Development and validation of a machine learning algorithm to identify patients with atypical hemolytic uremic syndrome from US claims data

Genevieve Lyons,[1] Moh-Lim Ong,[1] Raj Bandaru,[1] Radhika Gangaraju,[2] Jones Samuel,[3] Yan Wang[1]

[1]Alexion, AstraZeneca Rare Disease, Boston, MA, USA; [2]Division of Hematology and Oncology, Department of Medicine, The University of Alabama at Birmingham, Birmingham, AL, USA; [3]Renal Associates of Baton Rouge, Baton Rouge, LA, USA

MSR98

INTRODUCTION

- Atypical hemolytic uremic syndrome (aHUS) is a rare disease characterized by microangiopathic hemolytic anemia, thrombocytopenia and renal damage that can lead to death if left untreated.[1]
- Lack of a specific International Classification of Diseases (ICD) code for aHUS (before 2022) has made it difficult to appropriately identify patients from claims databases to study aHUS outcomes.

OBJECTIVE

- To develop and validate an algorithm to identify patients with aHUS in claims data analysis.

METHODS

Study database

- Linked electronic health record and claims data from Optum's de-identified Market Clarity Data (January 2016 to January 2022) for patients with chronic kidney disease (ICD-10 code, N18) were used.

Study population eligibility

- ≥ 2 hemolytic uremic syndrome (HUS) diagnosis codes (ICD-10 code, D59.3) or HUS-related hemolytic anemia codes (ICD-10 codes, D59.4, D59.9) or ≥ 2 thrombotic microangiopathy (TMA) diagnosis codes (ICD-10 code, M31.1) within 6 months. The index date was the date of the first HUS or TMA diagnosis code.
- Records with ≥ 2 physician notes between 2 weeks before and 1 year after index date.
- No diagnosis code of other complement-mediated diseases (paroxysmal nocturnal hemoglobinuria, myasthenia gravis, neuromyelitis optica spectrum disorder, folate deficiency and sickle cell) within 6 months before or after index date.

Algorithm development and testing

- A rule-based algorithm was developed as the first step to incorporate relevant criteria for the identification of aHUS diagnosis based on literature reviews and clinician input.
- Three machine learning algorithms (elastic net, classification and regression trees, and random forest) were developed using treatment history and clinical features (renal, pulmonary, cardiovascular, hematological) before or after the index date.
 - Models were run as ensemble machine learning algorithms with the most important features (χ^2; $p < 0.05$) used by the algorithms to identify aHUS cases.
 - The elastic net machine learning algorithm was developed with two different probability thresholds (0.50 and 0.75) to allow the selection of parameters for optimal performance.
- Training/testing set: 70% and 30% of the population, respectively.
- Ground-truth of aHUS: patients with the term 'aHUS' or 'atypical hemolytic uremic syndrome' mentioned in ≥ 2 physician notes, without a negative sentiment, such as 'aHUS unlikely' and without positive mention of thrombotic thrombocytopenic purpura or with confirmed treatment of aHUS (induction and 1 maintenance dose; ≥ 5 eculizumab or ≥ 2 ravulizumab infusions).
- Performance measures in identifying aHUS: sensitivity, specificity, positive predictive value and area under the curve.
 - Assessed the sensitivity of identifying ever/never treated patients with aHUS (ever: ≥ 1 eculizumab or ravulizumab infusion).

RESULTS

1. Population

- Of the 5,800,383 patients in this data set, 1,992 met the study eligibility criteria and the majority were female and adult patients.
- Among the 1,992 patients included, 304 (15%) had aHUS based on the definition of ground-truth (**Table 1**).

2. Algorithm performance

- In the testing set, the rule-based algorithm did not provide a clear differentiation between patients with or without aHUS.
- The elastic net machine learning algorithm had the highest positive predictive value (85%) and the highest specificity (98%), with 73% sensitivity (**Table 2**).

