Poster #EE527

A Systematic Literature Review of the Economic Burden of Leber Hereditary Optic Neuropathy (LHON)

Khashayar Azimpour[1], Patricia Dorling[1], Chukwuebuka Dominic Igbelina[2], Anna Forsythe[2]
[1]Chiesi, Boston, MA, USA; [2]Cytel, Inc, Waltham, MA, USA

Introduction

- Leber Hereditary Optic Neuropathy (LHON) is a rare, inherited mitochondrial disorder that leads to severe, irreversible vision loss[1]
- It predominantly affects males, with onset usually occurring in early adulthood. Left untreated, affected patients remain visually disabled and blind in most cases for the rest of their lives[1]
- The economic impact of LHON is not well-characterized

Objective

- **This review aimed to comprehensively explore the economic burden of LHON, including healthcare resource utilization, costs, and findings from economic models**

Methods

- A systematic literature review (SLR) was performed following Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines to identify studies reporting on the economic burden in LHON as described in **Figure 1**

Figure 1. Economic SLR in LHON methods

Population	• Patients with LHON
Outcomes	• Direct and indirect costs components • Total cost • Healthcare resource use • Model inputs/model summary • Sensitivity analysis • Model outputs (QALYs, LYs gained, ICERs) • Any other measure of effectiveness reported together with costs
Study designs	• Budget impact analysis • Resource use studies • Cost/economic burden of illness studies • Cost-benefit analysis • Cost-consequence analysis • Cost-minimization analysis • Cost-utility analysis • Cost analysis • SLR and meta-analysis (for cross checking only)
Databases searched	• Embase, MEDLINE, Econlit, CENTRAL, CDSR, CCA, CMR, DARE, HTA submissions, and NHS EED (from inception to 2022) • Key conference proceedings (2019 to 2022) • Search date: 25th February 2022 • English language studies • No restrictions on geography, interventions, or comparator
Study selection	• Preliminary selection by two independent reviewers • Any discrepancies resolved by third reviewer
Data collection	• Data extracted by one reviewer and independently checked by a second, senior reviewer • Any discrepancies resolved by a third reviewer

Results

- Among 269 records identified in the SLR, five publications[2-6] from four original studies met the inclusion criteria (**Figure 2**)
- The study designs included:
 - Targeted literature search combined with a patient and caregiver survey[2,3]
 - Retrospective review of reimbursement decisions[4]
 - Retrospective chart review[5,6]
- The target populations reported by studies included: patients with inherited retinal disease (IRD) including LHON[2,3] and patients undergoing genetic testing including LHON[5,6]
- There were two studies from North America (US/Canada),[2,3,6] one from the UK,[5] and one from Finland[4]
- No health economic models were identified

