EE403

Direct and Indirect Costs Among Employed Patients with Myasthenia Gravis in Sweden

Qian Cai[1], Qiaoyi Zhang[2], Jakob Börsum[3], Alberto E. Batista[2], Peter Kunovszki[4], Gabriel Isheden[3], Kristin Heerlein[5]

[1]Janssen Global Services, Market Access, RWE, Titusville, USA; [2]Janssen Global Services, Market Access, Neuroscience, Titusville, USA; [3]SDS Life Science AB, Uppsala, Sweden; [4]Janssen Global Services, Market Access, RWE, Budapest, Hungary; [5]Janssen Global Services, Medical Affairs, Neuroscience, Neuss, Germany

INTRODUCTION

- Myasthenia gravis (MG) is a rare, chronic autoantibody disorder characterized by muscle weakness and fatigue that leads to hallmark symptoms including ptosis, shortness of breath and limb weakness.[1]
- The majority of MG patients suffer from limitations related to disease severity and often need life-long treatment.[2] Thereby, patients with MG often experience productivity loss and employment change.
- Economic evaluation on cost of MG in terms of all-cause direct costs and indirect costs due to absenteeism remain largely unexplored.

OBJECTIVE

- To estimate all-cause direct and indirect costs among employed patients with newly diagnosed (ND) vs pre-existing (PE) myasthenia gravis (MG) in Sweden.

METHODS

Data Sources

- Data were collected from four national registers provided by Statistics Sweden (SCB) and the National Board of Health and Welfare (NBHW) in Sweden.
 - National Patient Register (NPR): diagnoses, procedures, hospitalizations, and outpatient specialist services.
 - Prescribed Drug Register (PDR): dispensed items, amount, date of filled prescription.
 - Cause of Death Register: date and cause of death.
 - Longitudinal Integrated Database for Health Insurance and Labor Market Studies (LISA): socioeconomic characteristics.
- Data from the four national registers were linked through the unique personal identity number.

Study Patients

- Patients should have
 - ≥2 primary diagnosis for MG (ICD-10-SE: G70.0) filed at least 12 months apart within 24 months, with ≥1 primary MG diagnosis recorded by a neurologist, between 1/1/2010 and 12/31/2017.
 - Aged ≥18 years as of index date* and have ≥24 months follow-up period.
 - Record of acetylcholinesterase (AChE) inhibitors, corticosteroids (CS), non-steroidal immunosuppressive treatment (NSIST), immunoglobulin (IVIg/SCIg) or plasma exchange (PLEX) on or after the index date.
 - Standing employment at index calendar year and one year after index year.
- Patients were categorized into ND vs PE groups based on whether they had a MG diagnosis prior to the index date (back until January 1, 2001).

*Index date: Date of the first primary MG diagnosis

Study Measures

- Annual all-cause direct costs were defined as costs associated with any condition incurred from medical or pharmacy services within 12-month post-index period.
 - Medical costs were estimated by multiplying the diagnosis-related group (DRG) weight with cost per DRG weight for each recorded inpatient admission and outpatient specialist visit.
 - Pharmacy costs were estimated by summing patient-paid costs and the reimbursement paid by the county council for recorded prescription drugs.
- Annual all-cause indirect costs were calculated by multiplying days of sickness with the average gross daily wage (adding payroll taxes) in the Swedish population.
- All costs were adjusted to the 2019 values and subsequently converted to EUR using Sweden Central Bank 2019 average conversion rate.

Statistical Analyses

- Means, standard deviation (SD), medians and interquartile ranges (IQR) were reported for continuous variables and frequency and percentage were reported for categorical variables.
- To compare the difference in patient characteristics and outcome measures between ND and PE MG patients, chi-square tests or exact Fisher tests were used for categorical variables, t-tests were used for continuous variables, and Wilcoxon Mann-Whitney tests were used to compare distributional similarity between two non-normally distributed variables.
- All data analyses were conducted using SAS 9.4 (SAS Institute, Cary NC), and R version 4.0.5.

AUTOANTIBODY : MG

RESULTS

- A total of 370 employed MG patients were included of which 158 patients (42.7%) were ND MG and 212 (57.2%) were PE MG.
- Mean (±SD) age for all patients with MG at index date was 48.1 (±12.3) years.

