Relationships between Heterotopic Ossification Volume and Functional and Quality of Life Endpoints in Fibrodysplasia Ossificans Progressiva

CO77

Elisabeth J.M. Verburg-Baltussen[1], Niels Dunnewind[1], Sonja Kroep[1], Ben van Hout[1], Elaine A. Böing[2], Richard Keen[3]

[1]OPEN Health, Rotterdam, Netherlands; [2]Ipsen, Cambridge, MA, USA; [3]Royal National Orthopaedic Hospital, Stanmore, UK. Presenting author: Elaine A. Böing (elaine.boing@ipsen.com)

Background

- Fibrodysplasia ossificans progressiva (FOP) is an ultra-rare, genetic disorder of heterotopic ossification (HO) resulting in loss of joint function, progressive restriction of movement, and severe disability.[1]
- New HO volume has been accepted by regulators as a primary endpoint for trials in FOP.[2] However, its relationship with outcomes which are likely relevant in health technology assessments, such as joint function and quality of life (QoL), warrants additional exploration.

Objective

To examine the relationships between HO volume and functional and QoL endpoints in patients with FOP.

Methods

- Data from 171 patients from the MOVE trial and the FOP Natural History Study (NHS) were pooled for this analysis:
 - MOVE (NCT033126343) was a phase 3, multicenter, open-label trial of patients with FOP who received oral palovarotene.
 - The FOP NHS was an international, observational, non-interventional study of patients with FOP followed for 36 months.
- Relationships were examined between HO volume and functional and QoL outcomes (including annualized change in these outcomes; **Table 1**). Additionally, the extent to which HO in individual body regions affected functional and QoL outcomes was evaluated.
 - The Cumulative Analogue Joint Involvement Scale (CAJIS) assesses total body and regional mobility burden of FOP in 15 anatomic locations.
 - The FOP Physical Function Questionnaire (FOP-PFQ) is a disease-specific instrument that includes questions related to activities of daily living and physical functioning.
 - The Patient Reported Outcome Measure Information System (PROMIS) is a generic patient-reported outcome measure of physical and mental function.
- **Spearman correlation coefficients** were computed between the variables of interest at baseline and year 1 (annualized change).
- Various **mixed effect regression models** were fitted, including a patient-level random effect model, to account for correlations among repeated measures on the same patient. Model types aligning with the data characteristics of the outcome variables were used.
 - First, best model fit was assessed per Akaike Information Criterion (AIC) and Bayesian Information Criterion (BIC) using multiple predictor sets (selection per BIC is presented in this poster). For each outcome, two main predictor sets were compared:
 - HO volume in individual body regions (i.e., chest, arms, torso, hips and legs);
 - HO volume in upper and lower extremities.
 - Second, the regression coefficients of the models were examined in terms of magnitude, direction, and statistical significance to determine whether positive or negative relationships could be confirmed.

Results

- **Figure 1** presents the Spearman correlation coefficients. Compared with baseline, relationships at year 1 (annualized changes) were less pronounced.
- **Table 2** presents the results from converged mixed regression models:
 - As expected, regression coefficients generally confirmed positive relationships between HO volume and CAJIS/FOP-PFQ, and negative relationships between HO volume and PROMIS.
 - Statistically significant relationships ($p<0.05$) were found between HO volume in certain body regions and endpoint scores (e.g., HO volume in upper extremities and observed/annualized change in CAJIS scores, and FOP-FPQ observed scores).
 - HO volume in the upper or lower extremities was significantly associated with change in upper extremity CAJIS scores, observed FOP-PFQ scores, and changes in FOP-PFQ scores ($p<0.05$).

