EPH237

US Population at Elevated Risk for Cancer: Population Size and Characteristics

International Society for Pharmacoeconomics and Outcomes Research (ISPOR) 2023
May 7-10, 2023, Boston, MA

David C. Grabowski, PhD,[1] William Cance, MD,[2] Ze Cong, PhD[2] | [1]Harvard Medical School, Boston, MA; [2]GRAIL, LLC, Menlo Park, CA

INTRODUCTION

- An estimated 1.9 million cancer cases are projected to result in over 600,000 cancer deaths in the year of 2022, making cancer the second leading cause of death in the US[1]
- The national cancer-attributable costs for medical services and prescription drugs are estimated at over $200 billion annually[2]
- Prior research has established that cancer is associated with a range of risk factors
- Roughly 40% of cancer cases are linked to potentially modifiable behaviors[3]

OBJECTIVES

- To understand the presence of cancer risk factors in the US population:
 - To estimate the number of individuals in the US at risk for cancer
 - To describe the characteristics of these individuals

STUDY DESIGN: DATA

- Data were from the National Health Interview Survey (NHIS) main survey and cancer supplement in 2015
- In the main NHIS survey, respondents were asked about their personal history of cancer, history of smoking, body mass index, and health conditions, such as diabetes
- Meanwhile, in the supplement, they were asked about their family history of cancer, sun exposure, and alcohol consumption
- Based upon a literature review of frequently identified cancer risk factors, we identified eight risk factors that were available in either the main NHIS or cancer supplement survey:
 - ever smoked (smoked >100 cigarettes in their lives)
 - smoke now (whether people with a history of smoking still report smoking either every day or some days)
 - heavy alcohol consumption (consuming more than 3 standard drinks per day)
 - excess body weight (BMI>30)
 - diabetes (ever been told by a doctor that the respondent has diabetes)
 - ultraviolet (UV) exposure (any sunburns in the last year)
 - personal cancer history (ever been told by a doctor that the respondent has cancer)
 - family cancer history (biological parent or sibling ever had cancer)

STUDY DESIGN: METHODS

- In all analyses, we used sample weights to obtain nationally representative estimates
- The 2015 cancer supplement was only conducted for the person selected in the NHIS sample (questions were not asked about other household members). Because of this, we rescaled the weights by the total of all adult sample weights divided by the total of adult sample weights among those who completed the supplement
- Using these adjusted weights, we are then able to estimate both prevalence rates and population totals for the risk factors
- We examined the correlation of cancer risk factors
- We compared cancer risk factors by age, gender, race/ethnicity, educational attainment, and family income
- We examined comorbidities (e.g., hypertension and high cholesterol) by cancer risk (3+ risk factors vs <3 risk factors)

LIMITATIONS

- Although we found individuals with risk factors were more likely to have been screened at least once, we were unable to determine whether they have received their USPSTF recommended screenings over time
- We were only able to study those cancer risk factors that were present in the NHIS main file and cancer supplement
- Our results do not account for "survivor's bias" in which the count of individuals at risk for cancer excludes individuals who may have already died from cancer

KEY RESULTS: MORE THAN 200 MILLION US ADULTS (84% OF POPULATION) HAVE AT LEAST ONE RISK FACTOR FOR CANCER, WITH ~1/3 ADULTS HAVING 3 OR MORE

