Evolution of Market Dynamics Within Immunology After Biosimilar Introduction Within Europe

OP1

Xin Q[1], Keady S[1], Bodin M[1]

[1]Biogen International GmbH, Baar, Switzerland

Objectives

This study aims to understand the European evolution of market dynamics following the introduction of biosimilars within the immunology area.

Methods

The report investigated 19 biologic molecules and 4 Janus kinase (JAK) inhibitors with approved immunology indications. Consumption was based on IQVIA MIDAS sales data (2017 to 2021). This timeframe reflected the first significant uptake of biosimilars in the immunology space in 2017 and incorporated the full effect of biosimilar uptake in 2021. Molecule usage was measured in treatment days, derived from volume of standard units sold divided by average daily doses. To enable the comparison of molecule uptake across countries, a per-capita indicator treatment rate was used (dividing yearly total treatment days per molecule by population size). All average treatment rates used in this report are population weighted.

Results

Within immunology, there was significant increase in usage of biologics across all investigated European countries. Average consumption of anti-TNF molecules increased by 35% from 0.51 treatment days/capita in 2017 to 0.68 treatment days/capita in 2021 (Table 1). Whilst the use of reference molecules has decreased by 31%, biosimilar use has increased by 346% from 0.09 treatment days/capita in 2017 to 0.39 treatment days/capita in 2021. Substantial growth in consumption of other mechanism of actions (MOAs) were also identified (Table 1). Usage of interleukin inhibitors and JAK inhibitors have increased by 168% and 1,906% respectively from 2017 to 2021.

Despite the increase in treatment rates seen across every single investigated market, there is a wide gap in molecule usage between countries (Figure 1). In 2021, there was a 16-fold difference in treatment days per capita between the countries with the highest and lowest treatment rates. Ireland, Norway, and Sweden had an average of 2.22 treatment days/capita of all investigated molecules, whereas Belarus, Bosnia-Herzegovina, and Poland had an average of 0.14 treatment days/capita (Table 2).

Biosimilar penetration is not always positively correlated with anti-TNF utilization (Figure 2). Poland has one of the highest biosimilar penetrations (above 70% in 2021). However, the usage of anti-TNF molecules is still relatively low. On the contrary, Switzerland and Belgium have relatively high anti-TNF utilization, whereas the biosimilar penetration is rather low (21% and 30% respectively).

Table 1. Evolution of molecule usage from 2017 to 2021 by Mechanism of Action (MOA)

	2017	2021	Change in %
Average treatment rate (all MOA)	0.66	1.10	66%
Average treatment rate (anti-TNF total)	0.51	0.68	35%
Average treatment rate (anti-TNF biosimilar)	0.09	0.39	346%
Average treatment rate (anti-TNF reference molecule)	0.42	0.29	-31%
Average treatment rate (interleukin inhibitor)	0.11	0.29	168%
Average treatment rate (JAK inhibitor)	0.00	0.07	1906%

Numbers may not add up due to rounding

Table 2. Differences in molecule utilization between countries

	2017	2021
Sweden, Norway, Ireland: Total treatment days	27,823,479.66	46,348,809.58
Total population	20,142,054	20,852,361
Population weighted average treatment days	1.38	2.22
Belarus, Bosnia-Herzegovina, Poland: Total treatment days	3,298,822.95	6,999,208.98
Total population	50,785,349	50,384,797
Population weighted average treatment days	0.06	0.14
Difference	21.27	16.00

Numbers may not add up due to rounding

Figure 1. Evolution of molecule usage in immunology across Europe from 2017 to 2021

