A Systematic Literature Review to Assess the Level of Evidence in Facioscapulohumeral Muscular Dystrophy

SA7

Lianne Barnieh[1], Rachel Beckerman[1], Helena Emich[1], Katy Eichinger[2], **Adi Eldar-Lissai[3]**

[1]Maple Health Group, LLC, New York, NY, USA; [2]University of Rochester Medical Center, Rochester, NY, USA; [3]Fulcrum Therapeutics, Inc., Cambridge, MA, USA

BACKGROUND

- Facioscapulohumeral muscular dystrophy (FSHD) is a rare genetic muscle disorder that manifests itself as a progressive weakening and loss of skeletal muscles.[1]
- FSHD is caused by aberrant expression of *DUX4* in skeletal muscle, which leads to death of the muscle and its replacement by fat, resulting in skeletal muscle weakness and progressive disability.[2]
- At disease onset, muscles of the face (facio), shoulder girdle (scapulo), and upper arms (humeral) are primarily affected. As the disease progresses, muscle weakness and loss spreads to the arms, trunk and lower body.[2]
- Currently, there are no approved therapies for FSHD. Disease management focuses on supportive treatments with physical therapy playing a key role.[3]
- The study systematically reviewed and synthesized the literature to gain a better understanding of the available evidence on the burden of and unmet need in FSHD.

METHODS

- A systematic literature review (SLR) was conducted in accordance with the methodological principles of conduct.[4] The results of this review are reported according to the PRISMA guidelines.
- Search strategies were executed from inception to October 11, 2022, in three electronic databases: Embase, MEDLINE, and Cochrane. Searches were restricted to English publications. Electronic searches were supplemented with manual searches of relevant, non-indexed conference proceedings, limited to the last three years.
- Criteria for study inclusion were adults, adolescents, or children with FSHD, type 1 or type 2; studies reporting on one of the topics of interest (efficacy, safety, humanistic, economic outcomes; validity of outcome measures in FSHD; humanistic burden; economic burden; disease diagnosis; or disease classification); clinical trials, observational studies (including case reports and case studies), or endpoint validation studies; and English language.
- Study selection, first by title and abstract screening and then by performing full text review, was conducted by two independent reviewers, with conflicts resolved by a third reviewer.

CONCLUSIONS

- **Despite being the second most prevalent form of muscular dystrophy, FSHD is not well characterized in the literature.**
- **The lack of studies depicting the humanistic and economic burden makes it difficult to assess the true disease burden experienced by patients, their families, and the healthcare system.**
- **Only a few studies on diagnosis and disease management guidelines were identified, potentially explaining the variability in time to diagnosis and treatment patterns experienced by patients.**
- **Multiple unique outcomes to assess disease progression were identified, suggesting an opportunity to build consensus around both *what* and *how* to measure treatment benefit.**

RESULTS

- The database and manual searches yielded 2,211 full text articles and 754 conference abstracts after de-duplication. A total of 101 conference abstracts and 153 full text articles met inclusion criteria for data extraction (**Figure 1**).

