ASSESSING THE PUBLIC HEALTH IMPACT OF THE ADJUVANTED RESPIRATORY SYNCYTIAL VIRUS PREFUSION F PROTEIN VACCINE (RSVPreF3 OA) AMONG OLDER ADULTS IN THE UNITED STATES (US)

Molnar D[1], La E[2], Verelst F[1], Curran D[1], Poston S[2], Van Bellinghen L-A[3], Graham J[4]

[1] GSK, Wavre, Belgium; [2] GSK, Philadelphia, PA, USA; [3] CHESS in Health, Bonheiden, Belgium; [4] RTI Health Solutions, Research Triangle Park, NC, USA

Audio File

SCAN ME

EPH45

INTRODUCTION

Respiratory syncytial virus (RSV) is a **contagious pathogen** that causes acute respiratory illness (ARI) in individuals of all ages.[1]

RSV infection occurs annually in 3-7% of healthy older adults (OA) and in 4-10% adults who are at increased risk of severe RSV.[2]

This study estimates the **public health impact of vaccinating US adults aged ≥60 years** with a single dose of adjuvanted RSVPreF3 OA vaccine.

METHODS

Static Markov cohort model

- Evaluated the impact of adjuvanted RSVPreF3 OA vaccine on symptomatic RSV cases, morbidity, and mortality, compared with no vaccination among US adults aged ≥60 years.
- Used a 1-year time horizon (assuming no reinfections), with a 1-month cycle length.
- Classified RSV ARI cases as upper or lower respiratory tract disease cases (URTD or LRTD, respectively).

Inputs

- Population estimates, RSV epidemiology, and health care resource use inputs were obtained from standard US sources and published literature (see Supplement).
- Vaccination coverage rates were assumed to be the same as for influenza vaccines in the 2021-2022 season (60-64-year-olds: 52.4%; ≥65-year-olds: 73.9%).