Figure 2. PRISMA diagram of included studies

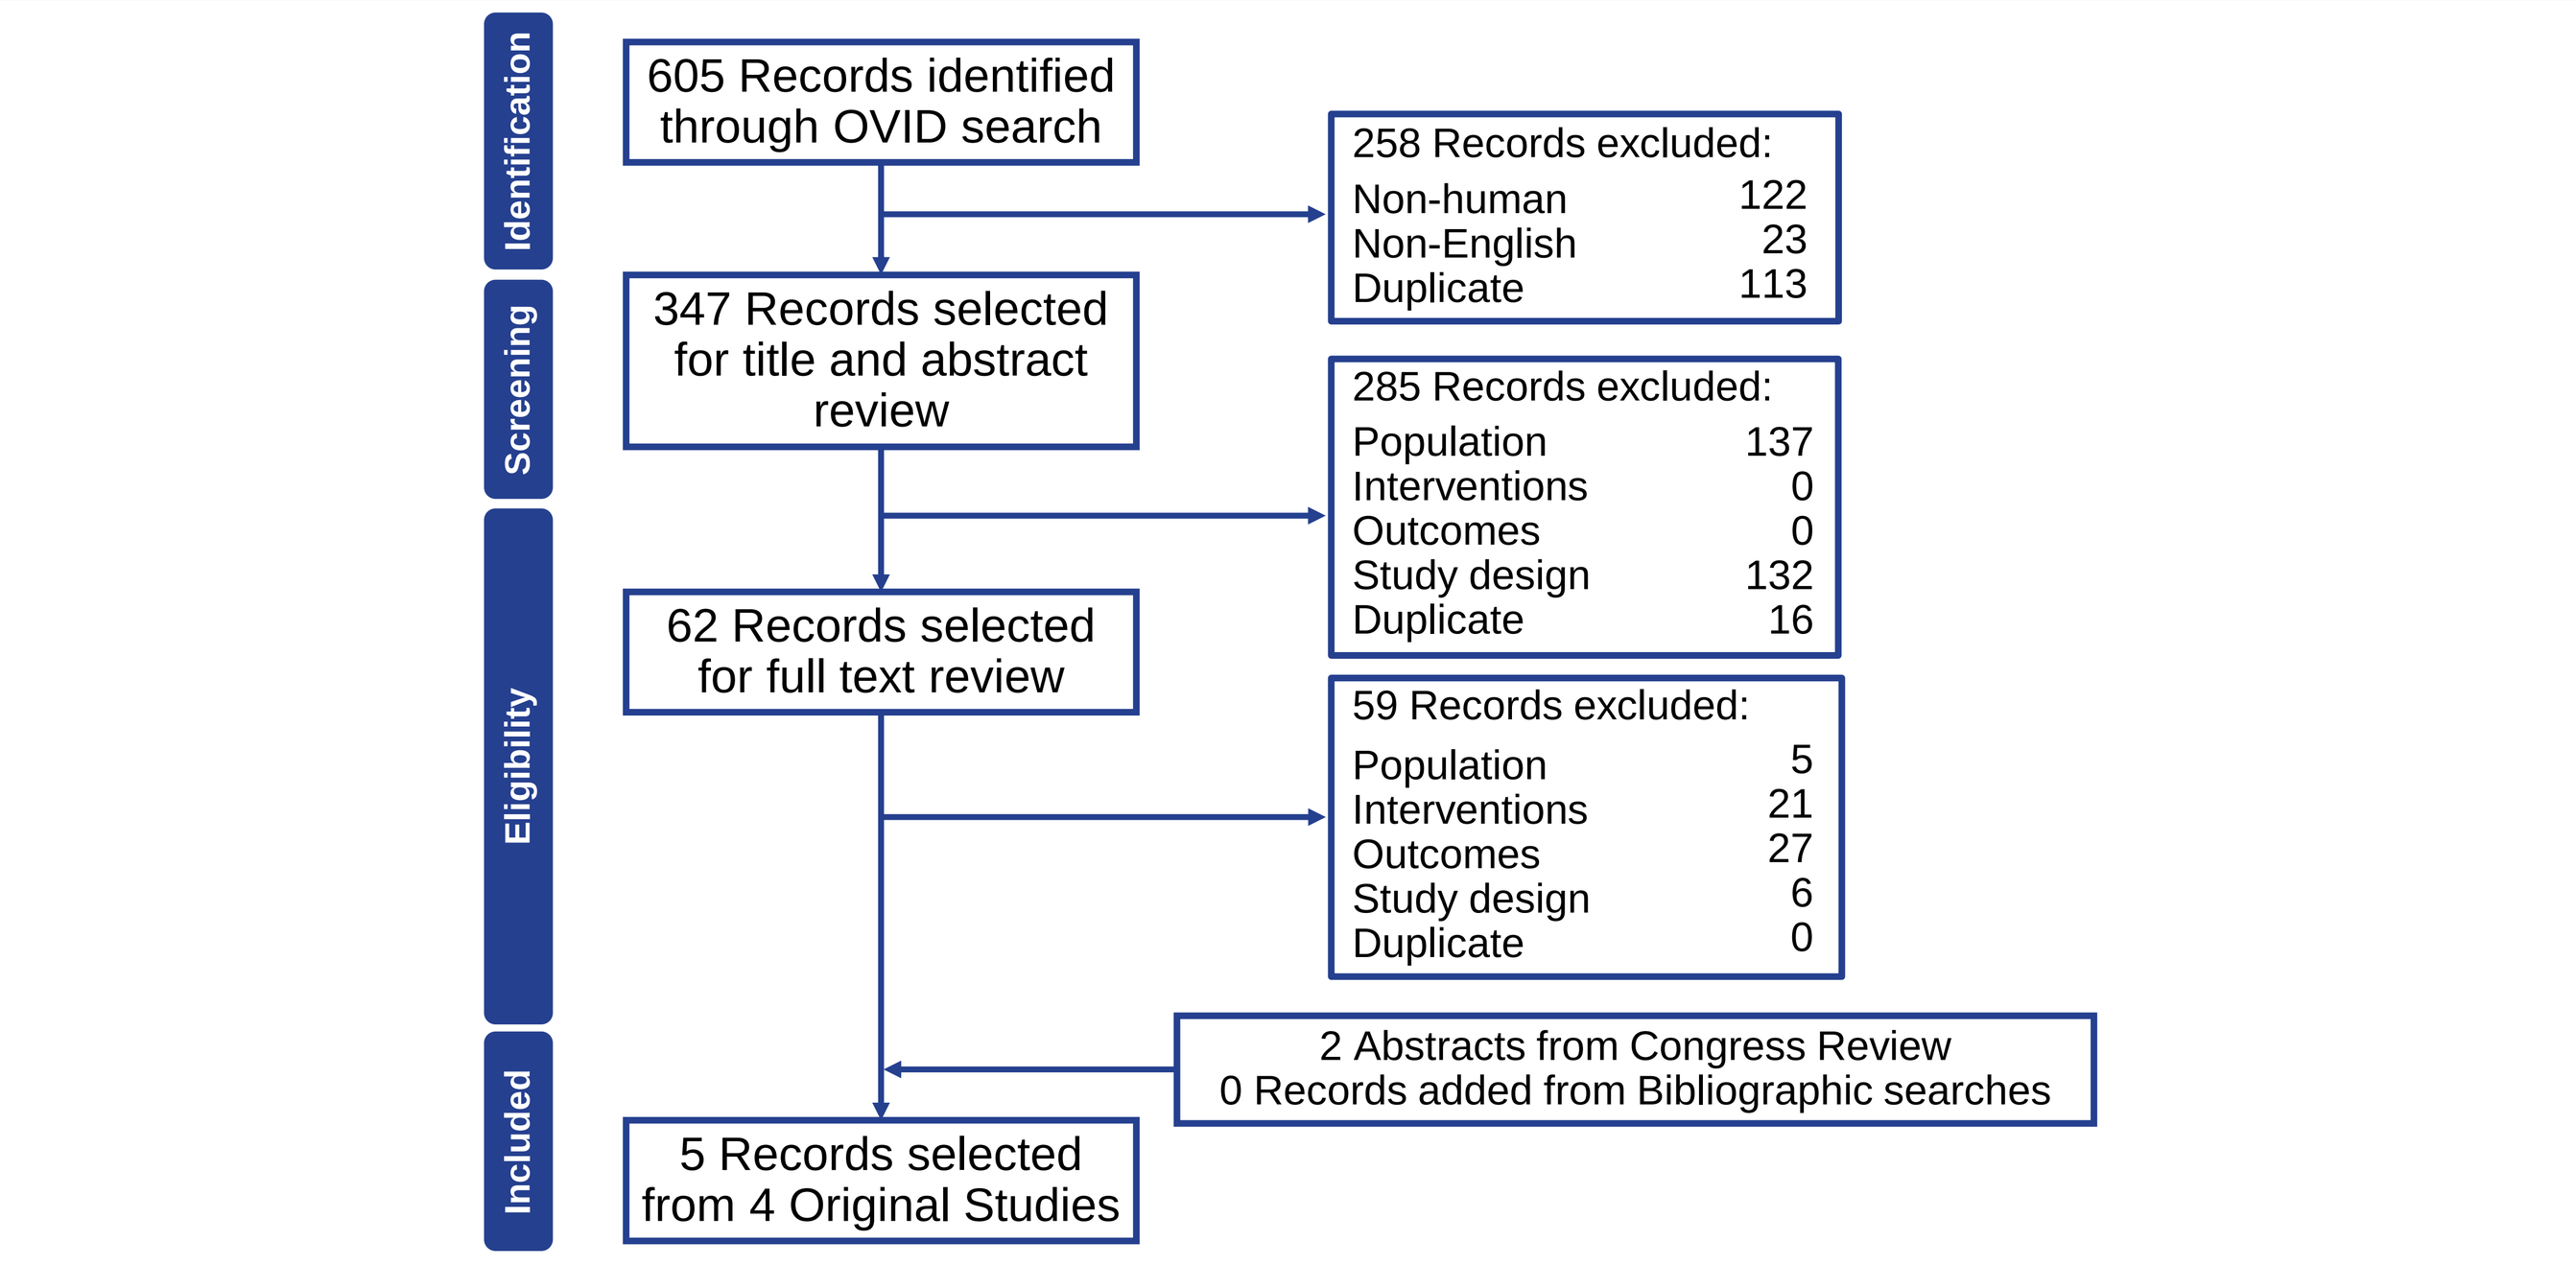
Identification
Screening
Eligibility
Included
605 Records identified through OVID search
258 Records excluded:
Non-human 122
Non-English 23
Duplicate 113
347 Records selected for title and abstract review
285 Records excluded:
Population 137
Interventions 0
Outcomes 0
Study design 132
Duplicate 16
62 Records selected for full text review
59 Records excluded:
Population 5
Interventions 21
Outcomes 27
Study design 6
Duplicate 0
2 Abstracts from Congress Review
0 Records added from Bibliographic searches
5 Records selected from 4 Original Studies