Figure 1. PRISMA Flow Diagram

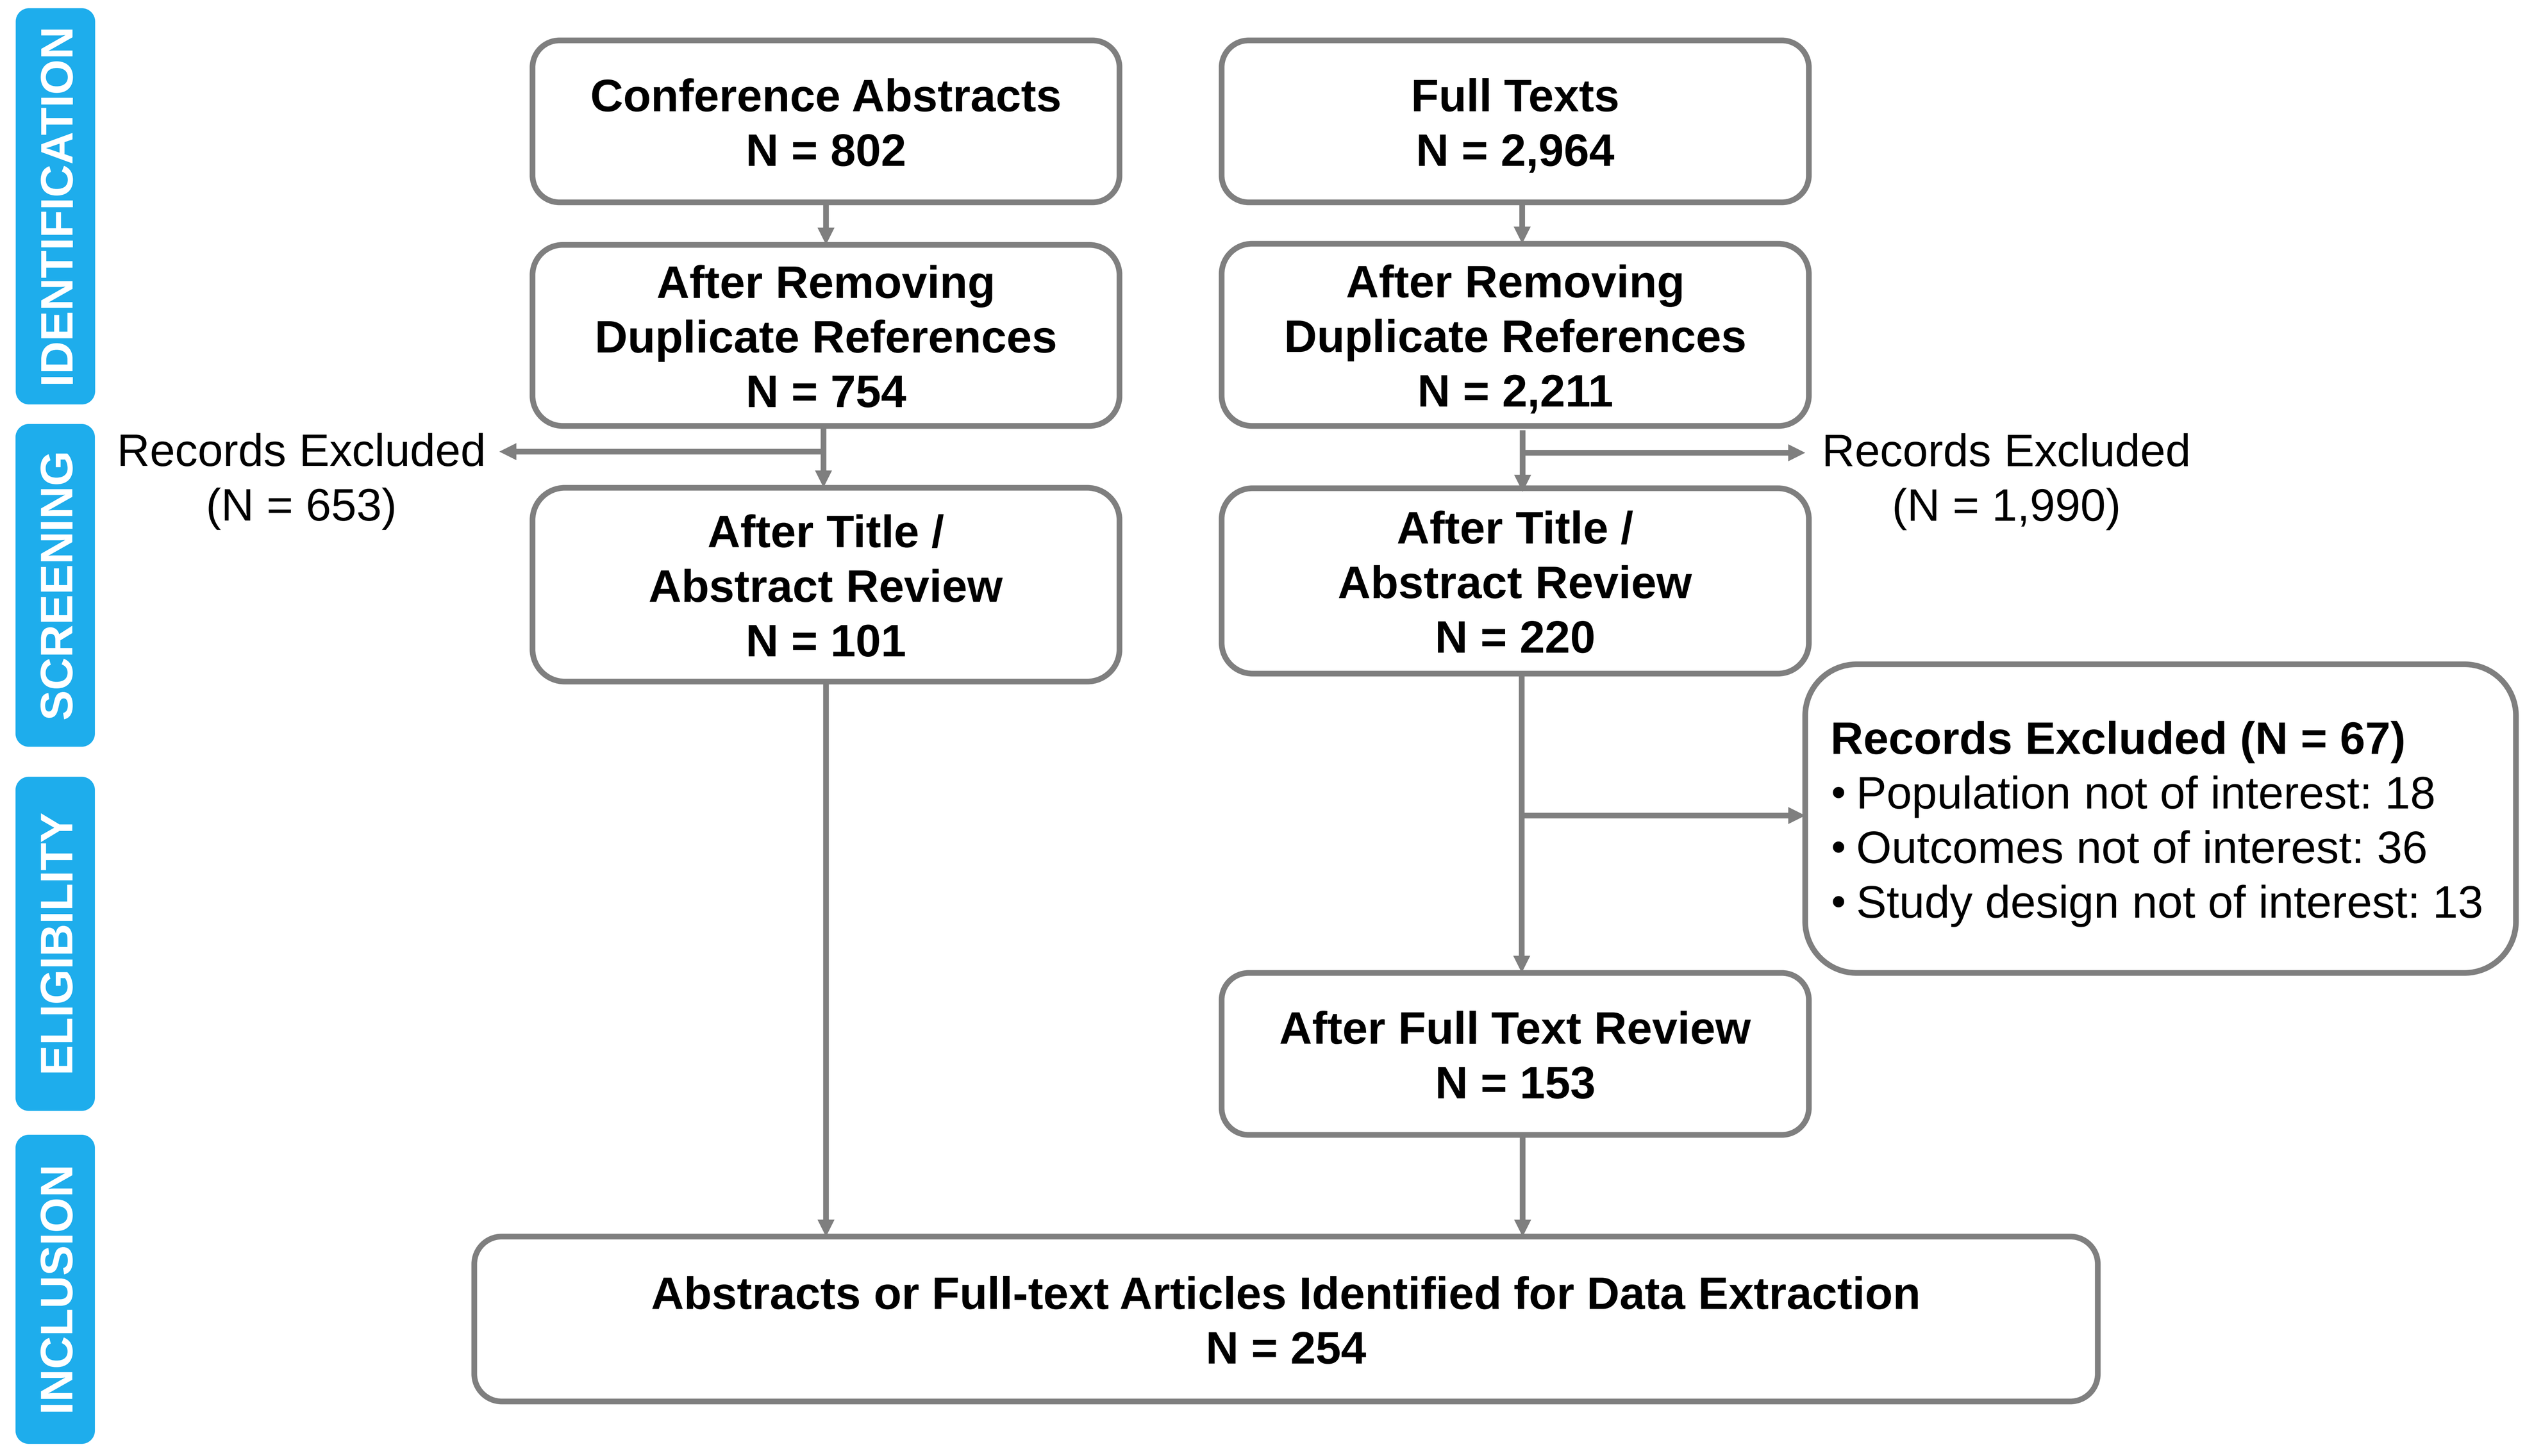
IDENTIFICATION
SCREENING
ELIGIBILITY
INCLUSION
Conference Abstracts N = 802
Full Texts N = 2,964
After Removing Duplicate References N = 754
After Removing Duplicate References N = 2,211
Records Excluded (N = 653)
Records Excluded (N = 1,990)
After Title / Abstract Review N = 101
After Title / Abstract Review N = 220
Records Excluded (N = 67)
• Population not of interest: 18
• Outcomes not of interest: 36
• Study design not of interest: 13
After Full Text Review N = 153
Abstracts or Full-text Articles Identified for Data Extraction N = 254

