Model Predictions for Lifetime Health Benefits of Mobocertinib and Current Treatment Options in Post-Platinum Patients with Locally Advanced or Metastatic NSCLC Harboring Epidermal Growth Factor Receptor Exon 20 Insertion Mutation in China

Genyong Zuo[1*], Yan Wang[2*], Yongbo Gao[3], Yan-Jun Zhang[3], Fanfan Zhu[3]

1. Center for Health Management and Policy Research, Shandong University, Jinan 250012, China; 2. Cancer Hospital, Chinese Academy of Medical Sciences, Beijing 100121, China; 3. Medical Affairs, Takeda (China) International Trading Company, Beijing 100027, China; * Zuo GY and Wang Y are co-first authors

CO214

INTRODUCTION

- Epidermal growth factor receptor (*EGFR*) mutation is a common pathogenic gene mutation in non-small cell lung cancer (NSCLC), and exon 20 insertion mutation (*EGFR*ex20ins) is a rare mutation, accounting for about 4%-12% of *EGFR* mutations [1].
- *EGFR*ex20ins exhibits high heterogeneity and is generally insensitive to most EGFR tyrosine kinase inhibitors (EGFR-TKIs). Chemotherapy regimens provide limited survival benefits that can not meet the medical needs in patients with *EGFR*ex20ins-mNSCLC.
- Mobocertinib is a first-in-class, potent, oral, irreversible TKI designed to selectively target *EGFR*ex20ins mutations. In September 2021, the FDA granted accelerated approval for mobocertinib to treat *EGFR*ex20ins-mNSCLC. In January 2023, China National Medical Products Administration approved mobocertinib for *EGFR*ex20ins-mNSCLC patients who had progressed on or after platinum-based chemotherapy.
- The evidence directly comparing mobocertinib with specific current treatments for previously treated patients with *EGFRe*x20ins-mNSCLC for health outcomes is lacking in China.

OBJECTIVES

- To estimate lifetime health benefits of mobocertinib and currently available treatments for Chinese patients with locally advanced or metastatic *EGFR*ex20ins-mNSCLC who had progressed on or after platinum-based chemotherapy.

Figure 1. METHODS: PARTITIONED SURVIVAL MODEL STRUCTURE

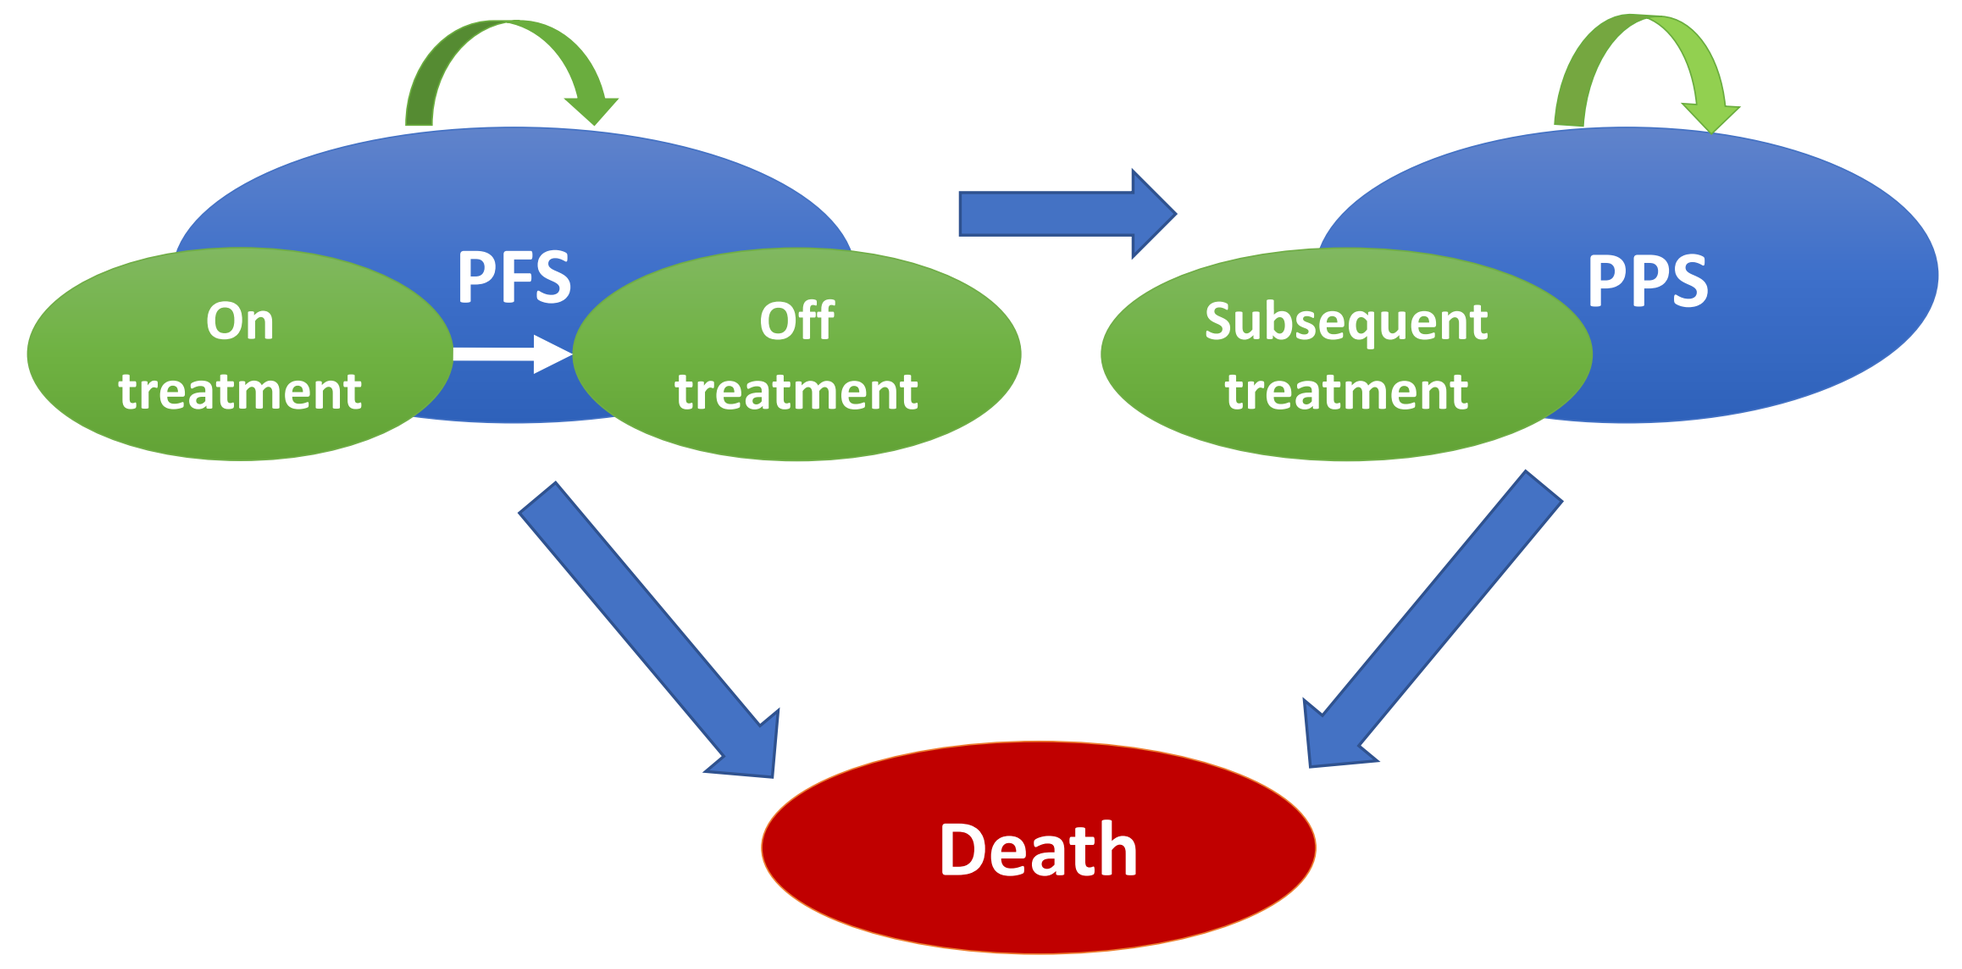
PFS
On treatment
Off treatment
PPS
Subsequent treatment
Death

