EPH85

A Cross-Sectional Assessment of Impact of COVID-19 Vaccine on Menstrual Cycle of Pharmacy and Medical Students of Qassim University, Saudi Arabia

Maryam Farooqui[1], Kholod Al Robaian[1], Ghana Alwanin[1], Rola Alzain[1], Shoaa Almutairi[1], Majd Alsaeed[1]

[1]Department of Pharmacy Practice, Unaizah College of Pharmacy, Qassim University, Saudi Arabia

Corresponding email : KholodAlrobaian1@gmail.com

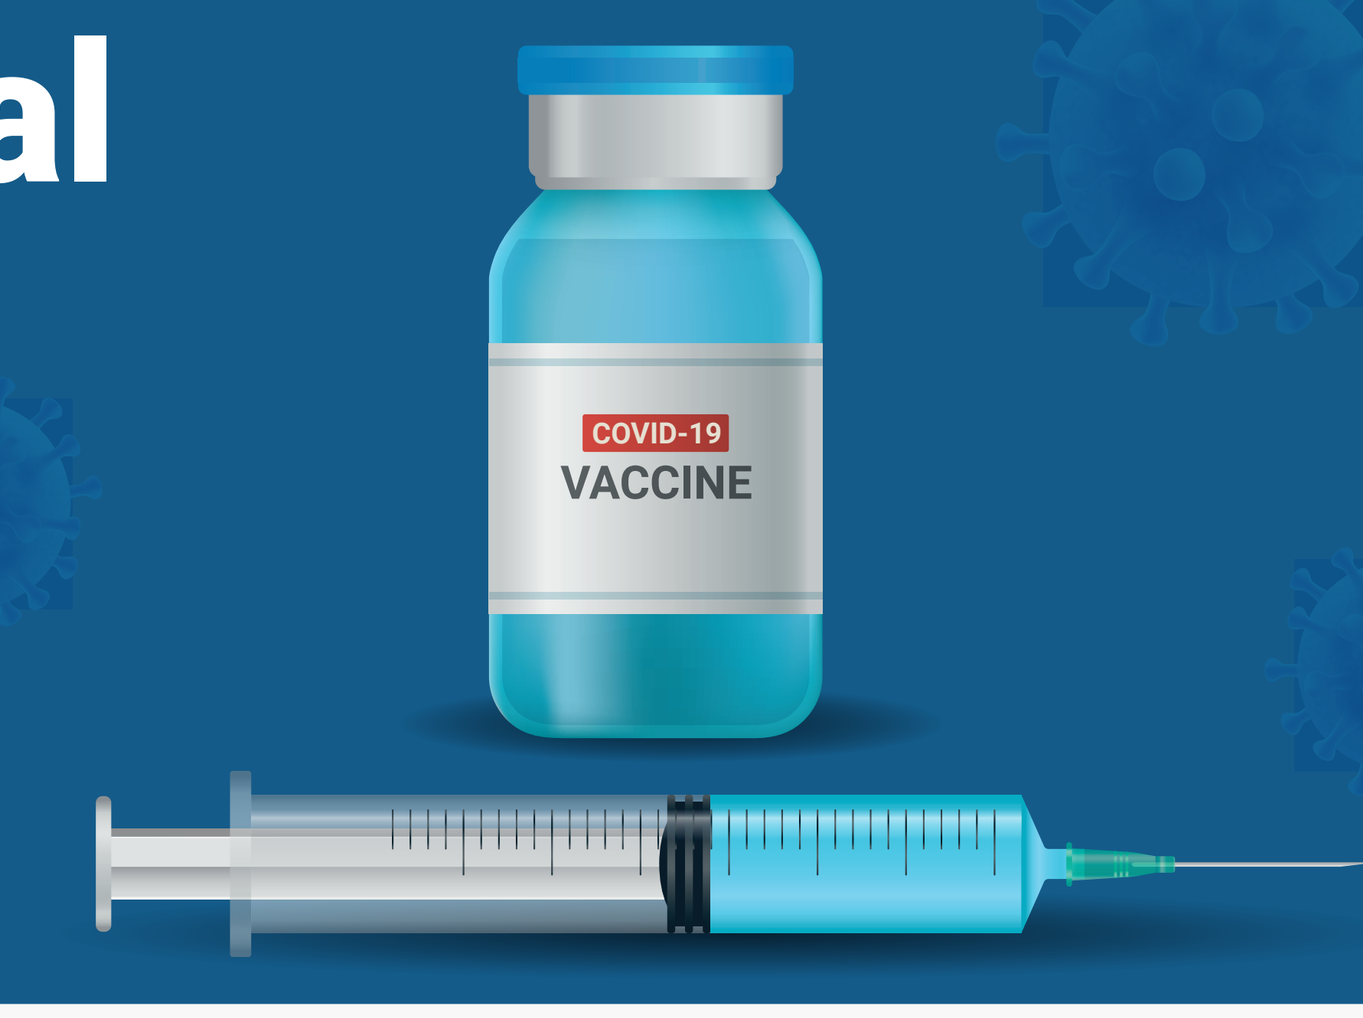
COVID-19
VACCINE

Introduction

COVID-19 pandemic has affected every area of human life, several vaccines were created and by the mid of 2021 nearly 3 billion doses were given out globally [1, 2]. Despite this, the Centers for Disease Control and Prevention (CDC) decided to deploy a real-time Vaccine Adverse Event Reporting System called VSafe to monitor any potential vaccine side effects [3]. There was a great deal of speculation that COVID-19 vaccine would have great impact on the reproductive health and may lead to menstrual abnormalities, including increased cycle length, pain, and bleeding. This emphasizes the importance of investigating menstrual abnormalities post COVID-19 vaccination [4,5].

