Incidence Rates of Infections and Association Between Lupus Nephritis and Serious Infections in Patients With Systemic Lupus Erythematosus: Systematic Literature Review and Meta-analyses

EPH90

Anubhav Nangia,[1] Evangelia Simou,[1] Shaun Abeysinghe,[1] Ann Colosia,[2] Meryem Bektas,[2] Jennifer Workman,[3] Shiyao Gao,[3] Natalia Bello[3]

[1] RTI Health Solutions, Manchester, United Kingdom; [2] RTI Health Solutions, Research Triangle Park, NC, United States; [3] Eli Lilly and Company, Indianapolis, IN, United States

BACKGROUND

- Patients with systemic lupus erythematosus (SLE) have a greater risk of infections compared with the general population and an increased susceptibility to life-threatening infections. Patients with SLE are more likely to die of infection than disease activity.[1]
- A dysregulated immune system and treatment with immunosuppressants contribute to the increased risk of infection.[1]
- Lupus nephritis (LN) is a more severe form of SLE that affects the kidneys and may require treatment with higher doses of glucocorticoids and more aggressive immunosuppressants such as mycophenolate mofetil, which can further exacerbate the risk of infections.[2]

OBJECTIVE

- The objective of this research was to perform meta-analyses (MAs) based on a systematic literature review (SLR) to quantify the incidence rates (IRs) of serious infections, fatal infections, herpes zoster (HZ), and tuberculosis (TB), as well as the association between LN and serious infections, in patients with SLE.

METHODS

- An SLR (conducted in Embase, MEDLINE, and MEDLINE In-Process) identified observational studies published between 1 January 2000 and 31 August 2020 that assessed the incidence or risk of any infections in patients with SLE, including those with LN.
- 2 researchers independently screened titles/abstracts followed by full texts against predefined eligibility criteria; disagreements over study relevance were resolved through consensus with a third researcher.
- Data were extracted from eligible full texts by 1 researcher and reviewed for accuracy against the source by an independent researcher. Quality assessments were performed by 1 researcher using the Critical Appraisal Skills Programme checklists.
- The metafor package in R was used for the MAs.[3] Primary analyses used random-effects (RE) models to derive pooled estimates of the IRs of serious infections, fatal infections, HZ, and TB in patients with SLE, as well as the unadjusted odds ratio (OR) of serious infections in patients with LN compared with those with nonrenal SLE.
- Between-study heterogeneity was assessed using Higgins I^2. Sensitivity analyses (SAs) using RE models were performed to validate the results through leave-one-out (LOO) analyses when minimal heterogeneity was observed ($I^2 < 40\%$) or to explore the sources of moderate-to-high heterogeneity ($I^2 > 40\%$) by excluding studies with potentially heterogenous populations.

RESULTS

- The SLR identified 86 studies that assessed any infection outcomes, of which 12 studies either recruited too few patients (< 100) to report a stable IR or reported outcomes in specific subgroups of patients that were not of interest (i.e., patients on peritoneal dialysis, or perinatal women). Therefore, data were extracted from 74 articles representing 73 studies in the SLR (Figure 1).
- Among the 25 studies included in the SLR that reported data on the outcomes of interest for the MAs, 22 studies were included in the MAs; 3 studies were excluded because they reported IRs of HZ[4-6] and/or TB[5] in patients with SLE without providing the raw data underlying the IRs, which were mandatory inputs in the statistical package used for the MAs.
- 18 studies were included in MAs of the IR of 1 or more of the following outcomes of interest in patients with SLE: serious infections (6 studies),[7-12] fatal infections (6 studies),[8,9,13-16] HZ (6 studies),[17-22] and/or TB (3 studies).[21,23,24] Additionally, 5 studies were included in the MA of the OR of serious infections in patients with LN compared with those with nonrenal SLE.[13,25-28]
- Based on the quality assessments performed in the SLR, most studies included in the MAs had a low risk of bias except 3 studies included in the MA of the IR of HZ, of which 2 studies had a medium risk of bias[17,22] and 1 study had an unclear risk of bias.[19]
- Figures 2-6 present the forest plots for the primary MA of each endpoint. For each endpoint, Table 1 shows the results of the primary MA, describes the SAs performed with corresponding results, and provides a rationale for each SA.
 - Pooled estimates from the primary MAs for the IRs of serious infections, fatal infections, HZ, and TB in patients with SLE were characterized by high heterogeneity ($I^2 > 70\%$).
 - Results of the SAs performed for the IRs of serious infections and HZ suggested that differences in background immunosuppressant use and geographic regions could not explain the high heterogeneity observed in the corresponding primary MAs, respectively.
 - Results of the SA performed for the IR of fatal infections suggested that the sources of high heterogeneity observed in the primary MA were 2 studies that recruited either patients with juvenile-onset SLE only or a healthier population relative to other studies that informed the endpoint; their exclusion reduced the I^2 to 0%. However, this SA should be interpreted cautiously because it was informed by only 4 studies.
 - Results of the SA performed for the IR of TB suggested that the high heterogeneity observed in the primary MA was due to 1 study; its exclusion reduced the I^2 to 31.72%, though a plausible explanation for this could not be identified based on an evaluation of the characteristics of this study relative to other studies that informed the endpoint. This SA should also be interpreted cautiously because it was informed by 2 studies only.
 - The pooled estimate from the primary MA for the OR of serious infections in patients with LN compared with patients with nonrenal SLE was characterized by low heterogeneity ($I^2 = 23.77\%$). Results from the leave-one-out SA validated the results of the primary MA, which suggested that patients with LN have statistically significantly higher odds of developing serious infections compared with patients with nonrenal SLE.

