Medical costs, hospitalizations, and length of stay associated with discontinuations in cycled oral antineoplastics among Medicare-eligible oncology patients.

Staskon, F., PhD; Witt, E.A., PhD; and Havern, L., PharmD

BACKGROUND

- Increasing utilization of cycled oral antineoplastic therapies (where dosages vary per monthly supply) among Medicare-eligible patients has generated interest in examining a discontinuation metric for adherence.
- Research is needed to demonstrate whether reduced gaps in cycled medication therapies (i.e., discontinuations) are associated with reduced total medical costs and hospitalization events.

OBJECTIVES

- To investigate the utilization of cycled antineoplastic therapies for various cancers among Medicare-eligible patients, and medical costs and outcomes associated with discontinuation gaps in monthly adherence.

METHODS

- We used a retrospective cohort design with Medicare-eligible patients 65 years or older from the MarketScan Supplemental and Coordination of Benefits Design database from 2019--2020.
- Discontinuations were calculated as gap in medication supply at intervals of 1.5*last supply in the last 6 months of 2020 (i.e., in last half of year).
- The measure was calculated using 8 generic products in 6 therapy classes as a single therapy at the patient level (see Table 1.).
- New to therapy/diagnosis was inferred from a look back on 2019 cancer diagnosis and medications. Possible indications for on-label end of therapy (not a discontinuation) on a subset of medications was inferred from number of fills in calendar year.
- Patients under 65 years, or without cancer diagnosis or continuous enrollment, or initiating medication within last 45 days of 6-months, or indications of hospice care or transplants were excluded.
- Cancer diagnosed patients were categorized as either having a discontinuation in their medication therapy or not within the later half-year of 2020.
- Logistic regression was used to model odds of discontinuations in 2020, examining the impact of COVID-19 and other variables. General linear models (GLM) examined differences in total medical costs in 2020 by discontinuation or other covariates. The odds of hospitalization were examined in a logistic model that included the binary discontinuations in 2020 and other covariates. Last, length of stay (LOS) was modeled with general linear models by discontinuation and other variables.
- Models included covariates of age (log transform, mean split), census region (Western vs. other), gender (female vs. male), new to therapy/diagnosis (or not), metropolitan area (or not), retail pharmacy (all vs. other), Covid-19 indication (yes or no), and presence of non-cancer Charlson comorbidity index categories (yes or no) and interaction between new to therapy and discontinuation.

RESULTS

- In 2020, 1,357 patients met metric criteria with a mean discontinuation rate of 16.7%.
- As reported in **Table 1**., the most utilized therapy classes per patient were Immunomodulators for Myelodysplastic Syndromes (53.5%) followed by Cyclin-Dependent Kinases (CDK) Inhibitors (23.6%) and Immunomodulators (11.4%).

Table 1. Distribution of therapy classes for 2020 on total fills

Therapy Class	Generic Products	Patient Count	% Patients
Antimetabolites	capecitabine	75	5.5%
Immunomodulators	pomalidomide	154	11.4%
Multikinase Inhibitors	sunitinib, lapatinib	21	1.6%
Cyclin-Dependent Kinases Inhibitors	palbociclib, ribociclib	371	27.3%
Imidazotetrazines	temozolomide	10	<1%
Immunomodulators for Myelodysplastic Syndromes	lenalidomide	726	53.5%

- In the 2020 sample, 64.0% were new to therapy/diagnosis and 5.2% were identified as having COVID-19 with either diagnostic or procedure codes (with 50.7% having both code indications). **Table 2**. shows the other covariate distributions.

