EE230

Real-World Healthcare Resource Utilization of Patients with Metastatic Breast Cancer: A Retrospective Cohort Analysis

Kent A. Hanson[1], Ashley S. Cha-Silva[1], Michael Hull[2], Mitchell DeKoven[2], Vamshi Ruthwik Anupindi[2], Riddhi P. Doshi[2], Gabrielle B. Rocque[3]

1 Pfizer, Inc., New York, NY, USA 2 IQVIA, Inc., Falls Church, VA, USA 3 Division of Hematology and Oncology, University of Alabama at Birmingham (UAB), Birmingham, Alabama, USA

Objective

This study described baseline demographics, clinical characteristics, and post-index HCRU and costs among all patients newly diagnosed with metastatic breast cancer (mBC) and a subset with the HR+/HER2+ molecular subtype

Conclusions

- Patients with mBC, particularly with HR+/HER2+ mBC, experienced high all-cause and mBC-related HCRU and costs, predominantly driven by pharmacy costs.
- All-cause HCRU and costs were higher during the initial three months after mBC diagnosis compared with the rest of the follow-up period.
- The higher economic burden during the first three months after mBC diagnosis, particularly among patients with HR+/HER2+ mBC, represents an opportunity to share information about anticipated treatment time requirements and to consider interventions aimed at reducing costs.
- With therapies targeting the HR+/HER2+ subtype (i.e., pertuzumab and trastuzumab) approaching loss of exclusivity, there is an opportunity to support innovation in HR+/HER2+ mBC therapeutic management.

Limitations: 1) While the patient selection algorithm excluded patients with confirmed mBC in the pre-index period, some patients with mBC who were diagnosed prior to the start of the study period may have been included in the sample. 2) Claims databases have inherent limitations affecting generalizability because the claims are collected for the purpose of reimbursement within the commercially-insured population. Results and conclusions are therefore limited to patients enrolled in managed care plans. 3) Presence of a diagnosis code on a medical encounter or outside claim may have not been conclusive or positive presence of disease, as the diagnosis code entered may be incorrect or included as rule-out criteria rather than actual disease.

References: 1. Gallicchio L, Devasia TP, Tonorezos E, Mollica MA, Mariotto A. Estimation of the number of individuals living with metastatic cancer in the United States. J Natl Cancer Inst. 2022; 114(11):1476-1483. **2.** Howlader N, Cronin KA, Kurian AW, Andridge R. Differences in Breast Cancer Survival by Molecular Subtypes in the United States. Cancer Epidemiol Biomarkers Prev. 2018; 27(6):619-626. **3.** Swain SM, Miles D, Kim SB, Im YH, Im SA, Semiglazov V, Ciruelos E, Schneeweiss A, Loi S, Monturus E, Clark E, Knott A, Restuccia E, Benyunes MC, Cortés J; CLEOPATRA study group. Pertuzumab, trastuzumab, and docetaxel for HER2-positive metastatic breast cancer (CLEOPATRA): end-of-study results from a double-blind, randomised, placebo-controlled, phase 3 study. Lancet Oncol. 2020 Apr; 21(4):519-530. **4.** Dai WF, Beca JM, Nagamuthu C, Liu N, de Oliveira C, Earle CC, Trudeau M, Chan KKW. Cost-effectiveness Analysis of Pertuzumab With Trastuzumab in Patients With Metastatic Breast Cancer. JAMA Oncol. 2022 Apr 1; 8(4):597-606. **5.** Huse DM, Russo PA, Vasey J. Expanding the evidence base in outcomes research: linking electronic medical records and claims data. ISPOR Connections Vol 19 No 3. May-June 2013.

Acknowledgement & Disclosures: Michael Hull, Mitchell DeKoven, Vamshi Ruthwik Anupindi, and Riddhi P. Doshi are employees of IQVIA, which received funding from Pfizer to conduct these analyses. Kent A. Hanson and Ashley S. Cha-Silva are employees of Pfizer. Gabrielle B. Rocque received funding from Pfizer for consultation on this this study. Edward Broughton, Rickard Sandin, and Stella Stergiopoulos of Pfizer provided input on the poster content.

Click or scan this quick response (QR) to download this poster along with associated material.

Presenting author: Dr. Kent Hanson
<Kent.Hanson@pfizer.com>

Presented at ISPOR 2023• May 7-10, 2023 • Boston, MA, USA

Background

- Recent estimates predict the number of US women living with mBC will increase from ~140,000 in 2018 to more than 160,000 by 2025.[1]
- Hormone receptor positive (HR+) and human epidermal growth factor receptor positive (HER2+) molecular subtypes account for nearly 10% of mBC cases.[2]
- Medications targeting HER2+ tumors (e.g., trastuzumab and pertuzumab) improve survival but may be cost burdensome to patients.[3,4]
- Recent real-world evidence regarding healthcare resource utilization (HCRU) and costs among patients with mBC and HR+/HER2+ mBC after the availability of these targeted therapies in the US is lacking.

