CO27

Real-World Treatment Response and Persistence among Black and Non-Black Patients with Metastatic Castration-Sensitive Prostate Cancer Treated with Apalutamide in a Urology Setting

Benjamin Lowentritt[1], Shawn Du[2], Carmine Rossi[3], Frederic Kinkead[3], Bronwyn Moore[3], Patrick Lefebvre[3], Dominic Pilon[3], Erik Muser[2], Ibrahim Khilfeh[2]

[1]Chesapeake Urology, Towson, MD, USA, MD, United States; [2]Janssen Scientific Affairs, LLC, Horsham, PA, USA; [3]Analysis Group, Inc., Montréal, QC, Canada

BACKGROUND

- Evidence from the Phase III TITAN trial demonstrated that apalutamide (APA), an androgen receptor signaling inhibitor (ARSI), in combination with androgen deprivation therapy (ADT), improved clinical outcomes among patients with metastatic castration-sensitive prostate cancer (mCSPC)[1,2], leading to the regulatory approval of APA plus ADT for treatment of mCSPC in September 2019[3]
- Reduction of prostate-specific antigen (PSA) observed in clinical trials of ARSIs has been associated with increased probability of achieving progression-free and/or overall survival outcomes, with patients achieving ≥90% reduction in baseline PSA having a greater likelihood of long-term benefit[4-7]
- Racial disparities in prostate cancer (PC) outcomes are well-documented[8], but real-world data describing treatment patterns, including adherence and persistence, and measures of APA effectiveness in racial subgroups of patients with mCSPC remains limited
- In a previous real-world study of Black patients with mCSPC treated with APA, the majority of men achieved ≥90% reduction in baseline PSA, but this study was limited by a small sample size (44 Black patients)[9]

OBJECTIVE

- To evaluate treatment persistence at 6 and 12 months and PSA reduction ≥90% (PSA90 response) from baseline among a larger cohort of Black and non-Black patients with mCSPC initiated on APA in US urology practices

METHODS

Data source

- Clinical data from Precision Point Specialty (PPS) Analytics collected as part of routine clinical care from 95 urology sites in the US (study period: 17 September 2018 – 1 April 2022)
- Data were de-identified and Health Insurance Portability and Accountability Act (HIPAA) compliant

Study design

- A retrospective, longitudinal cohort study of patients with mCSPC initiated on APA was conducted
- Index date was defined as the first dispensation of APA on or after 17 September 2019 (the US Food and Drug Administration [FDA] approval date for APA[3])
- Baseline patient characteristics were evaluated in the 12 months preceding the index date
- The observation period spanned from the index date to the earliest of end of clinical activity (including death) or end of data availability (1 April 2022)

Patient selection criteria

- Patients were included if they met the following criteria:
 - Male sex
 - ≥1 in-office medication dispensation for APA
 - Evidence of metastatic disease defined by the presence of bone, nodal, or visceral metastasis prior to or on the index date
 - ≥18 years of age on the index date
 - ≥12 months of clinical activity prior to the index date
- Patients were excluded if they met any of the following criteria:
 - Evidence of castration resistance prior to or on the index date, based on an algorithm utilizing presence of ADT and PSA levels[10] or an indicator for castration resistance as reported in the clinical data
 - Index date prior to the US FDA approval for APA on 17 September 2019
 - Prior use of an ARSI before the index date or the use of ≥2 ARSIs on the index date
 - Prior use of radiopharmaceuticals
- Concurrent use of ADT was not required for patients to be included in the APA cohort

PROSTATE CANCER

FIGURE 1: Patient identification flowchart

≥1 dispensation[a] for an ARSI *(first dispensation defined as the index date)*
N=10,841

↓

No more than 1 ARSI dispensed on the index date[a]
N=10,840 (100%)

↓

No prescription[b] for an ARSI other than that dispensed on the index date *(looking back at all available data)*
N=9,532 (87.9%)

↓

Initiated treatment with APA
N=1,739 (18.2%)

↓

mCSPC indication
N=784 (45.1%)

↓

Index date on or after 17 September 2019[c]
N=777 (99.1%)

↓

≥18 years of age at the index date
N=777 (100.0%)

↓

≥12 months of clinical activity prior to index date[d]
N=589 (75.8%)

↓

No previous treatment for radiopharmaceuticals
N=589 (100%)