Figure 1. PRISMA Chart of Studies Included at Each Stage of the SLR

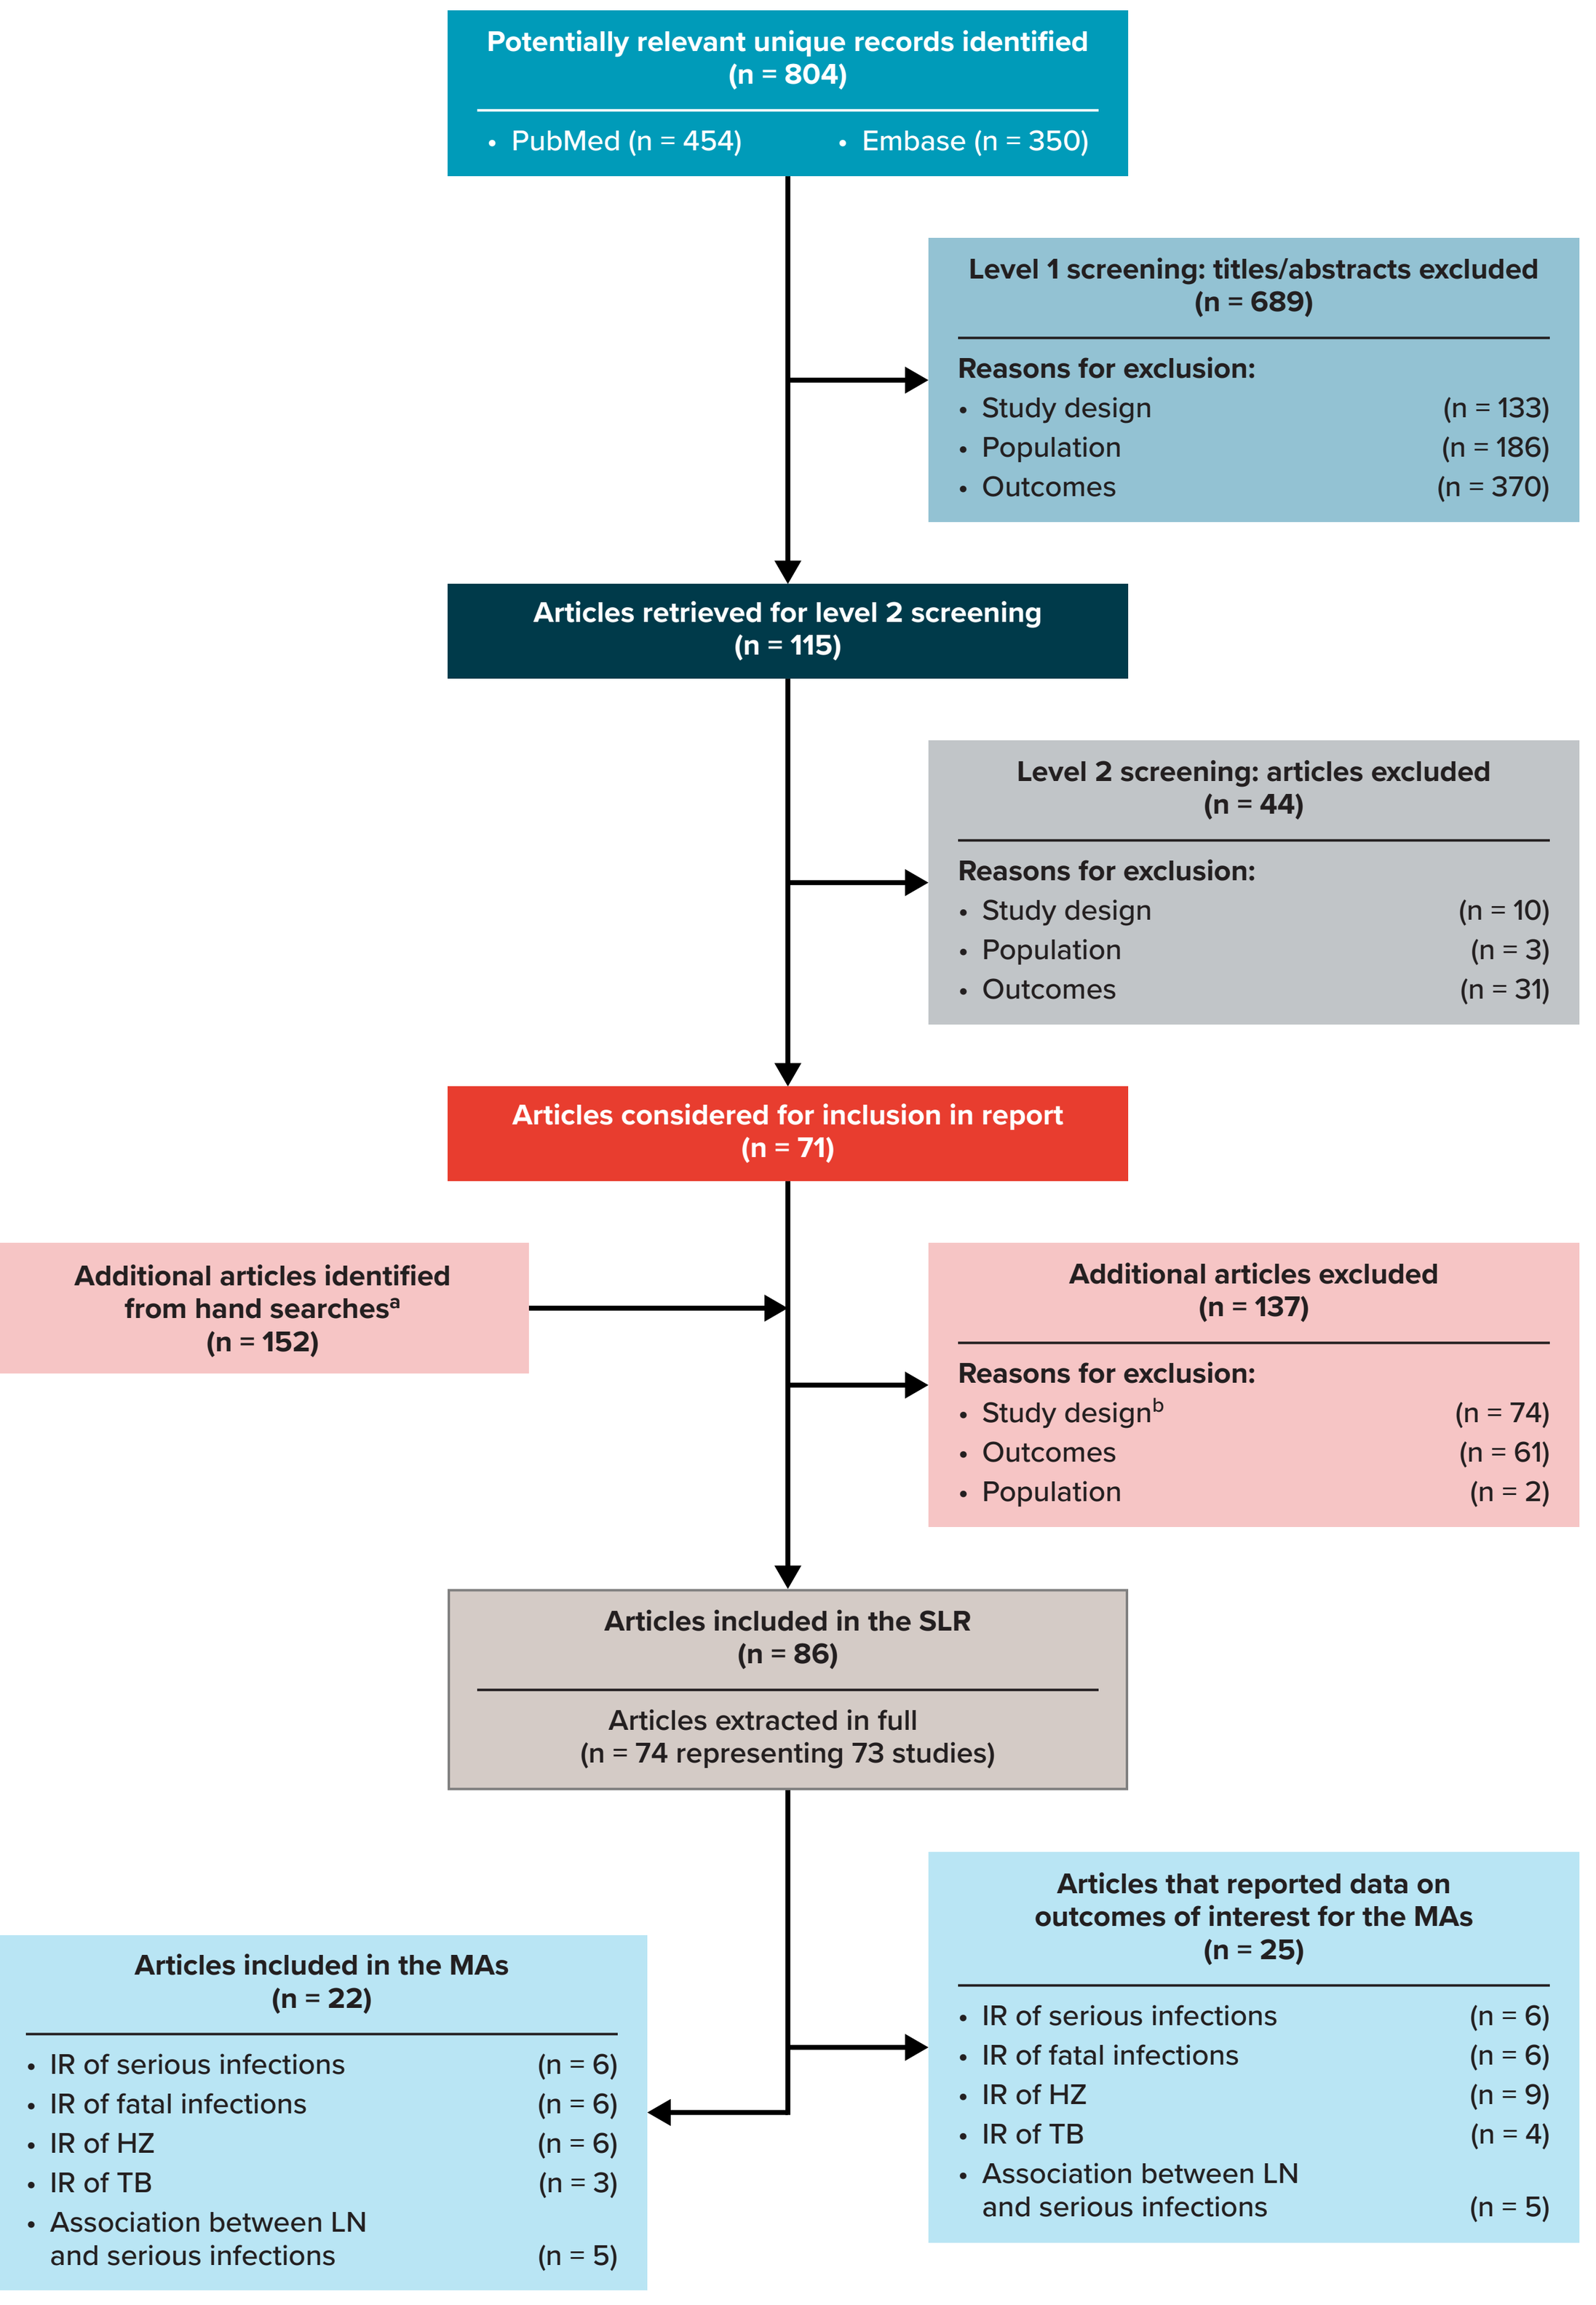
Potentially relevant unique records identified
(n = 804)
• PubMed (n = 454) • Embase (n = 350)
Level 1 screening: titles/abstracts excluded
(n = 689)
Reasons for exclusion:
• Study design (n = 133)
• Population (n = 186)
• Outcomes (n = 370)
Articles retrieved for level 2 screening
(n = 115)
Level 2 screening: articles excluded
(n = 44)
Reasons for exclusion:
• Study design (n = 10)
• Population (n = 3)
• Outcomes (n = 31)
Articles considered for inclusion in report
(n = 71)
Additional articles identified from hand searches[a]
(n = 152)
Additional articles excluded
(n = 137)
Reasons for exclusion:
• Study design[b] (n = 74)
• Outcomes (n = 61)
• Population (n = 2)
Articles included in the SLR
(n = 86)
Articles extracted in full
(n = 74 representing 73 studies)
Articles that reported data on outcomes of interest for the MAs
(n = 25)
• IR of serious infections (n = 6)
• IR of fatal infections (n = 6)
• IR of HZ (n = 9)
• IR of TB (n = 4)
• Association between LN and serious infections (n = 5)
Articles included in the MAs
(n = 22)
• IR of serious infections (n = 6)
• IR of fatal infections (n = 6)
• IR of HZ (n = 6)
• IR of TB (n = 3)
• Association between LN and serious infections (n = 5)