Table 2. Distribution of 2020 Dichotomous Covariates (n=1,357)

Age (75+ yrs)	Female	Western Region	Metro-area	New to Tx/Dx	Retail Only	Comor-bidity	COVID-19	Hospitalized
48.2%	60.8%	7.7%	38.3%	64.0%	61.5%	73.3%	5.2%	30.3%

- In a stepwise logistic regression, the 16.7% discontinuation rate in 2020 was predicted from hospital admissions, COVID-19 status, and covariates of age, female census region, retail pharmacy, metropolitan area, new to therapy/diagnosis, non-cancer comorbidity, as well an interaction term between COVID-19 and hospitalization. Only two variables remained in the significant model (X^2=64.4,p<.0001), with increased odds of discontinuation present for hospital admissions (p<.0001) and residing in a metropolitan area (p<.03).
- Note that neither a COVID-19 indication or the interaction term between COVID-19 and hospitalization were significant in predicting discontinuations.
- The simple correlation between COVID-19 indications and discontinuations was not significant in both the total sample (p<.49), where 19.7% of the 71 COVID-19 cases also discontinued, and also when controlling for hospitalizations (p<.39).
- The adjusted means from the GLM indicated the difference between total medical costs significantly increased among those who discontinued in 2020 compared to others ($11,977, p<.03), along with a significant interaction between discontinuations and new to therapy/diagnosis (p<.02).

RESULTS CONTINUED

- That interaction indicated that discontinuations had the most impact among new to therapy/diagnosis patients (a $25,034 increase, p<.0004) compared to non-new to therapy/diagnosis patients (a -$1,079 decrease, p<1). (See **Figure 1**. on adjusted total medical cost differences by discontinuation and interactions with new to therapy/diagnosis).
- Other significant covariates that increased total medical costs were new to therapy/diagnosis (p<.0001), comorbidities (p<.007), and Western census region location (p<.0001). Significantly lower medical costs were present for those using only retail pharmacies (p<.04), older in age (p<.002) and females (p<.0001).
- Finally, patients with indications of COVID-19 also had significantly higher total medical costs compared to non-infected patients ($18,145, p<.007).

Figure 1. Total Medical Costs: Differences in Adjusted Means between Discontinuation Cohorts Overall, and by New to therapy/diagnosis Cohorts.

