HPR157

COLLEGE OF PHARMACY
UNIVERSITY OF UTAH
L. S. SKAGGS PHARMACY INSTITUTE

Payer Perspectives on Health Insurance Coverage for Digital Therapeutics in the United States

Liang X[1], Gómez-Lumbreras A[1], Schippers S[1], Bhattacharjee S[2], Waters H[2], Hurwitz JT[3], Phillips K[1], Malone DC[1]
[1]Department of Pharmacotherapy, College of Pharmacy, University of Utah, Salt Lake City, UT, USA
[2]Otsuka Pharmaceutical Development & Commercialization Inc., Princeton, NJ, USA
[3]Center for Health Outcomes and Pharmacoeconomic Research, University of Arizona, Tucson, AZ, USA

Otsuka

Background

- Over the past several years Digital Therapeutics (DTx) have been marketed in the United States
- Because of the unique nature of DTx, there is considerable uncertainty around health insurance coverage of DTx applications (apps)

Objective

- To evaluate factors affecting health insurance coverage and reimbursement of DTx by United States payers and coverage decision-makers

Methods

- Cross-sectional qualitative study
- Online questionnaire and focus group sessions from August to October 2022
- Participants: managed care pharmacy and medical directors, health benefit consultants, and pharmacy benefit managers were recruited by snowball and convenience sampling
- Participants completed a pre-focus group questionnaire on their opinions on DTx products and their coverage
- The focus group evaluated the following topics: (1) evidence to evaluate DTx, (2) circumstances/attributes needed for DTx coverage, (3) barriers to DTx coverage, (4) coding issues and other factors influencing coverage as pharmacy or medical benefit, (5) benefit designs of DTx coverage, (6) indication and duration for DTx coverage, (7) reimbursement issues of DTx, (8) Food and Drug Administration (FDA) approval/policies associated with DTx
- Recorded transcriptions were analyzed by Atlas.ti software to determine frequency of topics mentioned by participants

Results

Table 1. Participants Characteristics and Perceptions of DTx

Participants Characteristics (N=21)	N (%) / Mean (SD)
Sex (Female)	6 (29)
Prefer not to answer	1(5)
Age (years) ≥ 50	16 (76)
Discipline	
Pharmacist	18 (86)
Physician	3 (14)
Position title	
Clinical pharmacist	3 (14)
Consultant	4 (19)
Medical leadership	3 (14)
Pharmacy leadership	11 (53)
Years of licensed professional	29.1 (10.0)
Years of health benefits experience	
≤15	3 (14)
>15	18 (86)
Experience with DTx products/apps [ranked none (0) to a lot (100)]	59.7 (29.6)
Organization	
Managed Care Organization	11 (52)
Health Benefit Consultant	4 (19)
PBM	3 (14)
HMO/Health Plan	2 (10)
Pharmacy Services Organization	1 (5)
Total members covered by health plan	8,635,632 (17,502,855)
Areas served by organization (can be more than one)	
Northeast	12 (57)
Midwest	10 (48)
West	14 (67)
South	10 (48)
Perceptions of Evidence and Current Coverage of DTx	
Clinical trial needed for evidence	19 (90)
Require FDA evaluation for a DTx to be covered	15 (71)
DTx currently covered by their organization	6 (29)

