Eradication of Helicobacter pylori with Vonoprazan-Bismuth Containing Quadruple Therapy in China: A Cost-Effectiveness Analysis

Poster No: EE475

Shengwen Zhu[1], Yan Li[1], Yashi Liu[1], Yanan Sheng[2], Hongchao Li[1]

1. Pharmacoeconomics Evaluation Research Center, School of International Pharmaceutical Business, China Pharmaceutical University, Nanjing, China.211198 , 2. Medical Affairs, Takeda (China) International Trading Company, 100006, Beijing, China

Objective

- The aim of this study was to determine the cost-effectiveness of vonoprazan-bismuth containing quadruple therapy (VBQT) compared with proton-pump inhibitor (PPI)-bismuth-containing quadruple therapy (BQT) in asymptomatic, Helicobacter pylori (H. pylori) infected populations from Chinese healthcare system perspective.

Methods

- Model type: A Markov model with a simulated cohort of 1,000,000 individuals was applied to evaluate the cost-effectiveness of different H. pylori eradication strategies (*Figure 1*).
- Primary outcomes: The main outcomes included gastric cancer incidence reduction, number of cause-specific deaths avoided, costs, quality-adjusted life year (QALY), and incremental cost-effectiveness ratio (ICER). And costs and health outcomes have been discounted at 5% per year.
- Data sources: Epidemiological parameters and health utilities used in the model were collected from published literatures or statistical bureaus.
- Uncertainty analysis: One-way sensitivity analysis and probabilistic sensitivity analysis was conducted to explore the influence of parameters on the uncertainty of the model.

