Healthcare Resource Utilization and Economic Burden of Hidradenitis Suppurativa in the USA Using MarketScan Claims Data

Christopher Sayed,[1] Amit Garg,[2] Jaco Voorham,[3] Tanja Tran,[4] Silky Beaty,[5] David Friesen,[6] Joslyn S Kirby[7]

Presented at ISPOR 2023 | May 7–10 | Boston, MA, USA

RWD181

Objective

To investigate the economic burden of hidradenitis suppurativa (HS) by evaluating healthcare resource utilization (HCRU) and associated costs of insured patients with hidradenitis suppurativa in the USA using MarketScan data.

Background

- HS is a chronic and inflammatory skin disease characterized by recurrent, painful deep lesions, and is associated with multiple comorbidities.[1]
- Patients with HS require frequent healthcare encounters. High-cost settings, such as emergency department and inpatient care, are used frequently by patients.[2] These costs are hypothesized to increase with disease severity.
- It is important for both patients, physicians and payers to better understand this economic burden based on healthcare resource utilization and associated costs of patients with HS.

Methods

- This observational cohort study used Merative® MarketScan® Commercial, Medicare, and Multi-State Medicaid claim databases.
- Included patients had ≥1 claim with a diagnostic code for HS on or before January 1, 2017, with continuous enrollment and medical and drug coverage during 2016. Patients on a capitated health plan on January 1, 2017 were excluded.
- Patients were censored when enrollment stopped, or upon switching to a capitated health plan.
- Healthcare resource types were reported according to the definitions provided in **Table 1**. They were based on HS-related claims, unless stated otherwise.
- All-cause and HS-related HCRU, measured as rate per 1,000 patient-years (kPY), and related costs in 2017 are presented stratified by obesity and disease severity.
- Disease severity was defined by two separate proxies:
 - History of biologic medication in 2016.
 - History of in/outpatient HS-specific surgery in 2016.

Results

- The mean age of the 54,912 included patients with HS was 40.0 years; 24.3% of patients were male (**Table 2**), 2.8% had a history of biologic medication and 16.4% had a history of HS-specific surgery.
- Patients with greater disease severity were more often obese, had more comorbidities, and used more HS-related medication than those with less severe HS (**Table 2**).
- The mean rate of HS-related outpatient claims per kPY was 726.6, making up only 3.9% of all-cause outpatient claims (**Table 1**, **Figure 1A**).
 - Results were consistent across insurance types (**Figure 1**).
- Compared with outpatient claims, rates of HS-related emergency department (14.4 per kPY) and inpatient claims (2.7 per kPY) were much lower; each were 1.7% of the all-cause claims (**Table 1**, **Figure 1A**).
- The claim rate ratio among HS-related outpatient claims was 1.6 for obesity/no obesity, 6.1 for biologic/no biologic and 3.5 for surgery/no surgery (**Figure 2**).
- However, rates for both HS-related emergency department and inpatient claims were 3.5–12 times higher in patients with severe disease.
- Among all patients, 81.7% used HS-related medication and 94.3% of HS-related claims were for medication. This covers 48.4% of all-cause medication claims and 89.1% of HS-related healthcare costs (**Table 3**, **Figure 3A**, **B**).
- Outpatient costs were the biggest contributor to all-cause costs at 41.0%, with 90.2% of patients utilizing this healthcare resource (**Figure 3C**, **Table 3**).
- Overall, total HS-related costs contributed to 17.9% of total all-cause costs.

Conclusions

There is considerable variation in HCRU and costs in patients with HS, partly driven by disease severity; outpatient and inpatient claims were greater in patients with severe disease.

Most HS-related resource utilization occurs with medication and in the outpatient setting.

A limitation of this study is that only inpatient and outpatient resources with an HS International Classification Disease code were considered, which may have led to an underestimation of costs of care related to HS.

The high proportion of non-HS-related healthcare resources in most patients suggests an important comorbidity burden in this patient population.

Summary

This observational cohort study investigated the **HCRU** of patients with HS in the **USA** using **Merative® MarketScan® Commercial, Medicare, and Multi-State Medicaid data**

Greater disease severity was associated with:

Obesity

Comorbidities

HS-related medication use

HS-related outpatient, inpatient and emergency department claim rates were **3.5–12 times higher** in patients with severe disease

Most resource utilization could be attributed to **medication** and in the **outpatient** setting

Table 1 Mean rate of HS-related and all-cause claims

Healthcare resource claim	Resource definition	HS-related claims (per kPY)	All-cause claims (per kPY)
Outpatient	Outpatient claims, excluding claims captured for emergency department attendance and the medication group	726.6	18,722.6
Dermatologist	Outpatient claims with a dermatologist specialist as provider type	151.5	416.9
Inpatient	Inpatient claims, minus emergency department claims; HS-related claims only considered those with HS code in the primary position	2.7	153.7
Emergency department	Emergency department claims	14.4	840.4
Medication	Pharmacy claims, plus medication application procedure claims	12,314.6	25,443.3
Surgery	Inpatient or outpatient claims for HS-related surgical procedures	211.2	–
Total	Sum of inpatient, outpatient, emergency department, and medication	13,058.2	45,160.1

HS-related outpatient, inpatient, and emergency department only included claims with an HS code; HS-related surgery and medication only included those used to treat HS.