- The most reported topics included "Outcome measures and validation" (n=75), "Treatment outcomes and clinical trials (n=67), and "Disease classification" (n=47) (**Figure 2**).

Figure 2. Topics Included in Full Text Articles and Conference Abstracts

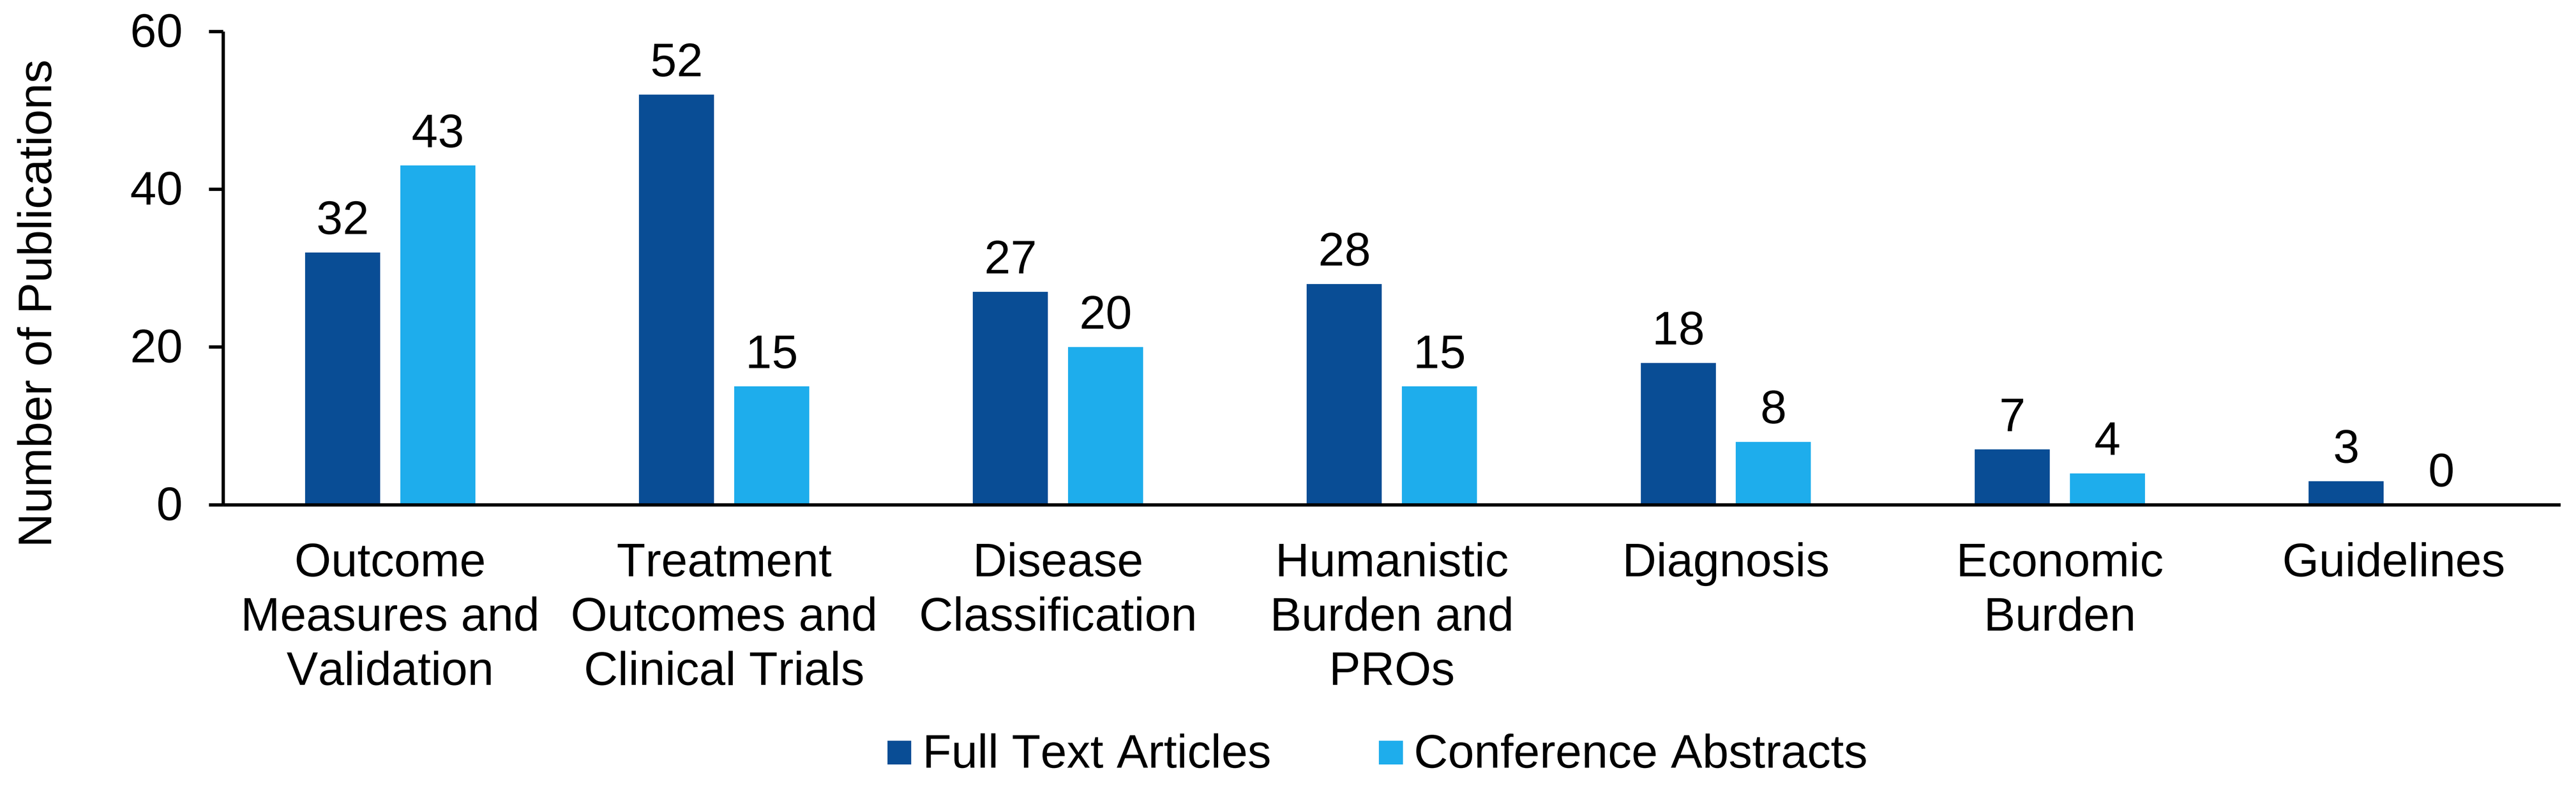
Number of Publications
60
40
20
0
32 43
52 15
27 20
28 15
18 8
7 4
3 0
Outcome Measures and Validation
Treatment Outcomes and Clinical Trials
Disease Classification
Humanistic Burden and PROs
Diagnosis
Economic Burden
Guidelines
Full Text Articles
Conference Abstracts