Figure 1. Participants' Opinions on Under What Benefit DTx Should be Reimbursed

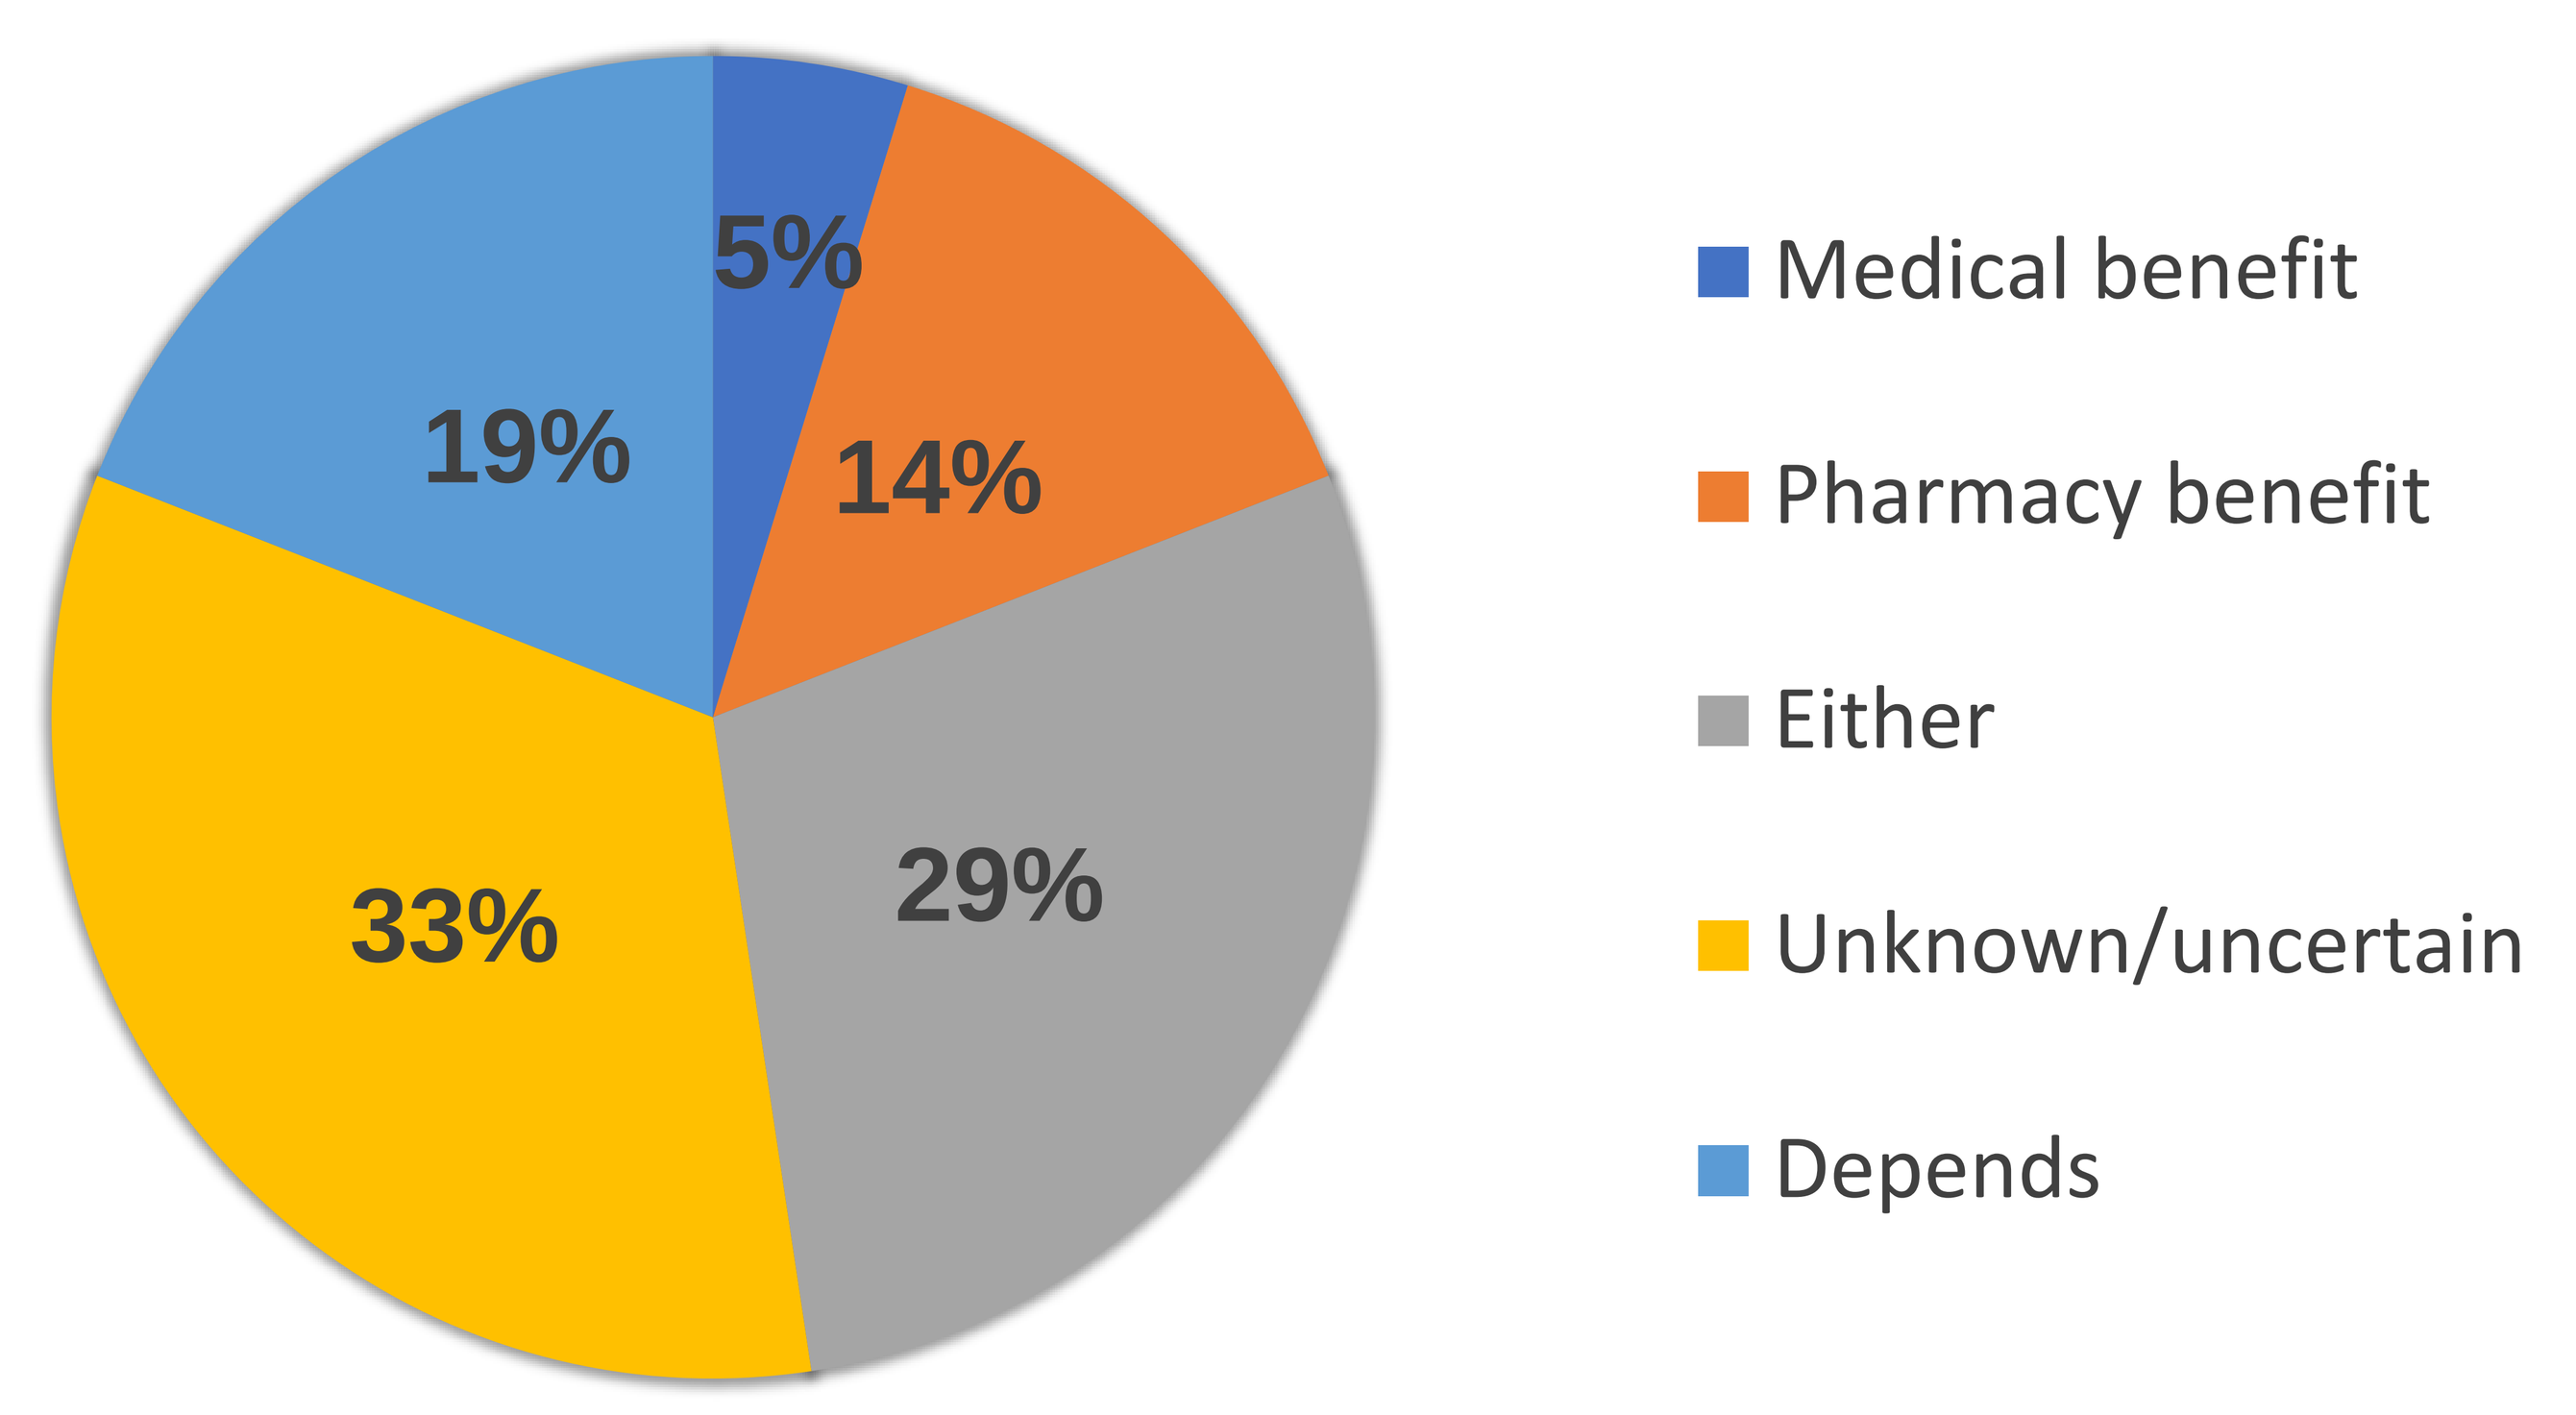
5%
14%
29%
33%
19%
Medical benefit
Pharmacy benefit
Either
Unknown/uncertain
Depends