Table 2 Baseline characteristics of patients

	Total (N=54,912)	With biologic at baseline (n=1,539)	With surgery at baseline (n=9,027)
Age, years, mean (SD)	40.0 (15.4)	41.3 (13.8)	40.3 (14.8)
Male, n (%)	13,354 (24.3)	395 (25.7)	2,232 (24.7)
Race, n (%)[a]			
n (% non-missing)	10,677 (19.4)	241 (15.7)	2,071 (22.9)
Black	5,821 (54.5)	115 (47.7)	1,219 (58.9)
White	4,347 (40.7)	113 (46.9)	773 (37.3)
Region, n (%)[b]			
n (% non-missing)	43,379 (79.0)	1,268 (82.4)	6,803 (75.4)
North Central	8,732 (20.1)	230 (18.1)	1,390 (20.4)
Northeast	7,585 (17.5)	199 (15.7)	1,217 (17.9)
South	22,803 (52.6)	707 (55.8)	3,571 (52.5)
West	4,259 (9.8)	132 (10.4)	625 (9.2)
Insurance type, n (%)			
Medicaid	11,459 (20.9)	269 (17.5)	2,213 (24.5)
Commercial/Medicare	43,453 (79.1)	1,270 (82.5)	6,814 (75.5)
Prior adalimumab use, n (%)	1,060 (1.9)	1,060 (68.9)	347 (3.8)
Prior excision or drainage surgery, n (%)	8,479 (15.4)	430 (27.9)	8,479 (93.9)
Any HS-related medication used in the baseline year, n (%)	47,807 (87.1)	1,506 (97.9)	8,299 (91.9)
Comorbidities in the previous year, n (%)			
Acne vulgaris	4,148 (7.6)	213 (13.8)	806 (8.9)
Obesity	14,558 (26.5)	507 (32.9)	3,176 (35.2)
Anxiety	9,680 (17.6)	346 (22.5)	2,050 (22.7)
Depression	8,657 (15.8)	335 (21.8)	1,895 (21.0)

[a]Medicaid data only; [b]Commercial/Medicare data only.

Table 3 Proportion of patients with at least one healthcare resource use

Healthcare resource claim	HS-related claims (%)	All-cause claims (%)
All resources	82.5	93.2
Outpatient	21.3	90.2
Inpatient	0.2	8.8
Emergency department	0.7	29.5
Medication[a]	81.7	87.1
In/outpatient surgery	11.4	–

[a]Includes biologics.