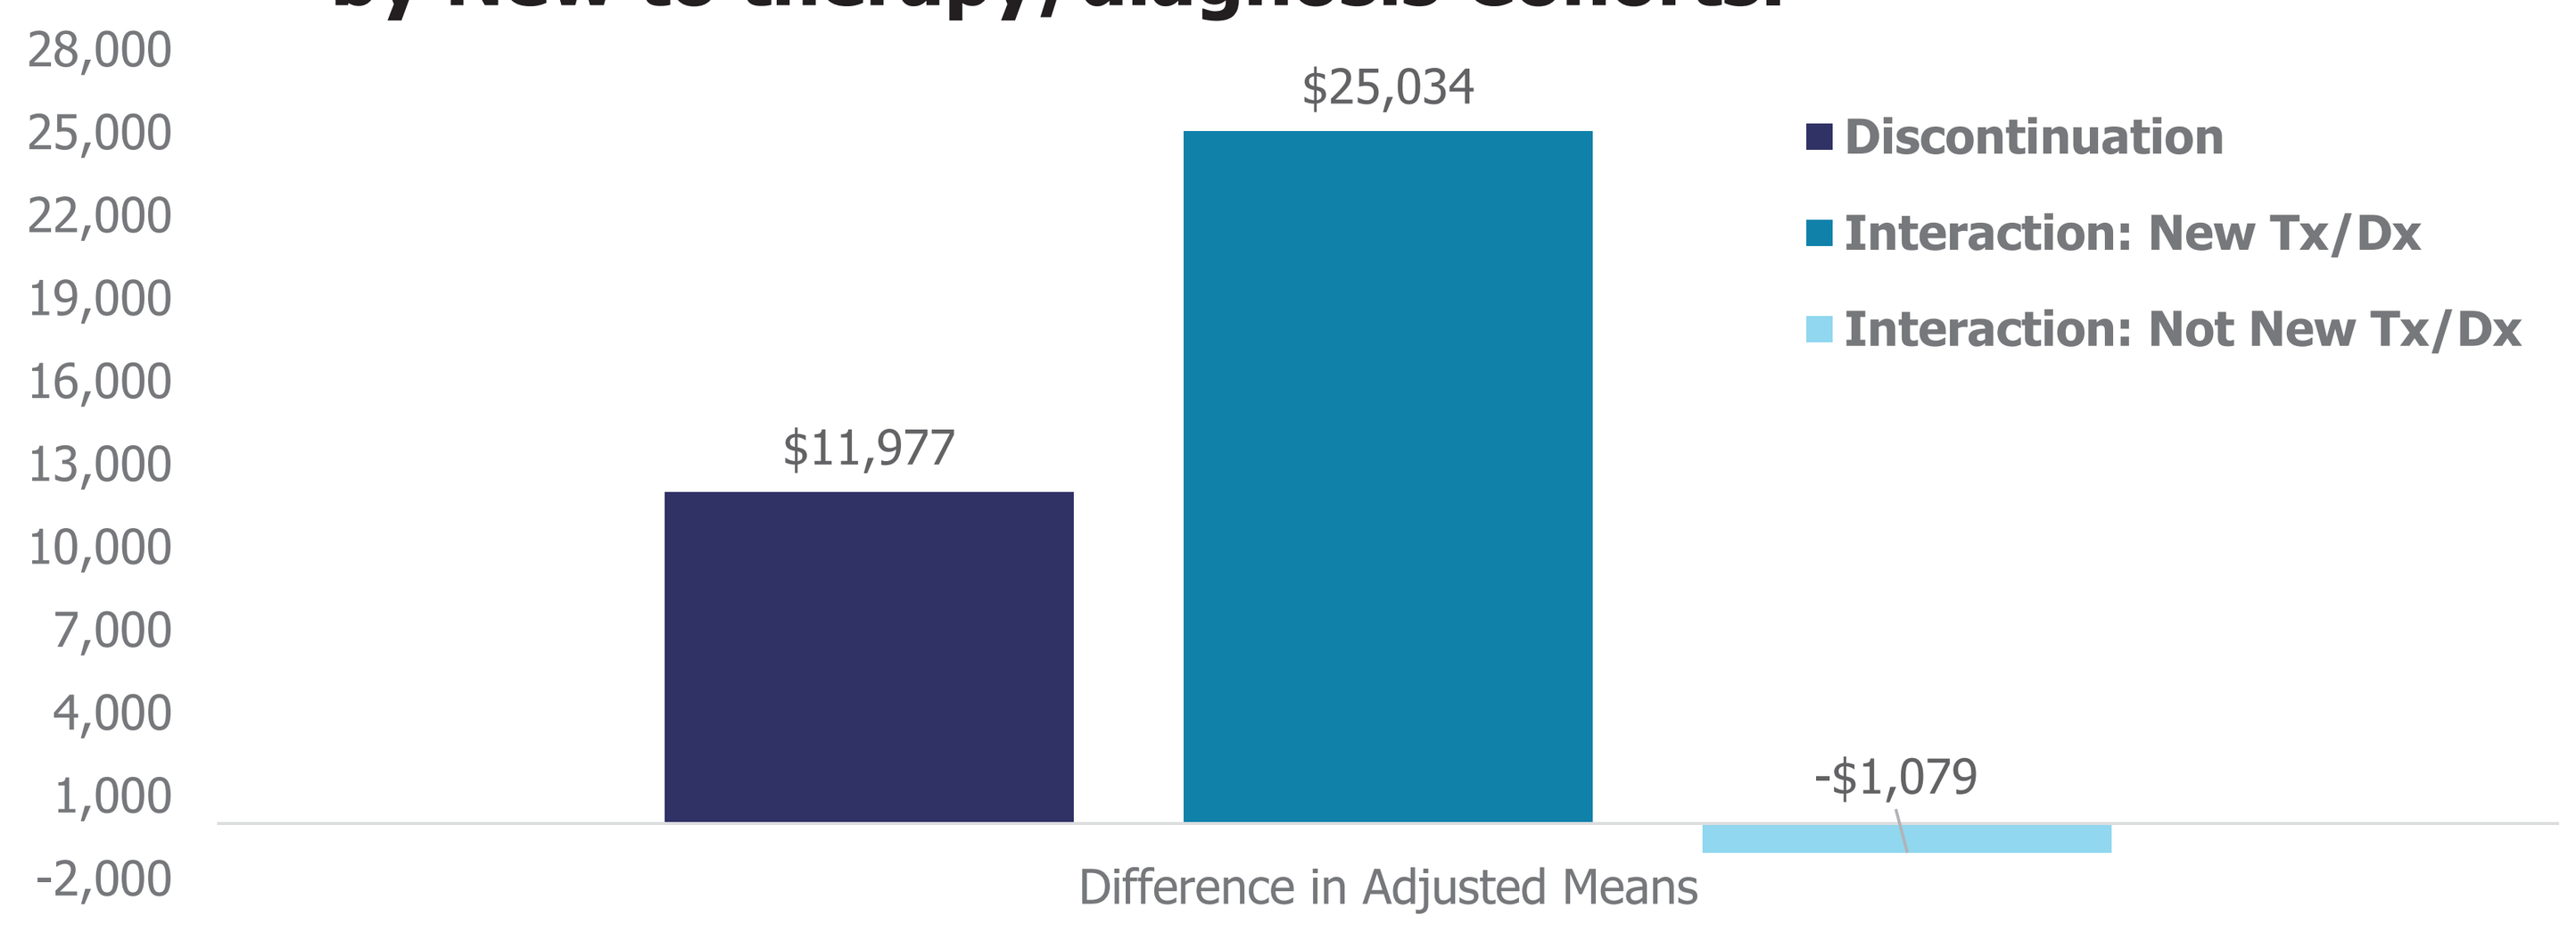
28,000
25,000
22,000
19,000
16,000
13,000
10,000
7,000
4,000
1,000
-2,000
$11,977
$25,034
-$1,079
Difference in Adjusted Means
Discontinuation
Interaction: New Tx/Dx
Interaction: Not New Tx/Dx