Table 1. Description of the data included in the analysis

Measure	Unit	Number of regions/questions	Conversion	Variables used in analysis
HO volume	mm^3	9	Normalized per region to a mean of 0 and a standard deviation of 1, pooling all timepoints	• Total HO volume • HO volume by body region • HO volume by upper and lower extremity
CAJIS	Score of 0,1, or 2	15	NA	• CAJIS total score • CAJIS upper and lower extremity score
FOP-PFQ	5-point Likert scale	28	Percentage of the worst possible score (PWPS)	• FOP-PFQ PWPS total score • FOP-PFQ PWPS upper and lower extremity
PROMIS	20- or 5-point Likert scales	10	Summary scores were converted into T-scores with a mean of 50 and a standard deviation of 10 points	• PROMIS Mental Health score • PROMIS Physical Health score

Figure 1. Spearman correlation coefficients a) at baseline and b) in year 1 (annualized change)

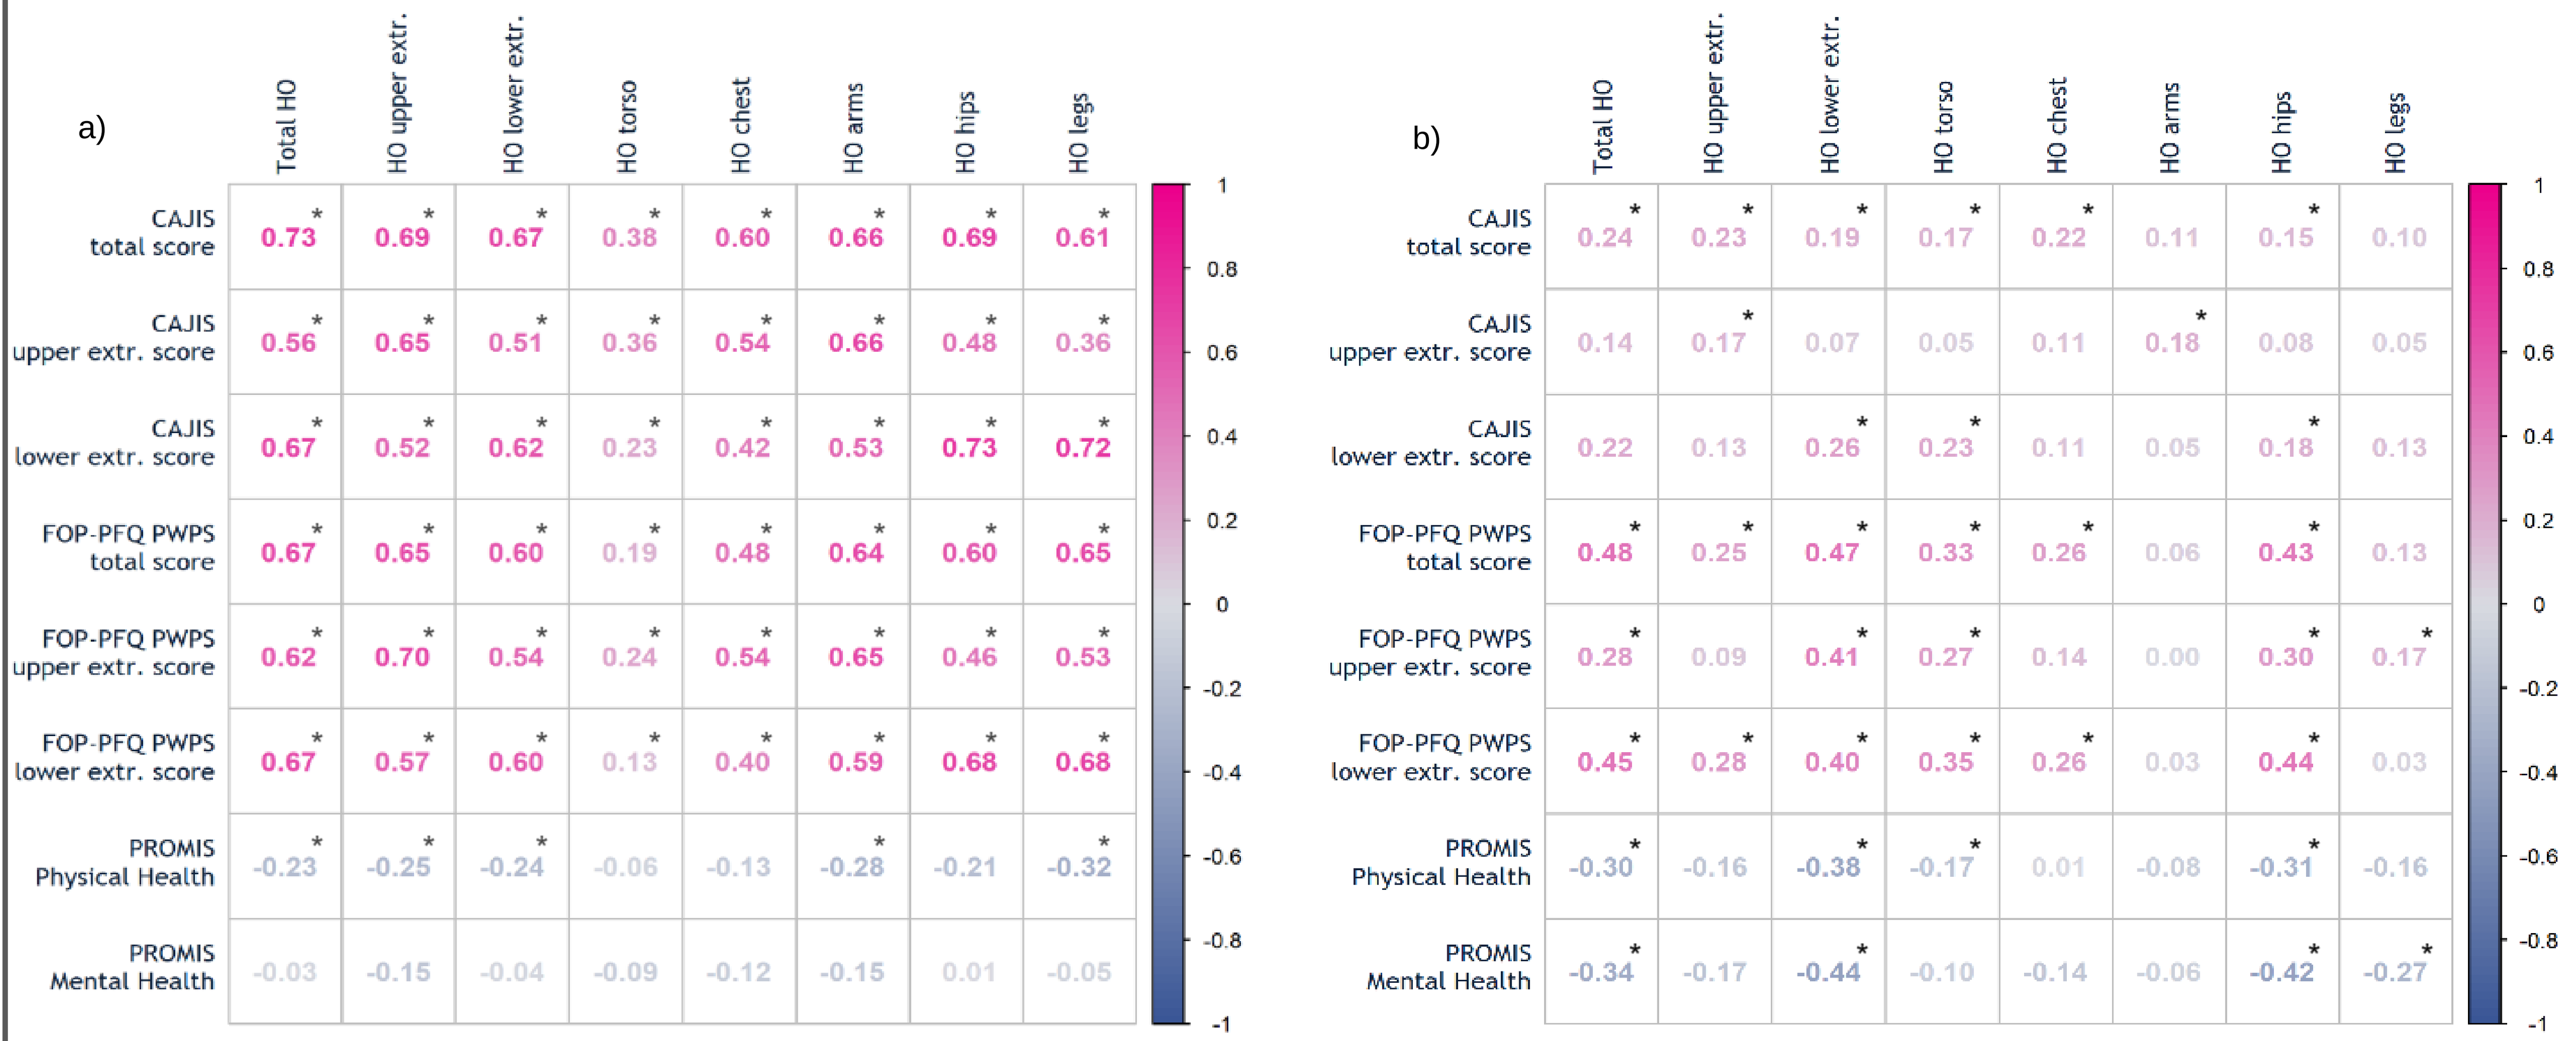
a)
Total HO
HO upper extr.
HO lower extr.
HO torso
HO chest
HO arms
HO hips
HO legs
CAJIS total score: 0.73* 0.69* 0.67* 0.38* 0.60* 0.66* 0.69* 0.61*
CAJIS upper extr. score: 0.56* 0.65* 0.51* 0.36* 0.54* 0.66* 0.48* 0.36*
CAJIS lower extr. score: 0.67* 0.52* 0.62* 0.23* 0.42* 0.53* 0.73* 0.72*
FOP-PFQ PWPS total score: 0.67* 0.65* 0.60* 0.19* 0.48* 0.64* 0.60* 0.65*
FOP-PFQ PWPS upper extr. score: 0.62* 0.70* 0.54* 0.24* 0.54* 0.65* 0.46* 0.53*
FOP-PFQ PWPS lower extr. score: 0.67* 0.57* 0.60* 0.13* 0.40* 0.59* 0.68* 0.68*