Total overall costs

- The estimated total financial and non-financial costs of LHON in the US and Canada in 2019 ranged from $84 million to $200 million (USD) and from $10 million to $42 million (CAD), respectively (**Figure 3**)[2]
- The breakdown of total costs is reported for the overall IRD population only:
 - Loss of wellbeing accounted for the largest share of total costs in both the US and Canada (**Figure 4**)[2,3]
 - Other costs included productivity losses (second largest), informal care, deadweight losses, health system (primary and secondary care, diagnostic tests, pharmaceuticals, vitamins and supplements, and medical research), and other costs (**Figure 4**)[2,3]
- Limited economic data was available for Europe:
 - A retrospective analysis of conditional reimbursement decisions by the Finnish Pharmaceutical Pricing Board on 23 medicines reported the reimbursement decision concerning a treatment for LHON fell within the annual cost range of €20,000 to €60,000 (based on list price)[4]

Figure 3. Total annual costs attributable to LHON in the US and Canada[2]

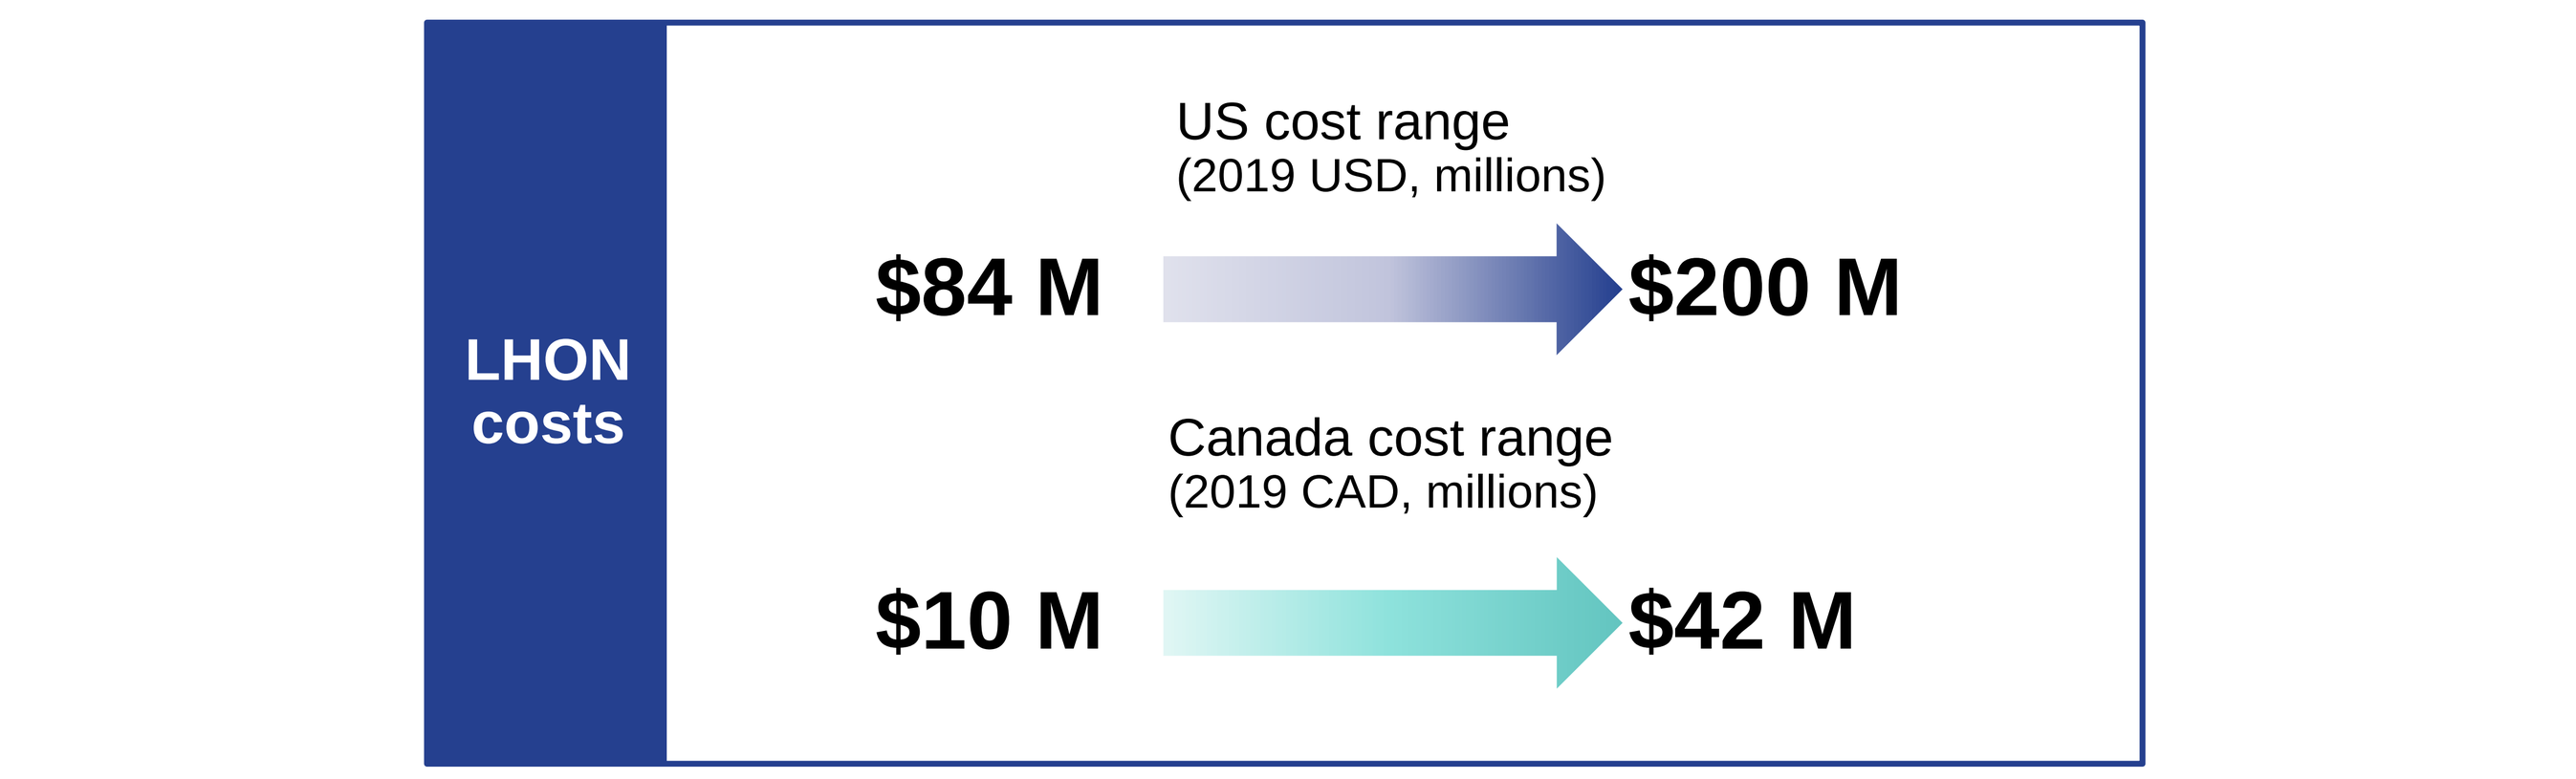
LHON costs
US cost range
(2019 USD, millions)
$84 M
$200 M
Canada cost range
(2019 CAD, millions)
$10 M
$42 M