[a] Reference lists of relevant SLRs, MAs, and health technology assessments identified in the review were hand-searched for further studies of interest.

[b] Includes duplicates of studies identified in the database records.

Figure 2. Forest Plot of the Primary MA of the IR of Serious Infections in Patients With SLE

Study	Person-years	IR/100 PY	95% CI	Weight (RE)
Pimentel–Quiroz et al.[7]	4,447.37	3.80	[3.25-4.40]	15.1%
Lertchaisataporn et al.[8]	217.00	12.44	[8.15-17.62]	12.6%
Terrier et al.[9]	181.00	6.63	[3.34-10.99]	12.1%
Feldman et al.[10]	3,438.40	8.99	[8.01-10.02]	15.1%
Herrinton et al.[11] Current Drug Use	1,526.00	2.56	[1.81-3.43]	14.8%
Herrinton et al.[11] Nonusers	2,090.00	2.49	[1.85-3.21]	15.0%
Rúa–Figueroa et al.[12]	24,143.84	2.92	[2.71-3.14]	15.3%
Random-effects model		**4.99**	**[2.78-7.82]**	**100.0%**

Heterogeneity: $I^2 = 98\%$, $\tau^2 = 0.0053$, $p < 0.01$ — IR of serious infections per 100 PY

Note: 7 data points from 6 studies were included. One study reported data separately for the first phase of drug use and the first phase of no drug use among patients, corresponding to the data points labeled "Herrinton et al.[11] Current Drug Use" and "Herrinton et al.[11] Nonusers," respectively, in the analysis. These data were not combined because patients contributed to both phases and could have had an infection in each—it is only the first infection experienced by a patient in a specified period that is of interest for an MA of IRs.

PY = person-year.

Figure 3. Forest Plot of the Primary MA of the IR of Fatal Infections in Patients With SLE

Study	Person-years	IR/100 PY	95% CI	Weight (RE)
Bosch et al.[13]	192.50	1.56	[0.19-3.95]	13.9%
Fernández–Nebro et al.[14]	213.46	0.94	[0.01-2.82]	14.4%
Lertchaisataporn et al.[8]	259.21	1.16	[0.14-2.93]	15.3%
Terrier et al.[9]	210.80	1.42	[0.17-3.61]	14.3%
Torrente–Segarra et al.[15]	5,543.00	0.07	[0.02-0.16]	21.3%
Wallace et al.[16]	2,416.00	0.04	[0.00-0.18]	20.8%
Random-effects model		**0.51**	**[0.03-1.41]**	**100.0%**

