Poster SA19

Kejal Jadhav
kejal.jadhav@novartis.com

Real-World Targeted Literature Review of Intolerance with Tyrosine Kinase Inhibitor Treatment for Chronic Myeloid Leukemia

Kejal Jadhav, BPharm, MS[1], Andrea Damon, PhD, CMPP[1], Daisy Yang, PharmD, BCOP[1], Dominick Latremouille-Viau, MSc[2], Dongyang Yang, PhD[2], Annie Guérin, MSc[2]

[1]Novartis Pharmaceuticals Corporation, One Health Plaza, East Hanover, New Jersey, 07936, United States;
[2]Analysis Group, Inc., 1190 avenue des Canadiens-de-Montréal, Montréal, Québec, H3C 1B3, Canada

Scan to obtain:
- Poster

https://www.medicalcongressposters.com/Default.aspx?doc=c5db1

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

CONCLUSIONS

- This targeted literature review suggests that intolerance to TKIs in CML is associated with unmet needs; intolerance was an important factor contributing to the clinical, economic, and humanistic burden of the disease
- Patient perspectives on prevention and management of intolerance in CML care is critical for making more informed treatment decisions and to evaluate clinical benefits of CML treatment strategies
- Publications related to intolerance reviewed in this study were mostly from studies conducted outside United States (US), and mostly reported findings regardless of line of therapies or severity grades, warranting further research in this field
- The heterogeneity of treatment intolerance definitions reflects the challenges in classifying intolerance in clinical practice and points to the need for consistency in definitions and clear guidelines in the US to help inform treatment decisions

This study was sponsored by Novartis Pharmaceuticals Corporation.

Poster presented at the 2023 Meeting of ISPOR – The Society for Health Economics and Outcomes Research held on 08–10 May 2023 in Boston, Massachusetts, USA.

BACKGROUND

- The outcomes of patients with chronic myeloid leukemia (CML) have significantly improved with tyrosine kinase inhibitors (TKIs) and standards for the management of CML have therefore moved from short-term survival to long-term outcomes and the achievement of early disease control
- New challenges appear in the management of CML and one of the clinical concerns is intolerance experienced by patients during TKI treatment
- Although certain adverse effects are common to most TKIs, they occur at varying frequencies depending on the TKI
- Treatment intolerance leads to poorer disease control

OBJECTIVE

- This targeted literature review examined the association of intolerance experienced by CML patients during TKI treatment and adherence, clinical, humanistic and economic outcomes in real-world setting

METHODS

- The targeted literature review identified records between January 2010 to October 2022
- Searches were conducted in PubMed using a combination of terms to identify association of TKI intolerance on different scopes
- A total of 225 unique articles were identified and screened based on title and abstract, 75 full-text articles were further assessed; 45 articles were included in this review for data extraction (**Figure 1**)