Schematic structure of the Markov model

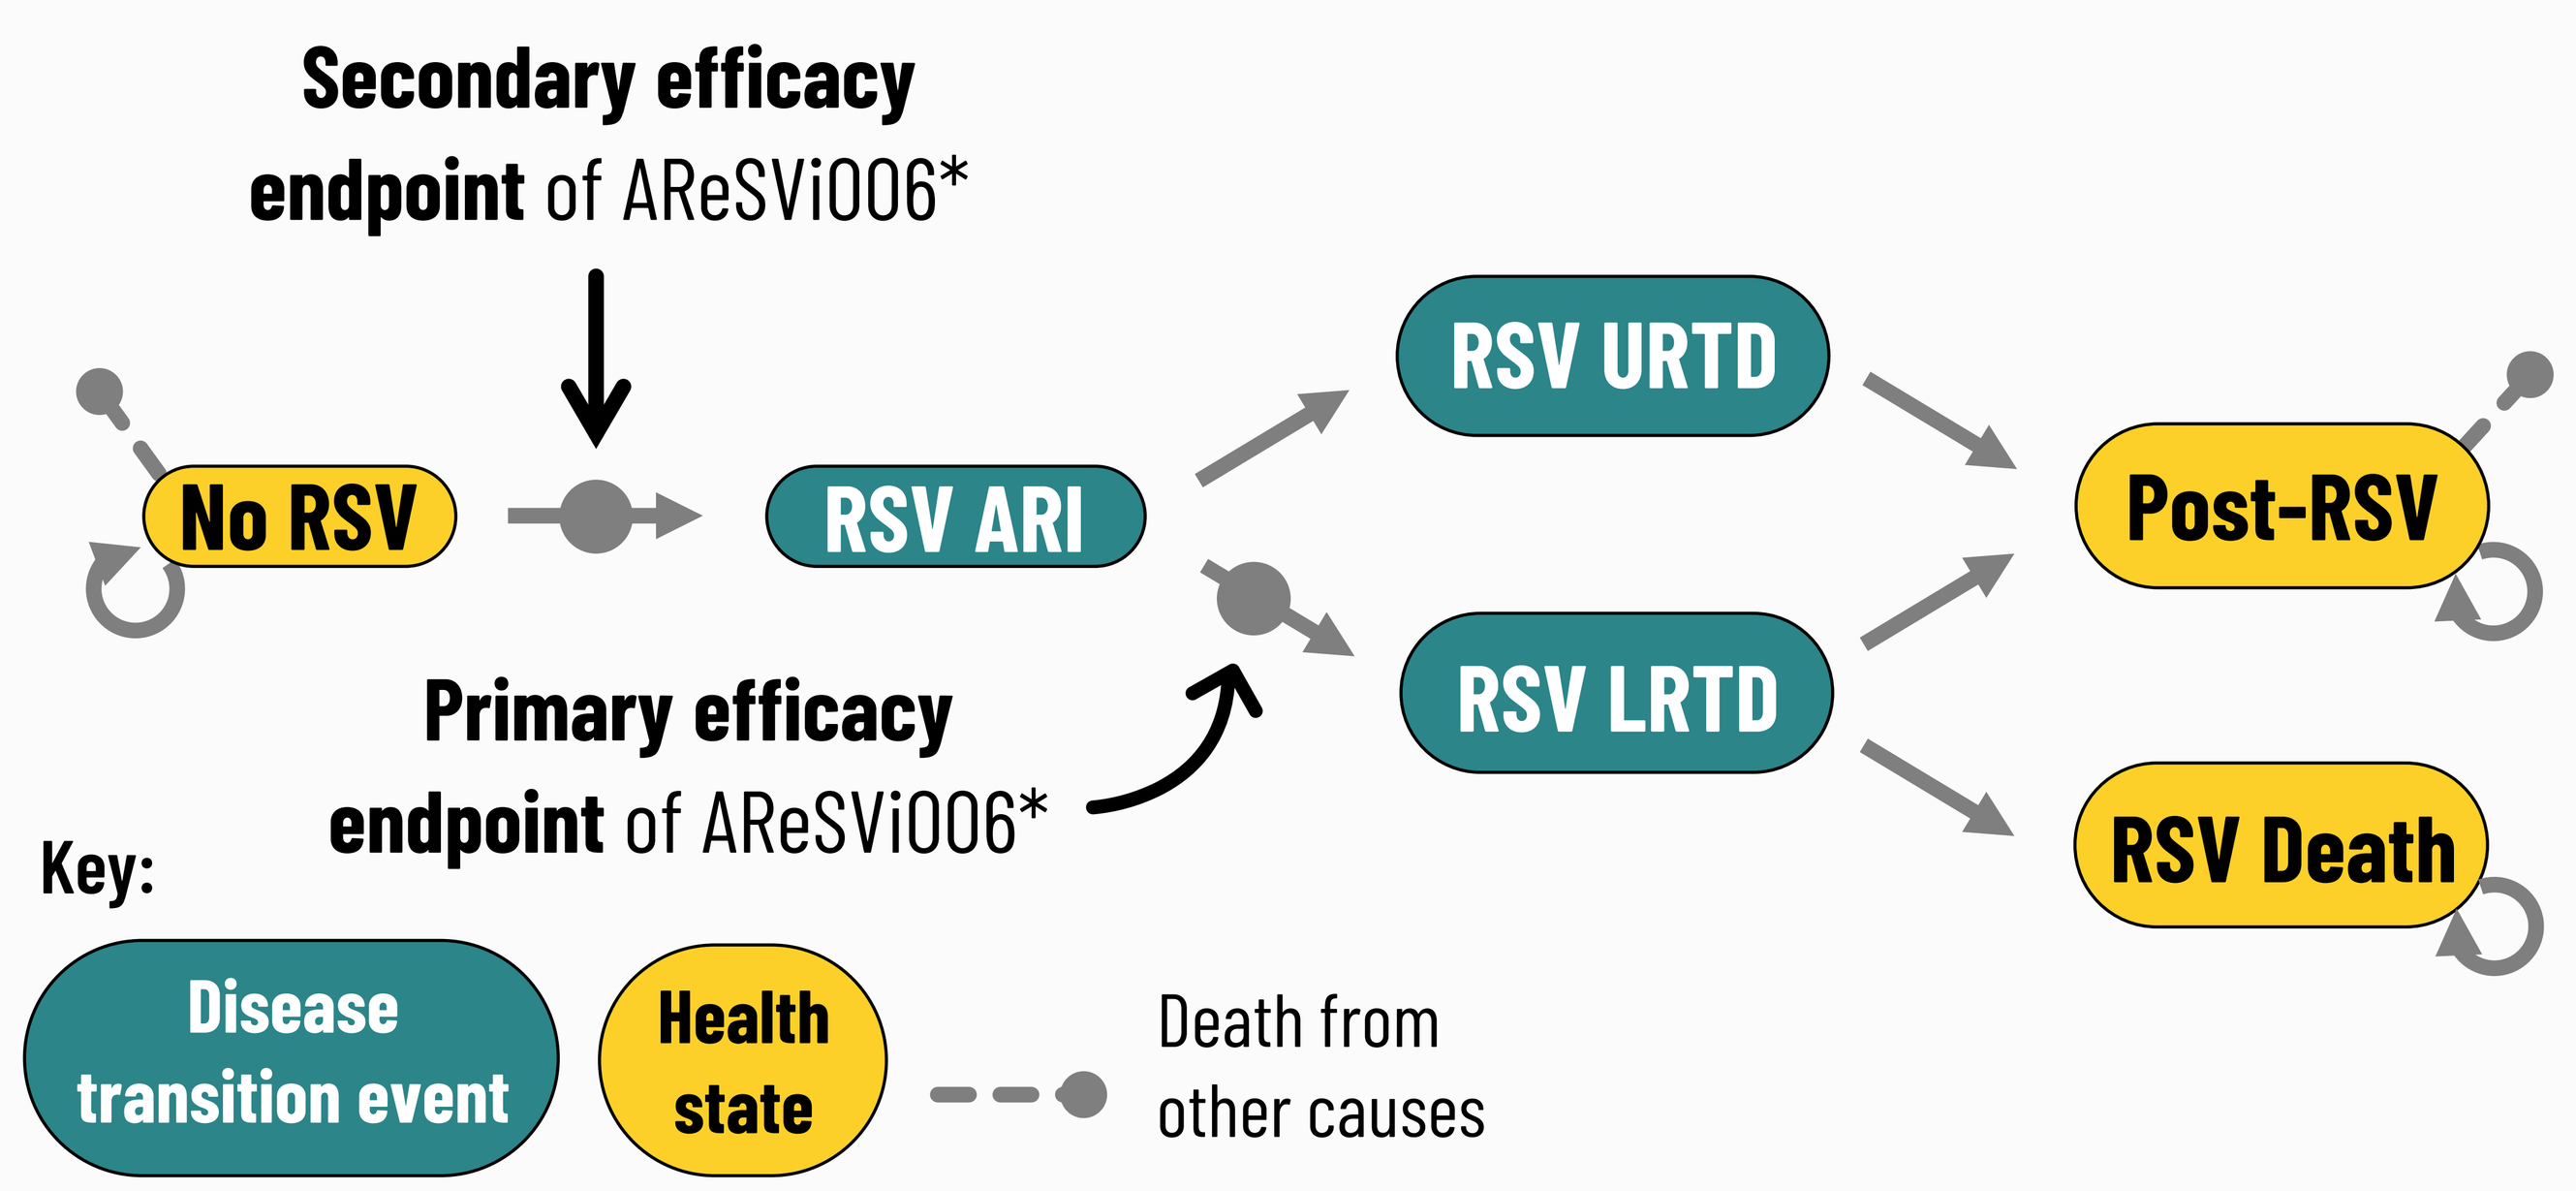
Secondary efficacy endpoint of AReSVi006*
No RSV
RSV ARI
RSV URTD
RSV LRTD
Post-RSV
RSV Death
Primary efficacy endpoint of AReSVi006*
Key:
Disease transition event
Health state
Death from other causes

