HSD84

Prescription antitussive use trajectories and their characteristics among patients with chronic cough in Florida

Seonkyeong Yang[1]; Shu Huang[1]; Juan M. Hincapie-Castillo[2]; Xuehua Ke[3]; Helen Ding[3]; Mandel R. Sher[4]; Bobby Jones[1]; Debbie L. Wilson[1]; Wei-Hsuan Lo-Ciganic[1]

[1]Department of Pharmaceutical Outcomes & Policy, College of Pharmacy, University of Florida, Gainesville, FL, USA; [2]Department of Epidemiology, University of North Carolina at Chapel Hill, Chapel Hill, NC, USA; [3]Center for Observational and Real-World Evidence (CORE), Merck & Co., Inc., Rahway, NJ, USA; [4]Sher Allergy Specialists, Largo, FL, USA; [5]Center for Drug Evaluation and Safety, College of Pharmacy, University of Florida, Gainesville, FL, USA

Background

- Chronic cough (CC), defined as cough lasting more than 8 weeks, can be caused by various respiratory/nonrespiratory conditions and other factors[1,2]
- CC can resolve when underlying causes are successfully treated; however, CC can also be refractory or unexplained[3]
 - Refractory CC (RCC) is CC that remains persistent after receiving an appropriate diagnostic workup and guideline-based management for underlying conditions
 - Unexplained CC (UCC) is CC that remains unexplained without clear etiology despite extensive clinical evaluation of underlying causes
- Emerging evidence suggests CC patients may have a distinct disease entity with neural dysregulation: cough hypersensitivity syndrome[4,5]
- Gabapentin is a potential pharmacological option for unexplained CC when risks and benefits are carefully evaluated at initiation and reassessed at 6 months before continuation[3]
- Common cough medications (CMs), including opioid antitussives (codeine-, hydrocodone-, and dihydrocodeine-containing formulations), benzonatate, and dextromethorphan,[6,7] are frequently used for symptomatic relief of cough despite a lack of proven efficacy and safety evidence for CC
- Little is known about prescription CM use patterns among CC patients in real-world settings

Objective

- To examine trajectories of prescription antitussive use among CC patients in Florida

Methods

- **Design:** Retrospective cohort study
- **Source:** 2012-2021 statewide clinical data from the OneFlorida Clinical Research Consortium
- **Study population**
 - **Inclusion criteria:** Adult patients (≥18 years) who met the CC algorithm, defined as having ≥3 clinical cough events (ie, a cough diagnosis or a prescription of opioid antitussives, benzonatate, or dextromethorphan) occurring within 120 days, with the first and last of the 3 events to be ≥8 weeks apart and any 2 of the 3 events to be ≥3 weeks apart (Figure 1)
 - Index date: The date of a patient's first qualifying cough event
 - Baseline period: 6 months prior to the index date
 - Trajectory measurement period: 12 months after the index date (including the index date)
 - **Exclusion criteria:** Patients (1) with any malignant cancer or any benign respiratory tumor diagnoses, (2) had an index date before July 1, 2012, or after January 1, 2021, or (3) had no encounter during the baseline period or had <2 encounters during the trajectory measurement period