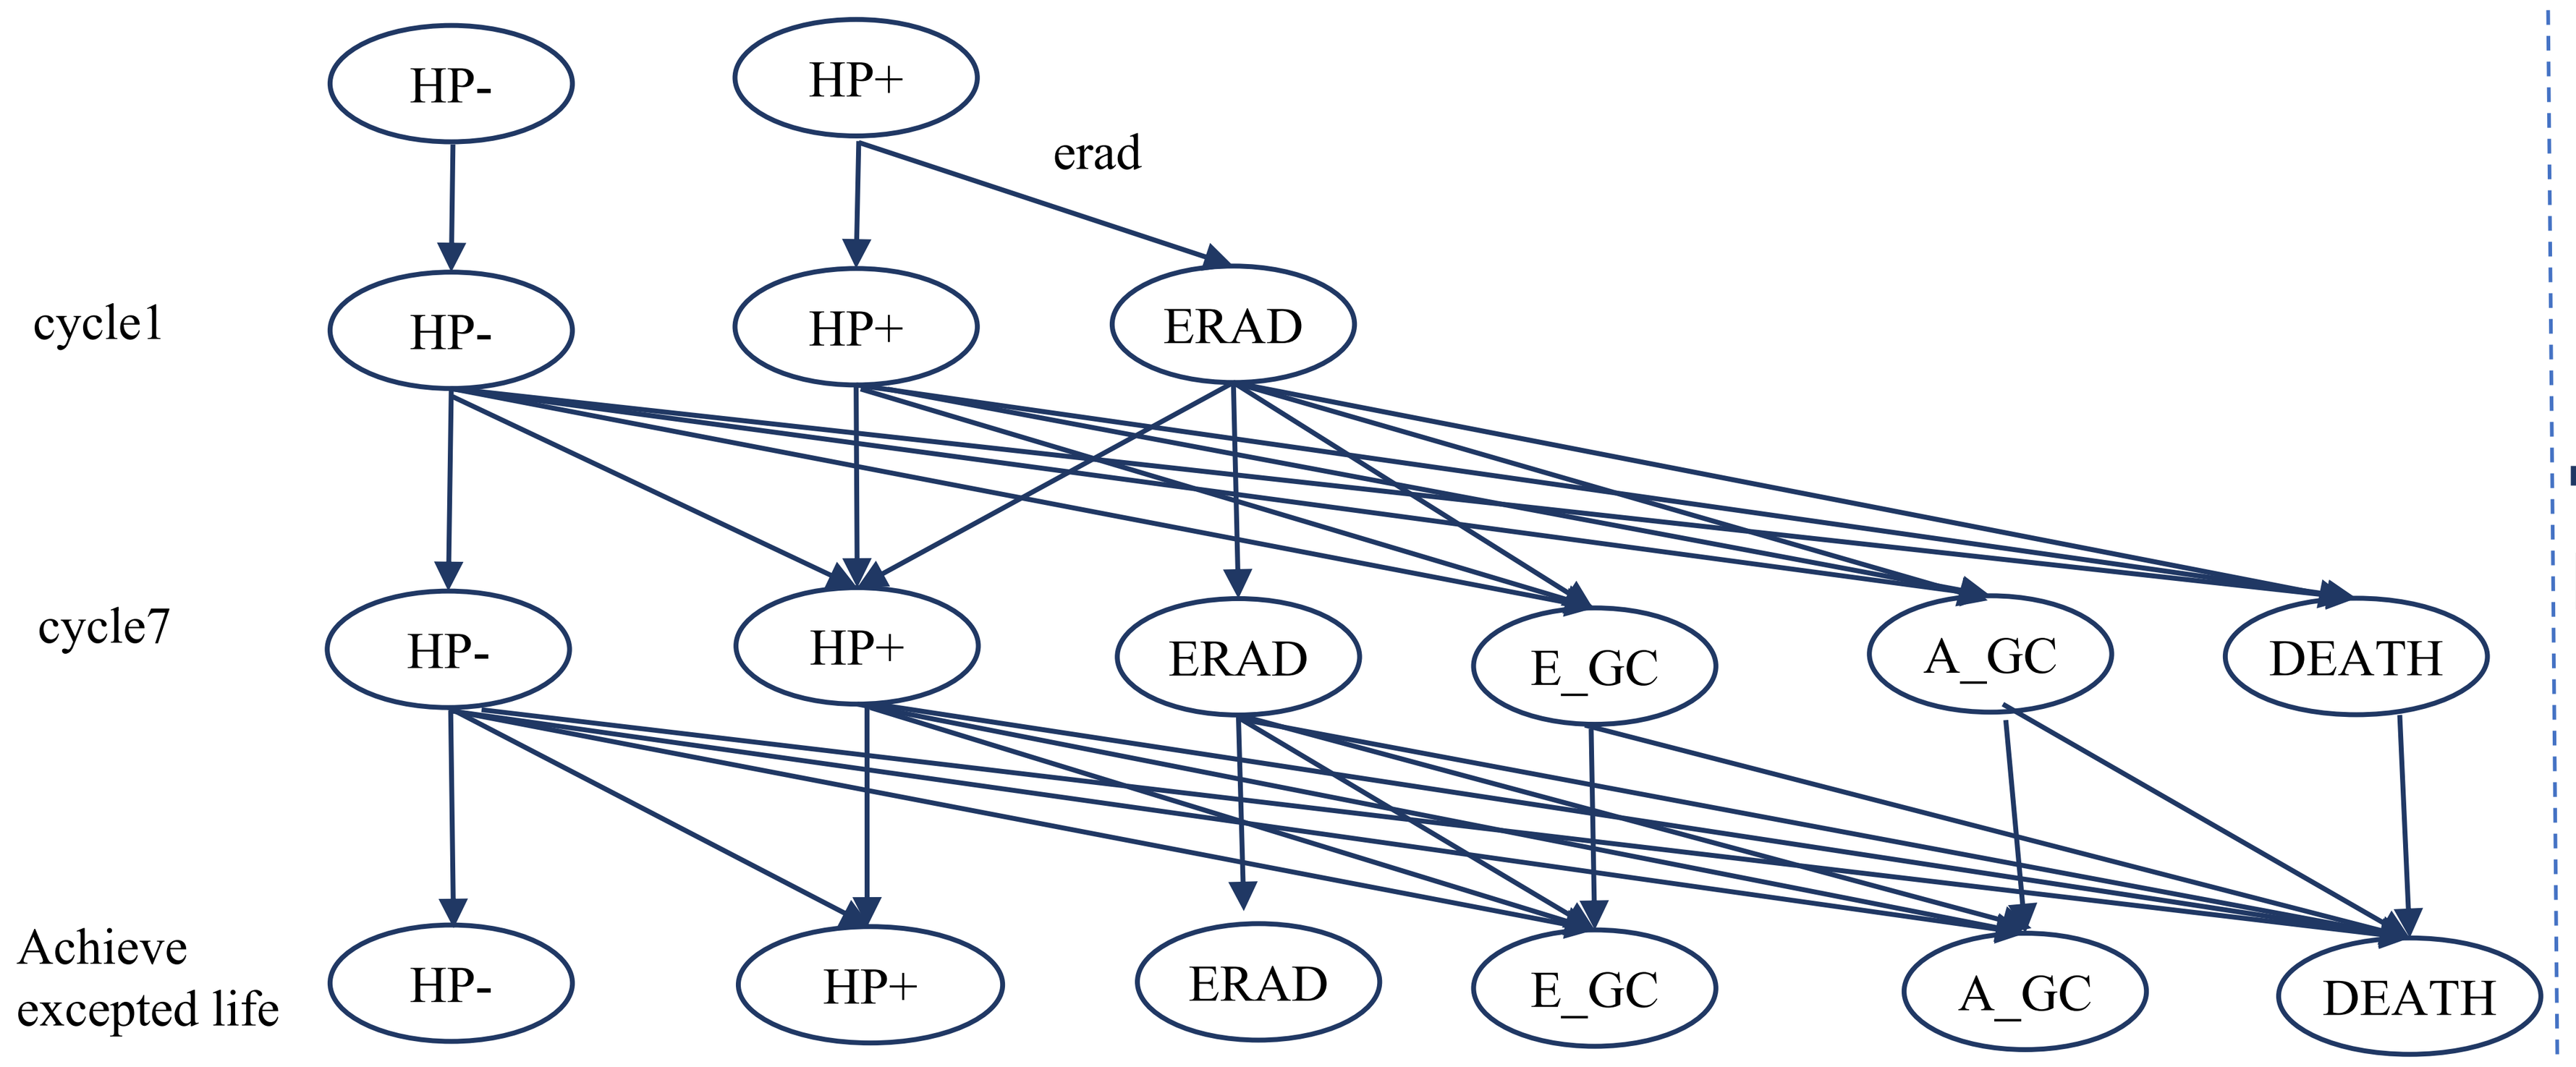
HP-
HP+
erad
cycle1
HP-
HP+
ERAD
cycle7
HP-
HP+
ERAD
E_GC
A_GC
DEATH
Achieve excepted life
HP-
HP+
ERAD
E_GC
A_GC
DEATH