*Numbers do not sum to 254 as some studies reported on two or more topics.

- Among studies included in the topic **"Treatment outcomes and clinical trials"**, the most identified study design was clinical trials (**Figure 3**).

Figure 3. Study Designs in "Treatment Outcomes and Clinical Trials" Studies

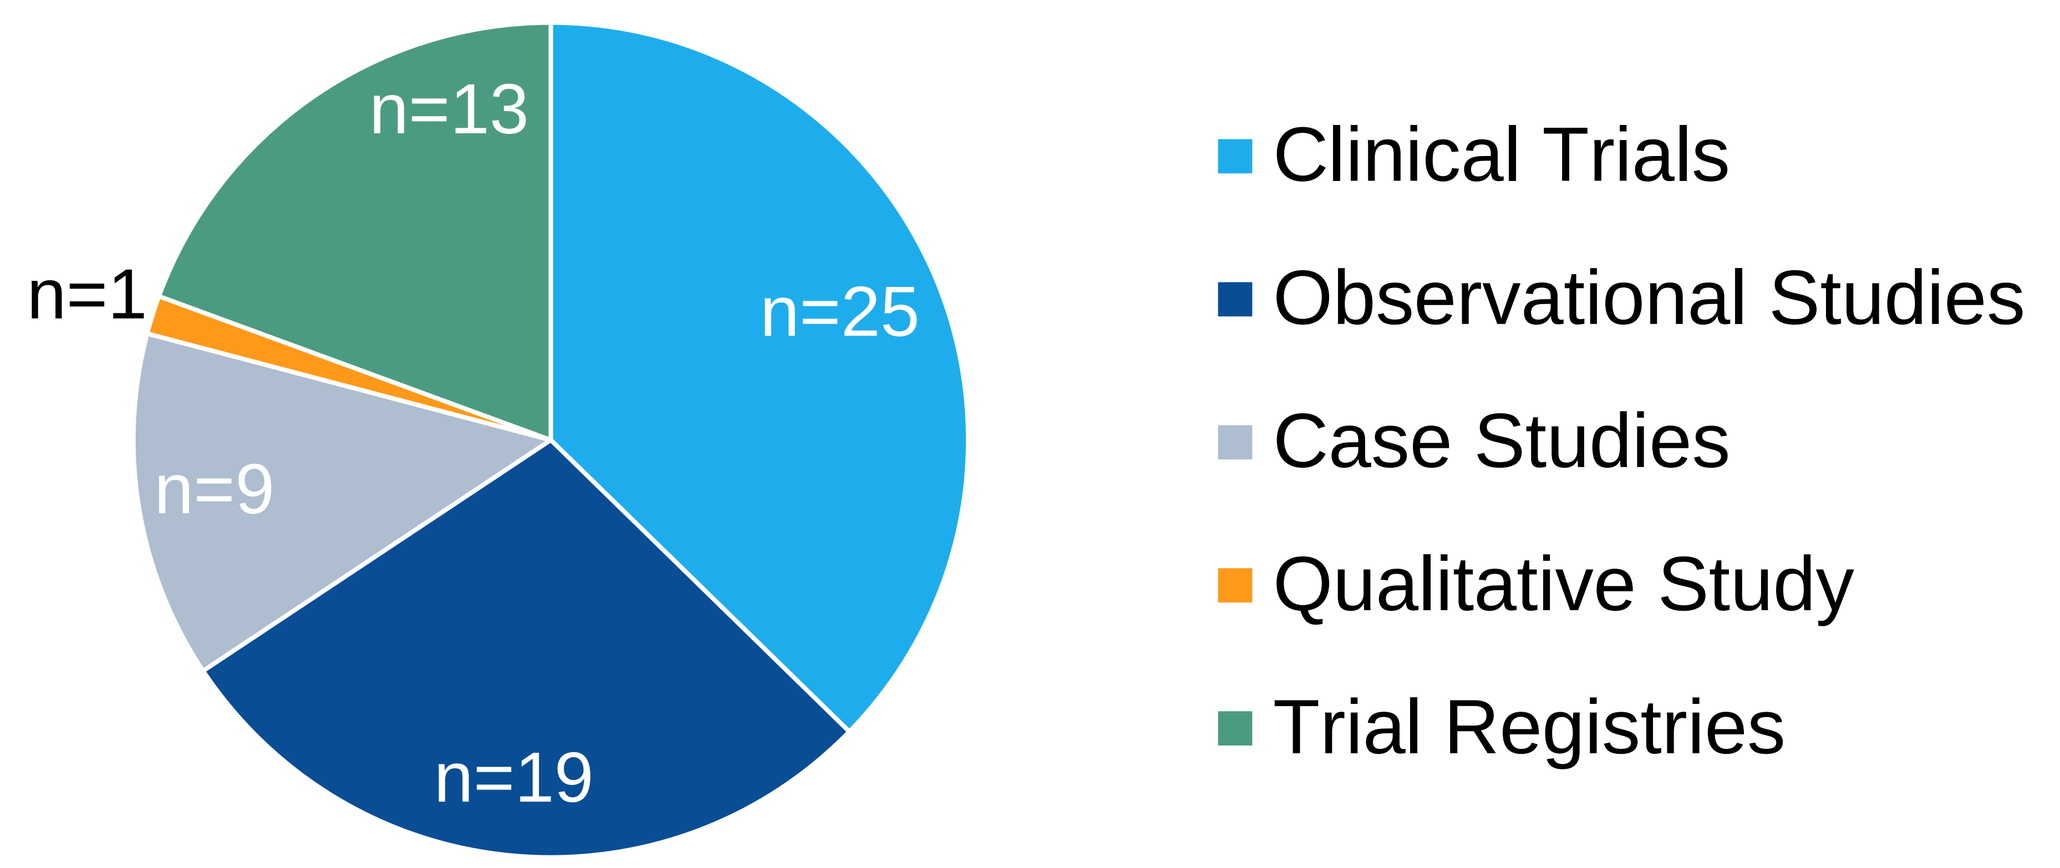
n=25
n=19
n=9
n=1
n=13
Clinical Trials
Observational Studies
Case Studies
Qualitative Study
Trial Registries

- Of the 34 full text articles included for data extraction in the topic "Treatment outcomes and clinical trials", 11 reported on pharmacological interventions, which included both disease-modifying therapies as well as nutritional supplements; 7 reported on physiological (exercise) interventions, which included strength training, high-intensity training, or aerobic conditioning; 13 reported on surgical interventions (either scapulothoracic arthrodesis or scapulopexy); and 3 reported on interventions that did not fit into the previous three categories.
- The number of unique outcomes for each of the intervention types for conference abstracts, full texts, and trial registries is shown in **Figure 4.**