↓

mCSPC APA cohort
N=589

↓

Black race	Non-Black race	Unknown race
N=98 (16.6%)	**N=424 (72.0%)**	**N=67 (11.4%)**

APA: apalutamide; ARSI: androgen receptor signaling inhibitor; mCSPC: metastatic castration-sensitive prostate cancer.
Notes:
a. Agent identified at index date was based on in-office medication dispensations only.
b. To confirm that patients were newly initiated on an ARSI as identified through dispensations, prescription information was examined to exclude patients with prescriptions for ARSIs prior to the index date.
c. The Food and Drug Administration (FDA) approved APA as treatment for mCSPC on 17 September 2019.
d. Clinical activity was defined as the period from the first to last record in the clinical database. Patients with no observation period after the index date were excluded.

Study outcomes

- Proportion of patients without a gap in APA treatment (i.e., treatment persistence) >60 days or >90 days by 6- and 12-months post-index
 - Treatment gaps were assessed from the end of the days of supply of the previous dispensation to the earliest of the next dispensation or the end of data availability
- Proportion of patients who achieved ≥90% reduction in PSA (i.e., PSA90) from the most recent baseline value at 3, 6, 9 and 12-months post-index
 - PSA90 response was assessed while patients remained on-treatment with APA among those with a baseline PSA value within 13 weeks prior to, and including, the index date and ≥1 post-index PSA test

Statistical analyses

- Patients were required to have complete observations during each specific time period to be included in each persistence calculation (i.e., at 6 and 12 months)
- Kaplan-Meier (KM) analysis was used to assess PSA90 response
 - PSA90 response was censored at the earliest of APA discontinuation (using a 90-day treatment gap to define discontinuation), initiation of another ARSI (i.e., abiraterone acetate, darolutamide, enzalutamide), use of radiopharmaceuticals, end of clinical activity, or end of data availability
 - Concurrent use of prednisone was not required for patients who switched to abiraterone acetate to be censored
- Outcomes were reported overall and stratified by Black and non-Black race
 - Patients with an unknown race were included in the overall APA cohort

RESULTS

Baseline characteristics

- A total of 589 patients with mCSPC initiated APA, of which 98 (16.6%) were Black, 424 (72.0%) were non-Black, and 67 (11.4%) had an unknown race (**Figure 1**)
- Overall, the mean age was 75.9 years (Black: 72.4 years; non-Black: 76.8 years) and the median time between metastasis and APA initiation was 1.5 months (Black: 2.2 months; non-Black: 1.5 months; **Table 1**)
- Concurrent ADT use was observed among 96.3% of patients overall (Black: 95.9%; non-Black: 96.0%)
- Overall, 81.2% of patients (Black: 76.5%; non-Black: 82.8%) had a baseline PSA test within 13 weeks of APA initiation, with a mean (median) PSA level of 19.2 (3.4) ng/mL (Black: 22.7 [3.3] ng/mL; non-Black: 18.3 [3.1] ng/mL)
- Overall, the mean (median) observation period was 388 (341) days (Black: 356 [307] days; non-Black 389 [337] days)

TABLE 1: Baseline characteristics, overall and among Black and non-Black subgroups