Objectives

This study attempts to investigate the menstrual cycle abnormalities among female students receiving COVID-19 vaccines.

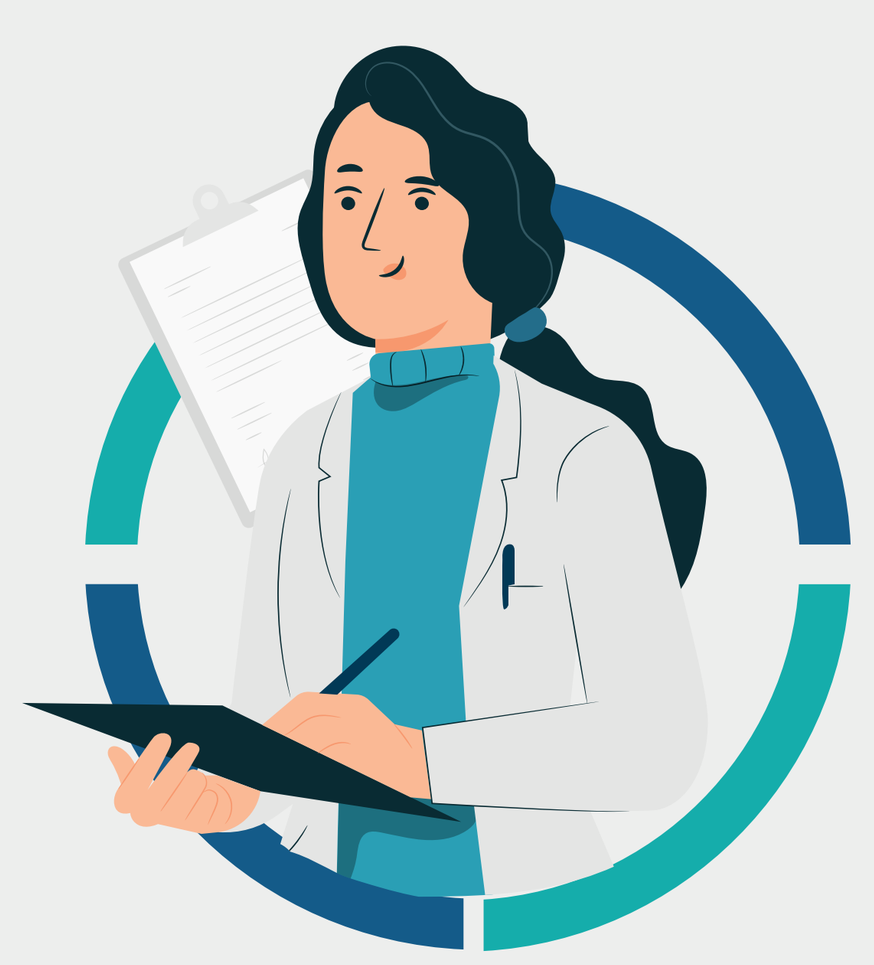

1. To determine the relationship between the COVID-19 vaccine and the menstruation.
2. To determine the impact of COVID vaccination on the menstrual cycle.

Methods

STUDY DESIGN, STUDY SAMPLE AND LOCATION:
A cross-sectional, questionnaire-based study was conducted among Females in Unaizah College of Pharmacy (UCP) and Unaizah Colleges of Medicine (UCM),

SAMPLING & SAMPLE SIZE:
A total of 299 enrolled females agreed to participate in the survey using a universal sampling method. A total of 76 Females who were using contraceptives, having history of polycystic ovary, hormonal disorders, recently delivered and menstrual irregularity or heavy bleeding were excluded from the study analysis. Prior to the participation, an explanatory statement detailing the objectives of the study was given to them and a consent was obtained.