TABLE 1. Socio-demographic and clinical characteristics of employed MG patients in Sweden

	All patients with MG	Newly diagnosed (ND) MG patients	Pre-existing (PE) MG patients	p-value
N Patients	370	158	212	
Age, year mean (SD)	48.1 (12.3)	48.7 (13.4)	47.6 (11.3)	0.401
Median (Q1-Q3)	49.0 [38.0, 58.0]	52.0 [37.3, 59.8]	48.0 [38.8, 75.0]	
Gender (n, %)				<0.001
Female	211 (57.0%)	70 (44.3%)	141 (66.5%)	
Education (n, %)				0.532
0-9 yrs	47 (12.7%)	17 (10.8%)	30 (14.2%)	
10-13 (high school)	162 (43.8%)	75 (47.5%)	87 (41.0%)	
13+ (post-secondary education)	154 (41.6%)	62 (39.2%)	92 (43.4%)	
Doctoral education	4 (1.1%)	2 (1.3%)	2 (0.9%)	
Missing	3 (0.8%)	2 (1.3%)	1 (0.5%)	
Individual income (n, %)				
<€ 20,000	49 (13.2%)	20 (12.7%)	29 (13.7%)	0.549
€ 20,000-30,000	82 (22.2%)	31 (19.6%)	51 (24.1%)	
>€ 30,000	238 (64.3%)	106 (67.1%)	132 (62.3%)	
Missing	1 (0.3%)	1 (0.6%)	0 (0%)	
CCI* (mean, SD)	2.63 (1.45)	2.91 (1.29)	2.42 (1.54)	0.001
Median (Q1-Q3)	3.00 [3.00, 3.00]	3.00 [3.00, 3.00]	3.00 [0, 3.00]	

*during 12-month pre-index period
CCI, Charlson Comorbidity index; MG, myasthenia gravis; SD, standard deviation

- The mean total annual costs of MG patients were €14,200, with direct costs of €8,714 and indirect costs of €5,486 (**Figure 2**).