Figure 1. Prevalence of US Population with Risk Factors for Cancer

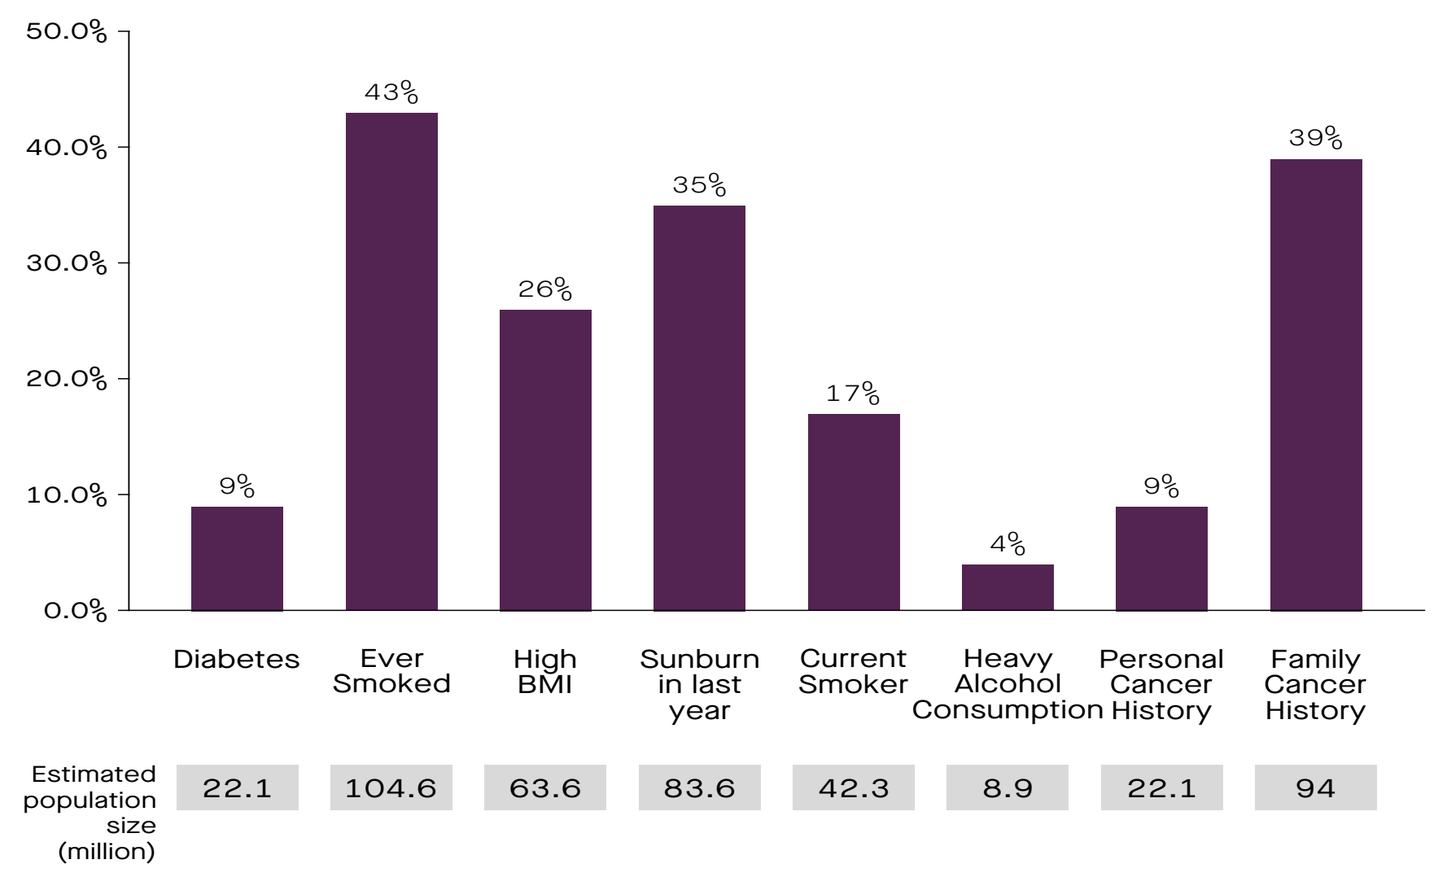
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
9%
43%
26%
35%
17%
4%
9%
39%
Diabetes
Ever Smoked
High BMI
Sunburn in last year
Current Smoker
Heavy Alcohol Consumption
Personal Cancer History
Family Cancer History
Estimated population size (million)
22.1
104.6
63.6
83.6
42.3
8.9
22.1
94

- Ever smoked was the most prevalent cancer risk factor, followed by family history of cancer and sunburn in the last year

Figure 2. Prevalence of US Population with Different Number of Risk Factors for Cancer