Figure 1. Flow diagram of articles inclusion

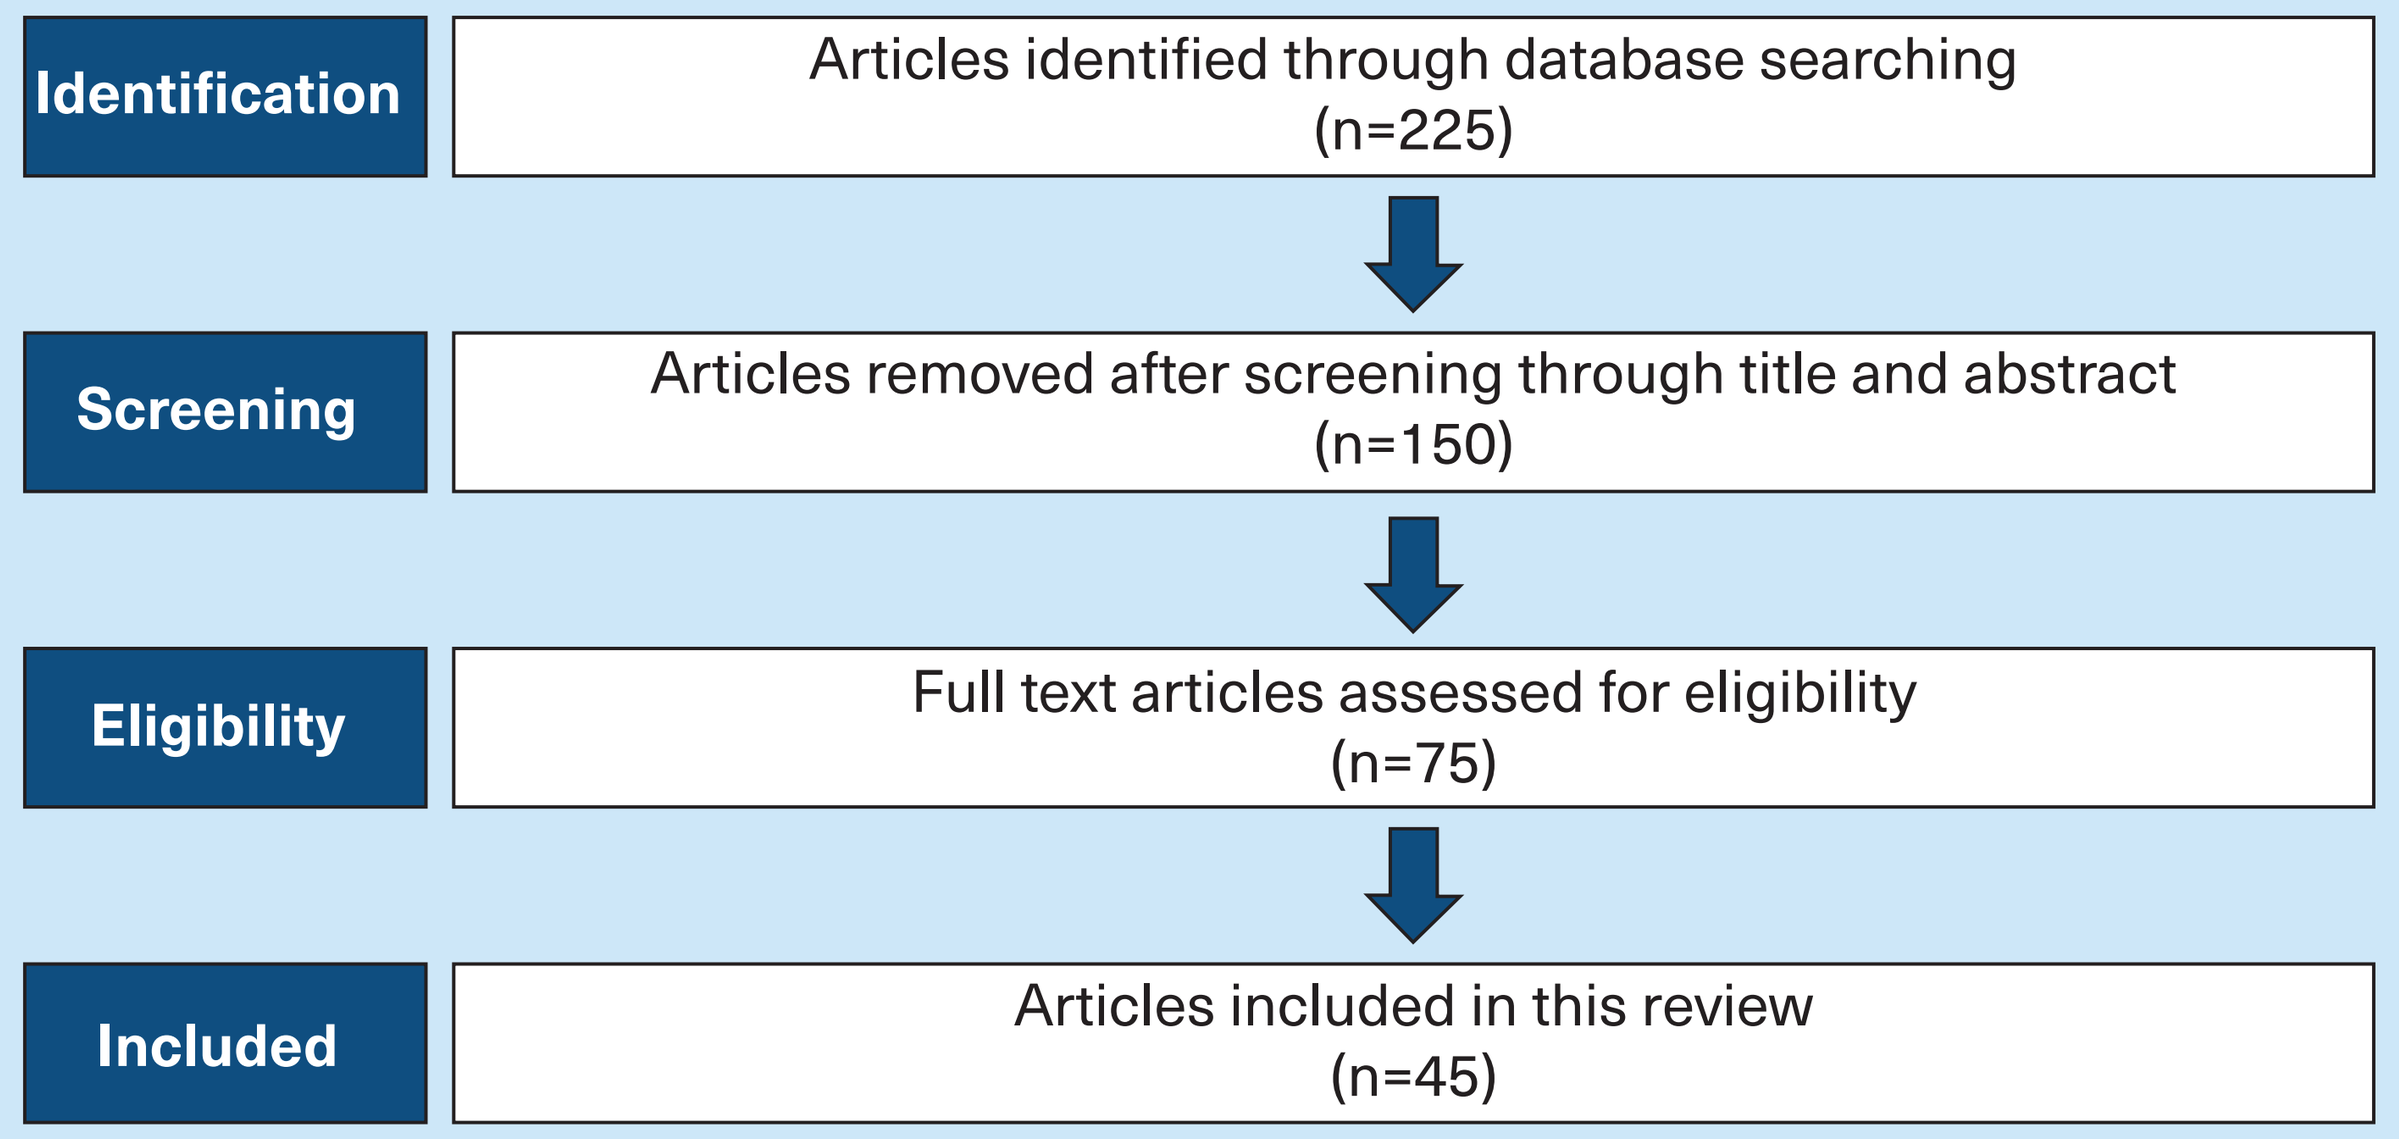
Identification
Articles identified through database searching (n=225)
Screening
Articles removed after screening through title and abstract (n=150)
Eligibility
Full text articles assessed for eligibility (n=75)
Included
Articles included in this review (n=45)