Methods

- This retrospective, observational study utilized linked data from IQVIA's Oncology Electronic Medical Record (OEMR) database and PharMetrics® Plus adjudicated health plan claims database. **Figure 1** illustrates the study design.
- The OEMR database comprises medical records data for 500,000+ cancer patients from 344+ community-based medical oncology practices and comprehensive cancer centers in 34 US states. PharMetrics® Plus comprises adjudicated claims for more than 190 million unique enrollees across the US.
- Baseline patient demographics, clinical characteristics, and total healthcare costs were assessed during the six-month pre-index period preceding the index date (date of initial mBC diagnosis).
- The overall post-index period was divided into an initial period and a variable continuation period, as defined in **Figure 1**. All-cause and mBC-related post-index HCRU and costs were assessed as 2021 USD per-patient-per-month (PPPM) for the overall period, the initial period, and the continuation period.

Figure 1: Study schematic

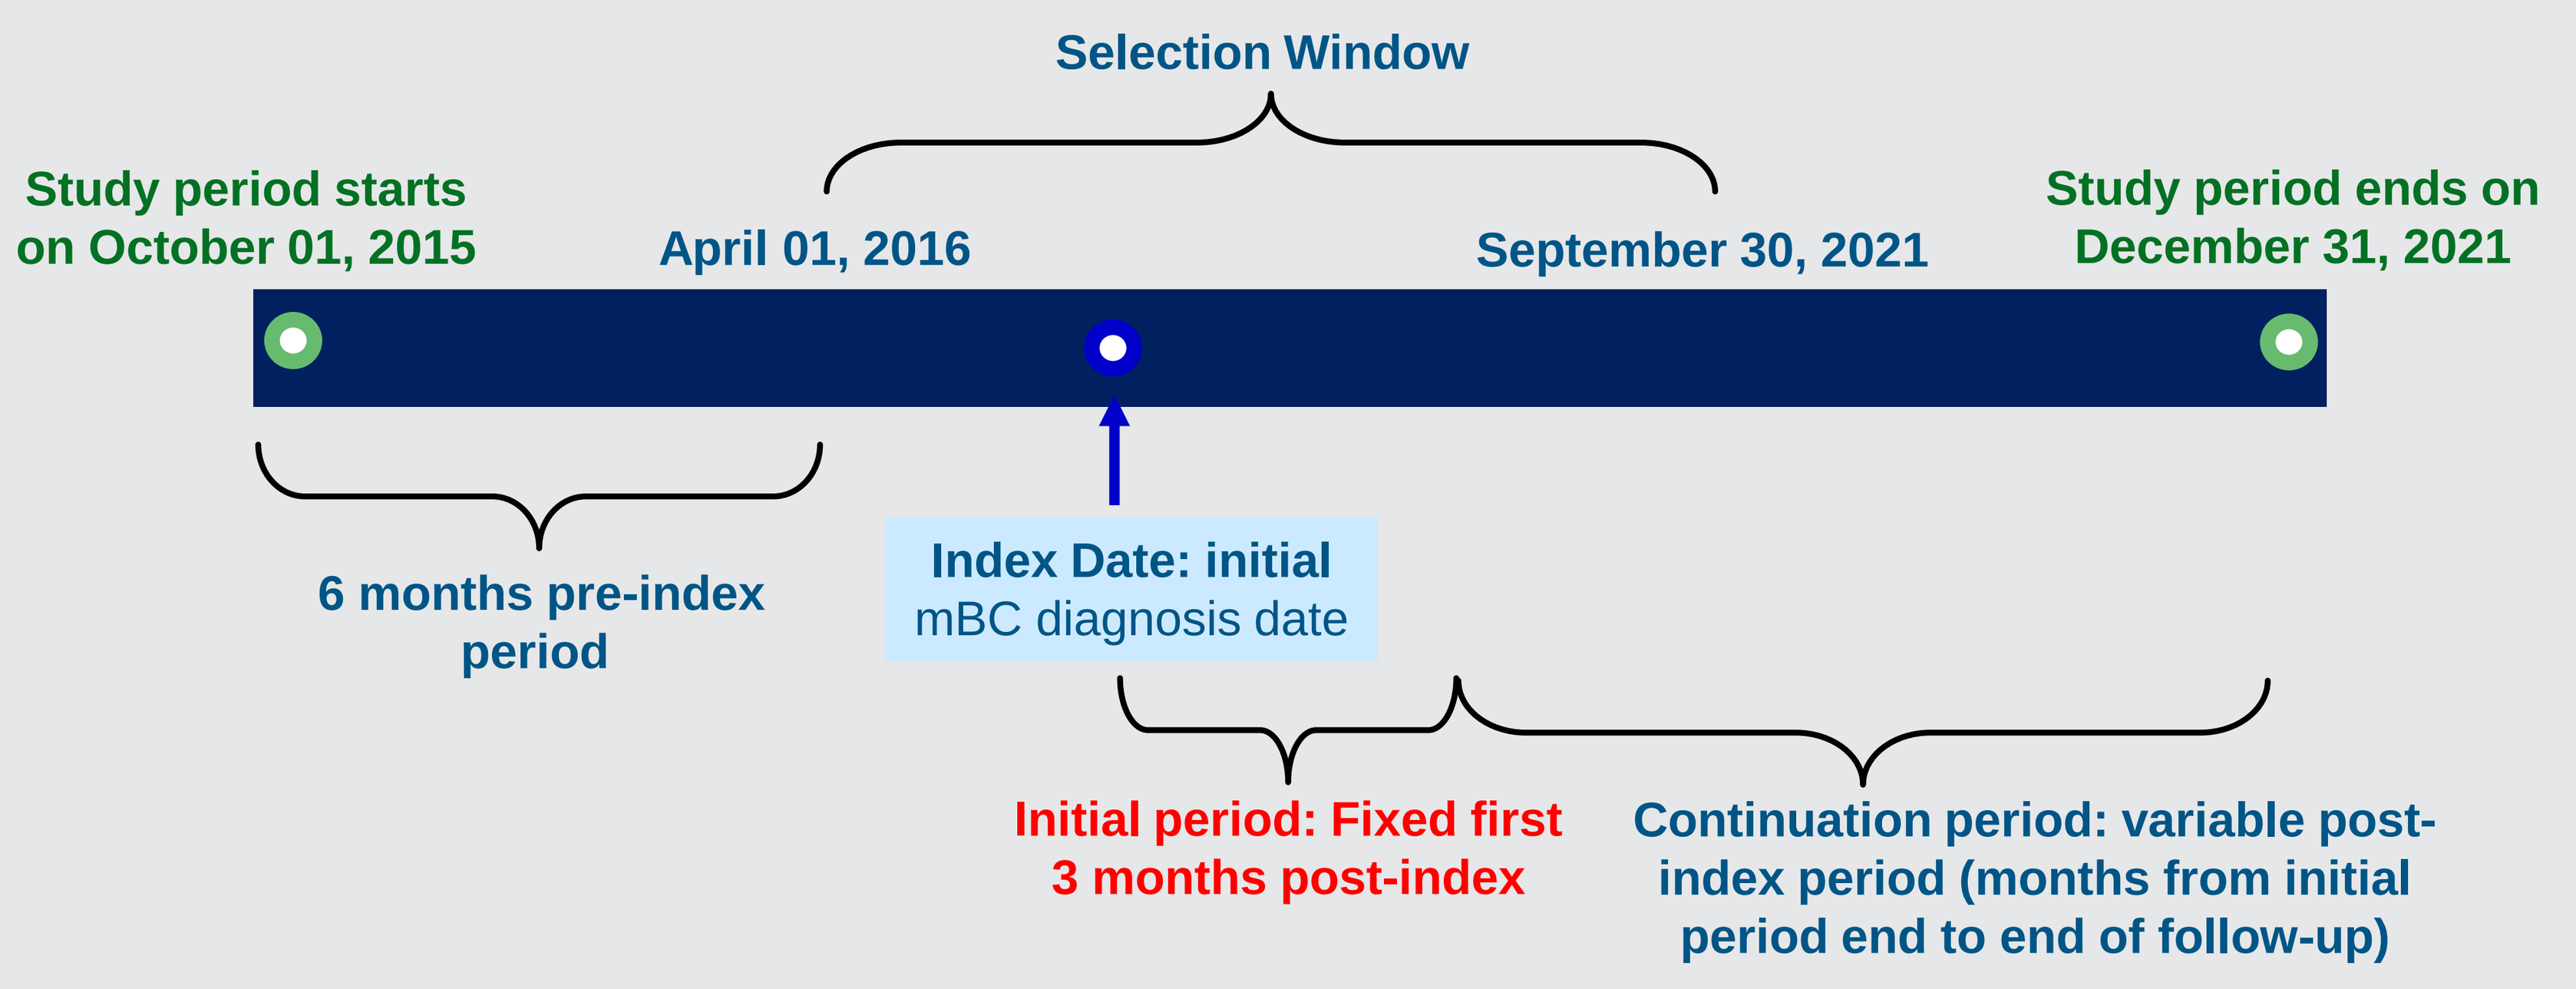
Selection Window
Study period starts on October 01, 2015
April 01, 2016
September 30, 2021
Study period ends on December 31, 2021
6 months pre-index period
Index Date: initial mBC diagnosis date
Initial period: Fixed first 3 months post-index
Continuation period: variable post-index period (months from initial period end to end of follow-up)

