which is often underpower.
- Non-steroidal aromatase inhibitors (NSAI) plus goserelin were assumed to have equal efficacy as NSAI alone in pre-menopausal population, since in China most patients receive goserelin during their treatment cycle.

Poster presented at the 2023 Meeting of ISPOR – The Society for Health Economics and Outcomes Research held on 08–10 May 2023 in Boston, Massachusetts, USA.

References

1. U.S. Department of Health and Human Services, National Institutes of Health, National Cancer Institute USA.gov, Accessed on 23 March 2023, Available from https://seer.cancer.gov/statfacts/html/breast-subtypes.html
2. Shah, M., et al., *Oncology* (Williston Park). 2018 May 15;32(5):216–22.
3. Piezzo, M.,et al., *Int J Mol Sci*. 2020 Sep 3;21(17):6400
4. Lee.K.W.C.,et al., *Breast Cancer Res Treat*. 2019 Feb;174(1):271–278
5. Sheikh, M. S.,et al., *Mol Cell Pharmacol*. 2021;13(3):9–12.
6. Hortobagyi, G. N.,et al., *Ann Oncol*. 2018 Jul 1;29(7):1541–1547
7. Hortobagyi, G. N.,et al., *N Engl J Med*. 2022 Mar 10;386(10):942–950
8. Robertson, J. F. R., et al., *Lancet*. 2016 Dec 17;388(10063):2997–3005
9. Finn, R. S., et al., *N Engl J Med*. 2016; 375:1925–1936
10. B. Xu, et al., *Ann Oncol*. 2021;32 (5),2021:S457
11. B. Xu, et al., *Ann Oncol*. 2022;33 (7),2021: S1384–S1385
12. Zhang, Q. Y.,et al., *Ther Adv Med Oncol*. 2020 Oct 22;12:1758835920963925
13. Johnston, S., et al., *NPJ Breast Cancer*. 2019 Jan 17;5:5
14. M.P. Goetz., et al., *Ann Oncol*. 2022;33 (7),2022: S1384
15. Lu, Y. S.,et al., 2022 Mar 1;28(5):851-859.
16. Tripathy, D., et al., *Lancet Oncol*. 2018 Jul;19(7):904-915

Funding

This research was sponsored by funds from Novartis Pharmaceuticals, Shanghai, China.

Disclosures

YY, LY, SC and YF are employees of Department of Oncology, The First Affiliated Hospital of Nanjing Medical University, Nanjing, China; **LC** and **YJ** are employees of Novartis Pharmaceuticals, Shanghai, China; **SR** was a former employee of Novartis Pharmaceuticals, Shanghai, China; **MS** is employee of Novartis Healthcare Pvt. Ltd., Hyderabad, India; **TT** is an employee Novartis Global Service Centre, Dublin, Ireland; **TB** is an employee of Novartis Corporate Center, Kuala Lumpur, Malaysia