METHODS

- The literature was searched to identify appropriate model inputs. Unadjusted single-arm meta-analysis was used to synthesize the identified evidence.
- If no efficacy and safety data for post-platinum EGFRex20ins-mNSCLC existed, evidence on same race (i.e., Asian) and second-line treatment of NSCLC was used.
- PFS and OS for mobocertinib were modeled using the lognormal and exponential distribution, respectively, as they had the lowest AIC (akaike information criterion) and BIC (bayesian information criterion) based on parametric survival analysis of the participants from Asian race in the mobocertinib trial. Due to lack of patient-level data for the comparators, their reported median PFS and OS were used to assume an exponential distribution.
- One-way and probabilistic sensitivity analyses (OWSA and PSA) were performed to assess the robustness of the results and parameters' uncertainty.

Table 1. COST-EFFECTIVENESS MODEL DESIGN

Model cohort	Chinese patients with *EGFR*ex20ins-mNSCLC who had progressed on or after platinum-based chemotherapy
Treatment regimen for assessment	mobocertinib
Control treatment regimens*	DTX/CDDP, DTX/CDDP/BEV, PEM/CDDP, PEM/CDDP/BEV, and high-dose osimertinib
Model outputs	Life years and QALYs
Model design	Partitioned survival model
Health states	PF, PP, and death
Adverse events+	Treatment-emergent adverse event grade≥3
Model cycle length	1 week
Time horizon	Lifetime

*BEV: bevacizumab; CDDP: cisplatin; DTX: docetaxel; PEM: pemetrexed; PF: progression-free; PP: post-progression; QALYs: quality-adjusted life years; The treatments are recommended by the CSCO or NCCN guidelines, or used in China. +AEs include leukopenia, constipation, hyponatraemia, nausea/vomiting, diarrhea, hepatic function abnormal, infection, hypertension, spinal-cord compression, interstitial pneumonia, stomatitis, rash, fatigue, anaemia, kidney function abnormal, decreased appetite, infusion related reaction, edema, alopecia, neuropathy peripheral, bacterial pneumonia, thrombocytopenia.

Table 2. MODEL INPUTS SUMMARY

Model inputs	Baseline	Model inputs	Baseline
Demographics		***Treatment efficacy***	
Age (years)	59.4	*Median OS (months)*	
Male (%)	34.5%	Mobocertinib	25.26
Body weight (kg)	60	DTX/CDDP	9.25[2-4,6-8,10]
Body surface area (m^2)	1.6	DTX/CDDP/BEV	11.65[11]
Treatment efficacy+		PEM/CDDP	13.46[2,15-19]
Median PFS (months)		PEM/CDDP/BEV	13.84[2,12,15-19]
Mobocertinib	7.59	High-dose osimertinib	15.02[20,22]
DTX/CDDP	2.83[2-9]		
DTX/CDDP/BEV	3.75[11-14]	***Quality of life (utility)***	
PEM/CDDP	3.39[2,15-19]	PFS	0.851
PEM/CDDP/BEV	3.75[11-14]	PPS	0.810
High-dose osimertinib	2.58[20-22]	disutility due to treatment-emergent adverse event grade≥3	0.048

* PFS: progression-free survival; OS: overall survival; + Mobocertinib is for Asian population (data cutoff: November 2020), its median PFS and OS are derived from lognormal and exponential distribution, respectively; Comparators are for Asian population (Chinese, Japanese, etc.) .

RESULTS

- In the base-case analysis (**Figure 3**), mobocertinib could gain 0.722 (vs. PEM/ CDDP/BEV) to 0.862 years (vs. high-dose osimertinib) of PFLYs, 1.269 (vs. high-dose osimertinib) to 1.993 years (vs. DTX/CDDP) of PPLYs, and 1.731 (vs. high-dose osimertinib) to 2.290 incremental QALYs (vs. DTX/CDDP).
- The OWSA showed that the incremental QALYs were mainly driven by additional PFLYs and PPLYs, the influential parameters included survival outcomes (PFS and overall survival) of the compared treatments and the utility of health states (**Figure 4**). Mobocertinib has the probability of 100.0% vs. all the comparators to gain more health benefits (total LYs and QALYs) based on the PSA (**Table 3**).