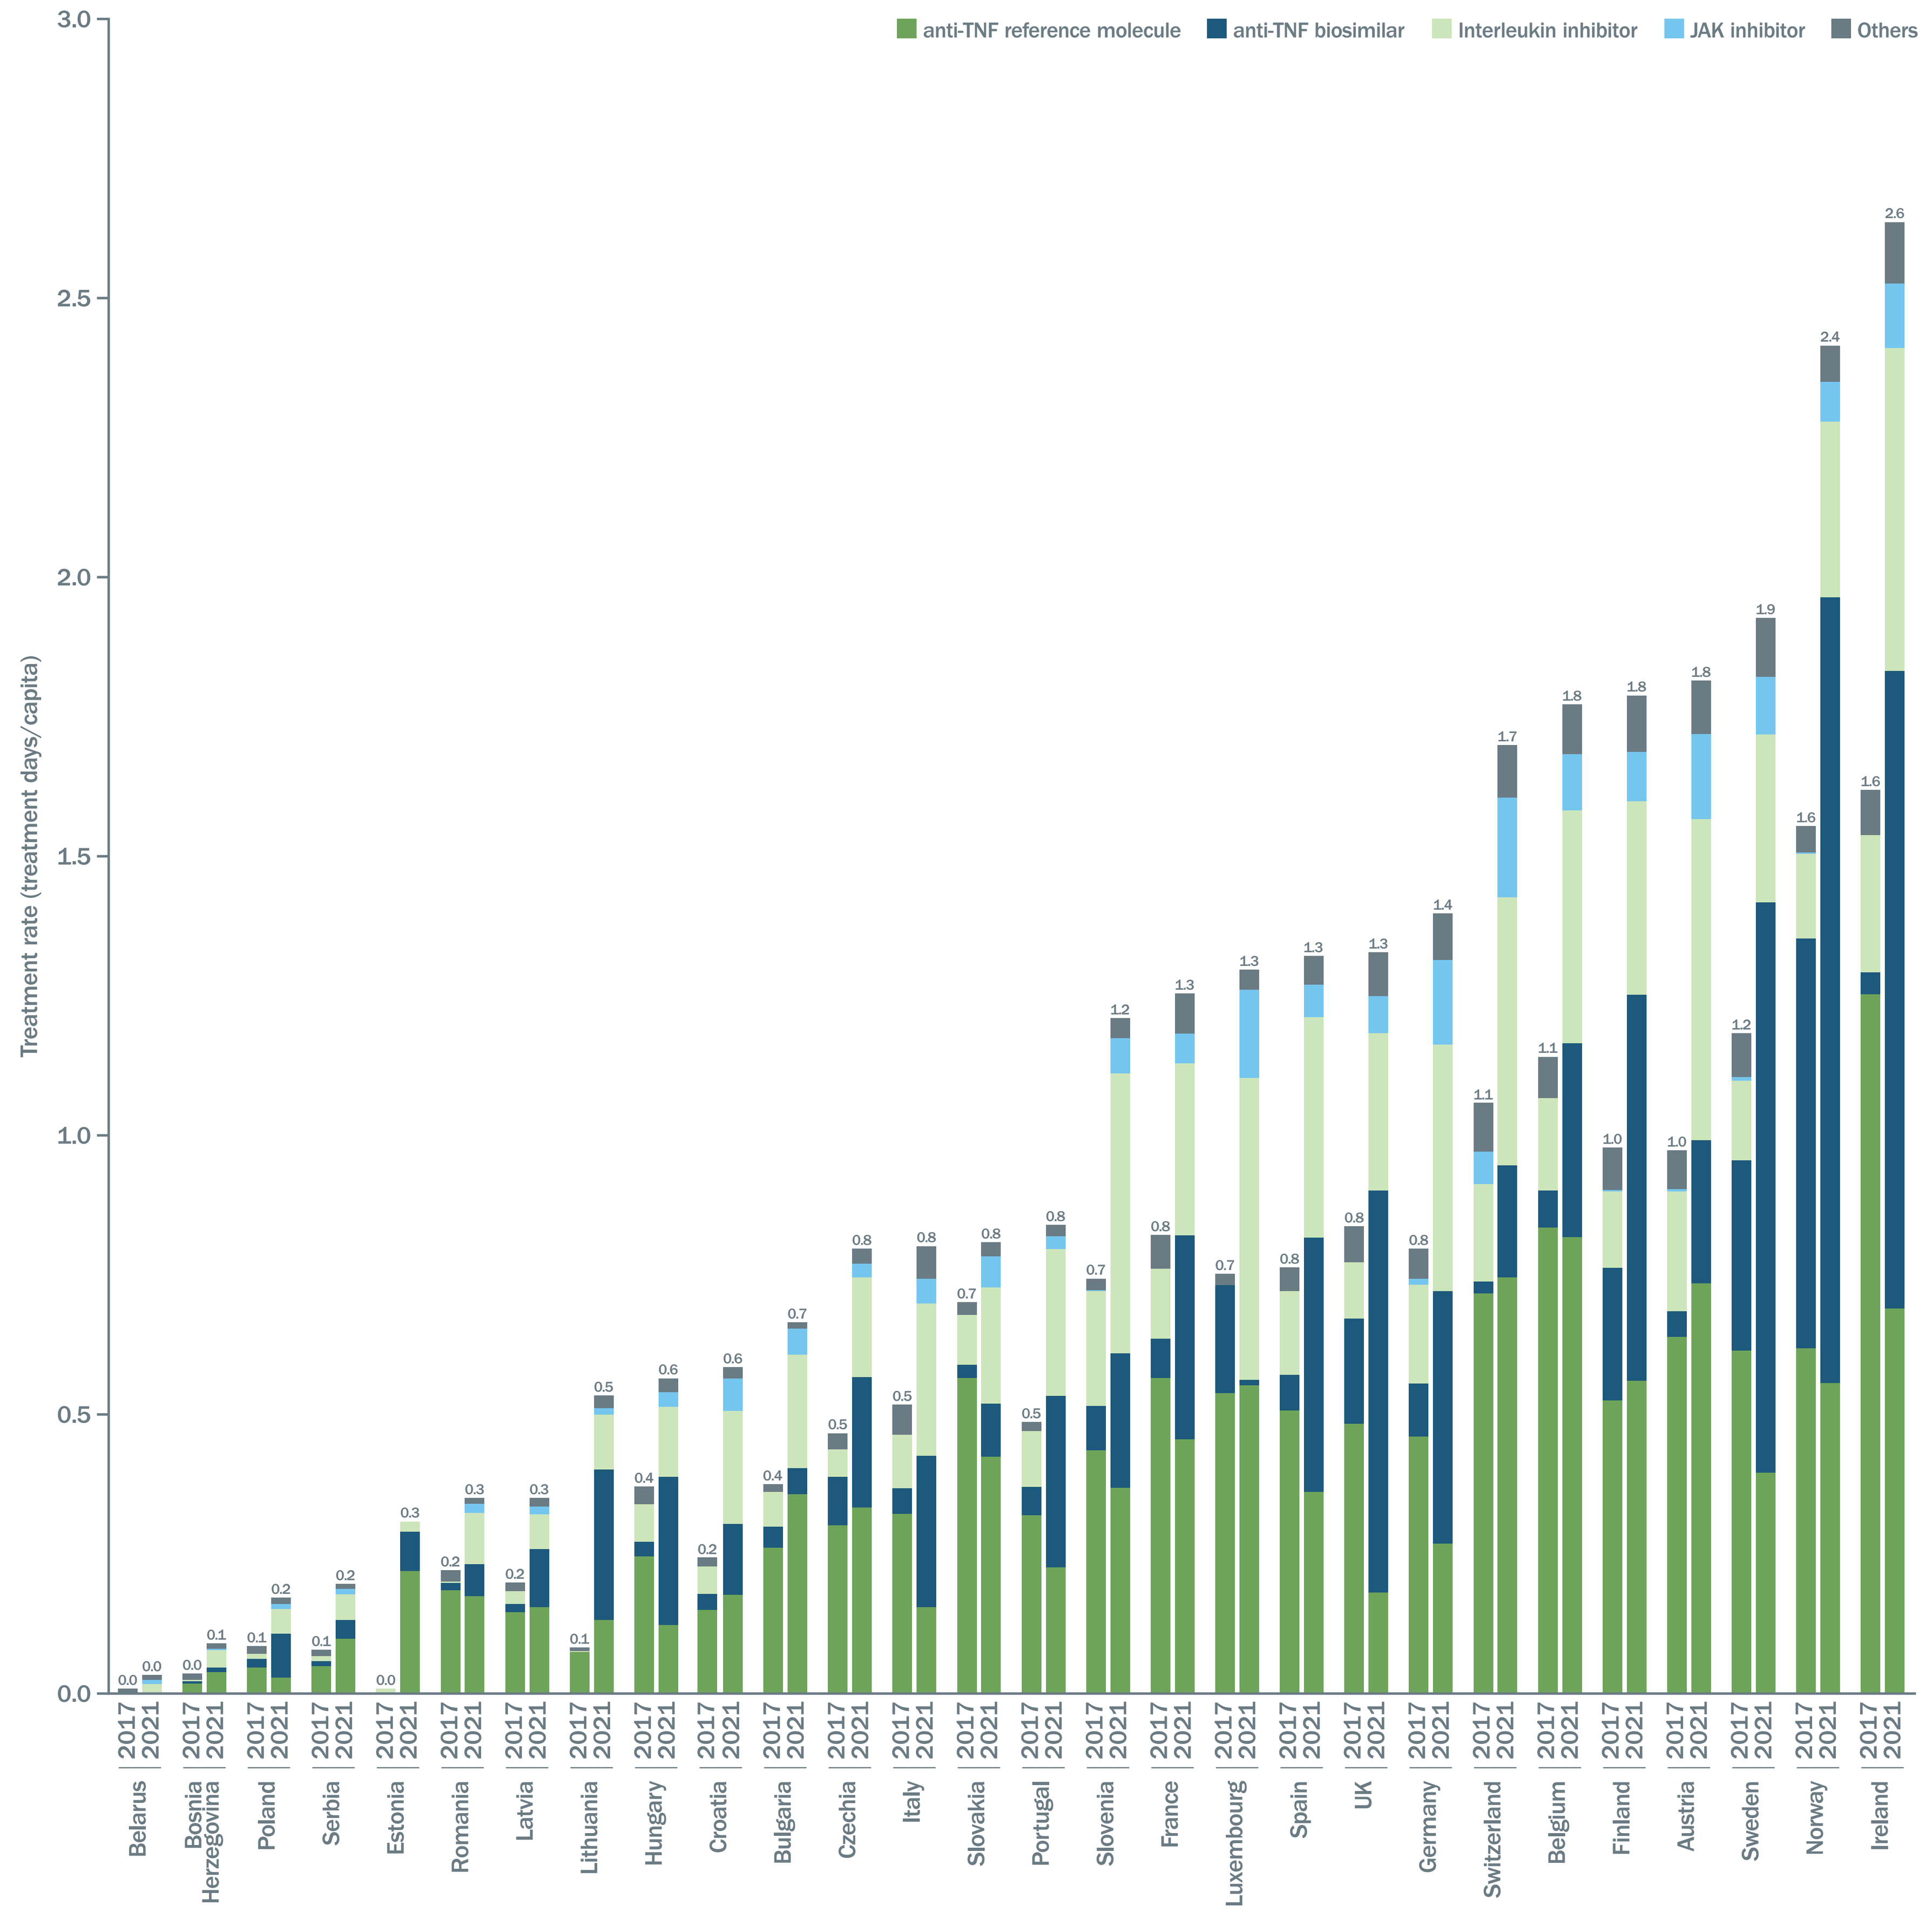
anti-TNF reference molecule
anti-TNF biosimilar
Interleukin inhibitor
JAK inhibitor
Others
Treatment rate (treatment days/capita)
Belarus, Bosnia Herzegovina, Poland, Serbia, Estonia, Romania, Latvia, Lithuania, Hungary, Croatia, Bulgaria, Czechia, Italy, Slovakia, Portugal, Slovenia, France, Luxembourg, Spain, UK, Germany, Switzerland, Belgium, Finland, Austria, Sweden, Norway, Ireland