Figure 1 Percentage of all-cause claims covered by HS-related claims

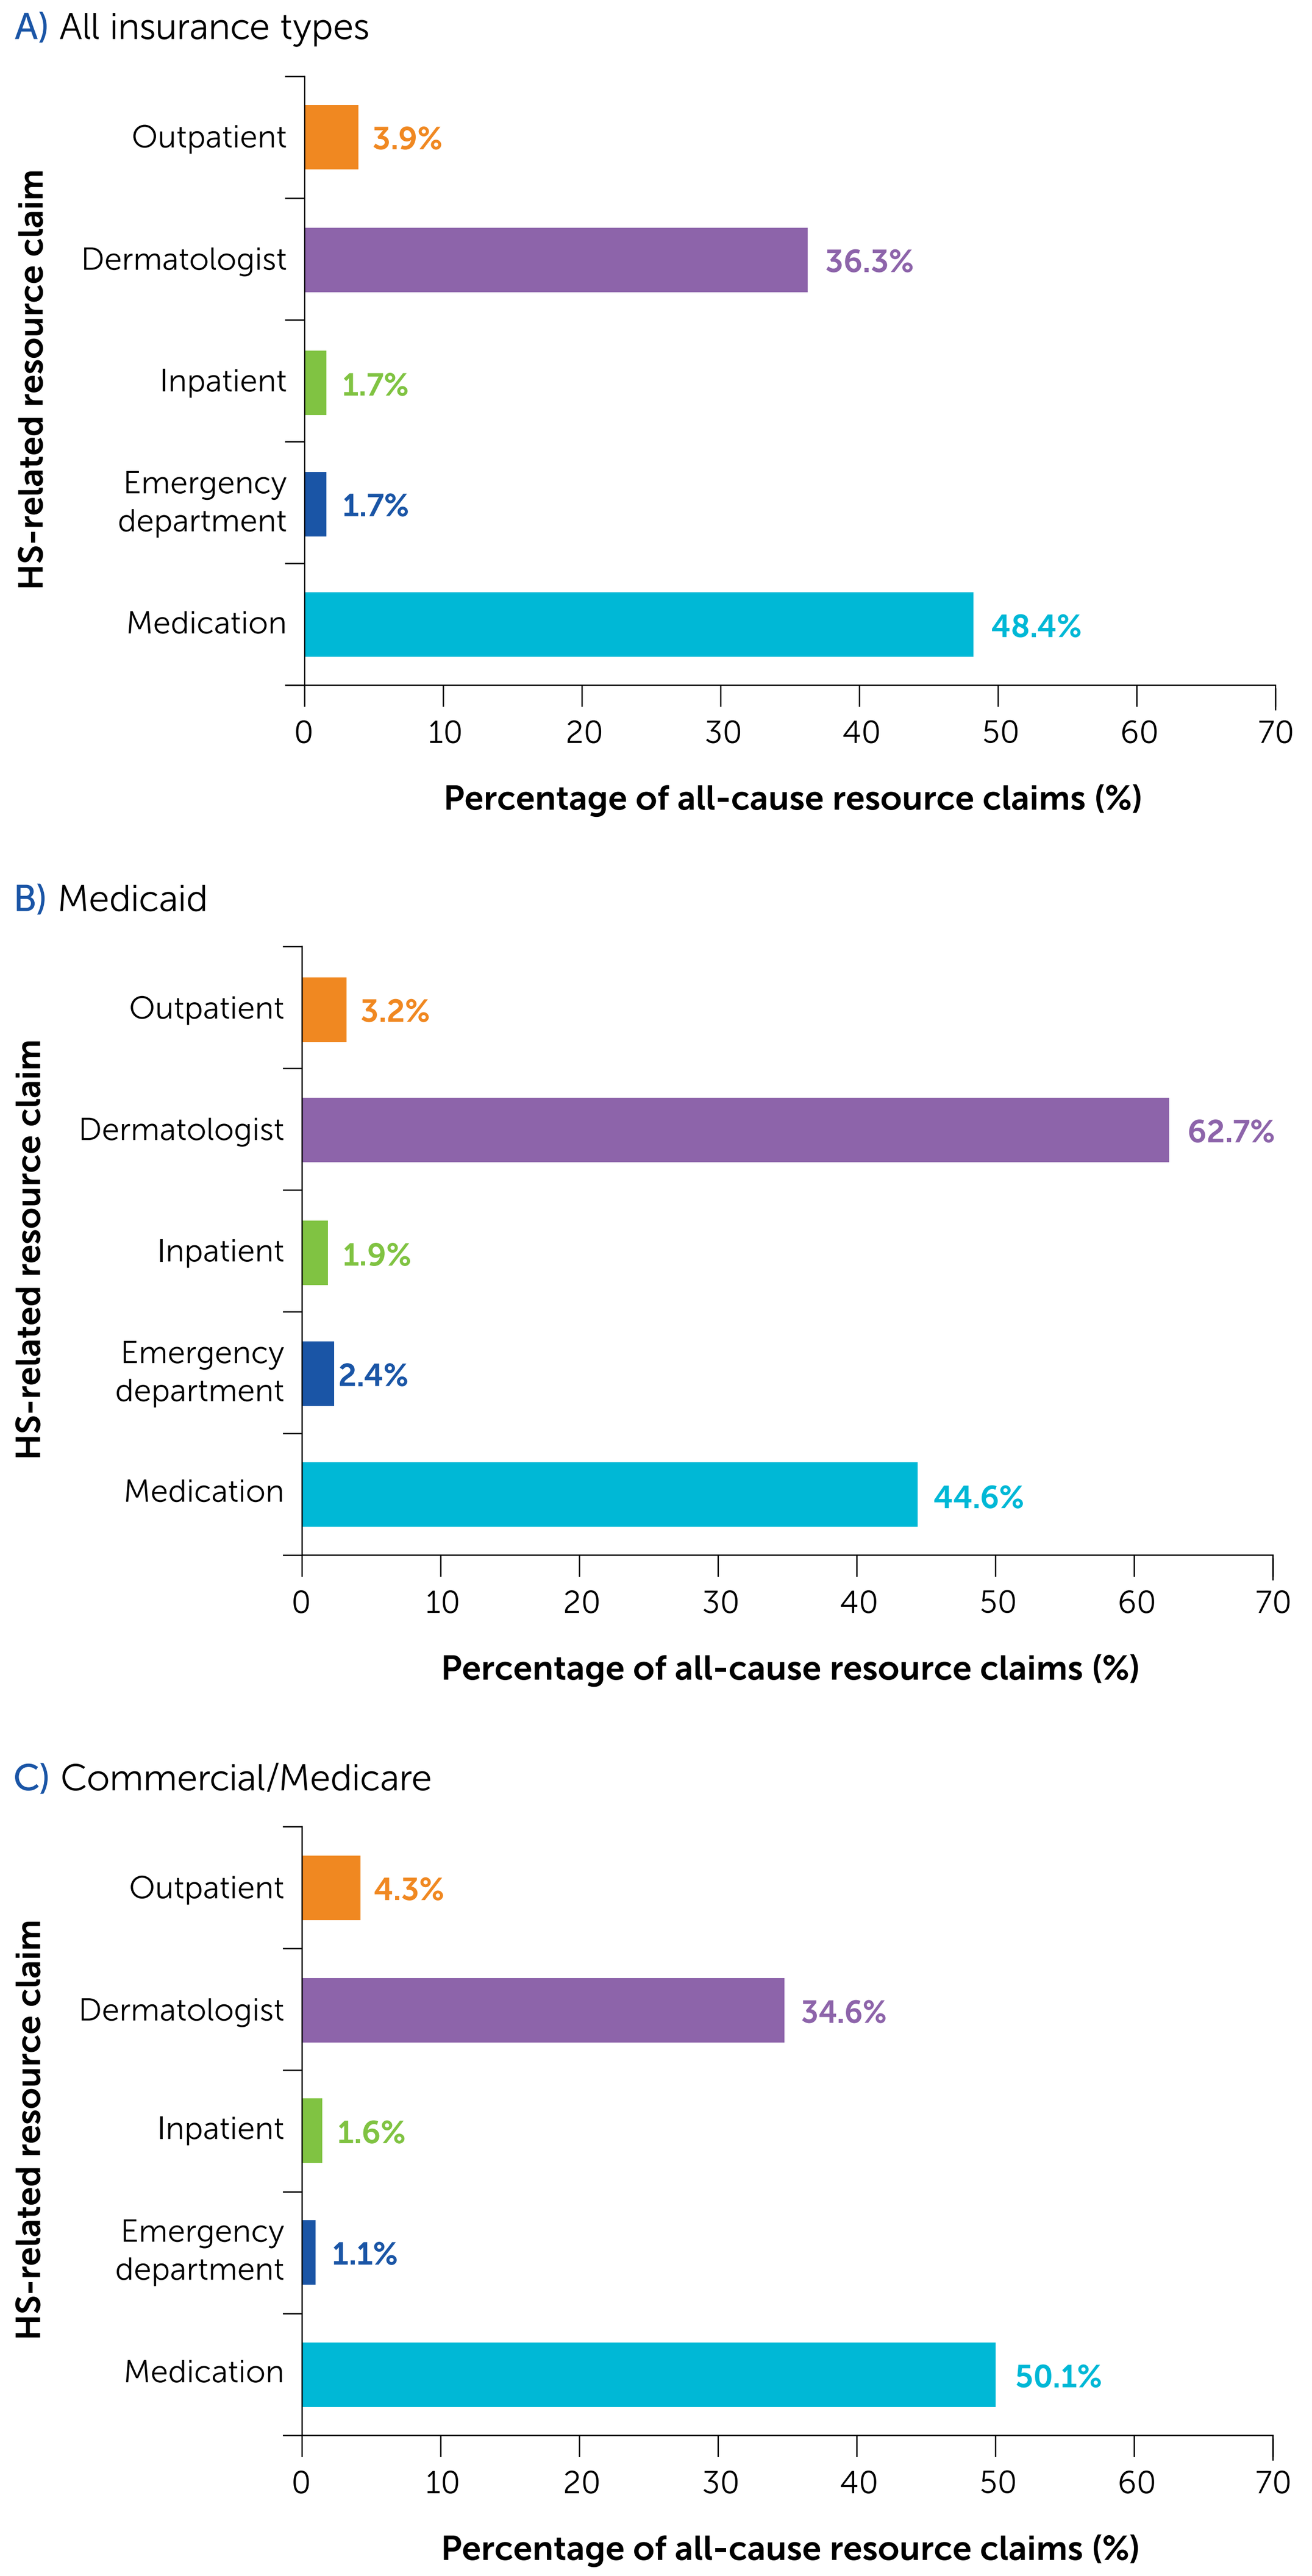
A) All insurance types
HS-related resource claim
Outpatient
Dermatologist
Inpatient
Emergency department
Medication
3.9%
36.3%
1.7%
1.7%
48.4%
0
10
20
30
40
50
60
70
Percentage of all-cause resource claims (%)
B) Medicaid
HS-related resource claim
Outpatient
Dermatologist
Inpatient
Emergency department
Medication
3.2%
62.7%
1.9%
2.4%
44.6%
0
10
20
30
40
50
60
70
Percentage of all-cause resource claims (%)
C) Commercial/Medicare
HS-related resource claim
Outpatient
Dermatologist
Inpatient
Emergency department
Medication
4.3%
34.6%
1.6%
1.1%
50.1%
0
10
20
30
40
50
60
70
Percentage of all-cause resource claims (%)