Figure 2. SIMULATED SURVIVAL CURVES

Figure 2a. Estimated PFS curves

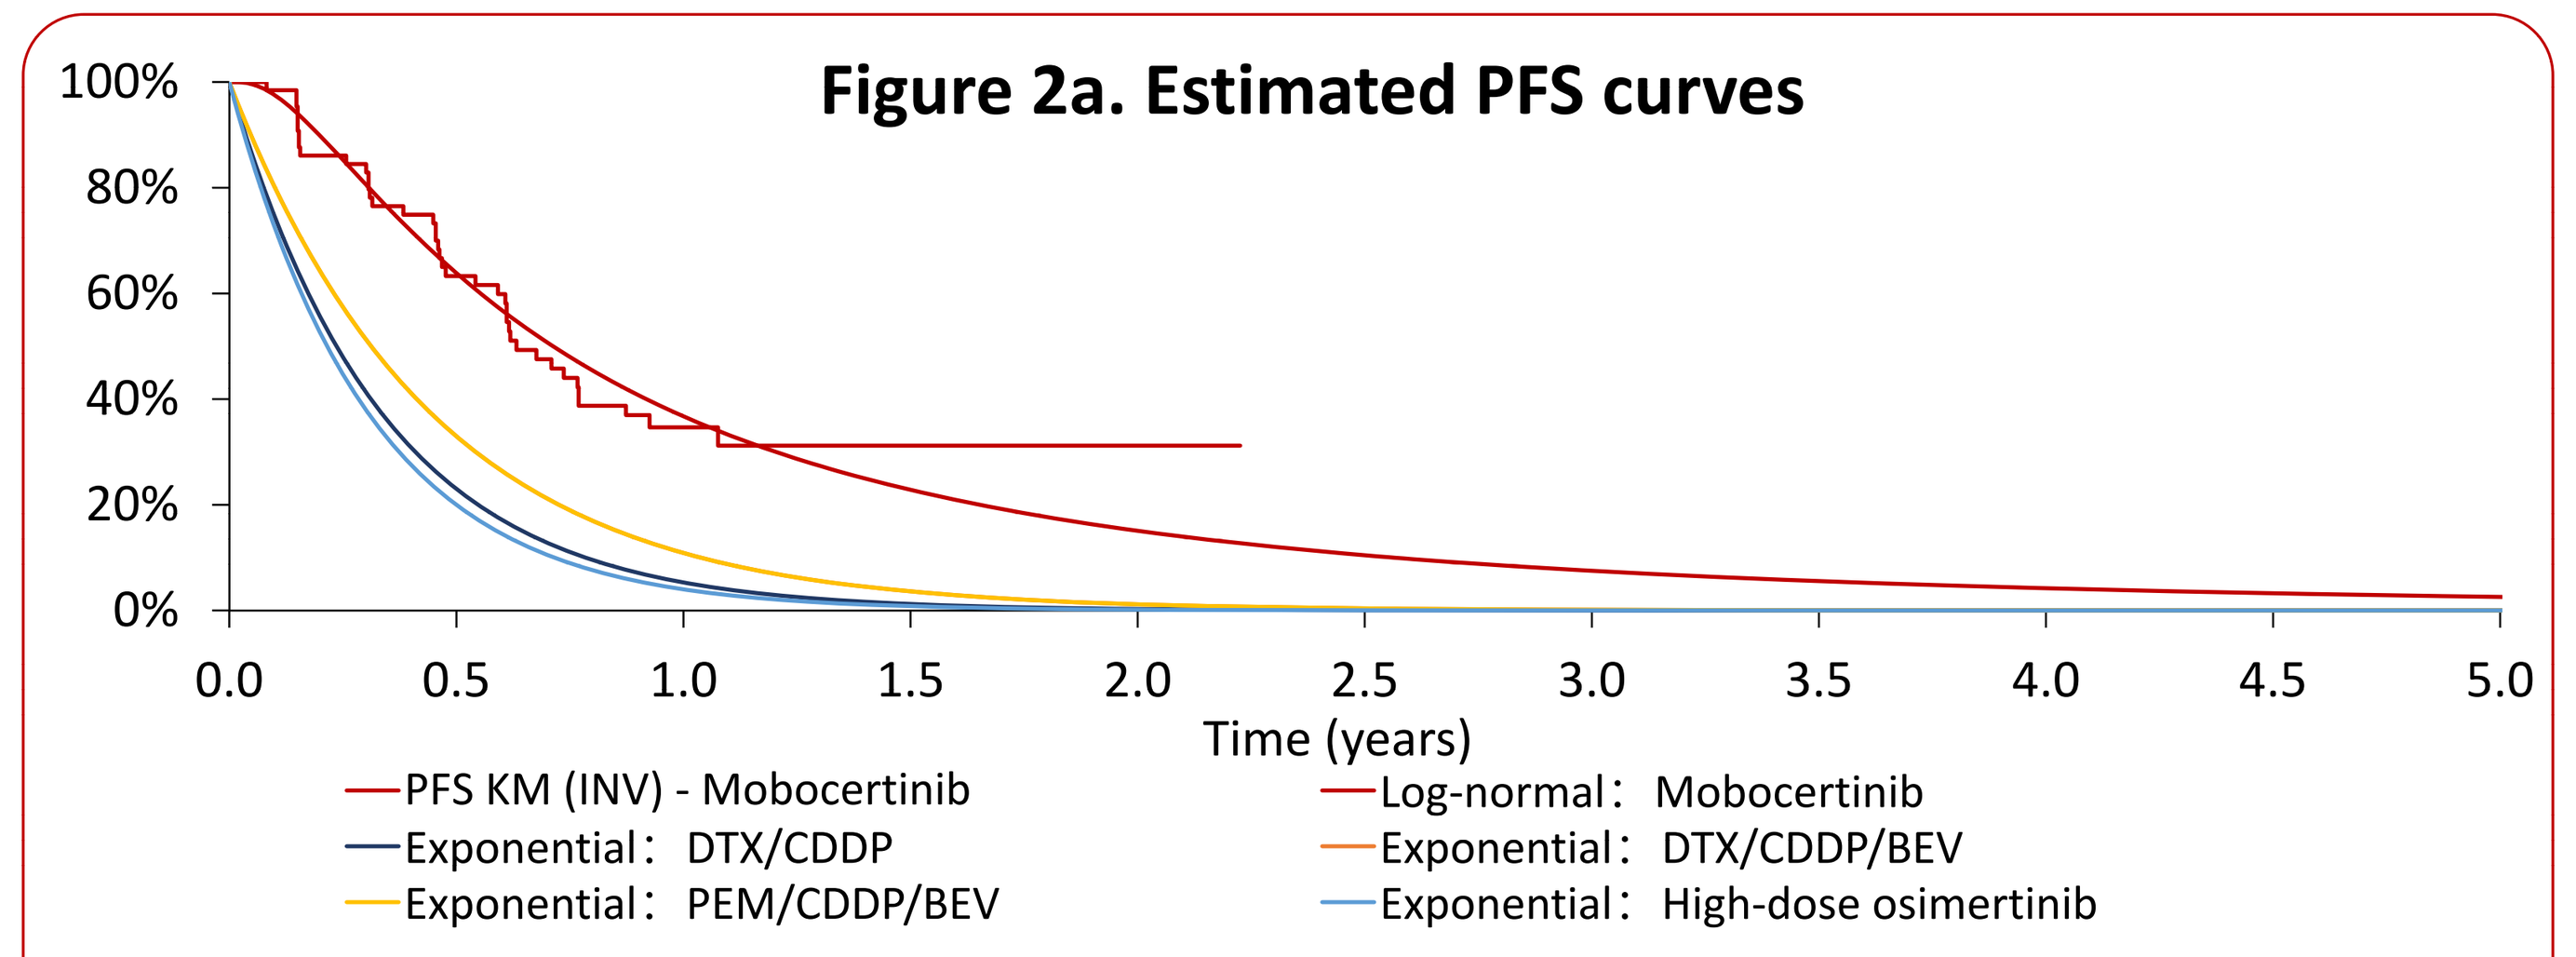
100%
80%
60%
40%
20%
0%
0.0 0.5 1.0 1.5 2.0 2.5 3.0 3.5 4.0 4.5 5.0
Time (years)
PFS KM (INV) - Mobocertinib
Log-normal： Mobocertinib
Exponential： DTX/CDDP
Exponential： DTX/CDDP/BEV
Exponential： PEM/CDDP/BEV
Exponential： High-dose osimertinib