STUDY TOOL:
To collect data, a self-administered questionnaire was adopted from a previous study {44}, after getting an official permission from the authors. Data was administered online created through Google Forms® . The questionnaire was first developed in English and was ensured for content and face validity.

STATISTICAL ANALYSIS:
All statistical analyses were performed using the Statistical Software Package for the Social Sci-ences version 23 for Windows (Chicago, IL). For descriptive statistics, we computed the means, standard deviations (SD), frequencies, and percentages. Pearson chi-square test or Fisher's exact test were used for the comparative analysis. p<0.05 was considered as significant.

ETHICAL APPROVAL:
The study was approved by Qassim ethics committee (Approval Number: 21-08-14).

Results

The menstruation abnormalities following COVID 19 vaccination

Menstruation bleeding quantity

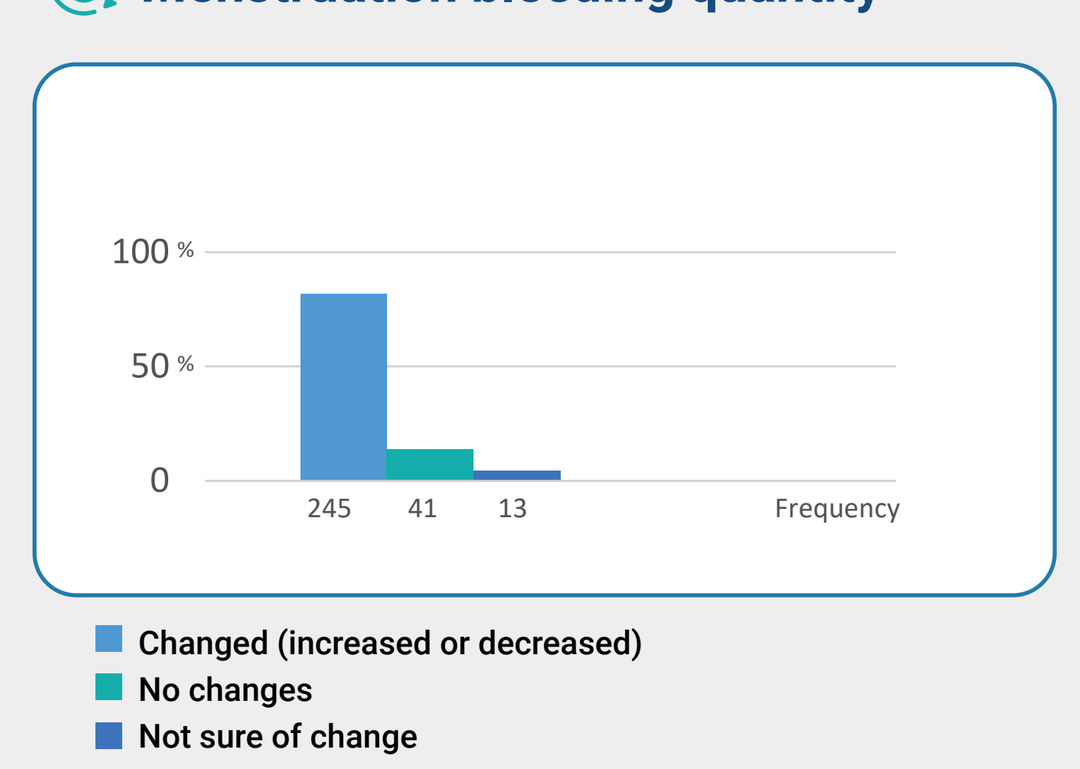
100 %
50 %
0
245
41
13
Frequency
Changed (increased or decreased)
No changes
Not sure of change