Figure 2 HS-related outpatient claims stratified by obesity and disease severity indicators

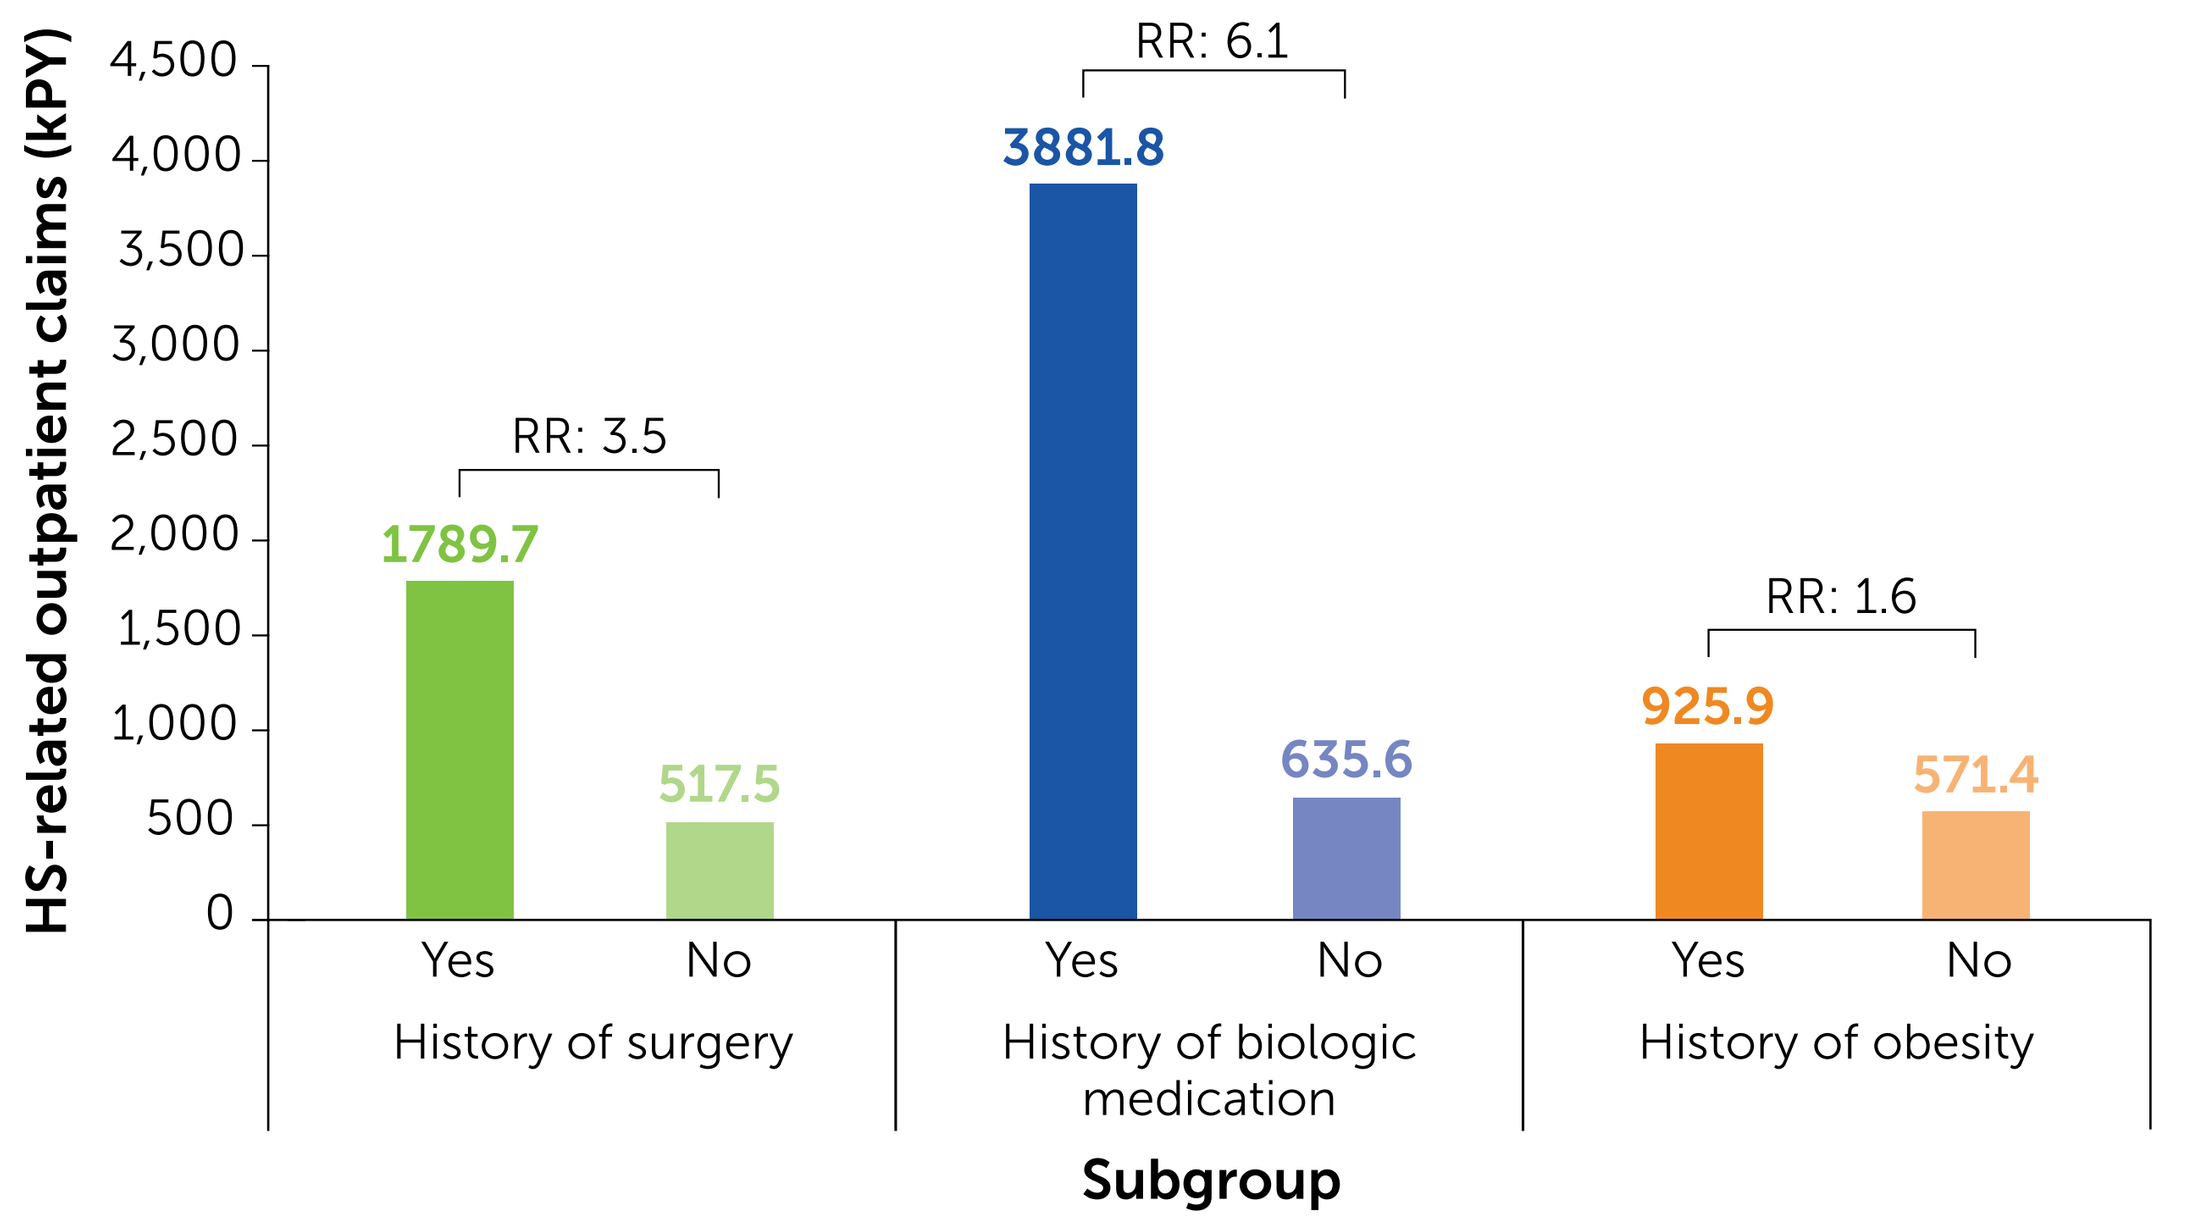
HS-related outpatient claims (kPY)
4,500
4,000
3,500
3,000
2,500
2,000
1,500
1,000
500
0
RR: 3.5
RR: 6.1
RR: 1.6
1789.7
517.5
3881.8
635.6
925.9
571.4
Yes
No
History of surgery
Yes
No
History of biologic medication
Yes
No
History of obesity
Subgroup