Note: This method using web-based search engine is not fully reproducible

RESULTS

Definitions of intolerance in a real-world setting

- Most of the publications included patient- or physician-reported intolerance without following specific guidelines or reporting clear definition of intolerance
- Only 3 out of 45 publications used Common Terminology Criteria for Adverse Events (CTCAE, version 4.0 and 5.0), 1 out of 45 used Functional Assessment of Chronic Illness Therapy (FACIT)-Fatigue scale, 3 out of 45 used WHO scale, and 1 out of 45 used the International Conference on Harmonisation Guidelines (ICH Steering Committee, 2003) to define intolerance

Intolerance and adherence

Study	Key Findings
Eliasson et al., 2011	Patients' reasons for not taking their medication as prescribed were to minimize side effects, such as nausea, tiredness and lack of energy
Chen et al., 2014	Adverse TKI effects and associated polypharmacy were the key concerns that impact on patients' adherence and alter their medicine-taking approach to maintaining long-term use of TKI. Adverse effects were perceived to be the key concern of treatment • Some patients perceived the incidence of side effect as an indicator for disease progression • Patients often mistook the abnormal laboratory results or adverse effects for occurrences of new diseases before they were confirmed by physicians • Some participants complained about the lack of relevant information to monitor the adverse effects
Kekäle et al., 2015	Medication-taking approach was reported to be negatively impacted by adverse drug reactions (ADRs). Patients had stopped taking the medication or reduced the dose when feeling worse
Al-Dewik et al., 2016	Adherent patients were significantly more likely to achieve optimal response compared to the nonadherent group • Adherent patients were significantly more likely to obtain optimal response (95%) compared to the nonadherent group (14.3%) • Half of the patient's sample developed one or more side effects such as fatigue, nausea, vomiting, headache, muscle pain, abdominal pain, skin rash/itching, orbital edema, leg edema, memory change, sunburn, weight gain, or infection
Unnikrishnan et al., 2016	Intentional nonadherence occurred in 43%-52% of the patients. CML-associated symptom scores were higher in nonadherent patients

Intolerance and humanistic outcomes

Study	Key Findings
Efficace et al., 2013	Chronic fatigue is the main factor limiting the quality of life (QoL) of CML patients who receive long-term TKI therapy • The magnitude of the QoL differences between patients with high versus those with low fatigue levels was very large • Fatigue greatly impacted on the extent to which the physical health and the emotional problems limited the work or other usual activities of patients in terms of time and performance • Higher levels of fatigue were more common in patients who reported having had job problems
Kekäle et al., 2015	TKI-related ADRs were common among CML patients irrespective of patient's adherence level. Patients who reported that ADRs had a negative influence on their daily QoL perceived more ADRs than those who did not experience a negative influence
Unnikrishnan et al., 2016	QoL scores, particularly the symptom scores associated with side effects, was significantly different between adherent and nonadherent patients • The proportion of nonadherence was 55%. • Multivariate analysis revealed global health status as the sole predictor of adherent behavior • Low QoL was associated with nonadherence to TKI in patients with CML
Unnikrishnan et al., 2017	Significant impairment of QoL was noted among patients with CML primarily due to the burden of symptom related to side effects
De Barros et al., 2019	A majority of CML patients faced professional consequences of their disease and treatments. ADRs were a major factor affecting the occurrence of work modifications, a change in the number of working hours was the most represented
Janssen et al., 2021	CML patients with severely TKI-induced fatigue were less physically active during the day, with regard to both light and moderate to vigorous intensity activity, on both week and weekend days: • There is a subset of CML patients particularly prone to TKI-induced fatigue • Severely fatigued patients have reduced levels of physical activity • Fatigued CML patients have impaired QoL and work fewer hours when compared to non-fatigued patients