FIGURE 2. Annual costs of MG patients

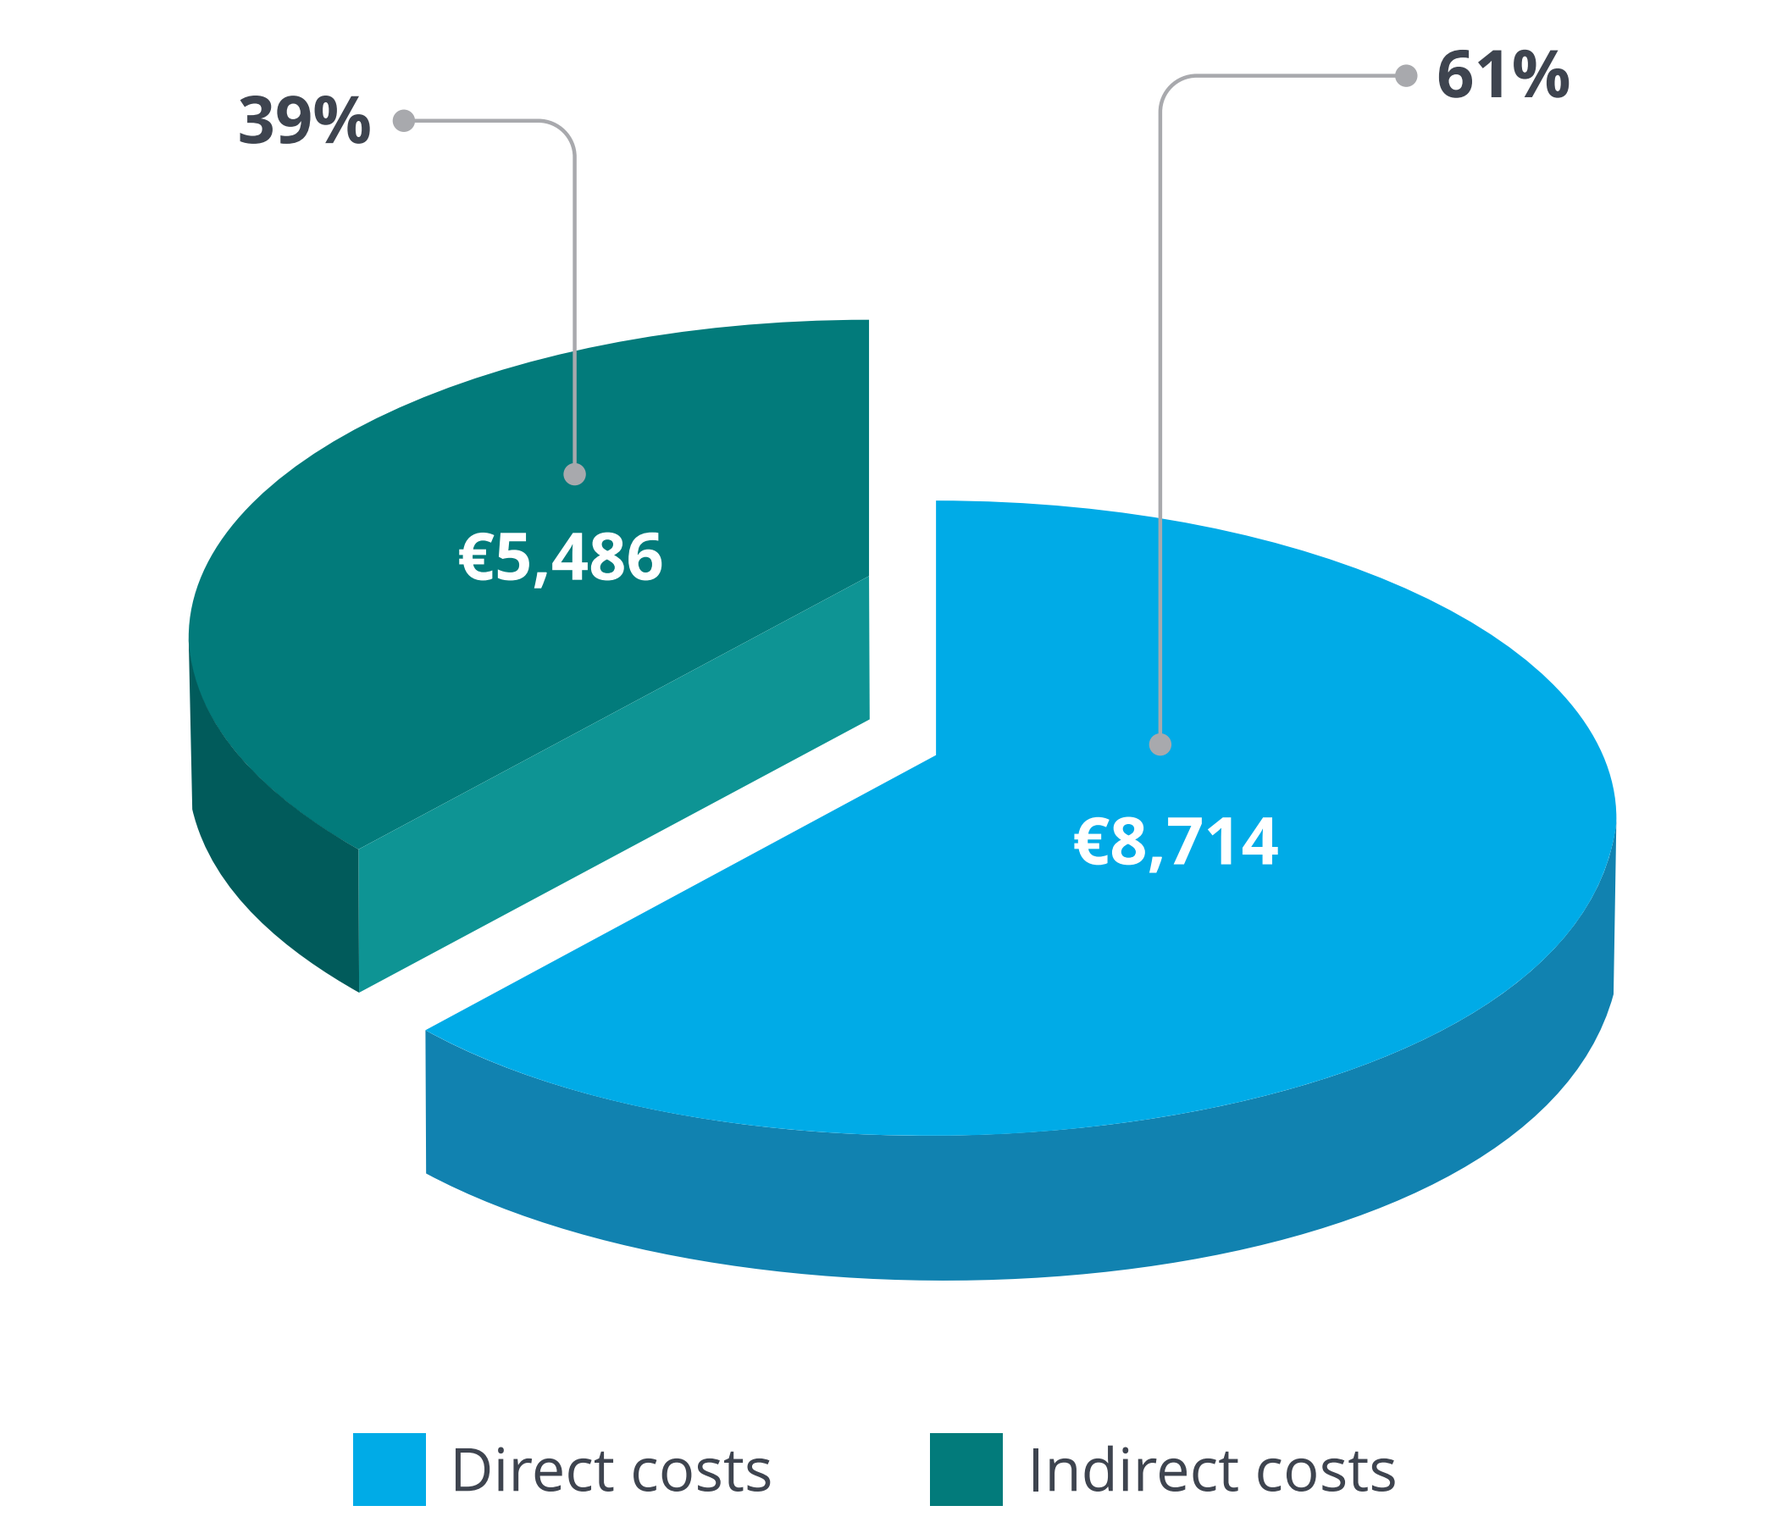
39%
61%
€5,486
€8,714
Direct costs
Indirect costs