Menstruation becomes irregular

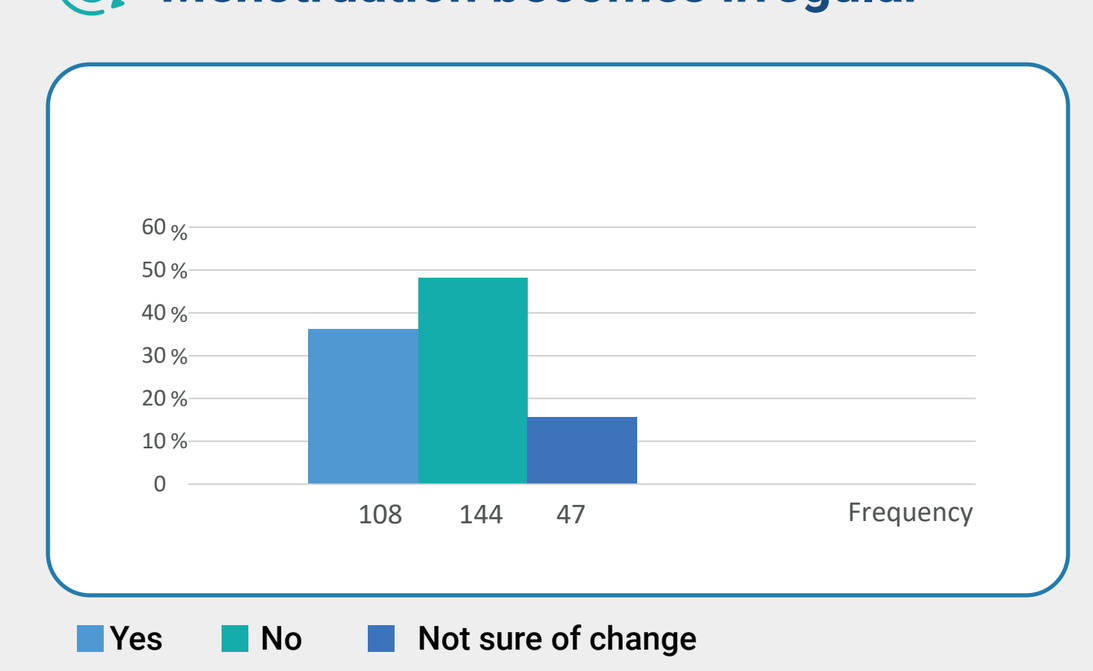
60 %
50 %
40 %
30 %
20 %
10 %
0
108
144
47
Frequency
Yes
No
Not sure of change

The length of menstruation changed

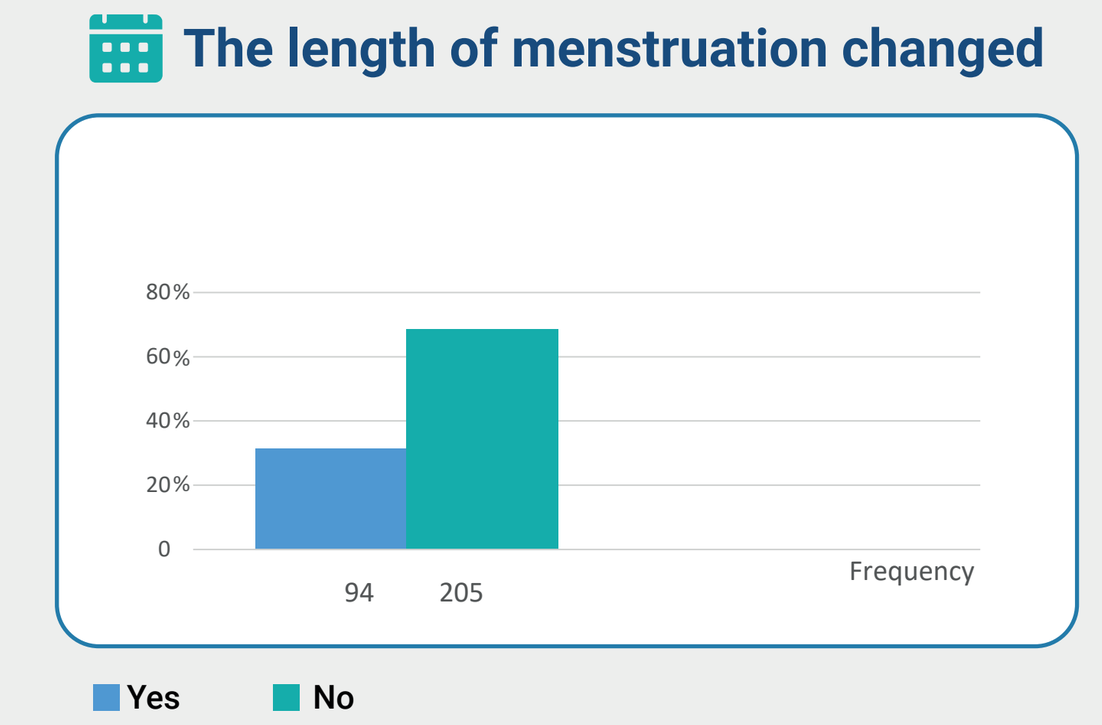
80%
60%
40%
20%
0
94
205
Frequency
Yes
No