	Overall N=589	Black N=98	Non-Black N=424
Age, mean ± SD [median]	**75.9 ± 7.7 [76.0]**	**72.4 ± 8.0 [72.0]**	**76.8 ± 7.4 [77.0]**
Year of index date, n (%)			
2019	51 (8.7)	7 (7.1)	33 (7.8)
2020	186 (31.6)	30 (30.6)	139 (32.8)
2021	288 (48.9)	42 (42.9)	213 (50.2)
2022	64 (10.9)	19 (19.4)	39 (9.2)
Time between metastases and index date, months, median [IQR]	**1.5 [0.5, 4.9]**	**2.2 [0.9, 5.7]**	**1.5 [0.5, 5.0]**
Metastasis type[a], n (%)			
Bone	420 (71.3)	67 (68.4)	312 (73.6)
Nodal	259 (44.0)	41 (41.8)	184 (43.4)
Visceral	18 (3.1)	2 (2.0)	13 (3.1)
Prior use of ADT[b], n (%)	**533 (90.5)**	**92 (93.9)**	**385 (90.8)**
Time between ADT initiation and index date, months, mean ± SD [median]	18.0 ± 29.5 [3.2]	13.5 ± 21.8 [3.4]	19.5 ± 31.1 [3.3]
Concurrent use of ADT[c], n (%)	**567 (96.3)**	**94 (95.9)**	**407 (96.0)**
Prior use of first-generation ARI[d], n (%)	**67 (11.4)**	**9 (9.2)**	**49 (11.6)**
Bone antiresorptive therapy, n (%)	**150 (25.5)**	**20 (20.4)**	**119 (28.1)**
PSA test within 13 weeks prior to and including the index date, n (%)	**478 (81.2)**	**75 (76.5)**	**351 (82.8)**
PSA level, ng/ml, mean ± SD [median]	19.2 ± 43.3 [3.4]	22.7 ± 50.3 [3.3]	18.3 ± 42.1 [3.1]
PSA doubling time, n (%)	**48 (8.1)**	**10 (10.2)**	**34 (8.0)**
PSA doubling time, months, mean ± SD [median]	9.5 ± 13.9 [5.6]	10.0 ± 8.8 [7.7]	8.9 ± 15.6 [4.3]
Gleason score[e], n (%)			
≤6	28 (4.8)	4 (4.1)	21 (5.0)
7	103 (17.5)	15 (15.3)	76 (17.9)
8	68 (11.5)	10 (10.2)	51 (12.0)
9	130 (22.1)	20 (20.4)	91 (21.5)
10	9 (1.5)	0 (0.0)	9 (2.1)
Not available	251 (42.6)	49 (50.0)	176 (41.5)
Next-generation imaging testing[f], n (%)	**166 (28.2)**	**24 (24.5)**	**121 (28.5)**
Time between most recent test and index date, months, mean ± SD [median]	2.6 ± 2.4 [1.8]	3.0 ± 2.1 [2.4]	2.5 ± 2.4 [1.7]

ADT: androgen deprivation therapy; ARI: androgen receptor inhibitor; IQR: interquartile range; PSA: prostate-specific antigen; SD: standard deviation.
Notes:
a. Types of metastases were not mutually exclusive.
b. Prior use of ADT medication was defined as any ADT administration at any time prior to (and excluding) the index date.
c. Concurrent use of ADT medication was defined as any ADT administration observed 180 days before or after the index date.
d. Prior use of first-generation ARI was defined as any prescription for bicalutamide, nilutamide, or flutamide, at any time prior to (and excluding) the index date.
e. Gleason score was evaluated at any time prior to and including the index date, with the most recent value reported.
f. Next-generation imaging testing was evaluated during the 12-month baseline period and included the index date.

Persistence

- Persistence at 6 months using >60- and >90-day gaps was 92.7% and 96.8% overall, 95.4% and 98.5% for Black patients, and 93.4% and 96.9% for non-Black patients (**Figure 2**)
- Persistence at 12 months using >60- and >90-day gaps was 89.1% and 94.9% overall, 93.3% and 97.8% for Black patients, and 89.3% and 94.4% for non-Black patients

FIGURE 2: Apalutamide treatment persistence using gaps of >60 days and >90 days of coverage, overall and among Black and non-Black subgroups[a]