Figure 2. Topics Mentioned by Participants in Focus Groups

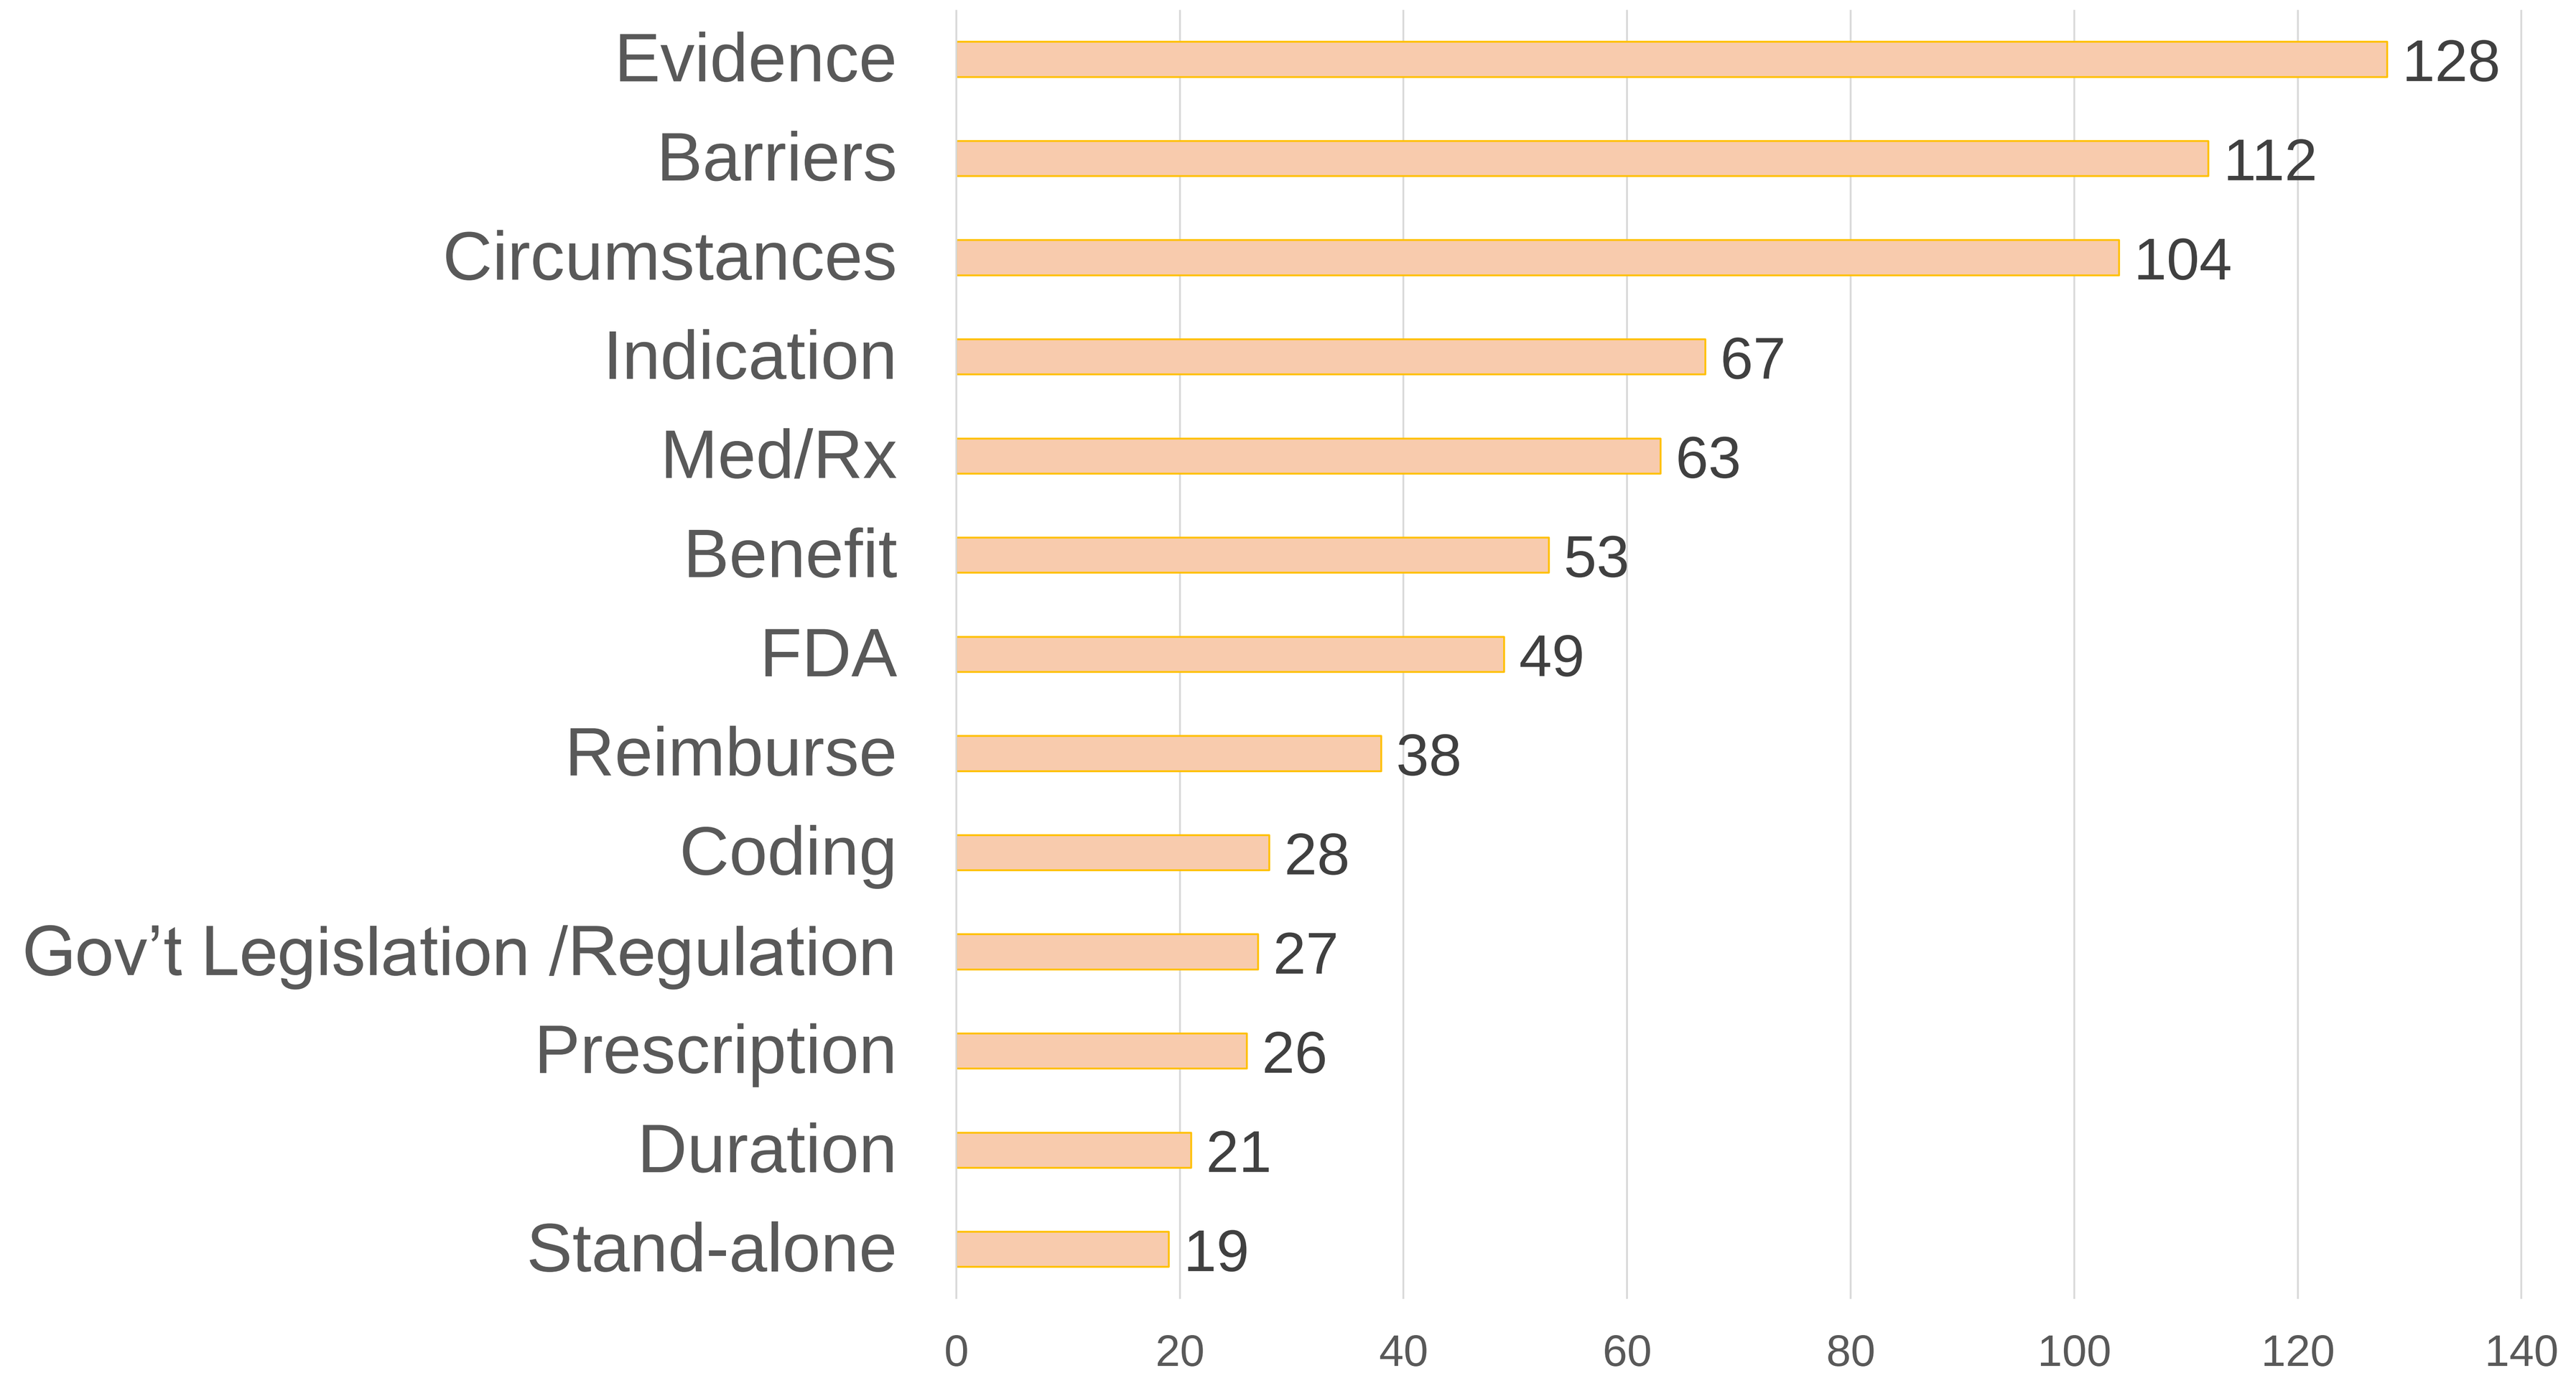
Evidence 128
Barriers 112
Circumstances 104
Indication 67
Med/Rx 63
Benefit 53
FDA 49
Reimburse 38
Coding 28
Gov't Legislation /Regulation 27
Prescription 26
Duration 21
Stand-alone 19
0
20
40
60
80
100
120
140