2019 populations: US: 328.3 million people; **Canada**: 37.6 million people

Figure 4. Breakdown of total IRD costs[2,3]

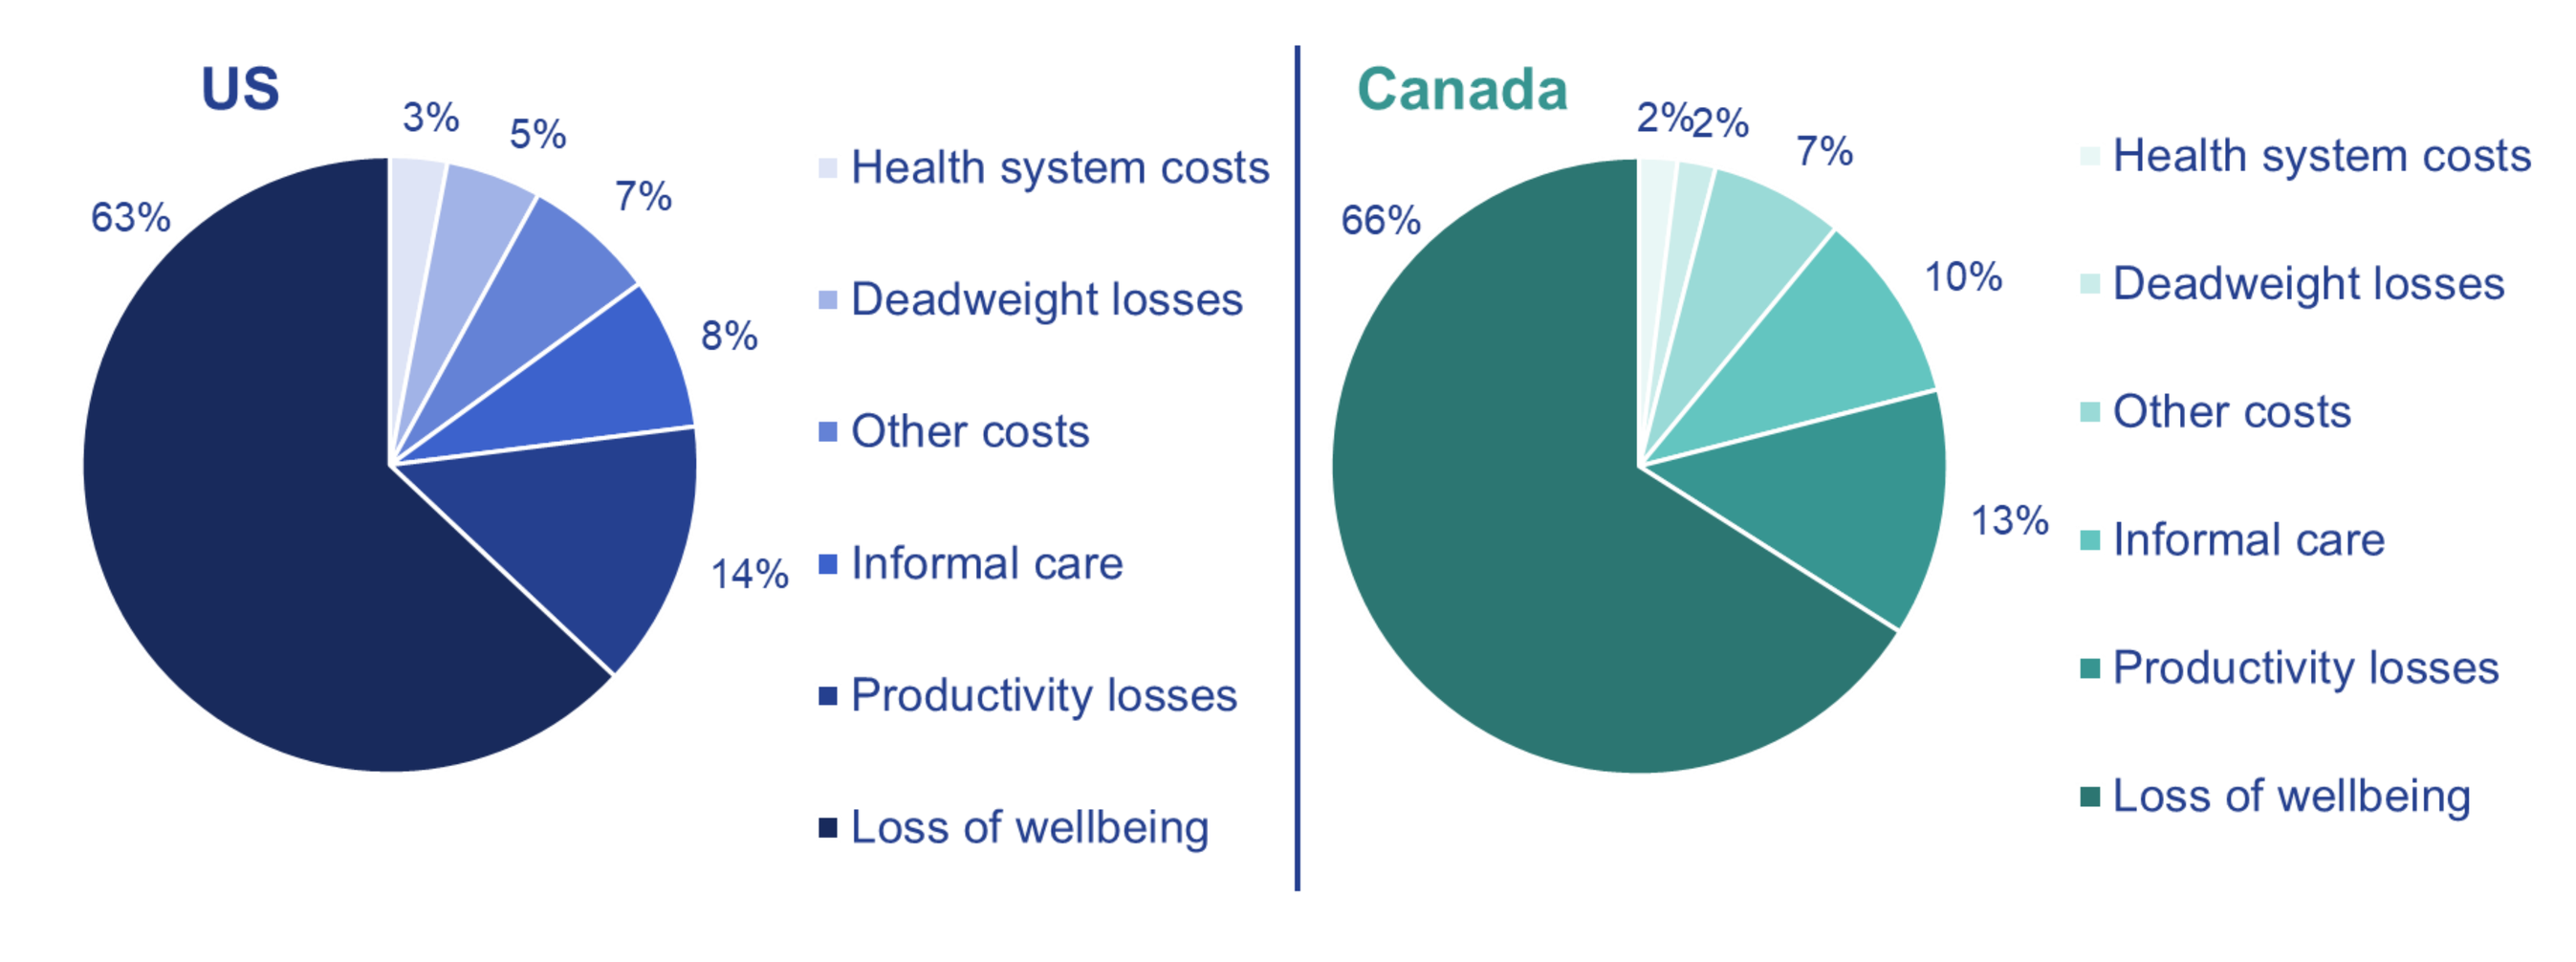
US
3%
5%
7%
8%
14%
63%
Canada
2%
2%
7%
10%
13%
66%
Health system costs
Deadweight losses
Other costs
Informal care
Productivity losses
Loss of wellbeing