Menstruation stopped

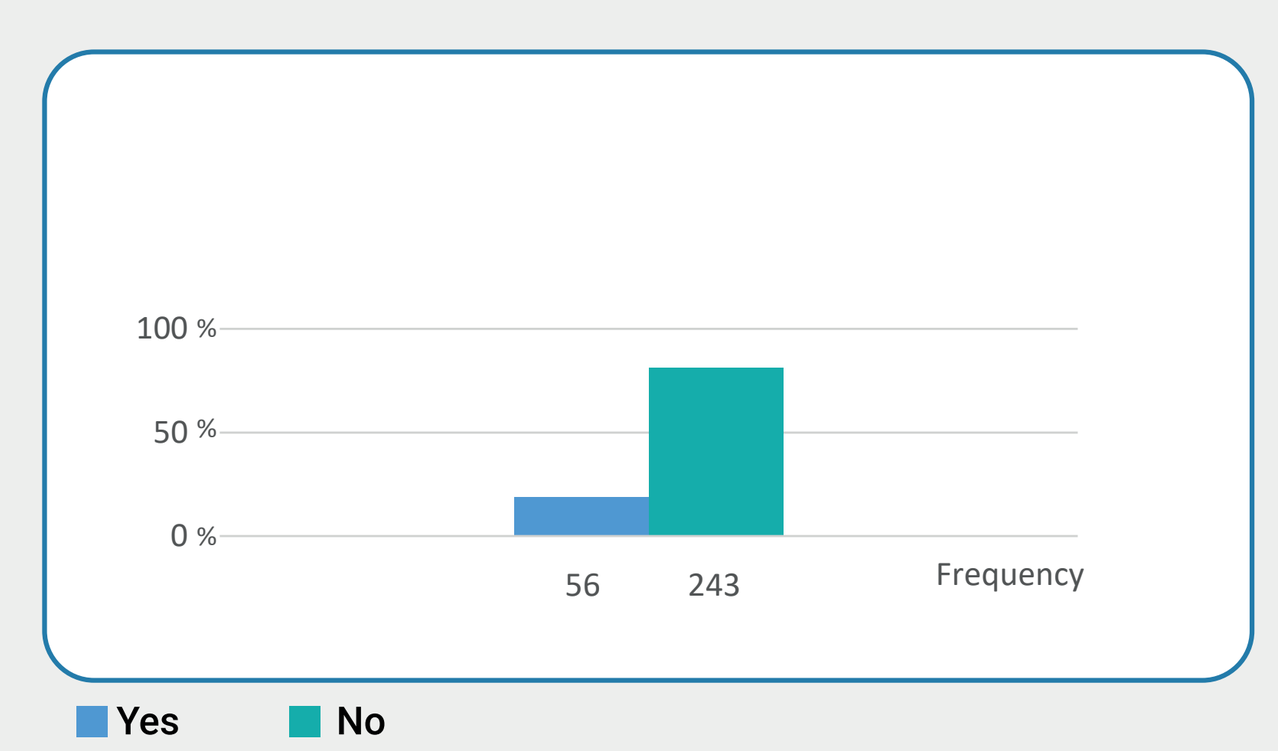
100 %
50 %
0 %
56
243
Frequency
Yes
No

Participants, who visited hospital to reduce the vaccination related side effects?