HP-: H. pylori-negative; HP+:H. pylori-positive; E_GC:early gastric cancer; A_GC:Advanced gastric cancer

Figure 1 Simplified model schematic

Results

- VBQT therapy brought an average of 0.05 incremental QALYs and saved ￥7.36 compared with the BQT group. Therefore, compared with BQT group, VBQT appears to be a dominant strategy (more QALYs gained, and less cost incurred). Meanwhile, VBQT group prevented 253 cases of early gastric cancer and 1012 cases of advanced gastric cancer (*Table 1*).

Table 1 Cost –Effectiveness Analysis Result

Interventions	Cost per capita	QALY per capita	Reduced number of E_GC	Reduced number of A_GC	ICER
BOT	2157.97	17.66	23577.11	94308.46	-
VBQT	2150.61	17.71	23324.22	93296.88	-
Incremental (VBQT vs BOT)	-7.36	0.05	-252.90	-1011.58	-143.40

VBQT, vonoprazan-bismuth containing quadruple ; BQT, proton-pump inhibitor (PPI)-bismuth-containing quadruple therapy; QALY, quality-adjusted life year; ICER, incremental cost-effectiveness ratio (ICER); *E_GC:early gastric cancer; A_GC:Advanced gastric cancer*

- In one–way sensitivity analysis, the cost-effectiveness was sensitive to infection rate, the cost of VBQT, the cost of BQT and the utility value of HP+ . The one-way sensitivity analyses showed that the results was not sensitive to any variables (*Figure 2*).
- The cost acceptability curve demonstrated that VBQT had a 52% probability of being more cost-effective than BQT when the willingness-to-pay (WTP) thresholds were 0. When the WTP was one times, two times and three times the GDP per capita, the probability of VBQT being cost-effective was close to 90% in all cases (*Figure 3*).

