EPH136

Available screening programs for the early identification of cancers: A targeted literature review

Aguiar-Ibáñez R[1]; Sharma S[2]; Tomassy J[3]; Puppala M[4]; Aktan G[5]

[1]Merck Canada Inc., Kirkland, QC, Canada; [2]Parexel International, Mohali, India; [3]Parexel International, Cracow, Poland; [4]Parexel International, Bangalore, India; [5]Merck & Co., Inc., Rahway, NJ, USA

Objectives

- To help diagnose certain cancers at an early stage and, in turn, assist to reduce the incidence of advanced/invasive cancers and subsequent mortality
- To understand the landscape of currently recommended screening programs across multiple tumors (criteria for screening, recommended frequency, and uptake) for the early identification of cancer across the following countries: Australia, Canada, France, Germany, Italy, Japan, LATAM (Argentina, Brazil, Mexico), Spain, UK, and the US

Methods

- A targeted literature review (TLR) was conducted on MEDLINE® and other publicly available sources, including up-to-date clinical guidelines (up to November 2022) to summarize currently recommended screening programs for the early detection of cancer
- The titles/abstracts of the identified literature (including relevant clinical guidelines) were first screened by a single reviewer to select potentially relevant studies, followed by full-text screening to define the final list for inclusion. At each stage, a sample of 20%-30% of the inclusion list was validated by an independent reviewer

Results

- Among the countries selected, population-based screening programs for breast (83%), cervical (83%), and colorectal cancer (83%) were the most commonly recommended across the included countries, followed by lung cancer (42%). Screening programs for gastric cancer and melanoma for general population; pancreatic and prostate cancer in high-risk population were only recommended in one country (8.3%) for each tumor type. Furthermore, no screening programs were identified for general or high-risk population for bladder, endometrial, ovarian, and renal cancer (**Table 1**)

Results

Table 1. National screening programs identified according to tumor type

Tumor type	Australia	Canada	France	Germany	Italy	Japan	LATAM	Spain	UK	US
Bladder	N	N	N	N	N	N	N	N	N	N
Breast	Y	Y	Y	Y	Y	Y	Y[c]	Y	Y	Y[a]
Cervical	Y	Y	Y	Y	Y	Y	Y[c]	Y	Y	Y
Colorectal	Y	Y	Y[a]	Y	Y	Y	Y[c]	Y	Y	Y[a]
Endometrial	N	N	N	N	N	N	N	N	N	N
Gastric	N	N	N	N	N	Y	N	N	N	N
Lung	Y[a]	N	N	Y[a]	N	Y	N	N	Y[a]	Y[a]
Melanoma	N	N	N	Y[b]	N	N	N	N	N	N
Ovarian	N	N	N	N	N	N	N	N	N	N
Pancreatic	N	N	N	N	N	N	N	N	N	Y[a]
Prostate	N	N	N	Y[a]	N	N	N	N	N	N
RCC	N	N	N	N	N	N	N	N	N	N

[a]Screening program not for general population, only for specific individuals based on their risk profile (eg, hereditary susceptibility, lifestyle risk factors).
[b]Screening program not recent; evidence lacking to show whether it is still in place.
[c]Screening program in Brazil among LATAM countries.

Breast cancer screening

- Age range and patient eligibility: 50-75 years old (40+ in US and Japan)[4]
- Screening methods:
 - Mammography[4]
 - Clinical examination[5]
- Screening frequency:

Figure 1. Proportion of countries by reported screening frequency for breast cancer[4]

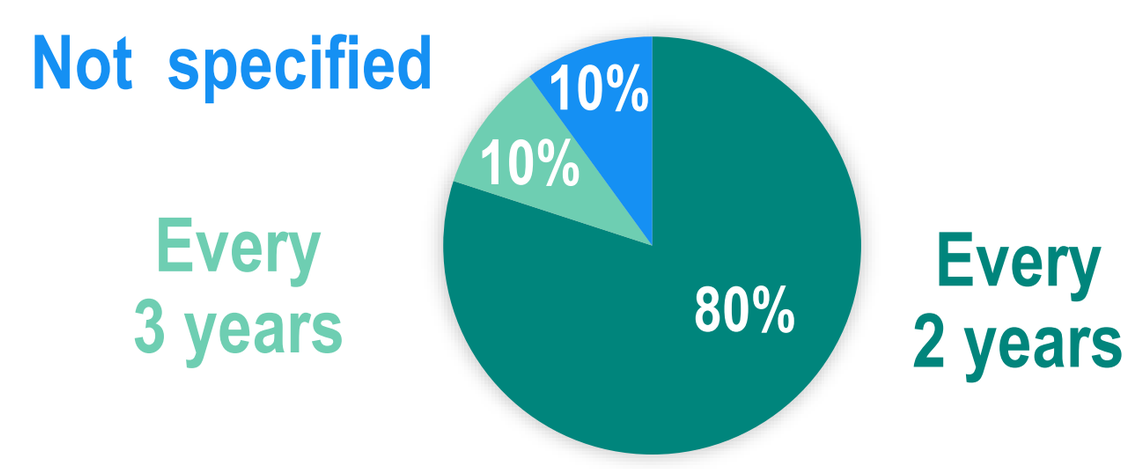
Not specified
10%
10%
Every 3 years
80%
Every 2 years