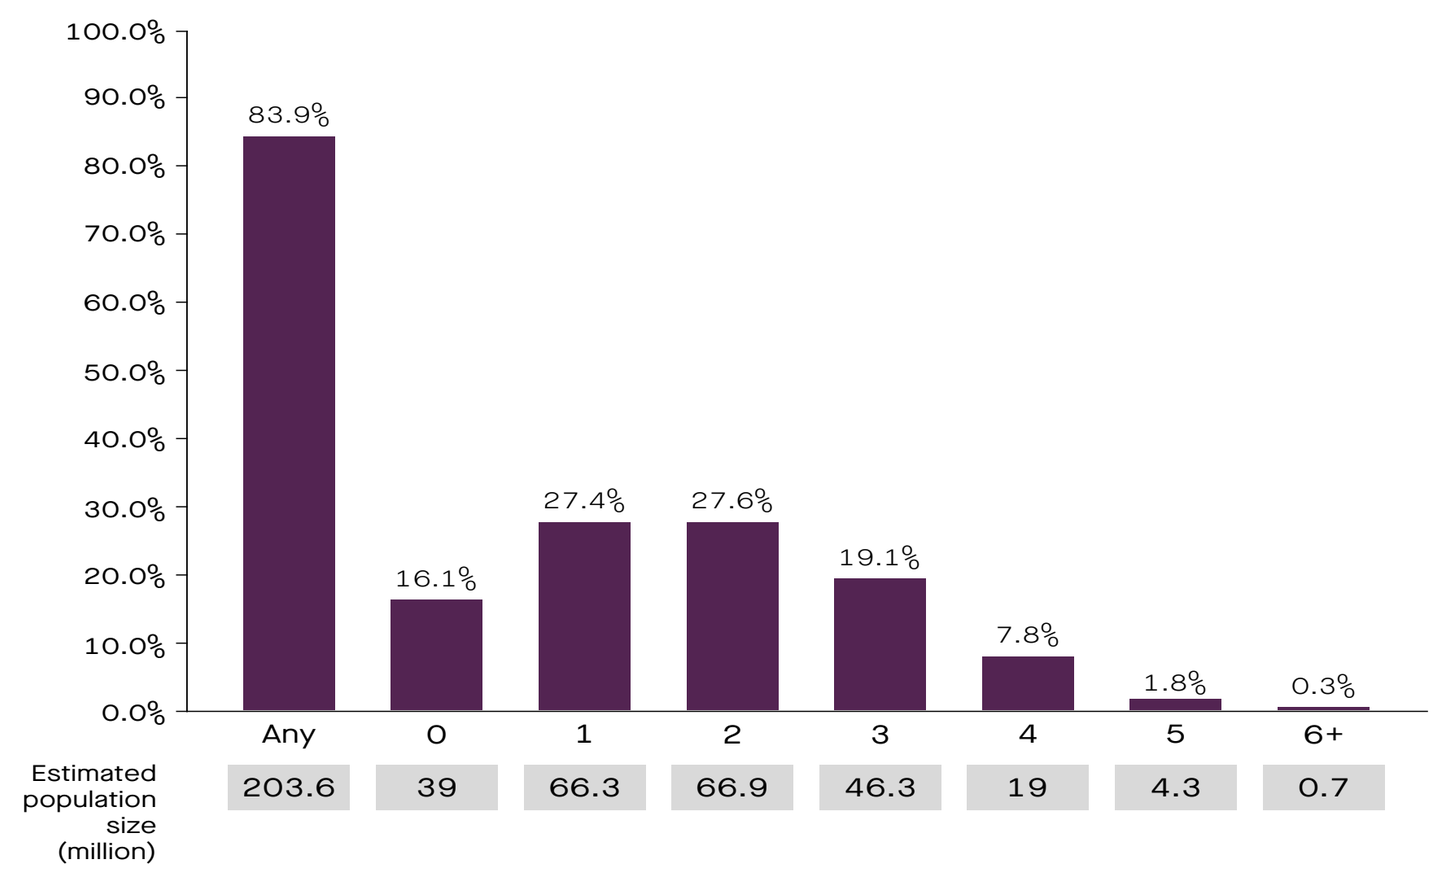
100.0%
90.0%
80.0%
70.0%
60.0%
50.0%
40.0%
30.0%
20.0%
10.0%
0.0%
83.9%
16.1%
27.4%
27.6%
19.1%
7.8%
1.8%
0.3%
Any
0
1
2
3
4
5
6+
Estimated population size (million)
203.6
39
66.3
66.9
46.3
19
4.3
0.7

- An estimated 203.6 million (84%) US adults have at least one of the eight risk factors for cancer, while 70.3 million (29%) have three or more