Results

Figure 2: Attrition Diagram

EHR record indicating mBC diagnosis
N=29,719

↓

Documented HR and HER2 status*
N=12,449

↓

OEMR and PharMetrics Plus linkage and continuous enrollment 6 mo prior to and ≥3 mo following index
N=458

↓

Females aged ≥18 years at index N=447

↓

No prior non-breast primary cancers identified pre-index
N= 359 (44 HR+/HER2+ mBC)

Documented using immunohistochemical analysis or fluorescence in situ hybridization test results

BASELINE CHARACTERISTICS

- The mean (SD) age of patients in the all-mBC cohort and HR+/HER2+ subset was 56.0 (12.0) years and 53.3 (10.6) years, with a high proportion of patients aged 55-64 years (38.2% and 40.9%) with commercial/self-insured health plan coverage (87.2% and 95.5%), respectively.
- Nearly two-thirds of the patients had a modified Charlson Comorbidity Index score of 0 (excluding cancer comorbidities).

HEALTHCARE RESOURCE UTILIZATION

- The HR+/HER2+ subset had a higher utilization of all-cause ER visits compared to the entire cohort (50.0% vs. 41.5%, respectively) (**Table 1**).
- The mean number of all-cause HCRU was higher in the initial period than the continuation period for both the cohort and subset across all service categories (**Table 1**).
- MBC-related HCRU was lower during the initial period than in the continuation period within each category of HCRU assessed for the all-mBC cohort (**Table 2**).
- For the HR+/HER2+ subset, HCRU was higher in the initial period than in the continuation period for specific services including ≥1 mBC-related physician office visit, laboratory/pathology, outpatient surgical, and ancillary visit (**Table 2**).