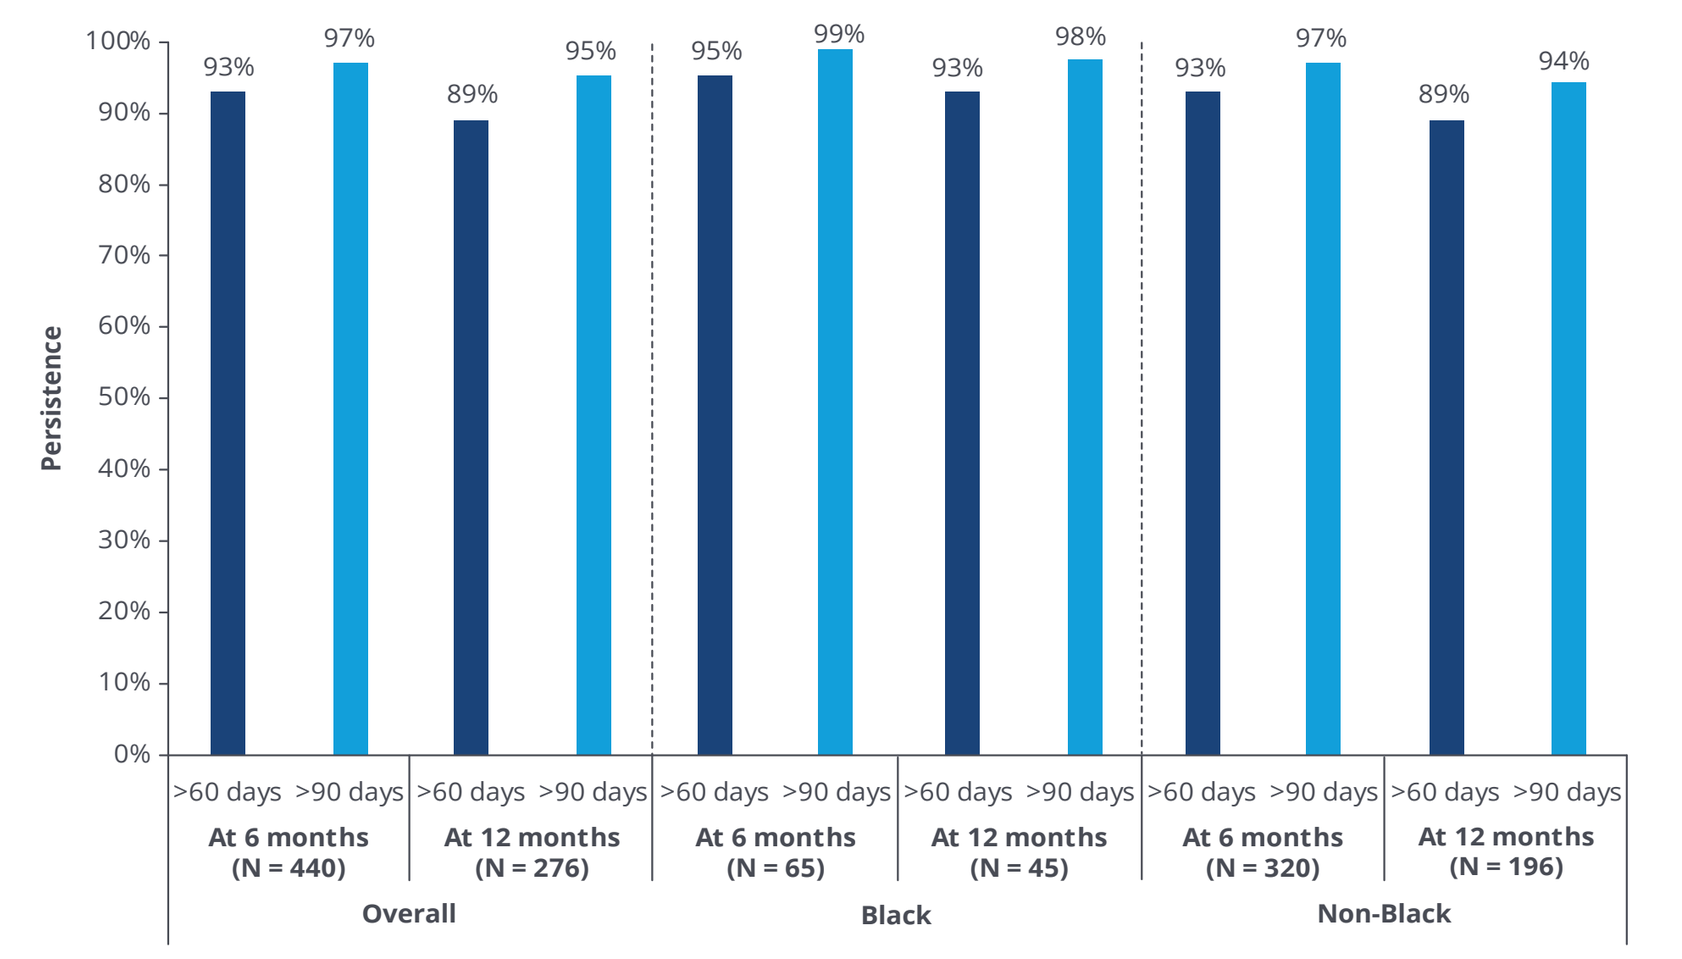
Persistence
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
93%
97%
89%
95%
95%
99%
93%
98%
93%
97%
89%
94%
>60 days
>90 days
At 6 months (N = 440)
At 12 months (N = 276)
At 6 months (N = 65)
At 12 months (N = 45)
At 6 months (N = 320)
At 12 months (N = 196)
Overall
Black
Non-Black

Note:
a. Patients with an unknown race were included in the overall population.