MG, myasthenia gravis

- During 12-month post-index period, patients with ND MG had higher all-cause inpatient admission rate (64.6% vs 34.9%, p<0.01), with 6.5 more hospitalization days (p<0.01); higher percentage of patients reported ≥ 1 sick leave (p<0.01), with 31 more days of sickness absence (53.2% vs 32.5%, p<0.01) (**Figure 1**).

FIGURE 1. Rate of all-cause inpatient admission and sickness absence in ND MG patients during 12-month post-index period

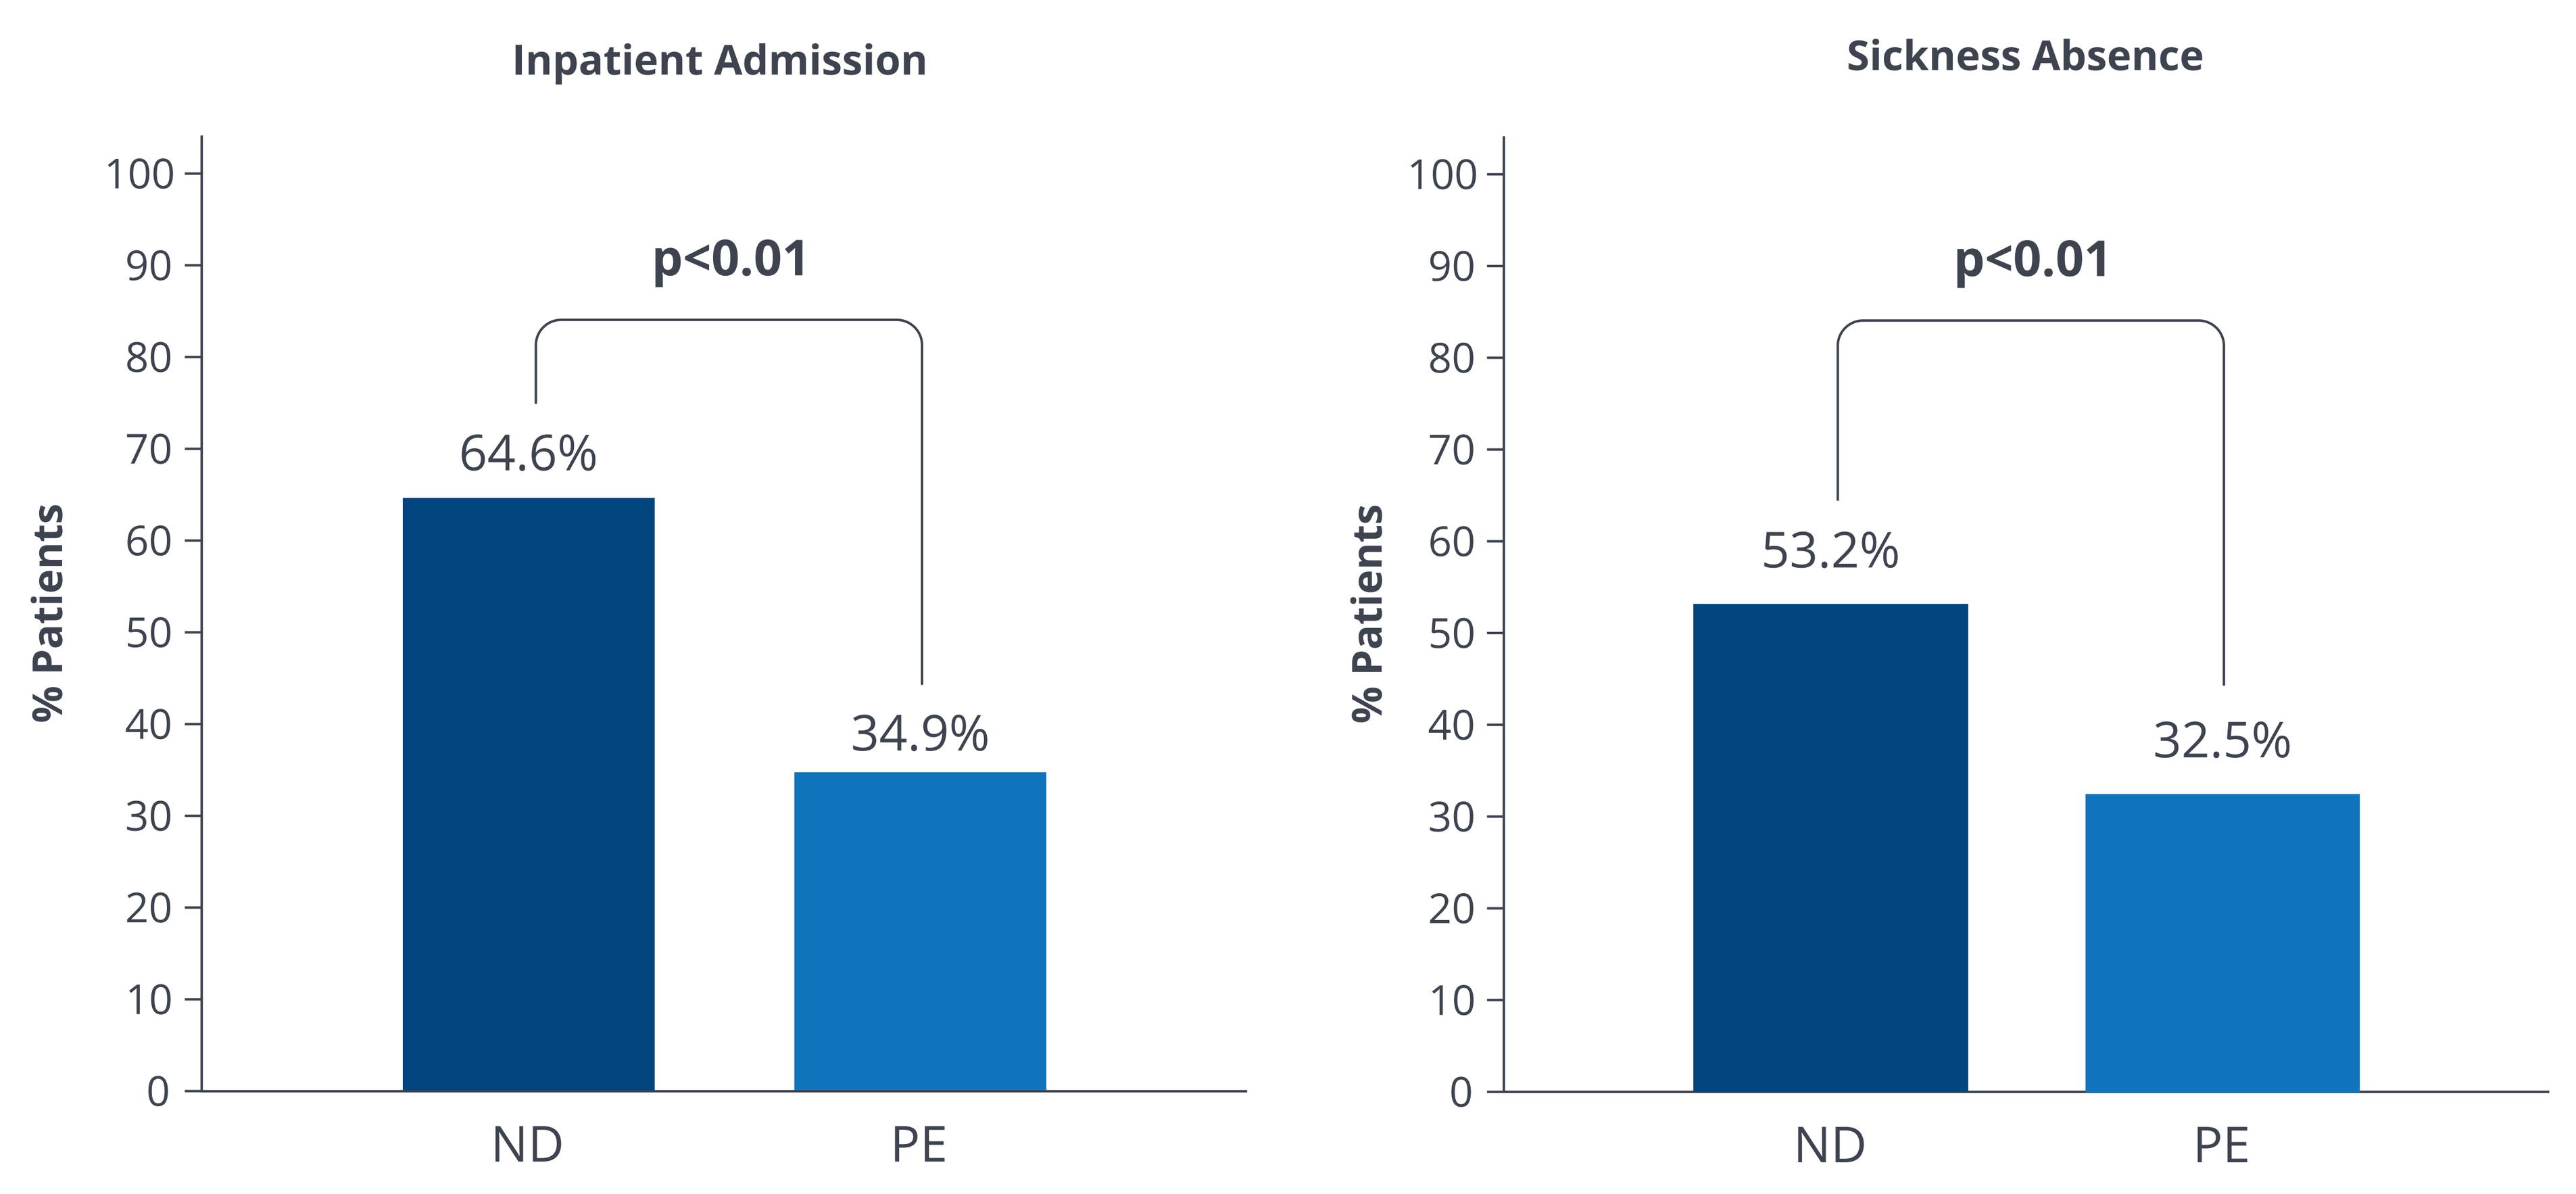
Inpatient Admission
Sickness Absence
% Patients
100
90
80
70
60
50
40
30
20
10
0
p<0.01
64.6%
34.9%
53.2%
32.5%
ND
PE