Figure 2b. Estimated OS curves

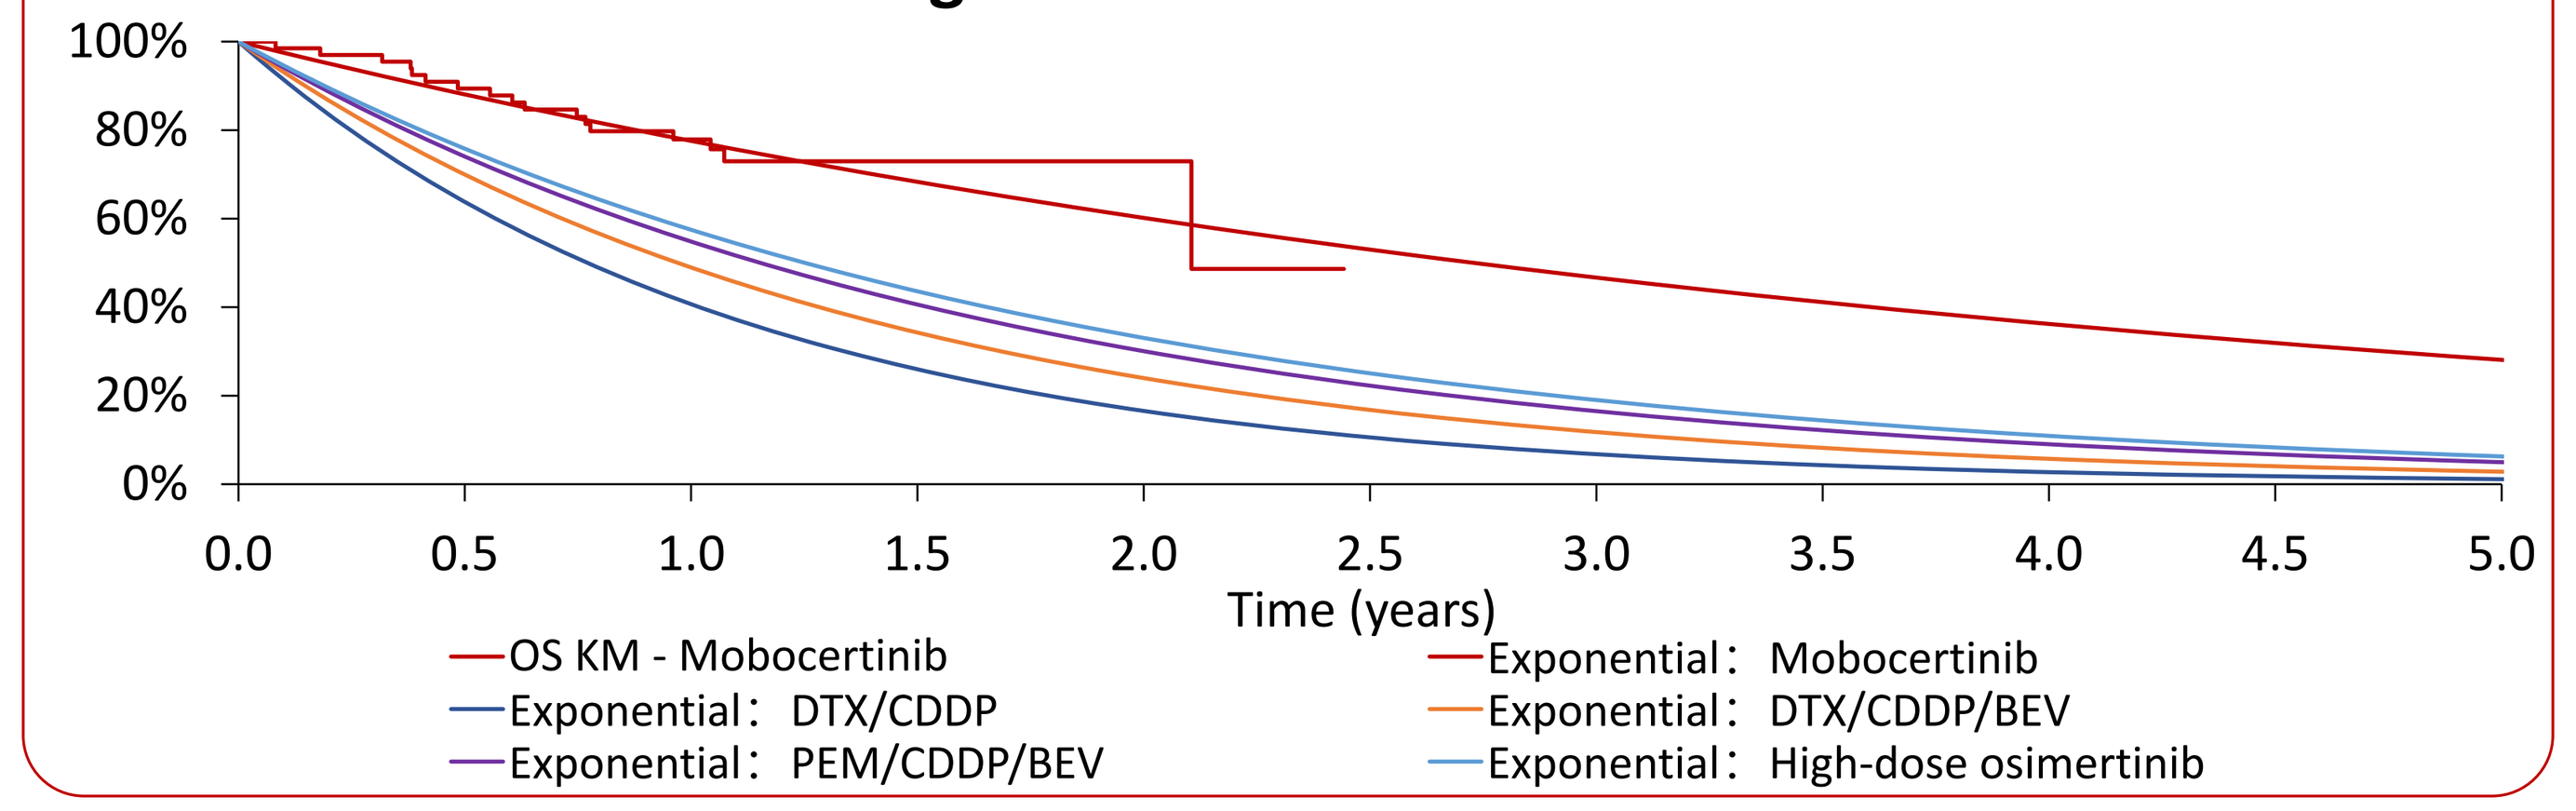
100%
80%
60%
40%
20%
0%
0.0 0.5 1.0 1.5 2.0 2.5 3.0 3.5 4.0 4.5 5.0
Time (years)
OS KM - Mobocertinib
Exponential： Mobocertinib
Exponential： DTX/CDDP
Exponential： DTX/CDDP/BEV
Exponential： PEM/CDDP/BEV
Exponential： High-dose osimertinib