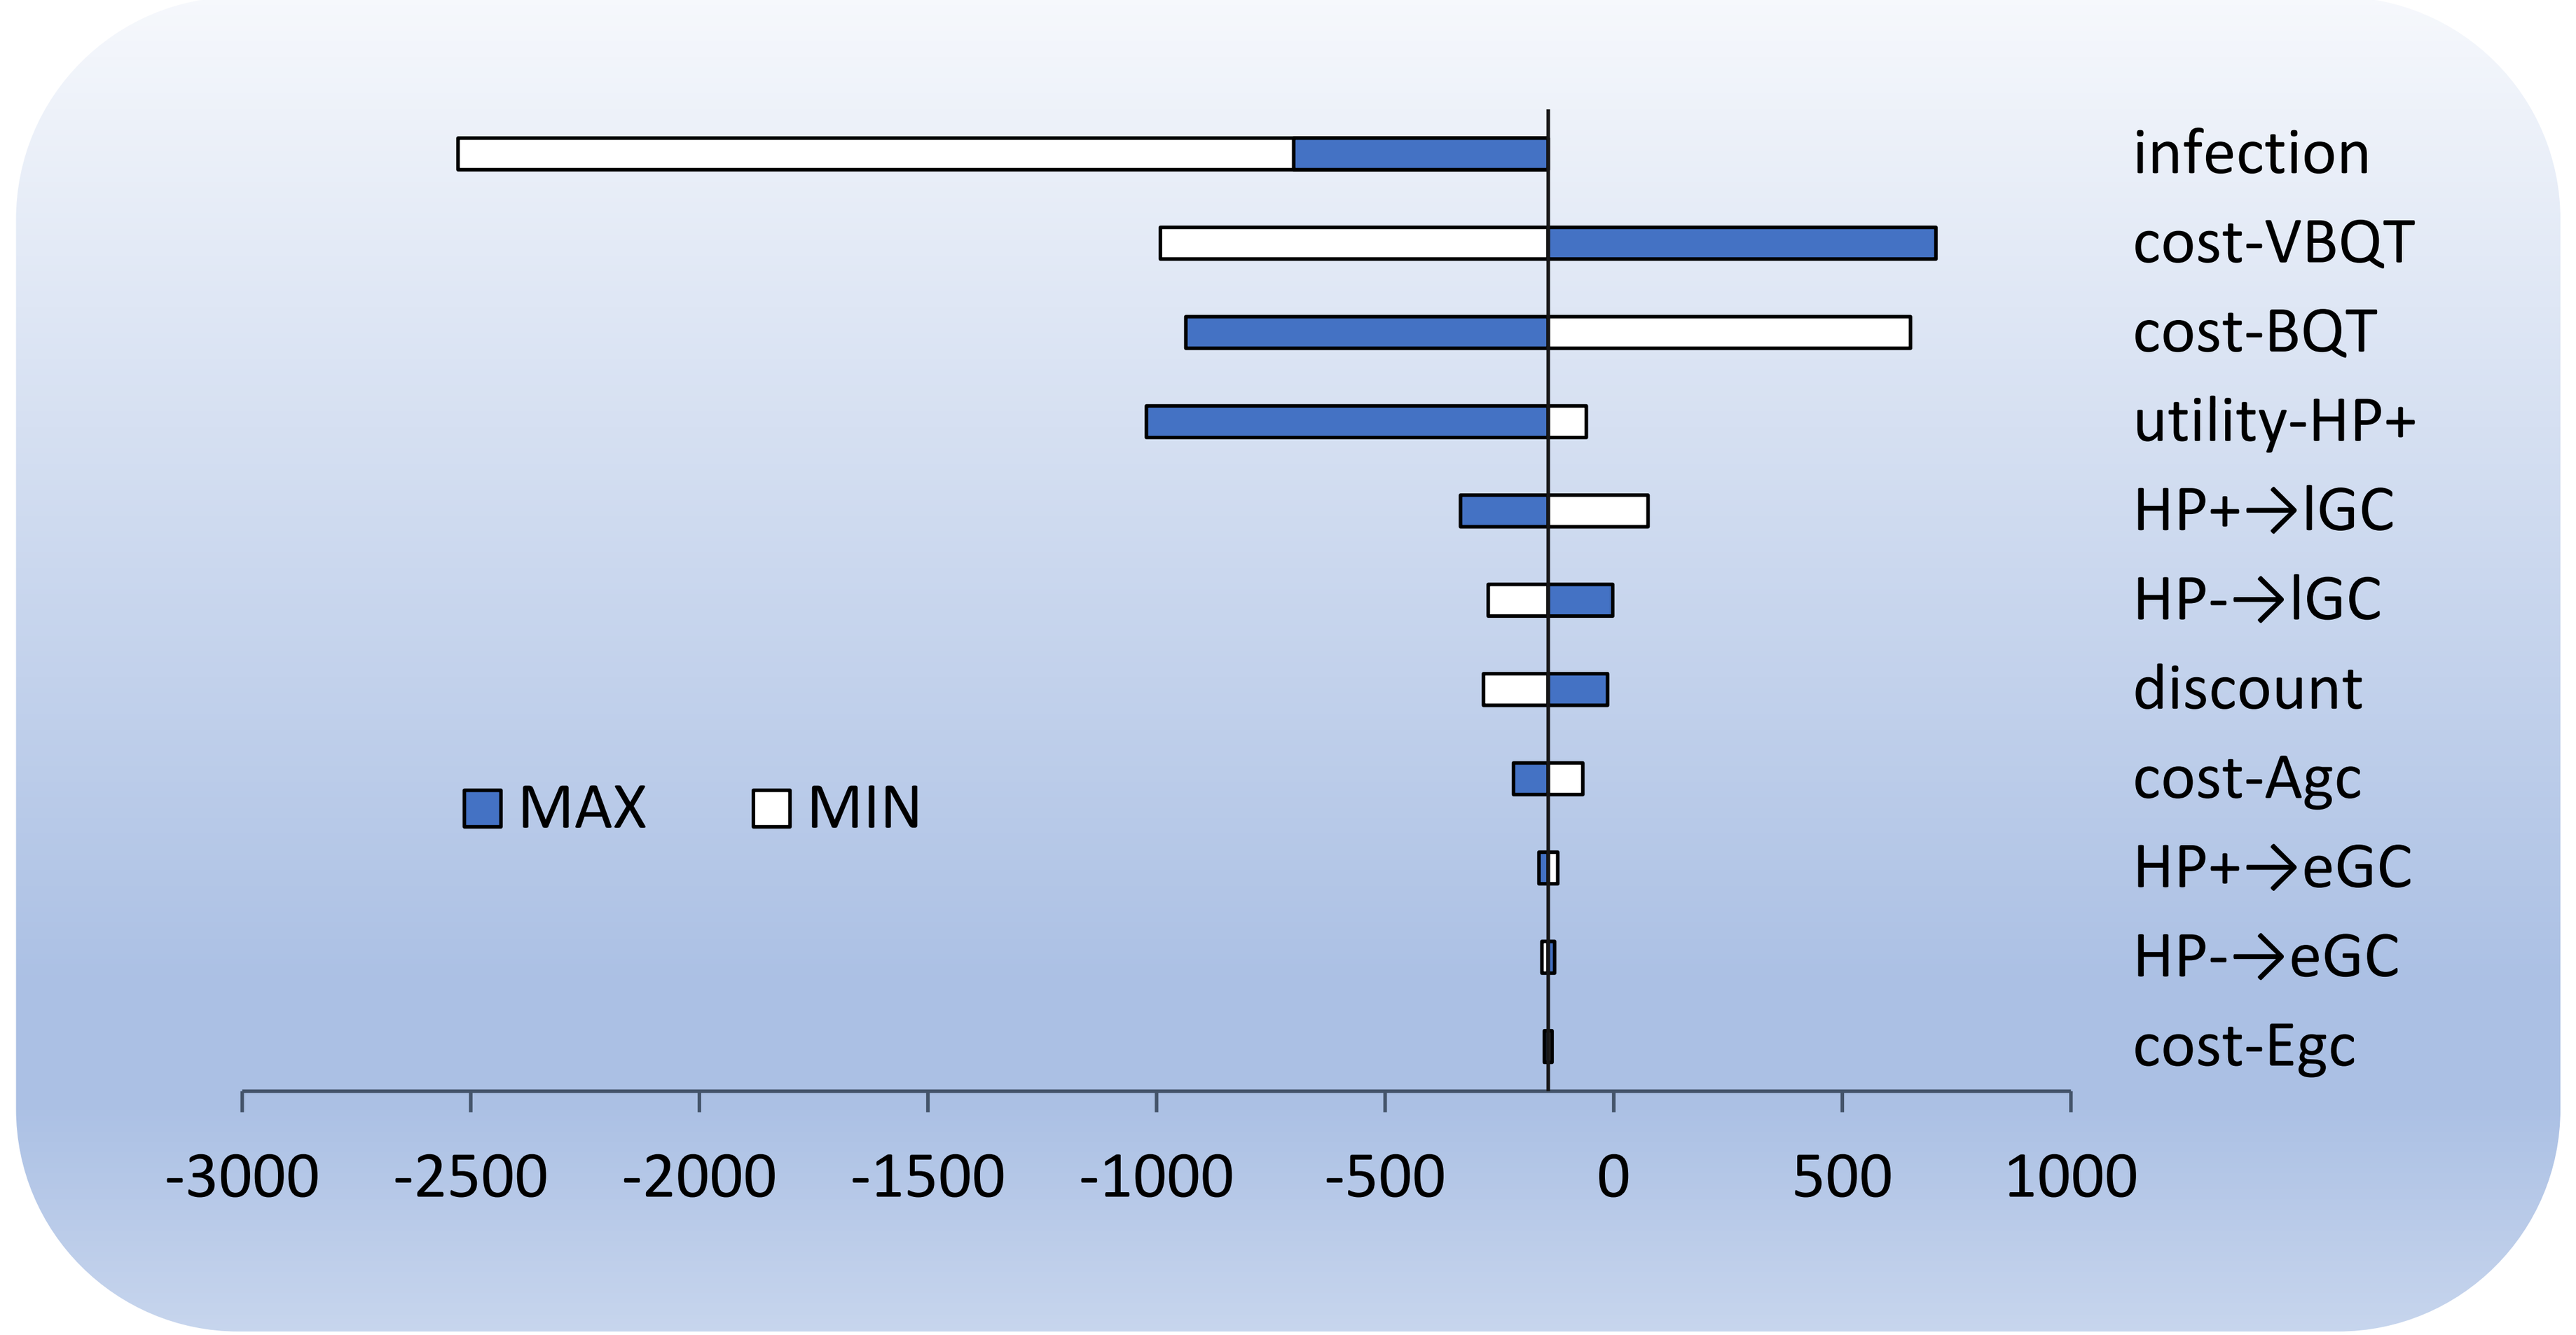
infection
cost-VBQT
cost-BQT
utility-HP+
HP+→lGC
HP-→lGC
discount
cost-Agc
HP+→eGC
HP-→eGC
cost-Egc
MAX
MIN
-3000
-2500
-2000
-1500
-1000
-500
0
500
1000