Numbers may not add up due to rounding. Market data: IQVIA MIDAS. Population data: The World Bank (www.worldbank.org).
Indications included: rheumatoid arthritis, Crohn's disease, ulcerative colitis, psoriasis, psoriatic arthritis, hidradenitis suppurativa, and ankylosing spondylitis.

- Anti-TNFs: adalimumab, certolizumab pegol, etanercept, golimumab, infliximab
- Interleukin inhibitors: tocilizumab, sarilumab, anakinra, ustekinumab, secukinumab, ixekizumab, brodalumab, bimekizumab, guselkumab, tildrakizumab, risankizumab
- JAK inhibitors: tofacitinib, upadacitinib, baricitinib, filgotinib
- Others: anti-integrin agent (vedolizumab), CD 80/86 co-stimulation inhibitor (abatacept), CD20-depleting antibody (rituximab)

Figure 2. Biosimilar penetration in 2021

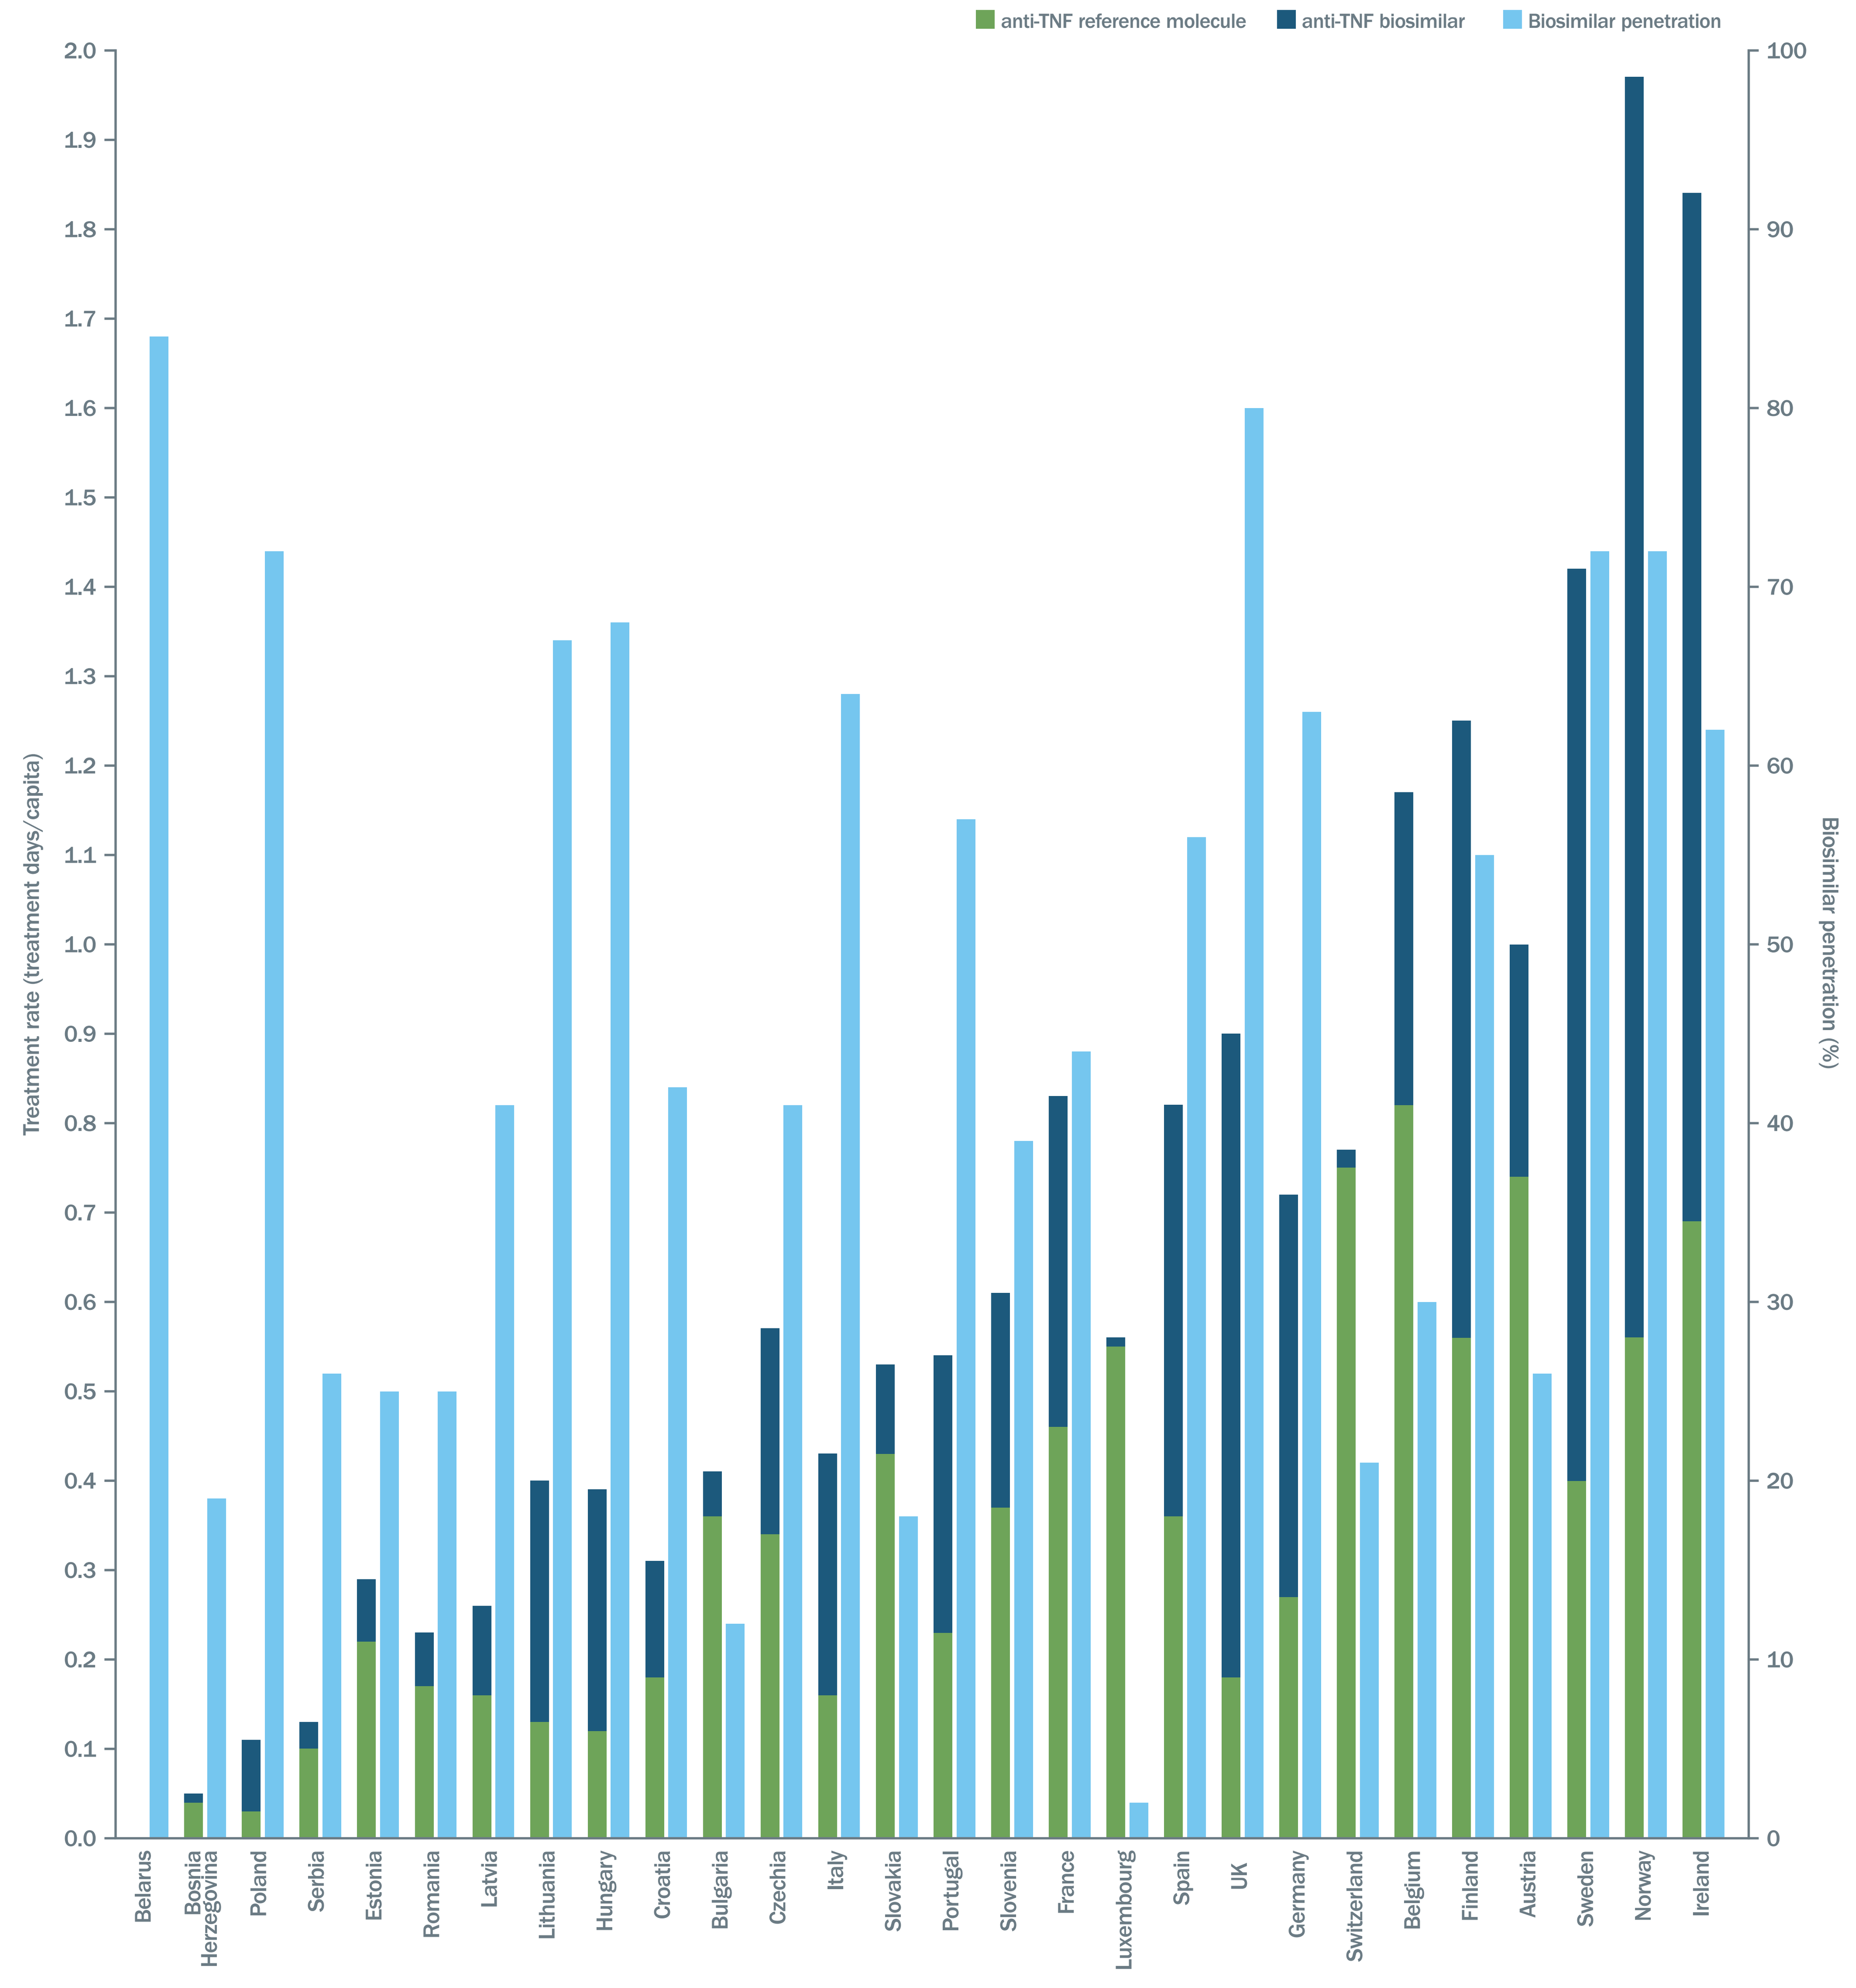
anti-TNF reference molecule
anti-TNF biosimilar
Biosimilar penetration
Treatment rate (treatment days/capita)
Biosimilar penetration (%)
Belarus, Bosnia Herzegovina, Poland, Serbia, Estonia, Romania, Latvia, Lithuania, Hungary, Croatia, Bulgaria, Czechia, Italy, Slovakia, Portugal, Slovenia, France, Luxembourg, Spain, UK, Germany, Switzerland, Belgium, Finland, Austria, Sweden, Norway, Ireland

Numbers may not add up due to rounding

Conclusions

The use of approved immunological therapies has increased significantly since the introduction of biosimilars within Europe. However, there remains a substantial inequality both in treatment rates and biosimilar penetration, primarily between Eastern and Western countries.

To mitigate inequalities, greater understanding of the combination of robust policies, risks and benefits of regionalized/centralized procurement models and promotion of sustainable pricing and reimbursement is required.

Disclosures of interest Q Xin, S Keady, and M Bodin are employees of Biogen. S Keady and M Bodin hold stock in Biogen.
Acknowledgments: Biogen International GmbH funded this study. Authors had full editorial control and provided final approval for all content. Editorial support for the preparation of this poster was provided by Springer Healthcare Ltd.

ISPOR 2023 | MAY 7–10 | Boston