Intolerance and clinical outcomes

Study	Key Findings
Latagliata et al., 2013	Severe hematologic and extrahematologic toxicities were observed in elderly patients. Occurrence of grade 3–4 extrahematologic toxicity was associated with lower rates of complete cytogenetic response (CCyR) and molecular response as well as worse overall survival (OS)
Unnikrishnan et al., 2016	Adherence to TKI over a long period is important for optimal outcomes in CML • QoL scores, particularly the symptom scores associated with side effects, were significantly different between adherent and nonadherent patients. Increased long-term symptom burden due to side effects could contribute to nonadherence • A higher proportion of adherent patients achieved deeper molecular responses
Fox et al., 2017	The most common adverse event (AE) was pleural effusions (PE) in 25% of patients and recurrence of effusions was high. Management of effusions varied substantially between institutions reflecting a lack of a consensus approach. Dose reductions and temporary drug cessations had minimal impact on response rate and survival; maintenance of MR4.5 could not be correlated with prior duration of MR4.5 in patients with effusions
Boddu et al., 2018	All 19 patients with a diagnosis of CML in chronic phase discontinued ponatinib due to resistance (n=13), toxicity (n=5), and to pursue stem cell transplantation (n=1). All 17 patients with a diagnosis of CML in accelerated phase discontinued ponatinib due to resistance (n=15) and toxicity (n=2). Long-term outcome of patients with ponatinib failure are poor with estimated one-year OS of 54% and event-free survival (EFS) rates of 40%
Cerveira et al., 2018	The reasons for TKI discontinuation among those with unsustained deep molecular response (DMR) included intolerance, toxicity issues, and pulmonary tuberculosis. More than 80% of patients who discontinued therapy relapsed and the relapse occurred within the first six months. These patients promptly resumed TKI therapy and regained at least major molecular response (MMR)
Ma et al., 2021	Among the patients who switched due to intolerance, 88% were able to achieve or maintain MMR with 5-year progression-free survival (PFS) 90.5% (95% confidence interval [CI] 90.4–90.6 %)

Intolerance and treatment patterns

Study	Key Findings
Intolerance was reported as a primary/main reason for treatment modification	
Abruzzese et al., 2019; Breccia et al., 2022; Fox et al., 2017; Gambacorti-Passerini et al., 2021; García-Gutiérrez et al., 2019; Hehlmann et al., 2019; Michallet et al., 2010	The main reasons for TKI *discontinuation* included side effects or intolerance (44%-75% of discontinuations), followed by lack of efficacy, primary resistance and acquired resistance
Bettiol et al., 2018; Ma et al., 2021; Marquinez-Alonso et al., 2020; Gambacorti-Passerini et al., 2021; Hehlmann et al., 2019; Tokuhira et al., 2018	• Intolerance was the most common primary reason for switching (43%-79% of switches), followed by primary resistance and acquired resistance • Among patients who switched to another TKI therapy due to an inadequate cytogenetic and/or molecular response, 79% required a change in therapy due to intolerance
Sánchez-Guijo et al., 2011	• 33% of patients interrupted therapy during follow-up, with a median duration of interruptions of 28 days, mainly caused by hematologic toxicity, accounting for 67% of the interruptions
Marquinez-Alonso et al., 2020; Nekoohesh et al., 2020	*Dose decrease* was observed in patients mainly due to AEs (57%-64% of dose decreases)
Intolerance was the only reason explored for treatment modification	
Boddu et al., 2019; Breccia et al., 2014; Devos et al., 2019; Huguet et al., 2019; Latagliata et al., 2016; Latagliata et al., 2021; Stagno et al., 2021	• Discontinuation was observed due to AEs and toxicity (15%-67% of study sample) with most of the discontinuation occurring in the first 12 months • Discontinuations due to toxicity occurred in 44% of patients with Charlson Comorbidity Index (CCI) score 0 and in 60% of patients with score ≥ 1
Scalzulli et al., 2021	Due to intolerance to the therapy, patients were reported to *switch* to another TKI (17% of study sample)
Claudiani et al., 2021; Szeto et al., 2022	Due to intolerable AEs and toxicity, patients had a dose *reduction* (28%-83% of study sample)

Intolerance and economic outcomes

Study	Results/Findings
Li et al. 2016	CML patients incur a total cost of \$1,409,466-\$1,422,122. Of this total direct healthcare cost, 0.5%-0.6% was attributed to AE-related costs
Seiter et al. 2018	The mean infection-related costs among patients with an infection during first-line TKI therapy was \$13,334 per patient per year (PPPY) and the median was \$877. The mean infection-related costs among patients with an infection during second-line TKI therapy was \$23,777 PPPY and the median was \$1,140
Seiter et al. 2018	The mean PE-related cost among patients with PE during the first-line therapy was \$14,250 PPPY and the median was \$3,038