Table 1. Study population

Characteristic, n (%)	Patients (N = 1,992)	
	Patients with aHUS (n = 304)	Patients without aHUS (n = 1,688)
Sex		
Female	187 (61.5)	951 (56.3)
Age group		
< 18 years	38 (12.5)	93 (5.5)
18–64 years	218 (71.7)	920 (54.5)
≥ 65 years	48 (15.8)	675 (40.0)

Table 2. Performance of machine learning algorithms in the testing set

Machine learning algorithm	Identification of aHUS				Identification of never treated aHUS	Identification of ever treated aHUS
	Sensitivity	Specificity	PPV	AUC	Sensitivity	Sensitivity
Elastic net						
Higher PPV[a]	73%	98%	85%	0.96	42%	84%
Lower PPV[b]	81%	94%	73%	0.96	58%	90%
Classification and regression tree	95%	79%	44%	0.93	83%	99%
Random forest	92%	79%	44%	0.94	83%	96%
Rule-based algorithm	72%	82%	42%	NR	21%	93%

[a]Elastic net algorithm with a probability threshold of 0.75.
[b]Elastic net algorithm with a probability threshold of 0.50.
aHUS, atypical hemolytic uremic syndrome; AUC, area under the curve; NR, not reported; PPV, positive predictive value.