Heterogeneity: $I^2 = 82\%$, $\tau^2 = 0.0023$, $p < 0.01$ — IR of fatal infections per 100 PY

Figure 4. Forest Plot of the Primary MA of the IR of HZ in Patients With SLE

Study	Person-years	IR/100 PY	95% CI	Weight (RE)
Borba et al.[17]	7,968.75	0.64	[0.48-0.83]	17.2%
Chakravarty et al.[18]	25,246.91	1.62	[1.47-1.78]	17.3%
Chen et al.[4]	1,000.00	3.77	[2.65-5.08]	0.0%
Chen et al.[19]	233,096.00	1.52	[1.47-1.57]	17.4%
Hata et al.[20]	707.64	5.37	[3.79-7.22]	15.2%
Hsu et al.[21]	89,256.00	2.89	[2.78-3.00]	17.4%
Imafuku et al.[22]	943.00	1.59	[0.87-2.51]	15.7%
Skare et al.[5]	100.00	0.27	[0.00-2.77]	0.0%
Yun et al.[6]	1,000.00	2.01	[1.22-3.00]	0.0%
Random-effects model		**1.94**	**[0.96-3.24]**	**100.0%**

Heterogeneity: $I^2 = 99\%$, $\tau^2 = 0.0024$, $p < 0.01$ — IR of HZ per 100 PY

Note: Chen et al.,[4] Skare et al.,[5] and Yun et al.[6] reported IRs without providing the raw data underlying the IRs, which were mandatory inputs in the statistical package used for the MA. Therefore, their IRs are included in the forest plot for completeness, but these studies did not contribute to the pooled estimates.

Figure 5. Forest Plot of the Primary MA of the IR of TB in Patients With SLE

Study	Person-years	IR/100 PY	95% CI	Weight (RE)
Erdozain et al.[23]	1,603.00	0.19	[0.02-0.47]	28.2%
Yun et al.[24]	1,898.73	0.79	[0.43-1.25]	29.7%
Hsu et al.[21]	89,256.00	0.38	[0.34-0.42]	42.1%
Skare et al.[5]	100.00	0.05	[0.00-2.10]	0.0%
Random-effects model		**0.31**	**[0.08-0.65]**	**100.0%**

Heterogeneity: $I^2 = 74\%$, $\tau^2 = 0.0003$, $p < 0.02$ — IR of TB per 100 PY

Note: Skare et al.[5] reported an IR without providing the raw data underlying the IR, which were mandatory inputs in the statistical package used for the MA. Therefore, the IR from this study is included in the forest plot for completeness, but it did not contribute to the pooled estimate.

Figure 6. Forest Plot of the Primary MA of the OR of Serious Infections in Patients With SLE and LN Compared With Patients With Nonrenal SLE

Study	Weight (RE)	95% CI
Bosch et al.[13]	0.5%	7.25 [2.83-18.56]
Feldman et al.[25]	96.7%	2.47 [2.32-2.63]
González–Echavarri et al.[26] 1Y	0.4%	3.02 [1.16-7.90]
González–Echavarri et al.[26] 2Y	0.2%	2.33 [0.62-8.74]
Jung et al.[27]	1.6%	1.97 [1.19-3.27]
Ruiz–Irastorza et al.[28]	0.6%	3.44 [1.47-8.05]
Random-effects model		**2.48 [2.33-2.64]**

Heterogeneity: $I^2 = 24\%$, $\tau^2 = 0.00$, $p < 0.26$ — OR of serious infections

Note: 6 data points from 5 studies were included. One study reported data separately for the first and second year of follow-up, corresponding to the data points labeled "González-Echavarri et al.[26] 1Y" and "González-Echavarri et al.[26] 2Y," respectively, in the analysis. These data were not combined, in order to avoid double-counting patients (a patient could have had an infection in the first and second year of follow-up separately). Therefore, LOO SAs performed for this endpoint that included both data points from the same study were informed by 5 data points from 4 studies.