Figure 1. Study design diagram

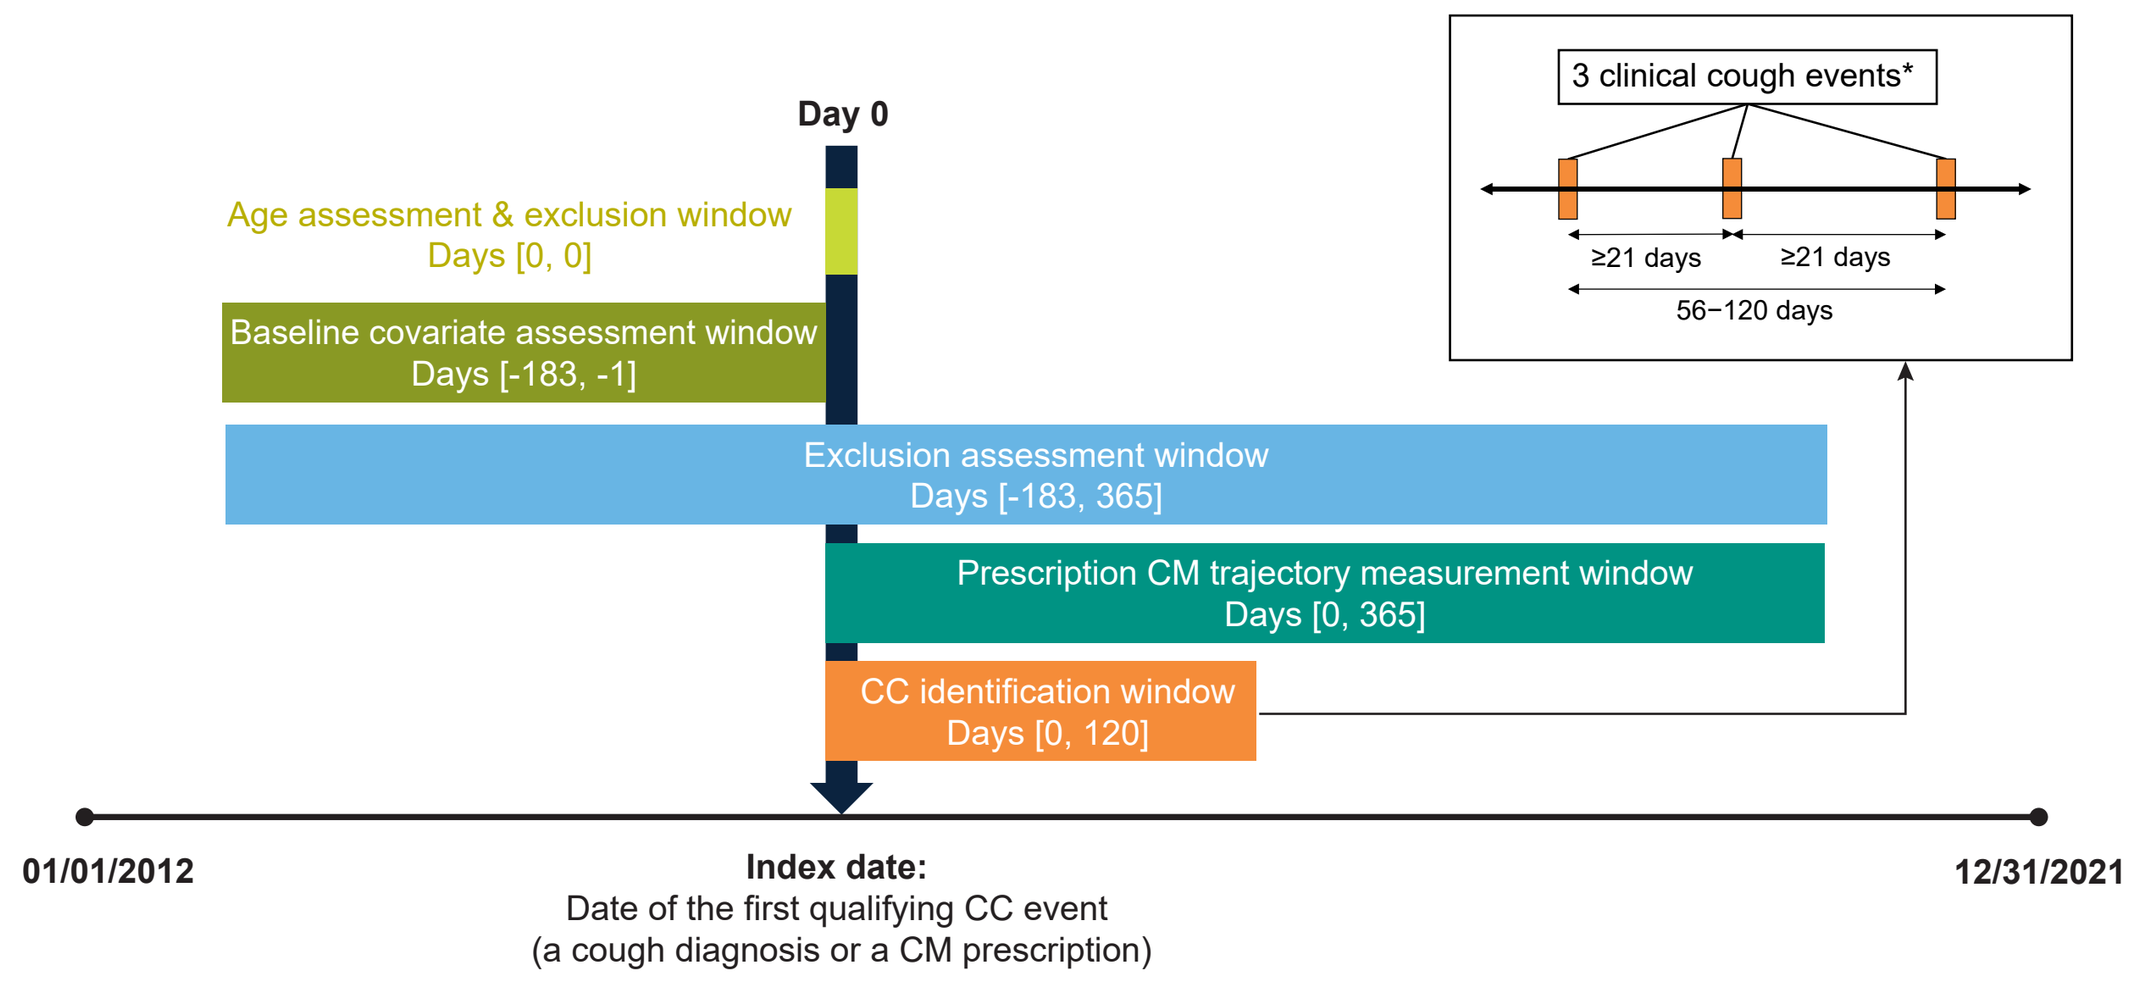
Day 0
Age assessment & exclusion window
Days [0, 0]
Baseline covariate assessment window
Days [-183, -1]
Exclusion assessment window
Days [-183, 365]
Prescription CM trajectory measurement window
Days [0, 365]
CC identification window
Days [0, 120]
3 clinical cough events*
≥21 days
≥21 days
56–120 days
01/01/2012
Index date:
Date of the first qualifying CC event
(a cough diagnosis or a CM prescription)
12/31/2021