3. Key clinical features identified from the elastic net machine learning algorithm

Figure 1. Feature importance[a] for the elastic net machine learning algorithm

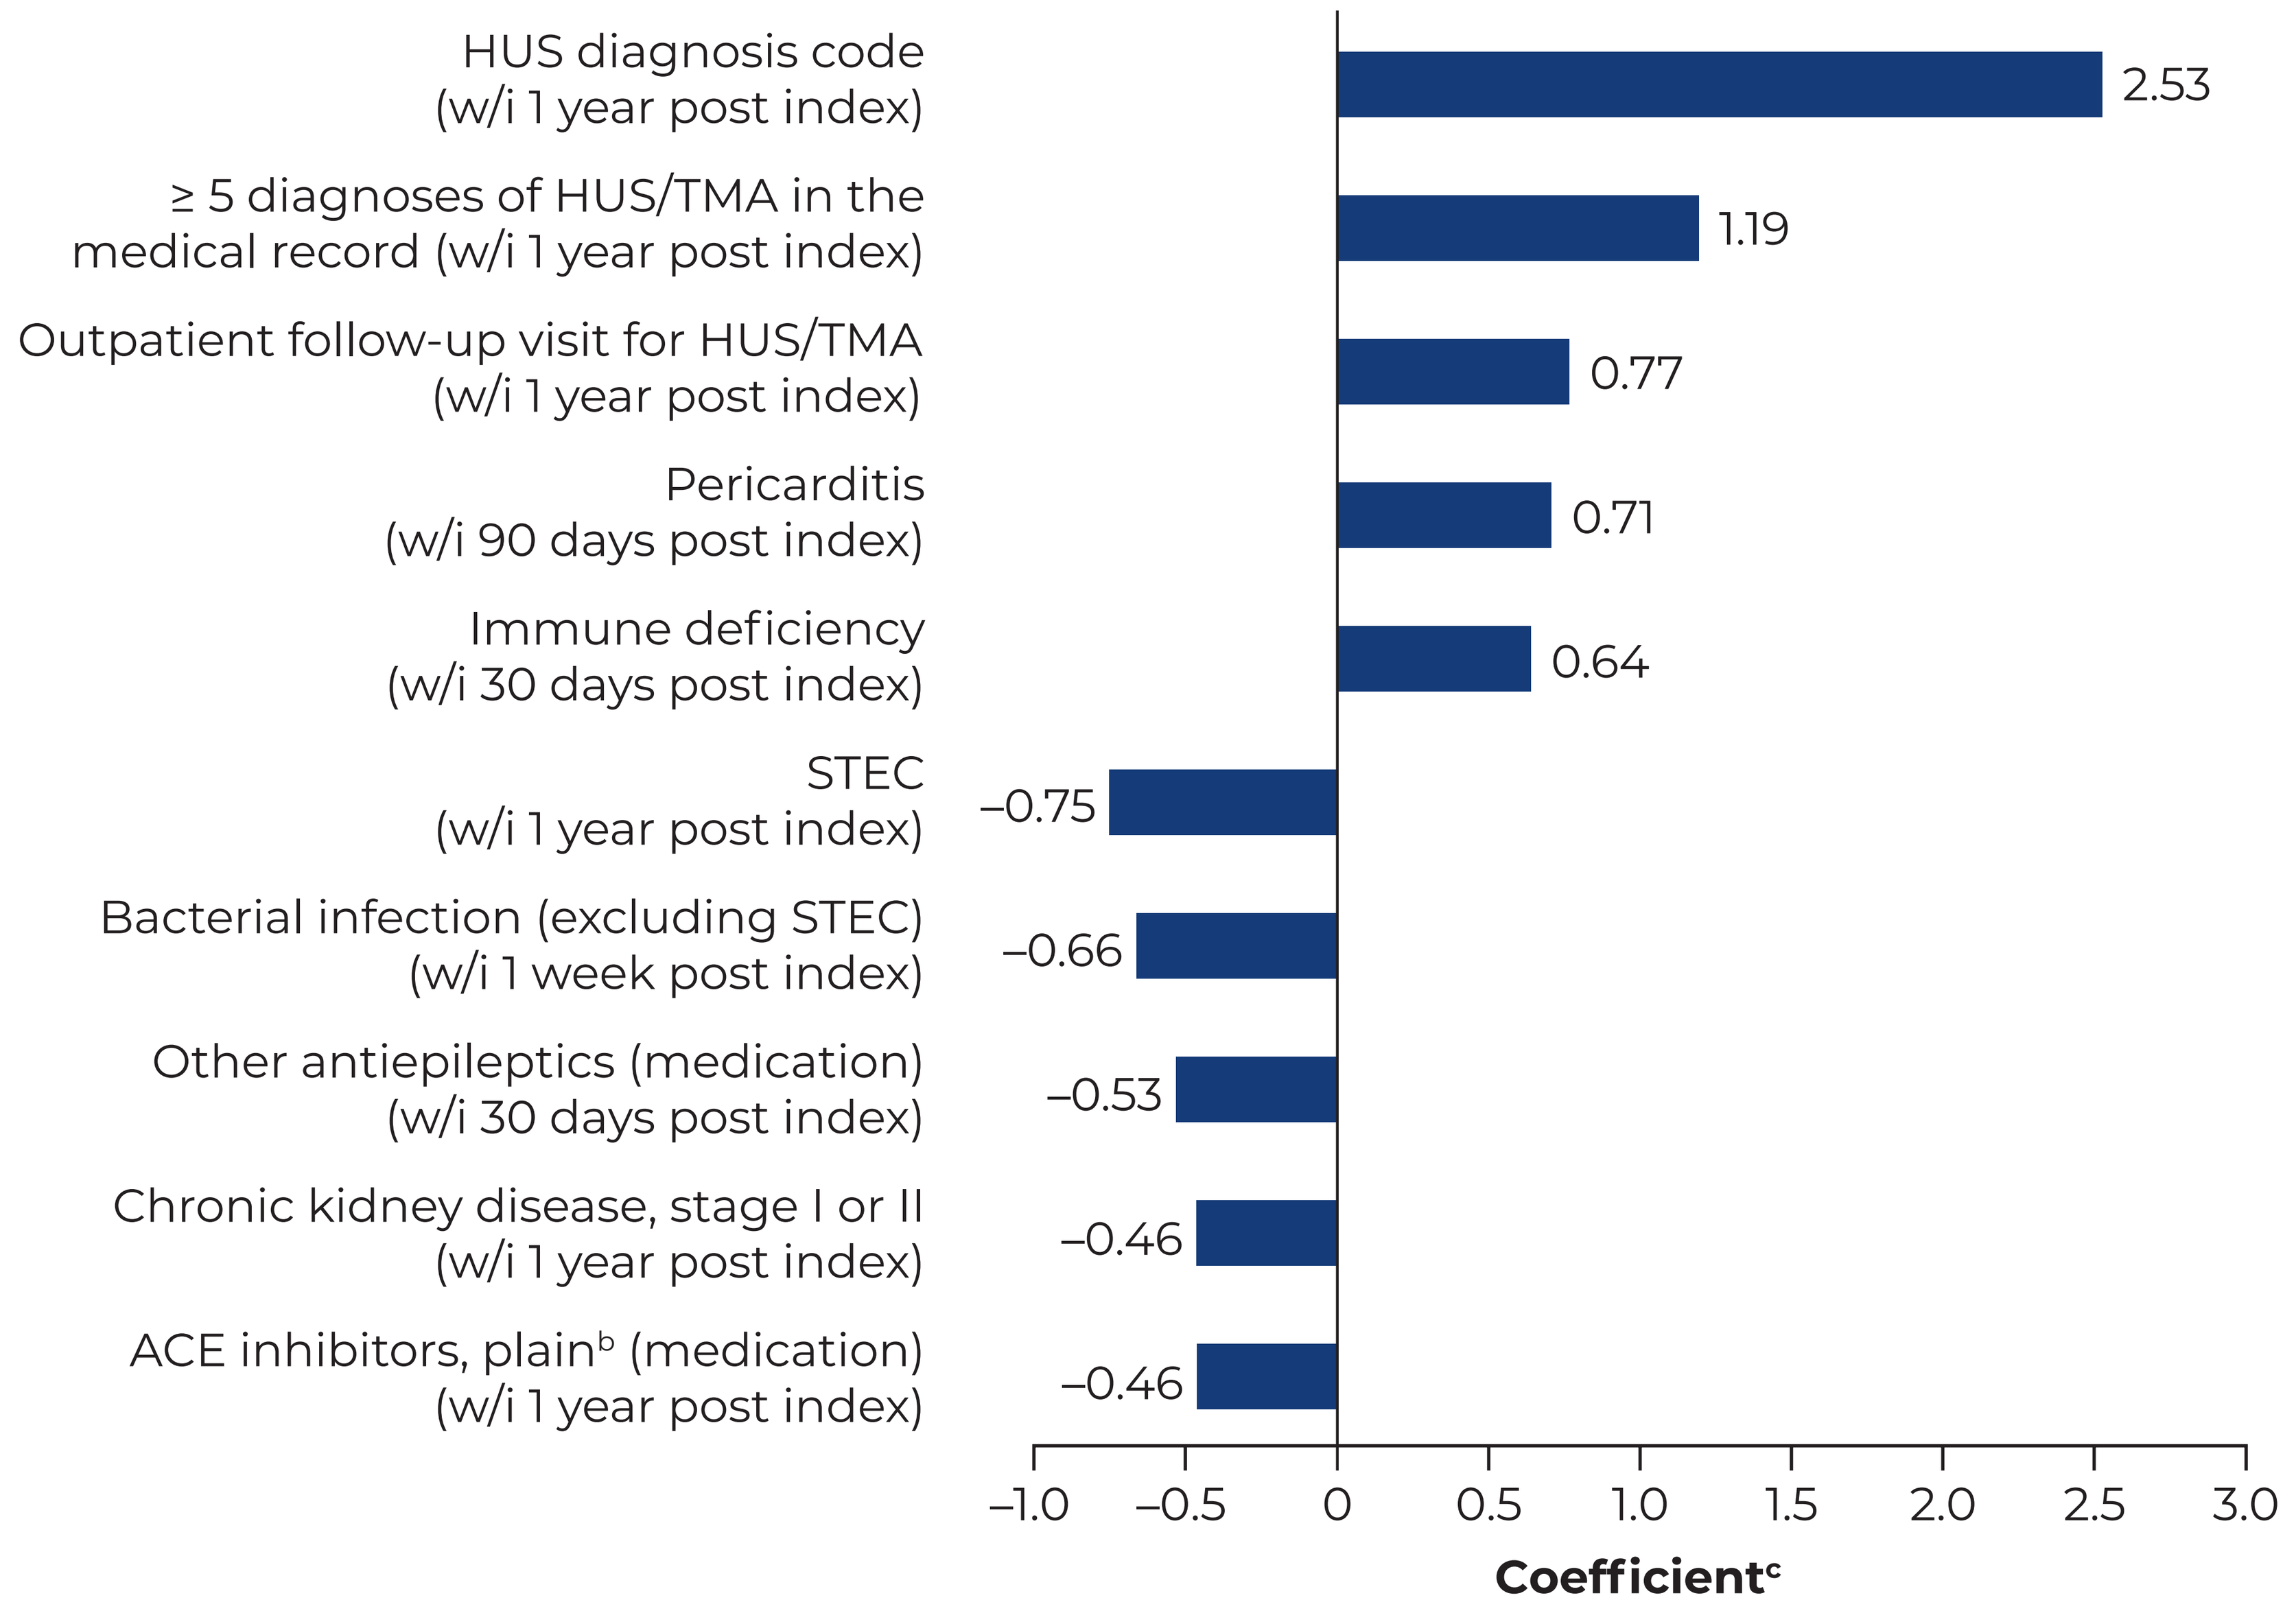
HUS diagnosis code (w/i 1 year post index)
2.53
≥ 5 diagnoses of HUS/TMA in the medical record (w/i 1 year post index)
1.19
Outpatient follow-up visit for HUS/TMA (w/i 1 year post index)
0.77
Pericarditis (w/i 90 days post index)
0.71
Immune deficiency (w/i 30 days post index)
0.64
STEC (w/i 1 year post index)
–0.75
Bacterial infection (excluding STEC) (w/i 1 week post index)
–0.66
Other antiepileptics (medication) (w/i 30 days post index)
–0.53
Chronic kidney disease, stage I or II (w/i 1 year post index)
–0.46
ACE inhibitors, plain[b] (medication) (w/i 1 year post index)
–0.46
–1.0 –0.5 0 0.5 1.0 1.5 2.0 2.5 3.0
Coefficient[c]