Table 1: Descriptive Statistics by Cancer Risk

	Share of those with:				
	Share of Full Sample	1 + Risk Factors	0 Risk Factors	3 + Risk Factors	<3 Risk Factors
White, non-Hispanic	0.693	0.730	0.501	0.798	0.650
Hispanic	0.140	0.122	0.234	0.087	0.162
Black	0.108	0.097	0.164	0.080	0.119
Asian	0.045	0.035	0.096	0.016	0.057
Non-White (Incl. Hispanic)	0.307	0.270	0.499	0.202	0.350
Less than High School	0.106	0.108	0.095	0.127	0.098
High School or Equivalent	0.232	0.240	0.194	0.283	0.212
Some College or Associates	0.317	0.323	0.283	0.350	0.303
4-Year College or More	0.128	0.121	0.161	0.084	0.145
$50,000+ household income	0.600	0.596	0.620	0.529	0.630
Age	47.64	48.70	42.13	51.73	45.97
Male	0.514	0.522	0.475	0.539	0.504
Ever had Skin Cancer Exam	0.253	0.268	0.166	0.284	0.240
Ever had pap smear test	0.956	0.963	0.919	0.980	0.946
Ever had colonoscopy	0.758	0.774	0.657	0.805	0.736
Ever had breast physical exam	0.533	0.548	0.411	0.588	0.502
Ever had mammogram	0.903	0.910	0.860	0.918	0.897
N	26,495	22,499	3,996	8,031	18,464

Author's calculations from 2015 National Health Interview Survey cancer supplement data.
Some cancer screening questions were filtered by age and sex - in these cases, the share of those asked is reported.

- Those with more risk factors are more likely to be white, male, older age on average, with less education, and less family income. Most cancer screening measures are positively associated with the number of risk factors.

Table 2: Correlations Among Cancer Risk Factors

	Smoke Ever	Smoke Now	Cancer History	Relative Cancer	Diabetes	High BMI	Sunburns	Alcohol
Smoke Ever	1	0.567	0.160	0.191	0.145	0.186	0.114	0.071
Smoke Now		1	0.029	0.091	0.047	0.078	0.085	0.086
Cancer History			1	0.134	0.117	0.080	0.010	-0.011
Family Cancer				1	0.113	0.125	0.282	0.040
Diabetes					1	0.202	0.008	-0.008
High Body Mass Index						1	0.095	0.028
Sunburns							1	0.103
Alcohol								1

Author's calculations from 2015 National Health Interview Survey cancer supplement data. Each cell reports the correlation coefficient between the row and column variables. All correlation coefficients are significant at the p<0.01 level.

- The risk factors are at least moderately correlated with one another, indicating that having a risk factor for cancer increases the probability of having other risk factors