PROMIS Physical Health: -0.23* -0.25* -0.24* -0.06 -0.13 -0.28* -0.21 -0.32*
PROMIS Mental Health: -0.03 -0.15 -0.04 -0.09 -0.12 -0.15 0.01 -0.05
1 0.8 0.6 0.4 0.2 0 -0.2 -0.4 -0.6 -0.8 -1
b)
CAJIS total score: 0.24* 0.23* 0.19* 0.17* 0.22* 0.11 0.15* 0.10
CAJIS upper extr. score: 0.14 0.17* 0.07 0.05 0.11 0.18* 0.08 0.05
CAJIS lower extr. score: 0.22 0.13 0.26* 0.23* 0.11 0.05 0.18* 0.13
FOP-PFQ PWPS total score: 0.48* 0.25* 0.47* 0.33* 0.26* 0.06 0.43* 0.13
FOP-PFQ PWPS upper extr. score: 0.28* 0.09 0.41* 0.27* 0.14 0.00 0.30* 0.17*
FOP-PFQ PWPS lower extr. score: 0.45* 0.28* 0.40* 0.35* 0.26* 0.03 0.44* 0.03
PROMIS Physical Health: -0.30* -0.16 -0.38* -0.17* 0.01 -0.08 -0.31* -0.16
PROMIS Mental Health: -0.34* -0.17 -0.44* -0.10 -0.14 -0.06 -0.42* -0.27*

