The Total Cost of Preeclampsia in the Ecuadorian Health System

EE296

Medina J[1], Ruiz C[2]
[1] Roche Ecuador, Quito, Ecuador,
[2] Universidad Central, Quito, Ecuador

OBJECTIVES: To determine the total cost of preeclampsia in the Ecuadorian Health System.

METHODS: The guide "Hypertensive disorders in pregnancy" 2016, The assessment of the study was carried out using the Tariff of Ecuador 2014; Regarding the drugs used, the prices were obtained from the list of official prices 2022 made by the Technical Secretariat of Pricing; as to the costs of the predictive markers of Preeclampsia were taken from the consultancy of direct and indirect costs.

RESULTS: Comprehensive care of the patient during pregnancy from 4 to 6 visits, determined an average value of 523 USD. Comprehensive care of the patient during pregnancy, determined an average value of 695 USD. The calculated value for the PLGF test is USD$30.89 and the value for SFlt-1 is $30.96 USD. For intensive care for a woman at high risk obstetrician, determined a value per day of 645.84 USD; For the intensive care of a newborn, determined a value per day of 931.50 USD; For the intensive care of a newborn, determined a value per day of 403.65 USD.

CONCLUSIONS: The fee generated by comprehensive care in high-risk patients obstetric care with the use of predictors of Preeclampsia is USD 1,080.76 for the second level of care and 1,157.82 for the third level of care. The fee generated by comprehensive care in high-risk patients without the use of predictors of Preeclampsia is USD 9,807.67 for the second level of care and 9,884.73 for the third level of care. It is concluded that it is more cost-effective to incorporate into high-risk prenatal care the Preeclampsia predictor markers, since it generates a saving of USD 8,726.91 for the second level of care and USD 8,726.91 for the third level of care.