*AReSVi006 (Adult Respiratory Syncytial Virus) phase III clinical trial[3]

Model assumes 50% of peak vaccine efficacy during month 1.

The benefits of RSV vaccination considered in the model include reductions in the probability of developing RSV ARI and RSV LRTD.

Vaccine efficacy and waning by month

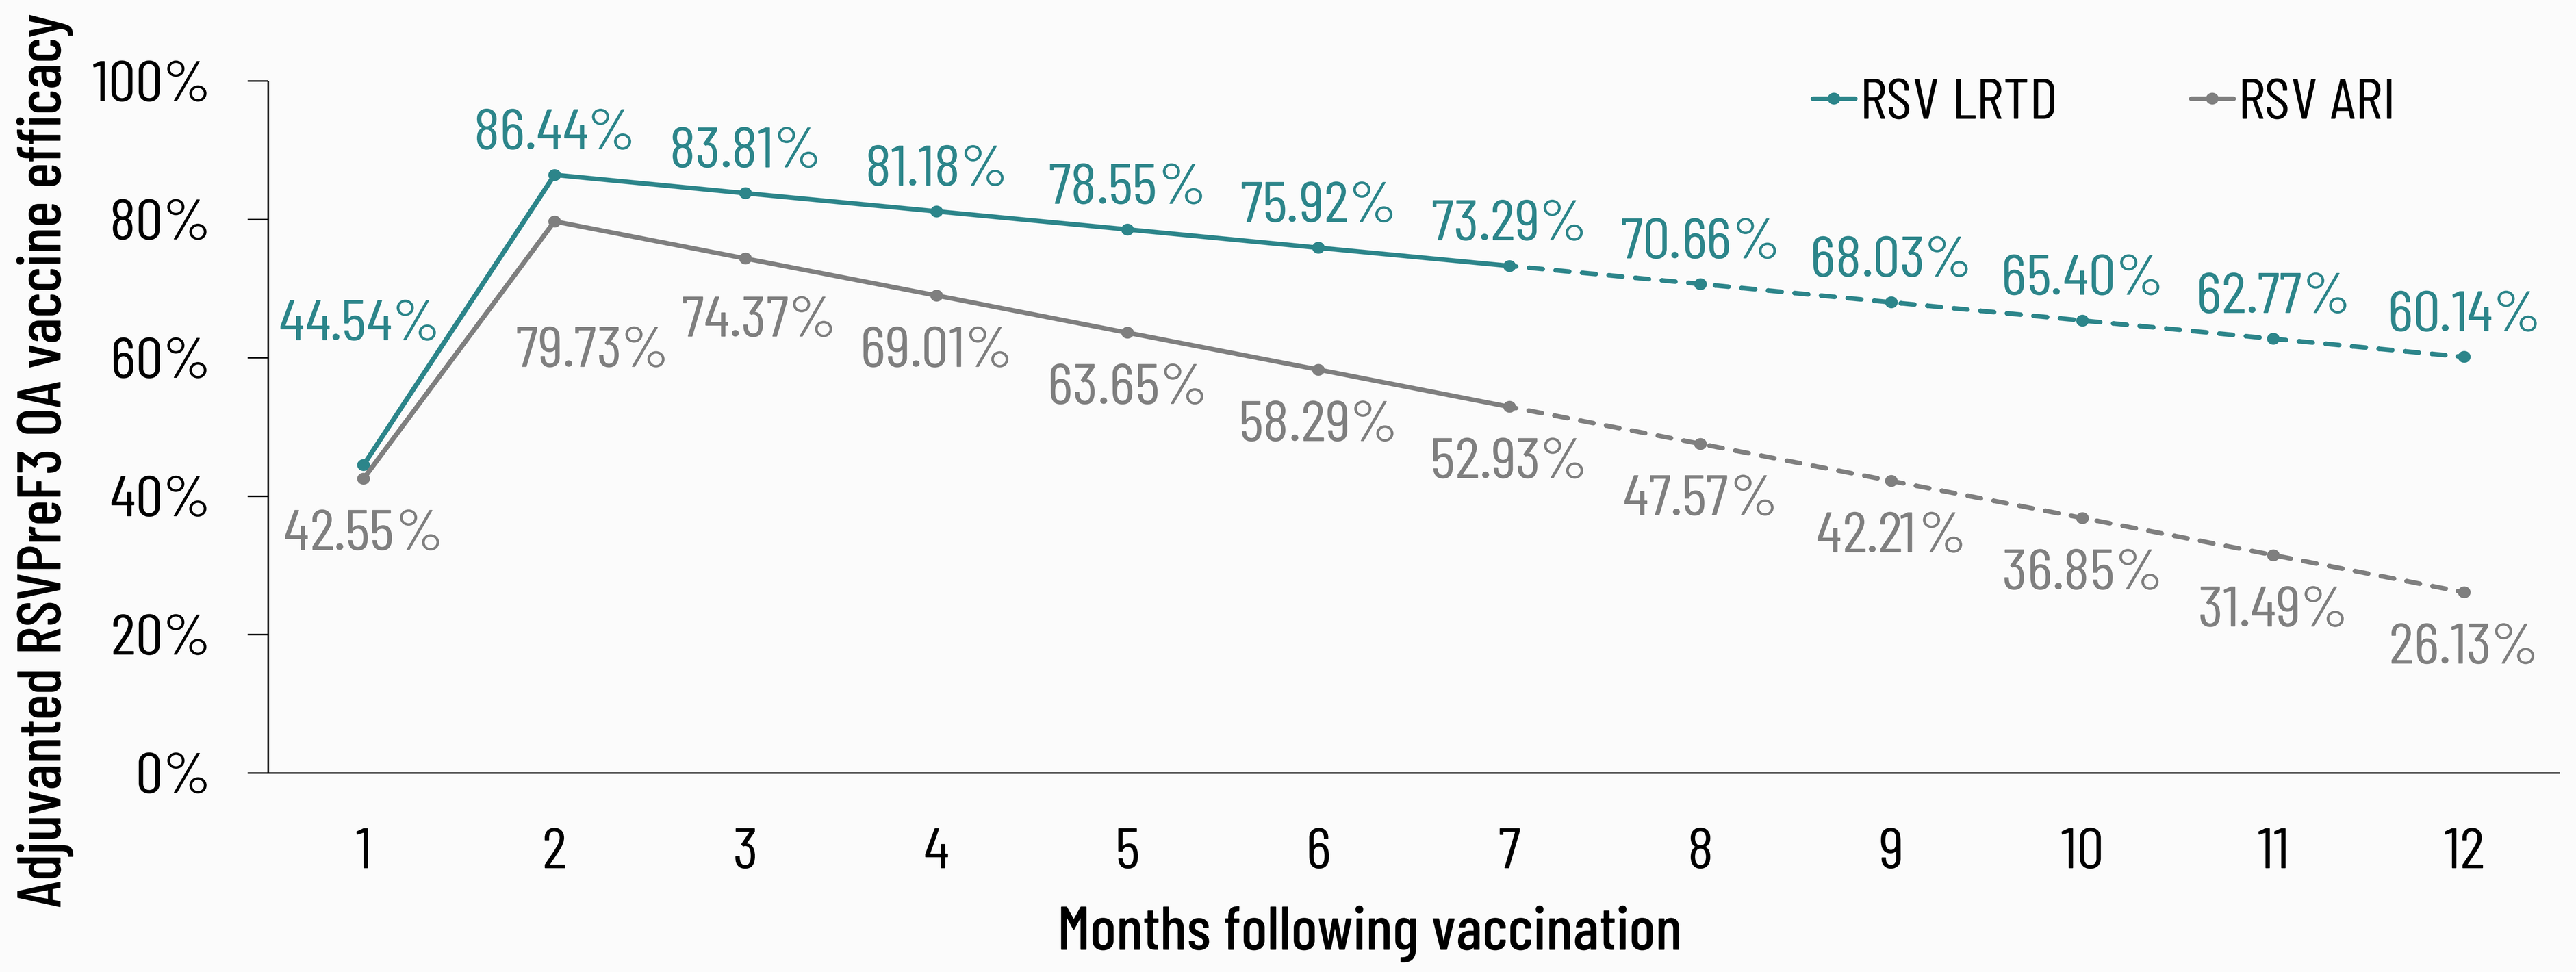
Adjuvanted RSVPreF3 OA vaccine efficacy
100%
80%
60%
40%
20%
0%
RSV LRTD
RSV ARI
44.54%
86.44%
83.81%
81.18%
78.55%
75.92%
73.29%
70.66%
68.03%
65.40%
62.77%
60.14%
42.55%
79.73%
74.37%
69.01%
63.65%
58.29%
52.93%
47.57%
42.21%
36.85%
31.49%
26.13%
1 2 3 4 5 6 7 8 9 10 11 12
Months following vaccination