*CC event: a cough diagnosis or a prescription of opioid antitussives, benzonatate, or dextromethorphan.

- **Statistical analysis**
 - We conducted a group-based trajectory modeling (GBTM) analysis to identify distinct prescription CM utilization trajectories over 12 months among CC patients
 - We estimated models with *traj* in STATA17 (StataCorp LLC, College Station, TX, USA) having the longitudinal count of the number of prescriptions per month for any prescription CM (opioid antitussives, benzonatate, or dextromethorphan) as the outcome variable and the month since the index date (1-12 months) as the time variable. Due to numerous on/off-label uses of gabapentin, gabapentin was not considered as CM in the current study[8]
 - GBTM outputs included estimated probabilities of group membership for each individual, estimated trajectory curves over time, and the proportion of membership in each group trajectory
 - We presented descriptive data for baseline characteristics of CC patients by prescription CM utilization trajectory groups
 - We compared characteristics across the prescription CM utilization trajectories using analysis of variance

Presented at ISPOR 2023; Boston, MA, USA; May 7-10, 2023.

Results

Prescription CM utilization trajectories identified in CC patients (Table 1)

- From a total of 16,697,875 individuals in the 2012-2021 OneFlorida data, we identified 15,566 CC patients (Figure 2)
- Among CC patients, we identified 3 distinct prescription CM utilization trajectories (Figure 3)

1. Group 1: No CM use (n=11,222; 72.1%)
2. Group 2: Declining CM use (n=4,105; 26.4%)
3. Group 3: Chronic CM use (n=239; 1.5%)

Table 1. Baseline sociodemographics and health care service use in CC patients by prescription CM utilization trajectory group

	All CC patients	Group 1 (No CM use)	Group 2 (Declining CM use)	Group 3 (Chronic CM use)	
	n=15,566	n=11,222	n=4105	n=239	*P*-value
Sociodemographics, %					
Age					**0.96**
<65 years	72.3	72.3	72.2	71.6	
≥65 years	27.7	27.7	27.8	28.5	
Female	70.2	69.4	72.6	70.7	0.002
Race					**<0.001**
White	62.3	62.8	61.5	55.7	
Black	27.6	26.6	29.7	36.8	
Other	10.1	10.7	8.8	7.5	
Ethnicity					**<0.001**
Hispanic	21.4	23.8	15.3	*	
Non-Hispanic	76.6	74.2	82.7	85.8	
Unknown	2.0	2.1	2.0	*	
Payer					**<0.001**
Medicare	48.1	47.7	48.6	56.9	
Medicaid	38.3	43.9	24.4	16.3	
Commercial	9.0	4.3	21.0	22.2	
Other	4.6	4.1	6.1	4.6	
Residency					**<0.001**
Metropolitan counties	88.7	89.1	87.6	90.8	
Nonmetropolitan counties	6.3	5.8	7.7	7.1	
Unknown	4.9	5.1	4.7	2.1	
Health care service use, %					
≥1 Hospitalization	25.7	26.4	23.6	31.0	<0.001
No. of emergency department visits					**<0.001**
0	49.3	46.9	55.5	54.4	
1	17.8	18.2	16.8	14.2	
≥2	33.0	34.9	27.7	31.4	
No. of outpatient visits					**<0.001**
0	6.8	6.4	8.0	8.4	
1	8.3	7.9	9.4	7.1	
2-4	20.7	20.4	21.7	13.0	
≥5	64.2	65.3	61.0	71.6	

CC, chronic cough; CM, cough medication; HIPAA, Health Insurance Portability and Accountability Act; SD, standard deviation.
*A percentage from a count of patients less than 11 or used to derive a count of patients less than 11 in other cells was not reportable according to HIPAA Privacy Rule.