- Among the 1,273 candidate features, k = 272 passed the feature selection pruning step and were used in model training. The top 5 features with the highest positive magnitude and negative magnitude are listed (**Figure 1**).
- A positive coefficient indicates that a patient with this feature is more likely to have aHUS instead of non-aHUS, while a negative coefficient indicates that a patient with this feature is less likely to have aHUS.
- The magnitude of the coefficient indicates the relative weight of the feature for predicting whether a patient has aHUS.

[a]≥ 5 patients with or without ground-truth aHUS.
[b]World Health Organization Anatomical Therapeutic Codes C09AA01–16.
[c]The coefficients represent adjusted coefficients in the algorithm used to identify aHUS from the linked electronic health record and claims database.
ACE, angiotensin-converting enzyme; (a)HUS, (atypical) hemolytic uremic syndrome; STEC, Shiga toxin-producing *Escherichia coli*; TMA, thrombotic microangiopathy; w/i, within.

CONCLUSIONS

This study demonstrated that the machine learning approach may address limitations of the traditional rule-based algorithms and may help improve the methodology of identifying patients with aHUS in claims data to better understand the aHUS treatment landscape and outcomes.

The elastic net machine learning algorithm may be a useful tool for future claims data analyses for more accurate identification of patients with aHUS.

PLAIN LANGUAGE SUMMARY

Why did we perform this research?

Atypical hemolytic uremic syndrome (aHUS) is a rare disease that can cause damage to the small blood vessels in the body's organs, most often in the kidneys, which can lead to death if left untreated. Before 2022, there was no specific way of recording in a database that a patient had aHUS, making it difficult to study patient outcomes using electronic health record and insurance claims data. This study aimed to develop and validate a machine learning algorithm (a computer program that uses statistical techniques to identify patterns in data and predict outputs) to classify patients with aHUS using anonymous information from a database of electronic health records and claims.

How did we perform this research?

Electronic health record and claims data of patients with chronic kidney disease were used to develop and test three machine learning algorithms. Among the 1,992 patients included, 15% had confirmed aHUS based on physician notes. Algorithm performance was measured by positive predictive value (proportion of patients who were predicted to have aHUS who did have confirmed aHUS), specificity (proportion of patients without aHUS who were correctly predicted) and sensitivity (proportion of patients with aHUS who were correctly predicted) for each algorithm.

What were the findings of this research and what are the implications?

The top-performing algorithm had an 85% positive predictive value, 98% specificity and 73% sensitivity. In addition, several clinical features (renal, pulmonary, cardiovascular, hematological and others) were associated with either an increased or decreased chance of being identified as aHUS. This machine learning algorithm has the potential to permit more accurate identification of a larger group of patients with aHUS in large databases, which could provide an improved understanding of the treatment of patients with aHUS.

Acknowledgments

Medical writing support was provided by Elena Sugrue, PhD, of Oxford PharmaGenesis, Oxford, UK and was funded by Alexion, AstraZeneca Rare Disease, Boston, MA, USA.

Disclosures

This study was sponsored by Alexion, AstraZeneca Rare Disease, Boston, MA, USA. GL, **MLO**, **RB** and **YW** are employees and stockholders of Alexion, AstraZeneca Rare Disease, Boston, MA, USA. **RG** served as a consultant to Sanofi and Alexion, and participated in an advisory board for Sanofi. **JS** is a speaker for Alexion and Bayer Pharmaceuticals.

Reference

1. Raina R *et al. Ther Apher Dial* 2019;23:4–21.

Poster presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 2023 Annual Meeting, May 7–10, 2023, Boston, MA, USA

Corresponding author email address: genevieve.lyons@alexion.com