References

Abruzzese E et al. *Mediterr J Hematol Infect Dis.* 2019;11(1):e2019025. Al-Dewik NI et al. *Clin Med Insights Oncol.* 2016;10:95-104. Bettiol A et al. *Clin Drug Investig.* 2018 Sep;38(9):837-44. Boddu P et al. *Leuk Lymphoma.* 2018;59(6):1312-22. Boddu P et al. *Leuk Lymphoma.* 2019;60(13):3172-80. Bonifacio M et al. *Pharmacoecon Open.* 2022;6(1):95-104. Breccia M et al. *Leuk Res.* 2014;38(10):1173-6. Breccia M et al. *Br J Haematol.* 2022;198(6):965-73 Chen LC et al. *Int J Clin Pharm.* 2014;36(1):120-7. Cerveira N et al. *BMC Cancer.* 2018;18(1):1245. Claudiani S et al. *Br J Haematol.* 2021;193(2):346-55. Cony-Makhoul P et al. *Br J Haematol.* 2018;180(3):356-64. De Barros S et al. *Ann Hematol.* 2019;98(3):615-23. Devos T et al. *Acta Haematol.* 2019;142(4):197-207. Efficace F et al. *Leukemia.* 2013;27(7):1511-9. Eliasson L et al. *Leuk Res.* 2011;35(5):626-30. Fox LC et al. *Blood Adv.* 2017.15;1(13):802-11. Gambacorti-Passerini C et al. *Eur J Haematol.* 2021;106(1):82-9. García-Gutiérrez V et al. *Ann Hematol.* 2019;98(2):321-30. Geelen IGP et al. *Haematol.* 2017;102(11):1842-9. Hehlmann R et al. *Am J Hematol.* 2019;94(1):46-54. Huguet F et al. *Br J Haematol.* 2019;187(5):615-26. Janssen L et al. *Haematol.* 2021;106(7):1876-82. Kekäle M et al. *Patient Prefer Adherence.* 2015;9:1733-40. Kuo CY et al. *Ther Adv Hematol.* 2018;9(3):65-78. Lang AS et al. *J Clin Pharm Ther.* 2015;40(4):391-7. Latagliata R et al. *Drugs Aging.* 2013;30(8):629-37. Latagliata R et al. *Neoplasia.* 2016;18(9):536-40. Latagliata R et al. *Hematol Oncol.* 2021;39(3):401-408. Li N et al. *J Med Econ.* 2017;20(4):328-36. Ma CE et al. *Leuk Res.* 2021;111. Marquinez-Alonso I et al. *Eur J Hosp Pharm.* 2020;27(5):299-301. Michallet M et al. *Curr Med Res Opin.* 2010;26(2):307-17. Nekoohesh L et al. *Int J Hematol Oncol Stem Cell Res.* 2020;14(1):11-18. Sánchez-Guijo FM et al. *Leuk Res.* 2011;35(9):1184-7. Scalzulli E et al. *Ann Hematol.* 2021;100(5):1213-9. Seiter K et al. *Adv Ther.* 2018;35(10):1671-85. Seiter K et al. *ISPOR* 2018. Stagno F et al. *Acta Oncol.* 2021;60(11):1527-33. Szeto AH et al. *Ann Pharmacother.* 2022;56(7):753-63. Tokuhira M et al. *Med Oncol.* 2018;35(3):38. Unnikrishnan R et al. *Clin Lymphoma Myeloma Leuk.* 2016;16(6):366-71. Unnikrishnan R et al. *Indian J Med Paediatr Oncol.* 2017;38(2):165-8.

Acknowledgements

Medical writing support was provided by Loraine Georgy, PhD, and we would like to thank Yan Meng, PhD, for her contribution, both employees of Analysis Group, Inc., which has received funding from Novartis Pharmaceuticals Corporation.