Loss of wellbeing

- Loss of wellbeing, defined as reduced quality of life, was estimated using disability-adjusted life years[2]
- For patients with IRD, loss of wellbeing accounted for the largest share of total costs, ranging from $51 million to $120 million (USD) in the US and from $6 million to $26 million (CAD) in Canada (**Table 1**)[2]

Table 1. Estimated breakdown of costs for LHON based on overall IRD costs[2]

2019 Costs		US (USD, M)	Canada (CAD, M)
Financial costs	Health system costs		
	IRD (range)	963.8 to 2,216.8	37.8 to 144.3
	LHON (range)*	5.78 to 13.30	0.23 to 0.87
	Productivity costs		
	IRD (range)	1,706.2 to 4,056.0	205.2 to 839.2
	LHON (range)*	10.24 to 24.34	1.23 to 5.04
Non-financial costs	Loss of wellbeing		
	IRD (range)	8,431.7 to 20,043.6	1,071.4 to 4,381.9
	LHON (range)*	50.59 to 120.26	6.43 to 26.29
	Deadweight losses		
	IRD (range)	706.0 to 1,662.9	38.3 to 155.7
	LHON (range)*	4.24 to 9.98	0.23 to 0.93

* Costs for LHON comprised 0.6% of the estimated total costs of all IRDs in the US and Canada.[2] This proportion is used to estimate the financial and non-financial costs associated with LHON

Cost of genetic testing

- A retrospective review of a regional genetics laboratory in the UK reported that among the 107 requests for neurogenetic tests in 82 patients, 6 requests (5.6%) were for LHON[5]
- The median cost per genetic diagnostic test was £225 (range £95 to £950)[5]
- A retrospective review of an Ocular Genetics Clinic in the US reported that up to 46% of patients suspected to have a genetic condition did not undergo genetic testing; approximately 59% of these patients declined to test for financial reasons[6]

Limitations

- There was a paucity of results and the majority of articles identified were abstracts with limited reporting of study methods and results. Both these factors limited the degree to which we could compare and contrast disease and treatment costs associated with LHON
- The SLR was limited to English-language publications. While this is generally considered acceptable for HTA submission, it presents a language bias and risk of exclusion of relevant studies not published in English
- Most of the evidence was generated from a few western countries, possibly limiting insights into the global costs associated with the treatment of LHON

Conclusions

- **This SLR revealed that there are various gaps in the evidence regarding the economic burden of LHON**
- **Further, there are no published health economic models specific to LHON**
- **Despite limited available data, the indirect economic burden of LHON appears to be substantial, particularly for loss of wellbeing**
- **For comprehensive health economic evaluations that best support decision-making, a greater understanding of the true economic and humanistic burden of LHON is needed**

Funding: This study was funded by Chiesi USA Inc. Medical writing services were provided by Leah Wiltshire of Cytel, Inc.

Abbreviations: CAD, Canadian dollar; HCRU, healthcare resource utilization; ICER, incremental cost-effectiveness ratio; IRD, Inherited Retinal Disease; LHON, Leber hereditary optic neuropathy; LY, life year; M, million; PICOS, Population, Intervention, Comparator, Outcomes, Study design; PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses; QALY, quality-adjusted life year; SLR, systematic literature review; USD, US dollar

References: 1) NORD's Rare Disease Database. https://rarediseases.org/rare-diseases/leber-hereditary-optic-neuropathy/. 2) Gong J, Cheung S, Fasso-Opie A, et al. The Impact of Inherited Retinal Diseases in the United States of America (US) and Canada from a Cost-of-Illness Perspective. *Clin Ophthalmol.* 2021;15:2855-2866. 3) Daly A, Cheung S, Galvin O. Cost of illness study of inherited retinal diseases in the USA and Canada. presented at: ARVO Annual Meeting Abstract; 2021; 4) Hahl J, Kosunen M, Pudas H, Renlund M. First 27 months of conditional reimbursement in Finland. *Value in Health*. 2013;22(S721). 5) Appleton J, Nicholl D, Fulton P, Macdonald F. Ten years of neurogenetic requests: to test or not to test? presented at: Association of British Neurologists (ABN) joint meeting with the Royal College of Physicians (RCP); 2013. 6) Lowery R, Dehnel J, Schaefer G, Uwaydat S. Rates of diagnostic genetic testing in a tertiary ocular genetics clinic. *Ophthalmic Genet.* 2020 41(3):271-274.