Figure 2. Study sample flowchart

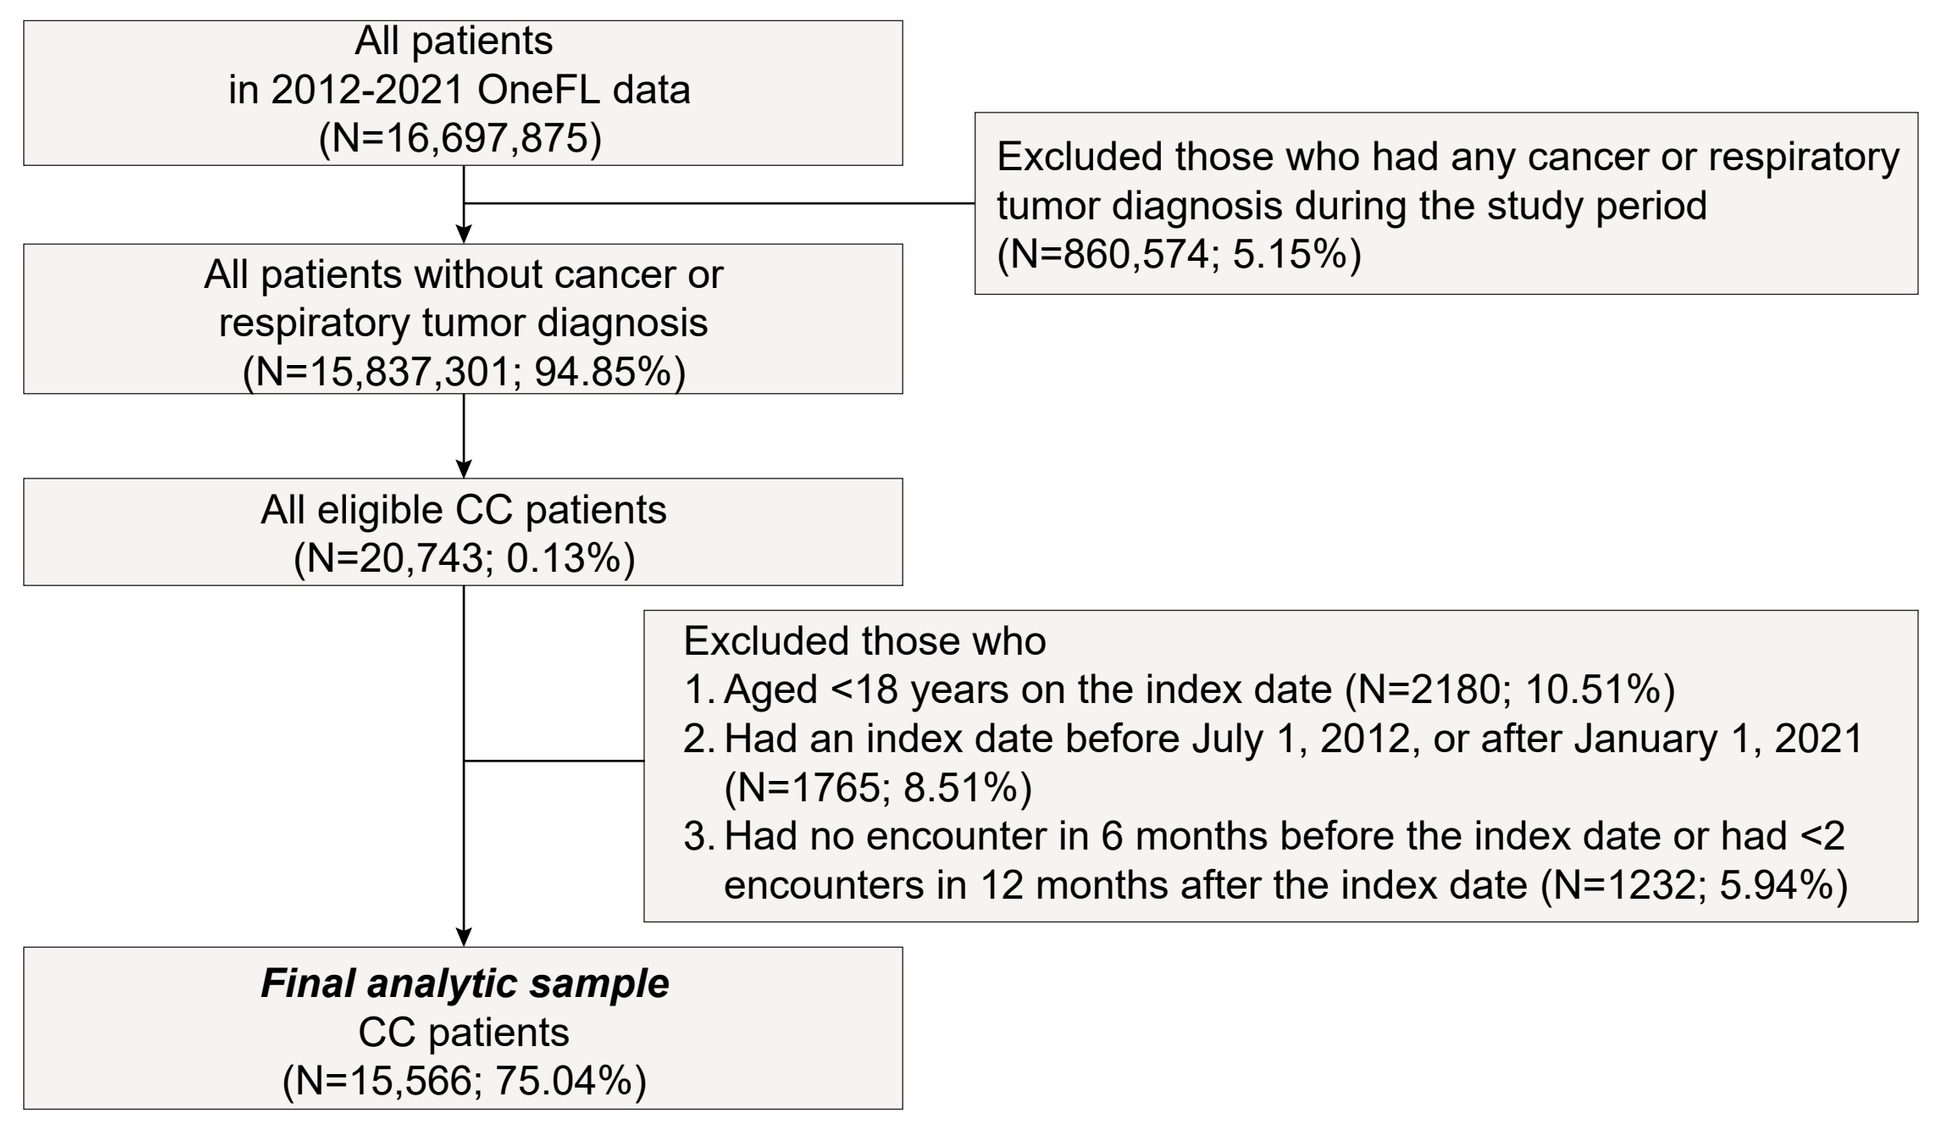
All patients
in 2012-2021 OneFL data
(N=16,697,875)
Excluded those who had any cancer or respiratory tumor diagnosis during the study period
(N=860,574; 5.15%)
All patients without cancer or respiratory tumor diagnosis
(N=15,837,301; 94.85%)
All eligible CC patients
(N=20,743; 0.13%)
Excluded those who
1. Aged <18 years on the index date (N=2180; 10.51%)
2. Had an index date before July 1, 2012, or after January 1, 2021
(N=1765; 8.51%)
3. Had no encounter in 6 months before the index date or had <2 encounters in 12 months after the index date (N=1232; 5.94%)
Final analytic sample
CC patients
(N=15,566; 75.04%)