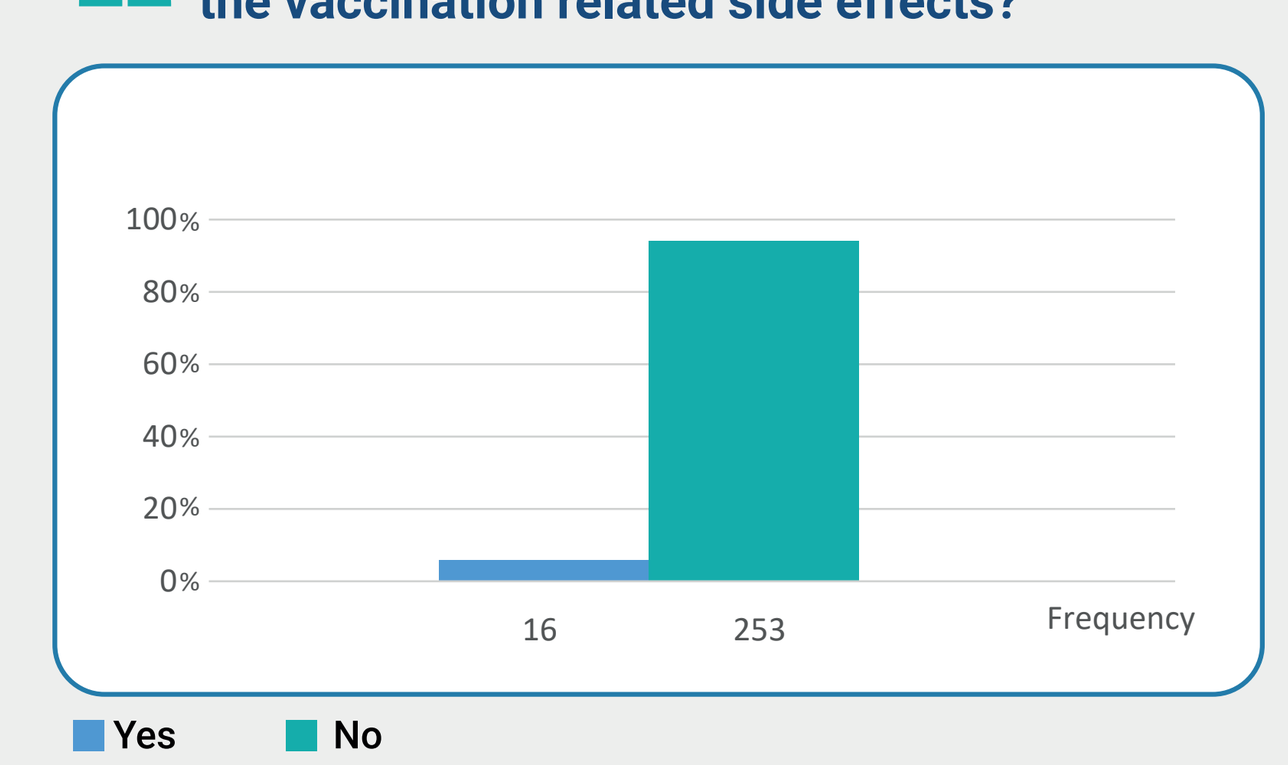
100%
80%
60%
40%
20%
0%
16
253
Frequency
Yes
No

Factors Associated with Menstrual Abnormalities After COVID-19 Vaccine

Variables	Yes (n=232)	No (n=67)	Total (299)	P-value
Age in years				
<18	5 (86.3%)	1 (16.7%)	6	0.71
18-24	198 (80.5%)	62 (19.5%)	260	
25-35	29 (87.9%)	4 (12.2%)	33	
Marital status				
Single	0 (0.0%)	255		0.92
Married	0 (0.0%)	42		
Divorced	0 (0.0%)	2		
Have you been affected by the covid-19 virus?				
Yes	70 (85.4%)	12 (14.6%)	82	0.03*
No	176 (91.7%)	16 (8.3%)	192	
Not Sure	22 (88.0%)	3 (12%)	25	
There is menstruation at the time of vaccination?				
Yes	57 (93.4%)	4 (6.6%)	61	0.44
No	181 (89.2%)	22 (10.8%)	203	
Not Sure	30 (85.7%)	5 (14.3%)	35	

* P < 0.05 (significant), the p-value was calculated using a chi-squared test or Fischer exact test.