Table 1: All-cause HCRU in all-mBC cohort and HR+/HER2+ subset

Categories*		Overall period		Initial period		Continuation period	
		Proportion of utilizers	Mean (SD) number of visits	Proportion of utilizers	Mean (SD) number of visits	Proportion of utilizers	Mean (SD) number of visits
All-mBC cohort							
Outpatient services	Prescription fills	96.7%	5.5 (4.1)	94.4%	8.0 (6.7)	95.3%	5.5 (8.8)
	Physician office visits	96.7%	1.9 (1.4)	92.5%	2.6 (2.1)	94.2%	1.9 (2.1)
	Lab/pathology	97.2%	4.0 (3.1)	88.6%	5.9 (5.3)	93.0%	3.6 (3.5)
	Radiology	95.0%	1.7 (2.0)	79.7%	2.7 (4.1)	87.5%	1.4 (2.1)
	Surgical services	79.9%	0.3 (0.5)	54.0%	0.6 (0.9)	67.1%	0.2 (0.4)
	Ancillary/ other	96.7%	4.4 (3.9)	88.0%	7.1 (6.4)	92.5%	4.1 (7.3)
Emergency room visits		41.5%	0.1 (0.1)	13.9%	0.1 (0.2)	35.4%	0.1 (0.1)
Inpatient stays		35.1%	0.1 (0.1)**	9.7%	0.4 (0.1)**	29.8%	0.1 (0.2)**
HR+/HER2+ cohort							
Outpatient services	Prescription fill	100.0%	6.9 (4.0)	100.0%	12.4 (7.9)	97.7%	6.1 (4.1)
	Physician office visits	100.0%	2.3 (1.5)	100.0%	3.0 (1.7)	97.7%	2.2 (1.8)
	Lab/pathology	97.7%	4.6 (3.1)	93.2%	7.7 (4.5)	93.2%	4.1 (3.4)
	Radiology	97.7%	2.4 (3.2)	86.4%	3.0 (4.4)	86.4%	1.7 (2.1)
	Surgical services	86.4%	0.4 (0.3)	68.2%	0.9 (0.9)	63.6%	0.2 (0.4)
	Ancillary/ other	100.0%	5.8 (4.1)	95.5%	9.4 (5.5)	95.5%	5.2 (4.8)
Emergency room visits		50.0%	0.1 (0.1)	27.3%	0.1 (0.2)	38.6%	0.1 (0.2)
Inpatient stays		31.8%	0.1 (0.1)**	13.6%	0.3 (0.0)**	27.3%	0.1 (0.1)**

**Computed among patients with ≥1 inpatient stay

Table 2: mBC-related HCRU in all-mBC cohort and HR+/HER2+ subset

Categories*		Overall period		Initial period		Continuation period	
		Proportion of utilizers	Mean (SD) number of visits	Proportion of utilizers	Mean (SD) number of visits	Proportion of utilizers	Mean (SD) number of visits
All-mBC cohort							
Outpatient services	Prescription fills	85.2%	1.1 (1.0)	74.1%	1.5 (1.6)	82.2%	1.2 (3.3)
	Physician office visits	90.5%	1.3 (1.1)	84.4%	1.9 (1.5)	86.9%	1.2 (1.9)
	Lab/pathology	89.7%	3.1 (2.9)	79.7%	4.9 (4.9)	84.7%	2.7 (3.2)
	Radiology	87.7%	1.4 (1.9)	72.7%	2.2 (3.8)	78.6%	1.2 (2.0)
	Surgical services	69.4%	0.2 (0.4)	46.5%	0.4 (0.7)	52.6%	0.1 (0.3)
	Ancillary/ other	88.3%	3.4 (3.4)	78.3%	5.9 (5.8)	84.7%	3.2 (7.0)
Emergency room visits		27.6%	0.0 (0.1)	9.5%	0.0 (0.2)	22.8%	0.0 (0.1)
Inpatient stays		6.7%	0.0 (0.0)**	2.2%	0.4 (0.1)**	4.5%	0.1 (0.0)**
HR+/HER2+ cohort							
Outpatient services	Prescription fill`	97.7%	2.0 (1.3)	93.2%	3.1 (2.0)	95.5%	1.9 (1.4)
	Physician office visits	97.7%	1.6 (1.2)	97.7%	2.4 (1.3)	95.5%	1.5 (1.5)
	Lab/pathology	95.5%	3.7 (2.8)	93.2%	6.6 (4.2)	90.9%	3.1 (2.8)
	Radiology	93.2%	2.1 (3.1)	75.0%	2.5 (4.4)	81.8%	1.4 (2.0)
	Surgical services	81.8%	0.2 (0.3)	63.6%	0.7 (0.8)	56.8%	0.2 (0.3)
	Ancillary/ other	95.5%	4.7 (3.4)	93.2%	8.5 (5.3)	90.9%	4.0 (3.6)
Emergency room visits		31.8%	0.0 (0.1)	13.6%	0.1 (0.2)	25.0%	0.0 (0.1)
Inpatient stays		4.5%	0.1 (0.1)**	0.0%	0.0 (0.0)**	4.5%	0.1 (0.1)**

**Computed among patients with ≥1 inpatient stay

HEALTHCARE COSTS

- The mean (SD) all-cause PPPM costs in the all-mBC cohort and HR+/HER2+ subset were $9,601 ($8,933) and $15,848 ($11,382) during the overall post-index period, respectively, driven predominantly by pharmacy costs (**Figure 2**).
- The mean all-cause total and service-category specific healthcare costs were higher during the initial period compared with the continuation period for both cohort and subset (**Figure 2**).