Figure 3. BASE CASE ANALYSIS

Figure 3a. Total life years

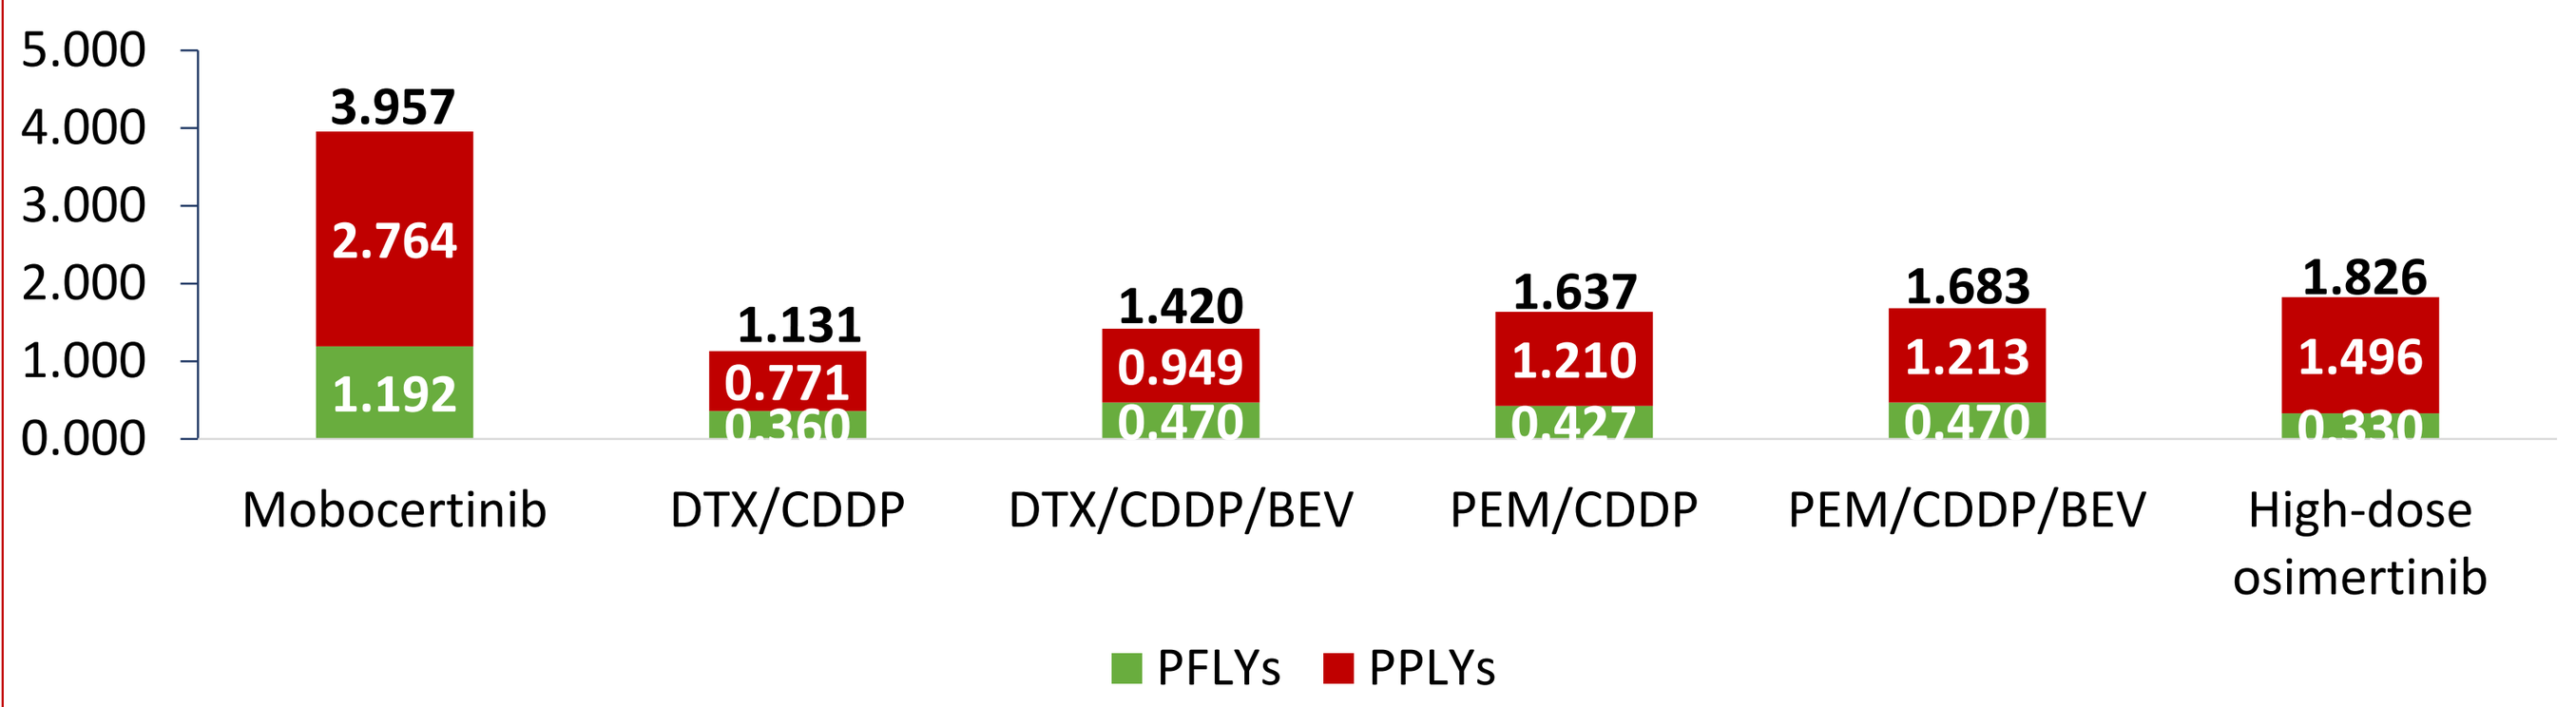
5.000
4.000
3.000
2.000
1.000
0.000
3.957
2.764
1.192
Mobocertinib
1.131
0.771
0.360
DTX/CDDP
1.420
0.949
0.470
DTX/CDDP/BEV
1.637
1.210
0.427
PEM/CDDP
1.683
1.213
0.470
PEM/CDDP/BEV
1.826
1.496
0.330
High-dose osimertinib
PFLYs
PPLYs

*PFLYs: progression-free life years; PPLYs: post-progression life years.