- Screening of younger patients (<50 years), or increased frequency was generally limited to high-risk patients (i.e., hereditary/genetic susceptibility, increased breast density)[6]
- Program uptake ranges (national estimates): 42.6% (France, 2021) to 55% (Australia, 2018-2019)

Cervical cancer screening

- Age range and patient eligibility: 20-79 years old; sexually active patients[4]
- Screening methods:

 Cytology test, approx. every 3 years
 +
 HPV test (secondary screening method implemented in routine testing of patients 30+ years old), approx. every 5 years
- Variations were found between cervical screening regarding frequency of testing and the timing for initiation of HPV testing among younger patients (<30 years old)
- Program uptake (national estimates): 54%-56% (Australia 2016-2017)[7]; data not identified for other countries

Colorectal cancer screening

- Age range and patient eligibility: 50-74 years old (40+ in Japan, 60+ in UK)[4]
- Screening methods:
 - Fecal immunochemical/fecal occult blood test
 - Sigmoidoscopy/colonoscopy
- Programs recommended regular fecal immunochemical/occult blood tests among the specified patient population (every 1-2 years)[4]
- Sigmoidoscopy/colonoscopy often used less regularly or as a secondary screening method following a positive test result
- Program uptake (national estimates): 28.9% (France, timeframe N/A)[8] to 43.5% (Australia 2018-2019).[7] Data not identified for 8/10 programs

Lung cancer screening

- Age range and patient eligibility: National screening program only in Japan; limited to patients (50-80 years) perceived to be at high risk (i.e., heavy smokers)[4]
- Metrics used to define "*heavy smoker*" varied between screening programs
- Screening methods:
 - CT scan/low-dose CT scan
 - Chest x-ray and Sputum cytology (Japan only)[9]
- Frequency: Not reported in 3/5 countries (Australia: every 0.25-2 years based on risk; US: annually)[10-12]
- ESMO recommends that referral to smoking cessation program should follow low-dose CT scan for lung cancer screening[13]
- Program uptake (national estimates): 5.8% (US, 2022);[14] data not identified for other countries

Gastric cancer screening

- Age range and patient eligibility: ≥50 years old[1]
- Screening methods: x-ray or endoscopy[1]
- Screening frequency: Every 2-3 years[1]
- Program uptake data not identified

Prostate cancer screening

- Age range and patient eligibility: >45 years old[2]
- Screening methods: Digital rectal examination, prostate/genital examination[2]
- Screening frequency: Annually[2]
- Program uptake data not identified

Melanoma screening

- Age range and patient eligibility: >35 years old[3]
- Screening methods: Full body visual skin examination[3]
- Screening frequency: Every 2 years[3]
- Program uptake data not identified

Conclusions

- Population-based screening programs have been implemented across countries to identify patients with breast, cervical, and colorectal cancers (who represent a large proportion of new cases). Variation exists regarding screening methods used, frequency of testing, and eligible patient populations
- The implementation of screening programs for other tumor types has been limited to specific countries (gastric, melanoma) or restricted to high-risk populations, as defined by lifestyle factors or hereditary susceptibility (i.e., lung, pancreatic, prostate). No population-based screening programs were identified for other cancer types

References
1. Fan X, et al. *Chin J Cancer Res.* 2021;33(2):168-180.
2. Bundesgesundheitsministerium.de: https://www.bundesgesundheitsministerium.de/krebsfrueherkennung.html. Accessed Nov. 8 2022.
3. Katalinic A, Eisemann N, Waldmann A. *Dtsch Ärztebl Int.* 2015;112(38):629-634.
4. Ebell MH, Thai TN, Royalty KJ. *Public Health Rev.* 2018;39:7.
5. Hamashima C, et al. *Jpn J Clin Oncol.* 2016;46(5):482-492.
6. American Society of Breast Surgeons. https://www.breastsurgeons.org/docs/statements/Position-Statement-on-Screening-Mammography.pdf. Accessed Nov. 10, 2022.
7. Australian Institute of Health and Welfare (AIHW). Cancer Screening. https://www.aihw.gov.au/reports/cancer-screening/nbcsp-monitoring-report-2021/summary. Accessed Nov. 8, 2022
8. Sante publique France: https://www.santepubliquefrance.fr/. Accessed Nov. 8, 2022.
9. Poon C, et al. *Health Policy.* 2022;Epub Jun 7. DOI: 10.1016/j.healthpol.2022.06.003.
10. Cancer Australia, Australian Government. 2020. Report on the Lung Cancer Screening Enquiry. ISBN Online: 978-1-74127-356-4.
11. Centers for Medicare & Medicaid Services. 2015. Screening for Lung Cancer with LDCT (CAG-00439N). https://www.cms.gov/medicare-coverage-database/view/ncacal-decision-memo.aspx?proposed=N&NCAId=274 Accessed Nov. 8, 2022.
12. US preventative services task force. 2021. Final Recommendation Statement: Lung Cancer Screening https://www.uspreventiveservicestaskforce.org/uspstf/recommendation/lung-cancer-screening Accessed Nov. 8 2022.
13. Postmus PE, et al. *Ann. Oncol. 2017;*28(suppl_4):iv1-21.
14. American Lung Association. State of Lung Cancer Report. 2022. https://www.lung.org/research/state-of-lung-cancer. Accessed April 11, 2023.

Presented at: ISPOR 2023; Boston, MA; May 7-10, 2023.

Copyright © 2023 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.