Linear regression model fitted based on the AReSVi006 phase III clinical trial (median follow-up time: 6.7 months) to **estimate peak vaccine efficacy** (intercept) and **monthly waning** rate (slope) for RSV ARI and RSV LRTD.[3]

- **Peak efficacy**
 - RSV LRTD: 89.07%
 - RSV ARI: 85.09%
- **Monthly waning**
 - RSV LRTD: 2.63%
 - RSV ARI: 5.36%

Dashed lines represent extrapolation beyond the median trial follow-up period of the first season in the AReSVi006 phase III clinical trial.

Supplementary details on methods available through QR-code scanning

RESULTS

The model estimates that, compared to no vaccination, adjuvanted RSVPreF3 OA vaccine would substantially reduce the burden of RSV among US adults aged ≥60 years by preventing RSV cases and their associated health care resource use, antibiotic use, complications, and deaths.

A total of 55,282,554 individuals received the RSVPreF3 OA vaccine in the scenario with vaccination

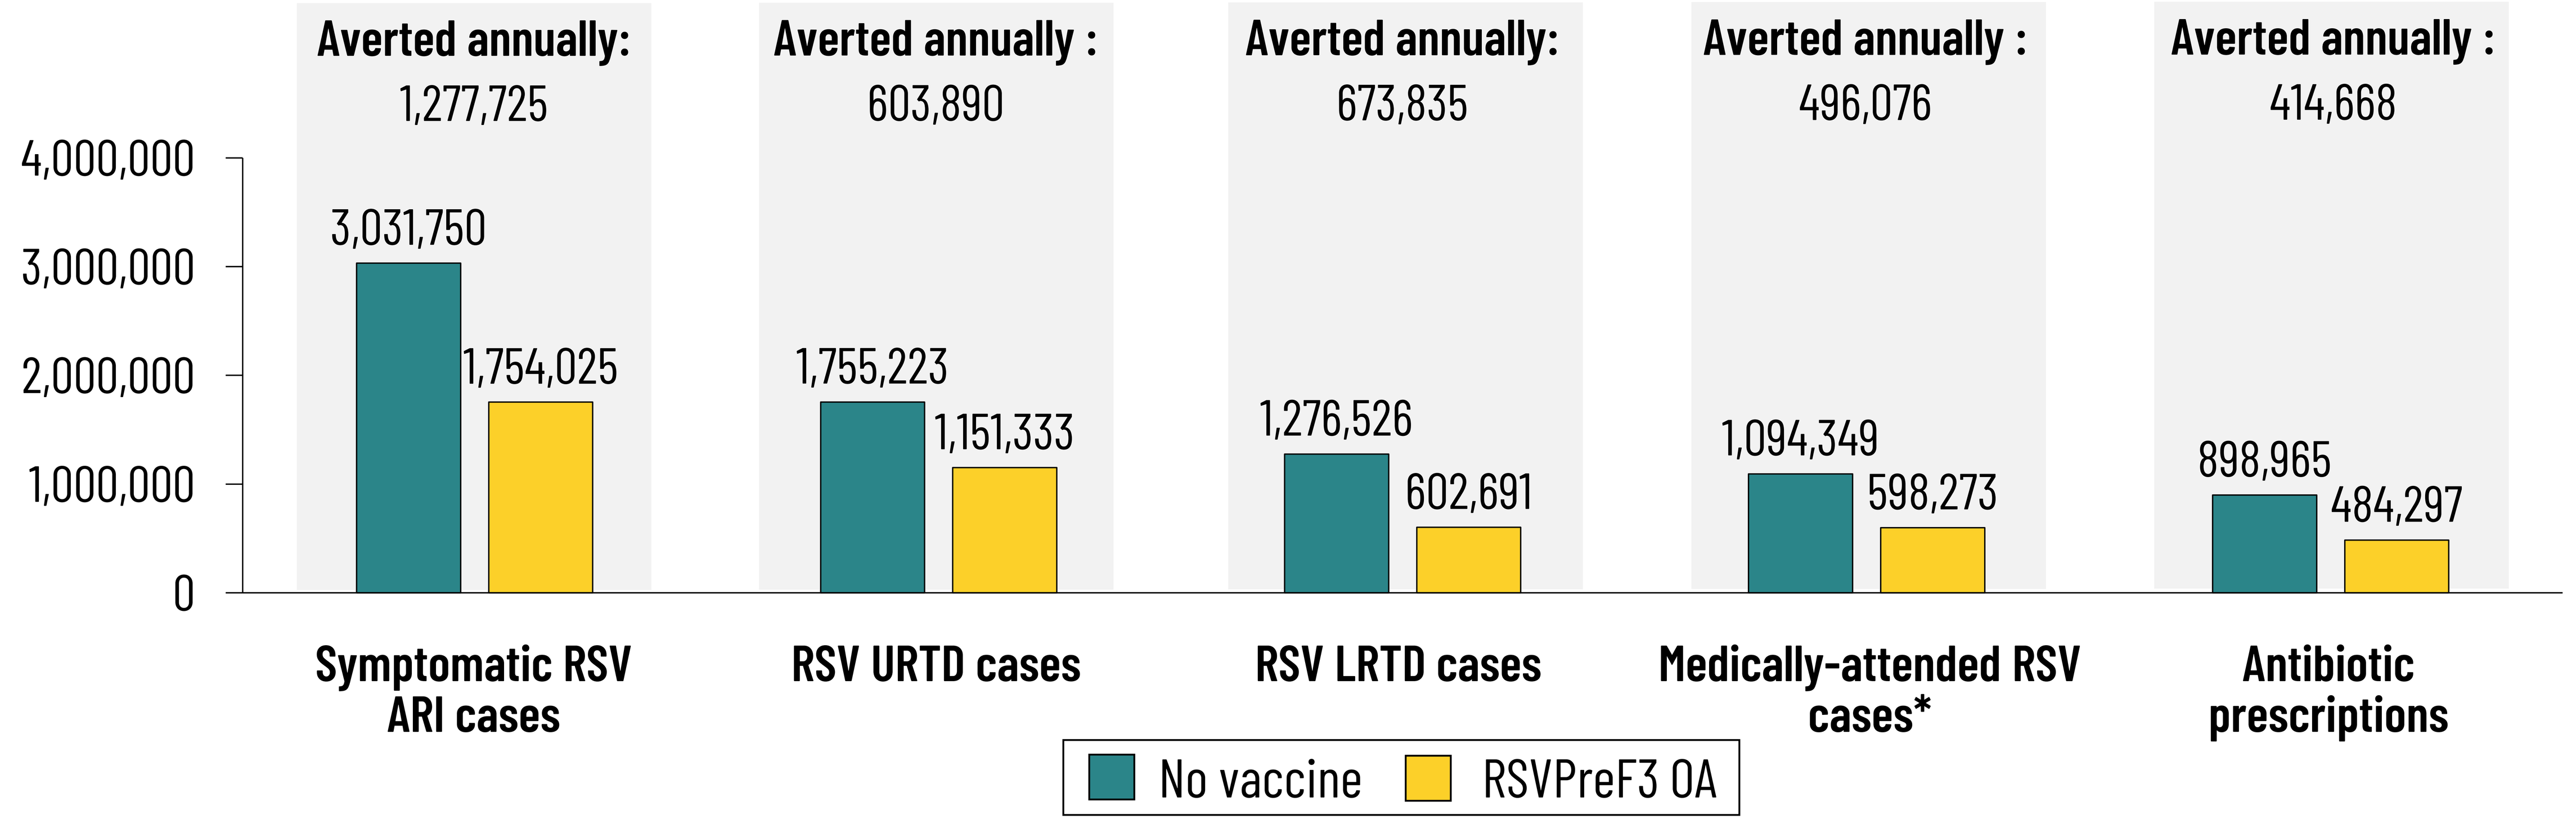
Averted annually: 1,277,725
Averted annually : 603,890
Averted annually: 673,835
Averted annually : 496,076
Averted annually : 414,668
4,000,000
3,000,000
2,000,000
1,000,000
0
3,031,750
1,754,025
1,755,223
1,151,333
1,276,526
602,691
1,094,349
598,273
898,965
484,297
Symptomatic RSV ARI cases
RSV URTD cases
RSV LRTD cases
Medically-attended RSV cases*
Antibiotic prescriptions
No vaccine
RSVPreF3 OA