Figure 3. Prevalence of Risk Factors

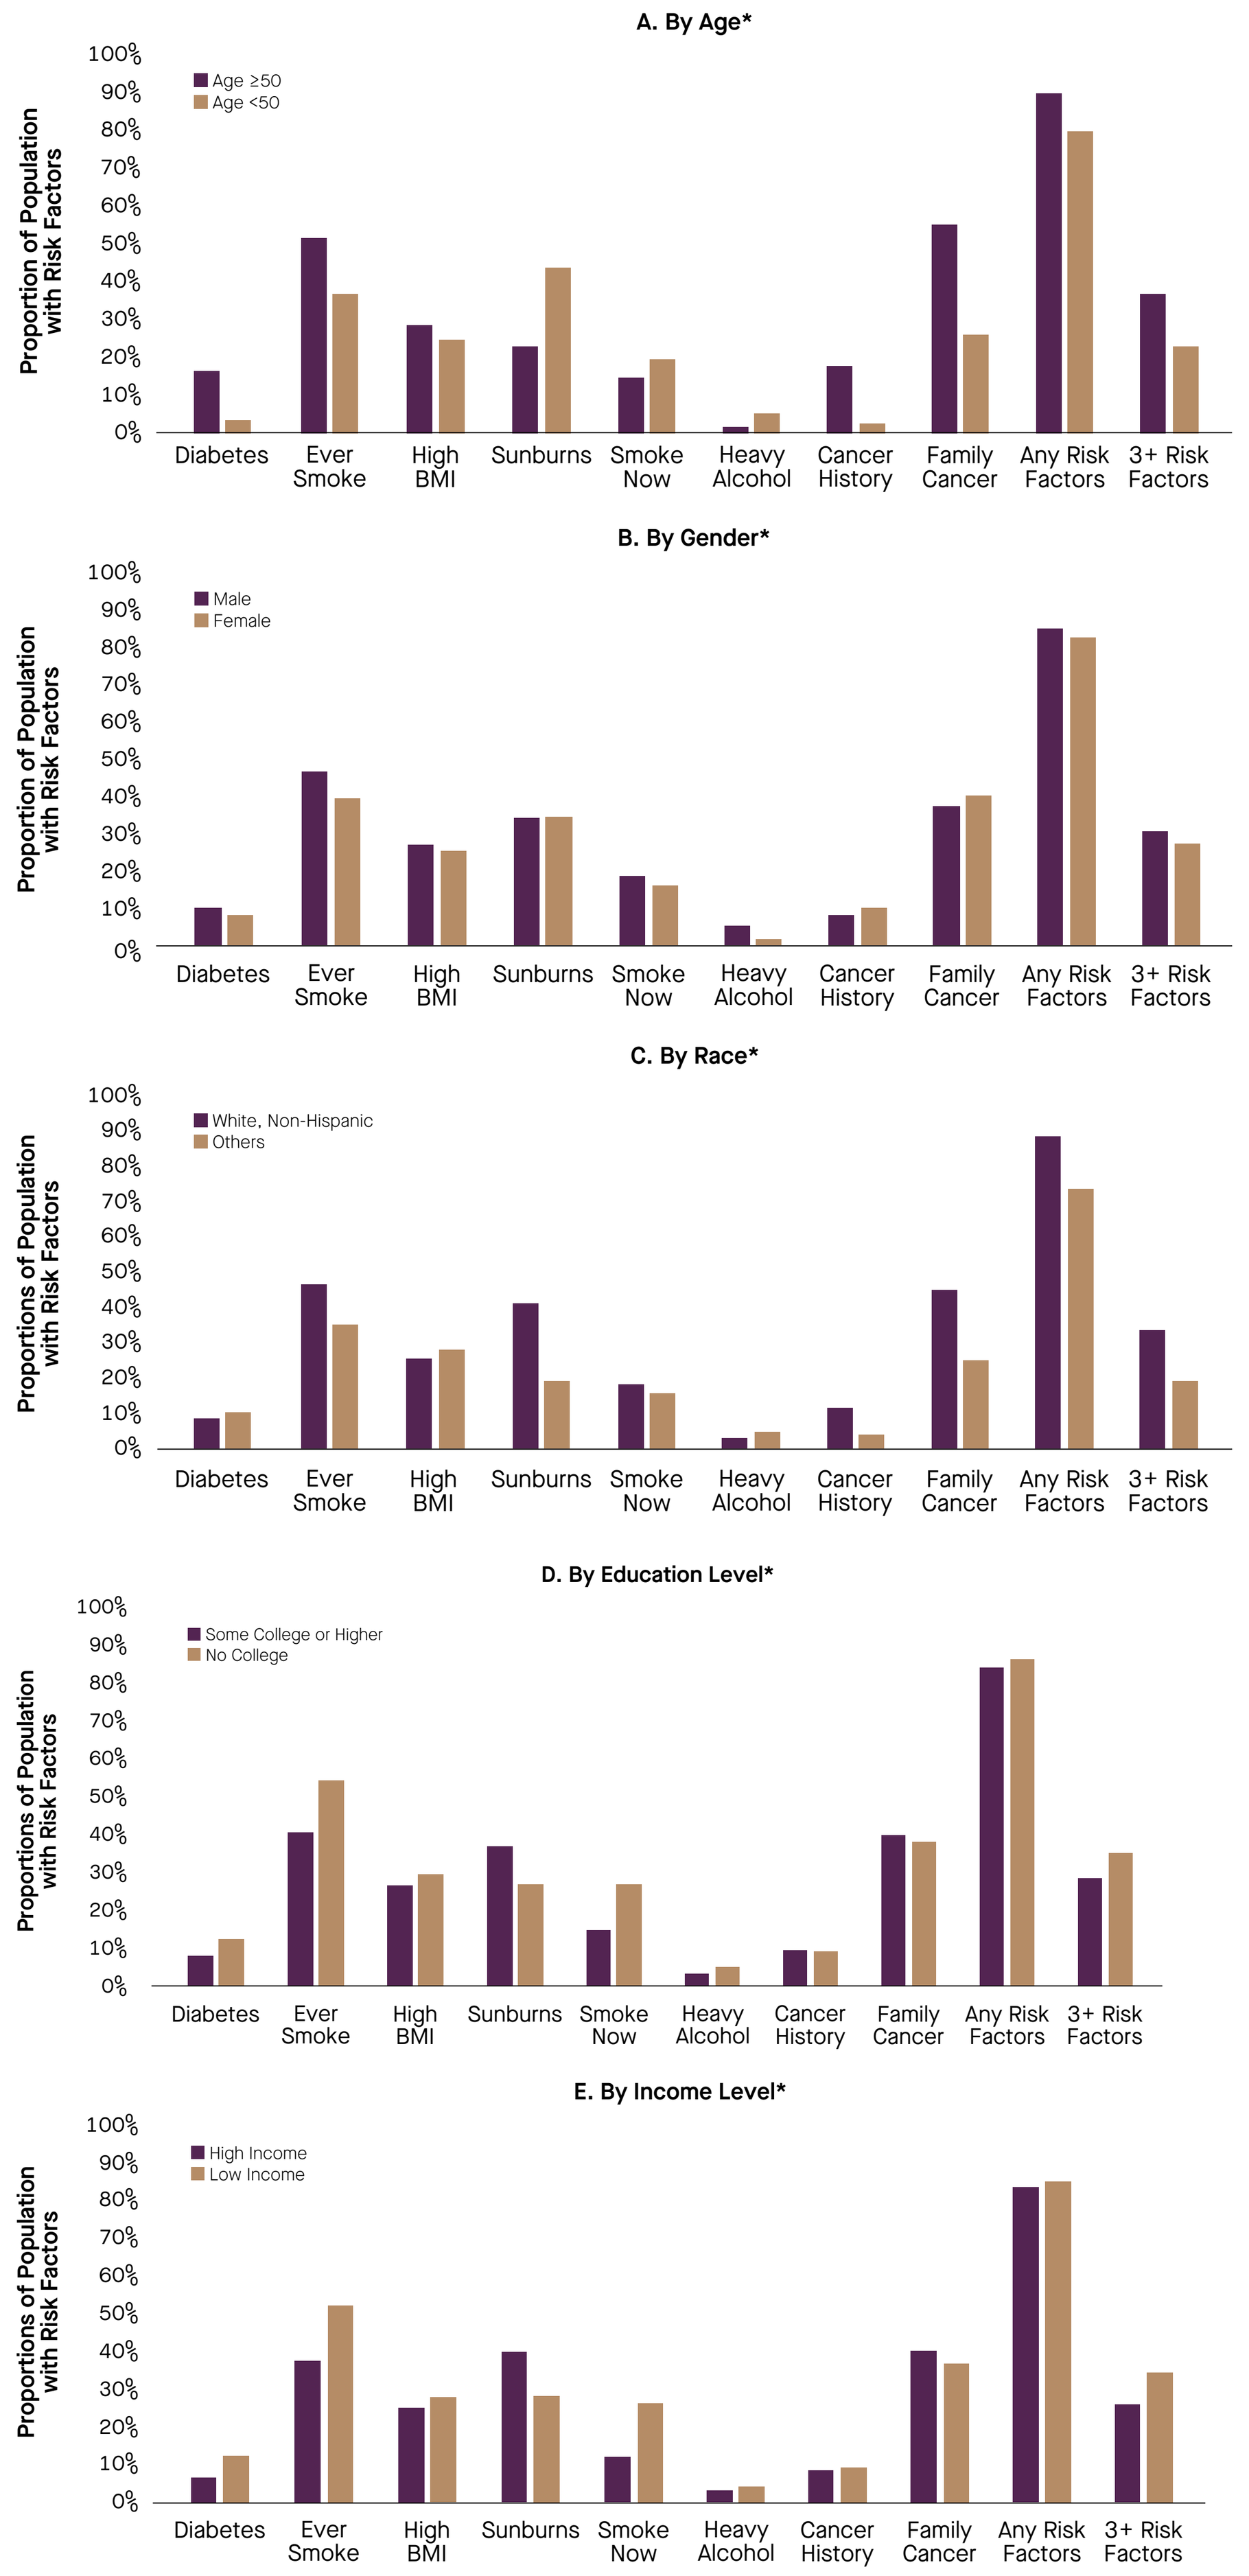
A. By Age*
Age ≥50
Age <50
B. By Gender*
Male
Female
C. By Race*
White, Non-Hispanic
Others
D. By Education Level*
Some College or Higher
No College
E. By Income Level*
High Income
Low Income
Proportion of Population with Risk Factors
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Diabetes
Ever Smoke
High BMI
Sunburns
Smoke Now
Heavy Alcohol
Cancer History
Family Cancer
Any Risk Factors
3+ Risk Factors