Table 1. Primary and Sensitivity Analysis Performed for Each Endpoint

Endpoint (no. of data points included)	Primary analysis result: pooled estimate (95% CI) and Higgins I^2	SA description	SA result: pooled estimate (95% CI) and Higgins I^2	SA rationale
IR of serious infections in patients with SLE (n = 7)[a]	• 4.99 (2.78-7.82) per 100 PYs • I^2 = 97.52%	Subgroup of studies in which all patients received IMSPs (n = 3)[9-11]	• 5.66 (2.26-10.55) per 100 PYs • I^2 = 97.56%	Potential heterogeneity due to differences in background IMSP use because IMSPs are known to increase the risk of infections[29]
		Subgroup of studies in which only some patients used IMSPs (n = 3)[7,8,12]	• 5.50 (1.53-11.87) per 100 PYs • I^2 = 94.62%	
IR of fatal infections in patients with SLE (n = 6)	• 0.51 (0.03-1.41) per 100 PYs • I^2 = 82.28%	Exclusion of 2 studies (n = 4)[8,9,13,14]	• 1.24 (0.56-2.15) per 100 PYs • I^2 = 0%	Potential heterogeneity due to 2 studies that recruited either patients with juvenile-onset SLE only or a healthier population relative to other studies[15,16]
IR of HZ in patients with SLE (n = 6)	• 1.94 (0.96-3.24) per 100 PYs • I^2 = 99.28%	Subgroup of studies conducted in Asia (n = 3)[20-22]	• 3.07 (1.37-5.42) per 100 PYs • I^2 = 88.84%	Potential heterogeneity due to differences in geographic region because there is an increased risk of HZ and relatively lower use of AMs, which are known to be protective against infections, in Asian populations relative to populations from the Americas[30,31]
		Subgroup of studies conducted in the Americas (n = 3)[17-19]	• 1.22 (0.66-1.95) per 100 PYs • I^2 = 96.63%	
IR of TB in patients with SLE (n = 3)	• 0.31 (0.08-0.65) per 100 PYs • I^2 = 73.74%	LOO analysis scenario 1 (n = 2)[21,24]	• 0.53 (0.20-1.01) per 100 PYs • I^2 = 83.43%	Primary analysis was informed by 3 studies only, and at least 2 studies are needed for an MA; was deemed reasonable to explore the impact of the exclusion of each study
		LOO analysis scenario 2 (n = 2)[21,23]	• 0.34 (0.21-0.48) per 100 PYs • I^2 = 31.72%	
		LOO analysis scenario 3 (n = 2)[23,24]	• 0.44 (0.04-1.22) per 100 PYs • I^2 = 84.96%	
OR of serious infections in patients with LN compared with patients with nonrenal SLE (n = 6)[b]	• 2.48 (2.33-2.64) • I^2 = 23.77%	LOO analysis scenario 1 (n = 5)[25-28;c]	• 2.47 (2.32-2.63) • I^2 = 0%	Validate the results of the primary analysis where minimal heterogeneity was observed (I^2 < 40%)
		LOO analysis scenario 2 (n = 5)[13,26-28;c]	• 3.05 (1.88-4.95) • I^2 = 34.56%	
		LOO analysis scenario 3 (n = 5)[13,25,27,28]	• 2.48 (2.32-2.64) • I^2 = 37.45%	
		LOO analysis scenario 4 (n = 5)[13,25,27,28]	• 2.48 (2.33-2.64) • I^2 = 38.93%	
		LOO analysis scenario 5 (n = 5)[13,25,26,28;c]	• 3.03 (2.07-4.43) • I^2 = 30.49%	
		LOO analysis scenario 6 (n = 5)[13,25-27;c]	• 2.48 (2.32-2.64) • I^2 = 33.16%	

AM = antimalarial; CI = confidence interval; IMSP = immunosuppressant.

[a] 7 data points from 6 studies were included in this analysis (see footnote of Figure 2 for an explanation).

[b] 6 data points from 5 studies were included in this analysis (see footnote of Figure 6 for explanation).

[c] 5 data points from 4 studies were included in this analysis (see footnote of Figure 6 for explanation).