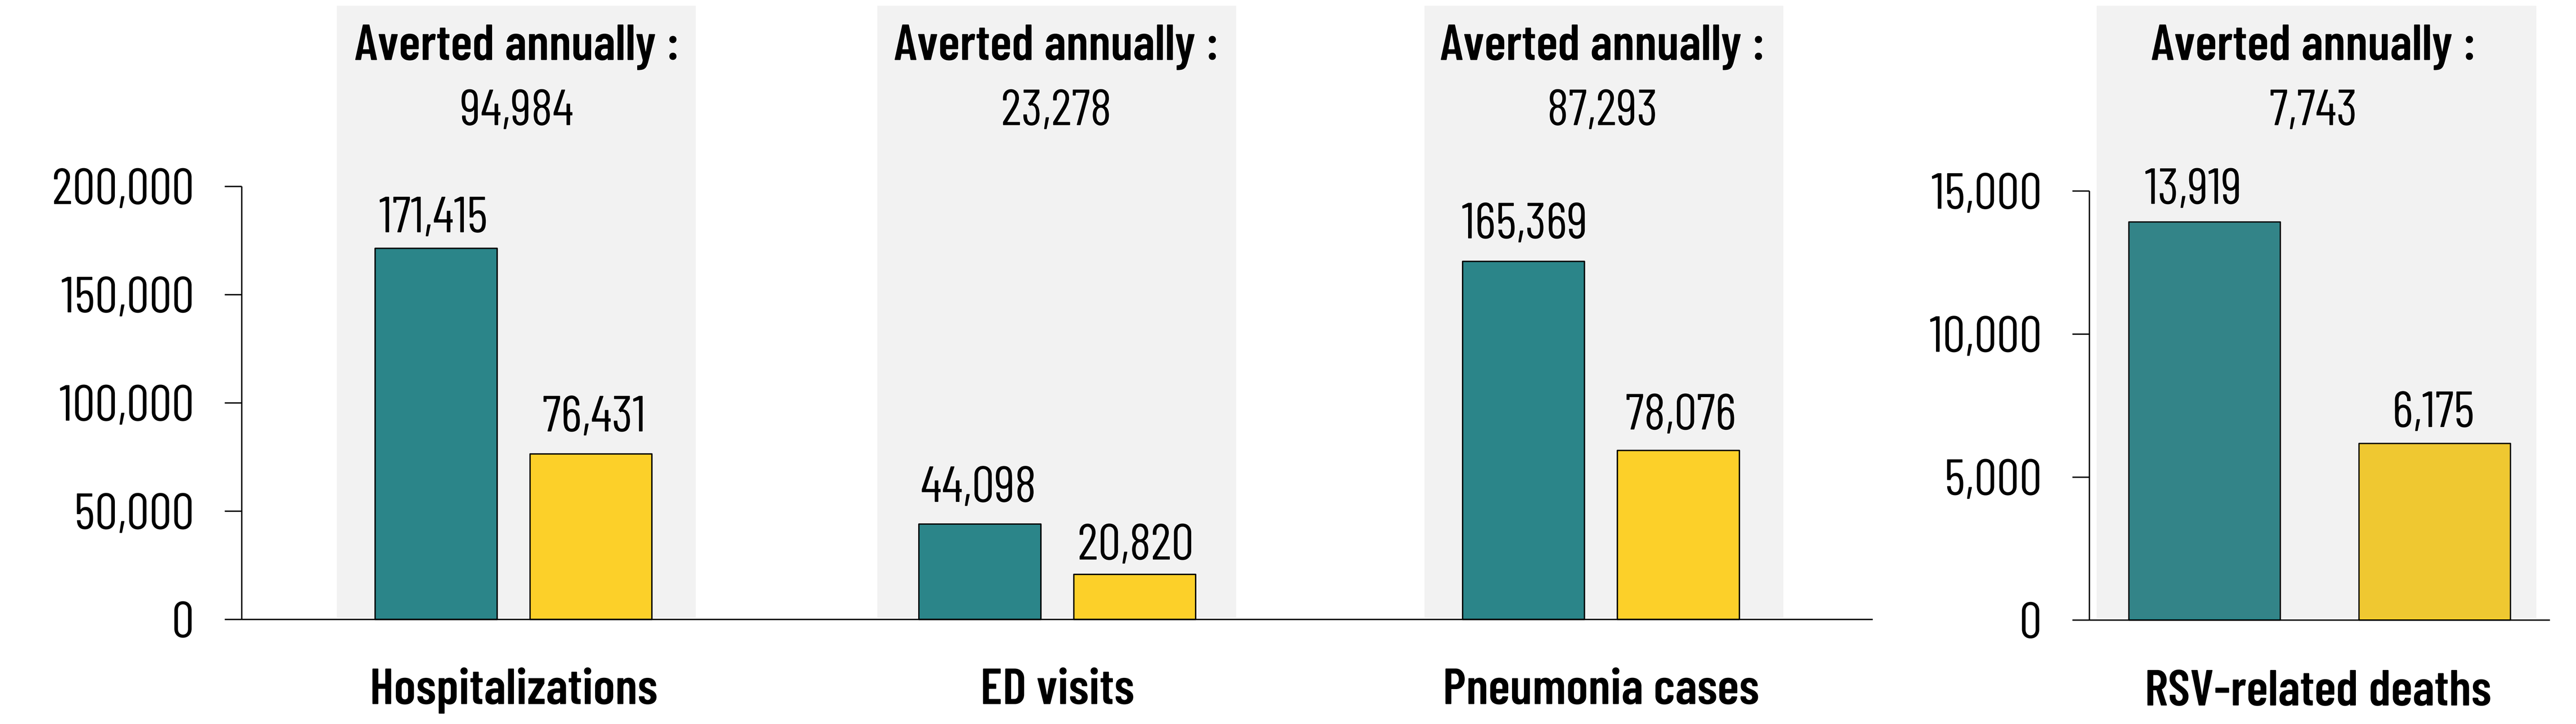
Averted annually : 94,984
Averted annually : 23,278
Averted annually : 87,293
Averted annually : 7,743
200,000
150,000
100,000
50,000
0
171,415
76,431
44,098
20,820
165,369
78,076
15,000
10,000
5,000
0
13,919
6,175
Hospitalizations
ED visits
Pneumonia cases
RSV-related deaths