PSA testing

- Most patients (Overall: 80.8%; Black: 76.5%; Non-Black: 81.6%) had ≥1 on-treatment PSA test, with nearly all of these patients having their first PSA test within 6 months following APA initiation (Overall: 99.2%; Black: 97.3%; Non-Black: 99.7%) (**Table 2**)
- The mean (median) number of on-treatment PSA tests per year was 4.3 (3.8) overall, 4.1 (3.6) among Black patients, and 4.3 (3.9) among non-Black patients

TABLE 2: On-treatment PSA testing, overall and among Black and non-Black subgroups

	Overall N=589	Black N=98	Non-Black N=424
Patients with ≥1 PSA test[a], n (%)	**476 (80.8)**	**75 (76.5)**	**346 (81.6)**
Within 3 months of observation	429 (90.1)	66 (88.0)	311 (89.9)
Within 6 months of observation	472 (99.2)	73 (97.3)	345 (99.7)
Number of follow-up PSA tests per year, mean ± SD [median]	**4.3 ± 3.5 [3.8]**	**4.1 ± 3.7 [3.6]**	**4.3 ± 3.5 [3.9]**
Patients with PSA test on average every 3 months, n (%)	276 (46.9)	44 (44.9)	200 (47.2)
Patients with PSA test on average every 6 months, n (%)	454 (77.1)	70 (71.4)	334 (78.8)

ARSI: androgen receptor signaling inhibitor; PSA: prostate-specific antigen; SD: standard deviation.
Note:
a. PSA-related outcomes were assessed only while the patient was on apalutamide, with follow-up time censored at the earliest of apalutamide discontinuation (using a 90-day treatment gap to define discontinuation), initiation of a new ARSI or the use of radiopharmaceuticals, end of clinical activity (including death), or end of data availability (1 April 2022).

PSA90 response

- The median time to PSA90 response was 3.2 months overall, 3.8 months for Black patients, and 3.1 months for non-Black patients (**Figure 3**)
 - Among patients achieving a PSA90 response, PSA90 was achieved on the first PSA test among 68.0% of patients overall, 77.1% of Black patients, and 67.4% of non-Black patients
- By 6-months post-index, PSA90 response was achieved by 67.4% of patients overall, 64.7% of Black patients, and 67.5% of non-Black patients (**Figure 3**)
 - PSA90 response was achieved by 68% of patients overall in the post-hoc analysis of TITAN by 6 months[5]
- By 12-months post-index, PSA90 response was achieved by 72.3% of patients overall, 73.1% of Black patients, and 70.8% of non-Black patients (**Figure 3**)
 - PSA90 response was achieved by 71% of patients overall in the post-hoc analysis of TITAN by 12 months[5]

FIGURE 3: Kaplan-Meier rates of PSA response ≥90%, overall and among Black and non-Black subgroups[a,b]