Figure 3b. Total QALYs

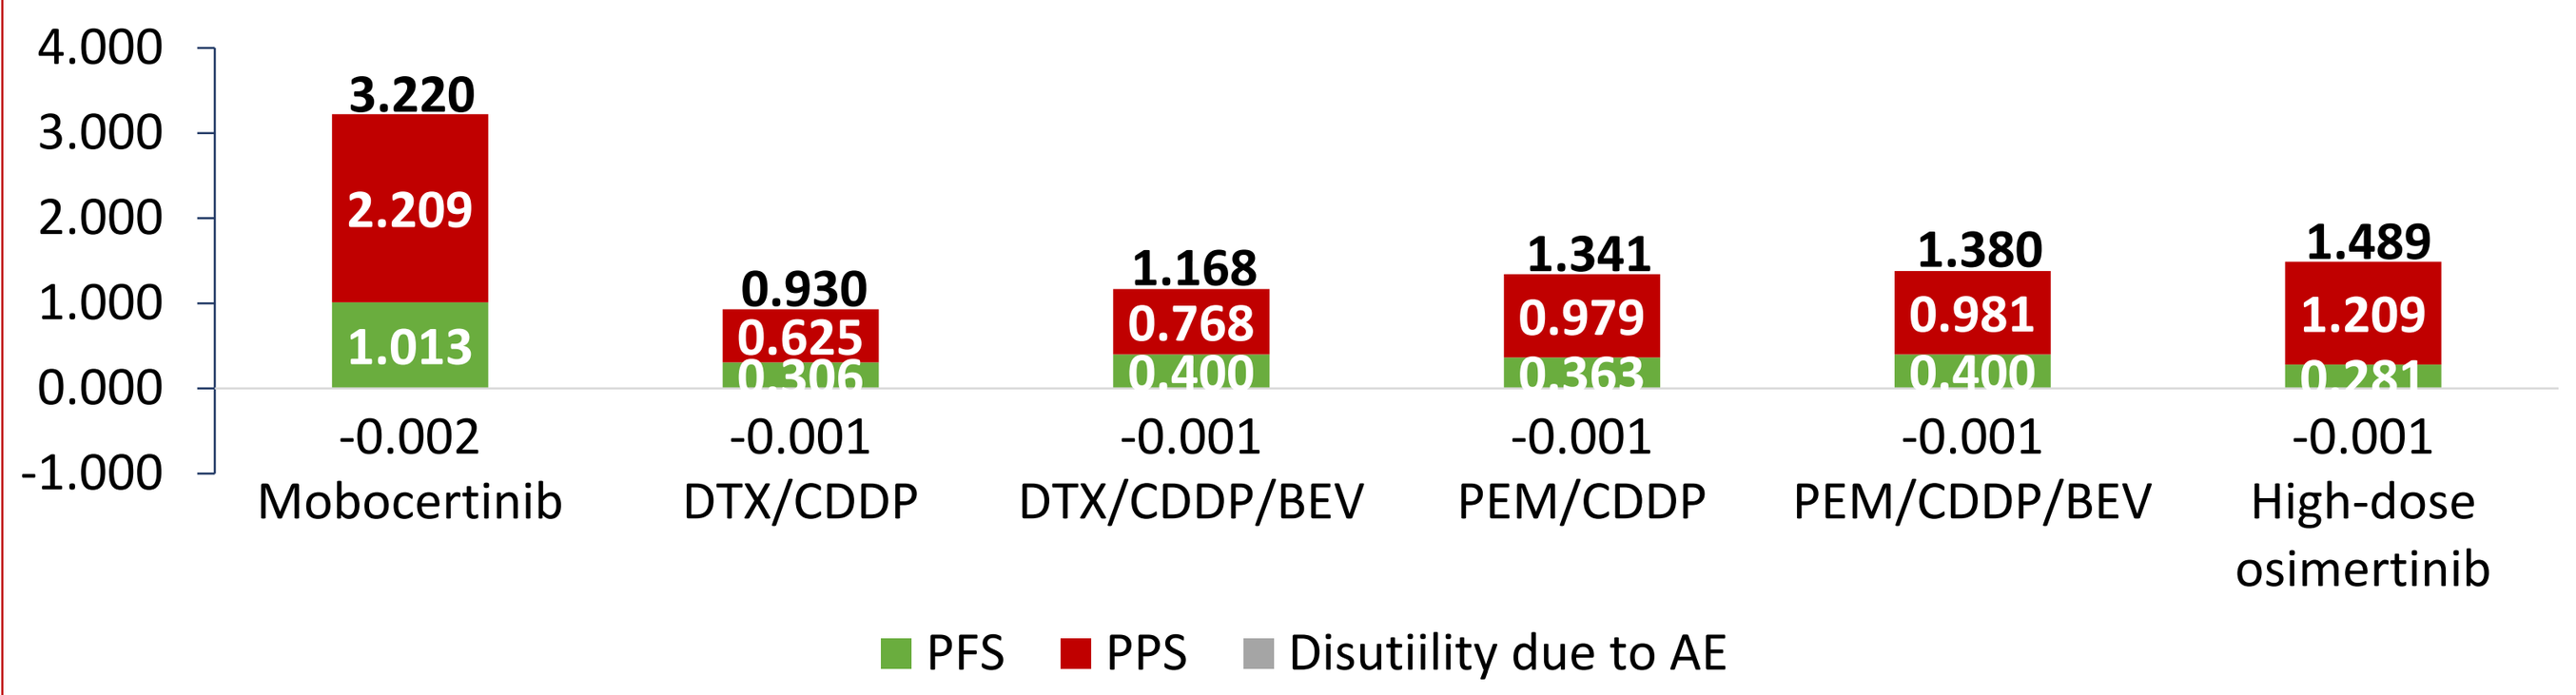
4.000
3.000
2.000
1.000
0.000
-1.000
3.220
2.209
1.013
-0.002
Mobocertinib
0.930
0.625
0.306
-0.001
DTX/CDDP
1.168
0.768
0.400
-0.001
DTX/CDDP/BEV
1.341
0.979
0.363
-0.001
PEM/CDDP
1.380
0.981
0.400
-0.001
PEM/CDDP/BEV
1.489
1.209
0.281
-0.001
High-dose osimertinib
PFS
PPS
Disutiility due to AE

*Mobocertinib had slightly higher disutility due to AEs (adverse events), for it provided additional treatment time for patients to benefit from the efficacy meanwhile a longer time to be exposed for the risk of AEs..

Figure 4. ONE-WAY SENSITIVITY ANALYSIS (OWSA)