** Because each medically-attended case was assumed to have 1 outpatient visit: medically-attended RSV cases = number of modeled outpatient visits.*

Outcome	Number needed to vaccinate to avoid one case
RSV ARI case	43
RSV LRTD case	82
Medically-attended RSV case	111
RSV hospitalization	582
RSV-related death	7,139

LIMITATIONS

The current analysis does not capture benefits of RSV vaccination beyond the 1-year time horizon

The model assumes RSV-related deaths only occur in hospitalized cases.

The model does not account for disease attenuation from the vaccine in breakthrough cases.

CONCLUSIONS

- ✓ Assuming the same vaccination coverage as for influenza vaccines, the adjuvanted RSVPreF3 OA vaccine has the potential to avert nearly 1.3 million symptomatic RSV cases, 95,000 RSV-related hospitalizations, and 8,000 RSV-related deaths annually among US adults aged ≥60 years vs. no vaccination.
- ✓ To achieve this modeled public health impact in real-world practice, efforts will be needed to support RSV vaccination among older adults (e.g., through patient and healthcare provider education on RSV disease and vaccines).

Presenting author: Elizabeth La, liz.m.la@gsk.com

ISPOR 2023 | May 7-10, 2023 | Boston, MA, USA

Funding: GlaxoSmithKline Biologicals SA (GSK study identifier: VEO- 000319); **Acknowledgments:** Business & Decision Life Sciences c/o GSK (writer: Sarah Fico); Jessica Pickett, Zinan Yi, Will Herring and Ilse Van Vlaenderen

© 2023 GSK