Figure 3 Percentage of total costs and total HCRU by healthcare resource

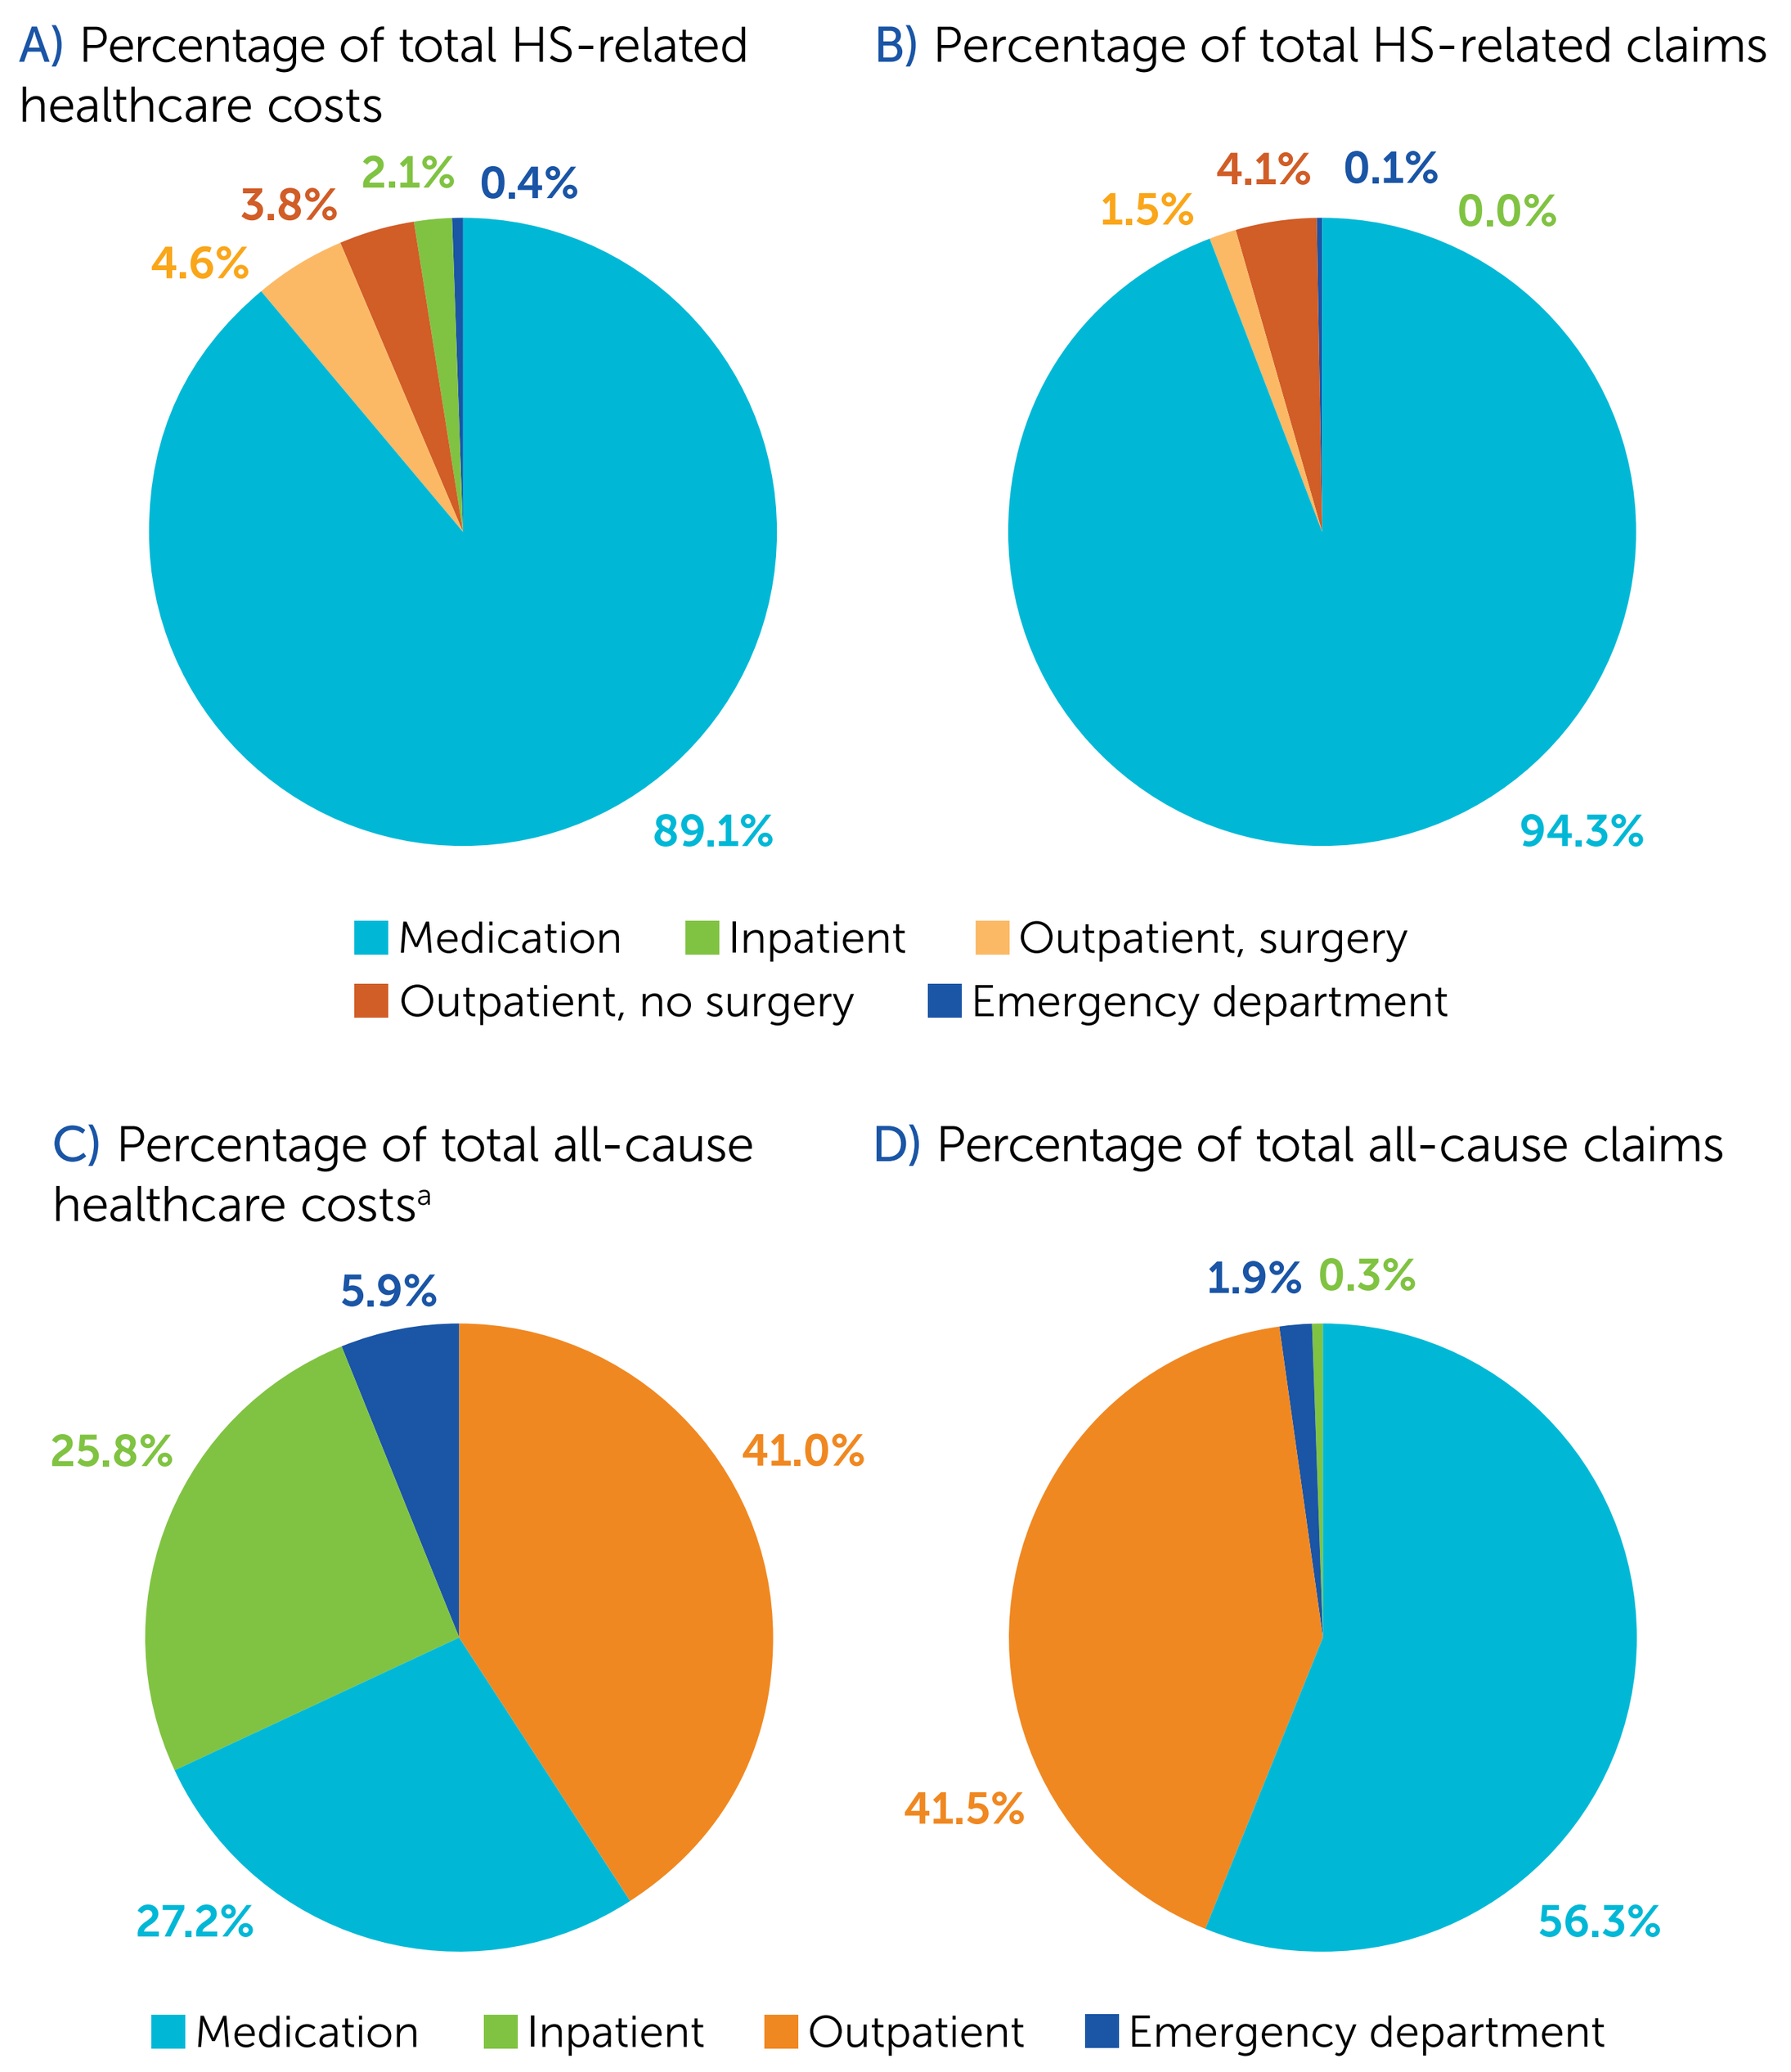
A) Percentage of total HS-related healthcare costs
4.6%
3.8%
2.1%
0.4%
89.1%
B) Percentage of total HS-related claims
1.5%
4.1%
0.1%
0.0%
94.3%
Medication
Inpatient
Outpatient, surgery
Outpatient, no surgery
Emergency department
C) Percentage of total all-cause healthcare costs[a]
5.9%
25.8%
41.0%
27.2%
D) Percentage of total all-cause claims
1.9%
0.3%
41.5%
56.3%
Medication
Inpatient
Outpatient
Emergency department