AVERAGE SAVINGS WITH PRE-ECLAMPSIA FACTORS IN THE SECOND AND THIRD LEVEL OF CARE PER PATIENT IN THE SNS ECUADOR

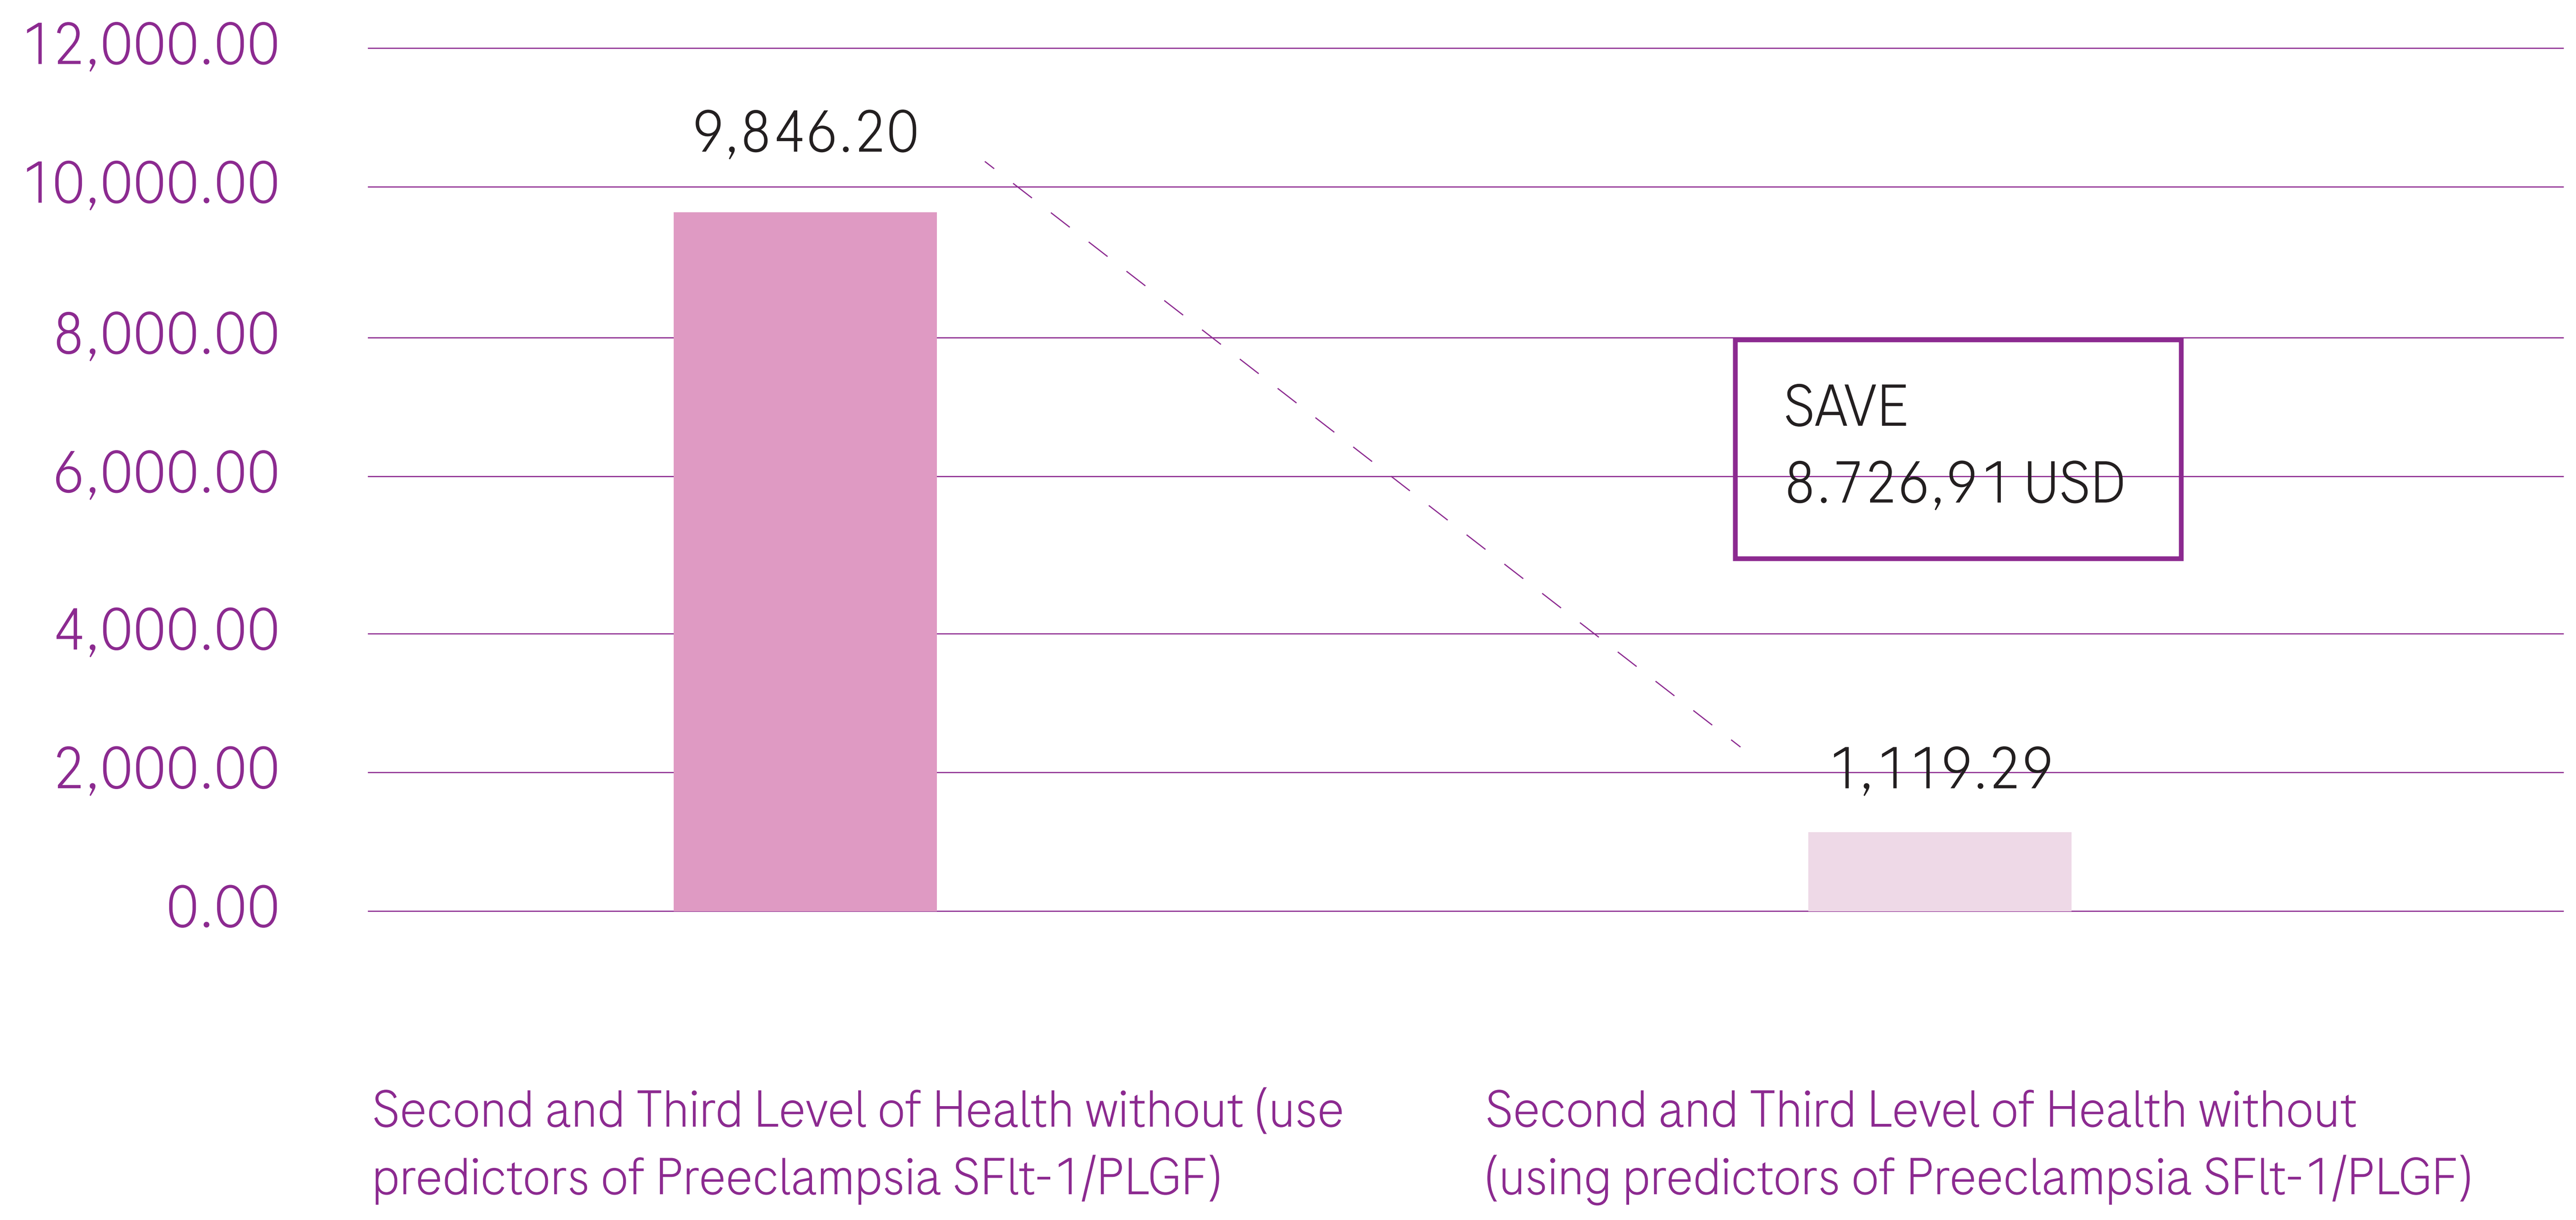
12,000.00
10,000.00
8,000.00
6,000.00
4,000.00
2,000.00
0.00
9,846.20
1,119.29
SAVE
8.726,91 USD
Second and Third Level of Health without (use predictors of Preeclampsia SFlt-1/PLGF)
Second and Third Level of Health without (using predictors of Preeclampsia SFlt-1/PLGF)