DISCUSSION

- The pooled IRs from the primary MAs of serious infections, fatal infections, and HZ in patients with SLE were higher than the IRs of these infections in the general population or in suitable controls (i.e., patients without SLE) where these were reported in our evidence base.[4,6,11,13,18,22] Although the primary MAs were characterized by high heterogeneity, these observations align with published literature that suggests that patients with SLE have an increased susceptibility to infections.[1]
- No study in our evidence base reported the IR of TB in the general population or in suitable controls (i.e., patients without SLE). However, a recently published MA concluded that patients with SLE have a significantly higher risk of developing TB than the general population or healthy controls, though there was high heterogeneity in that MA.[32]
- Our finding that patients with LN have 2.48 times higher odds of developing serious infections compared with patients with nonrenal SLE is aligned with the results of a recently published MA, which suggested that renal involvement is a significant risk factor for infections in patients with SLE, though there was high heterogeneity in that MA.[33]

CONCLUSIONS

- Patients with LN have statistically significantly higher odds of developing serious infections relative to patients with nonrenal SLE.
- Pooled estimates of the IRs of serious infections, fatal infections, HZ, and TB in patients with SLE should be interpreted cautiously because high heterogeneity was observed across these endpoints.

REFERENCES

1. Ioannou Y, et al. Postgrad Med J. 2002;78(924):599-606.
2. Mageau A, et al. Autoimmun Rev. 2019;18(7):733-7.
3. Viechtbauer W. J Stat Softw. 2010;36(3):1-48.
4. Chen HH, et al. Clinics (Sao Paulo). 2011;66(7):1177-82.
5. Skare TL, et al. Einstein (Sao Paulo). 2016 Jan-Mar;14(1):47-51.
6. Yun H, et al. Arthritis Rheumatol. 2016 Sep;68(9):2328-37.
7. Pimentel-Quiroz VR, et al. Lupus. 2019;28(9):1101-10.
8. Lertchaisataporn K, et al. J Clin Rheumatol. 2013;19(3):115-20.
9. Terrier B, et al. Arthritis Rheum. 2010;62(8):2458-66.
10. Feldman CH, et al. Arthritis Rheumatol. 2017;69(2):387-97.
11. Herrinton LJ, et al. J Rheumatol. 2016;43(8):1503-9.
12. Rúa-Figueroa Í, et al. Semin Arthritis Rheum. 2017;47(1):38-45.
13. Bosch X, et al. Lupus. 2006;15(9):584-9.
14. Fernández-Nebro A, et al. Lupus. 2012;21(10):1063-76.
15. Torrente-Segarra V, et al. Semin Arthritis Rheum. 2020;50(4):657-62.
16. Wallace DJ, et al. Arthritis Rheumatol. 2019;71(7):1125-34.
17. Borba EF, et al. J Clin Rheumatol. 2010;16(3):119-22.
18. Chakravarty EF, et al. Lupus. 2013;22(3):238-44.
19. Chen SY, et al. Infection. 2014;42(2):325-34.
20. Hata A, et al. Infection. 2011;39(6):537-44.
21. Hsu CY, et al. Arthritis Res Ther. 2019;21(1):211.
22. Imafuku S, et al. Dermatol Ther. 2019;9(1):117-33.
23. Erdozain JG, et al. Lupus. 2006;15(4):232-5.
24. Yun JE, et al. Clin Exp Rheumatol. 2002;20(2):127-32.
25. Feldman CH, et al. Arthritis Rheumatol. 2015;67(6):1577-85.
26. González-Echavarri C, et al. Lupus. 2018;27(14):2253-61.
27. Jung JY, et al. Sci Rep. 2019;9(1):9704.
28. Ruiz-Irastorza G, et al. Arthritis Res Ther. 2009;11(4):R109.
29. Suzuki K, et al. Indian J Rheumatol. 2022;10.4103/injr.injr_78_22.
30. Curran D, et al. Infect Dis Ther. 2022;11(1):389-403.
31. Miyazaki C, et al. J Med Econ. 2020;23(7):786-99.
32. Pego-Reigosa JM, et al. Rheumatology (Oxford). 2021;60(1):60-72.
33. Yuan Q, et al. Semin Arthritis Rheum. 2020;50(5):1022-39.

CONTACT INFORMATION

Anubhav Nangia, MSc
Senior Research Statistician

RTI Health Solutions

2nd Floor, The Pavilion
Towers Business Park
Wilmslow Road
Didsbury
United Kingdom

Phone: +44(0)161.446.1734
Email: anangia@rti.org

The power of **knowledge.** The value of **understanding.**

Presented at: ISPOR 2023; 7-10 May 2023; Boston, Massachusetts, United States