Type of vaccine at the first dose

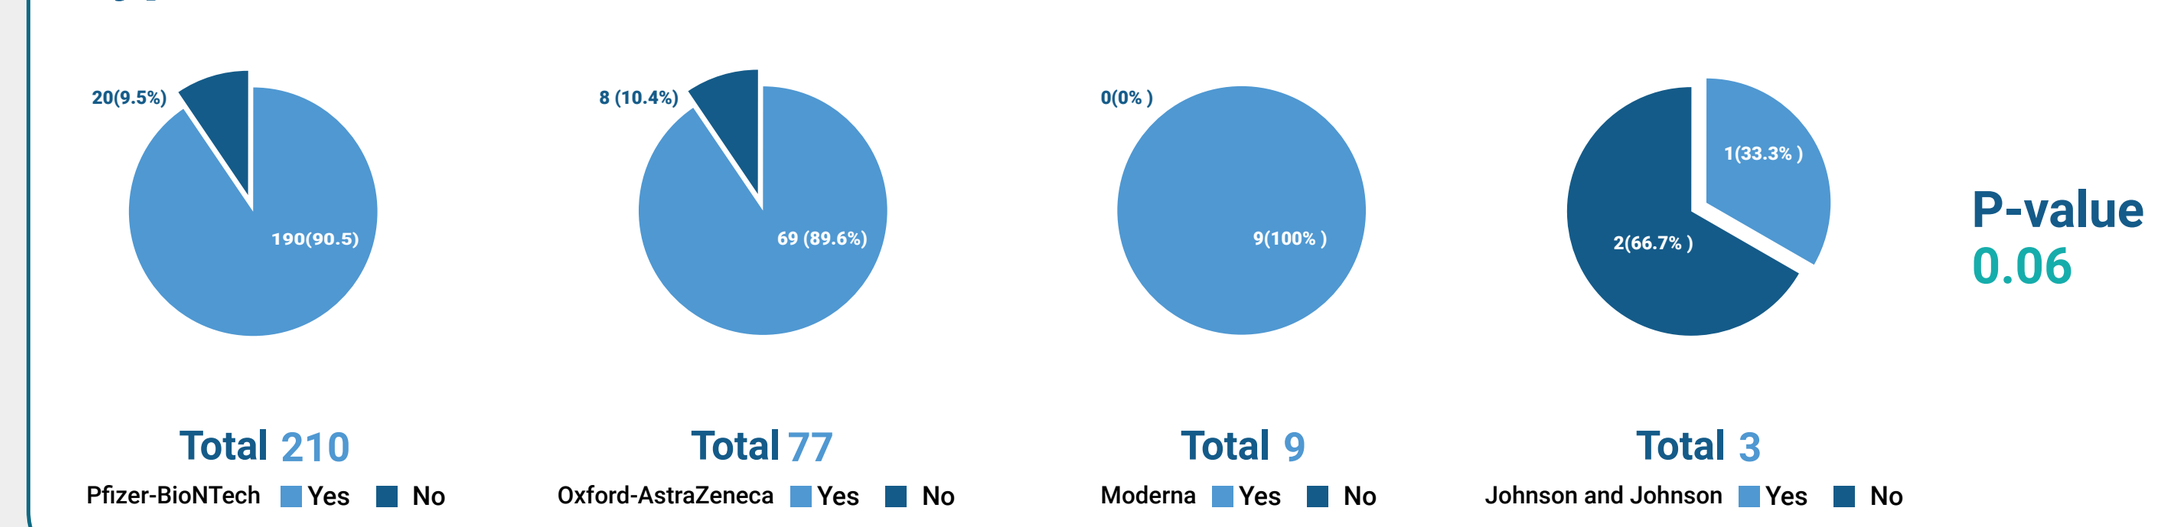
20(9.5%)
190(90.5)
Total 210
Pfizer-BioNTech Yes No
8 (10.4%)
69 (89.6%)
Total 77
Oxford-AstraZeneca Yes No
0(0%)
9(100%)
Total 9
Moderna Yes No
1(33.3%)
2(66.7%)
Total 3
Johnson and Johnson Yes No
P-value
0.06

Type of vaccine at the second dose

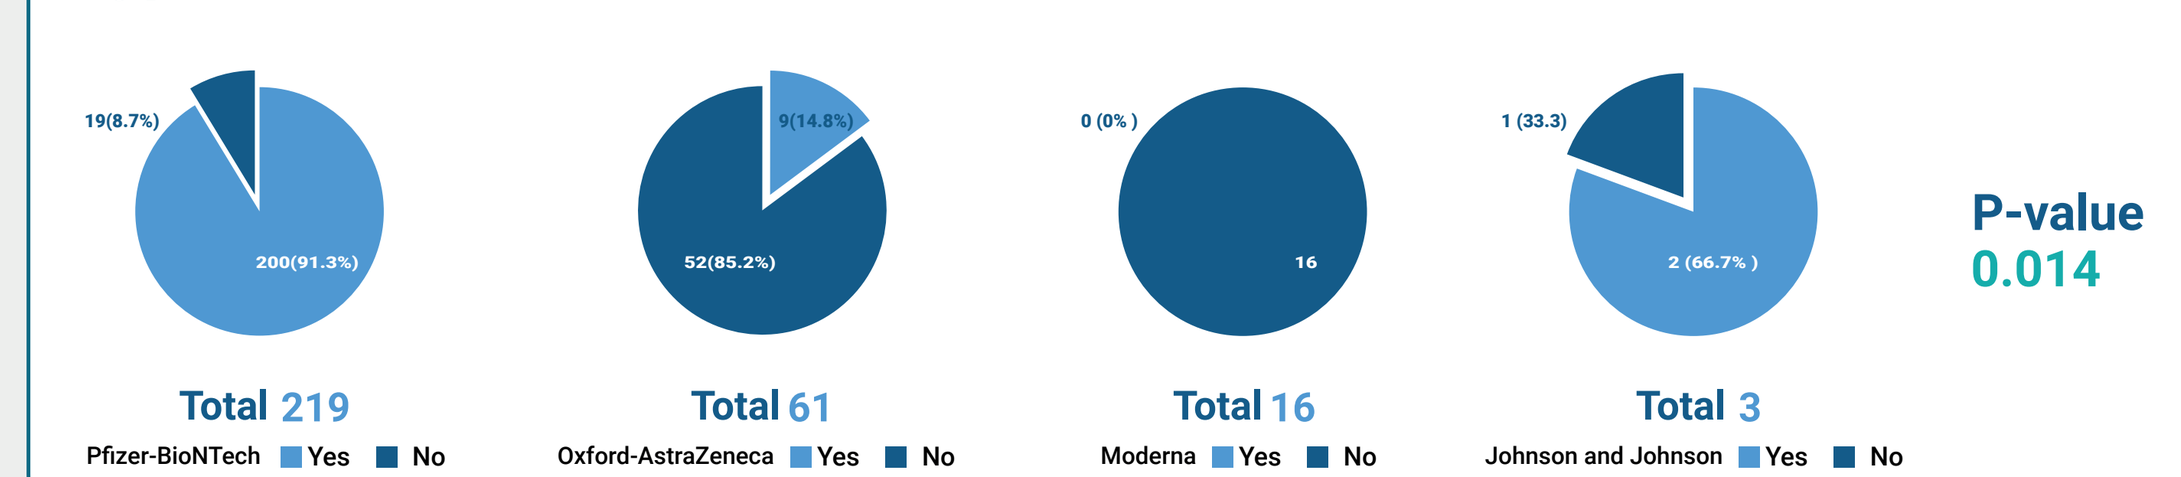
19(8.7%)
200(91.3%)
Total 219
Pfizer-BioNTech Yes No
9(14.8%)
52(85.2%)
Total 61
Oxford-AstraZeneca Yes No
0 (0%)
16
Total 16
Moderna Yes No
1 (33.3)
2 (66.7%)
Total 3
Johnson and Johnson Yes No
P-value
0.014