Figure 3. Prescription CM utilization trajectories identified in CC patients

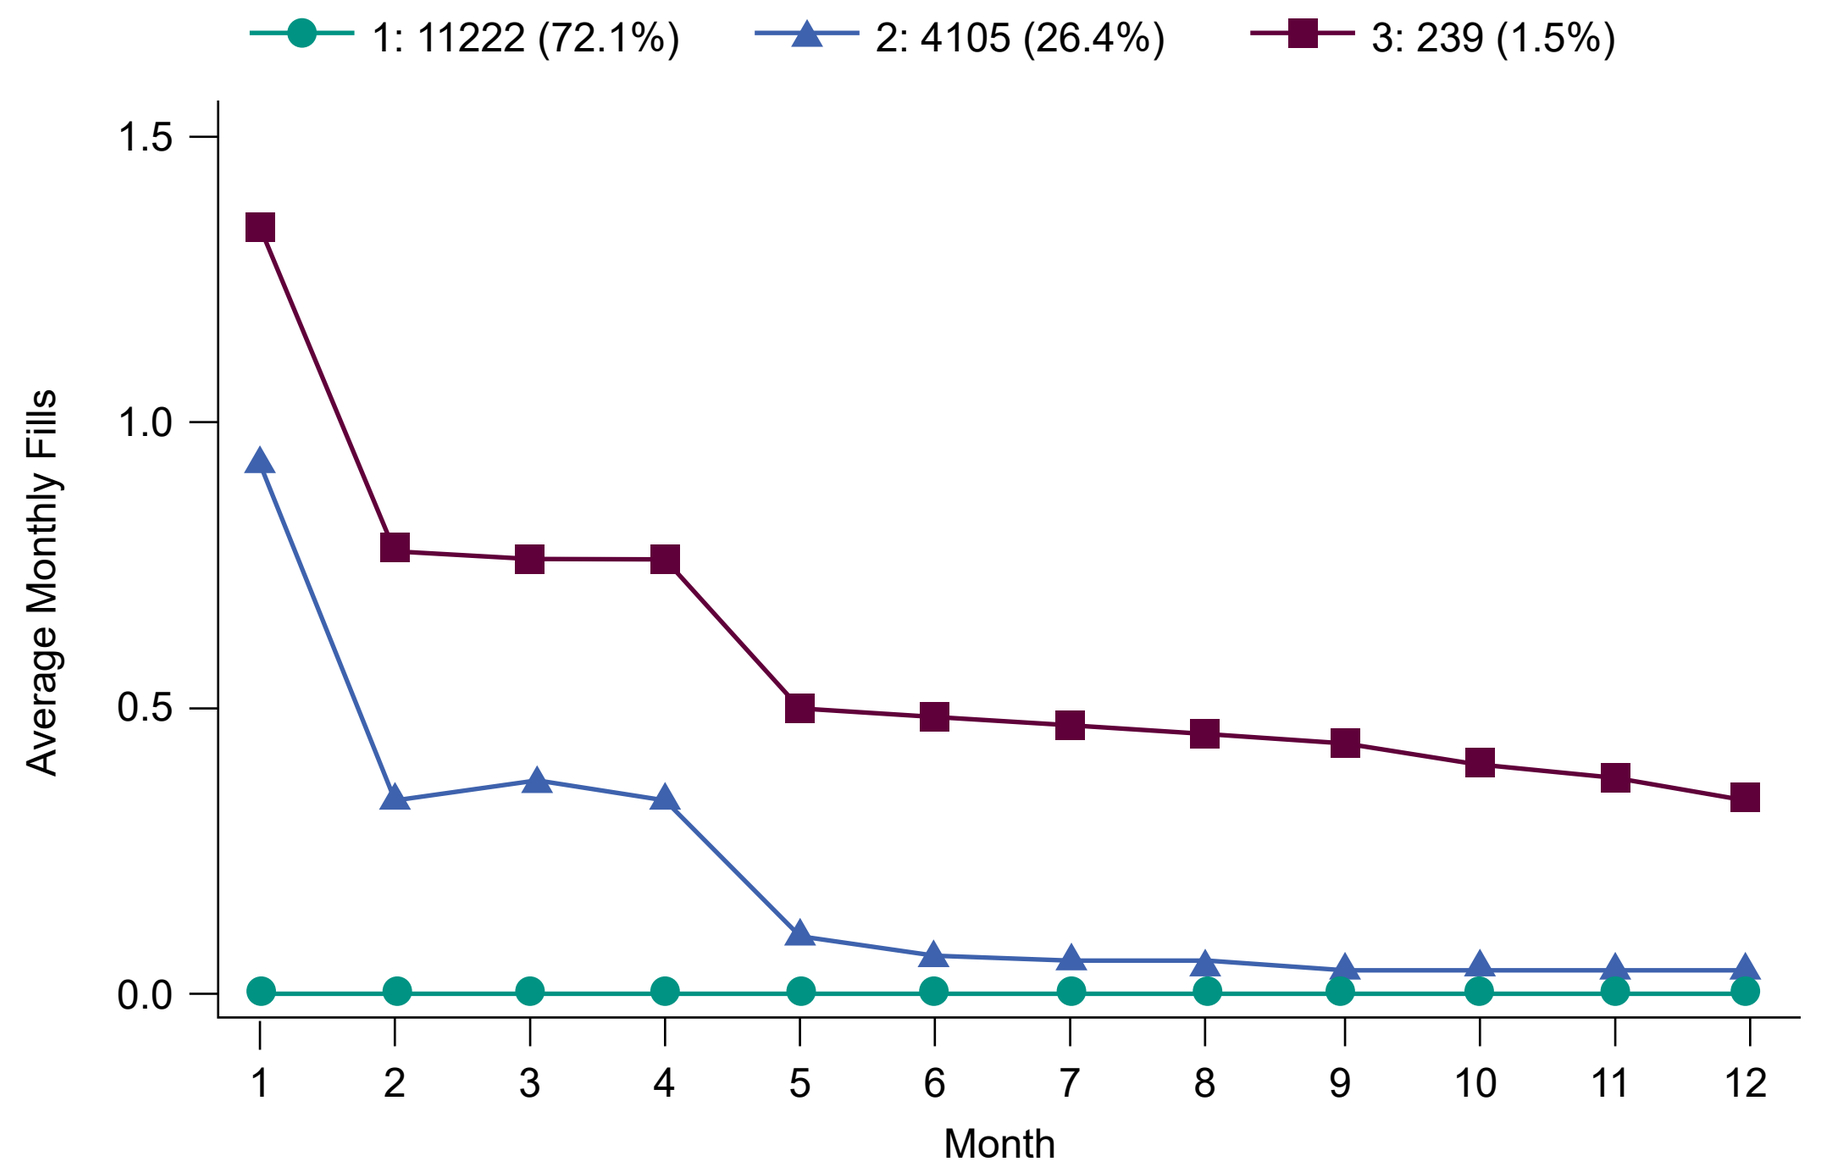
1: 11222 (72.1%)
2: 4105 (26.4%)
3: 239 (1.5%)
Average Monthly Fills
1.5
1.0
0.5
0.0
1
2
3
4
5
6
7
8
9
10
11
12
Month