*Statistically significant ($p<0.05$)

CONCLUSIONS

- The Spearman correlation coefficients, as well as results from the mixed regression models, confirm logical, significant relationships between HO volume and CAJIS and FOP-PFQ.
- There are insufficient data to confirm or reject a relationship between HO volume and PROMIS.
- These results help to better understand the associations between HO volume and functional and QoL endpoints in FOP.

Table 2. Direction and significance of relationships between HO volume in body regions and functional and QoL endpoints

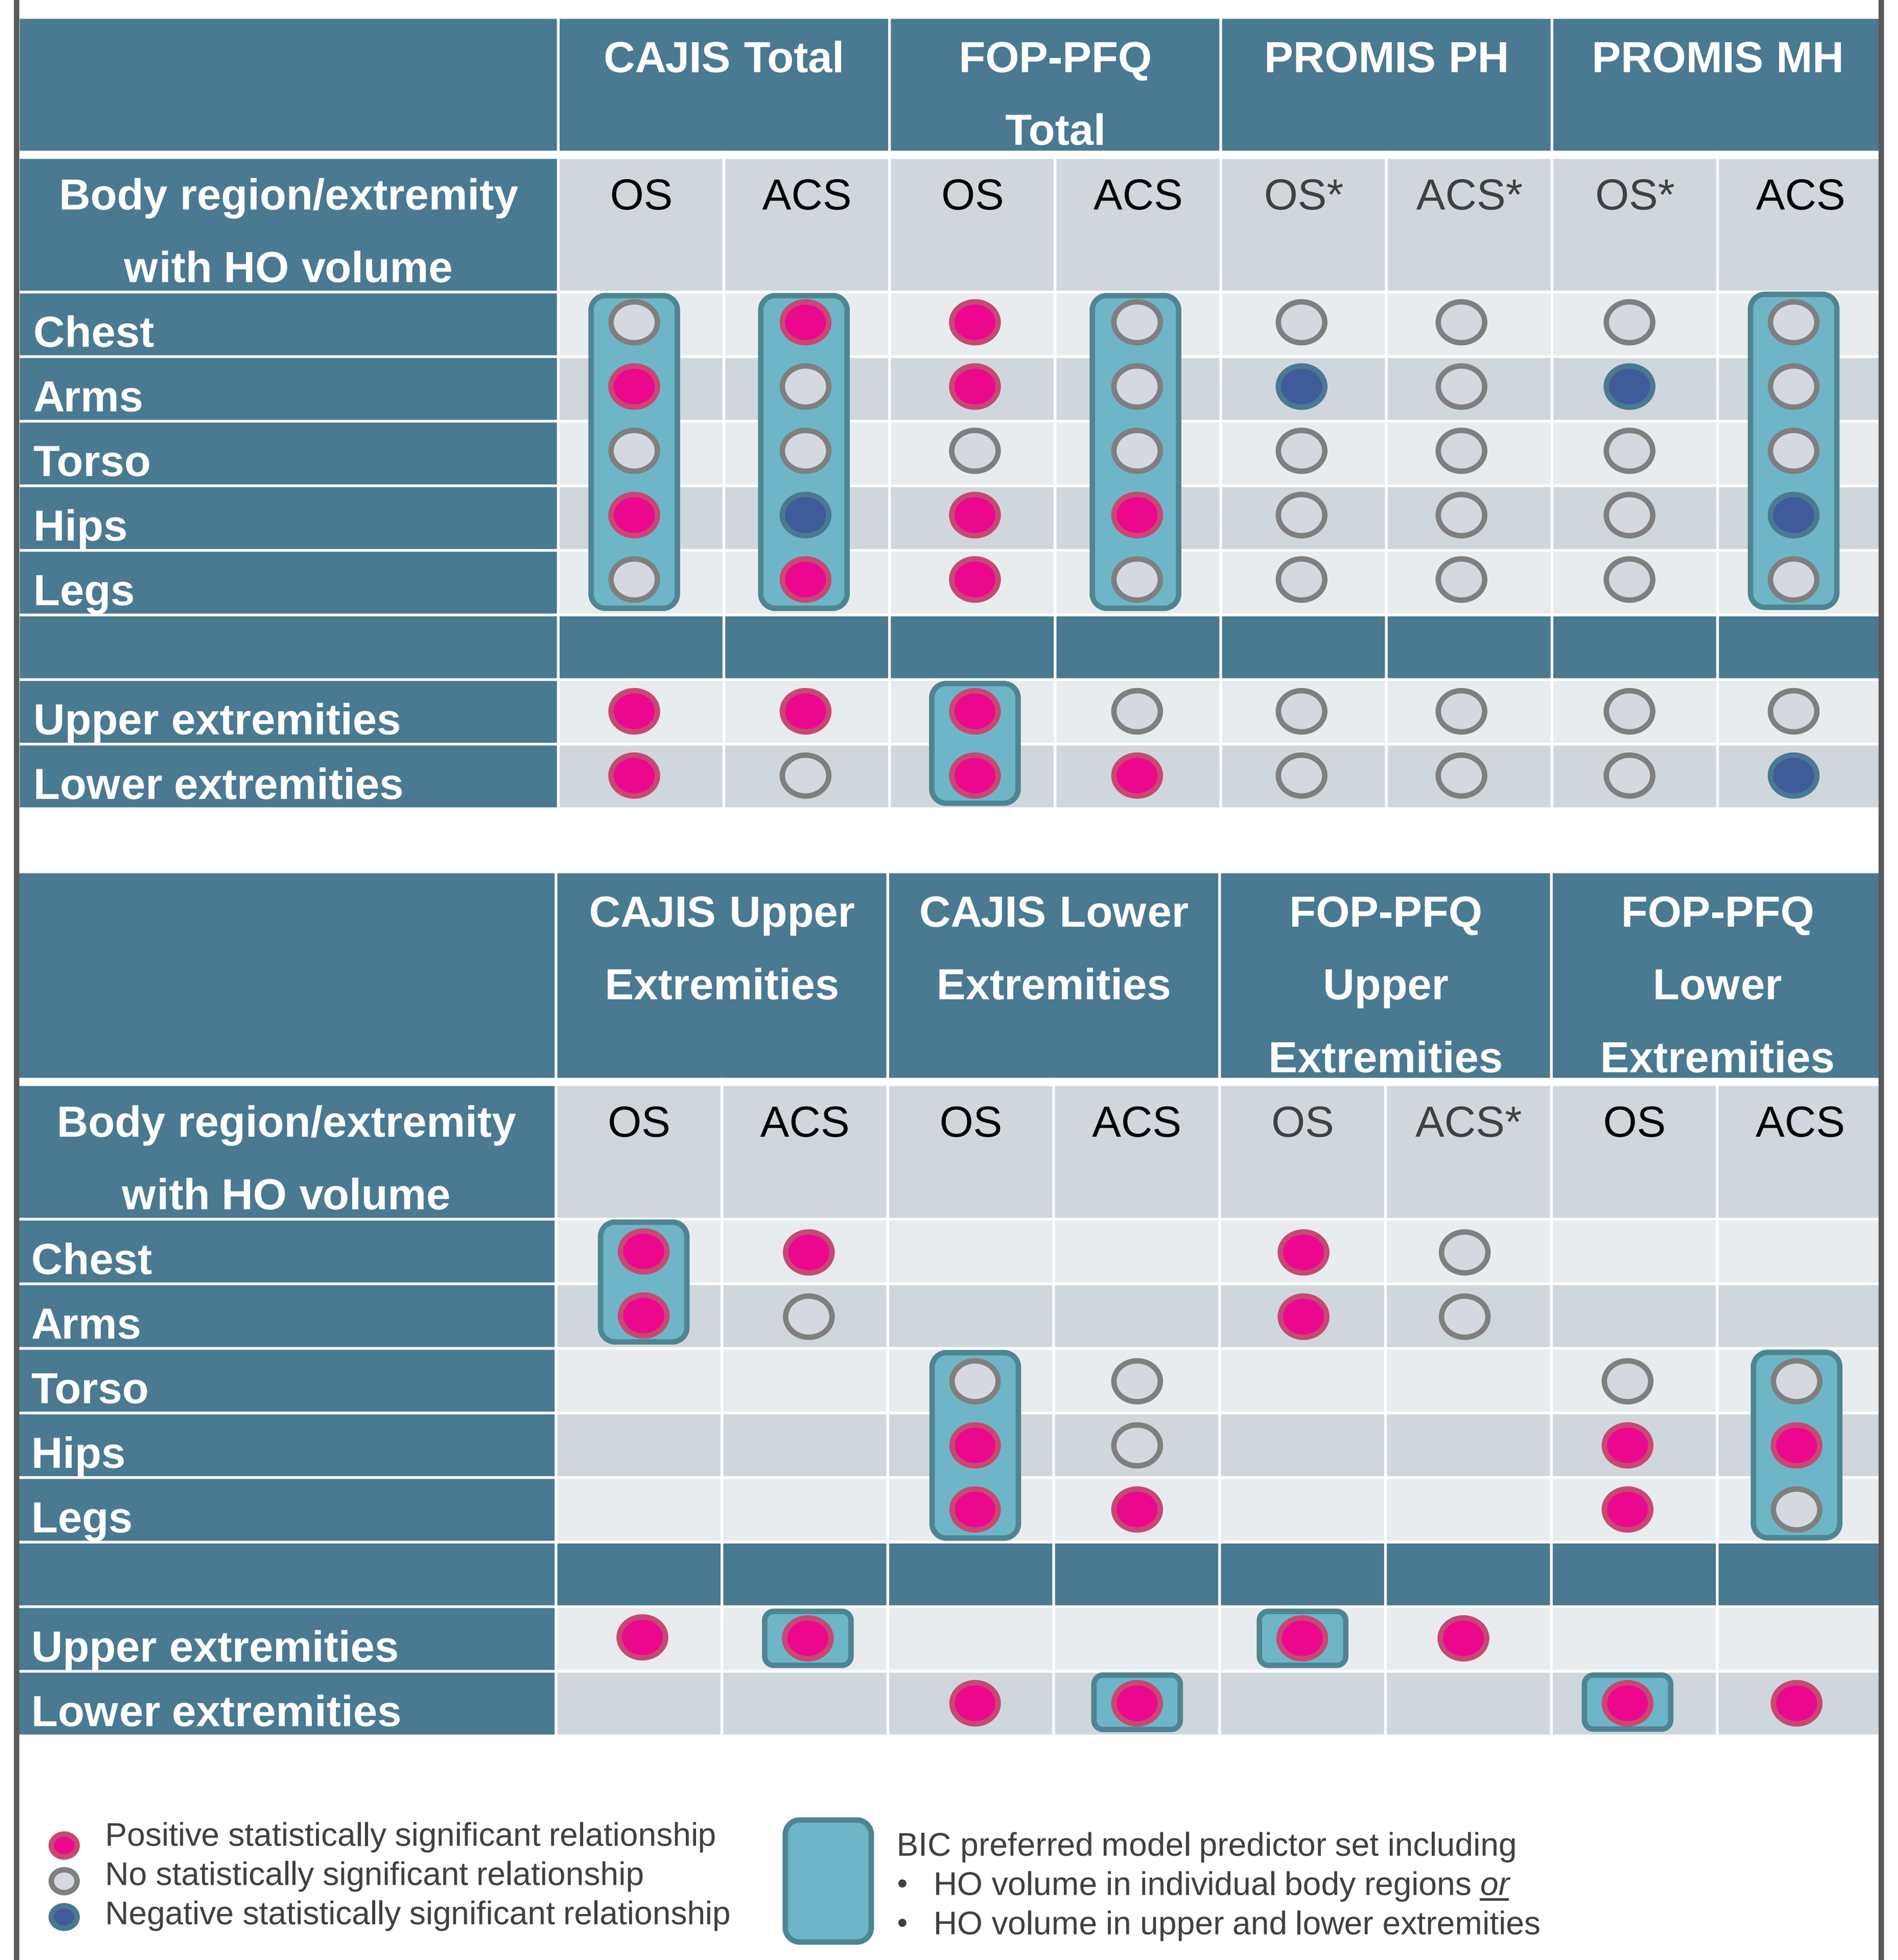
CAJIS Total
FOP-PFQ Total
PROMIS PH
PROMIS MH
Body region/extremity with HO volume
OS ACS OS ACS OS* ACS* OS* ACS
Chest
Arms
Torso
Hips
Legs
Upper extremities
Lower extremities
CAJIS Upper Extremities
CAJIS Lower Extremities
FOP-PFQ Upper Extremities
FOP-PFQ Lower Extremities
OS ACS OS ACS OS ACS* OS ACS
Positive statistically significant relationship
No statistically significant relationship
Negative statistically significant relationship
BIC preferred model predictor set including
• HO volume in individual body regions *or*
• HO volume in upper and lower extremities