Figure 4. Number of Outcomes Identified for "Treatment Outcomes and Clinical Trials"

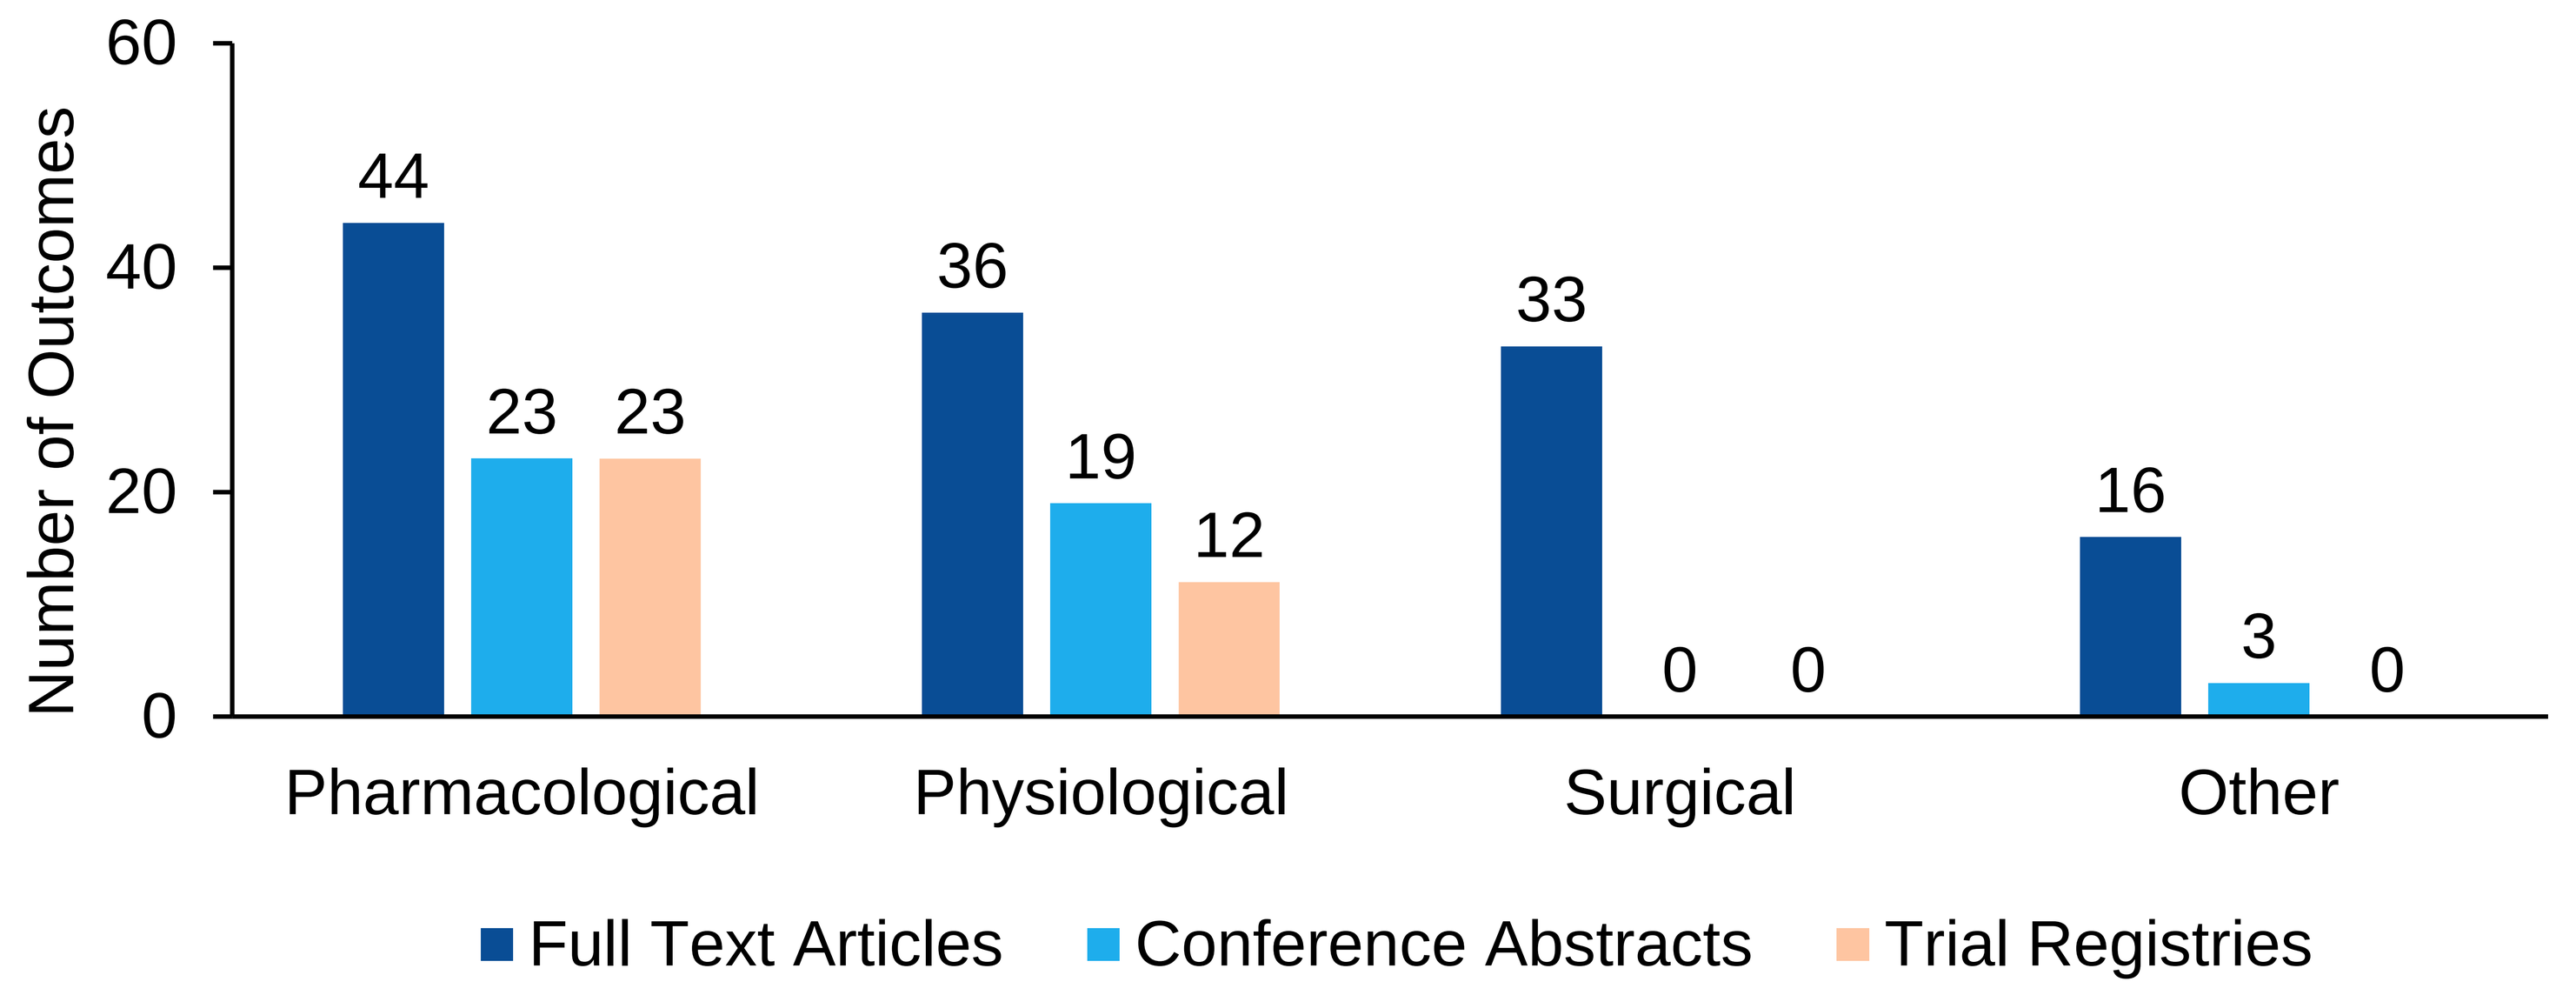
Number of Outcomes
60
40
20
0
44 23 23
36 19 12
33 0 0
16 3 0
Pharmacological
Physiological
Surgical
Other
Full Text Articles
Conference Abstracts
Trial Registries

DISCLOSURES

AE-L is a current employee and shareholder at Fulcrum Therapeutics. KE has served on advisory boards or has received consulting fees from Biogen, Acceleron, PTC Therapeutics, Ionis Pharmaceuticals, Fulcrum Therapeutics, Dyne Therapeutics, Avidity Biosciences, and Roche. Maple Health Group received consulting fees for this work.

REFERENCES

1. Huml *et al.*, *Ther Innov Regul Sci* 2020; 54(1): 144-50. 2. FULCRUM Therapeutics. About FSHD. 2022. https://www.fulcrumtx.com/therapeutic-focus/fshd/. 3. Preston *et al.*, Facioscapulohumeral Muscular Dystrophy. In: [Internet] G, editor.; 2020. 4. University of York Centre for Reviews and Dissemination. Systematic Reviews: CRD's guidance for undertaking reviews in health care, 2008.

ABBREVIATIONS

FSHD = facioscapulohumeral muscular dystrophy; PRISMA = Preferred Reporting Items for Systematic Reviews and Meta-Analyses; PRO = patient-reported outcome; SLR = systematic literature review