Baseline characteristics of CC patients by prescription CM utilization trajectory group

- CC patients were predominantly <65 years (72.3%), women (70.2%), White (62.3%), and non-Hispanic (76.6%) (Table 1)
- CC patients with chronic prescription CM use were more likely to be Black, non-Hispanic, and have Medicare, ≥1 hospitalization, and ≥5 outpatient visits than other CC patients (Table 1)
- Among all CC patients, CC patients with chronic prescription CM use tend to have the highest prevalence of selected respiratory conditions, including chronic obstructive pulmonary disease (36.0%), asthma (32.6%), cough (not meeting the CC definition) (29.3%), and bronchitis (25.9%) (Table 2)
- Among all CC patients, CC patients with chronic prescription CM use had the highest prevalence of selected nonrespiratory conditions, including musculoskeletal disorders (63.6%), hypertension (60.7%), coronary artery disease (37.2%), and obesity (35.2%) (Table 2)
- Among all CC patients, CC patients with chronic prescription CM use showed the highest use of respiratory and other medications at baseline (Table 3)

Table 2. Baseline conditions in CC patients by prescription CM utilization trajectory group

	All CC patients	Group 1 (No CM use)	Group 2 (Declining CM use)	Group 3 (Chronic CM use)	
	n=15,566	n=11,222	n=4105	n=239	*P*-value
Selected respiratory conditions,† %					
Acute URTI	19.6	19.6	19.6	19.7	1.00
Allergic rhinitis	11.7	11.3	12.7	16.3	0.005
Asthma	22.8	22.5	23.1	32.6	0.001
Bronchitis	18.7	18.8	17.9	25.9	0.007
Chronic obstructive pulmonary disease	28.0	29.5	23.4	36.0	<0.001
Chronic sinusitis	5.7	5.9	4.7	12.1	<0.001
Cough‡	25.9	27.4	21.5	29.3	<0.001
Obstructive sleep apnea	9.6	8.8	11.5	14.2	<0.001
Pertussis	6.6	6.7	6.2	7.5	0.48
Pneumonia	12.8	13.6	10.5	11.7	<0.001
Selected nonrespiratory conditions,† %					
Anxiety disorders	26.0	26.1	25.1	33.9	0.008
Atrial fibrillation	7.4	7.4	7.3	8.8	0.68
Coronary artery disease	28.9	28.7	28.8	37.2	0.02
Gastroesophageal reflux disease	26.8	26.7	26.5	34.7	0.02
Heart failure	12.8	13.1	11.7	16.7	0.02
Hypertension	55.0	55.2	54.2	60.7	0.11
Mood disorders	27.9	29.5	23.8	23.4	<0.001
Musculoskeletal disorders	57.5	58.0	55.7	63.6	0.006
Nonopioid substance use disorders	9.1	9.5	8.1	10.5	0.03
Obesity	23.9	22.6	27.0	35.2	<0.001
Other immune disorders	5.4	5.4	5.4	6.3	0.82
Peripheral vascular disease	5.2	5.6	4.3	*	0.008
Sleep disturbance	7.9	7.7	8.1	17.6	<0.001
Vomiting	5.0	5.3	4.2	*	0.008

CC, chronic cough; CM, cough medication; URTI, upper respiratory tract infection.
*A percentage from a count of patients less than 11 was not reportable according to HIPAA Privacy Rule.
†Due to space limits, the table only includes those conditions that at least 5% of all CC patients had.
‡Cough not qualified as CC based on the CC definition in Figure 1.