Figure 3: All-cause healthcare costs in all-mBC cohort and HR+/HER2+ subset

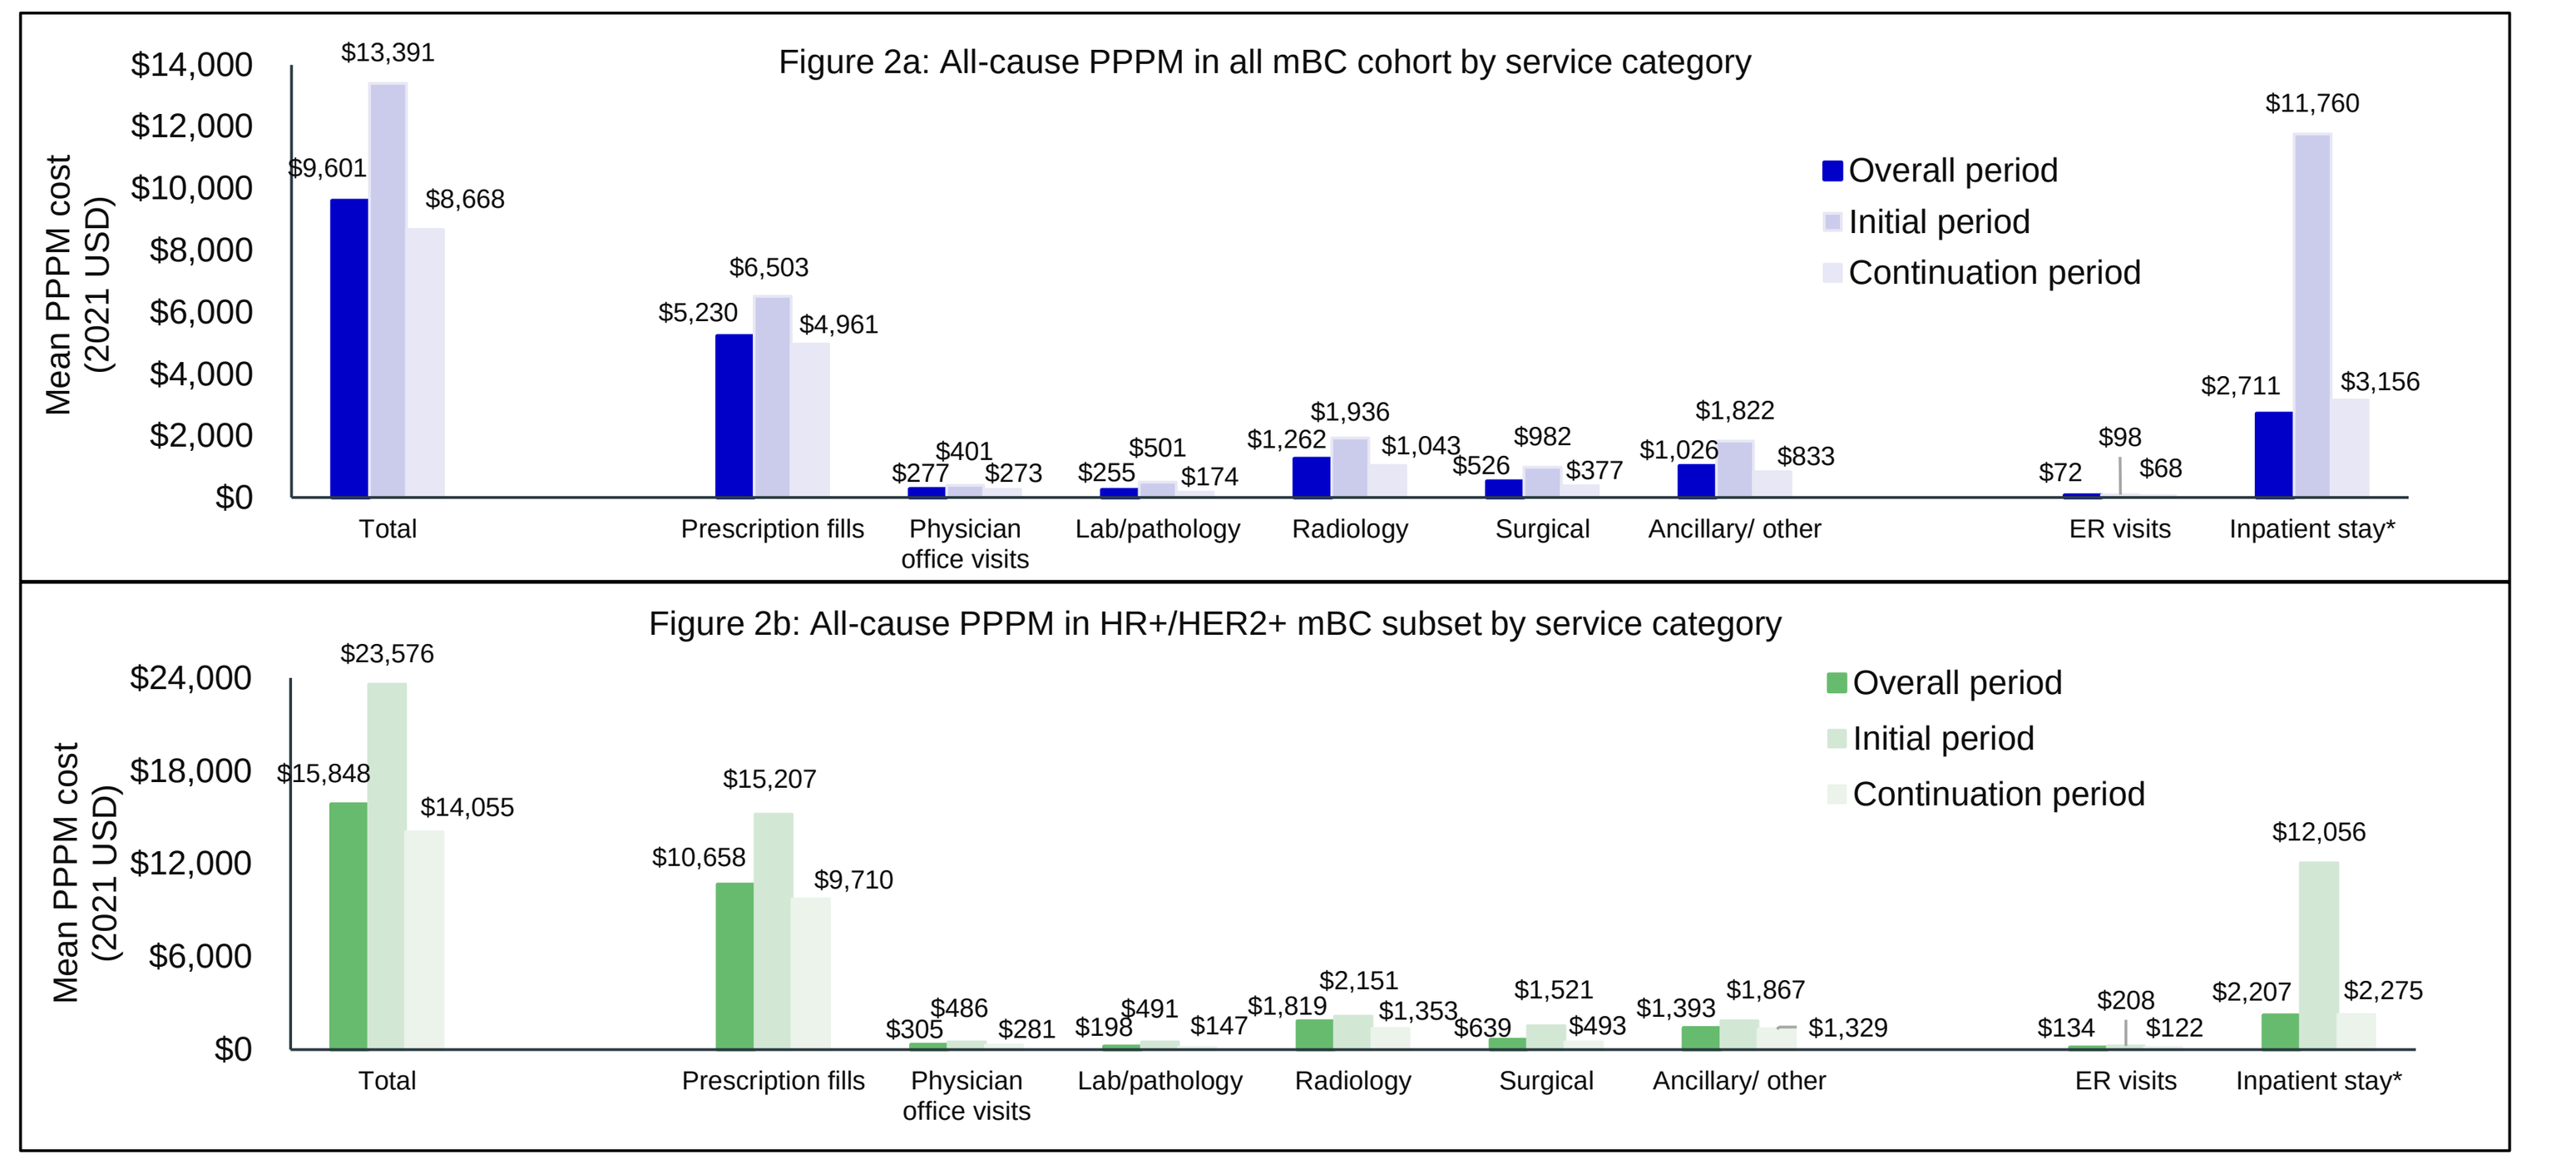
Figure 2a: All-cause PPPM in all mBC cohort by service category
Mean PPPM cost (2021 USD)
Overall period
Initial period
Continuation period
Total
Prescription fills
Physician office visits
Lab/pathology
Radiology
Surgical
Ancillary/ other
ER visits
Inpatient stay*
Figure 2b: All-cause PPPM in HR+/HER2+ mBC subset by service category

- The mean (SD) mBC-related total PPPM costs in the all-mBC cohort and HR+/HER2+ subset were $6,991 ($7,189) and $12,712 ($10,126), respectively, with pharmacy costs accounting for about half of the total costs (**Figure 3**).
- The mean total mBC-related PPPM costs were higher during the initial period compared with the continuation period for the all-mBC cohort ($9,267 vs $6,337) and HR+/HER2+ subset ($17,416 vs $11,460) (**Figure 3**).