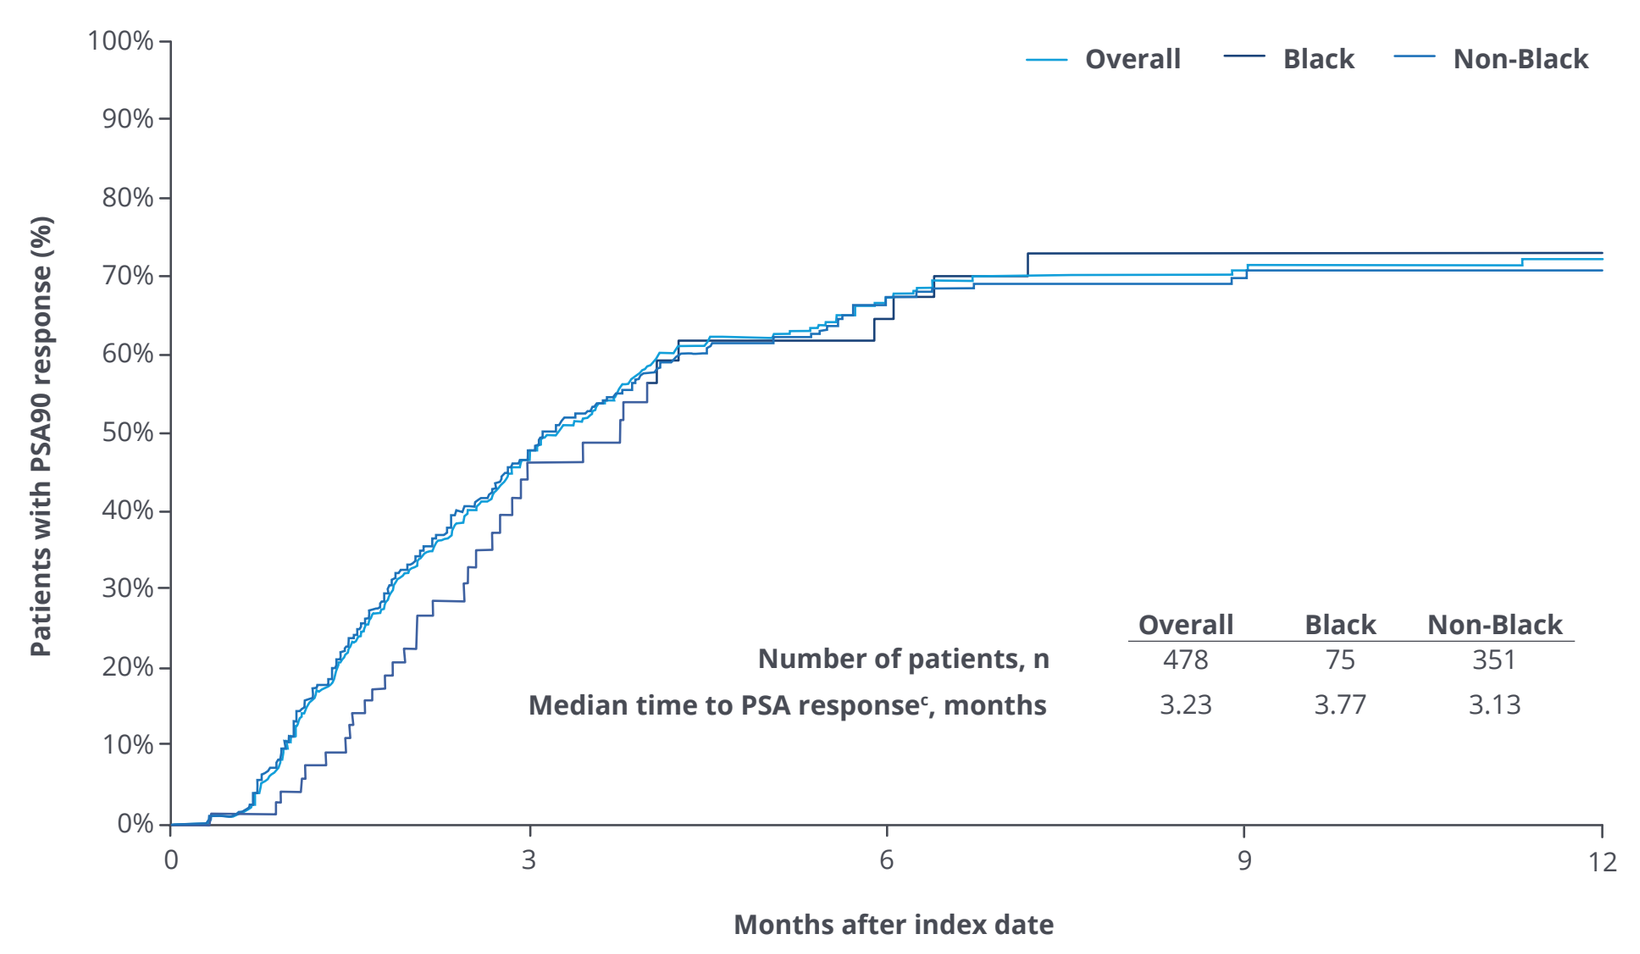
Overall
Black
Non-Black
Patients with PSA90 response (%)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0
3
6
9
12
Months after index date
Overall Black Non-Black
Number of patients, n 478 75 351
Median time to PSA response[c], months 3.23 3.77 3.13