Med/Rx: Medical or Pharmacy Benefit

Figure 3. Word Cloud from Focus Group Transcripts

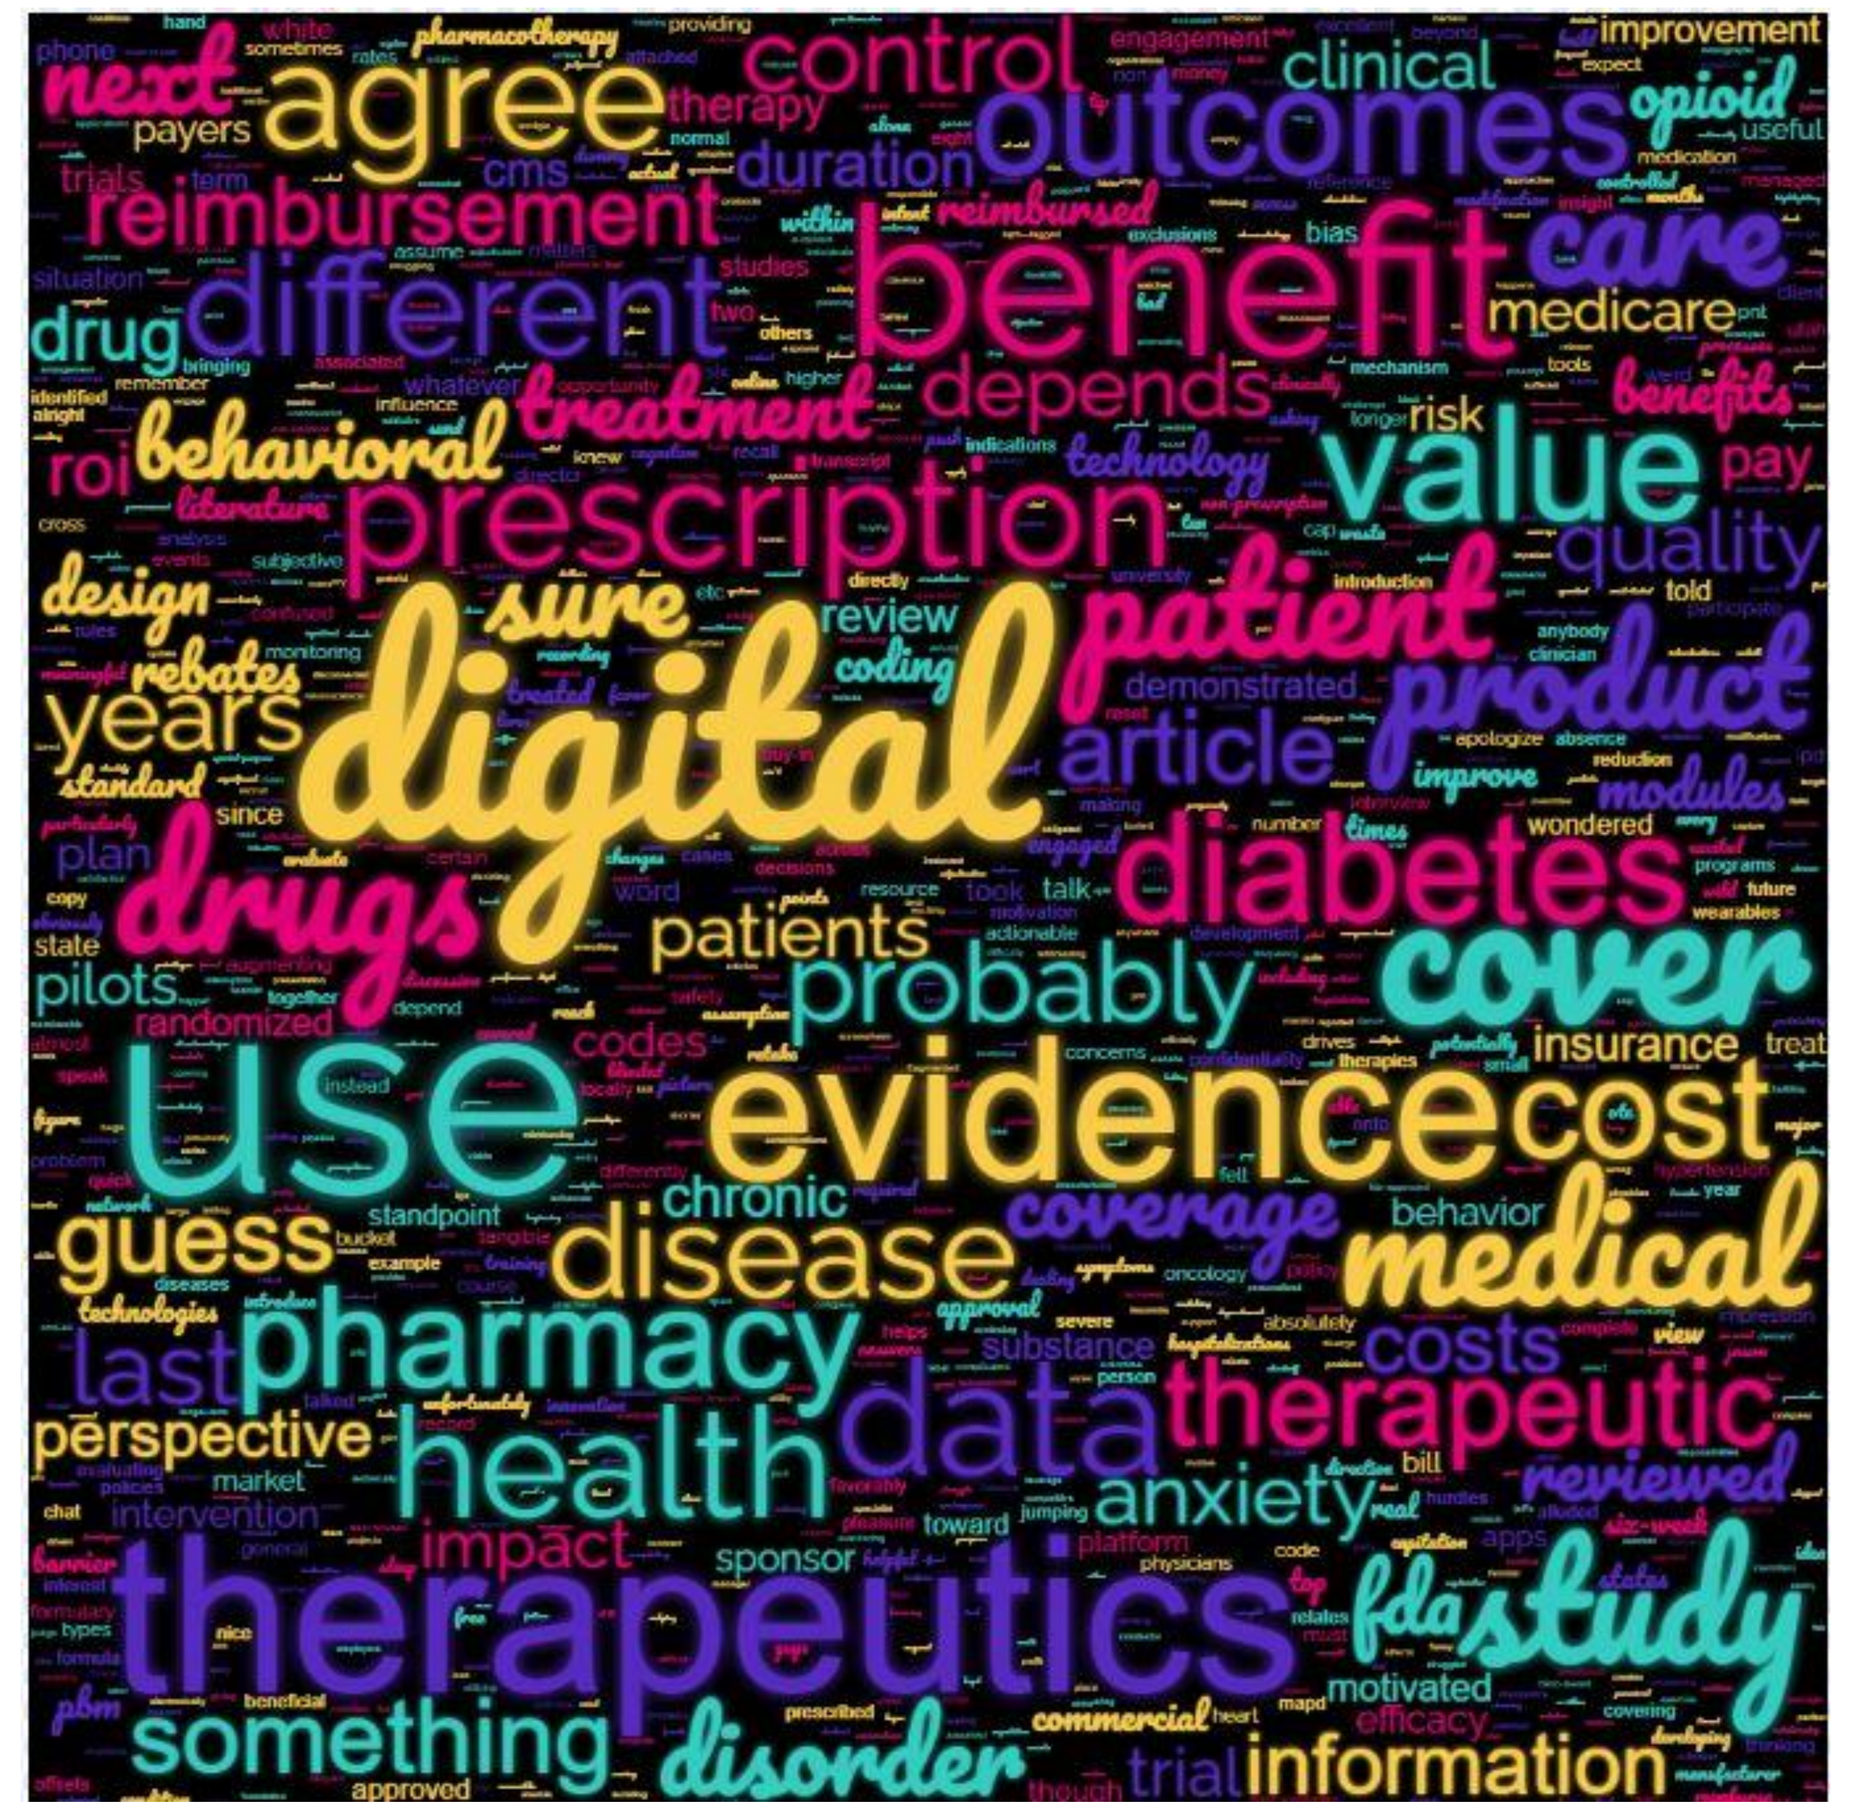
digital
therapeutics
evidence
benefit
use
outcomes
agree
different
reimbursement
prescription
value
patient
product
drugs
diabetes
cover
probably
medical
cost
disease
pharmacy
health
data
therapeutic
study
care
treatment
behavioral
years
guess
something
disorder
information
control
clinical
depends
article
coverage
anxiety
impact

Quotes

- **Evidence**: "…*with the limited published evidence that's out there, we're not seeing long-term value proposition and long-term effectiveness or cost-effectiveness assessments…"*
- **Gov't Legislation /Regulation:** *"I think Medicare covering it would probably, you know, urge payer to be able to covered under the commercial plan"*
- **Pharmacy/medical benefits**: *"It just depends on the disease"*
- **Barriers**: *"In what populations? How long? Do they use technology? What's the durability or the stickiness of the effect and to gain insights from pilots before we figure out? Is there an ROI and how do we deploy it?"*

Conclusions

Health insurance coverage of DTx is not yet common, but most participants believe that:

- FDA review/approval will be required by most organizations to include DTx as a covered benefit
- Payers expect high-quality evidence as a condition of coverage
- Prescription apps (circumstances) were preferred over non-prescription apps from a coverage perspective
- Benefit policies will need to be modified to accommodate DTx products

For more information or a reprint of this poster, please contact Ainhoa G Lumbreras. Email: Ainhoa.Gomez-Lumbreras@utah.edu

Funding: Otsuka Pharmaceutical Development & Commercialization, Inc.

Poster presented at ISPOR Annual Meeting, May 7-10, 2023, Boston, MA