*No significant model available for comparison
Abbreviations: ACS: annualized change score; BIC: Bayesian information criteria; MH: mental health; OS: observed score; PH: physical health.

Abbreviations ACS: annualized change score; AIC: Akaike information criterion; BIC: Bayesian information criteria; Extr.: extremities; CAJIS: Cumulative Analogue Joint Involvement Scale; FOP-PFQ: FOP Physical Function Questionnaire; HO: heterotopic ossification; MH: mental health; NA: not applicable; OS: observed score; PH: physical health; PROMIS: Patient-Reported Outcomes Measurement Information System; PWPS: percentage of the worst possible score.

References **1.** Kaplan FS et al. Best Pract Res Clin Rheumatol. 2008;22(1):191–205; **2.** Hsiao EC et al. Br J Clin Pharmacol. 2019;85(6): 1199–1207; **3.** ClinicalTrials.gov. An Efficacy and Safety Study of Palovarotene for the Treatment of Fibrodysplasia Ossificans Progressiva (MOVE); **4.** ClinicalTrials.gov. A Natural History Study of Fibrodysplasia Ossificans Progressiva (FOP).

Author Contributions Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **EVB, ND, SK, BH, EAB**; Drafting of the publication, or revising it critically for important intellectual content: **EVB, ND, EAB, RK**; Final approval of the publication: **EVB, ND, SK, BH, EAB, RK**.

Disclosures EVB, ND, SK, BH: Employees of OPEN Health; **EAB**: Employee and shareholder of Ipsen; **RK**: Research Investigator: Clementia/Ipsen, Kyowa Kirin, Regeneron; Advisory board: IFOPA FOP Registry Medical Advisory Board, International Clinical Council on FOP.

Medical Writing Support The authors thank Carla Kraan and Ana Moura of OPEN Health for design support, which was sponsored by Ipsen in accordance with Good Publication Practice guidelines.

Presented at ISPOR US 2023 | Boston, Massachusetts, USA | May 7–10, 2023

This study was sponsored by Ipsen

To download the poster, please scan the QR code.
Copies of this e-poster obtained through the QR code are for personal use only and may not be reproduced without written permission from the authors.