	3 Months	6 Months	9 Months	12 Months
Number of patients at risk[d], n (%)				
Overall	173 (36.2)	82 (17.2)	49 (10.3)	34 (7.1)
Black	24 (32.0)	13 (17.3)	7 (9.3)	5 (6.7)
Non-Black	130 (37.0)	60 (17.1)	37 (10.5)	25 (7.1)
Kaplan-Meier rates				
Overall	48.0%	67.4%	71.0%	72.3%
Black	46.4%	64.7%	73.1%	73.1%
Non-Black	47.8%	67.5%	70.0%	70.8%

ARSI: androgen receptor signaling inhibitor; PSA: prostate-specific antigen.
Notes:
a. Patients with an unknown race were included in the overall population.
b. Among patients with baseline PSA within 13 weeks prior to and including index date.
c. PSA response was defined as the first decline for a follow-up PSA value of 90% or more from the closest PSA to the index date in the 12-month baseline period (including index date).
d. Refers to the population at risk of having the event at that point in time (i.e., patients who have not had the event and have not been lost to follow-up or censored). The observation period was defined as the time from the index date to the earliest of index treatment discontinuation (using a 90-day treatment gap to define discontinuation), initiation of a new ARSI or radiopharmaceuticals, end of clinical activity (including death), or end of data availability (1 April 2022).

LIMITATIONS

- This study relied upon clinical data that may contain inaccuracies or omissions (e.g., diagnosis dates, treatment start dates) and does not capture any diagnoses, medical services, or prescription fills obtained outside of the network of urology practices
- Miscoding or misclassification in the clinical record may introduce selection and information biases in terms of identifying the castration-sensitive population
- This study was limited to patients who received medications through in-office dispensations, and thus may not be representative of patients receiving external dispensations or patients from urology practices without in-office dispensations
- The database represents the community urology perspective and may not be representative of the entire population of patients with mCSPC in the US, which may limit the generalizability of the results to other settings

KEY TAKEAWAY

This real-world study of patients with mCSPC initiating APA in a urology setting observed high treatment persistence and robust PSA response rates overall and among both Black and non-Black patients

CONCLUSIONS

This real-world study demonstrated high APA treatment persistence for both Black and non-Black patients with mCSPC, with non-persistence (>90 day treatment gap) at 12 months observed in fewer than 6% of both Black and non-Black patients, suggesting that, once started, few patients discontinued APA treatment over 12 months

This study shows that, in the real-world setting, the majority of Black and non-Black patients with mCSPC treated with APA achieved rapid, deep PSA response by 6 months after starting treatment[9]

ACKNOWLEDGEMENTS

This study was sponsored by Janssen Scientific Affairs, LLC.

DISCLOSURES

B. Lowentritt is an employee of Chesapeake Urology Associates and has received consulting fees from Janssen Scientific Affairs, LLC. S. Du, E. Muser, and I. Khilfeh are employees of Janssen Scientific Affairs, LLC and stockholders of Johnson & Johnson. C. Rossi, F. Kinkead, B. Moore, P. Lefebvre, and D. Pilon are employees of Analysis Group, Inc., a consulting company that has provided paid consulting services to Janssen Scientific Affairs, LLC.

REFERENCES:

1. Chi KN, et al. *N Engl J Med.* 2019;381(1):13-24. **2.** Chi KN, et al. *J Clin Oncol.* 2021;39(20):2294-2303. **3.** FDA approves apalutamide for metastatic castration-sensitive prostate cancer. 2019; https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-apalutamide-metastatic-castration-sensitive-prostate-cancer. **4.** Saad F, et al. *Eur Urol.* 2022;81(2):184-192. **5.** Chi KN, et al. Prostate-Specific Antigen Kinetics in Patients With Advanced Prostate Cancer Treated With Apalutamide: Results from the TITAN and SPARTAN Studies. In: Poster presented at AUA 2021 Meeting, September 11, 2021. **6.** Matsubara N, et al. *Eur Urol.* 2020;77(4): 494-500. **7.** Fizazi K, et al. Darolutamide (DARO) tolerability from extended follow up and treatment response in the phase 3 ARAMIS trial. Poster presented at ASCO21 Virtual Scientific Program, June 4-8, 2021. **8.** Chowdhury-Paulino IM, et al. *Prostate Cancer Prostatic Dis.* 2022;25(3):397-402. **9.** Bivins VM, et al. *Future Oncol.* 2022;18(32):3595-3607. **10.** Freedland SJ, et al. *Curr Med Res Opin.* 2021;37(4):609-622.

Poster presented at ISPOR 2023; May 7–10, 2023; Boston, MA & online.