Table 3. Baseline medication use in CC patients by prescription CM utilization trajectory group

	All CC patients	Group 1 (No CM use)	Group 2 (Declining CM use)	Group 3 (Chronic CM use)	
	n=15,566	n=11,222	n=4105	n=239	*P*-value
CMs, %					
Benzonatate	2.7	0.8	7.3	15.9	<0.001
Dextromethorphan	1.0	0.3	2.5	8.0	<0.001
Opioid antitussives	1.6	0.4	4.1	15.9	<0.001
Respiratory medications,† %					
H1 antihistamines	9.6	5.1	20.7	32.2	<0.001
ICS/LABA combination	5.1	2.9	10.4	19.7	<0.001
Nasal corticosteroids	6.0	3.1	13.0	20.9	<0.001
SABA singly inhaled	10.9	6.3	22.2	33.9	<0.001
SABA/SAMA combination	5.0	2.7	10.5	21.8	<0.001
Other medications,† %					
ACE inhibitors	6.8	4.0	13.7	19.7	<0.001
Antibiotics for respiratory conditions	19.4	10.7	41.1	59.4	<0.001
Antidepressants	10.8	5.9	23.1	32.2	<0.001
Benzodiazepines	8.4	4.9	16.6	33.5	<0.001
Gabapentinoids	5.5	2.9	11.7	19.3	<0.001
Muscle relaxants	5.8	2.8	12.7	26.4	<0.001
Opioid analgesics	13.7	7.6	28.4	50.6	<0.001
Oral corticosteroids	8.4	4.2	18.6	31.0	<0.001
Proton pump inhibitors	11.4	6.8	23.1	30.1	<0.001
Polypharmacy, %					
≥3 different medications	28.1	17.2	55.3	74.1	<0.001

CC, chronic cough; CM, cough medication; ACE, angiotensin converting enzyme; H1, histamine-1 receptor; ICS, inhaled corticosteroid; LABA, long-acting beta-agonist; SAMA, short-acting muscarinic-antagonist; SABA, short-acting beta-agonist.
†Due to space limits, the table only includes respiratory and other medication use that at least 5% of all CC patients had.

Conclusions

- In Florida, over 70% of CC patients did not have any CM prescriptions, which may indicate that most of the CC patients received only treatment for underlying conditions of CC or used only over-the-counter CM for cough management
- CC patients with chronic prescription CM use was low. Chronic antitussive use may indicate unmet or suboptimal management of CC in a subset of CC patients, potentially those with RCC/UCC
- CC patients with chronic prescription CM use had substantial burdens of health care utilization, comorbidities, and medication use at baseline

Disclosures

This research was funded by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. Xuehua Ke and Helen Ding are employees of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, and own stock in Merck & Co., Inc., Rahway, NJ, USA. Mandel R. Sher has received consulting fees from Merck & Co., Inc. for this study, research funding from Bayer, NeRRe, Bellus, and Shionogi unrelated to this study, and consulting fees from Bayer, Bellus, Merck, NeRRe, Nocion, Shionogi, and Soundable Health unrelated to this study. Wei-Hsuan Lo-Ciganic has received research funding from Bristol Myers Squibb unrelated to this study.

References

1. Song WJ, et al. *Eur Respir J.* 2015;45:1479-1481. doi:10.1183/09031936.00218714
2. Irwin RS, et al. *Chest.* 2006;129:1S-23S. doi:10.1378/chest.129.1_suppl.1S
3. Gibson P, et al. *Chest.* 2016;149:27-44. doi:10.1378/chest.15-1496
4. Morice AH. *Lung.* 2010;188(suppl 1):S87-S90. doi:10.1007/s00408-009-9185-z
5. Chung KF, et al. *Lancet Respir Med.* 2016;4:934-935. doi:10.1016/S2213-2600(16)30373-3
6. Weinberger M, Hendeles L. *Allergy Asthma Proc.* 2018;39:169-176. doi:10.2500/aap.2018.39.4117
7. *Med Lett Drugs Ther.* 2018;60(1562):206-208.
8. Zhou L, et al. *J Clin Med.* 2019;9(1):83. doi:10.3390/jcm9010083

Contact Information

Seonkyeong Yang, University of Florida (yang.se@ufl.edu)

Copies of this presentation obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without permission of the authors.

https://bit.ly/3nONvfi

Copyright © 2023 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.