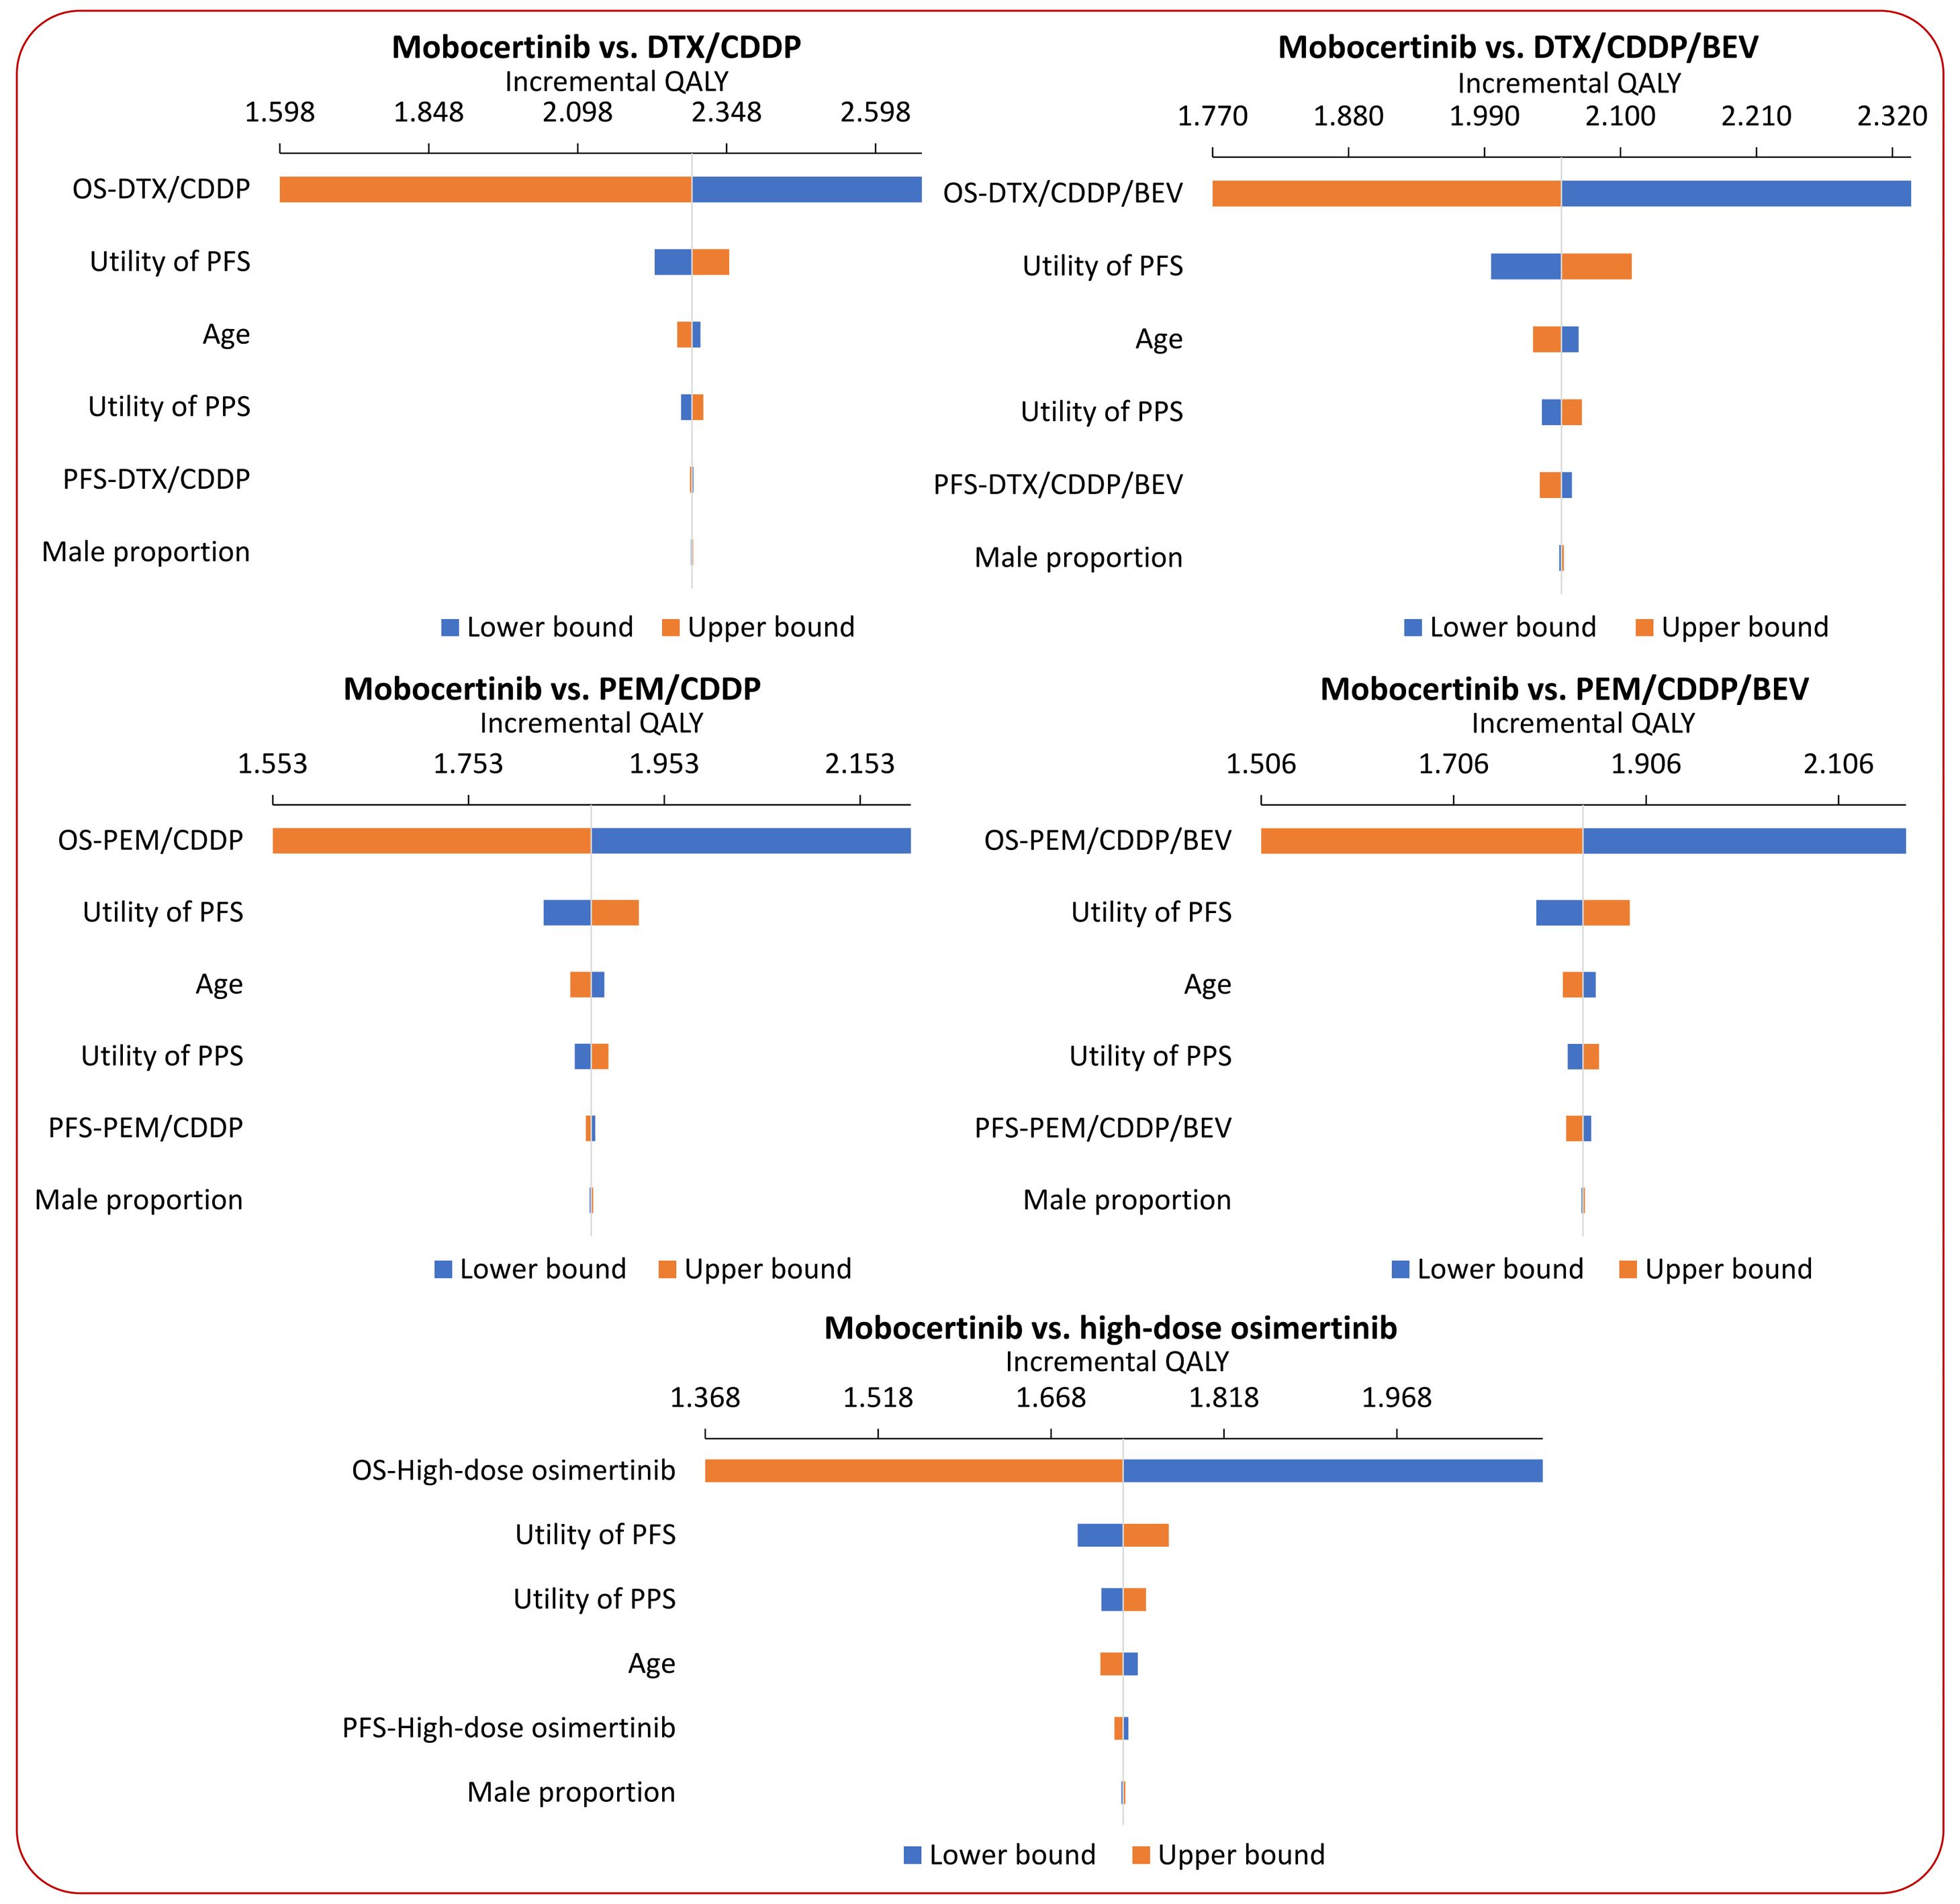
Mobocertinib vs. DTX/CDDP
Incremental QALY
1.598 1.848 2.098 2.348 2.598
OS-DTX/CDDP
Utility of PFS
Age
Utility of PPS
PFS-DTX/CDDP
Male proportion
Lower bound
Upper bound
Mobocertinib vs. DTX/CDDP/BEV
Incremental QALY
1.770 1.880 1.990 2.100 2.210 2.320
OS-DTX/CDDP/BEV
Utility of PFS
Age
Utility of PPS
PFS-DTX/CDDP/BEV
Male proportion
Lower bound
Upper bound
Mobocertinib vs. PEM/CDDP
Incremental QALY
1.553 1.753 1.953 2.153
OS-PEM/CDDP
Utility of PFS
Age
Utility of PPS
PFS-PEM/CDDP
Male proportion
Lower bound
Upper bound
Mobocertinib vs. PEM/CDDP/BEV
Incremental QALY
1.506 1.706 1.906 2.106
OS-PEM/CDDP/BEV
Utility of PFS
Age
Utility of PPS
PFS-PEM/CDDP/BEV
Male proportion
Lower bound
Upper bound
Mobocertinib vs. high-dose osimertinib
Incremental QALY
1.368 1.518 1.668 1.818 1.968
OS-High-dose osimertinib
Utility of PFS
Utility of PPS
Age
PFS-High-dose osimertinib
Male proportion
Lower bound
Upper bound