MG, myasthenia gravis; ND, newly diagnosed; PE, Pre-existing

- The mean all-cause costs for ND and PE MG patients were €19,900 and €14,200, respectively.
- Compared to the PE group, patients with ND MG incurred €6435 higher direct costs (p<0.01), with an incremental difference of €4916 due to inpatient costs (**Figure 3**).

FIGURE 3. Total direct and indirect costs for ND and PE MG patients during 12-month post-index period

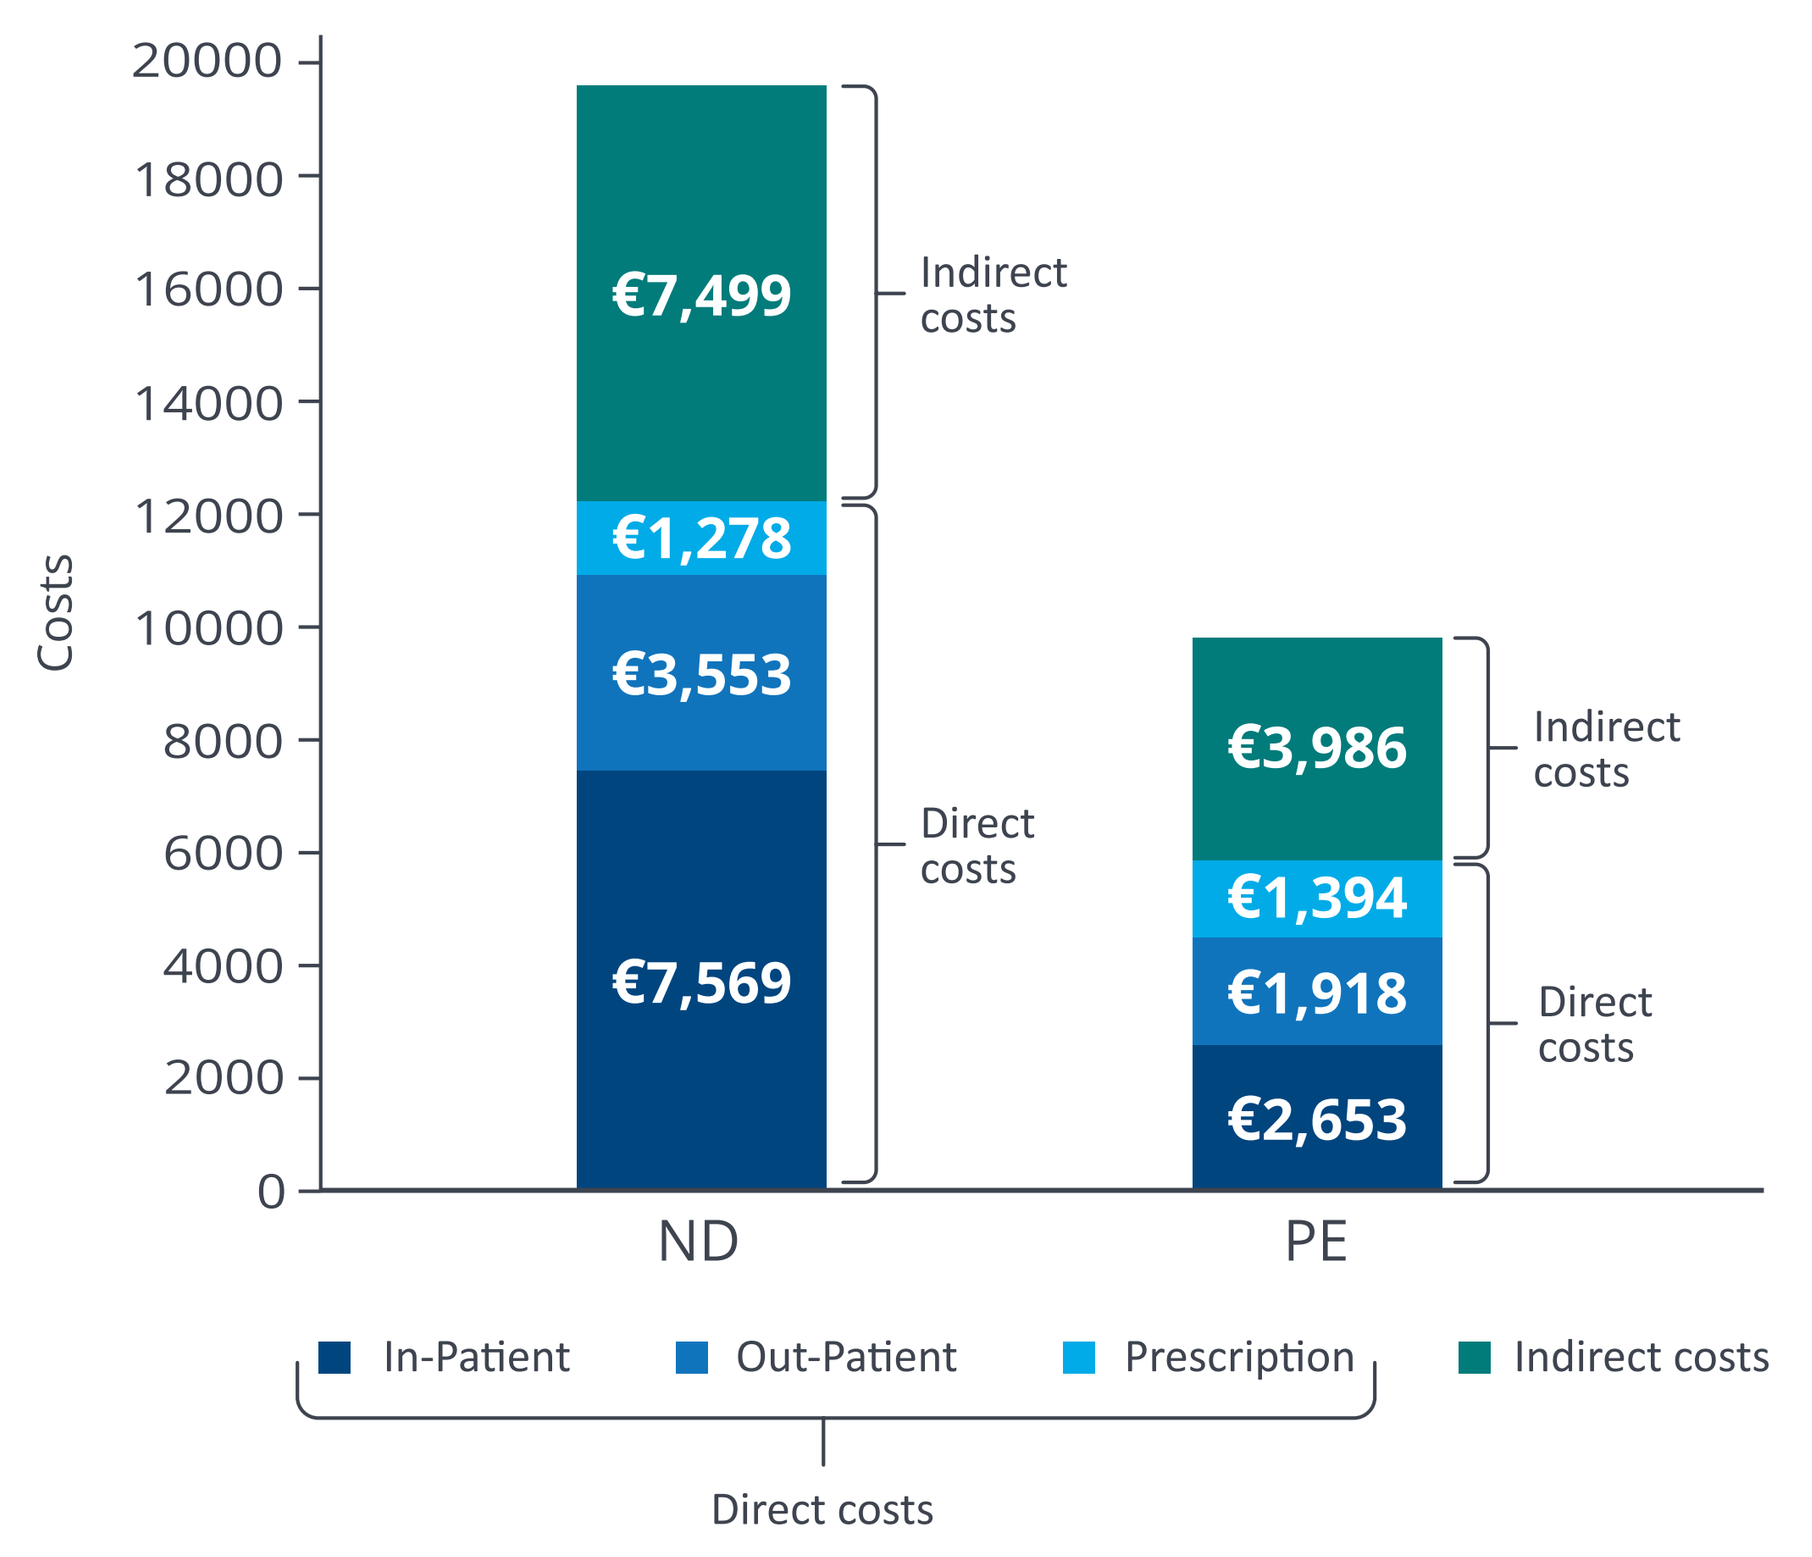
Costs
20000
18000
16000
14000
12000
10000
8000
6000
4000
2000
0
€7,499
€1,278
€3,553
€7,569
€3,986
€1,394
€1,918
€2,653
Indirect costs
Direct costs
ND
PE
In-Patient
Out-Patient
Prescription
Indirect costs
Direct costs