- Odds of hospitalization were significantly increased by discontinuations (2.7,p<.0001), COVID-19 infection (6.3,p<.0001). new to therapy/ diagnosis (2.2,p<.0001), and comorbidities (3.5,p<.0001). Being female significantly reduced odds of hospitalization (0.63,p<.0005).
- Overall, LOS was also significantly higher for the discontinued cohort (7.7 days, p<.03) and new to therapy/diagnosis (3.6 days, p<.04), but the interaction of these effects was not significant.
- For COVID-19 infection, there were a significantly longer length of non-cancer diagnosis for LOS (4.6 days ,p<.009)

CONCLUSIONS

- Among Medicare-eligible patients, medication adherence (fewer gaps in medication coverage) on cycled oral antineoplastics can lead to lower medical costs, fewer hospitalization events, and short LOS as compared to lower adherence levels. This was independent of COVID-19 indications that were associated with increased costs, hospitalization, and non-cancer LOS.
- In addition, these results help validate the methodology presented on discontinuations. This metric can be used for yearly reporting requirements by implementing filters controlling for possible end of therapy, late medication starts in the given year, and allowing product switching within a therapy class.

Presented at the 2023 International Society for Pharmacoeconomics and Outcomes Research Conference (ISPOR 2023), May 7 – 10, Boston, MA. This research was approved by Advarra IRB (#Pro00039505) and funded internally by Walgreen Co. All authors are employees of Walgreen Co at time of analysis for whom this research was conducted. Please contact: research@walgreens.com. ©2023 Walgreen Co. All rights reserved.