Figure 2 Tornado diagram

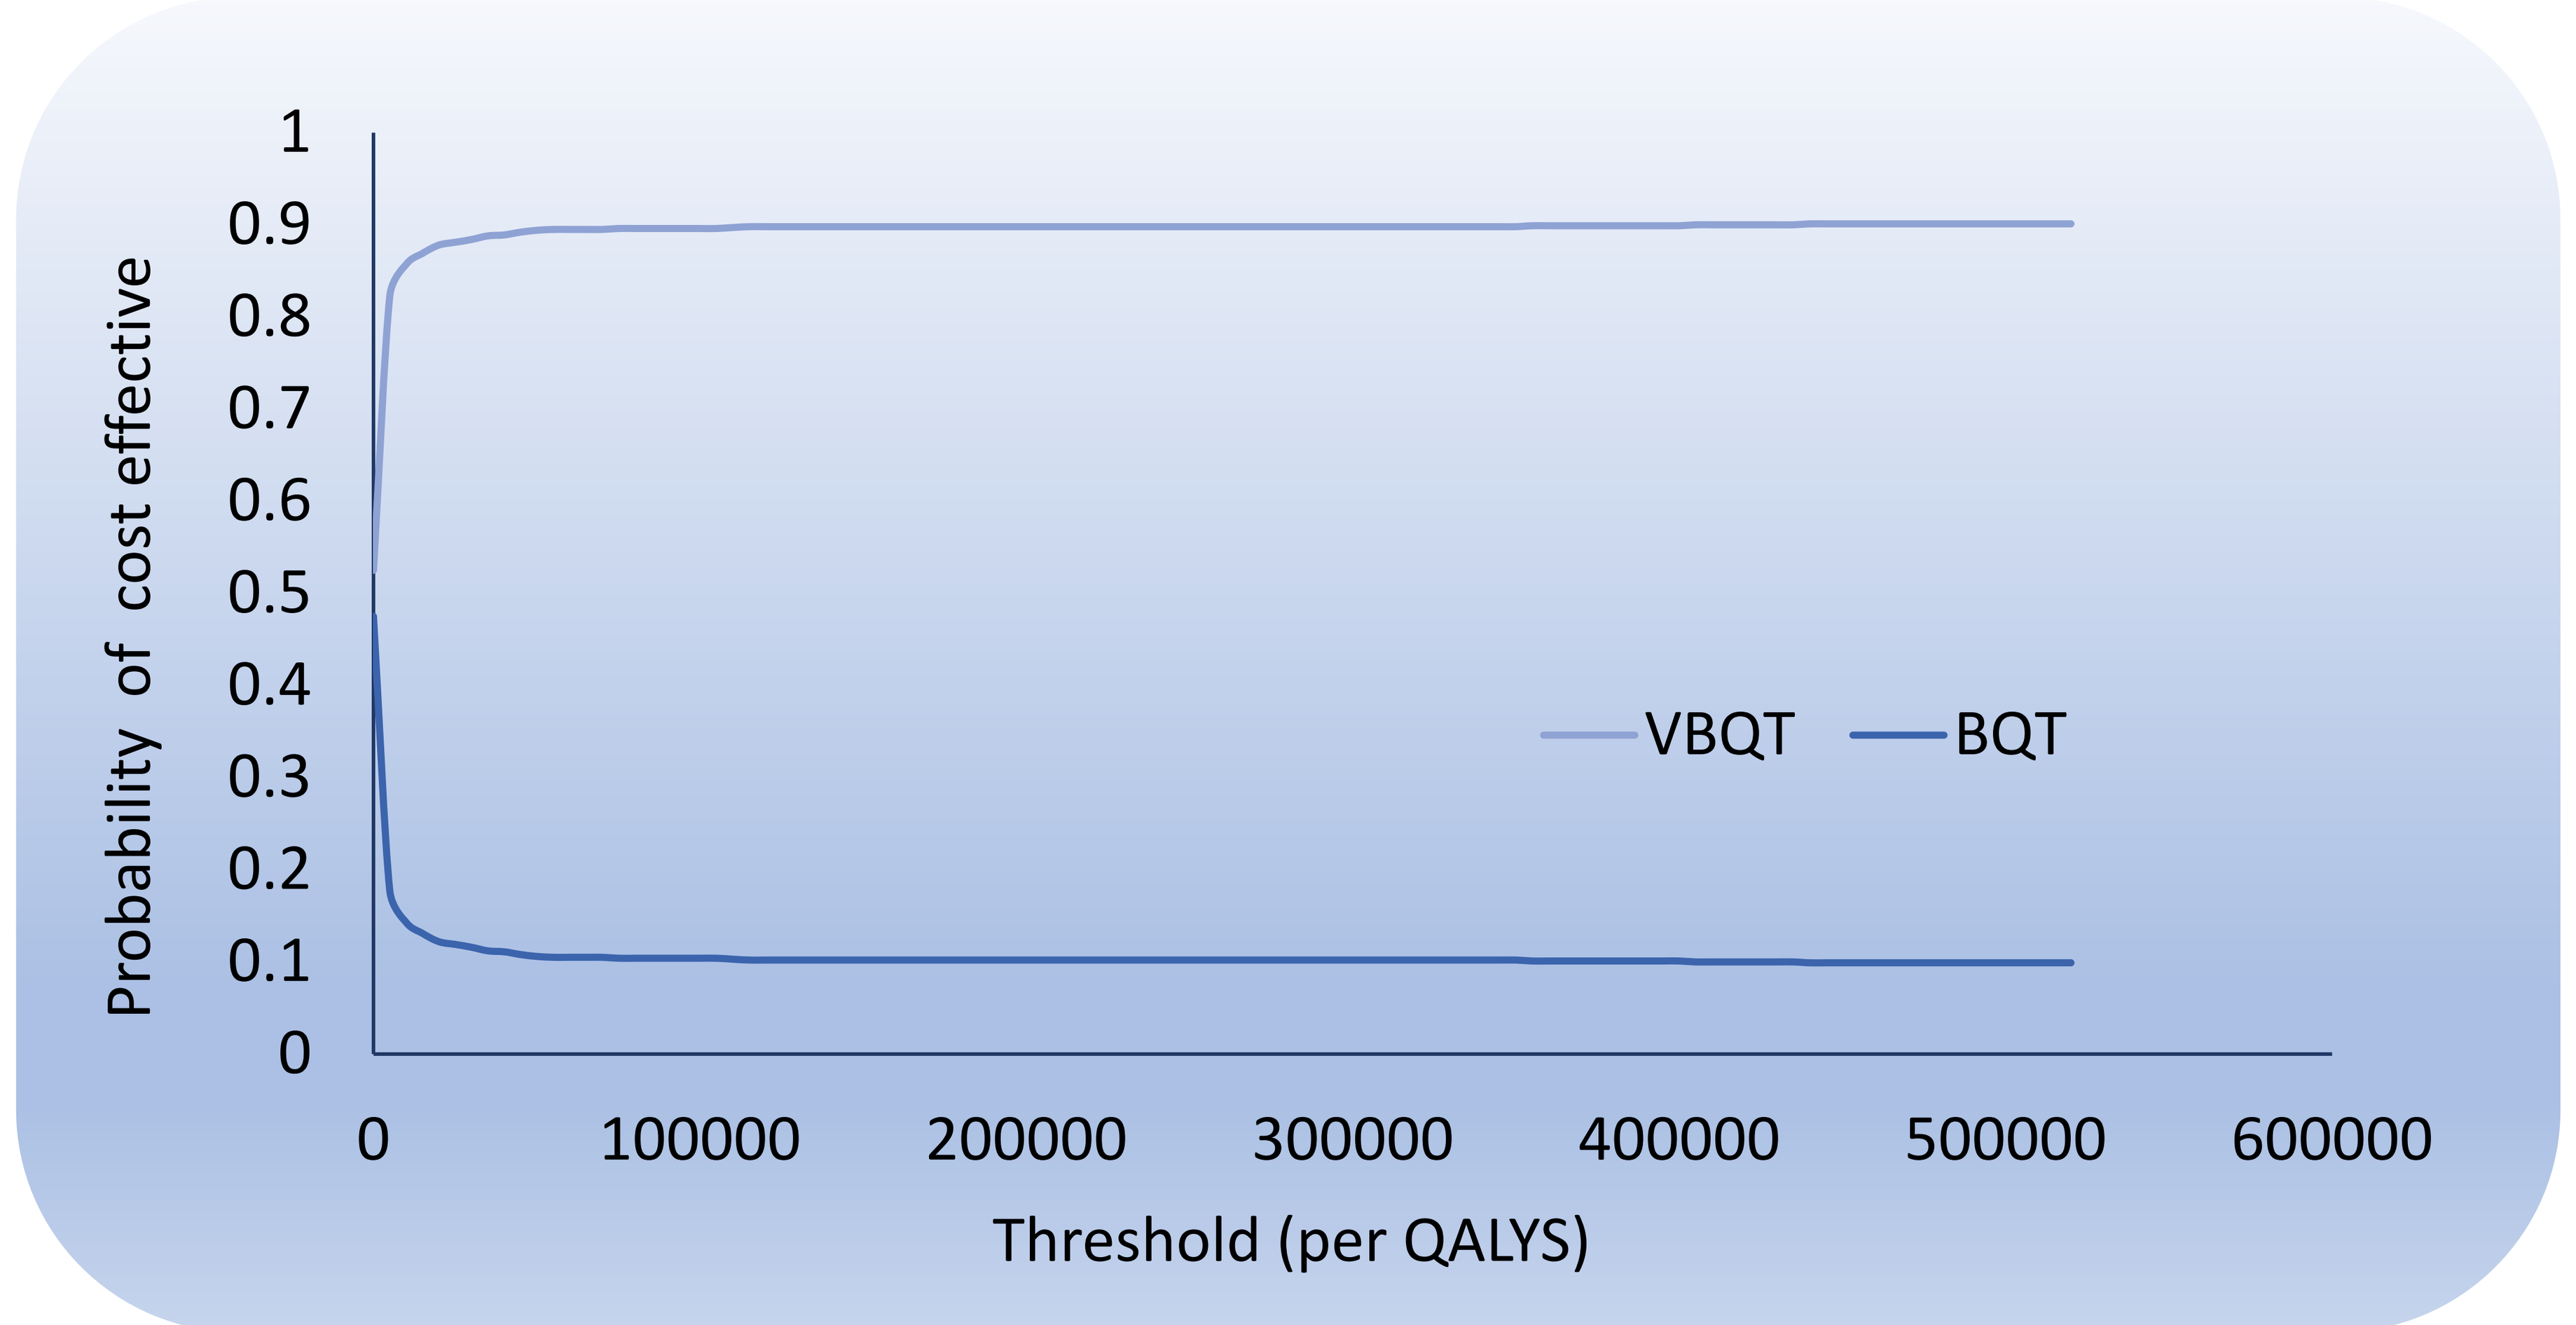
Probability of cost effective
1
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
VBQT
BQT
0
100000
200000
300000
400000
500000
600000
Threshold (per QALYS)

VBQT, vonoprazan-bismuth containing quadruple ; BQT, proton-pump inhibitor (PPI)-bismuth-containing quadruple therapy; QALY, quality-adjusted life year

Figure 3 Cost -Effectiveness Acceptability Curve

Conclusion

- VBQT may be more effective in reducing the number of gastric cancers, lowering costs, and increasing QALYs compared to BQT therapy. Therefore, VBQT would be a cost-effective treatment compared with BQT for the eradication of H. pylori among patients with asymptomatic infection in China.

Disclosure

- This study was funded by Takeda (China) International Trading Company. Yanan Sheng is an employee of Takeda (China) International Trading Company

Presented at ISPOR 2023, 9:00AM-10:00PM, May 10, 2023, Boston Conference & Exhibition Center, Boston, Massachusetts, USA.