Receive third dose

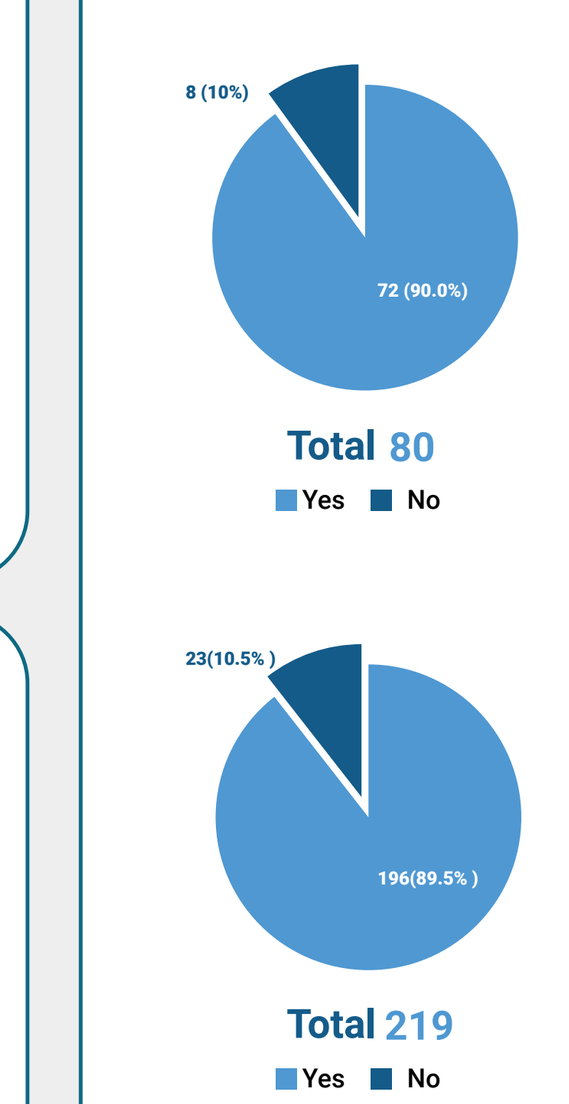
8 (10%)
72 (90.0%)
Total 80
Yes No
23(10.5%)
196(89.5%)
Total 219
Yes No

P-value
0.91

* P < 0.05 (significant), the p-value was calculated using a chi-squared test or Fischer exact test.

Conclusion

This study offers early evidence that women who get the COVID-19 vaccine may develop menstrual irregularities, such as increased menstrual cycle lengths and longer menstrual periods.These anomalies may have an effect on women's daily activities and ultimately degrade their general quality of life.Our study also offers preliminary proof that these symptoms can be temporary and self-limiting. Nevertheless, women are less likely to engage in health-seeking activities as a result of the changing priorities brought about by the COVID-19 epidemic.Therefore, it is crucial to inform women and medical professionals about menstruation problems following vaccination.To pinpoint the temporal relationship between menstrual cycle alterations and the various COVID-19 vaccination types, more prospective cohort studies are required.