[a]Note that these values have been rounded, and so add to 99.9%.

HCRU: healthcare resource utilization; **HS:** hidradenitis suppurativa; **kPY:** rate per 1,000 patient-years; **RR:** rate ratio; **SD:** standard deviation.

Institutions: [1]UNC School of Medicine, Department of Dermatology, Chapel Hill, NC, USA; [2]Donald and Barbara Zucker School of Medicine at Hofstra/Northwell, Hempstead, NY, USA; [3]Data to Insights Research Solutions, Lisbon, Portugal; [4]UCB Pharma, Brussels, Belgium; [5]UCB Pharma, Smyrna, GA, USA; [6]Parasol Ltd, Warrington, UK; [7]Department of Dermatology, Penn State Milton S. Hershey Medical Center, Hershey, PA, USA.

References: [1]Ingram JR et al. J Eur Acad Dermatol Venereol. 2022;36(9):1597–605; [2]Kirby JS et al. JAMA Dermatol. 2014;150(9):937–44.

Author Disclosures: CS: Investigator for AbbVie, Chemocentryx, GSK, Incyte, InflaRx, Novartis, and UCB Pharma; consultancy fees from AbbVie, Alumis, Incyte, InflaRX, Logical Images, Sonoma Biotherapeutics, and UCB Pharma; speaker for AbbVie and Novartis. **AG:** Consultant for AbbVie, Aclaris Therapeutics, Anaptys Bio, Aristea Therapeutics, BMS, Boehringer Ingelheim, Incyte, Insmed, Janssen, Novartis, Pfizer, Sonoma Biotherapeutics, UCB Pharma, Union Therapeutics, Ventyx biosciences, and Viela Biosciences, and receives honoraria. Reported receiving research grants from AbbVie, UCB Pharma, National Psoriasis Foundation, and C3; co-copyright holder of the HS-IGA and HiSQoL instruments. **TT**, **SB:** Employees and shareholders of UCB Pharma. **JV**, **DF:** Independent contractors for UCB Pharma. **JK:** Reports personal fees from AbbVie, ChemoCentryx, CSL Behring, DermTech, Incyte, Insmed, Janssen, MoonLake, Novartis, and UCB Pharma; personal fees and grants from Incyte. Co-copyright holder of HiSQOL. **Acknowledgments:** This study was funded by UCB Pharma. The authors acknowledge Susanne Wiegratz, MSc, UCB Pharma, Monheim, Germany, for publication coordination, Lucy-Paige Willows, BSc, Costello Medical, Cambridge, UK, for medical writing, and editorial assistance, and the Costello Medical Creative team for design support. All costs associated with development of this poster were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit:
https://ucbposters.com/?PosterID=ISPOR2023_RWD181

Poster ID: ISPOR2023_RWD181
Link expiration: May 24th 2023