- Those aged ≥50 years were more likely to have risk factors for cancer, except for the risk factors of current smoking and heavy alcohol consumption
- Males were more likely to have risk factors for cancer, except for the sunburn risk factor
- The White, non-Hispanic population were more likely to be smokers, have sunburns, have a personal and family history of cancer, and more risk factors; they were also less likely to have diabetes, a high BMI, and heavy alcohol consumption
- The population without a college education were more likely to have diabetes, smoke, have a high BMI, heavy alcohol consumption, and more risk factors, whereas the population with some college education or more were more likely to have sunburns and a family history of cancer
- The population with low income levels were more likely to have diabetes, smoke, have a high BMI, heavy alcohol consumption, and more risk factors, whereas the population with high income levels were more likely to have sunburns and cancers in the family

*All differences between groups are statistically significantly different with p<0.01 except Sunburn and Cancer History.

Figure 4. Comorbidity Conditions Among Populations with Different Number of Risk Factors*

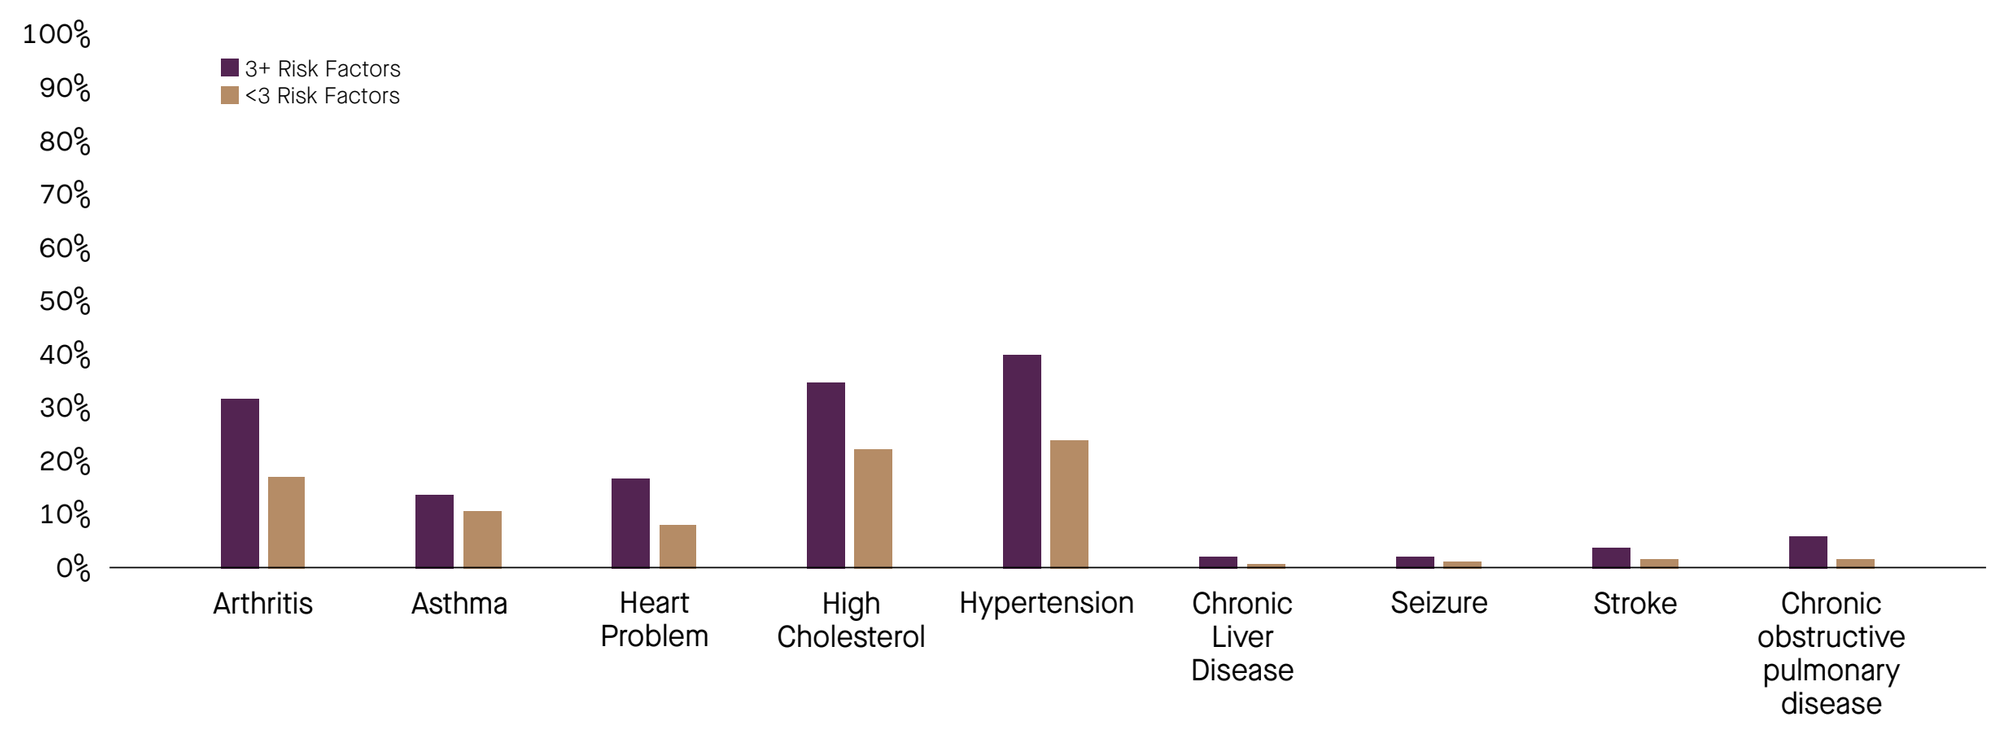
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
3+ Risk Factors
<3 Risk Factors
Arthritis
Asthma
Heart Problem
High Cholesterol
Hypertension
Chronic Liver Disease
Seizure
Stroke
Chronic obstructive pulmonary disease