MG, myasthenia gravis; ND, newly diagnosed; PE, Pre-existing

REFERENCES

1. NINDS. Myasthenia Gravis Fact Sheet. NIH Publication No. 20-768. Publication date March 2020.
2. Narayanaswami P, Sanders DB, Wolfe G et al. International consensus guidance for management of myasthenia gravis: 2020 update. *Neurology* (2021) 96:114–22.

Presented at the ISPOR, May 07-10, 2023, Boston, MA, USA.

CONCLUSIONS

- Employed patients with ND MG incurred significantly higher economic burden than those with PE MG.
- Indirect costs accounted for nearly 40% of total all-cause costs among employed MG patients, with significantly higher costs in the first year compared to later years.
- Early diagnosis of disease and effective treatment may be important factors to reduce the burden in ND MG patients.

STRENGTHS AND LIMITATIONS

- This study used nationwide population-level registers to evaluate the outcomes which allows for the consistency, completeness, and accuracy of data collection and linkage.
- Primary healthcare data are not reported on a national level; therefore, there is lack of information on primary care diagnosis and initial treatment.
- No specific drug information on infusion therapy (such as IVIg) is available in the NPR; thus, costs of these treatments are underestimated.

ACKNOWLEDGEMENTS
Kalpana Tilekar, Ph.D (SIRO Clinpharm Pvt. Ltd., India) provided writing assistance and Robert Achenbach (Janssen Global Services, LLC) provided additional editorial support for the development of this poster.

DISCLOSURES
Qian Cai, Qiaoyi Zhang, Alberto E. Batista, Peter Kunovszki and Kristin Heerlein are or were employees or contractors of Janssen Pharmaceuticals and may own stock or stock options in Johnson & Johnson. Jakob Borsum and Gabriel Isheden are or were employees of SDS Lifescience AB, Uppsala, Sweden.