Table 3. PROBABILISTIC SENSITIVITY ANALYSIS (PSA)

Mobocertinib vs.	Incremental life years			Incremental QALYs		
	MEDIAN	95%CI		MEDIAN	95%CI	
		Lower	Upper		Lower	Upper
DTX/CDDP	2.970	1.508	5.335	2.405	1.241	4.275
DTX/CDDP/ BEV	2.548	1.084	4.916	2.060	0.901	3.910
PEM/CDDP	2.331	0.867	4.698	1.885	0.726	3.738
EPM/CDDP /BEV	2.285	0.821	4.652	1.848	0.689	3.701
High-dose osimertinib	2.142	0.678	4.510	1.737	0.577	3.594

- Based on 5,000 Monte-Carlo simulation results

CONCLUSIONS

- Mobocertinib had additional lifetime health benefits (PFLYs, PPLYs, LYs and QALYs) compared with existing, post-platinum treatments of *EGFR*ex20ins-mNSCLC in China. The model outputs were robust to variations in the OWSA and PSA.

DISCLOSURE AND ACKNOWLEDGEMENTS

- Yongbo Gao, Yan-Jun Zhang and Fanfan Zhu are employees of Takeda (China) International Trading Company, and hold Takeda stocks. This study was funded by Takeda (China) International Trading Company.
- Medical writing assistance was provided by Qianyi Gong and Wendong Chen of Changsha Normin Health Technology Ltd. and funded by Takeda (China) International Trading Company.

REFERENCES: **1**. Riess JW, Gandara DR, Frampton GM, et al. J Thorac Oncol. 2018, 13(10):1560-1568.; **2.** Kato R, et al. J Immunother Cancer. 2020;8(1):e000350.; **3.** Kawaguchi T, et al. J Clin Oncol. 2014;32(18):1902-1908.; **4.** Liu QC, et al. Chin J Cancer Prev Treat. 2016;23(6):389-93.; **5.**He Y, Fan QX. CSMO. Beijing,2014. p. 535; **6.** Miura S, et al. Oncol Lett. 2011;2(6):1059-1064.; **7.** Yang JC, et al. JTO Clin Res Rep. 2021;2(3):100142.; **8.** Sugawara S, et al. Int J Clin Oncol. 2019;24(5):485-493.; **9.** Kasahara K, et al. Eur J Cancer. 2015;51: S620.; **10.** Wang LL, et al. Oncol Progress, 2018, 16(15):4.; **11.** Jiang H, Cheng Y. J Thorac Oncol. 2018;13(12 Supplement):S1061.; **12.** Yang G, et al. Lung Cancer. 2020;145:186-194.; **13.** Naoki K, et al. Oncol Lett. 2017;13(5):3285-3290.; **14.** Li SY. Suzhou University. 2017.; **15.** Sun JM, et al. Cancer. 2012;118: 6234-42.; **16.** Hattori Y, et al. Cancer Chemother Pharmacol. 2014;73(1):17-23.; **17.** Sato A, et al. J Thorac Oncol. 2011;6: S1213-S4.; **18.** Katakami N, et al. J Thorac Oncol. 2012;7: S73.; **19.** Hong T, et al. J Cancer Res Clin Oncol. 2012;138(2):285-291.; **20.** Eiki I, et al. AACR 2020.; **21.** Yang GJ, et al. Lung Cancer. 2021;152:39-48.; **22.** Kim TM, et al. Ann Oncol. 2019, 30(Supplement_5):v628.;