Figure 4: mBC-related healthcare costs in all mBC cohort and HR+/HER2+ subset

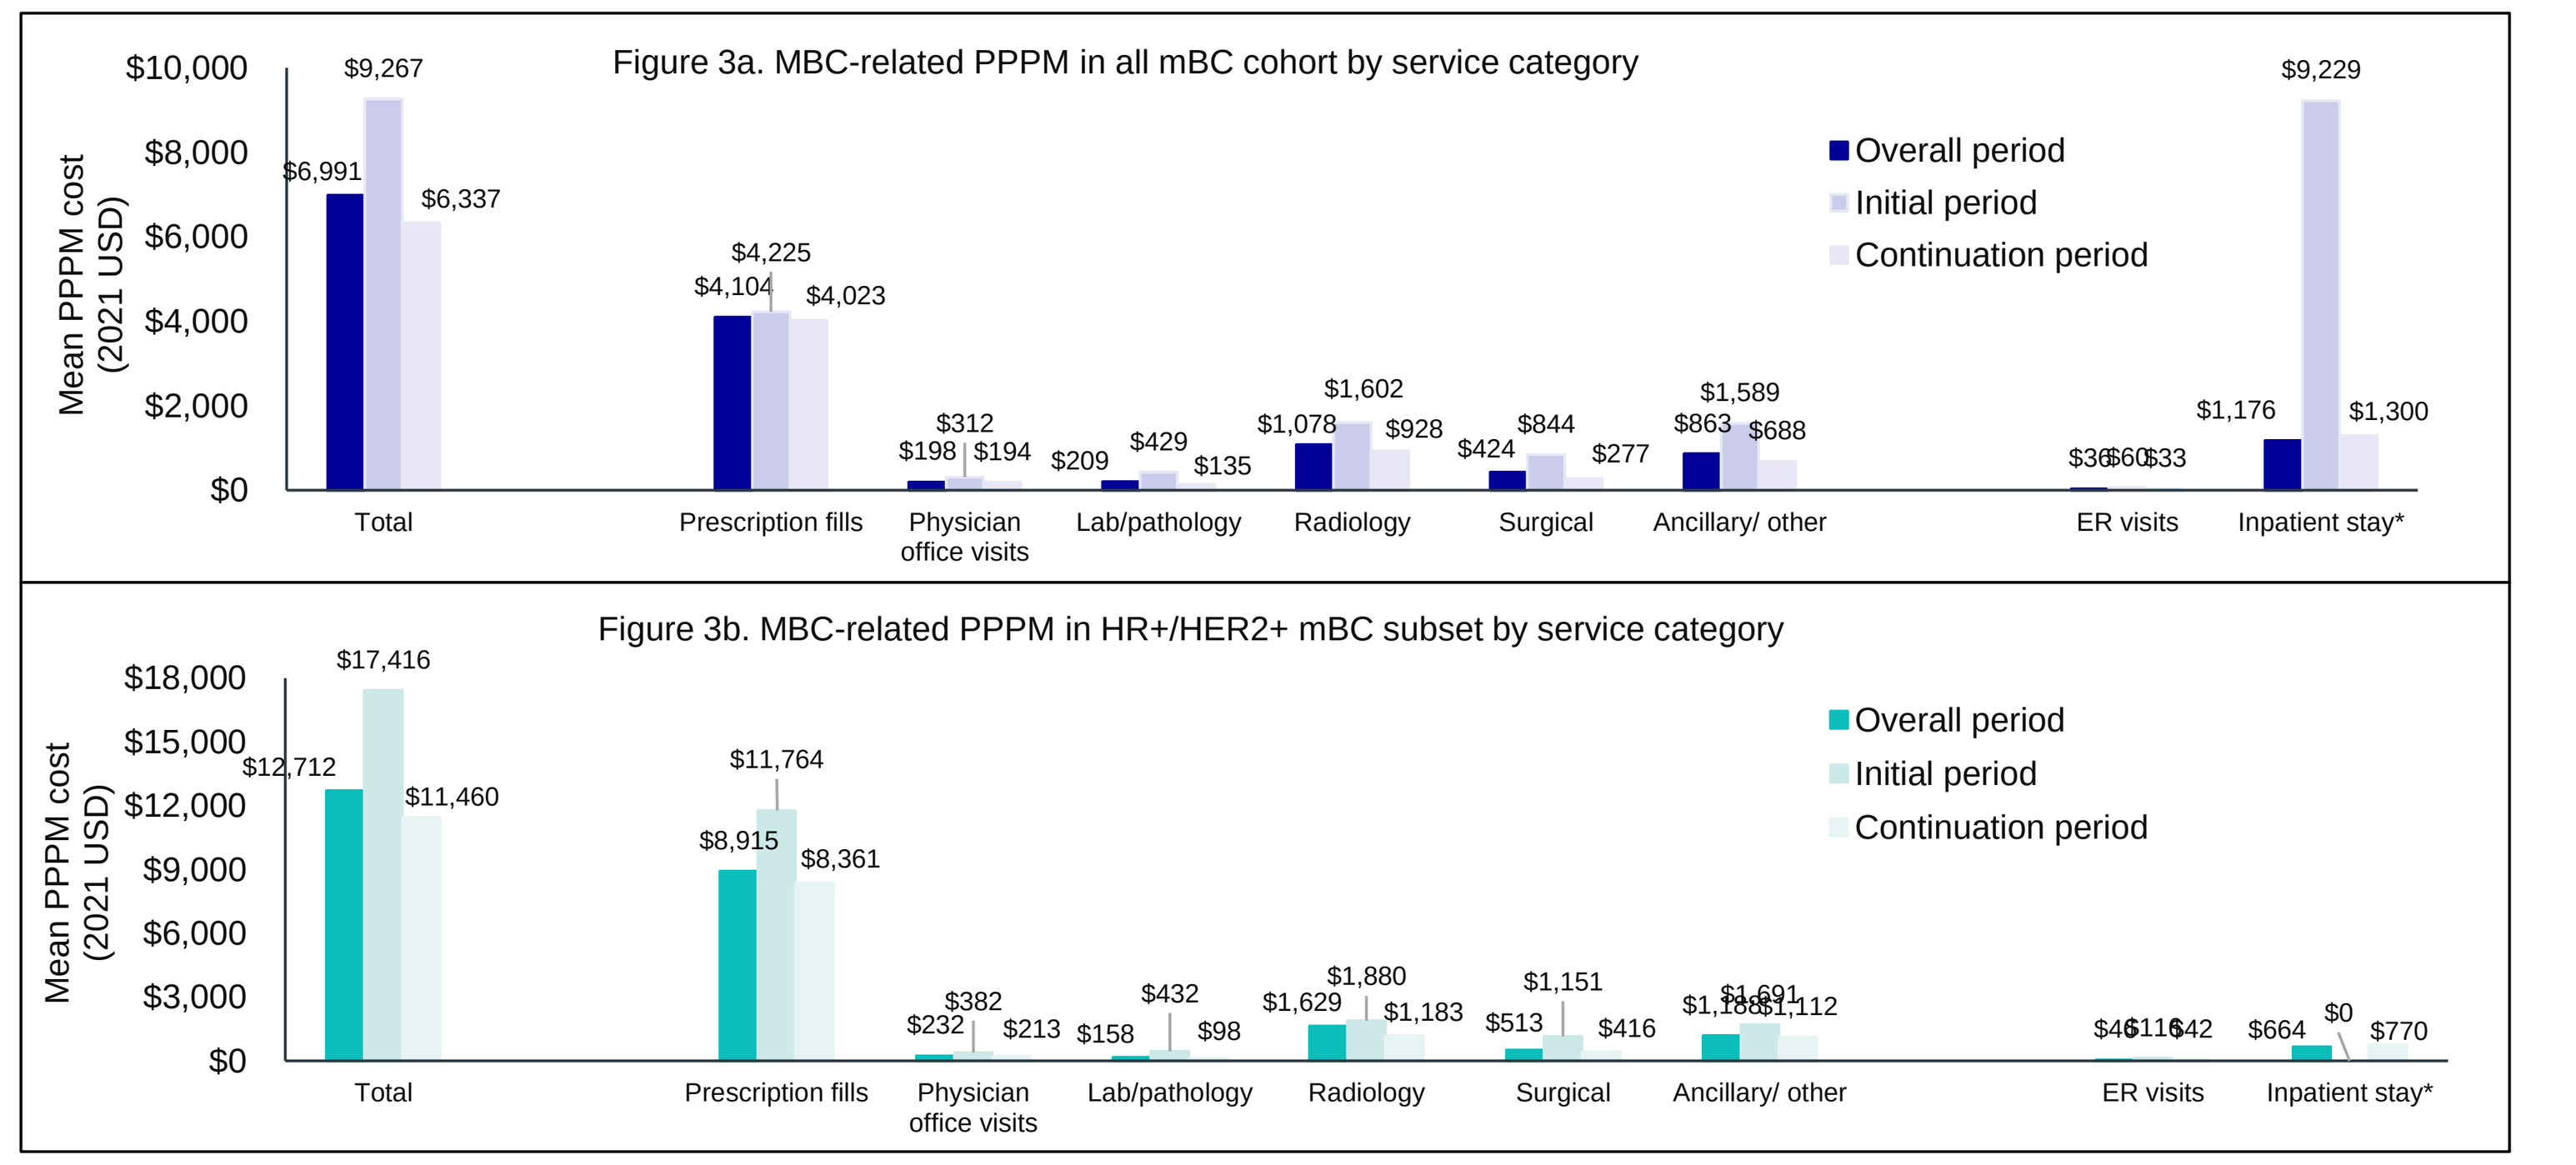
Figure 3a. MBC-related PPPM in all mBC cohort by service category
Mean PPPM cost (2021 USD)
Overall period
Initial period
Continuation period
Total
Prescription fills
Physician office visits
Lab/pathology
Radiology
Surgical
Ancillary/ other
ER visits
Inpatient stay*
Figure 3b. MBC-related PPPM in HR+/HER2+ mBC subset by service category

Copyright ©2023. All rights reserved.