- The population with 3 or more risk factors were more likely to have comorbid conditions

*All differences between groups are statistically significantly different with p<0.01.

CONCLUSIONS

- Risk factors for cancer are prevalent within the US adult population
- Across eight well-known cancer risk factors, over 200 million Americans or 84% of the adult population had at least one risk factor
- Those with a cancer risk factor tended to have other risk factors
 - Over 70 million or 29% of adult Americans have 3+ risk factors
- Individuals with cancer risk factors were more likely to be White non-Hispanic, older, male, with low income, low education, have ever received a cancer screening, and have comorbidities such as hypertension

IMPLICATIONS FOR POLICY AND PRACTICE

- The large number of Americans at risk for cancer suggests continued efforts are needed to address potentially modifiable risk factors such as smoking, high alcohol use, and sunburn
- Regardless of whether the behavior is modifiable, we need to ensure access to early screening, testing, and treatment
- The relationship between race and cancer risk factors is nuanced and should be used to help target public health messaging, screening, and treatment
 - Non-Hispanic White adults have a higher number of risk factors for cancer, while several risk factors are higher for non-white or Hispanic adults including diabetes, alcohol consumption, and body mass index
- Further policies are needed to address the disparity in the underlying risk factors and target screening and treatments to lower income and lower education individuals

References

1. Siegel RL, et al. *CA Cancer J Clin.* 2022;72(1):7-33.
2. Mariotto AB, et al. *Cancer Epidemiol Biomarkers Prev.* 2020;29(7):1304-1312.
3. Islami F, et al. *CA Cancer J Clin.* 2018;68(1):31-54.

Disclosures

Study funded by GRAIL, LLC. ZC and WC are employees of GRAIL, LLC. DCG received funding from GRAIL, LLC to conduct this study. All financial relationships disclosed at abstract submission.

Acknowlegements

Study funded by GRAIL, LLC. Graphic assistance provided by Prescott Medical Communications Group (Chicago, IL). Research assistance provided by Charles Hanzel.